An analysis of stakeholders' contribution to high-WAC and low-WAC adalimumab products in 2025 under the Inflation Reduction Act in the United States

EE353

Background

- In 2022, the US government announced the Inflation Reduction Act (IRA), which included several provisions to lower prescription drug costs for Medicare patients and reduce federal government spending. In 2025, the legislation will introduce a $2,000 hard cap on out-of-pocket spending, while mandating commercial payers to cover 60% of costs from catastrophic coverage.
- In 2023, nine biosimilars of adalimumab were launched in the US. Most products were launched with slightly lower list prices, and among them, five have launched with significantly discounted versions (low-WAC, with discount rates between 55% and 86%) compared to the originator.
- In this study, our aim is to compare the cost of high-WAC and low-WAC adalimumab biosimilars for the Medicare program by estimating the amount that each stakeholder would spend in 2025.

Methods

- Costs levied on each stakeholder: the government, commercial plans, manufacturers, and beneficiaries were compared under the Medicare Part D standard eligibility group scheme. We assumed the annual cost of treatment would be about $85,000 for high-WAC products and $17,000 for low-WAC products.

Table 1. Total Amount Paid by Stakeholders in 2025

	Patient	Commercial	Manufacturer	Medicare
High WAC	$ 2,000.00	$ 51,252.54	$ 16,409.85	$ 15,831.85
Low WAC	$ 2,000.00	$ 10,215.31	$ 2,730.77	$ 2,152.77

Results

- The study results showed that in 2025, patient out-of-pocket spending for low-WAC and high-WAC drugs would be the same.
- Commercial plan's contributions would be $51,253 and $10,215 for high-WAC products and low-WAC products, respectively. Commercial plan's financial burden accounts for 59.7% of the annual cost of treatment with adalimumab.
- For manufacturers, contributions would be $16,410 for high-WAC, and $2,731 for low-WAC products.
- Medicare contributions would be $15,832 for high-WAC products, and $2,153 for low-WAC products.

Figure 1. Medicare Part D cost-sharing structure changes

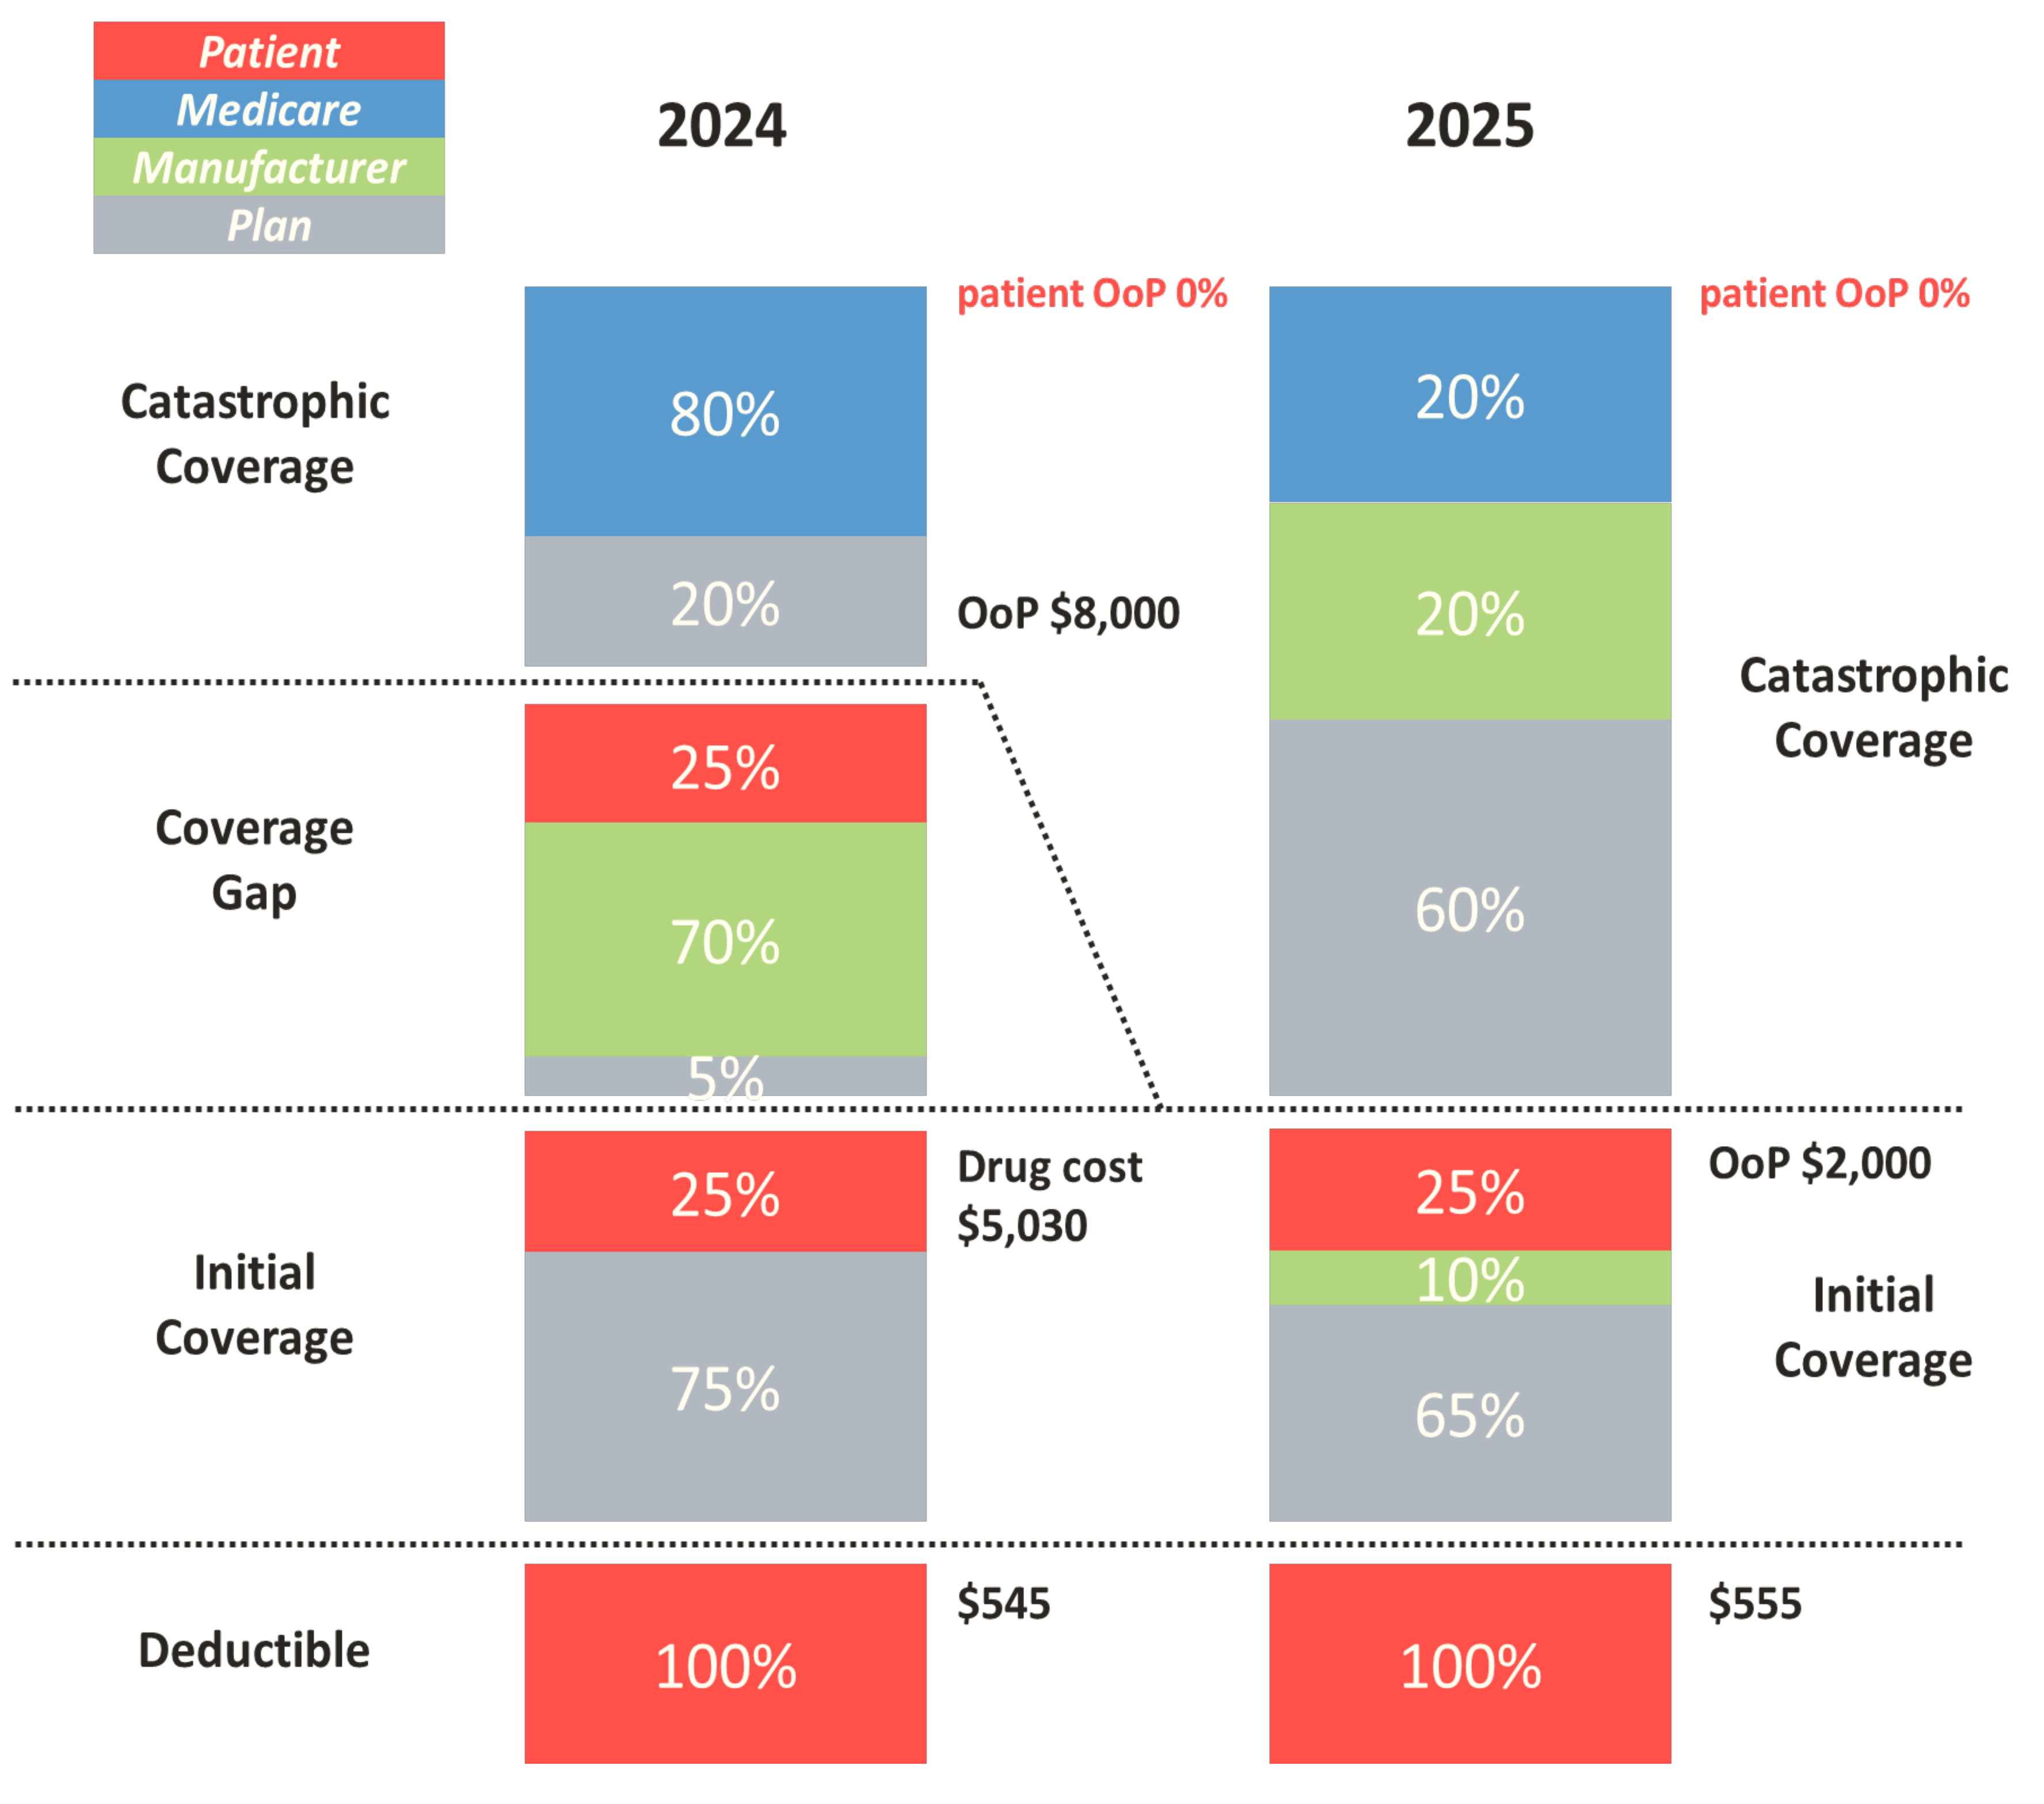

Patient
Medicare
Manufacturer
Plan
2024
2025
Catastrophic Coverage
80%
20%
patient OoP 0%
OoP $8,000
20%
20%
60%
patient OoP 0%
Catastrophic Coverage
Coverage Gap
25%
70%
5%
Initial Coverage
25%
75%
Drug cost $5,030
25%
10%
65%
OoP $2,000
Initial Coverage
Deductible
100%
$545
100%
$555

Conclusion

- Comparing each stakeholder's contribution to the Medicare program in 2025, the financial burden for Medicare, commercial plans and manufacturers is significantly reduced in low-WAC products compared to that of high-WAC products.

Conflict of Interest: *The authors are full-time employees of Celltrion Inc.*

References

* Centers for Medicare and Medicaid Services (CMS). Inflation Reduction Act and Medicare. 2023. Available at: https://www.cms.gov/inflation-reduction-act-and-medicare. Accessed Dec 2023.
* Micromedex. Red Book drug price database. 2024. Available at https://www.micromedexsolutions.com/home/dispatch. Accessed Jan 2024.