EPH167

Real-world treatment patterns, sequencing, and unmet need in metastatic colorectal cancer

Desai K, Shao C, Li S, Hair GM, Liu L, Jain R, Amonkar MM

Merck & Co., Inc., Rahway, NJ, USA

Background

- Colorectal cancer is the third most commonly occurring cancer in the United States[1]
- In 2024, it is estimated that over 150,000 new cases of colorectal cancer will be diagnosed and approximately 53,000 colorectal cancer-related deaths will occur in the United States[1]
- An estimated 20% to 25% of patients with newly diagnosed colorectal cancer already have metastatic disease at the time of diagnosis,[2-4] and approximately 70% of patients with colorectal cancer develop metastasis[4]
- The most common standard of care first-line therapy for microsatellite stable metastatic colorectal cancer (mCRC) consists of FOLFOX (folinic acid, fluorouracil, and oxaliplatin) or FOLFIRI (folinic acid and irinotecan), with or without additional targeted therapy[5]
- Real-world data of treatment pathways of mCRC in the United States are limited[6]
- To help assess the burden of disease, there is a need to understand the real-world treatment patterns and sequencing by line of therapy in the mCRC population

Methods

- This was a retrospective observational cohort analysis utilizing the Optum's de-identified Clinformatics® Data Mart Database (CDM) from January 1, 2015 to December 31, 2021
- Eligible subjects were ≥18 years of age at the time of mCRC diagnosis, had ≥2 independent diagnoses for a secondary malignant neoplasm
- Patients enrolled in a clinical trial or those with multiple primaries were excluded
- Descriptive analyses reported treatment use by line of treatment
 - A line of therapy begins on the administration/fill date of a mCRC treatment
 - All agents with an administration/fill date within 30 days of line initiation will be considered a part of the treatment regimen in that line
 - The end of the line was defined as a treatment gap of at least 120 days, the addition of a new chemotherapy agent (but not a targeted agent) at least 30 days after the start of the initial agent, or the end of continuous enrollment[7]

Results

- A total of 18,656 adults with mCRC were included in the analysis

Demographic and clinical characteristics

- More than half (55%) of the study population were male, 64% were white, and the mean (SD) age was 67 (13) years (Table 1)
- The most common comorbidities were diabetes without chronic complications (41%), mild liver disease (32%), and chronic pulmonary disease (20%) (Table 2)
- Over half of the patients had 1-2 metastases (54%), and the most prevalent site of metastasis was liver (62%), followed by lymph nodes (51%) and thorax (37%) (Table 2)

Table 1. Demographic characteristics[a]

Characteristic	Study population (N=18,656)
Sex	
Male	10,168 (54.5)
Female	8,482 (45.5)
Unknown	6 (<0.1)
Race	
White	11,919 (63.9)
Black	2,458 (13.2)
Hispanic	1,947 (10.4)
Asian	542 (2.9)
Unknown	1,790 (9.6)
Age at metastatic diagnosis date in years, mean (SD)	67 (13)
Age group	
≥65	11,493 (61.6)
<65	7,163 (38.4)
Region	
South	8,604 (46.1)
Midwest	4,471 (23.9)
West	3,247 (17.4)
East	2,259 (12.1)
Unknown	75 (0.4)
Insurance type	
Medicare	11,978 (64.2)
Commercial	6,678 (35.8)

SD, standard deviation.
[a]Values presented as N (%) unless indicated otherwise; [b]The index date is defined as the mCRC metastatic diagnosis date.

Table 2. Clinical characteristics at the metastatic diagnosis date[a]

Characteristic	Study population (N=18,656)
Comorbidity[b]	
Diabetes without chronic complications	3,195 (17.1)
Mild liver disease	2,527 (13.5)
Chronic pulmonary disease	1,607 (8.6)
Peripheral vascular disease	1,223 (6.6)
Congestive heart failure	1,214 (6.5)
Diabetes with chronic complications	1,186 (6.4)
Renal disease	1,088 (5.8)
Charlson Comorbidity Index, mean (SD)	**1.82 (1.17)**
Number of metastases	
1-2	10,124 (54.3)
3-4	5,785 (31.0)
≥5	2,747 (14.7)
Anatomical sites of metastasis[c]	
Liver	11,477 (61.5)
Lymph nodes	9,485 (50.8)
Thorax	6,892 (36.9)
Digestive system	5,130 (27.5)
Retroperitoneum and peritoneum	4,410 (23.6)
Other	6,293 (33.7)
Tumor location	
Colon	14,829 (79.5)
Rectal	3,221 (17.3)
Both	606 (3.2)
Tumor sidedness	
Left	8,844 (47.4)
Right	5,186 (27.7)
Both	281 (1.5)
Unspecified	4,345 (23.2)

CRC, colorectal cancer; IQR, interquartile range; SD, standard deviation.
[a]Values presented as N (%) unless indicated otherwise.
[b]Comorbidities present in <10% of the study population are not shown.
[c]Anatomical sites of metastasis that occur <5% are not shown. They include adrenal gland and kidney (4.5%), brain and spinal cord (4.2%), genital organs (3.2%), other urinary organs (3.0%), ovary (2.7%), skin (1.9%), other parts of the nervous system (1.4%), and breast (0.4%).

Treatment patterns

- FOLFOX-based regimens were most commonly prescribed in the first line (L1) (48%), with decreasing use (<20%) in subsequent lines of therapy (L2-L4) (Table 3, Figure 1)
- FOLFIRI-based regimens were commonly used as second line of therapy (L2) (33%)
- The use of postchemotherapy standard of care (regorafenib or TAS-102 monotherapy) was low (<20%) in any individual line of therapy, but usage of these treatments increased from the first and second lines to the third and fourth lines of therapy

Table 3. Treatments used, by line of therapy[a]

Drug class	L1 (N=11,514)	L2 (N=4,492)	L3 (N=1,491)	L4 (N=438)
Combination therapy				
FOLFOX +/- bevacizumab	5,480 (47.6)	738 (16.4)	251 (16.8)	67 (15.3)
FOLFIRI +/- bevacizumab	1,294 (11.2)	1,497 (33.3)	280 (18.8)	65 (14.8)
Other bevacizumab combinations[b]	865 (7.5)	464 (10.3)	138 (9.3)	29 (6.6)
FOLFIRINOX +/- bevacizumab	254 (2.2)	59 (1.3)	22 (1.5)	8 (1.8)
Other chemotherapy/targeted therapy combination	141 (1.2)	183 (4.1)	55 (3.7)	20 (4.6)
Regorafenib/TAS-102−based combination	35 (0.3)	40 (0.9)	54 (3.6)	26 (5.9)
Monotherapy				
Cetuximab/panitumumab-containing regimen	745 (6.5)	539 (12.0)	179 (12.0)	56 (12.8)
5-FU	691 (6.0)	110 (2.5)	27 (1.8)	10 (2.3)
Irinotecan	94 (0.8)	138 (3.1)	33 (2.2)	7 (1.6)
TAS-102	125 (1.1)	228 (5.1)	237 (15.9)	85 (19.4)
Oxaliplatin	784 (6.8)	73 (1.6)	23 (1.5)	8 (1.8)
Regorafenib	110 (1.0)	37 (0.8)	31 (2.1)	10 (2.3)
Other chemotherapy/targeted monotherapy	896 (7.8)	386 (8.6)	161 (10.8)	47 (10.7)

[a]Values presented as N (%).
[b]Excludes TAS-102 + bevacizumab combination therapy.

Figure 1. Sankey diagram of treatment sequences, by line of therapy

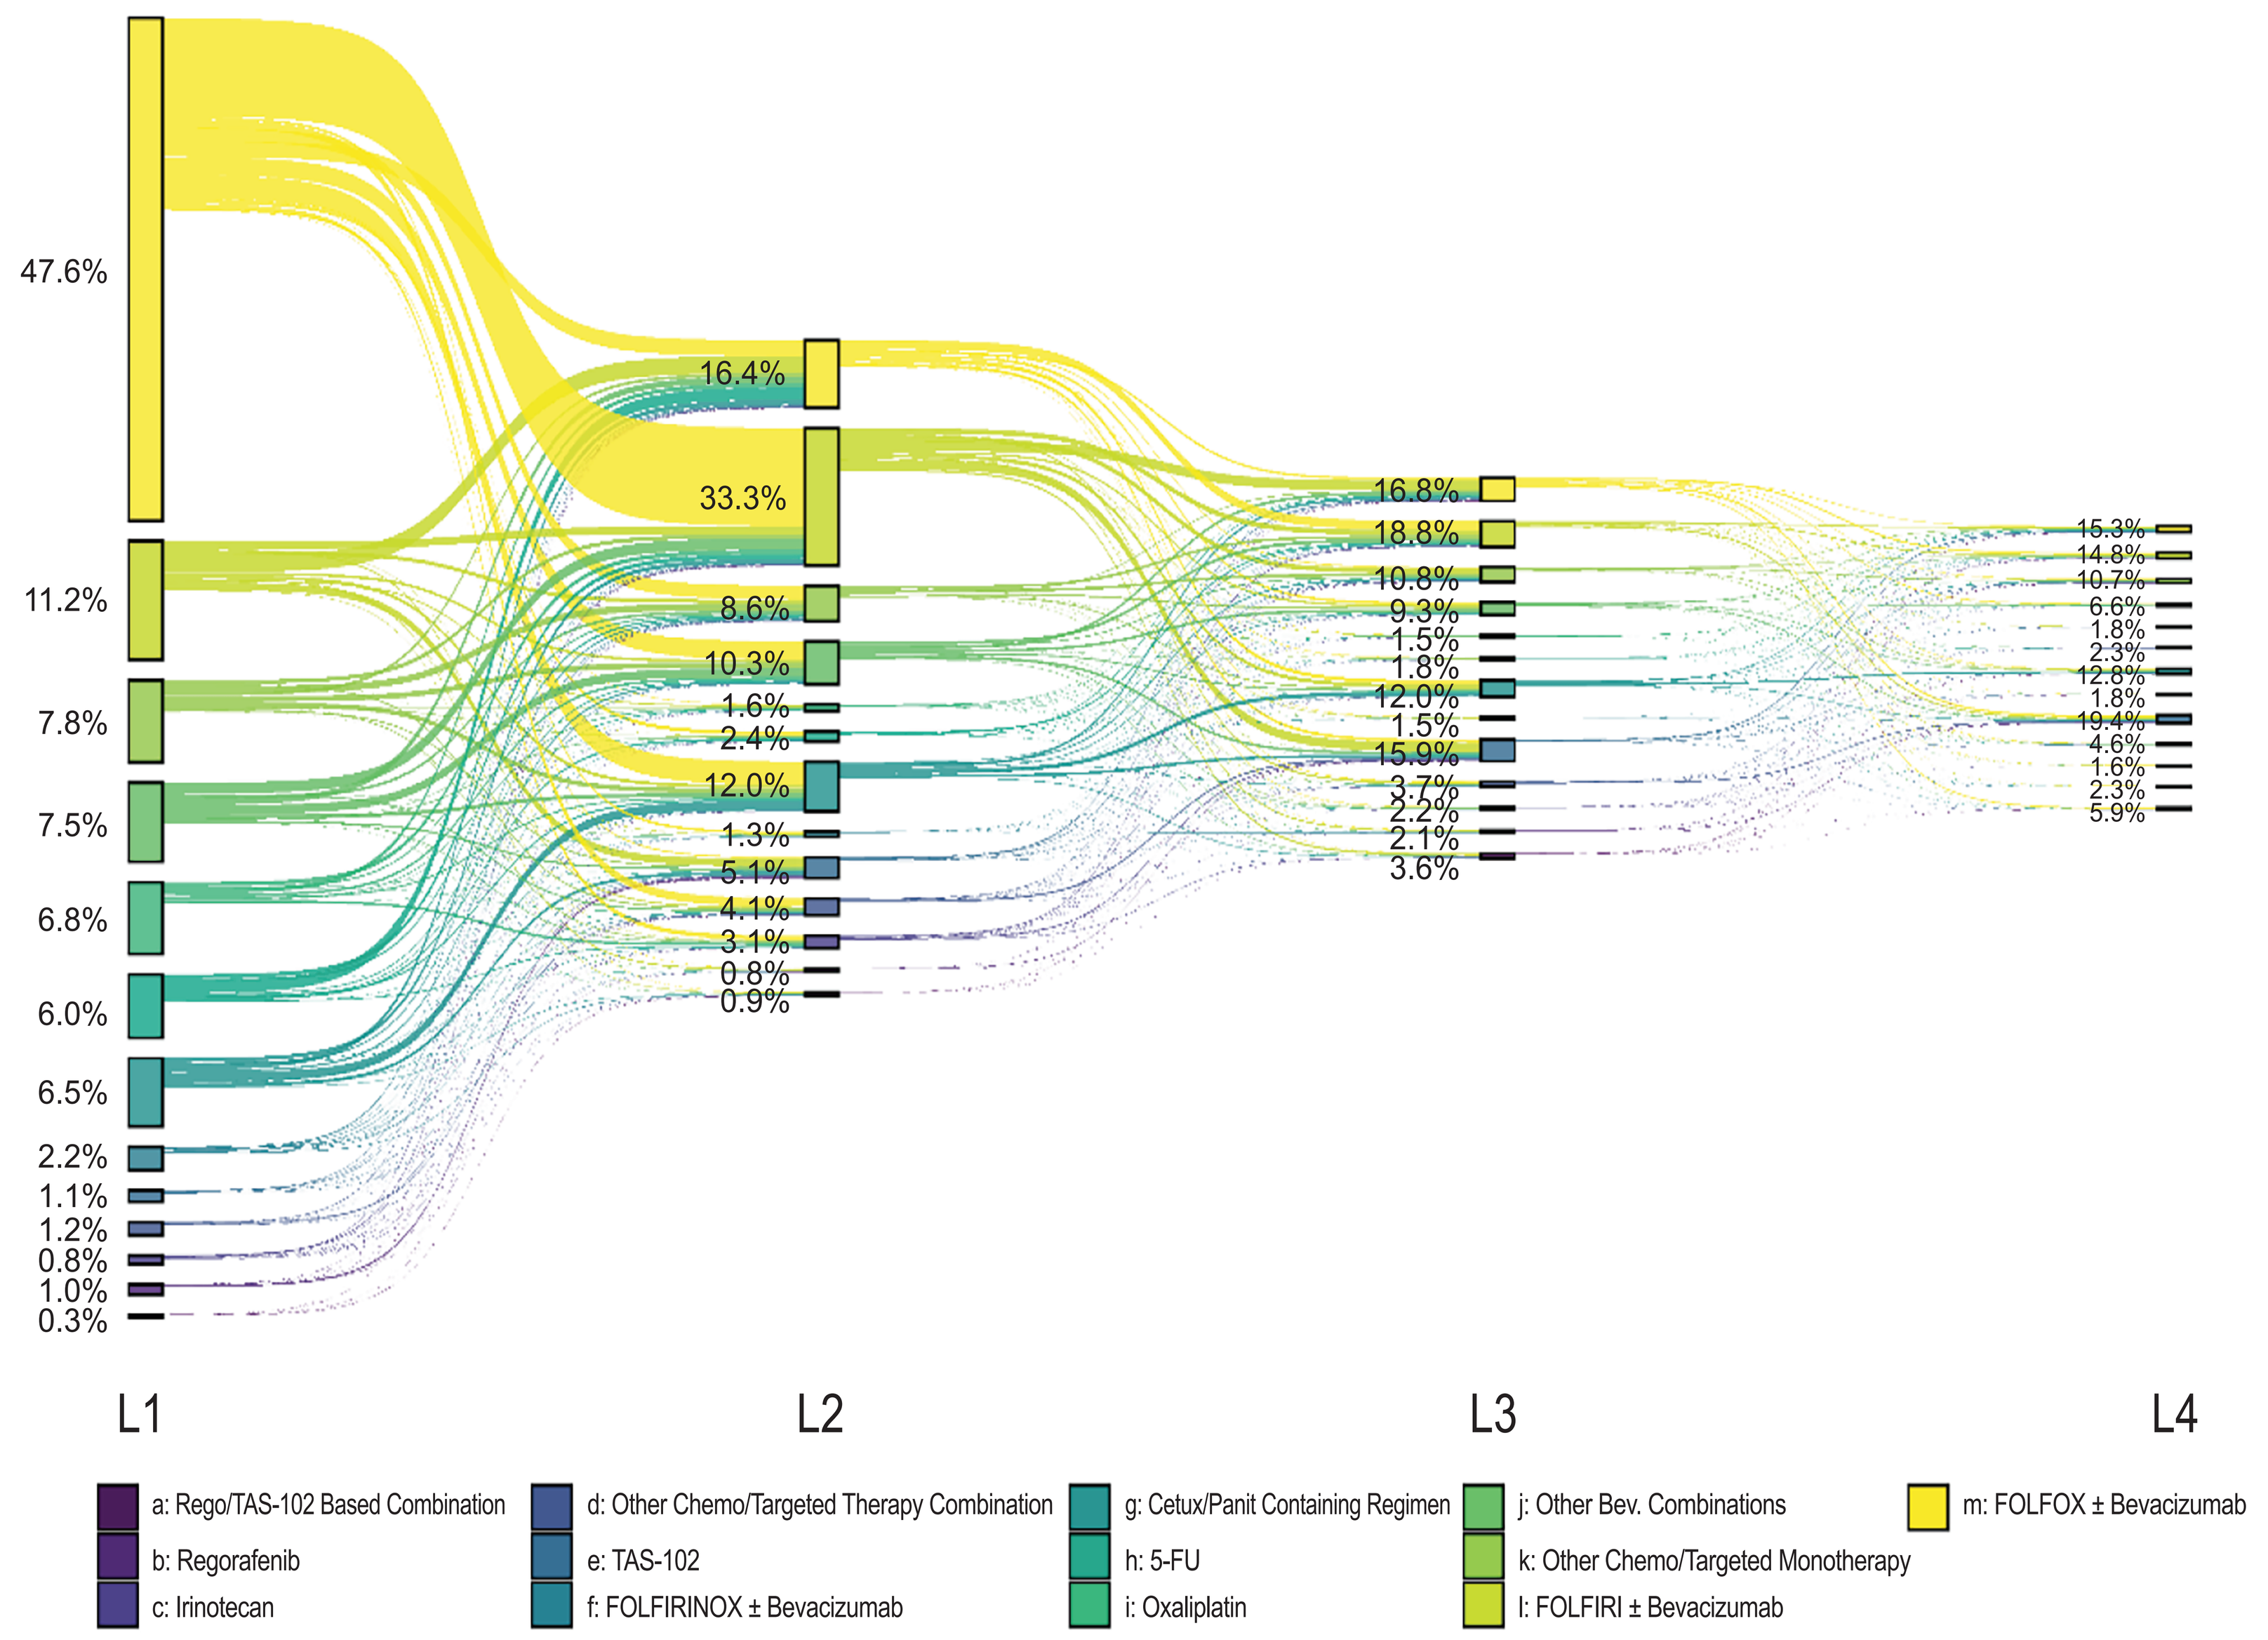
L1
L2
L3
L4
a: Rego/TAS-102 Based Combination
b: Regorafenib
c: Irinotecan
d: Other Chemo/Targeted Therapy Combination
e: TAS-102
f: FOLFIRINOX ± Bevacizumab
g: Cetux/Panit Containing Regimen
h: 5-FU
i: Oxaliplatin
j: Other Bev. Combinations
k: Other Chemo/Targeted Monotherapy
l: FOLFIRI ± Bevacizumab
m: FOLFOX ± Bevacizumab

Limitations

- Generalizability: The study included individuals with commercial and Medicare Advantage plans and therefore may not be generalizable to populations with other types or no insurance
- Documentation bias: Data-entry errors and missing data may affect the results of this analysis

Conclusions

- Real-world treatment patterns in mCRC are varied, with a wide variety of chemotherapy and targeted therapy utilized in later lines of therapy
- Regorafenib and TAS-102 monotherapy, standard-of-care options among previously treated advanced colorectal cancer patients, were not frequently used
- Minimal use of standard-of-care therapies and substantial recycling of chemotherapy combinations in the relapsed/refractory setting were observed, pointing to a high level of unmet need in this population

References

1. Siegel RL, et al. *CA Cancer J Clin.* 2024;74:(1):12-49.
2. National Cancer Institute. Cancer Stat Facts: Colorectal Cancer. Accessed March 18, 2024. https://seer.cancer.gov/statfacts/html/colorect.html
3. Elferink MA, et al. *Int J Colorectal Dis.* 2015;30:(2):205-212.
4. Amonkar MM, et al. *Cancer Treat Res Commun.* 2023;36:100712.
5. Morris VK, et al. *J Clin Oncol.* 2023;41:(3):678-700.
6. Moss B, et al. *Future Oncol.* 2023;19:(26):1809-1821.
7. Meng W, et al. *J Biomed Inform.* 2019;100:103335.

Presented at ISPOR; Atlanta, GA, USA; May 5-8, 2024.

Copyright © 2024 Merck & Co., Inc., Rahway, NJ, USA and its affiliates. All rights reserved.