CO119

Indirect treatment comparison and cost-minimization analysis between riociguat and selexipag to WHO functional class III pulmonary arterial hypertension (PAH) patients

Ricardo Saad[1]; Daniela Foli[1]; Glauco Britto[1]; Camila Roubik[2]; Rodolfo Mattar[1]; Gabriela Roncato[1]

[1] Bayer SA, São Paulo, Brazil; [2] Previously Bayer SA, São Paulo, Brazil

BACKGROUND

- Pulmonary arterial hypertension (PAH) is a pathophysiological disorder that can involve multiple clinical conditions that result in right ventricle overload and, consequently, heart failure (HF). This HF is responsible for the symptoms and limitations of the disease and is segmented into four functional classes (FC). Patients in WHO Functional Class I and II generally have low risk of mortality, in WHO FC-III, intermediate risk, and in WHO FC-IV, high risk[1,2]
- The pharmacological treatment for PAH is based on drugs from the classes of phosphodiesterase 5 inhibitors (PDE5i, e.g., sildenafil), guanylate cyclase stimulators (e.g., riociguat), prostacyclin analogs (e.g., iloprost) or agonists (e.g., selexipag), and endothelin-1 receptor antagonists (ERA, e.g., ambrisentan/bosentan)[3]
- The PAH treatment normally starts with a combination of PDE5i + ERA. However, after the follow-up, if the patient did not achieve the low risk, according to the guideline of the European Society of Cardiology and European Respiratory Society, it is possible to add selexipag to the ongoing therapy or replace the PDE5i to riociguat[1]
- The pivotal trials evaluating riociguat and selexipag for the treatment of PAH are REPLACE[4] and GRIPHON[5], respectively, however, there are no direct comparison studies between these trials. A published indirect treatment comparison (ITC) showed no statistically significant differences in efficacy between the two therapeutic approaches; however, it presented some limitations related to differences in the studies designs[6]

OBJECTIVE

- The present analysis aimed to address these limitations by conducting additional ITC scenarios, comparing riociguat + ERA versus selexipag + ERA + PDE5i. Additionally, a cost-minimization analysis (CMA) was performed from a Brazilian public health perspective

METHODS

- To further minimize limitations related to the differences between the REPLACE[4] and GRIPHON[5] studies, we conducted three additional indirect comparison analysis using the Bucher method, complementary to the analysis published by Ornstová *et al.*[6]
- To select a population more closely aligned with the treatment recommended in the Brazilian guidelines for PAH patients (triple therapy regimen with selexipag), the study by Coghlan *et al.*[7] was utilized in two of the three analyses. Coghlan *et al.*[7] reported results from the GRIPHON[5] trial of patients only on triple therapy (selexipag + ERA + PDE5i) across different WHO-FCs, including WHO-FC III. This was used for comparison with patients from the REPLACE[4] trial that included only WHO-FC III patients
- Therefore, with Coghlan *et al.*[7] data, it was possible to make a fairer comparison between the two therapies, since it was possible to select patients receiving selexipag in triple therapy and WHO-FC III combined, data which were not available in the publication of the GRIPHON[5] trial
- Similarly, to enhance the robustness of the evidence, our analyses included only patients from the REPLACE study using riociguat in dual therapy with ERA
- Through the R software, using the "*meta*" and "*metafor*" packages, with the odds ratio (OR) values and standard errors of each comparison of riociguat/selexipag versus their comparators, it was possible to calculate the indirect OR between riociguat + ERA and selexipag + ERA + PDE5i in patients with WHO-FC III receiving prior dual therapy (PDE5i + ERA)
- Since it is not possible to assert superiority in efficacy of one therapy compared to another, we conducted a cost-minimization analysis (CMA) to compare yearly costs between the two regimens based on label posology and publicly available prices

RESULTS

Scenario 1

- Scenario 1 analysis targeted WHO FC-III patients treated with riociguat + ERA vs. WHO FC-III patients treated with selexipag + ERA + PDE5i comparing “not achieving the primary endpoint” of each trial. This is, patients who did not improve in the REPLACE trial (worsened, stabilized, or died) vs. patients who worsened or died in the GRIPHON trial, excluding patients who stabilized
- For the calculation of the OR for riociguat, only patients receiving dual therapy from the REPLACE trial were included in the analysis (71.2%), all of them being WHO-FC III
- For selexipag, patients in WHO FC-III and receiving triple therapy were included (21.2%), as reported in the study published by Coghlan *et al.*[7]
- The indirect analysis using the Bucher method resulted in an OR of 0.77 (95% CI 0.33 – 1.79) for riociguat vs. selexipag, favoring riociguat numerically, but without statistical significance

Figure 1. Indirect comparison result of scenario 1 including patients who did not reach the primary endpoint of each trial

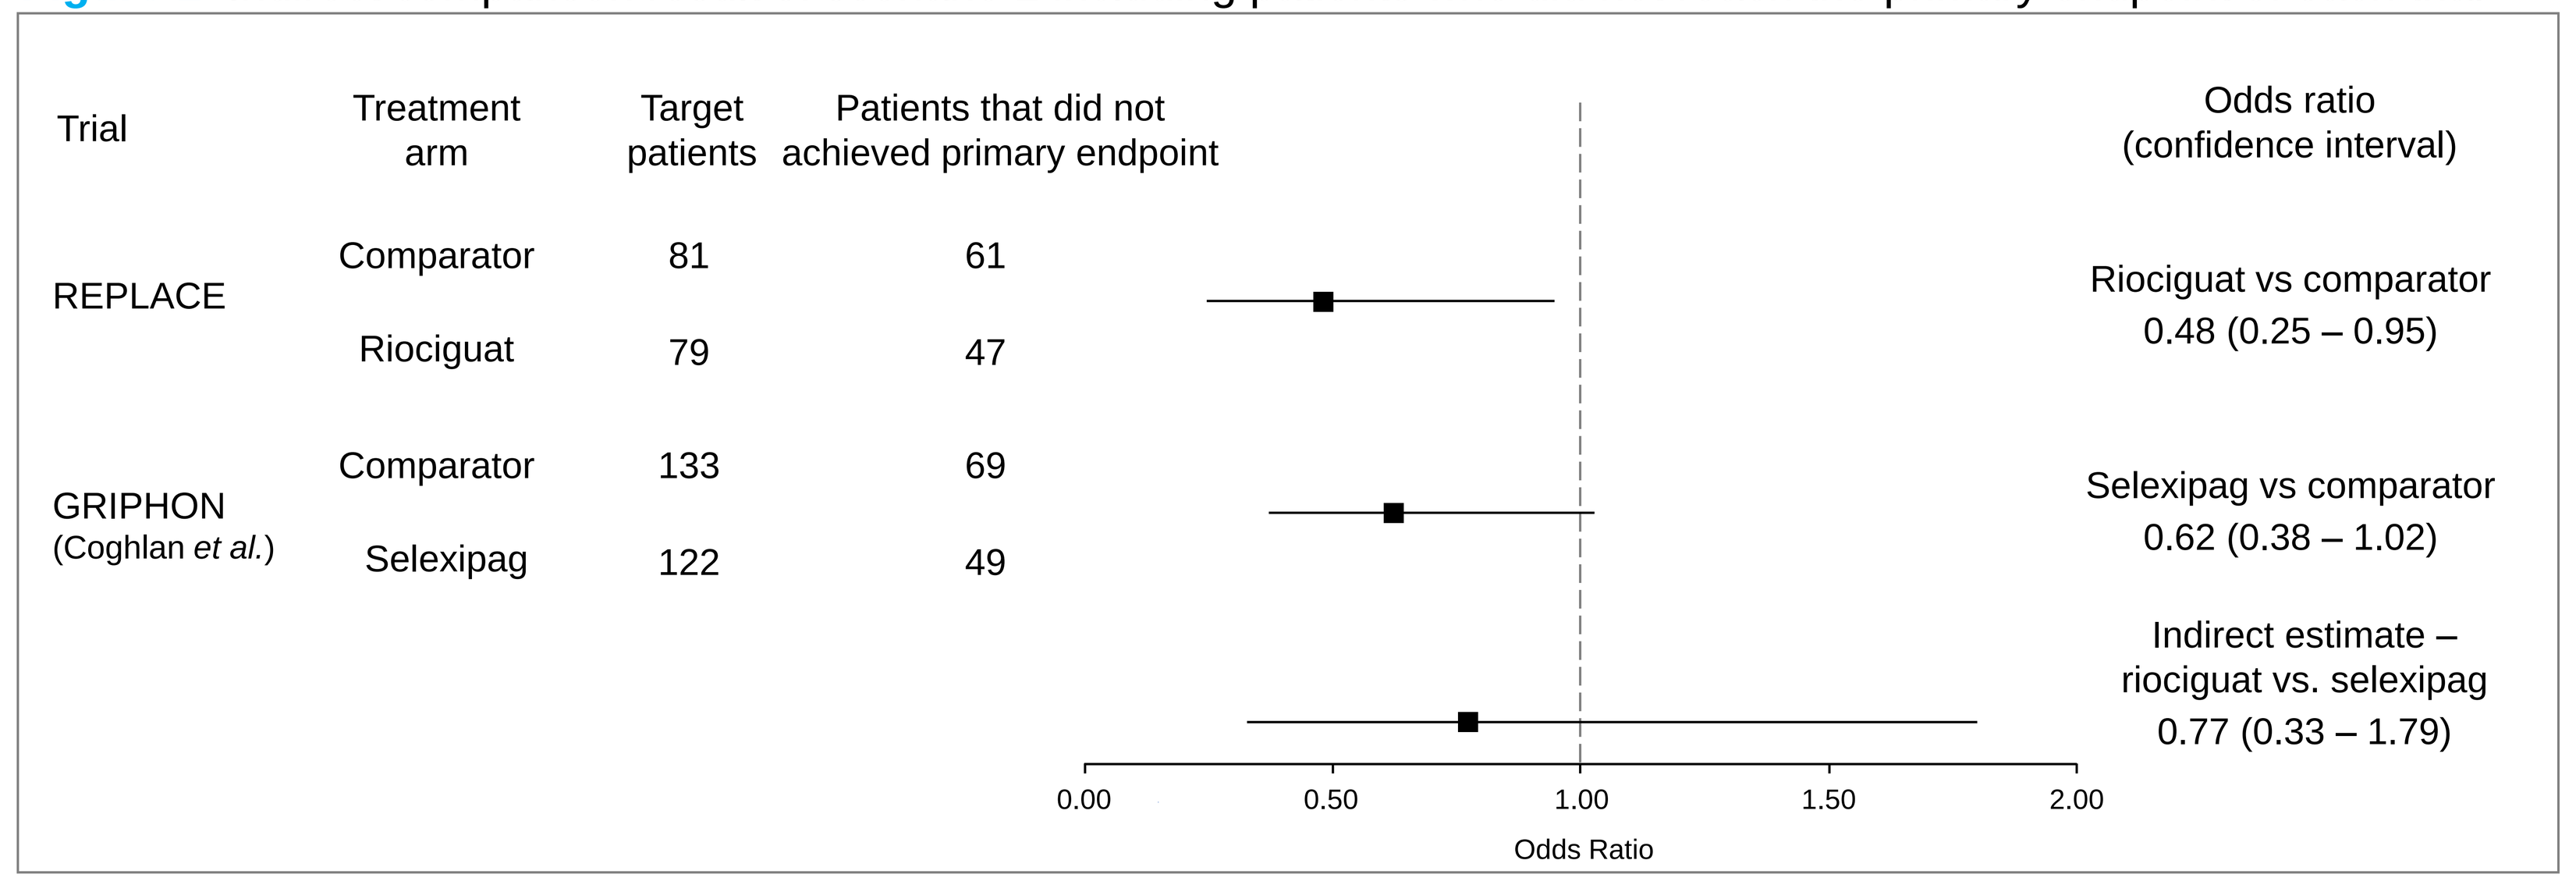
Trial
Treatment arm
Target patients
Patients that did not achieved primary endpoint
Odds ratio (confidence interval)
REPLACE
Comparator
81
61
Riociguat
79
47
Riociguat vs comparator
0.48 (0.25 – 0.95)
GRIPHON (Coghlan et al.)
Comparator
133
69
Selexipag
122
49
Selexipag vs comparator
0.62 (0.38 – 1.02)
Indirect estimate – riociguat vs. selexipag
0.77 (0.33 – 1.79)
0.00
0.50
1.00
1.50
2.00
Odds Ratio

Scenario 2

- Scenario 2 analysis targeted WHO FC-III patients treated with riociguat + ERA vs. WHO FC-III patients treated with selexipag + ERA + PDE5i comparing the morbimortality endpoint
- Target patients for riociguat and selexipag were the same as scenario 1
- However, in this analysis, only data from patients who worsened or died in the REPLACE[4] trial were used, excluding those who stabilized the disease (neither worsened nor improved), making the comparison with selexipag even fairer, given that the primary outcome of the GRIPHON[5] trial was only for patients who worsened or died (morbidity/mortality). In other words, the same outcomes were considered in this analysis
- The indirect analysis using the Bucher method resulted in an OR of 0.26 (95% CI 0.03 – 2.31) for riociguat vs. selexipag, favoring riociguat numerically, but without statistical significance

Figure 2. Indirect comparison result of scenario 2 including patients who worsened or died in each trial

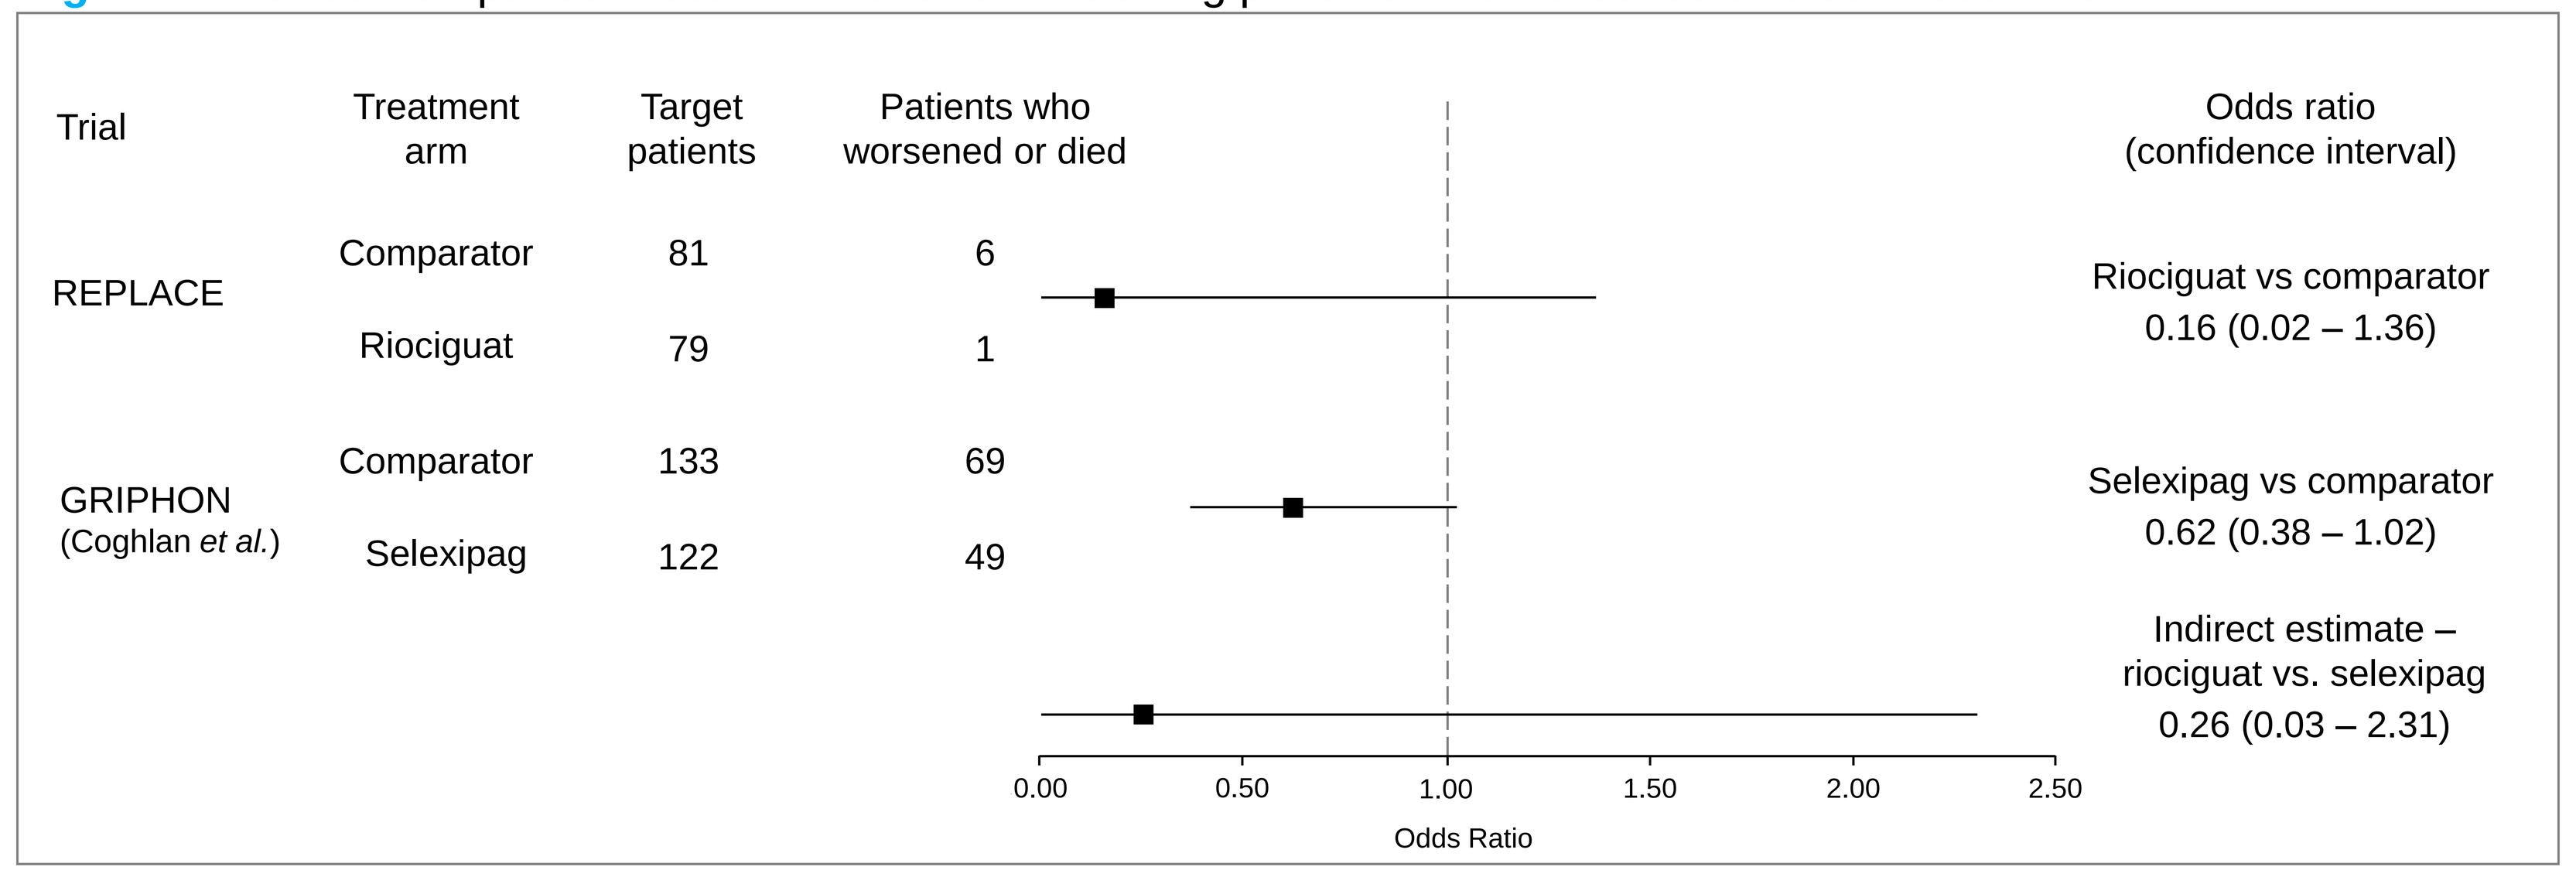
Trial
Treatment arm
Target patients
Patients who worsened or died
Odds ratio (confidence interval)
REPLACE
Comparator
81
6
Riociguat
79
1
Riociguat vs comparator
0.16 (0.02 – 1.36)
GRIPHON (Coghlan et al.)
Comparator
133
69
Selexipag
122
49
Selexipag vs comparator
0.62 (0.38 – 1.02)
Indirect estimate – riociguat vs. selexipag
0.26 (0.03 – 2.31)
0.00
0.50
1.00
1.50
2.00
2.50
Odds Ratio

Scenario 3

- Scenario 3 analysis targeted WHO FC-III patients treated with riociguat + ERA vs. any WHO FC patients treated with selexipag + ERA + PDE5i considering “not achieving the primary endpoint” of each trial
- Once more, only patients receiving dual therapy from the REPLACE[4] trial were included in the analysis (71,2%), all of them in WHO-FC III
- For selexipag, in a more conservative perspective, we considered the available subgroup data from the GRIPHON[4] trial, which consisted of patients on triple therapy, regardless of WHO FC.
- The indirect analysis using the Bucher method resulted in an OR of 0.93 (95% CI 0.41 – 2.07) for riociguat vs. selexipag, favoring riociguat numerically, but without statistical significance

Figure 3. Indirect comparison result of scenario 3 including patients who did not reach the primary endpoint of each trial

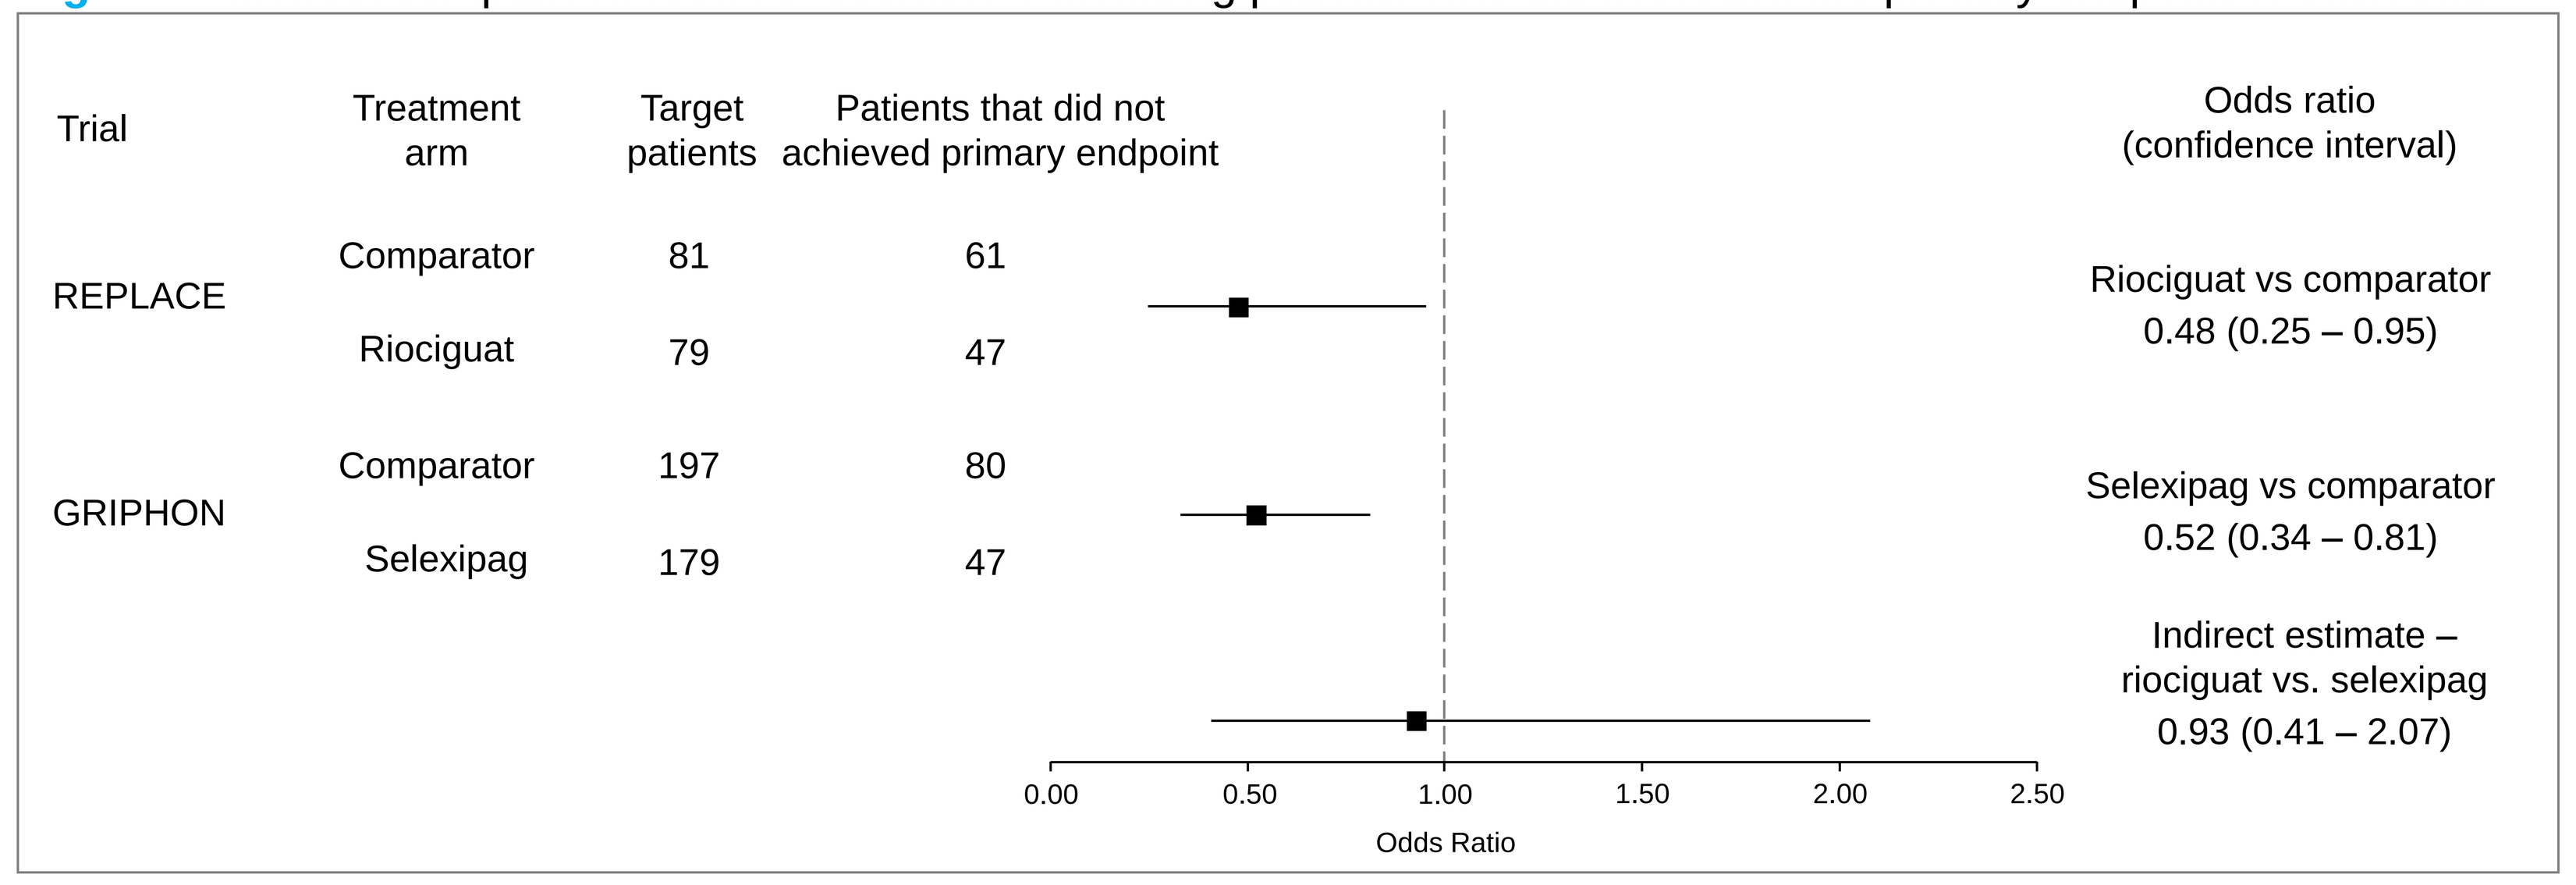
Trial
Treatment arm
Target patients
Patients that did not achieved primary endpoint
Odds ratio (confidence interval)
REPLACE
Comparator
81
61
Riociguat
79
47
Riociguat vs comparator
0.48 (0.25 – 0.95)
GRIPHON
Comparator
197
80
Selexipag
179
47
Selexipag vs comparator
0.52 (0.34 – 0.81)
Indirect estimate – riociguat vs. selexipag
0.93 (0.41 – 2.07)
0.00
0.50
1.00
1.50
2.00
2.50
Odds Ratio

- Table 1 summarizes the results of all scenarios analyses

Table 1. Results of the scenarios

Scenario	Endpoint	Patients REPLACE	Patients GRIPHON	Source	Riociguat vs. Selexipag (OR [95% IC])
1	Patients who did not achieve the primary endpoint.	WHO-FC III on dual therapy.	WHO-FC III on triple therapy.	REPLACE[4] and Coghlan *et al.*[7]	0.77 (0.33 – 1.79)
2	Patients with the endpoint of worsening or death.	WHO-FC III on dual therapy.	WHO-FC III on triple therapy.	REPLACE[4] and Coghlan *et al.*[7]	0.26 (0.03 – 2.31)
3	Patients who did not achieve the primary endpoint.	WHO-FC III on dual therapy.	Triple therapy regardless of WHO-FC.	REPLACE[4] and GRIPHON[5]	0.93 (0.41 – 2.07)

Cost-minimization analysis

- Table 2 shows the prices, number of tablets used per patients per year and yearly costs

Table 2. Prices, number of tablets per patients per year and yearly costs

Drug	Price per tablet (BRL)	Price source	Tablets/year	Yearly costs (BRL)
Riociguat	90.47	Proposed price to Brazilian MoH	1,095 (t.i.d.)	99,065
Selexipag	133.20	SIGTAP (8)	730 (b.i.d.)	97,236
Ambrisentan (ERA)	25.72	SIGTAP (8)	365 (q.d.)	9,388
Bosentan (ERA)	8.83	SIGTAP (8)	730 (b.i.d.)	6,446
Sildenafil (PDE5i)	11.80	BPS/SIAG (9)	1,095 (t.i.d.)	12,921
			Riociguat + ERA*	**106,932**
			Selexipag + ERA* + PDE5i	**118,024**
			Cost savings of riociguat + ERA* vs. selexipag + ERA* + PDE5i	**11,092**

MoH: Ministry of health; t.i.d.: three times per day; b.i.d: two times per day; q.d.: once per day
*Considering 48.3% usage for ambrisentan and 51.7% for bosentan from DATASUS
Note: Macitentan (ERA) and Tadalafil (PDE5i) do not have regulatory approval in Brazil, therefore were not included in the cost analysis

- Yearly costs per patient for riociguat + ERA was BRL 106,932 vs. BRL 118,024 for selexipag + ERA + PDE5i, showing a cost saving of BRL 11,092 per year for riociguat + ERA treatment

CONCLUSIONS

- Our complementary analyses compared to those published by Ornstová *et al.*[6] led to the same conclusion: it was not possible to identify a statistically significant difference in efficacy between riociguat + ERA vs. selexipag + ERA + PDE5i
- Even with different data collection approaches, comparing different WHO FC and outcomes, the results were consistent
- However, concerning the annual treatment cost, the combination of riociguat + ERA proved to be less costly when compared to the therapy of selexipag + ERA + PDE5i

REFERENCES

1. Humbert, M. et al. 2022 ESC/ERS Guidelines for the diagnosis and treatment of pulmonary hypertension. Eur. Heart J. 43, 3618–3731 (2022).
2. Ministério da Saúde. Protocolo Clínico e Diretrizes Terapêuticas: Hipertensão Pulmonar(2023).
3. Fernandes, C. J. et al. Atualização no Tratamento da Hipertensão Arterial Pulmonar. Arq. Bras. Cardiol. 117, 750–764 (2021).
4. Hoeper, M. M. et al. Switching to riociguat versus maintenance therapy with phosphodiesterase-5 inhibitors in patients with pulmonary arterial hypertension (REPLACE): a multicentre, open-label, randomised controlled trial. Lancet Respir. Med. 9 , 573–584 (2021).
5. Sitbon, O. et al. Selexipag for the Treatment of Pulmonary Arterial Hypertension. N. Engl. J. Med. 373, 2522–2533 (2015).
6. Ornstová, Eva., Tužilab, Jan., Chadimováac, Kateřina., Mlčocha, Tomáš. & Doležalad, Tomáš. Indirect treatment comparison and cost-minimization analysis of riociguat versus selexipag in patients with pulmonary arterial hypertension. Expert Rev. . Pharmacoecon. Outcomes Res.
7. Coghlan JG, Channick R, Chin K, Di Scala L, Galiè N, Ghofrani HA, Hoeper MM, Lang IM, McLaughlin V, Preiss R, Rubin LJ, Simonneau G, Sitbon O, Tapson VF, Gaine S. Targeting the Prostacyclin Pathway with Selexipag in Patients with. with Pulmonary Arterial Hypertension Receiving Double Combination Therapy: Insights from the Randomized Controlled GRIPHON Study. Am J Cardiovasc Drugs. 2018 Feb;18(1):37-47. doi: 10.1007/s40256-017-0262-z. PMID: 29307087; PMCID: PMC5772136.
8. Brasil. Sistema de Gerenciamento da Tabela de Procedimentos, Medicamentos e OPM do SUS - SIGTAP. Ministério da Saúde. DATASUS.
9. Brasil. Banco de Preços em Saúde (BPS) e Sistema Integrado de Administração de Serviços Gerais (SIASG).

Presented at ISPOR 2024, May 5–9, Atlanta, GA

Corresponding author: Ricardo Saad (ricardo.saad@bayer.com)