RWD119

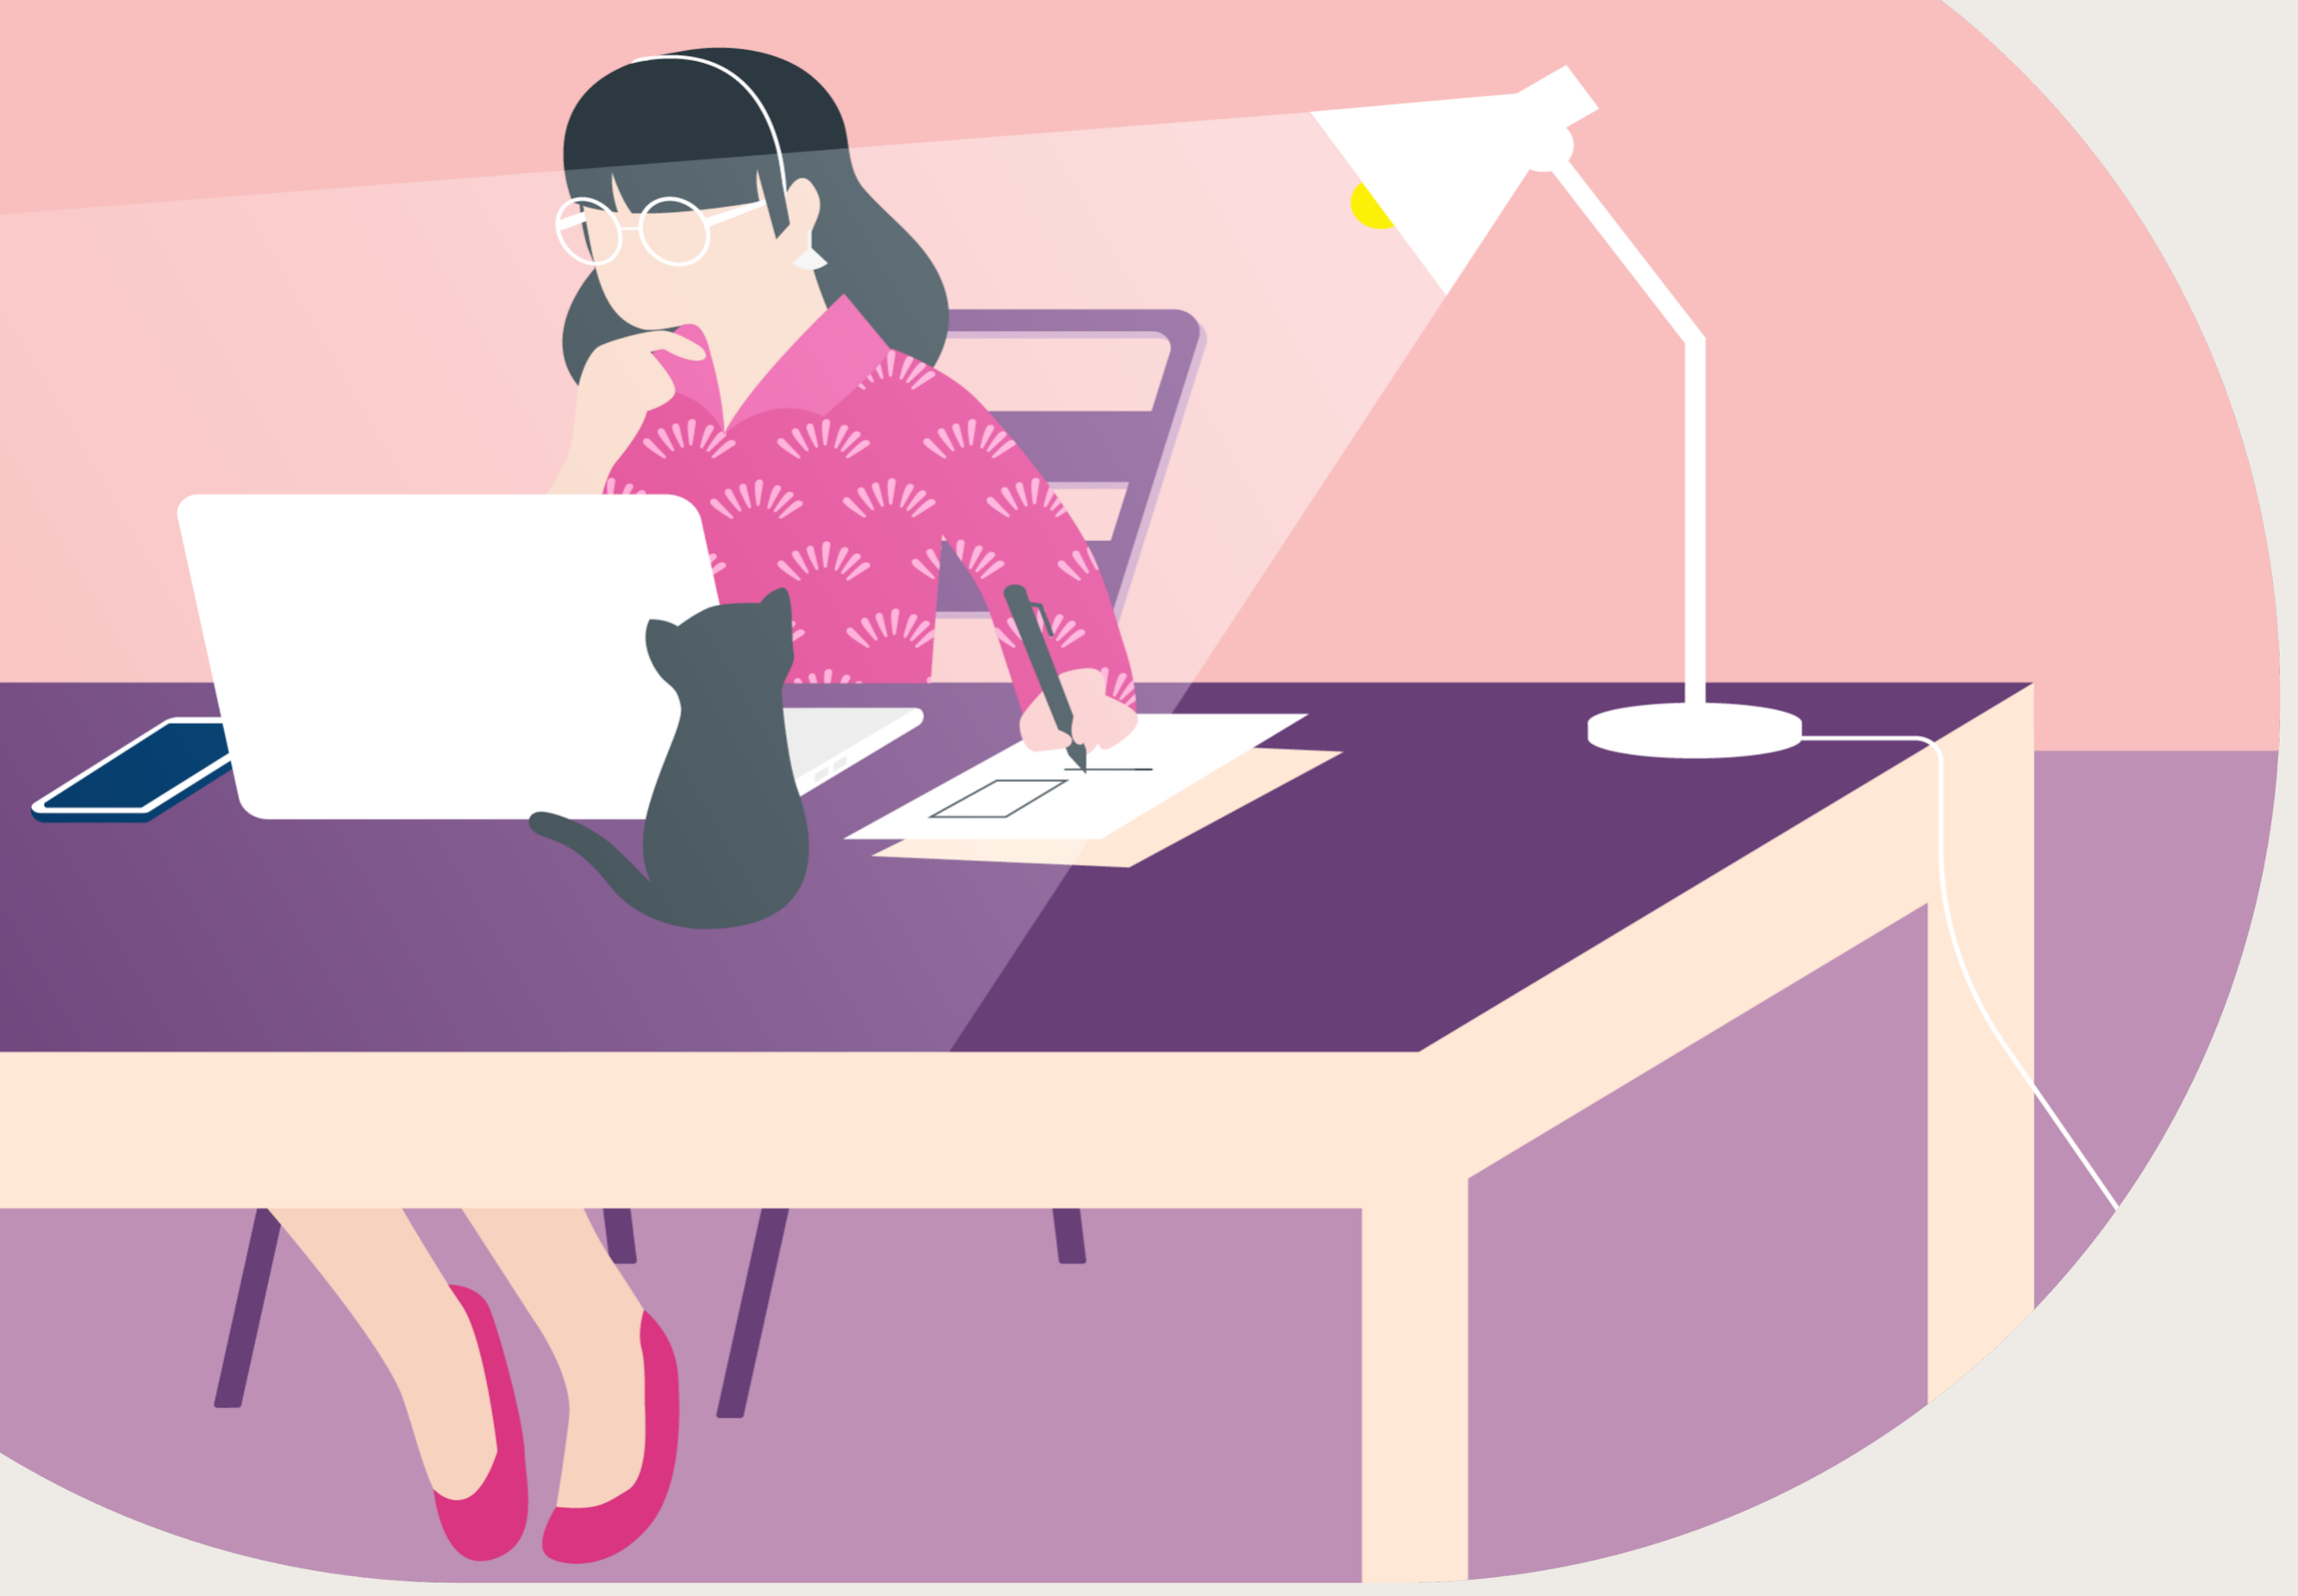

A Methodological Approach Using Sentiment Analysis (SA) of Online Medical Forums as a Source of Real-World Data on Patient Experiences

Andrea N. Cimino,[1] Curtis Culbertson,[1] Eren Watkins,[2] Jim Li[2] & Sandra Wangeshi[1]
1. Rogue Scholar Consulting, Baltimore, MD & 2. Organon, Jersey City, NJ

Online reviews of products used to treat medical conditions are a rich source of real-world data. Analyzing these data is a novel alternative to patient interviews and focus groups. The methods described here have broad application across diseases, new and emerging therapeutic areas, and for outcomes research evidence generation

WebMD.com Review

KO841610 | 25-34 | Female | On medication for less than 1 month | Patient 8/4/2018

Condition: Vaginosis caused by Bacteria

Overall rating 4.0 ★★★★☆

Effectiveness ★★★★☆ **Ease of Use** ★★★★☆ **Satisfaction** ★★★★☆

I find reviews to be helpful for people looking for real life answers like myself. Information is valuable. I had been dealing with BV for 6 long torturous months. The fishy smell was embarrassing and it would make me nauseous after while. When I got tested from my gynecologist, she told me that I was borderline BV....so they refused to give me anything. After a 2nd visit and more compliants from me because it would come back every single month after I took OTC med every month....my doctor finally prescribed me with Product A The rx was like $120 even with my insurance! Luckily, my dr gave me a coupon and it dropped the price down to around $40. I inserted the vaginal cream that same night (it's only one dose). Ever since I inserted it, I've had no more fishy smell and no more thin milky discharge....and it's been 2 months so far that I'm BV free! I'm so relieved! However, I did get a yeast infection that keeps coming back after I've tried several treatments (I tried three over-the-counter products). BV gone but YI is still lingering around. I'm currently trying boric acid suppositories for my reoccurring YI. I hope it works. But as far as Product A for BV, it worked for me! I'm 2 months free of BV and I was getting them every month for the last 7 months! I would recommend Product A but please beware of possible yeast infection or UTI. I hope that this information helps. **Read Less** ^

01 Pharmacovigilance & Ethics

Data are anonymous, complicating the AE/SE reporting without explicit policies

IRB may deem as research not involving human subjects

02 Data Extraction & Cleaning

Custom web scraper can extract all text, demographics, star ratings, & dates into a relational SQL database

Clean text of cases, special characters, emojis, contractions and typos

03 Tokenization & Determinations

Word-, sentence-, and review-level tokens provide different levels of analysis

Reviews that contain multiple topics need to be parsed out to determine the topical theme (see left)

04 Compare & Validate SA Packages

SA packages use different lexicons & scoring algorithms. Consider building a lexicon for a disease state of interest

Validate scores with a human review

05 Quantitative Analyses

Use reviewer attributes, start ratings, and SA scores to test hypotheses

Line graphs, bar charts, and word clouds provide additional details to make product comparisons (see below)

06 Qualitative Analyses

Themes can be summarized using qualitative description for a rich, straight description of an experience without the imposition of a theoretical or interpretative lens

DETERMINATION PROPORTIONS

Attribute: Product characteristics
Use: Reviewers' advice on how to use product
Side Effects: Intended or unintended effect (excludes "no side effects")
Value: Efficacy, price, worth, or usefulness
Adherence: Reviewers who stopped taking it
Condition: Disease state and its impact
Cross Product: Other products used & efficacy
Doctor: Interactions with a provider
Neutral: Dose, encouragement, questions

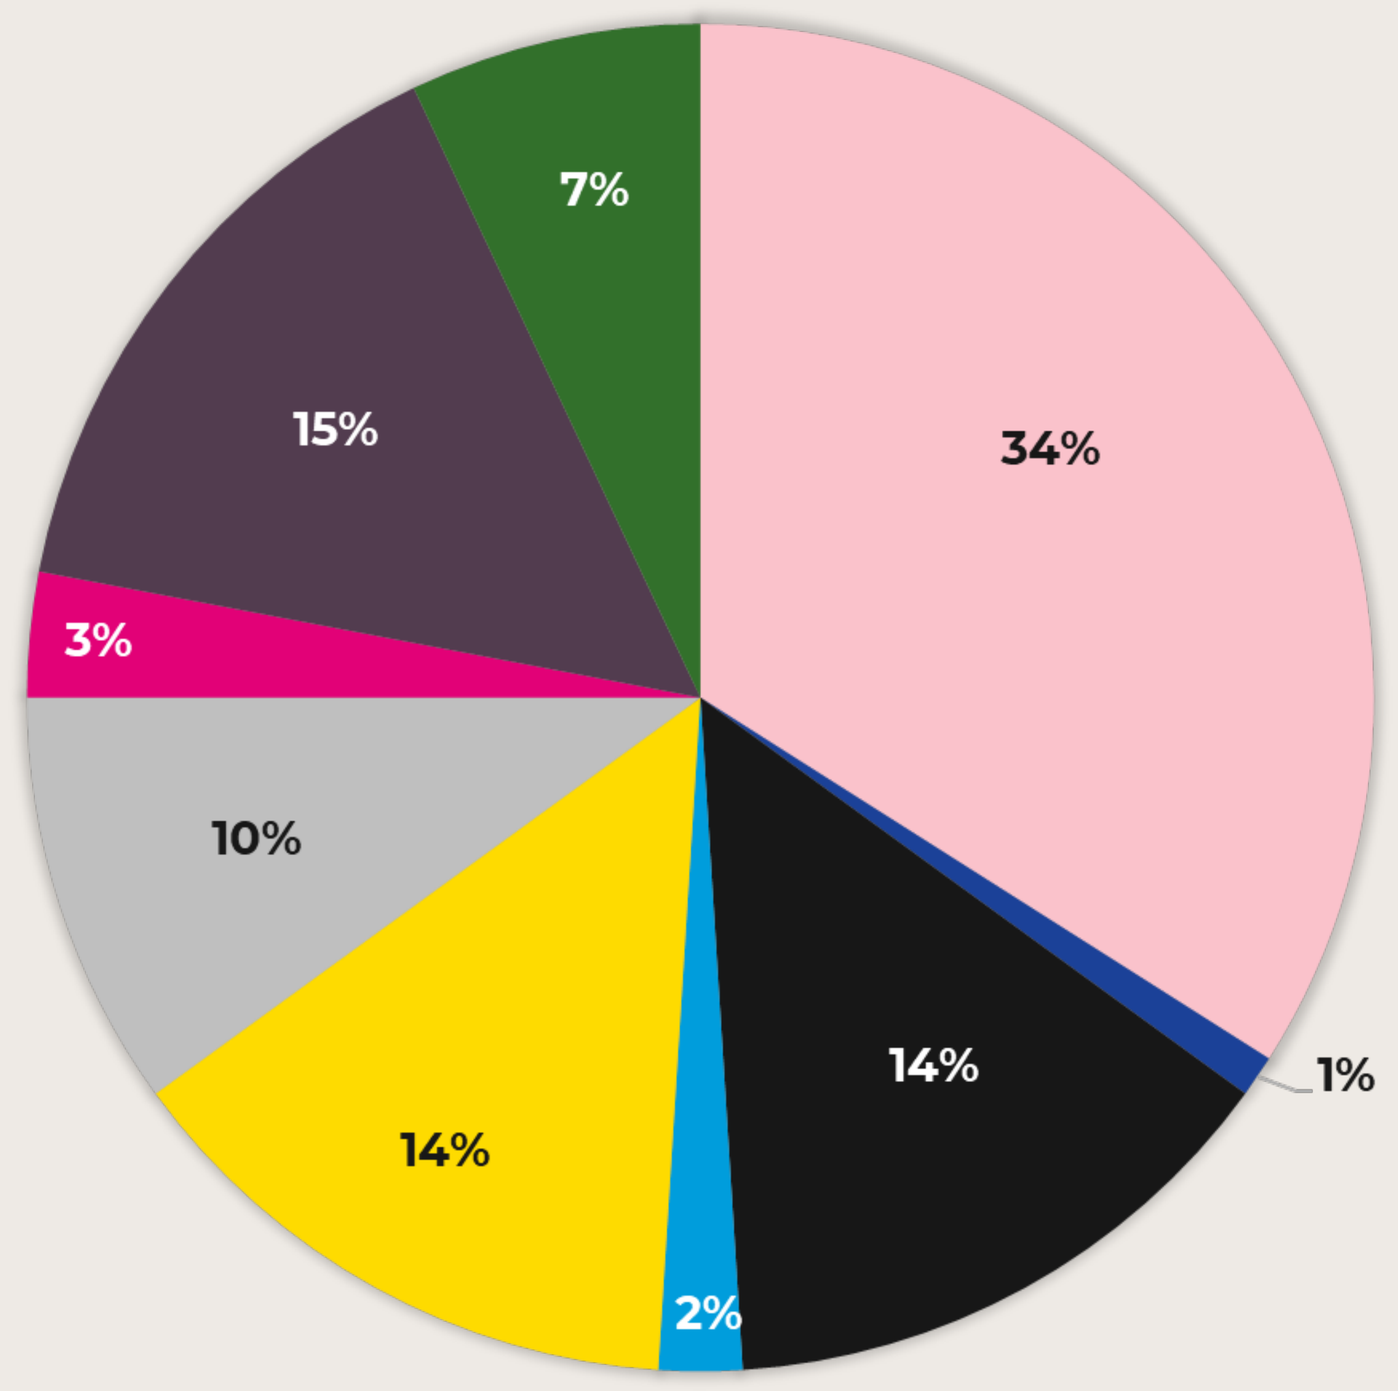
7%
34%
15%
3%
10%
14%
14%
1%
2%

LINE CHARTS & WORD CLOUDS

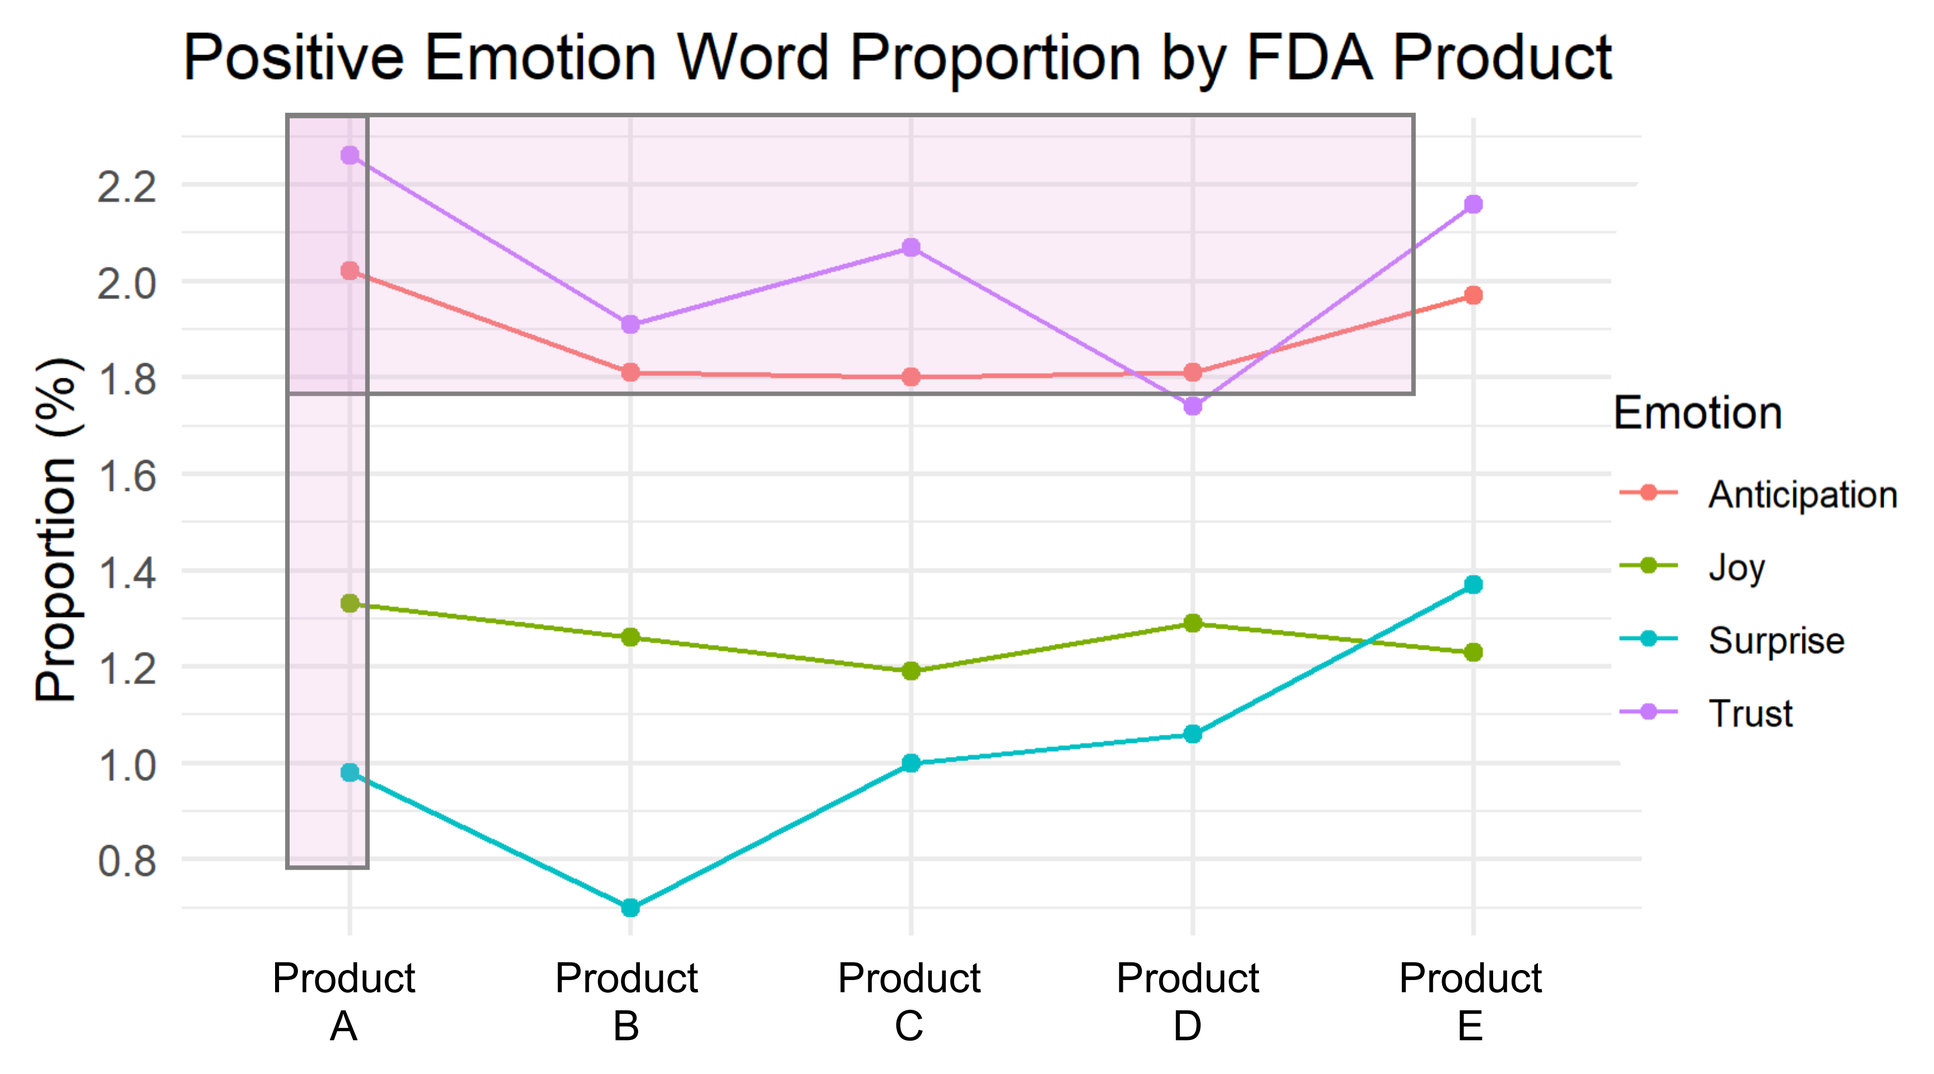
Positive Emotion Word Proportion by FDA Product
Proportion (%)
2.2
2.0
1.8
1.6
1.4
1.2
1.0
0.8
Product A
Product B
Product C
Product D
Product E
Emotion
Anticipation
Joy
Surprise
Trust

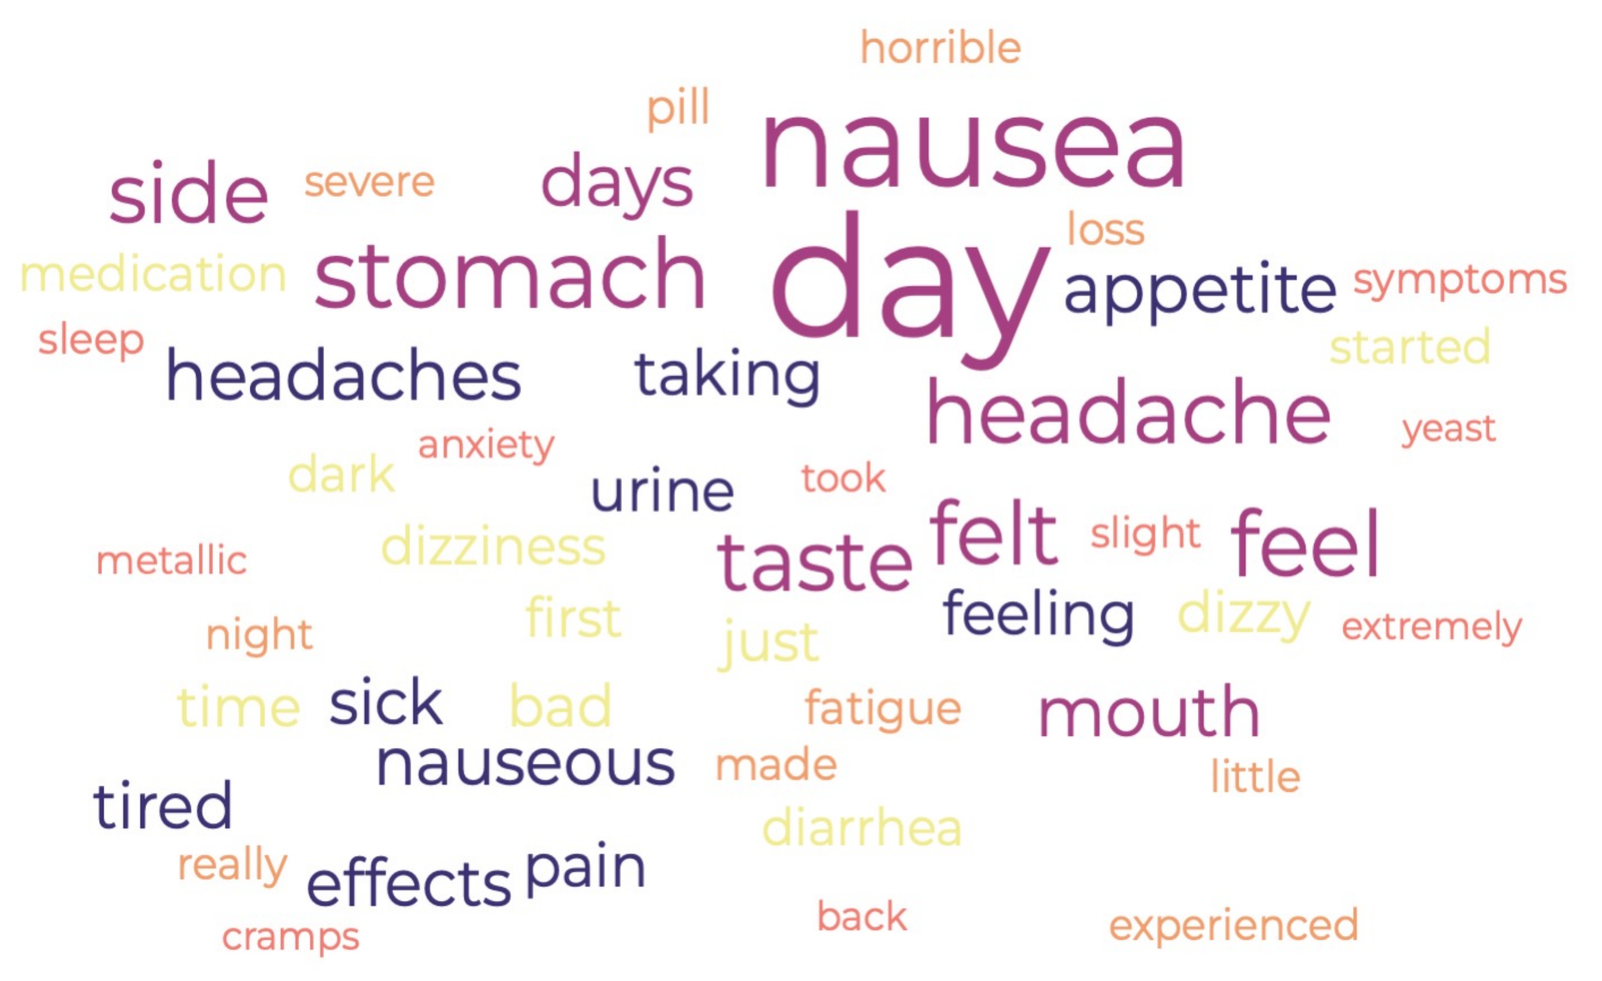
horrible
pill
side
severe
days
nausea
medication
stomach
day
loss
appetite
symptoms
sleep
headaches
taking
headache
started
yeast
anxiety
dark
urine
took
metallic
dizziness
taste
felt
slight
feel
night
first
just
feeling
dizzy
extremely
time
sick
bad
fatigue
mouth
nauseous
made
little
tired
diarrhea
really
effects
pain
cramps
back
experienced

THEMATIC ANALYSIS

*"**I didn't have** any of the **side effects** I read about. I did have **some "leakage"** the first couple of days and just used a liner. It's been a week and I seem to be back to my old self. I **would definitely use this again**, but this time **use the coupon**. With my insurance it **cost** me around **$125**."*

ROGUE SCHOLAR
CONSULTING

SCAN HERE
USE YOUR PHONE