HSD122

Medication Adherence, Persistence, and Treatment Switching in Patients Receiving Advanced Therapies for Rheumatoid Arthritis in Clinical Practice

Martin Bergman,[1] Patrick M. Zueger,[2] Yi Peng,[2] Richard Thielen[2]

[1]*Drexel University College of Medicine, Philadelphia, PA, USA;* [2]*AbbVie Inc., North Chicago, IL, USA*

OBJECTIVE

To evaluate 1-year treatment adherence, persistence, and switching for upadacitinib vs other advanced therapies (ATs) in TNF inhibitor (TNFi)-experienced patients with RA

CONCLUSIONS

Overall, treatment adherence was significantly higher, and rates of treatment discontinuation or switching were significantly lower for patients initiating upadacitinib compared with other ATs after recent TNFi treatment

The FDA safety warning for tofacitinib did not appear to significantly impact treatment persistence across AT classes

These data highlight the potential benefit of initiating upadacitinib after discontinuing a TNFi vs initiating another TNFi or non-TNFi AT, including other JAK inhibitors

Medical writing services provided by Natalie Mitchell, of Fishawack Facilitate, Ltd, part of Avalere Health, and funded by AbbVie.

AbbVie funded this study and participated in the study design, research, analysis, data preparation, interpretation of data, and the review and approval of the publication. All authors had access to relevant data and participated in the drafting, review, and approval of this publication. No honoraria or payments were made for authorship.

Financial arrangements of the authors with companies whose products may be related to the present report are listed as declared by the authors: M. Bergman reports consulting, advising, and serving on speakers' bureaus for AbbVie, Amgen, BMS, Genentech/Roche, Gilead, GSK, Janssen, Merck, Novartis, Pfizer, Sandoz, and Sanofi/Regeneron; and is a shareholder of J&J (parent company of Janssen) and Merck. P.M. Zueger, Y. Peng, and R. Thielen are employees of AbbVie, and may hold stock options.

References

1. Smolen JS, et al. *Ann Rheum Dis.* 2023;82:3–18.
2. Osterberg L, et al. *NEJM.* 2005;353(5):487–97.
3. AbbVie. AbbVie Receives FDA Approval of RINVOQ™ (upadacitinib), an Oral JAK Inhibitor For The Treatment of Moderate to Severe Rheumatoid Arthritis. Available at: https://news.abbvie.com/2019-08-16-AbbVie-Receives-FDA-Approval-of-RINVOQ-TM-upadacitinib-an-Oral-JAK-Inhibitor-For-The-Treatment-of-Moderate-to-Severe-Rheumatoid-Arthritis. Accessed March 8, 2024.
4. Thorley J. *Lancet Rheumatol.* 2021;3(11):e757.

Presented at the Professional Society for Health Economics and Outcomes Research (ISPOR), May 5–8, 2024, Atlanta, GA, USA

INTRODUCTION

- Following treatment failure with an initial TNF inhibitor (TNFi), guidelines recommend patients with RA be treated with advanced therapies (ATs) with alternative mechanisms of action[1]
- Poor adherence and persistence on ATs contribute to suboptimal outcomes in patients with RA[2]
- It is important to evaluate these outcomes with upadacitinib (UPA), the most recently approved AT for RA,[3] following treatment with TNFi to help inform treatment decision-making

METHODS

- This retrospective study used data from the Merative™ MarketScan® Research Database (August 2018–April 2023) **(Figure 1)**
- Eligible patients had received a first prescription claim for:

METHODS CONTINUED

 - JAK inhibitors (JAKis): UPA, tofacitinib (TOF), baricitinib (BAR)
 - TNFis: adalimumab (ADA), etanercept (ETA)
 - Non-TNFi biologics: abatacept (ABA), tocilizumab (TOC)
- Outcomes were assessed during 12-month follow-up:
 - Adherence: percentage of patients who were adherent to treatment, defined as proportion of days covered (PDC) ≥ 80%
 - Persistence: discontinuation or treatment switch rate 12 months after index treatment initiation, and mean or median days on treatment in 12 months after initiation
 - Discontinuation was defined as a gap in treatment of ≥ 60 days based on pharmacy or medical claims
- Adjusted odds ratios (aOR) and adjusted hazard ratios (aHR) with 95% CIs were estimated for comparisons of treatment adherence and treatment discontinuation or switching, respectively
 - Models were adjusted for age, sex, Charlson Comorbidity Index, and number of ATs received anytime before the index date
- Stratified analyses were conducted to determine the potential impact of the FDA warning (September 1, 2021)[4] on JAKi safety on treatment persistence of the included ATs
 - Patients were stratified according to index medication follow-up occurring fully before, fully after, or with overlap of the FDA warning date
- Subgroup analyses evaluated treatment persistence among patients with use of a single TNFi as their only prior AT

Figure 1. Study Design

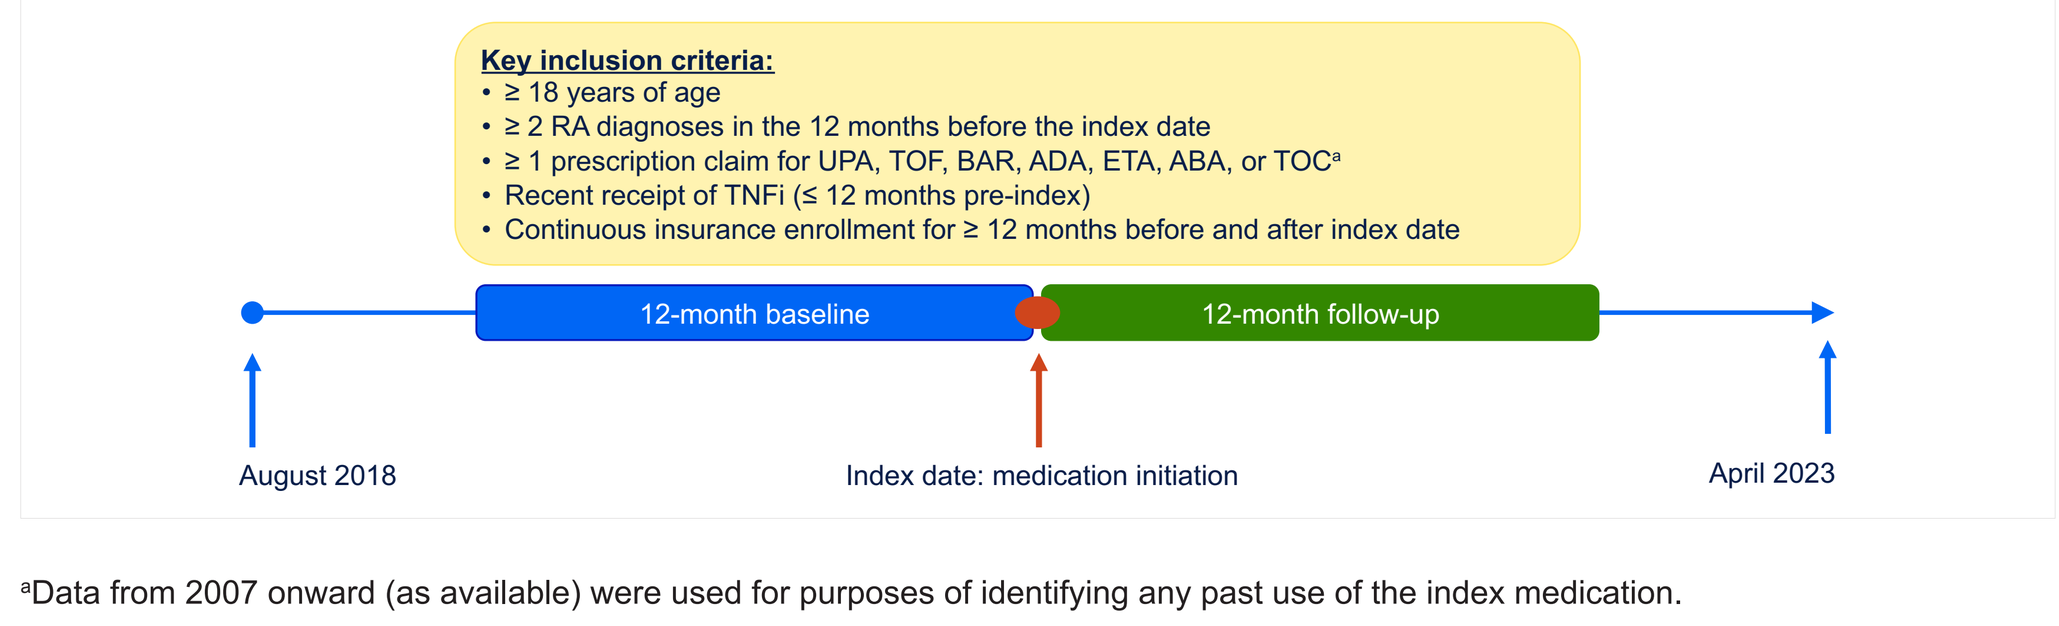
Key inclusion criteria:
• ≥ 18 years of age
• ≥ 2 RA diagnoses in the 12 months before the index date
• ≥ 1 prescription claim for UPA, TOF, BAR, ADA, ETA, ABA, or TOCa
• Recent receipt of TNFi (≤ 12 months pre-index)
• Continuous insurance enrollment for ≥ 12 months before and after index date
12-month baseline
12-month follow-up
August 2018
Index date: medication initiation
April 2023

[a]Data from 2007 onward (as available) were used for purposes of identifying any past use of the index medication.

RESULTS

- Demographic characteristics and comorbidity burden were similar between ATs, while ADA and ETA initiators had used fewer prior ATs **(Table 1)**
- At 1 year, compared with UPA initiators, patients initiating each other AT were significantly less likely to be adherent to treatment **(Figure 2)** and significantly more likely to discontinue or switch treatment **(Figure 3)**
- Median time on treatment was greater for UPA initiators (>366 days) compared with TOF (289 days), BAR (149 days), ADA (252 days), ETA (211 days), ABA (282 days), and TOC (239 days) through 1 year **(Figure 4)**
- Discontinuation or switch rates for index medications with follow-up occurring fully before, fully after, or with overlap of the FDA warning on JAKi safety were comparable to the overall results **(Figure 5)**
- Among patients with use of a single TNFi as their only prior AT, UPA initiators had the lowest rate of discontinuation or switch at 1 year compared to each other AT (UPA 47.4%; TOF 54.7%; BAR 71.4%; ADA 54.9%; ETA 63.9%; ABA 52.8%; TOC 62.0%)

Table 1. Baseline Characteristics

Characteristic	UPA N = 926	TOF N = 872	BAR N = 39	ADA N = 769	ETA N = 751	ABA N = 982	TOC N = 384
Age (years), mean ± SD	51.6 ± 9.6	50.9 ± 9.9	50.2 ± 8.0	50.5 ± 10.5	49.6 ± 10.8	51.8 ± 10.3	51.0 ± 10.9
Female, n (%)	740 (79.9)	698 (80.1)	29 (74.4)	610 (79.3)	592 (78.8)	805 (82.0)	310 (80.7)
Charlson Comorbidity Index, mean ± SD	1.5 ± 1.0	1.5 ± 1.0	1.5 ± 1.0	1.6 ± 1.0	1.5 ± 1.1	1.7 ± 1.1	1.6 ± 1.1
Number of prior advanced therapies, mean ± SD	2.1 ± 1.5	1.8 ± 0.9	3.1 ± 1.9	1.3 ± 0.7	1.3 ± 0.7	2.0 ± 1.0	2.3 ± 1.3
Number of prior TNFi, mean ± SD	1.5 ± 0.7	1.5 ± 0.6	1.7 ± 0.8	1.1 ± 0.3	1.1 ± 0.3	1.6 ± 0.7	1.7 ± 0.8

ABA, abatacept; ADA, adalimumab; BAR, baricitinib; ETA, etanercept; TNFi, TNF inhibitor; TOC, tocilizumab; TOF, tofacitinib; UPA, upadacitinib.

Figure 2. Adherence (PDC ≥ 80%) at 1 Year Among TNFi-Experienced Patients With RA Initiating Advanced Therapies

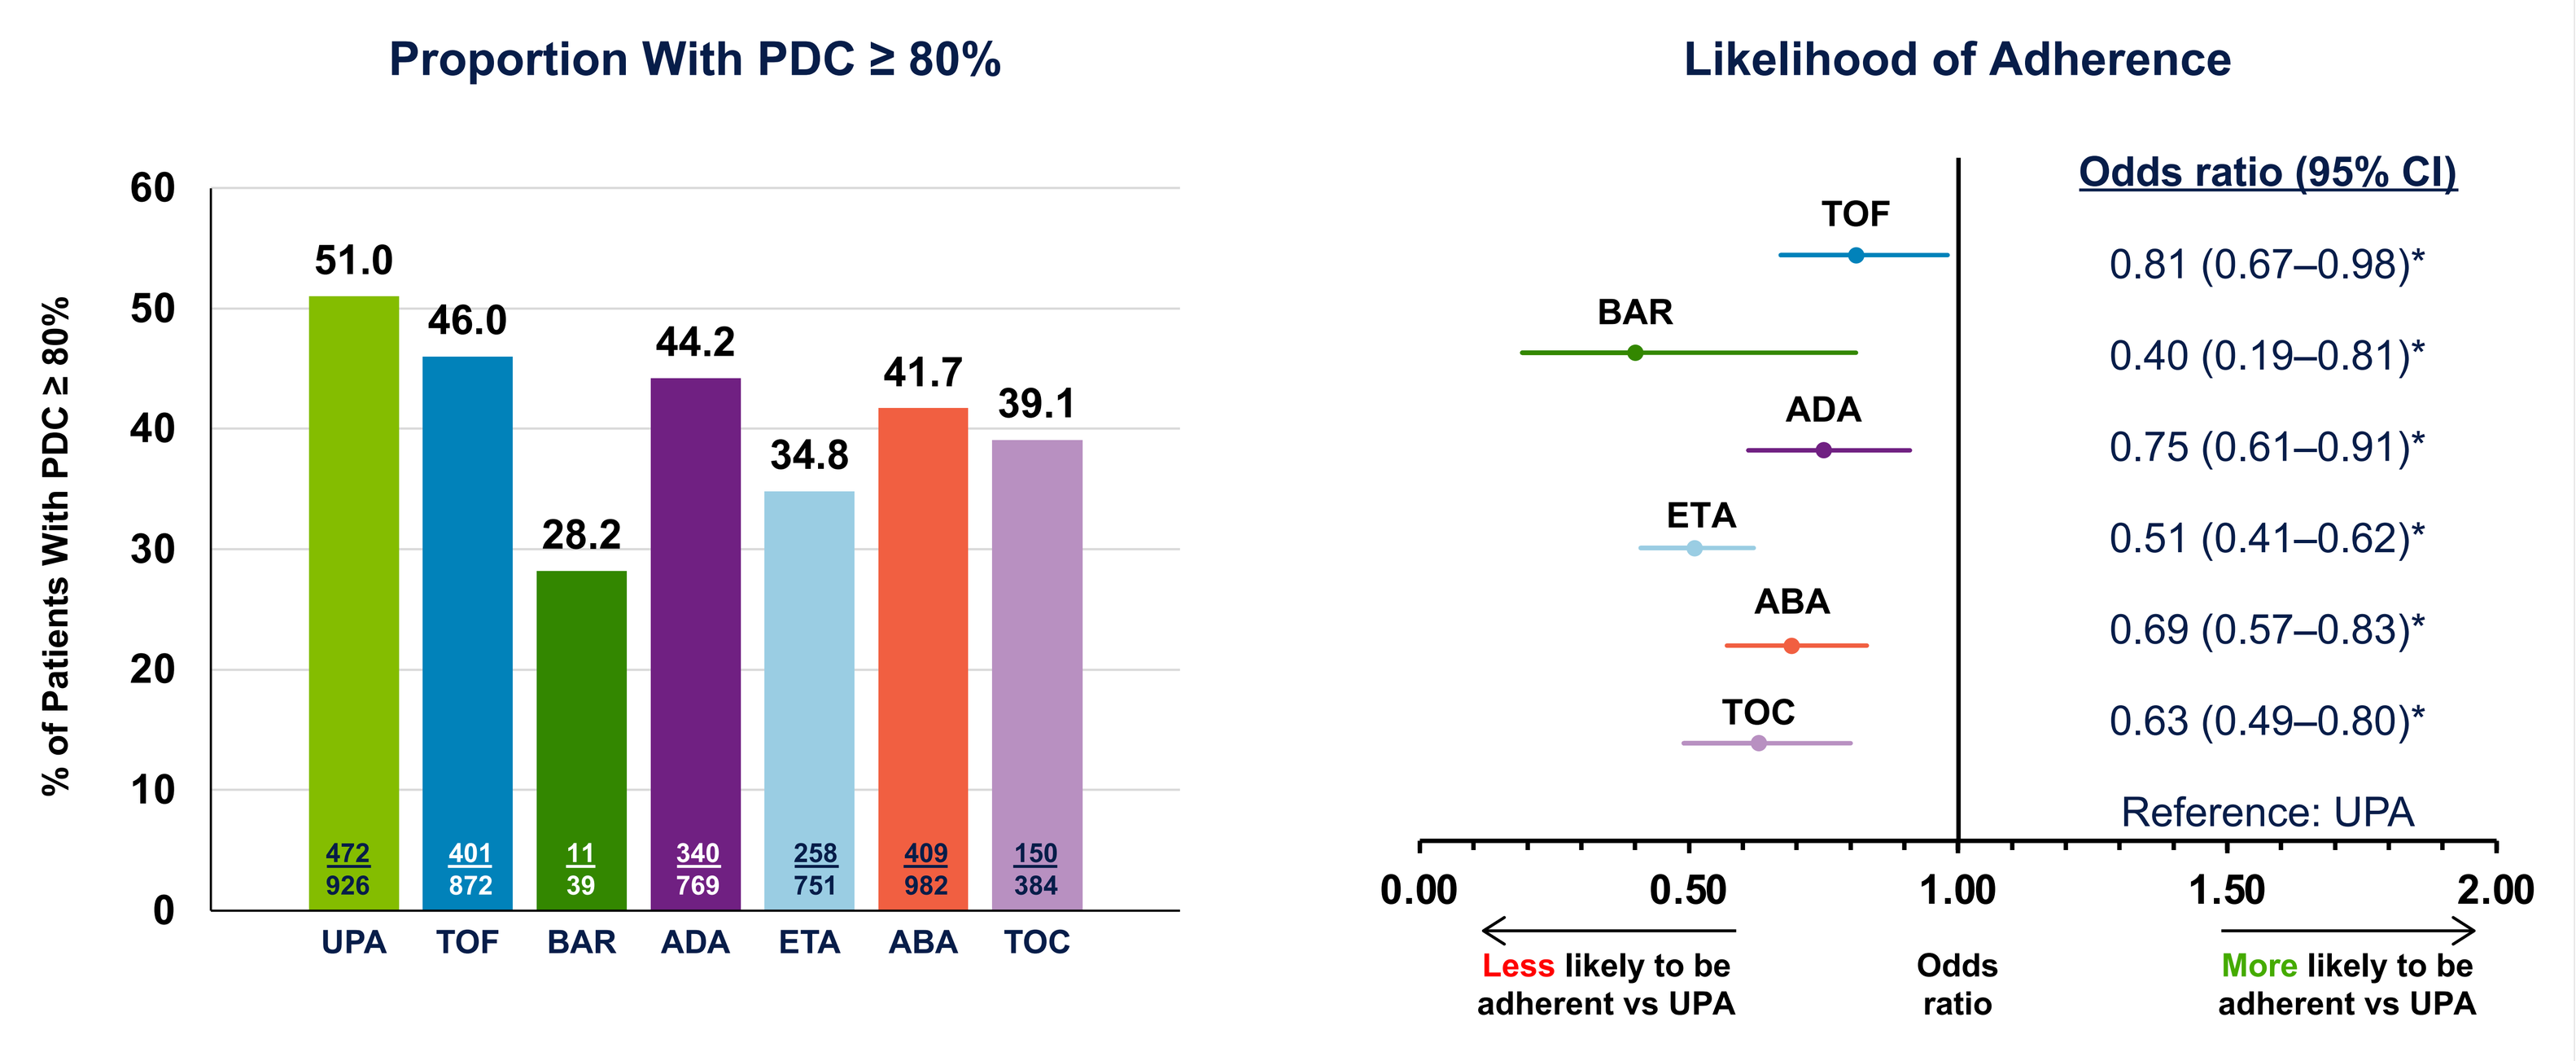
Proportion With PDC ≥ 80%
% of Patients With PDC ≥ 80%
51.0
46.0
28.2
44.2
34.8
41.7
39.1
472/926
401/872
11/39
340/769
258/751
409/982
150/384
UPA TOF BAR ADA ETA ABA TOC
Likelihood of Adherence
Odds ratio (95% CI)
TOF 0.81 (0.67–0.98)*
BAR 0.40 (0.19–0.81)*
ADA 0.75 (0.61–0.91)*
ETA 0.51 (0.41–0.62)*
ABA 0.69 (0.57–0.83)*
TOC 0.63 (0.49–0.80)*
Reference: UPA
0.00 0.50 1.00 1.50 2.00
Less likely to be adherent vs UPA
Odds ratio
More likely to be adherent vs UPA

ABA, abatacept; ADA, adalimumab; BAR, baricitinib; ETA, etanercept; PDC, Proportion of Days Covered; TNFi, TNF inhibitor; TOC, tocilizumab; TOF, tofacitinib; UPA, upadacitinib.
*P <.05.

Figure 3. Treatment Discontinuation or Switch Rates Through 1 Year Among TNFi-Experienced Patients With RA Initiating Advanced Therapies

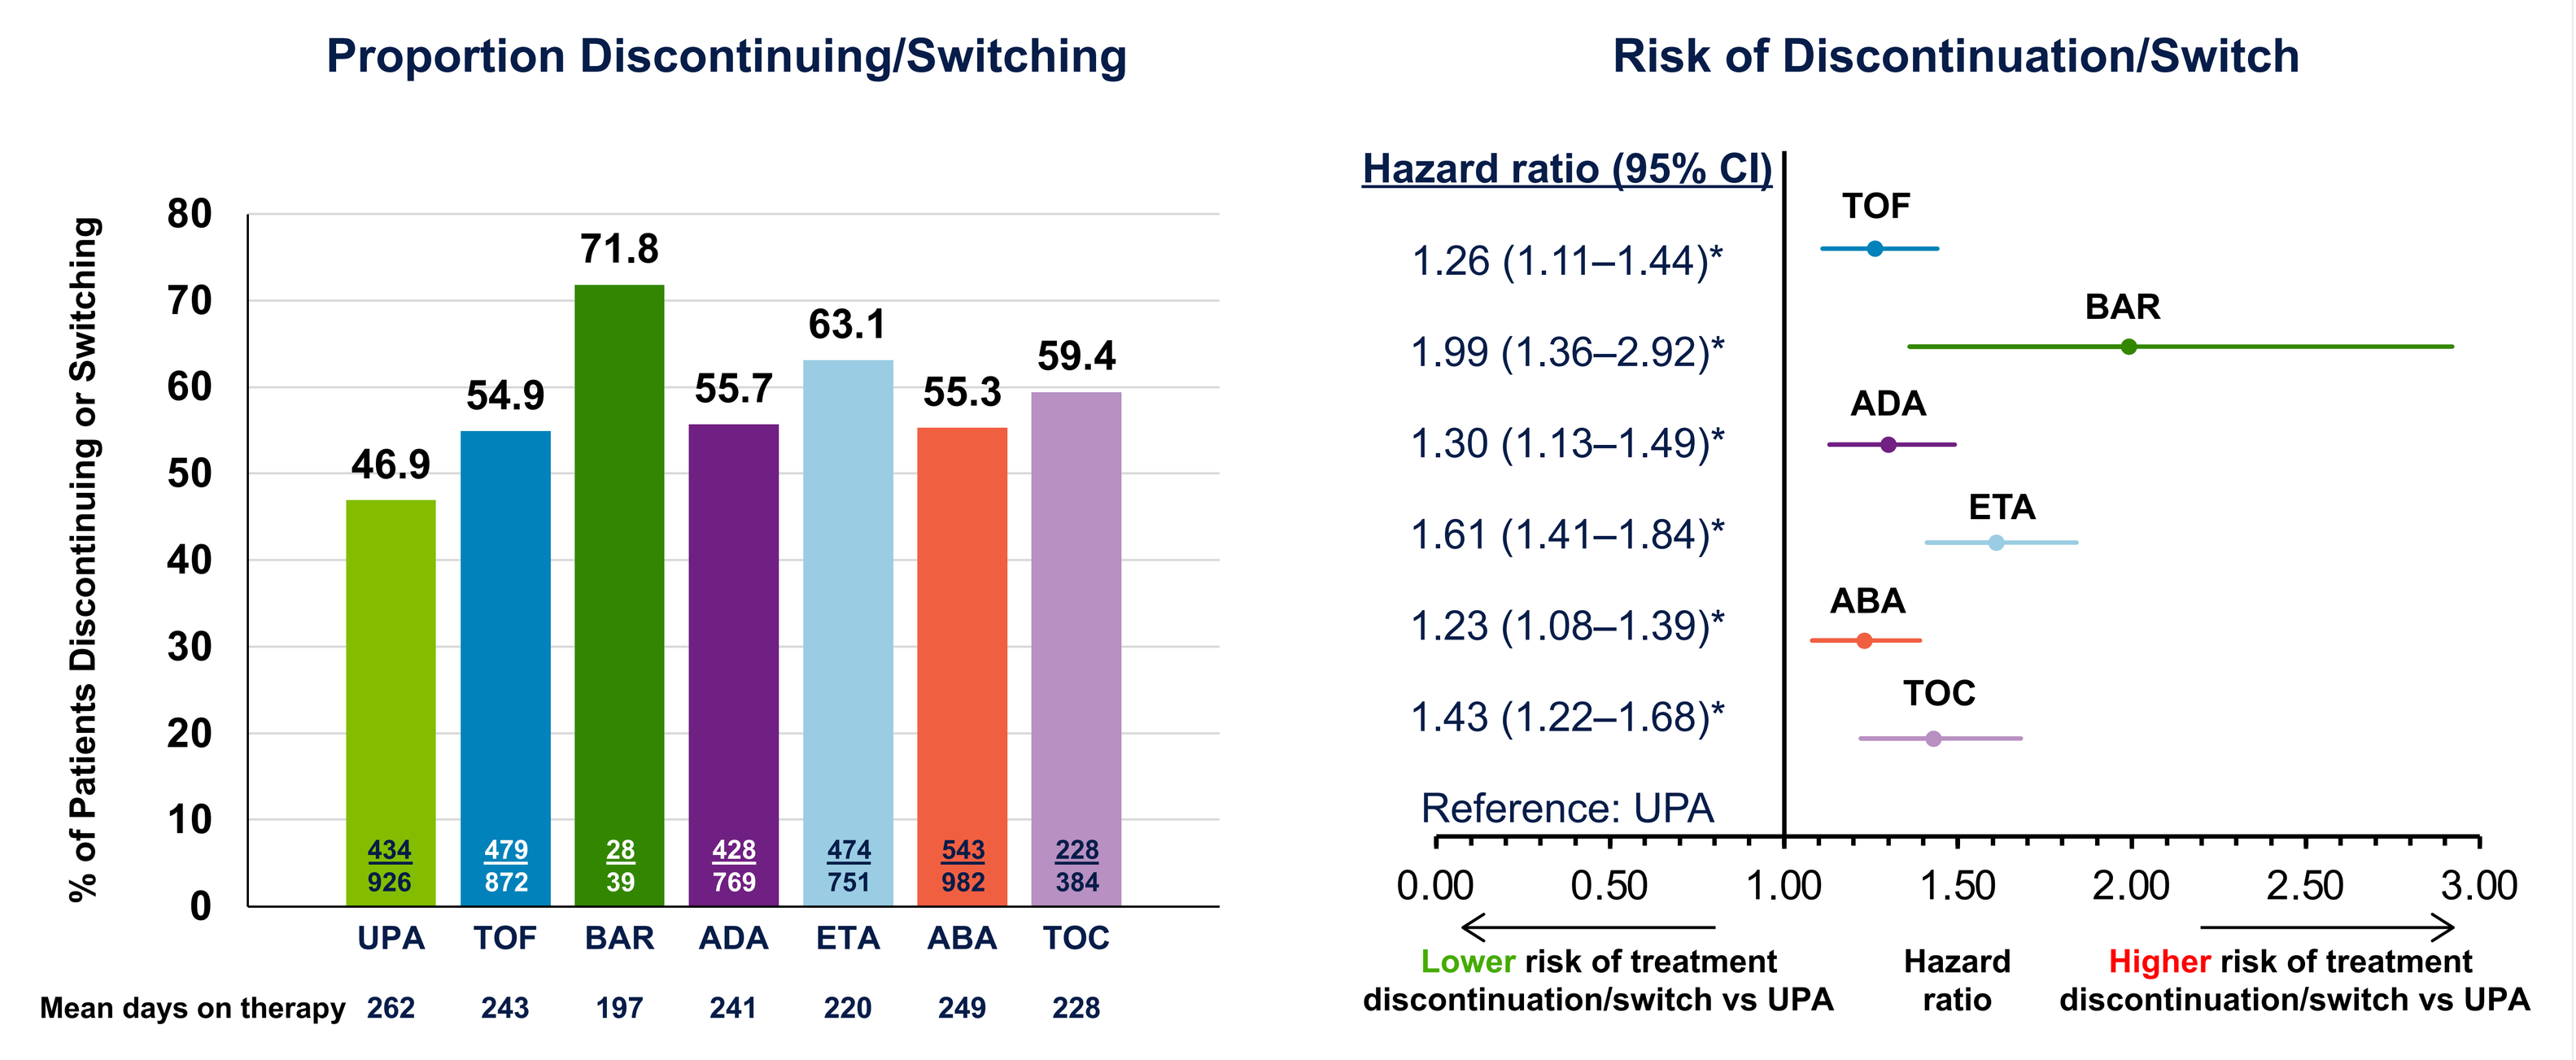
Proportion Discontinuing/Switching
% of Patients Discontinuing or Switching
46.9
54.9
71.8
55.7
63.1
55.3
59.4
434/926
479/872
28/39
428/769
474/751
543/982
228/384
UPA TOF BAR ADA ETA ABA TOC
Mean days on therapy 262 243 197 241 220 249 228
Risk of Discontinuation/Switch
Hazard ratio (95% CI)
TOF 1.26 (1.11–1.44)*
BAR 1.99 (1.36–2.92)*
ADA 1.30 (1.13–1.49)*
ETA 1.61 (1.41–1.84)*
ABA 1.23 (1.08–1.39)*
TOC 1.43 (1.22–1.68)*
Reference: UPA
0.00 0.50 1.00 1.50 2.00 2.50 3.00
Lower risk of treatment discontinuation/switch vs UPA
Hazard ratio
Higher risk of treatment discontinuation/switch vs UPA

ABA, abatacept; ADA, adalimumab; BAR, baricitinib; ETA, etanercept; TNFi, TNF inhibitor; TOC, tocilizumab; TOF, tofacitinib; UPA, upadacitinib.
*P <.05.

Figure 4. Kaplan-Meier Curve for Probability of Remaining on Treatment[a] Through 1 Year Among TNFi-Experienced Patients With RA Initiating Advanced Therapies

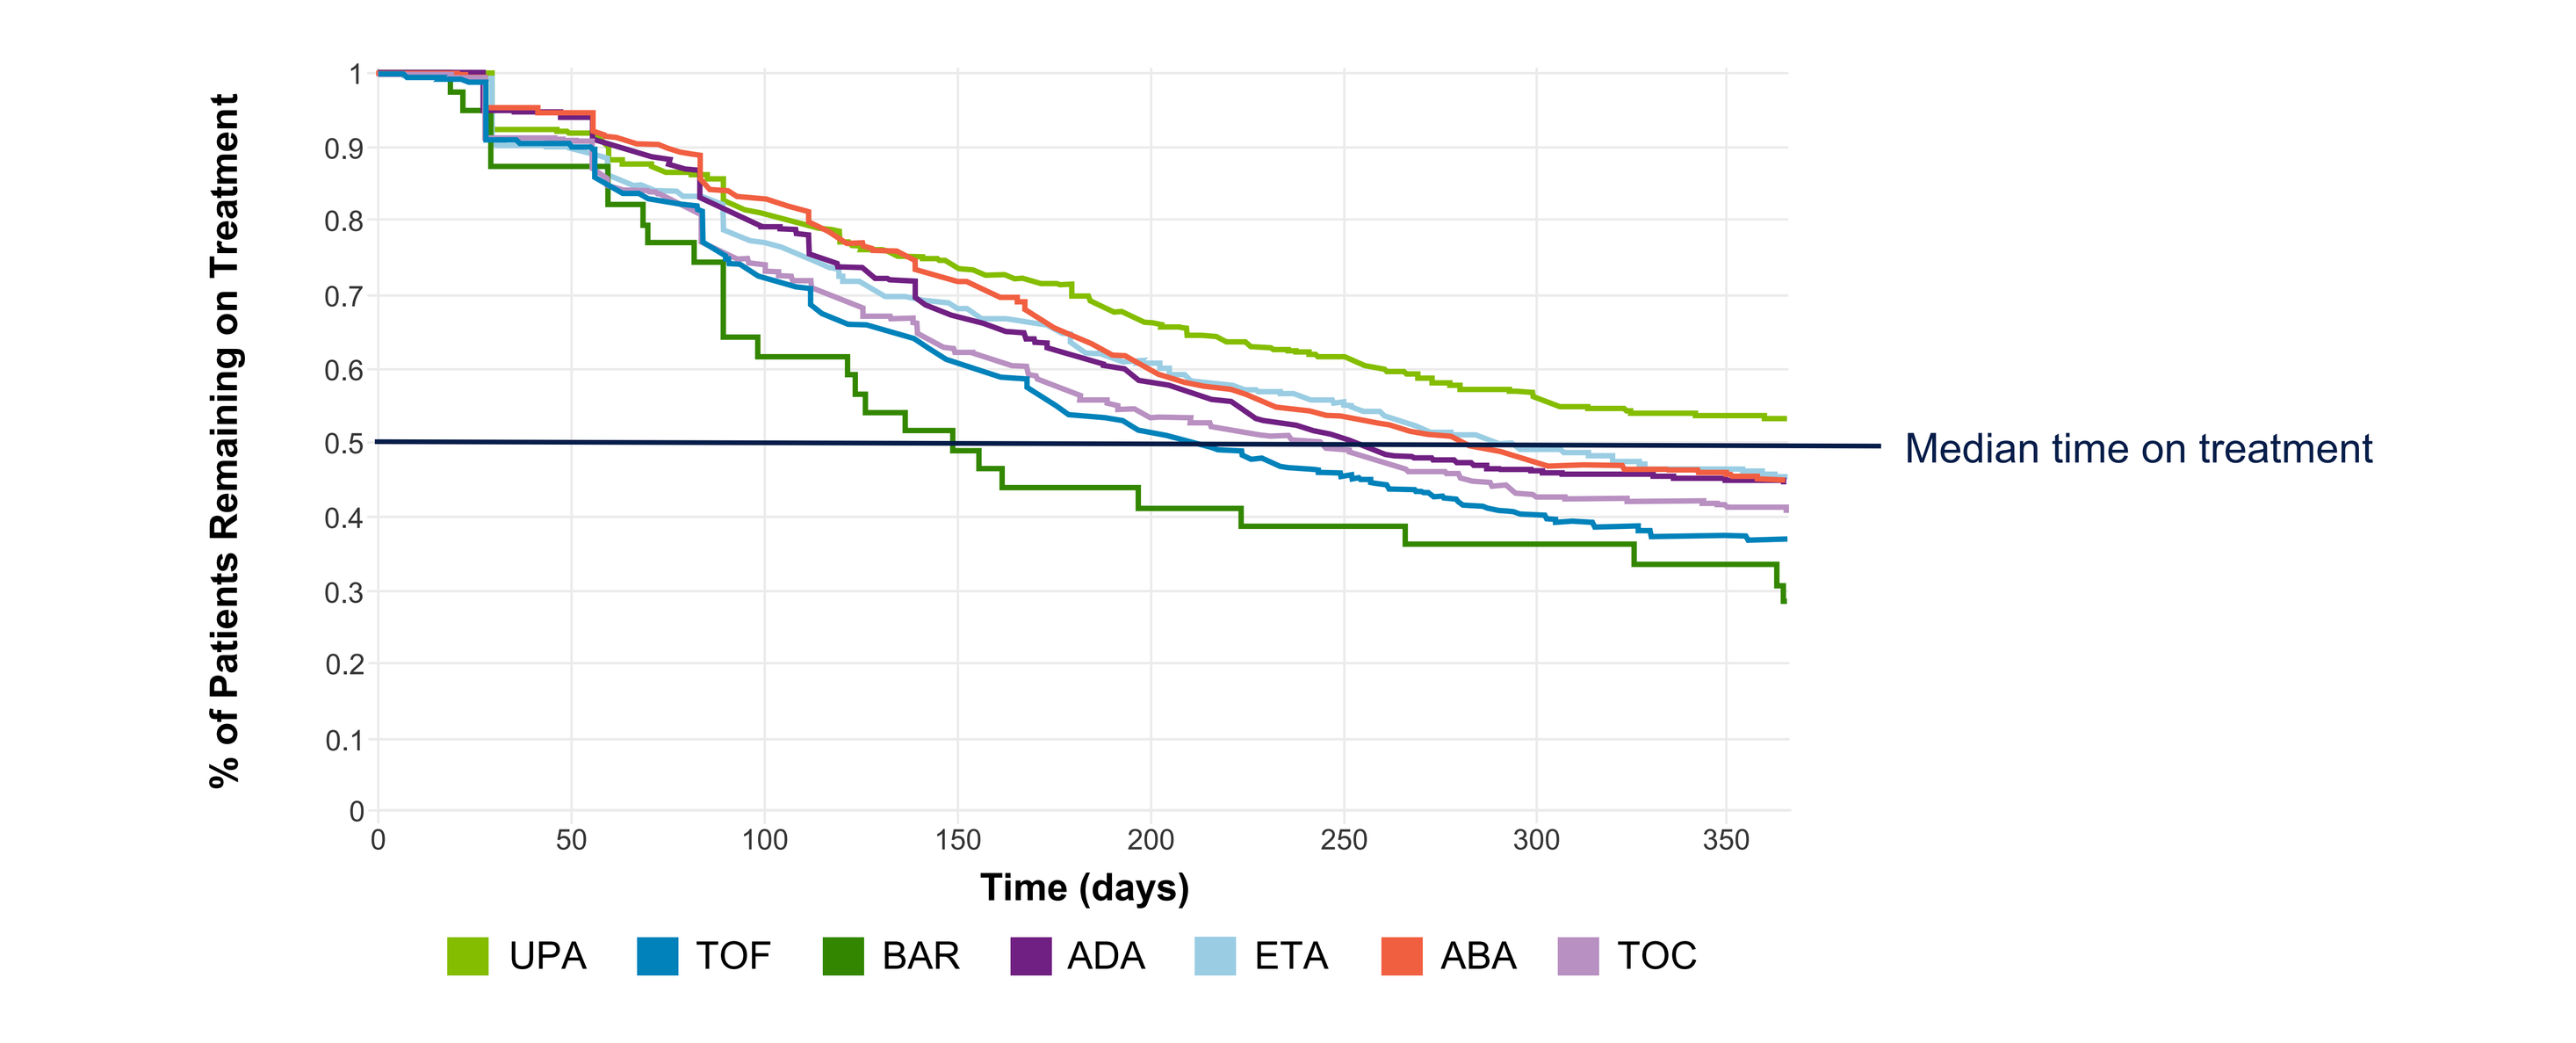
% of Patients Remaining on Treatment
Median time on treatment
Time (days)
UPA TOF BAR ADA ETA ABA TOC

ABA, abatacept; ADA, adalimumab; BAR, baricitinib; ETA, etanercept; TNFi, TNF inhibitor; TOC, tocilizumab; TOF, tofacitinib; UPA, upadacitinib.
[a]Patients who had not discontinued or switched treatment.

Figure 5. Treatment Discontinuation or Switch Rates Through 1 Year Stratified by Patients With Index Medication Follow-Up Occurring Before, After, or With Overlap of the FDA Safety Warning (September 1, 2021) Issued for TOF

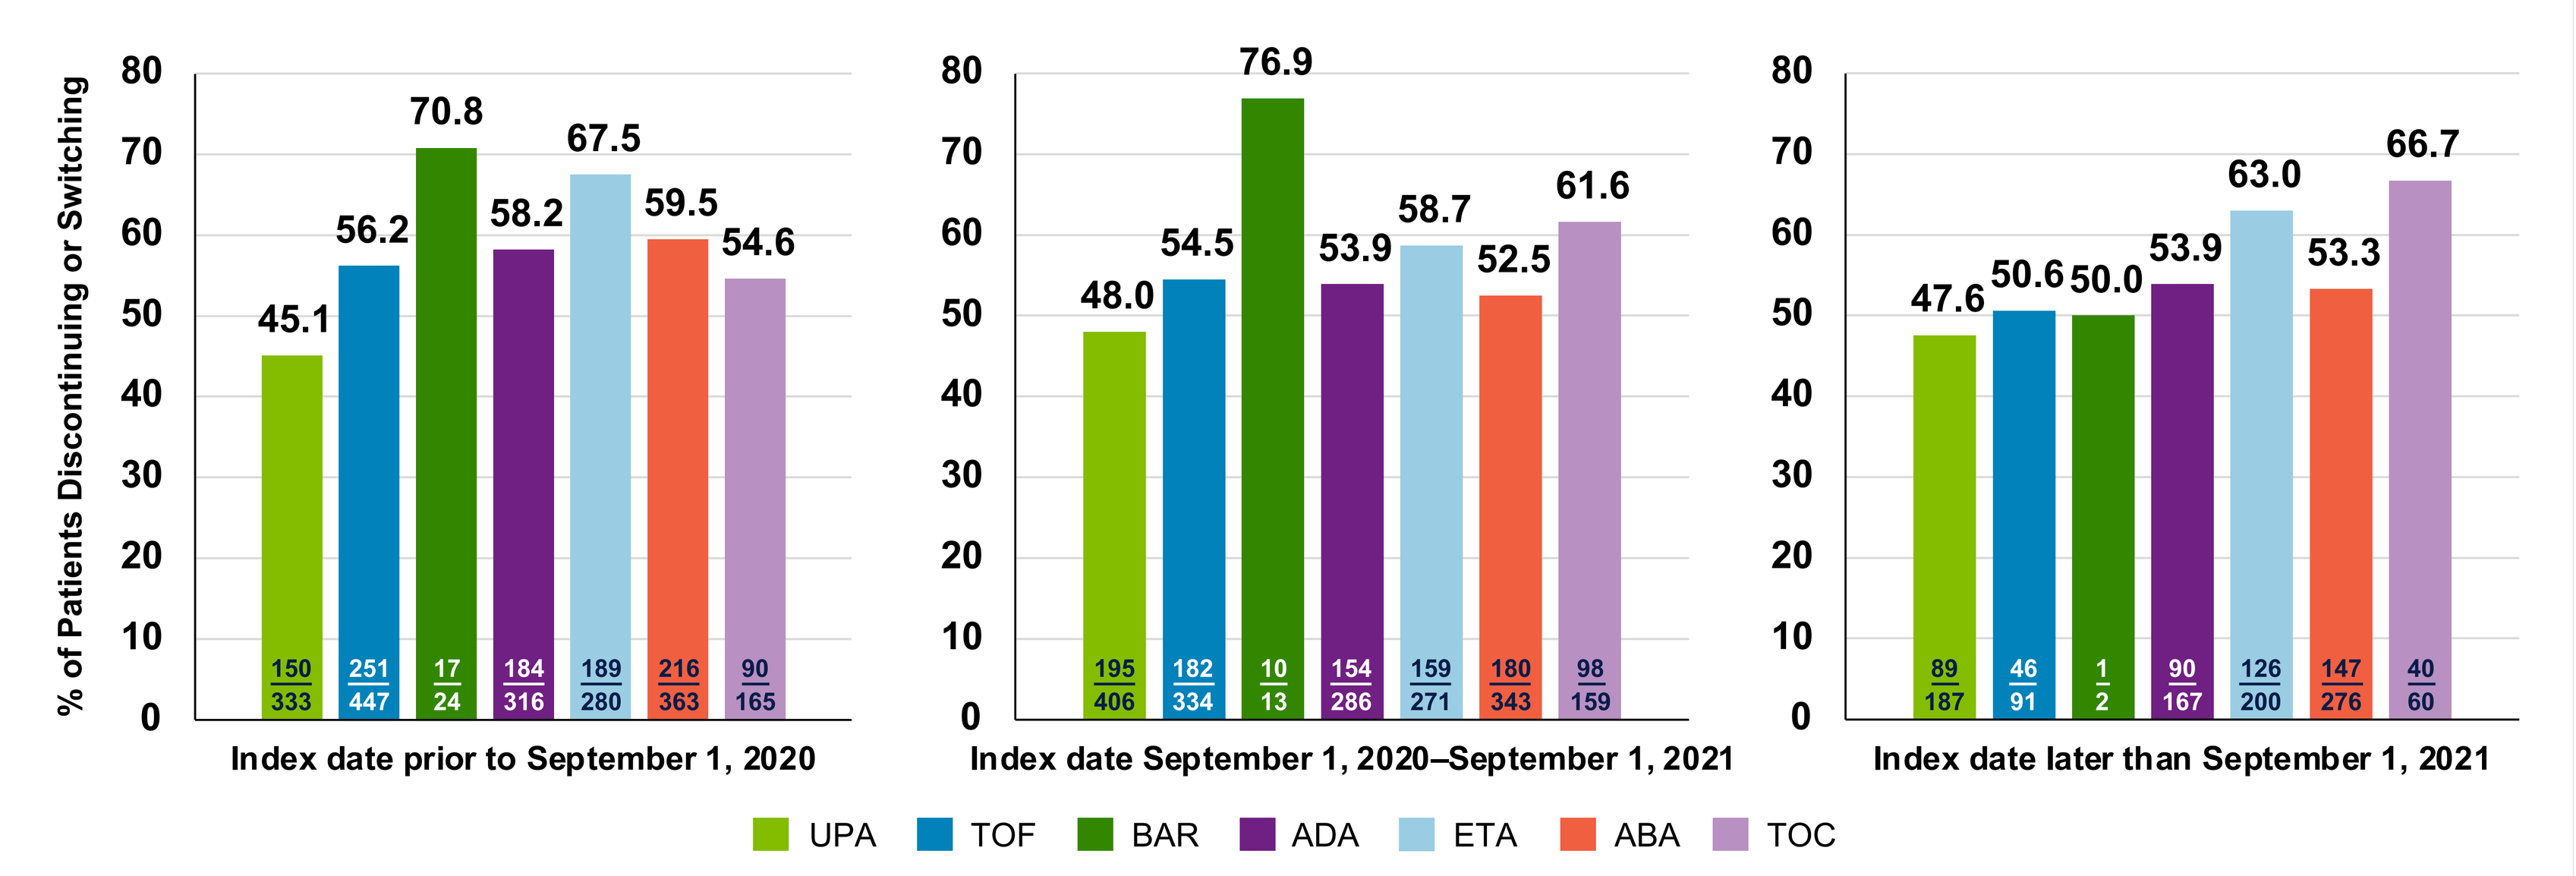
% of Patients Discontinuing or Switching
Index date prior to September 1, 2020
45.1 56.2 70.8 58.2 67.5 59.5 54.6
150/333 251/447 17/24 184/316 189/280 216/363 90/165
Index date September 1, 2020–September 1, 2021
48.0 54.5 76.9 53.9 58.7 52.5 61.6
195/406 182/334 10/13 154/286 159/271 180/343 98/159
Index date later than September 1, 2021
47.6 50.6 50.0 53.9 63.0 53.3 66.7
89/187 46/91 1/2 90/167 126/200 147/276 40/60
UPA TOF BAR ADA ETA ABA TOC

ABA, abatacept; ADA, adalimumab; BAR, baricitinib; ETA, etanercept; TOC, tocilizumab; TOF, tofacitinib; UPA, upadacitinib.