Poster number: CO210

Real-World Effectiveness and Treatment Patterns of Venetoclax-based Regimens among Patients with Chronic Lymphocytic Leukemia (CLL) Treated in Community Settings

Marjan Zakeri[1], Nnadozie Emechebe[2], Rozina Chowdhery[3], Dureshahwar Jawaid[2], Hasan Alhasani[2], Beenish S Manzoor[2*]

[1]University of Houston, Houston, TX, USA; [2]AbbVie Inc., North Chicago, IL, USA; [3]Northwest Cancer Centers, Crown Point, IN, USA.
*Presenting author

OBJECTIVE

To assess frontline (1L) and second-line (2L) CLL real-world (RW) treatment patterns and outcomes in patients with chronic lymphocytic leukemia (CLL) in community settings

CONCLUSIONS

This study is among the first studies in a burgeoning body of evidence to report RW effectiveness in venetoclax-based regimens, including median duration of therapy

Venetoclax-based regimens are effective in RW community settings, as seen with low rates of subsequent treatment initiation and significantly prolonged time to next treatment or death (TTNT-D)

This study confirms the RW benefit of venetoclax-based regimens as demonstrated in clinical trials with respect to sustained treatment-free remission

For additional information or to obtain a PDF of this poster

Scan QR code to download an electronic version of this presentation and other AbbVie ISPOR 2023 scientific presentations:

https://abbvie1.outsystemsenterprise.com/GMAEventPublications/Assets.aspx?ConferenceId=574

QR code expiration: 07 April 2024

To submit a medical question, please visit www.abbviemedinfo.com

Venetoclax is developed in a collaboration between AbbVie Inc. and Genentech Inc. AbbVie funded the study and participated in the study design, research, analysis, data collection, interpretation of the data, reviewing, and approval of the publication. All authors had access to relevant data and participated in the drafting, review, and approval of this publication. No honoraria or payments were made for authorship.

Medical writing support was provided by Joann Hettasch, PhD, of Fishawack Facilitate Ltd, part of Fishawack Health, and funded by AbbVie Inc.

Disclosures

MZ: nothing to disclose; **BSM, HA, DJ, NE:** employees of AbbVie, Inc. and may hold stock or stock options; **RC:** nothing to disclose.

References

1. Stilgenbauer S, et al. *Lancet Oncol.* 2016;17(6):768–778.
2. Venetoclax (Venclexta) Tablets. Available at: https://www.fda.gov/drugs/resources-information-approved-drugs/venetoclax-venclexta-tablets. Accessed March 6, 2023.
3. National Cancer Institute. Combination Therapy with Venetoclax Approved for Chronic Lymphocytic Leukemia. Available at: https://www.cancer.gov/news-events/cancer-currents-blog/2019/venetoclax-obinutuzumab-fda-approval-cll-sll. Accessed March 6, 2023.
4. Seymour JF, et al. *N Engl J Med.* 2018;378:1107–20.
5. FDA approves venetoclax for CLL and SLL. Available at: https://www.fda.gov/drugs/resources-information-approved-drugs/fda-approves-venetoclax-cll-and-sll. Accessed March 6, 2023.
6. Fischer K, et al. *N Engl J Med.* 2019;380:2225–36.

Presented at the ISPOR 2023 May 7-10; Boston, Massachusetts, USA

abbvie

INTRODUCTION

- In 2016, based on the results of a Phase II trial reporting 12-month progression-free survival (PFS) of 72% (95%CI 61.8–79.8)[1], venetoclax, a selective BCL2 inhibitor, received FDA approval for the treatment of patients with chronic lymphocytic leukemia (CLL) with a 17p deletion[2]
- In 2018, the FDA expanded the approval to include treatment of patients whose cancers have progressed after at least one previous treatment with or without the 17p deletion,[3] based on the MURANO trial results, which demonstrated that venetoclax plus rituximab (VenR) yielded longer rates of 2-year PFS than bendamustine plus rituximab (BR) in patients with CLL[4]
- Finally, in May 2019, the FDA approved venetoclax as a chemotherapy-free combination regimen with obinutuzumab for 1L therapy of patients with CLL.[5] This approval was based on the results which demonstrated that the combination of venetoclax plus obinutuzumab (VenO) resulted in longer PFS than chlorambucil-obinutuzumab (ClbO) in untreated patients with CLL[6]
- Although clinical trials have shown that venetoclax regimens substantially improve PFS in patients with CLL, there is currently little data available on the effectiveness of these regimens in RW clinical practice settings

METHODS

Study design and data source

- Retrospective, observational study using data from the ConcertAI RWD360 database
 - De-identified electronic health records (EHR) are from community practices across ~35 states in the US
- De-identified EHR are derived from patient clinical charts
- Eligible patients
 - Were diagnosed with CLL and were treated in the community setting
 - Received a venetoclax-based regimen in 1L or 2L on or before April 30, 2021 in the US
 - Had ≥12 months of follow-up after venetoclax initiation

Venetoclax-based treatment regimens

Regimen	Indication
Venetoclax + obinutuzumab (VenO)	1L CLL
Venetoclax + rituximab (VenR)	R/R CLL
Venetaclax monotherapy (VenMono)	R/R CLL with del(17p)/TP53 mutation

1L, frontline; CLL, chronic lymphocytic leukemia; R/R, relapsed/refractory.

Outcomes

- Median duration of therapy (mDOT); treatment duration was defined as the number of days from the start to end of treatment in each line of therapy
- Median time to next treatment or death (mTTNT-D), defined as the time in days from the date of treatment initiation to the date of start of next line initiation or death, being lost to follow-up, or end of study plus one day. Patients were censored at their last activity date in the database if there was no subsequent line initiation within the follow-up period
- Percentage of patients receiving each venetoclax-based regimen
- Percentage of patients who did not receive subsequent treatment

Data Analysis

- Data were summarized as median values for continuous variables and proportions for categorical variables
- Kaplan-Meier estimates were generated to evaluate TTNT-D

RESULTS

1L therapy

- Median age at 1L initiation was 65.5, 72.0 and 69.5 years for VenO, VenR and VenMono, respectively (**Table 1**)

Table 1. Baseline characteristics

Characteristics	1L			2L		
	VenO (n=76)	VenR (n=4)	VenMono (n=48)	VenO (n=22)	VenR (n=22)	VenMono (n=82)
Mean ± SD [median] age at diagnosis (yrs)	62.9 ± 9.7 [62.0]	64.3 ± 13.4 [61.0]	66.9 ± 9.3 [68.0]	66.6 ± 10.1 [67.0]	60.0 ± 14.2 [57.5]	63.6 ± 11.2 [63.0]
Mean ± SD [median] age at line initiation (yrs)	65.3 ± 9.9 [65.5]	71.0 ± 10.7 [72.0]	70.1 ± 9.3 [69.5]	71.0 ± 9.6 [73.0]	64.7 ± 12.3 [66.5]	69.5 ± 10.3 [67.0]
Mean ± SD [median] time from diagnosis to 1L therapy (months)	29.2 ± 35.6 [17.1]	81.2 ± 66.5 [84.8]	38.8 ± 41.9 [26.1]	—	—	—
Sex, n (%)						
Female	25 (32.9)	3 (75.0)	18 (37.5)	6 (27.3)	4 (18.2)	34 (41.5)
Male	51 (67.1)	1 (25.0)	30 (62.5)	16 (72.7)	18 (81.8)	48 (58.5)
Race, n (%)						
Asian	1 (1.3)	0	0	0	1 (4.5)	1 (1.2)
Black or African American	4 (5.3)	0	7 (14.6)	3 (13.6)	3 (13.6)	7 (8.5)
White	65 (85.5)	3 (75.0)	35 (72.9)	16 (72.7)	13 (59.1)	66 (80.5)
Other or Unknown Race	6 (7.9)	1 (25.0)	6 (12.5)	3 (13.6)	5 (22.7)	8 (9.8)
Ethnicity, n (%)						
Hispanic or Latino	0	0	0	1 (4.5)	1 (4.5)	2 (2.4)
Not Hispanic or Latino	62 (81.6)	3 (75.0)	29 (60.4)	18 (81.8)	16 (72.7)	63 (76.8)
Unknown	14 (18.4)	1 (25.0)	19 (39.6)	3 (13.6)	5 (22.7)	17 (20.7)
Region, n (%)						
Midwest	24 (31.6)	2 (50.0)	19 (39.6)	11 (50.0)	7 (31.8)	26 (31.7)
Northeast	18 (23.7)	2 (50.0)	4 (8.3)	2 (9.1)	5 (22.7)	14 (17.1)
South	34 (44.7)	0	17 (35.4)	8 (36.4)	7 (31.8)	27 (32.9)
West	0	0	6 (12.5)	1 (4.5)	2 (9.1)	11 (13.4)
Missing	0	0	2 (4.2)	0	1 (4.5)	4 (4.9)

VenMono, venetoclax monotherapy; VenO, venetoclax in combination with obinutuzumab; VenR, venetoclax in combination with rituximab; yrs, years.

- Among 128 patients in 1L therapy, VenO was the most common regimen followed by VenMono and VenR (**Figure 1**)

2L therapy

- Among 126 patients in 2L therapy, 62% received a prior-BTKi
- The most frequently prescribed 2L regimen was VenMono (65.1%)

Figure 1. Percentage of patients receiving VenO, VenR, or VenMono as 1L or 2L therapy

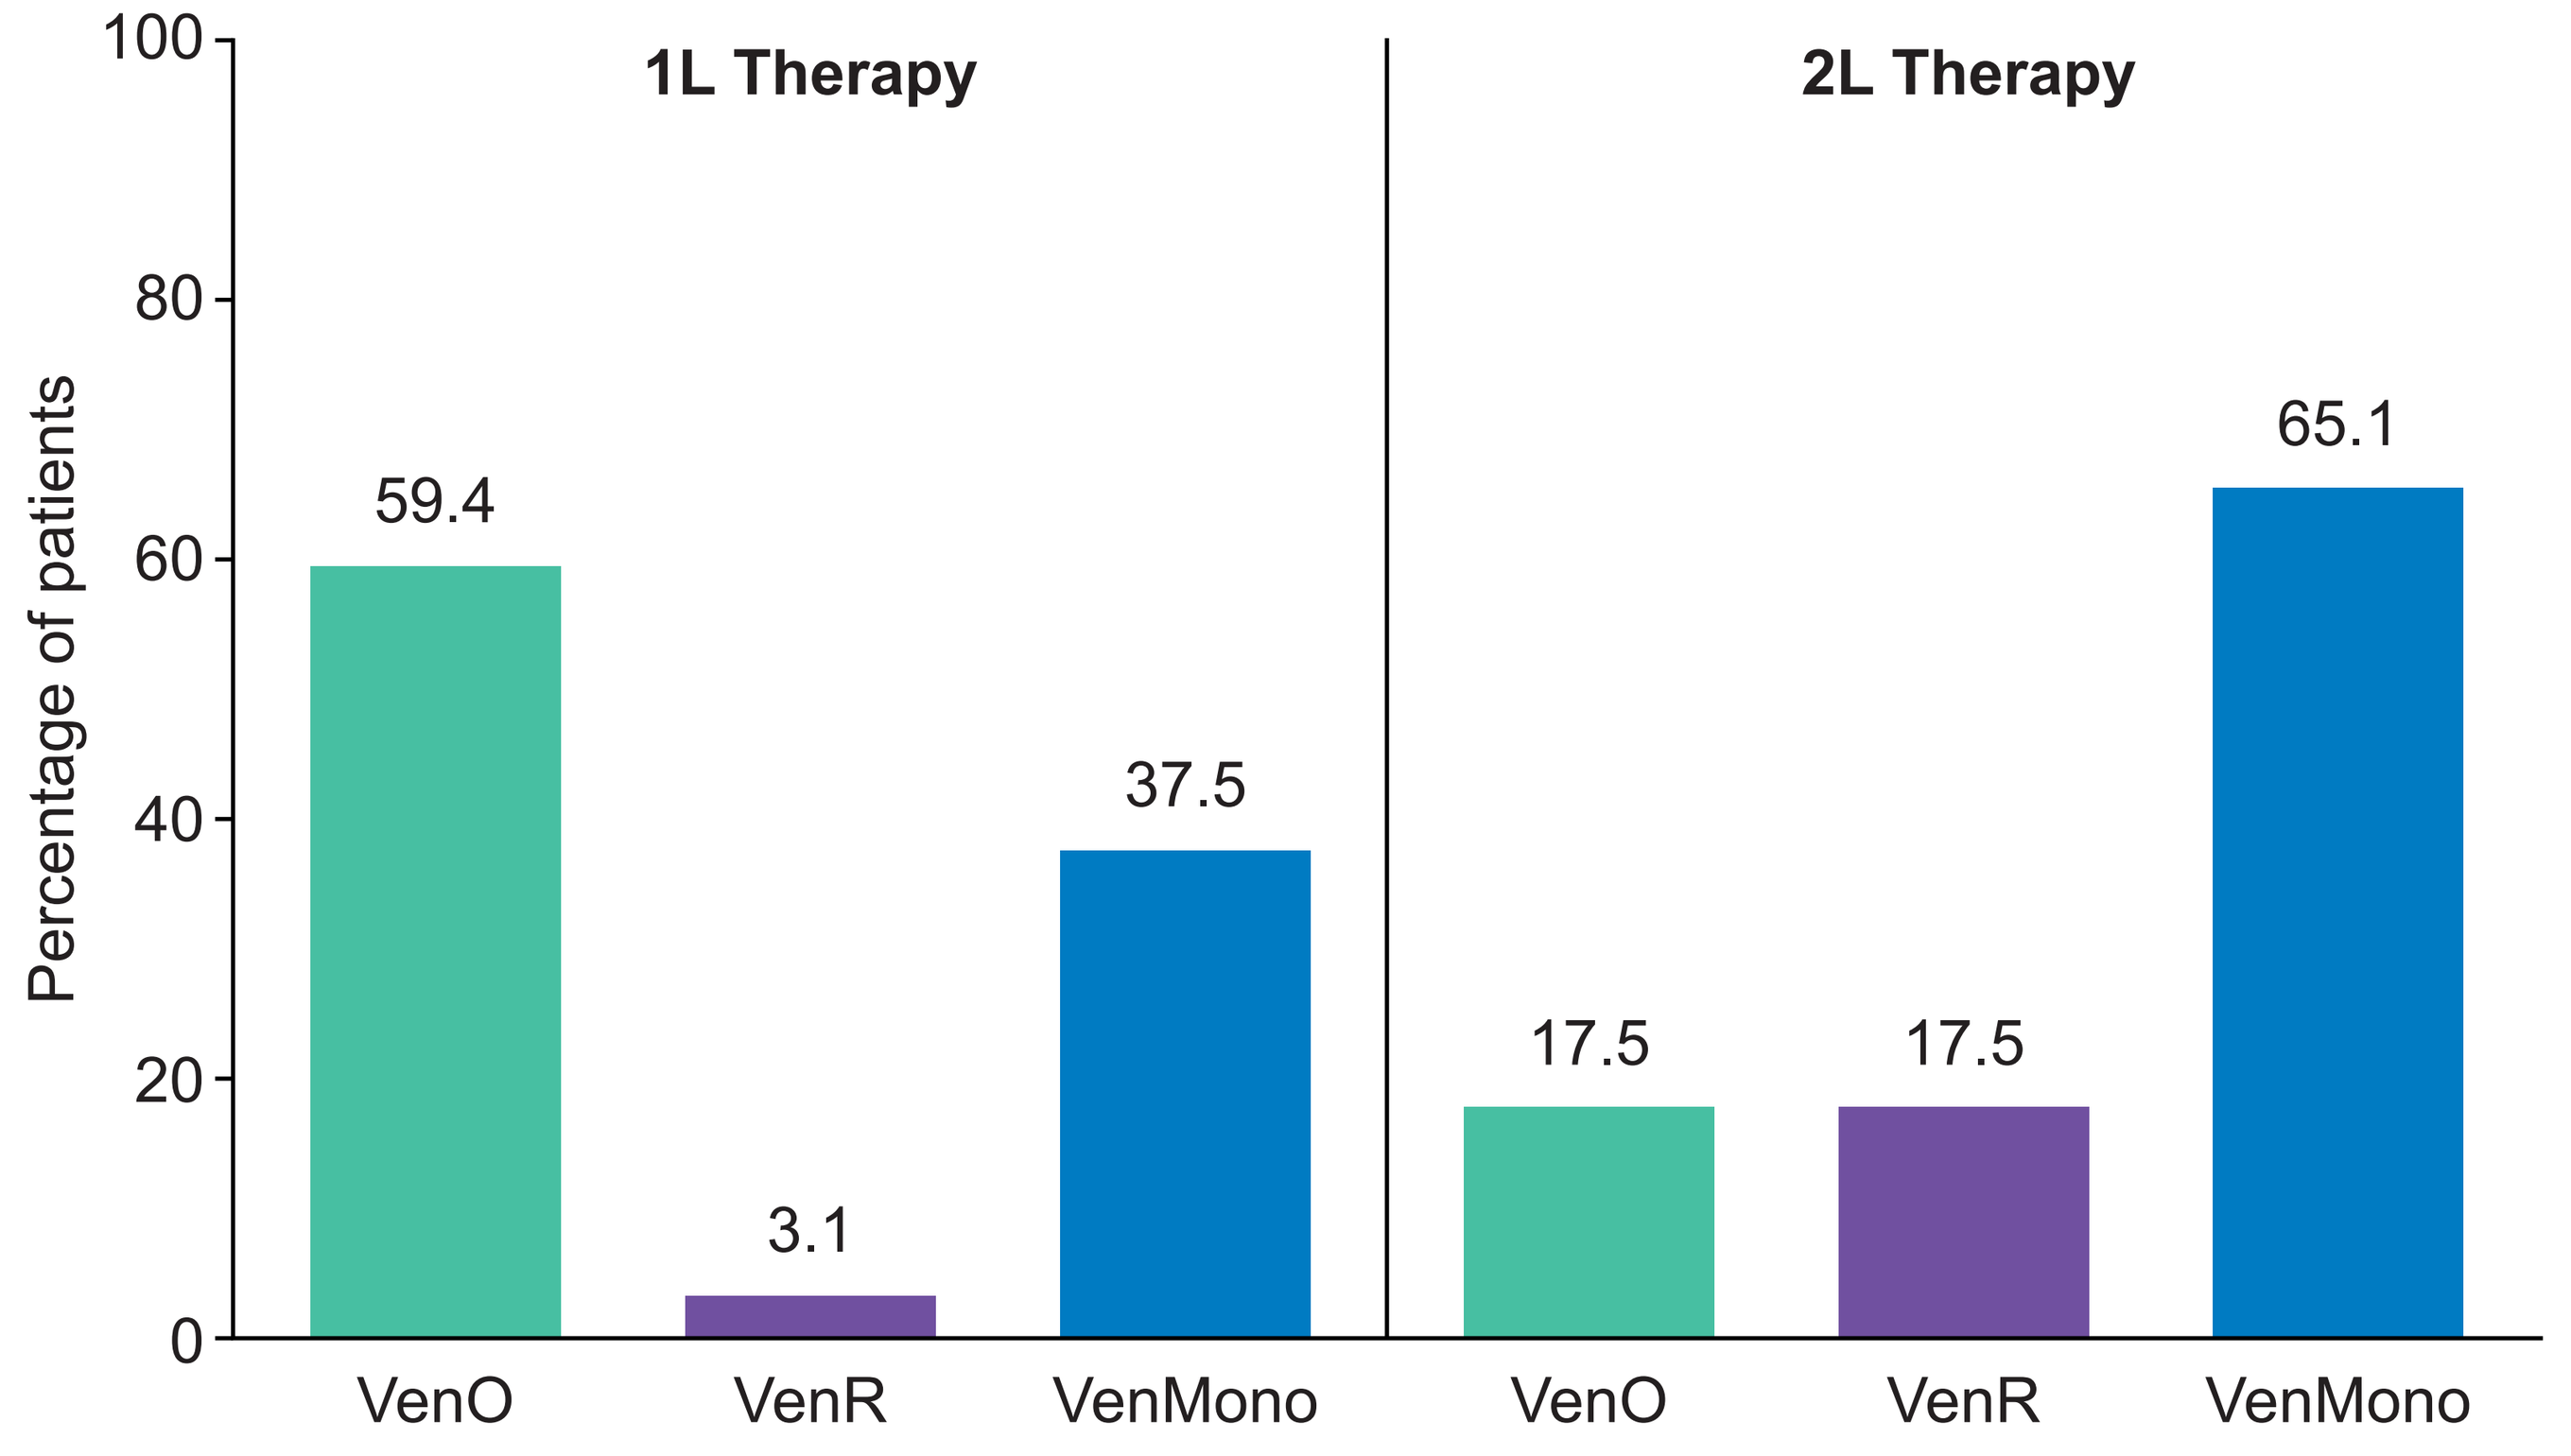
1L Therapy
2L Therapy
Percentage of patients
100
80
60
40
20
0
59.4
3.1
37.5
17.5
17.5
65.1
VenO
VenR
VenMono
VenO
VenR
VenMono

VenMono, venetoclax monotherapy; VenO, venetoclax in combination with obinutuzumab; VenR, venetoclax in combination with rituximab.

- Most patients did not receive subsequent treatment (73.4%), driven by patients on VenO (93.4%)
- Median duration of therapy (mDOT) was 12.7, 19.9, and 3.2 months for VenO, VenR and VenMono, respectively (**Figure 2**)
- At median follow-up (mFU) of 23.1 months, the median TTNT-D was not reached for patients on the VenO and VenR regimens
- Median TTNT-D for VenMono was 13.5 months

Figure 2. TTNT-D by regimen in 1L

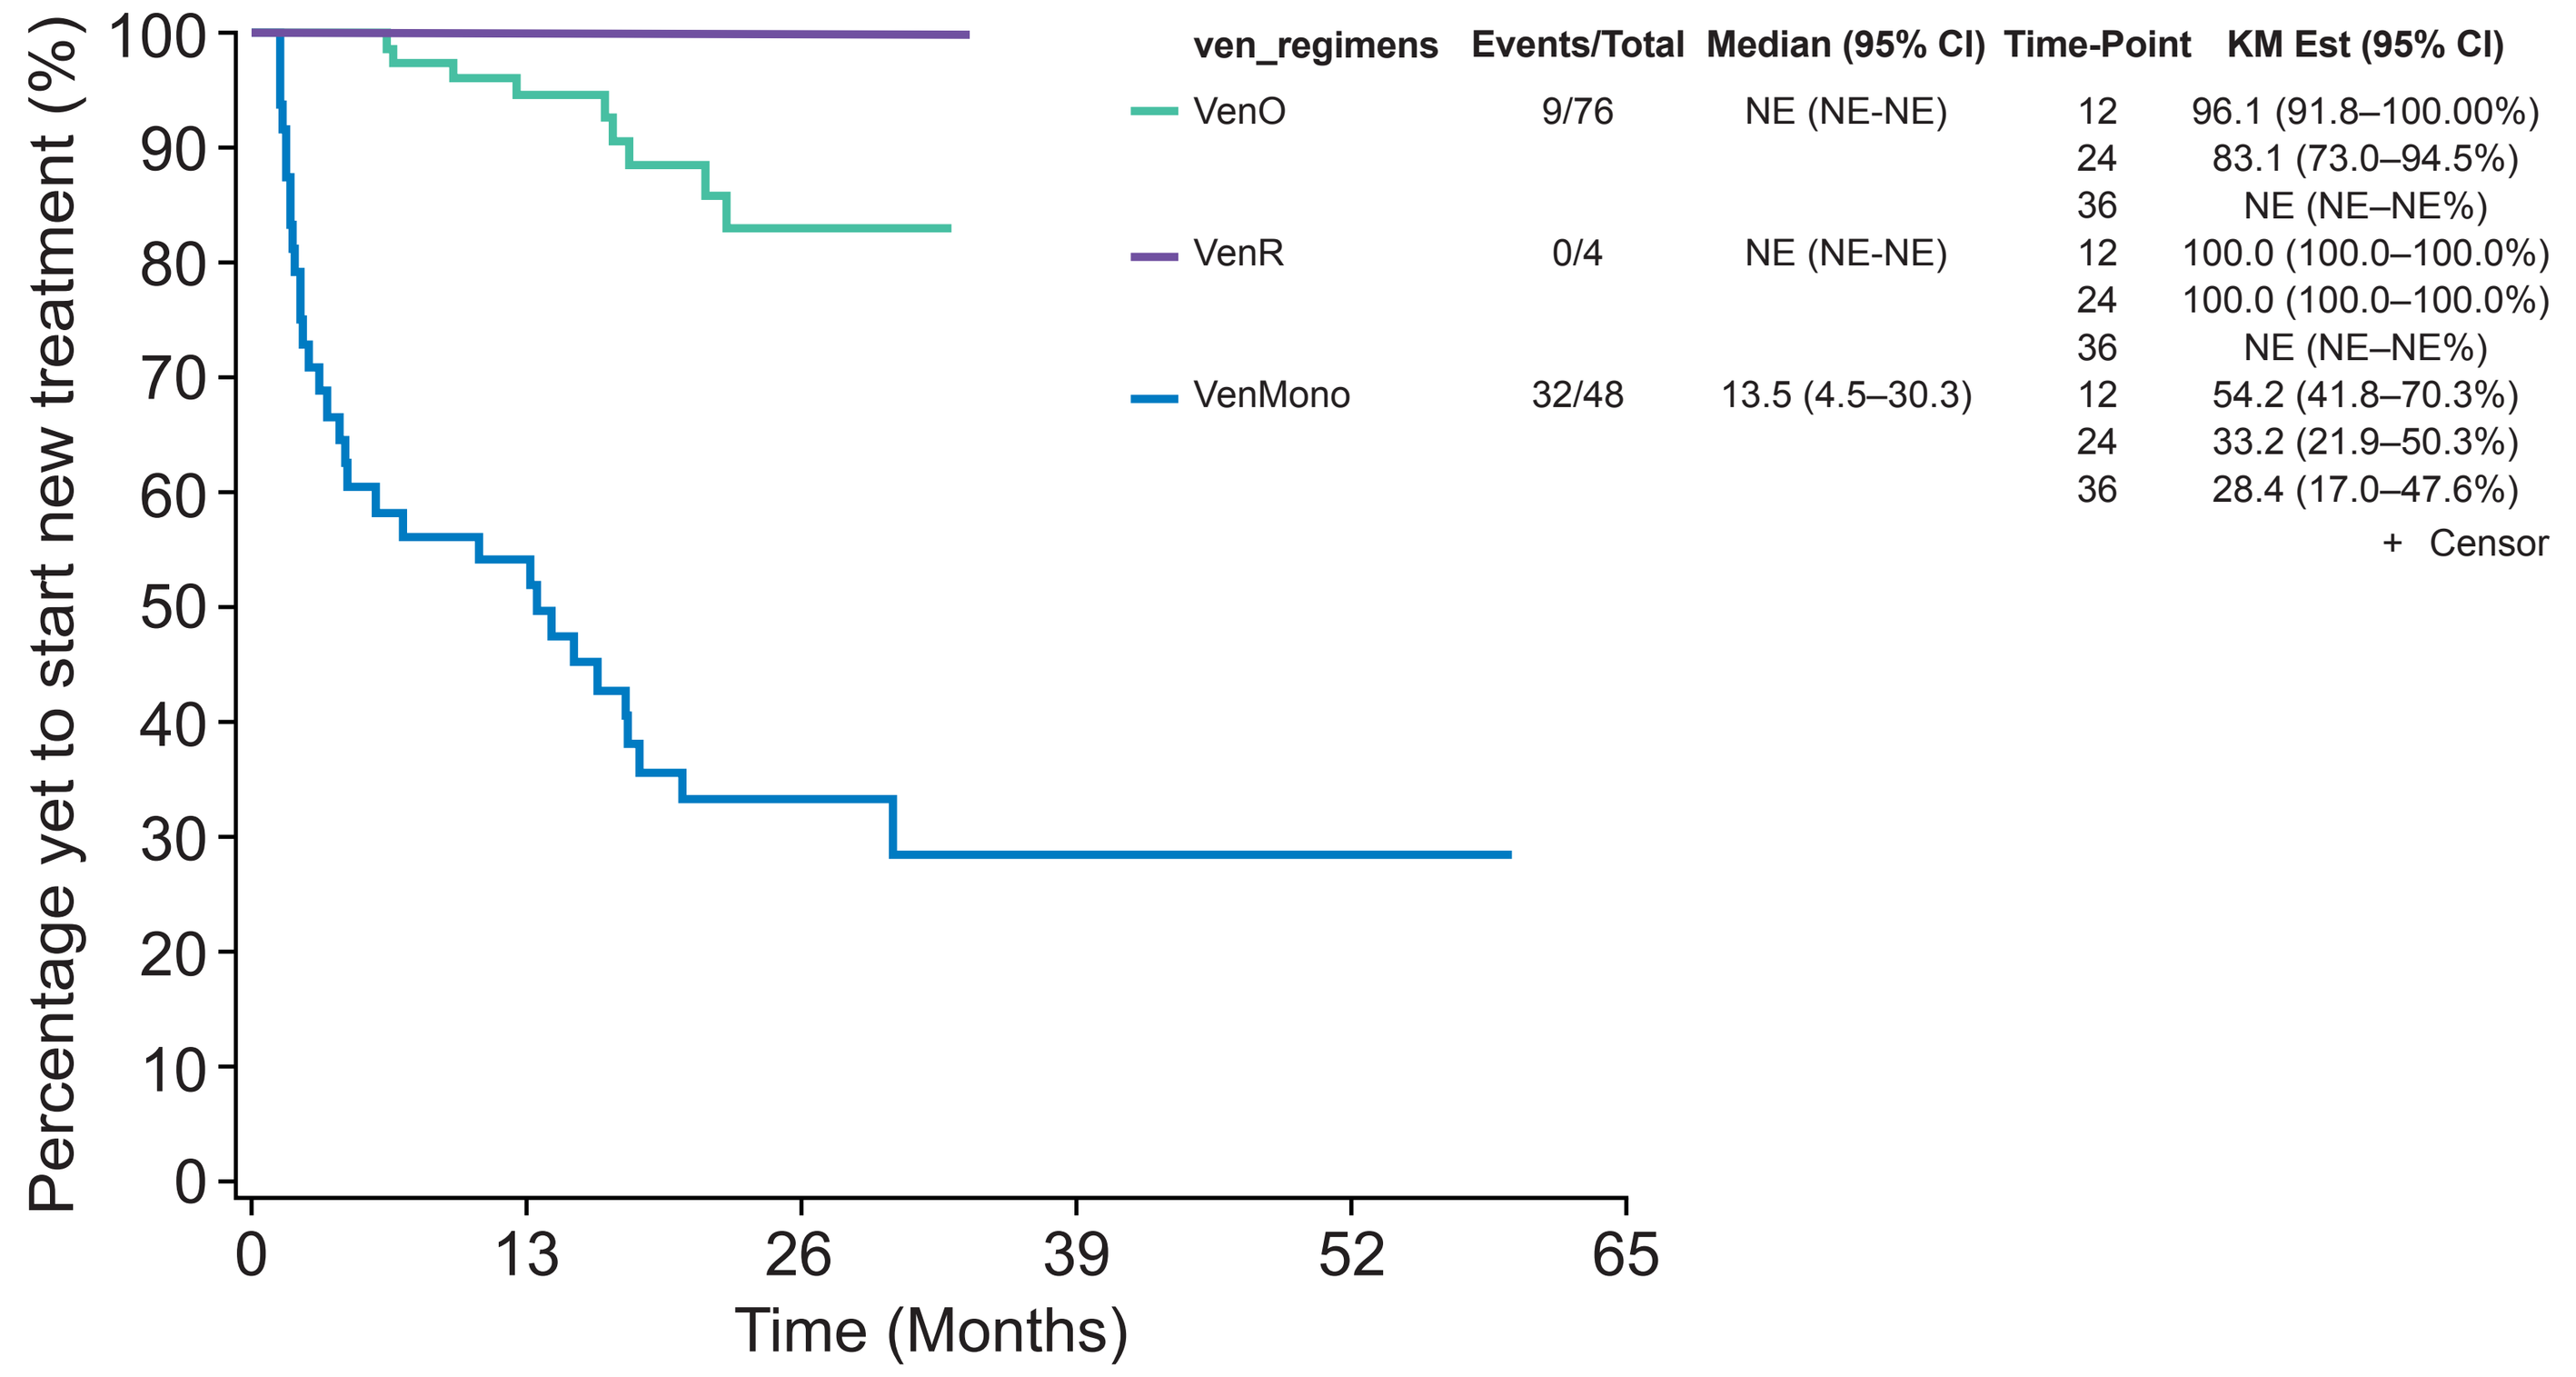
Percentage yet to start new treatment (%)
Time (Months)
0
13
26
39
52
65
ven_regimens
Events/Total
Median (95% CI)
Time-Point
KM Est (95% CI)
VenO 9/76 NE (NE-NE) 12 96.1 (91.8–100.00%) 24 83.1 (73.0–94.5%) 36 NE (NE–NE%)
VenR 0/4 NE (NE-NE) 12 100.0 (100.0–100.0%) 24 100.0 (100.0–100.0%) 36 NE (NE–NE%)
VenMono 32/48 13.5 (4.5–30.3) 12 54.2 (41.8–70.3%) 24 33.2 (21.9–50.3%) 36 28.4 (17.0–47.6%)
+ Censor

	VenO	VenR	VenMono
mDOT, months	12.7	19.9	3.2
mFU from venetoclax initiation, months	20.0	25.7	28.0

CI, confidence interval; KM, Kaplan-Meyer; mDOT, median duration of therapy; mFU, median follow-up; NE, not estimable; ven, venetoclax; VenMono, venetoclax monotherapy; VenO, venetoclax in combination with obinutuzumab; VenR, venetoclax in combination with rituximab; yrs, years.

- Similar to 1L therapy, 67.5% of patients did not receive additional treatment
- mDOT was 22.1, 12.1, and 9.5 months for VenR, VenO and VenMono, respectively (**Figure 3**)
- With a mFU of 27.5 months, median TTNT-D was not reached for patients on VenR and VenO, median TTNT-D was 27.9 months for VenMono

Figure 3. TTNT-D by regimen in the 2L

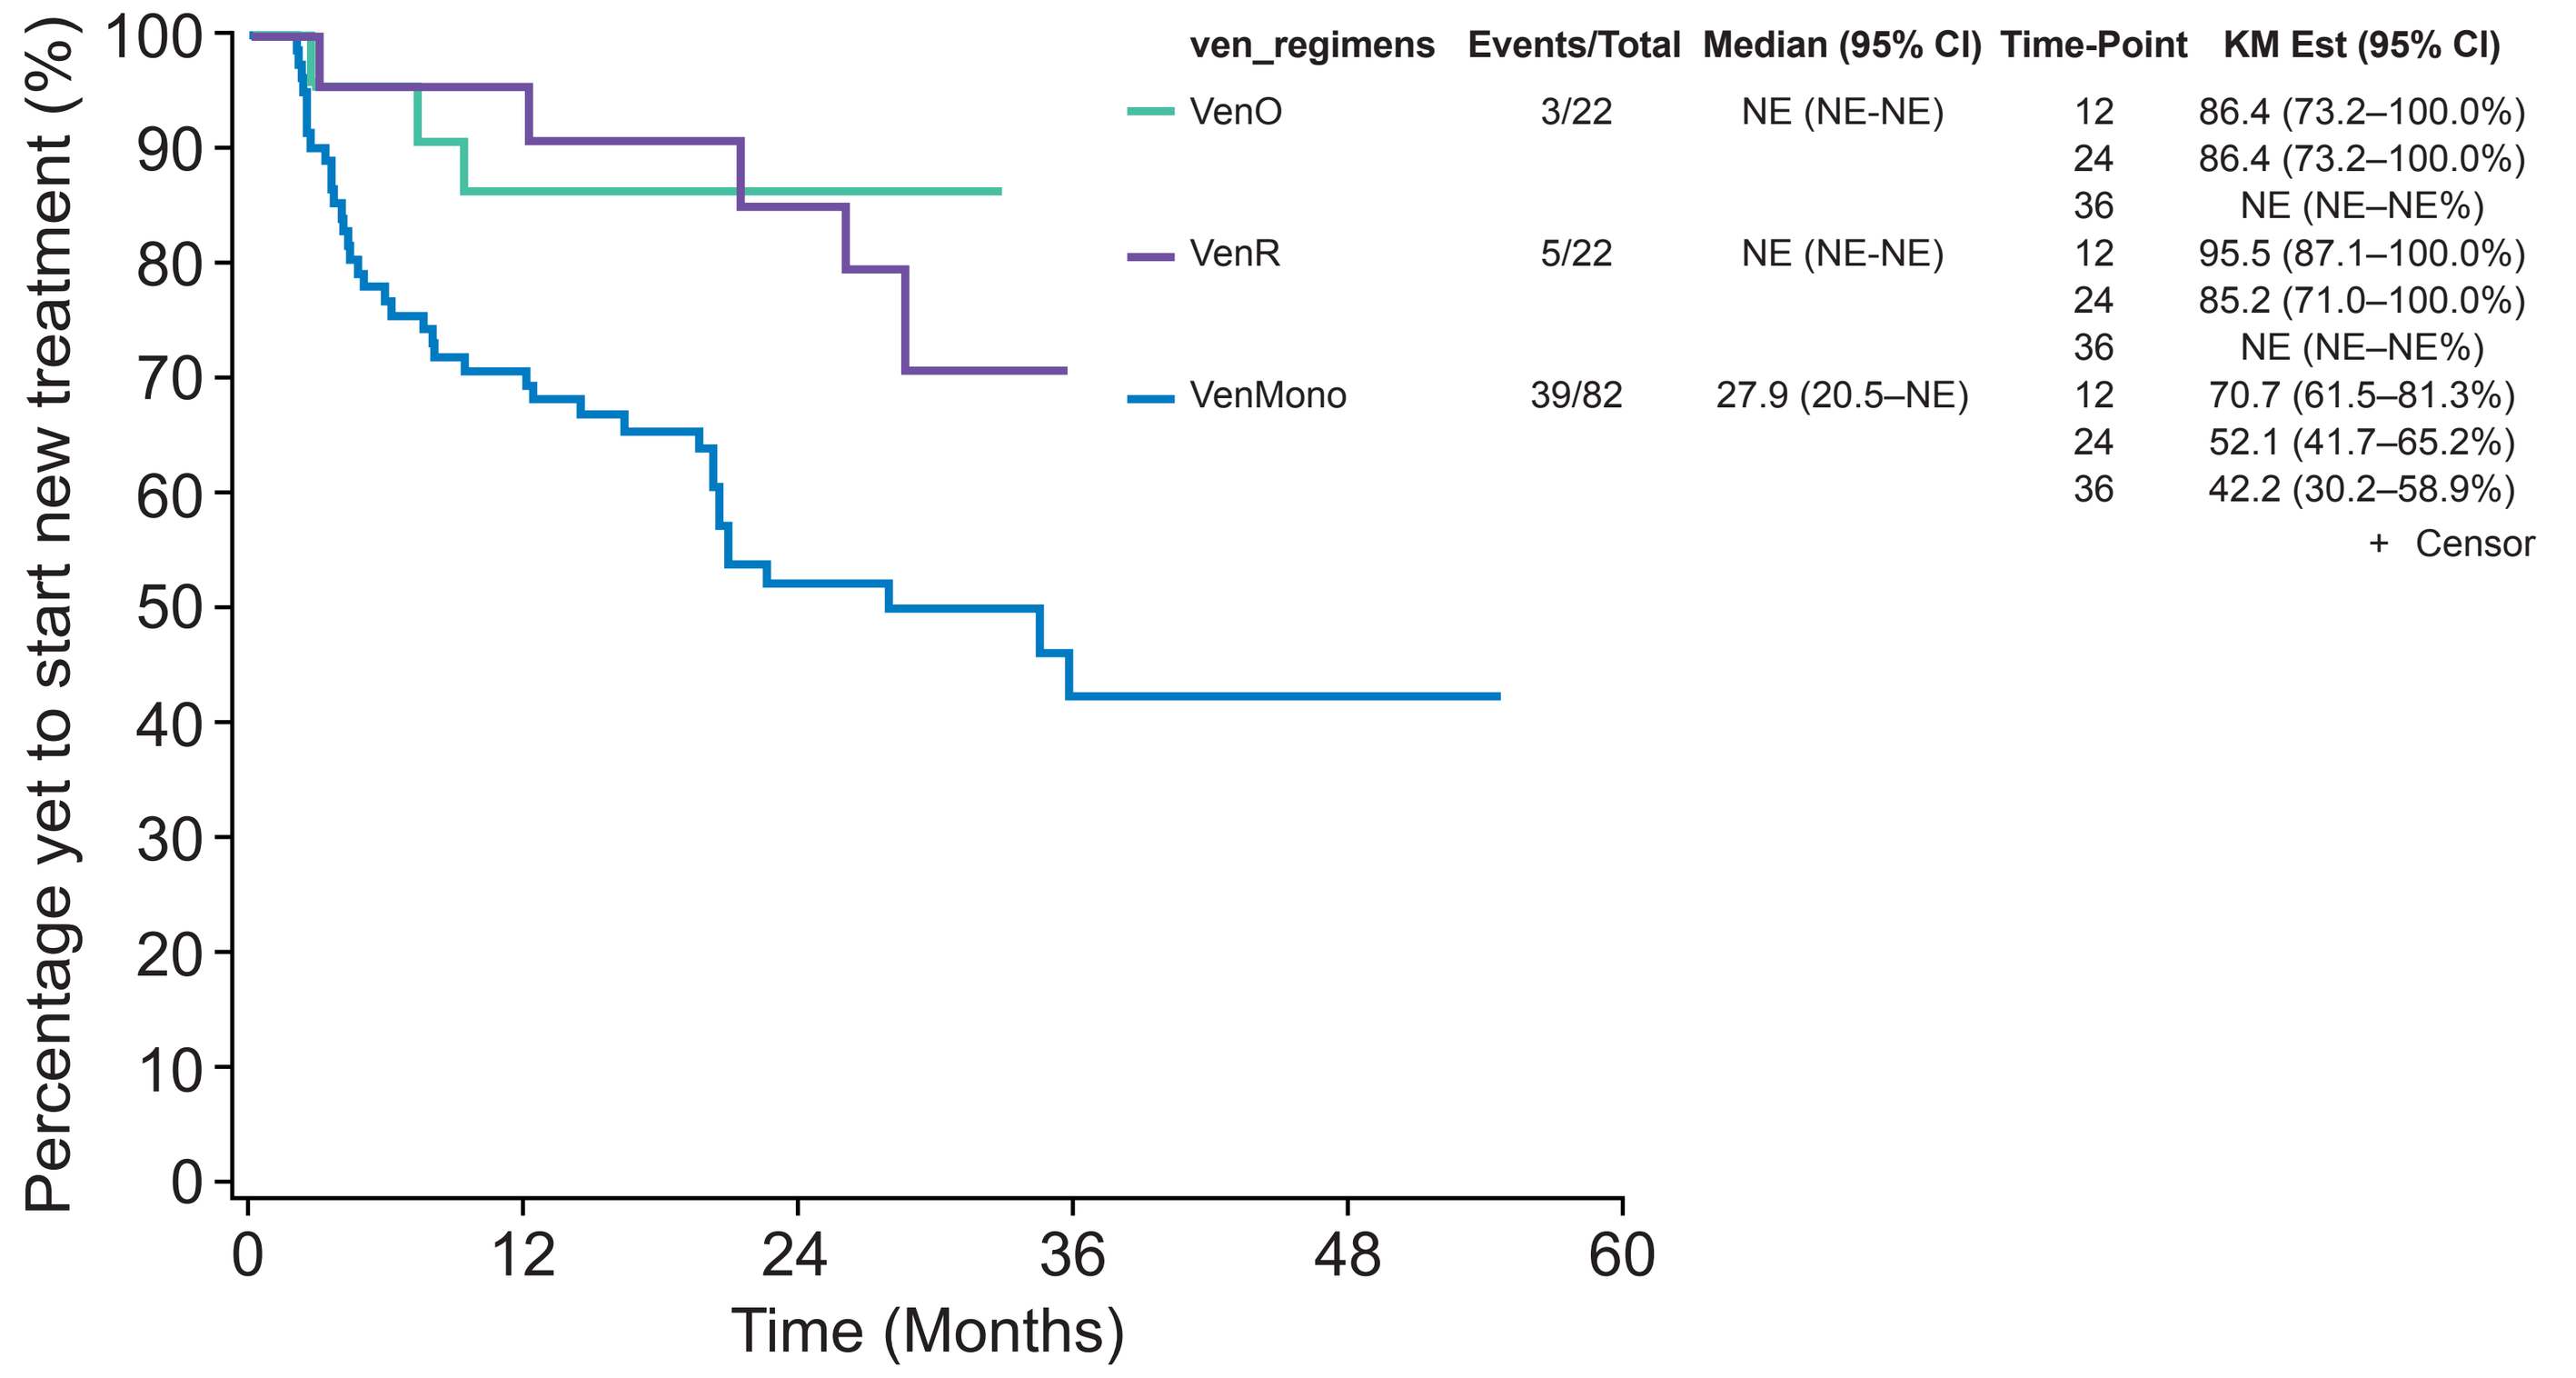
Percentage yet to start new treatment (%)
Time (Months)
0
12
24
36
48
60
ven_regimens
Events/Total
Median (95% CI)
Time-Point
KM Est (95% CI)
VenO 3/22 NE (NE-NE) 12 86.4 (73.2–100.0%) 24 86.4 (73.2–100.0%) 36 NE (NE–NE%)
VenR 5/22 NE (NE-NE) 12 95.5 (87.1–100.0%) 24 85.2 (71.0–100.0%) 36 NE (NE–NE%)
VenMono 39/82 27.9 (20.5–NE) 12 70.7 (61.5–81.3%) 24 52.1 (41.7–65.2%) 36 42.2 (30.2–58.9%)
+ Censor

	VenO	VenR	VenMono
mDOT, months	12.1	22.1	9.5
mFU time from venetoclax initiation, months	20.8	28.9	26.7

CI, confidence interval; KM, Kaplan-Meyer; mDOT, median duration of therapy; mFU, median follow-up; NE, not estimable; ven, venetoclax; VenMono, venetoclax monotherapy; VenO, venetoclax in combination with obinutuzumab; VenR, venetoclax in combination with rituximab; yrs, years.

Limitations

As with well-documented limitations for all real-world data sources, the information used in this study was collected as part of routine clinical practice, not for research purposes, and therefore selection and information biases may exist, including missing data and orders for oral drugs, which would require evidence of filled prescriptions or details on use of drug in physician notes. Furthermore, the generalizability of the results is limited to patients within the ConcertAI RWD360 database and could be impacted by smaller sample sizes.