Poster code: RWD57

Real-world treatment patterns of elderly patients with locally advanced squamous cell carcinoma of the head and neck in the US

E. Lu,[1] C. Frenkel,[2] M. Danese,[3] S. Bobiak,[1] S. Salmio,[4] N. Saba[5]

[1]EMD Serono, Billerica, MA, USA; [2]Atrium Health Levine Cancer Institute, Charlotte, NC, USA; [3]Outcomes Insights, Agoura Hills, CA, USA; [4]The healthcare business of Merck KGaA, Darmstadt, Germany; [5]Winship Cancer Institute of Emory University, Atlanta, GA, USA

SCOPE

- We report baseline demographics, clinical characteristics, and treatment patterns in elderly patients with newly diagnosed locally advanced squamous cell carcinoma of the head and neck (LA SCCHN) using data from the Surveillance, Epidemiology, and End Results (SEER)–Medicare linked database to better understand real-world clinical treatment practice in the US

CONCLUSIONS

- Real-world management of elderly patients with LA SCCHN most often includes concurrent adjuvant or definitive chemoradiotherapy (CRT) with cisplatin
- However, in this database analysis, 17% of patients did not receive any treatment; reasons for this are unknown but may include patient preference or concerns with treatment toxicities
- These data highlight a need for innovative and effective therapies for elderly patients and beyond with LA SCCHN

GET POSTER PDF

Copies of this poster obtained through this quick response (QR) code are for personal use only and may not be reproduced without permission from ISPOR and the authors of this poster

Correspondence: **Ember Lu,** ember.lu@emdserono.com

BACKGROUND

- In the US, head and neck cancer is the ninth most commonly diagnosed cancer, with 66,630 new cases and 14,620 deaths recorded in 2021[1]
- The majority of cases of head and neck cancer (≈90%) are squamous cell carcinomas[2]
- Approximately 60% of patients are diagnosed with LA SCCHN,[3] and standard of care is definitive CRT for unresected patients, or surgery followed by adjuvant radiotherapy (RT) with or without chemotherapy (CT)[3]
- To understand real-world clinical treatment practices, we assessed the treatment patterns in elderly patients with newly diagnosed LA SCCHN in the US population using data from the SEER–Medicare linked database

METHODS

Data source

- Real-world data were extracted from the SEER–Medicare linked database
 - Data from the SEER database include demographic, clinical, and cause-of-death information from several US cancer registries
 - Medicare data include claims for covered healthcare services
 - Personal identifiers were matched between the databases, providing comprehensive medical data for all selected patients
- Due to Medicare eligibility criteria, only patients aged ≥65 years were included in the linked database

Study design

- The study design is presented in **Figure 1**
- Patients were diagnosed between January 2014 and December 2017 and were followed up until diagnosis of subsequent cancer, change in enrollment in Medicare, death, or data cutoff in December 2019 (most recent data available for this analysis)
- Patients were divided into subgroups based on types of anticancer treatment received, and descriptive analyses were conducted
- As deidentified patient data were used, no ethical consent was required

Figure 1. Study design

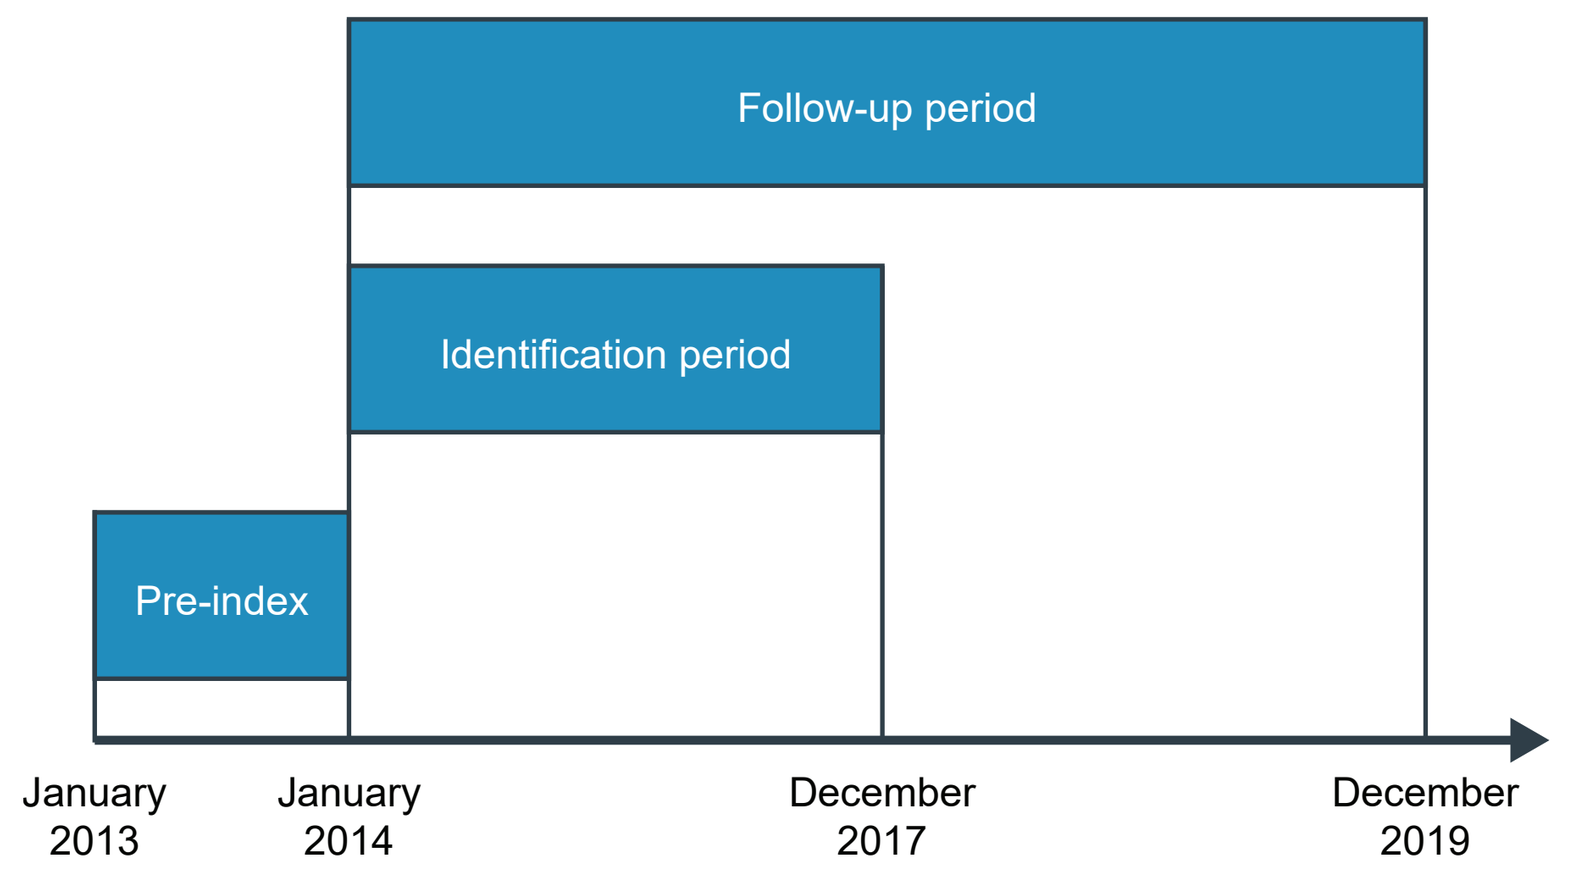

Follow-up period
Identification period
Pre-index
January 2013
January 2014
December 2017
December 2019

Inclusion and exclusion criteria

Inclusion criteria

- Patients were diagnosed with stage III, IVA, or IVB LA SCCHN in the oropharynx, oral cavity, larynx, or hypopharynx as their first and primary cancer
- Patients were required to have ≥1 year of Medicare eligibility prior to the diagnosis of their cancer; thus, patients included in the study were aged ≥66 years

Exclusion criterion

- The first diagnosis was made at autopsy or by death certificate

RESULTS

Patient demographics

- A total of 2,776 patients met the eligibility criteria (**Figure 2**)
- Patient demographics for the overall population and subgroups are presented in **Table 1**
- In the overall population:
 - Mean patient age was 73.6 years (standard deviation, 6.8 years), and 74.0% and 26.0% were male and female, respectively
 - Patients' race and ethnicity were reported as follows: 88.7% were White, 6.9% were Black, and 2.9% were Asian
 - Cancer was stage III in 27.1% of all patients, stage IVA in 65.9%, and stage IVB in 7.0%
 - Cancer of the oropharynx was the most common tumor type (48.6%), followed by oral cavity (25.1%), larynx (20.3%), and hypopharynx (5.9%)

Figure 2. Flow diagram of patient selection

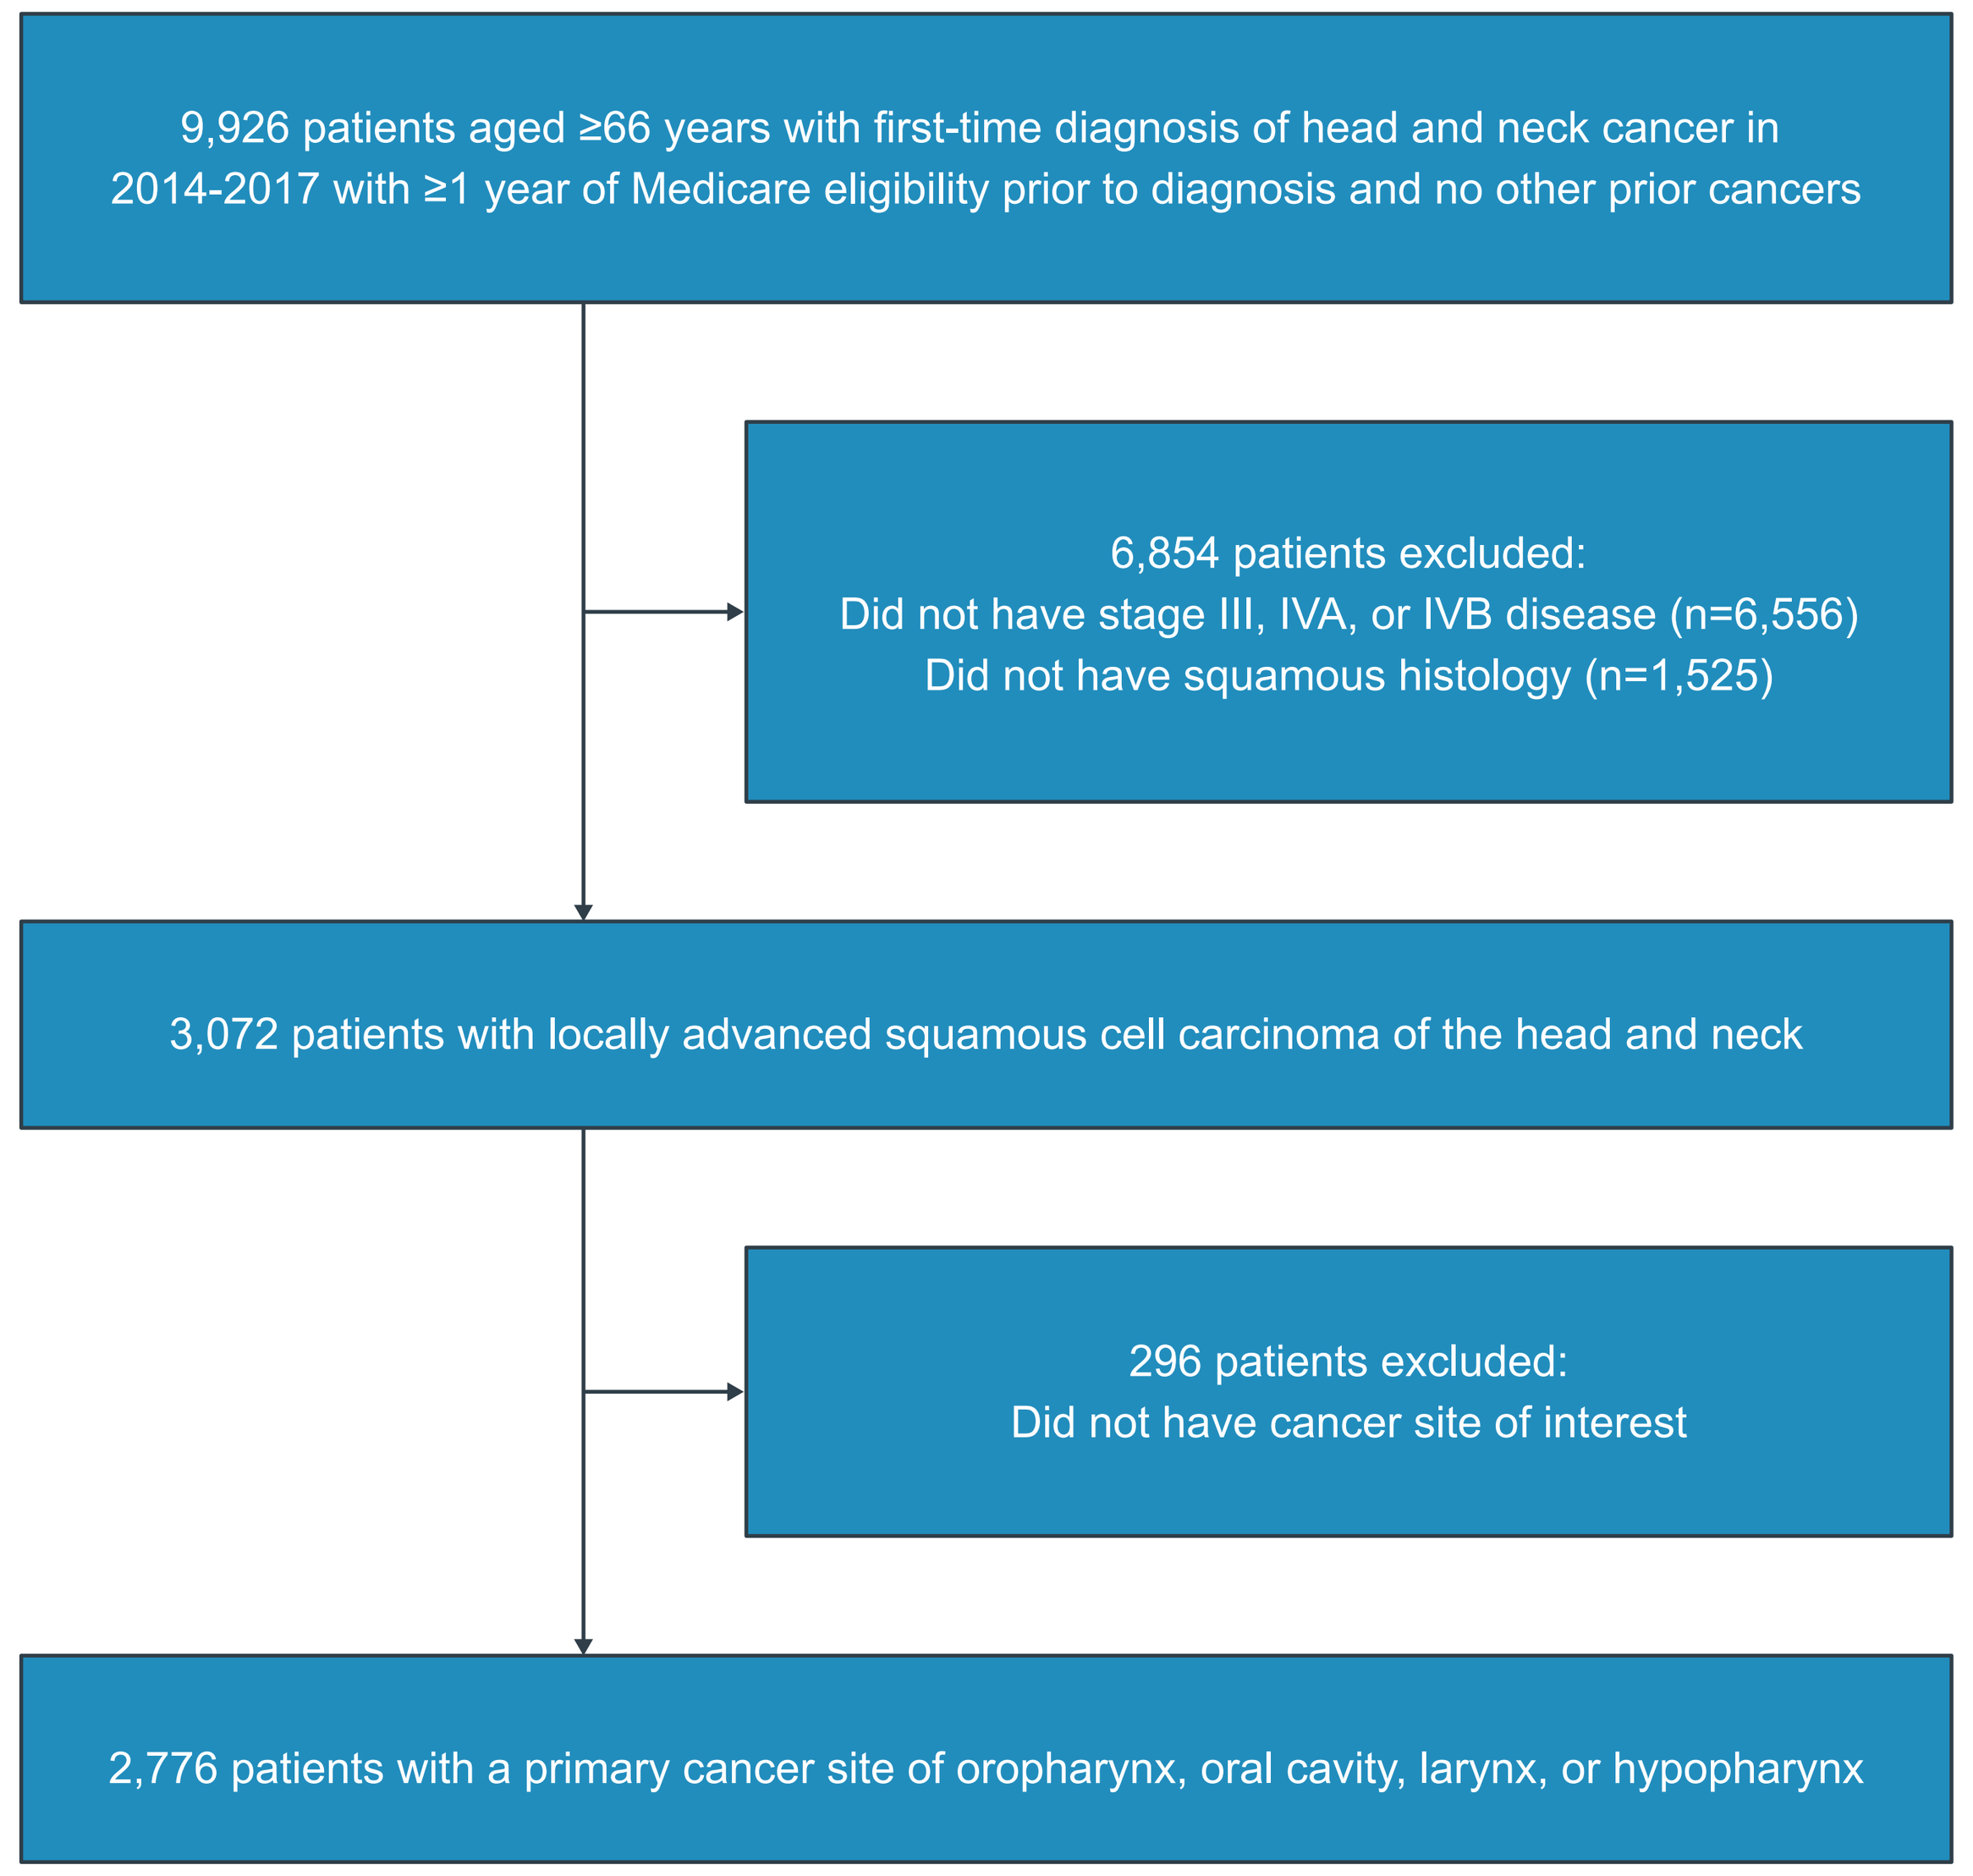

9,926 patients aged ≥66 years with first-time diagnosis of head and neck cancer in 2014-2017 with ≥1 year of Medicare eligibility prior to diagnosis and no other prior cancers
6,854 patients excluded:
Did not have stage III, IVA, or IVB disease (n=6,556)
Did not have squamous histology (n=1,525)
3,072 patients with locally advanced squamous cell carcinoma of the head and neck
296 patients excluded:
Did not have cancer site of interest
2,776 patients with a primary cancer site of oropharynx, oral cavity, larynx, or hypopharynx

Table 1. Patient demographics at baseline

	All patients (N=2,776)	Definitive non-surgical treatment (n=1,487)	Primary resection (n=753)
Age, mean (SD), years	73.6 (6.8)	73.3 (6.7)	73.7 (6.6)
Age, years, n (%)			
66-69	976 (35.2)	549 (36.9)	251 (33.3)
70-74	800 (28.8)	422 (28.4)	233 (30.9)
75-79	494 (17.8)	277 (18.6)	120 (15.9)
80-84	273 (9.8)	127 (8.5)	85 (11.3)
≥85	233 (8.4)	112 (7.5)	64 (8.5)
Sex, n (%)			
Male	2,055 (74.0)	1,145 (77.0)	490 (65.1)
Female	721 (26.0)	342 (23.0)	263 (34.9)
Race and ethnicity, n (%)			
White	2,461 (88.7)	1,321 (88.8)	689 (91.5)
Black	191 (6.9)	99 (6.7)	36 (4.8)
Asian	80 (2.9)	40 (2.7)	NR
Other	44 (1.6)	27 (1.8)	NR
Disease stage, n (%)			
III	752 (27.1)	416 (28.0)	198 (26.3)
IVA	1,829 (65.9)	962 (64.7)	523 (69.5)
IVB	195 (7.0)	109 (7.3)	32 (4.2)
Cancer site, n (%)			
Oropharynx	1,349 (48.6)	877 (59.0)	258 (34.3)
Oral cavity	698 (25.1)	146 (9.8)	402 (53.4)
Larynx	564 (20.3)	349 (23.5)	82 (10.9)
Hypopharynx	165 (5.9)	115 (7.7)	11 (1.5)

A total of 63 patients received CT alone, and 473 patients received no treatment. Values <11 or that can be calculated to be <11 are suppressed to comply with NCI privacy rules.
CT, chemotherapy; NR, not reported.

Treatment patterns

- **Figure 3** presents treatment patterns in all patients
 - Definitive non-surgical treatment was administered to 54% of patients (n=1,487)
 - 27% of patients (n=753) had primary resection
 - CT alone was received by 2% of patients (n=63)
 - No treatment was received by 17% of patients (n=473)
- **Figure 4** presents the treatment patterns for all patients by cancer site
- **Figure 5** presents the systemic chemotherapy agents administered to unresected patients who received definitive CRT alone and in those who had primary resection with adjuvant CRT
 - Cisplatin was received by >50% of patients in both groups
 - Cetuximab was received by 28% of unresected patients who received definitive CRT and 27% of patients who had primary resection with adjuvant CRT

Figure 3. Treatment patterns for all patients

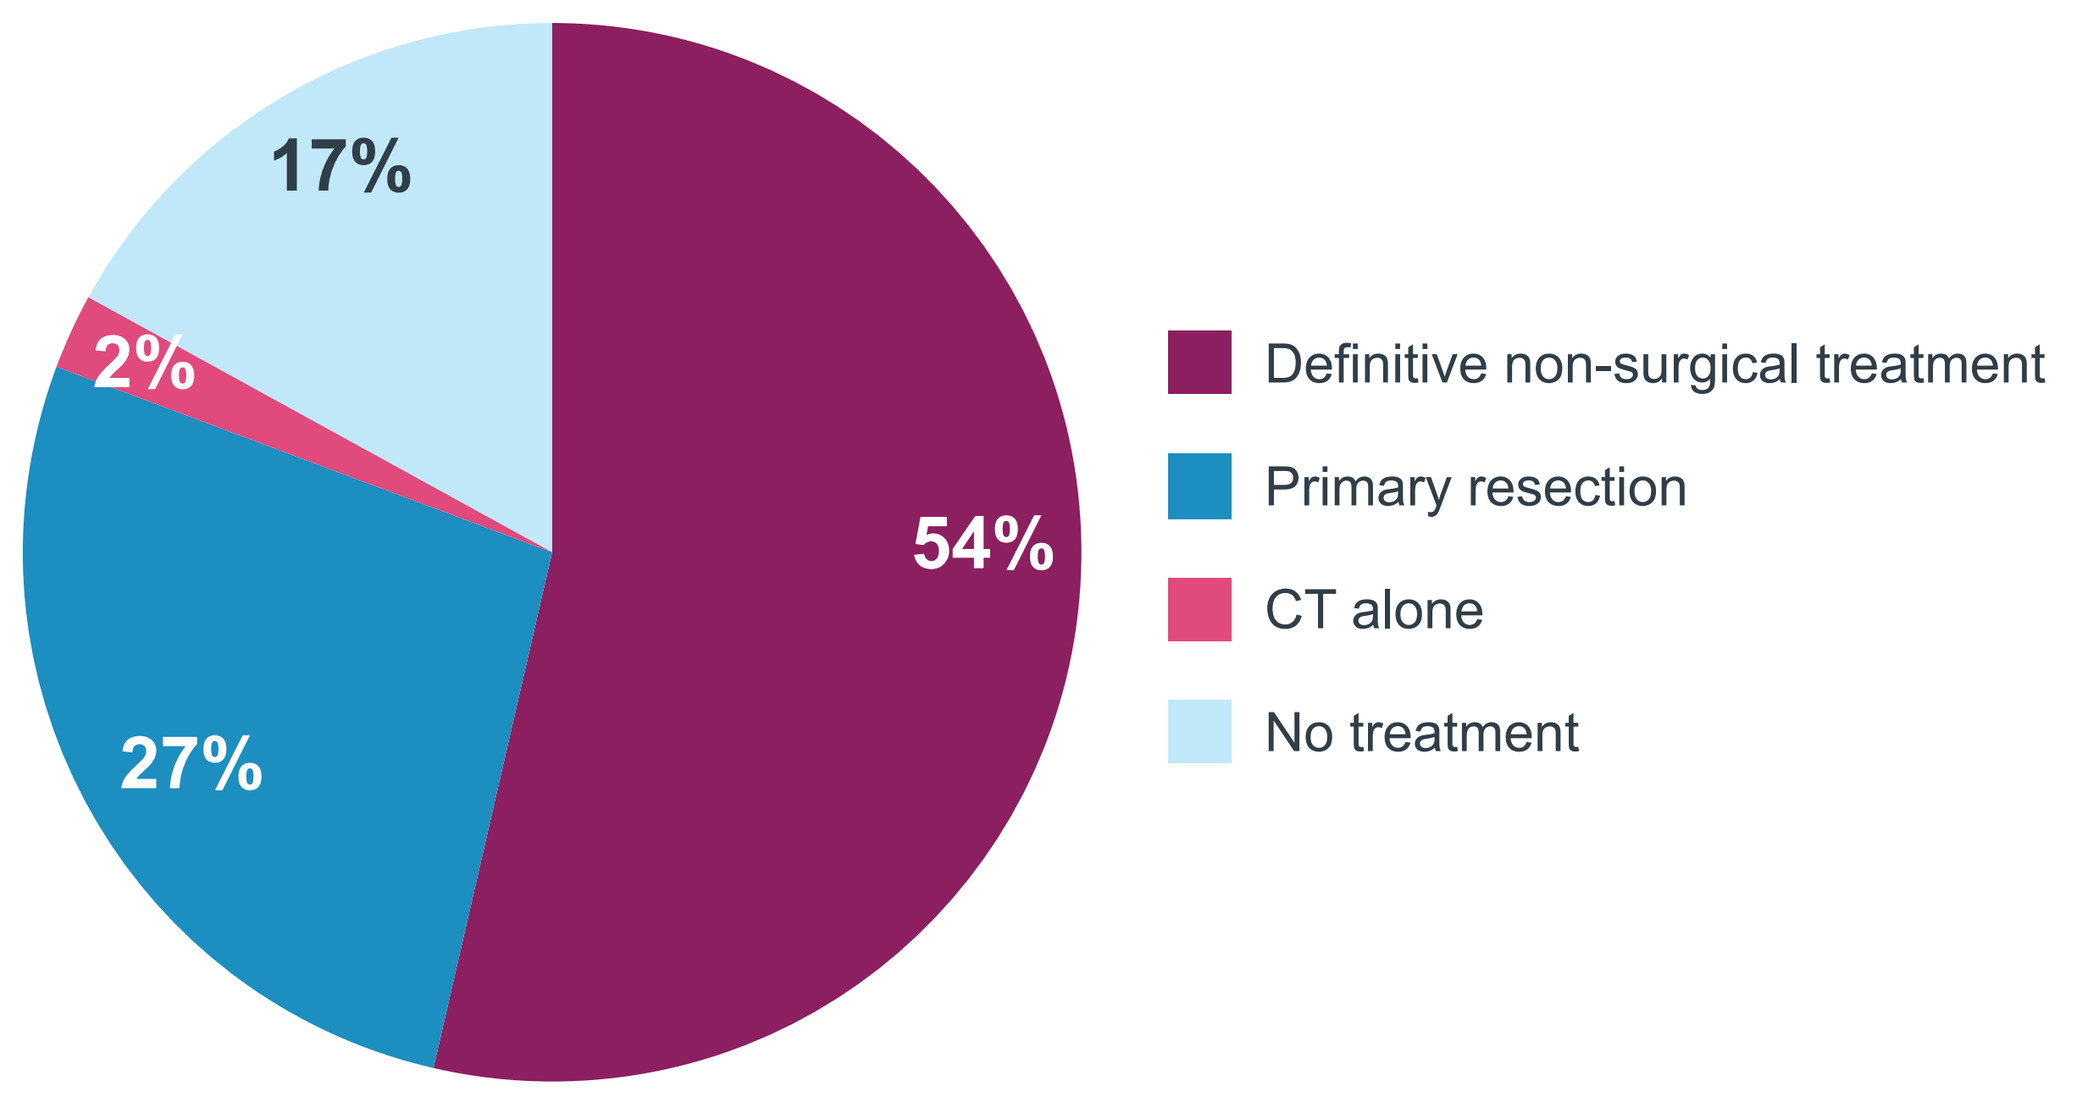

17%
2%
27%
54%
Definitive non-surgical treatment
Primary resection
CT alone
No treatment

Definitive non-surgical treatment (n=1,487)	n (%)	Primary resection (n=753)	n (%)
CRT alone	1,076 (72.4)	Surgery followed by CRT	276 (36.7)
RT alone	289 (19.4)	Surgery alone	231 (30.7)
CT followed by RT	102 (6.9)	Surgery followed by RT	221 (29.3)
RT followed by CT	20 (1.3)	Other	25 (3.3)

CRT, concurrent chemoradiotherapy; CT, chemotherapy; LA SCCHN, locally advanced squamous cell carcinoma of the head and neck; RT, radiotherapy.

Figure 4. Treatment patterns for all patients by cancer site

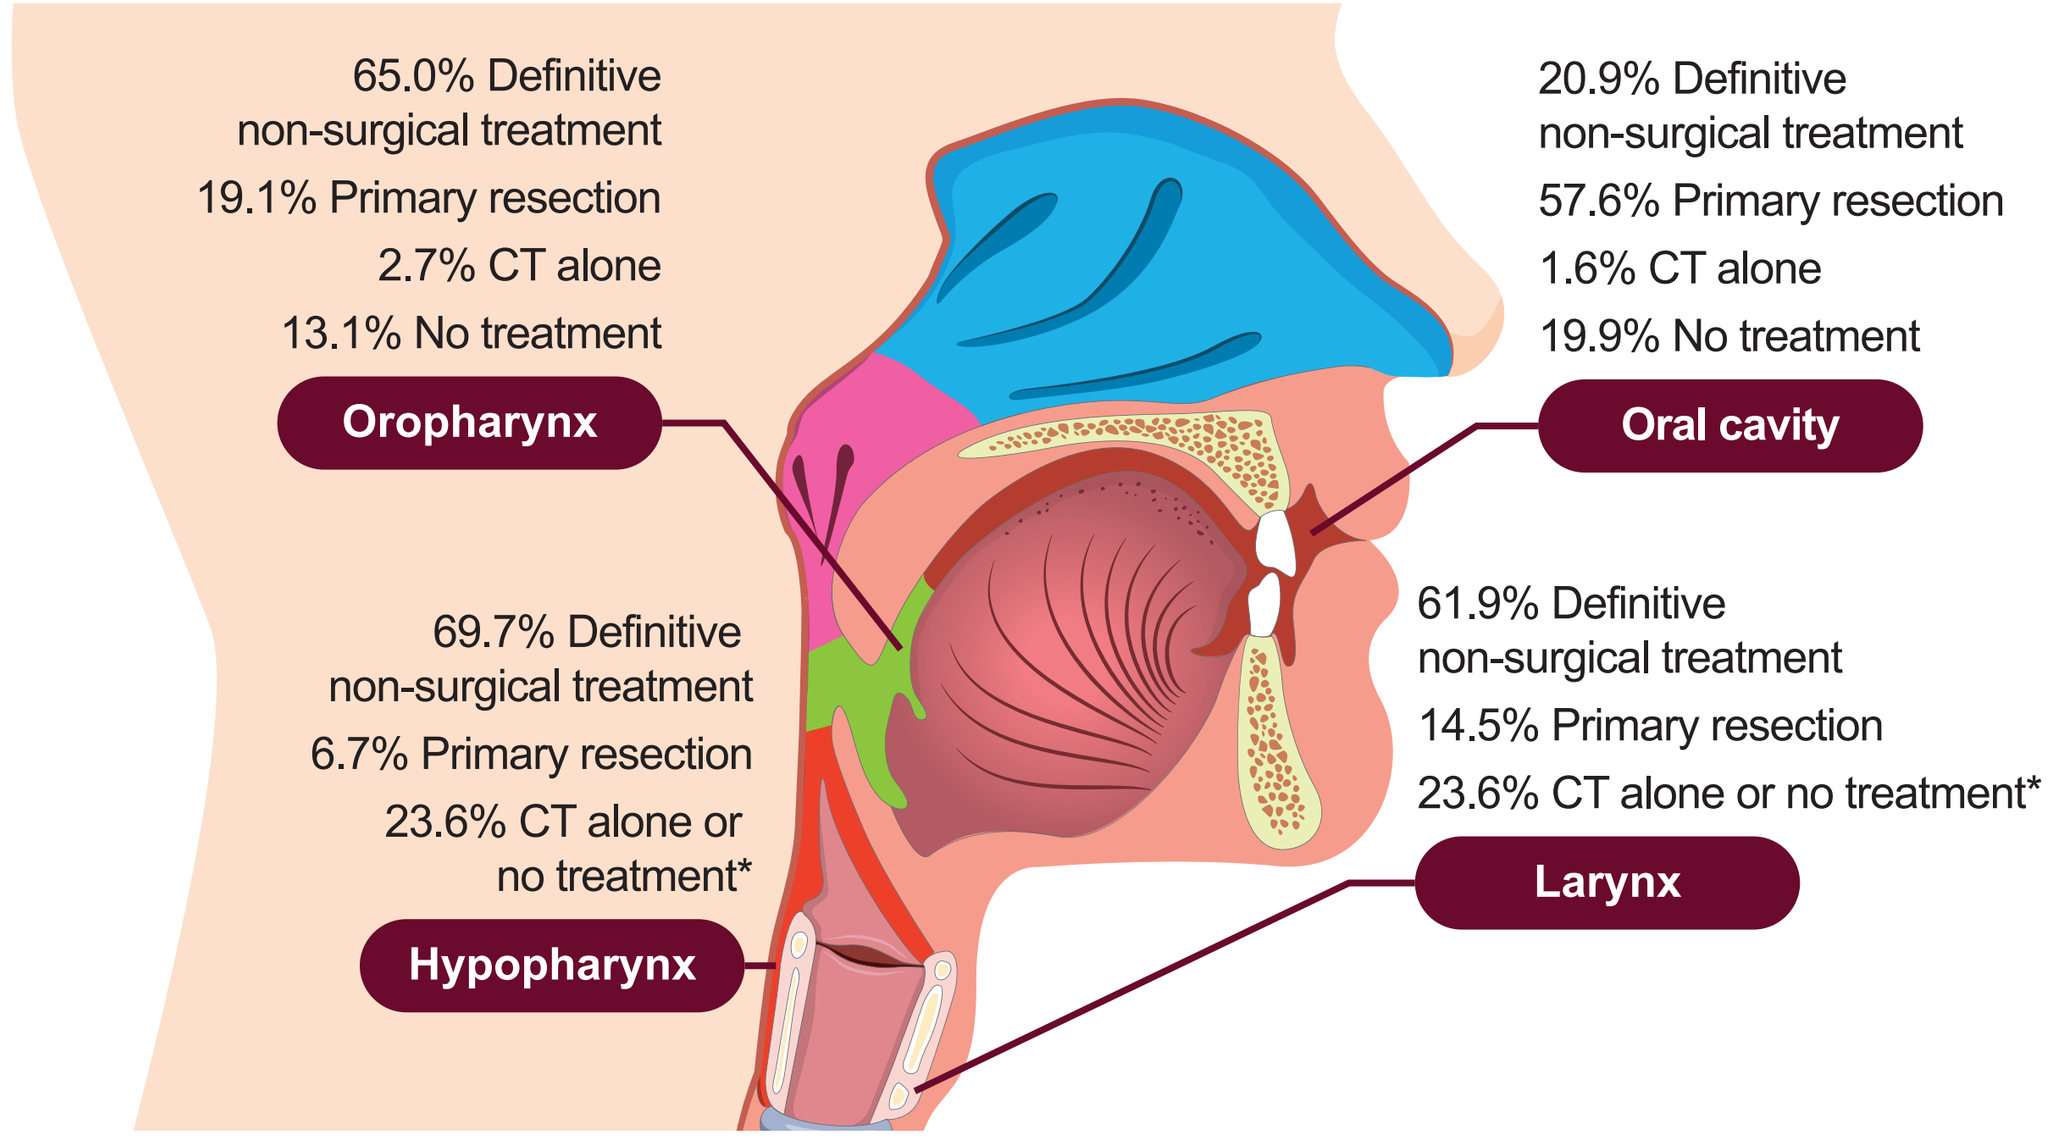

65.0% Definitive non-surgical treatment
19.1% Primary resection
2.7% CT alone
13.1% No treatment
Oropharynx
20.9% Definitive non-surgical treatment
57.6% Primary resection
1.6% CT alone
19.9% No treatment
Oral cavity
69.7% Definitive non-surgical treatment
6.7% Primary resection
23.6% CT alone or no treatment*
Hypopharynx
61.9% Definitive non-surgical treatment
14.5% Primary resection
23.6% CT alone or no treatment*
Larynx

CT, chemotherapy; NCI, National Cancer Institute.
*Data for treatments received by <11 patients (1.6%) are suppressed to comply with NCI privacy rules.

Figure 5. Systemic therapy for (A) unresected patients who received definitive CRT (n=1,076) or (B) patients who had primary resection with adjuvant CRT (n=276)

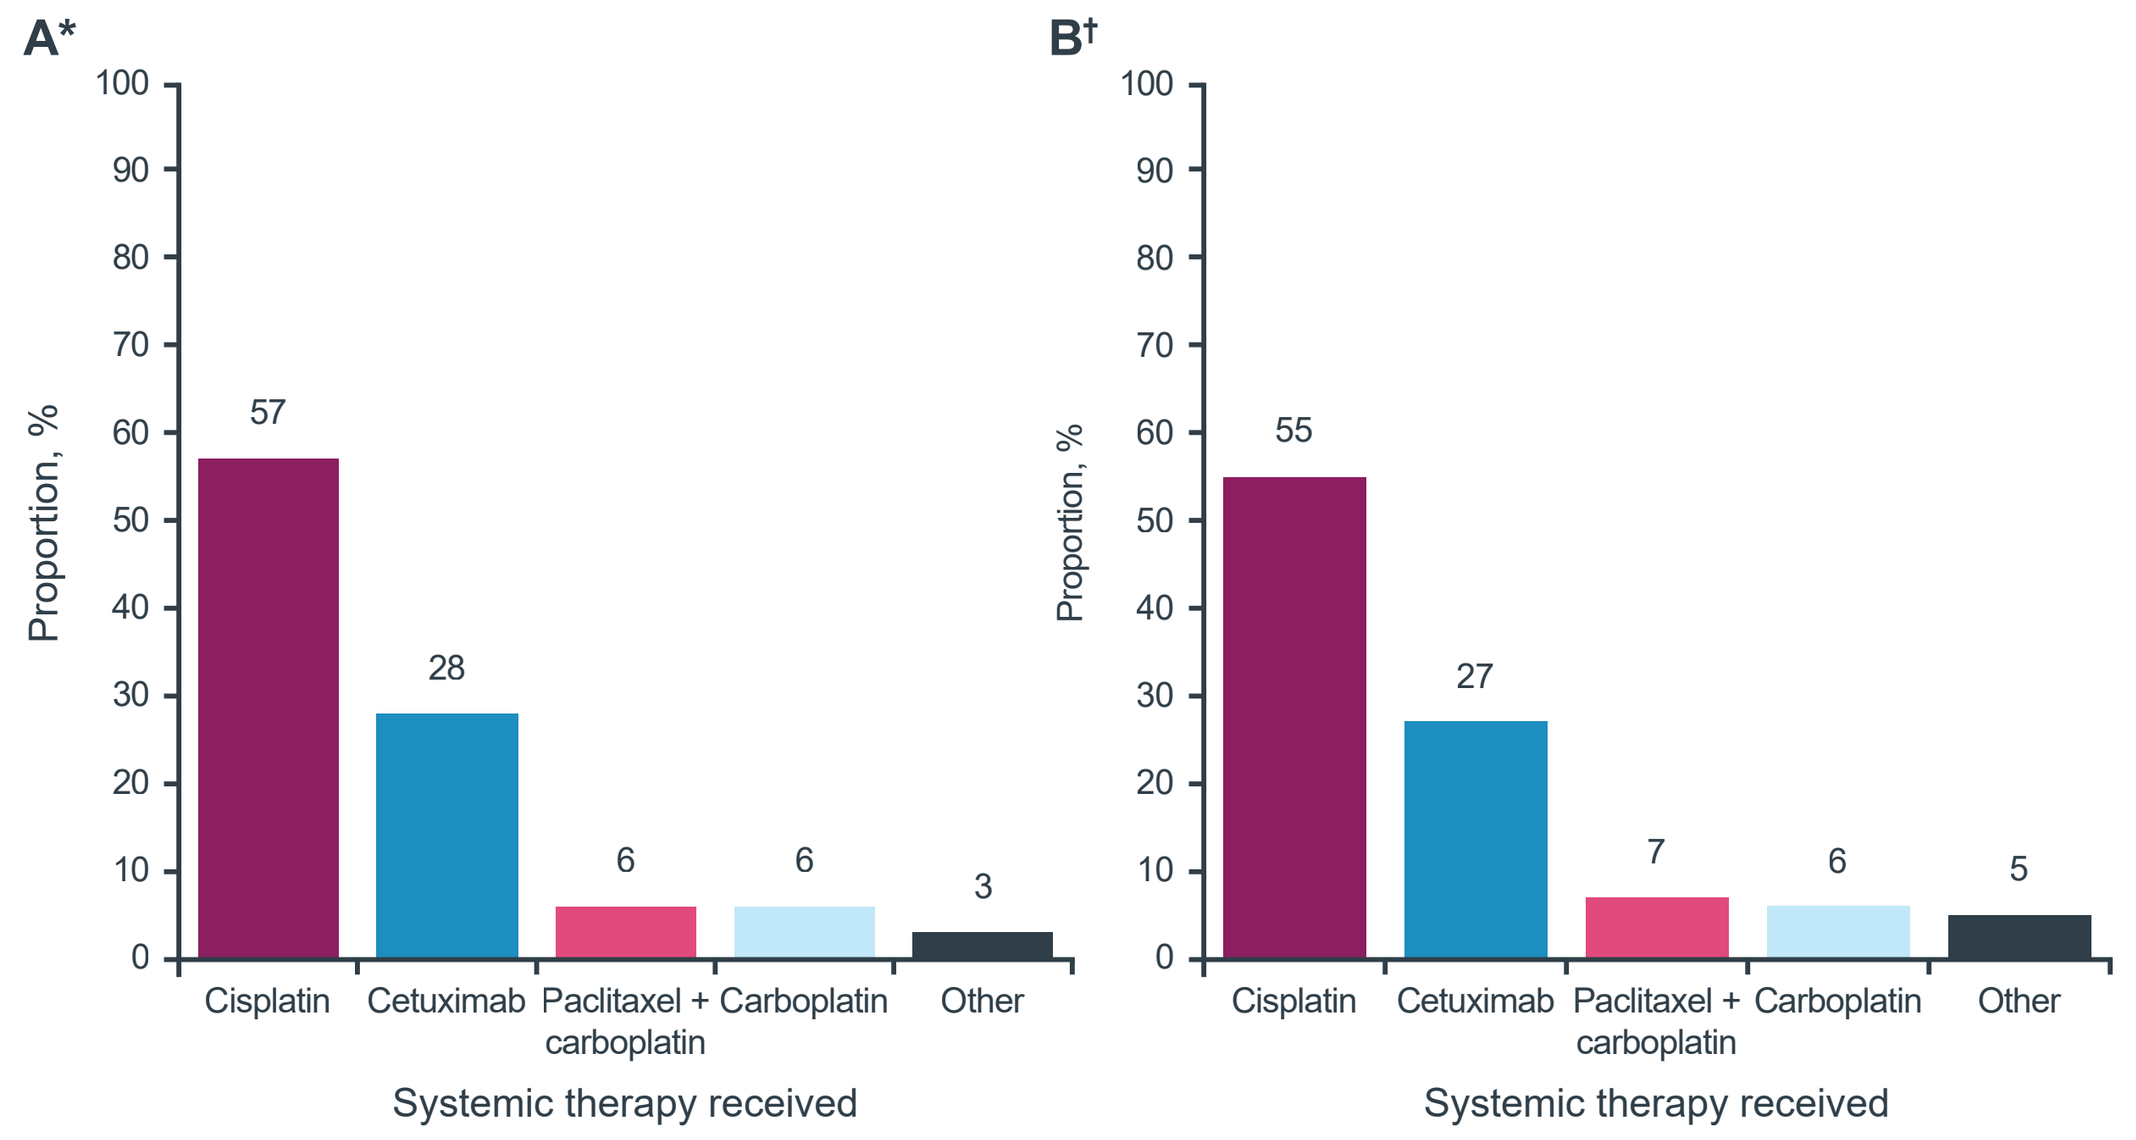

A*
B†
Proportion, %
57
28
6
6
3
55
27
7
6
5
Cisplatin
Cetuximab
Paclitaxel + carboplatin
Carboplatin
Other
Systemic therapy received

*Other treatments included aflibercept, aflibercept + cisplatin, cetuximab + carboplatin, cetuximab + cisplatin, cetuximab + paclitaxel + carboplatin, docetaxel + carboplatin, docetaxel + cisplatin, fluorouracil + carboplatin, fluorouracil + cisplatin, paclitaxel + cisplatin, and pembrolizumab + cisplatin. †Other treatments included cetuximab + cisplatin, cetuximab + docetaxel, cetuximab + docetaxel + cisplatin, docetaxel, docetaxel + carboplatin, docetaxel + cisplatin, fluorouracil + cisplatin, and paclitaxel + cisplatin.

Limitations

- Due to data lag, the latest available data are from 2019
- The study included patients aged ≥66 years; the population may not be representative of the wider LA SCCHN population

REFERENCES: 1. Surveillance, Epidemiology, and End Results Program. SEER cancer statistics review (CSR) 1975-2018. Accessed March 28, 2023. https://seer.cancer.gov/csr/1975_2018/browse_csr.php?sectionSEL=1&pageSEL=sect_01_table.01. **2.** Psyrri A, et al. Ann Oncol. 2014;25:2101-15. **3.** NCCN Clinical Practice Guidelines in Oncology. Head and neck cancers. V1, 2023. Accessed March 28, 2023. https://www.nccn.org/professionals/physician_gls/pdf/head-and-neck.pdf. **4.** Ahn MJ, et al. Oral Oncol. 2016;53:10-6. **5.** Falco A, et al. Cancer Manag Res. 2021;13:6689-703. **6.** Bower M, Waxman J. Oncology Lecture Notes. 3rd ed. Wiley; 2015. **7.** National Cancer Institute. Head and neck cancers. Accessed March 28, 2023. https://www.cancer.gov/types/head-and-neck/head-neck-fact-sheet.

DISCLOSURES: E. Lu reports employment at EMD Serono, Billerica, MA, USA. **C. Frenkel** has nothing to disclose. **M. Danese** reports employment at Outcomes Insights. **S. Bobiak** reports employment at EMD Serono, Billerica, MA, USA. **S. Salmio** reports employment at the healthcare business of Merck KGaA, Darmstadt, Germany. **N. Saba** reports serving in an advisory role for EMD Serono. **ACKNOWLEDGMENTS:** This study was sponsored by the healthcare business of Merck KGaA, Darmstadt, Germany (CrossRef Funder ID: 10.13039/100009945). Medical writing support was provided by Jamie Ratcliffe of Clinical Thinking and funded by the healthcare business of Merck KGaA, Darmstadt, Germany (CrossRef Funder ID: 10.13039/100009945).

Presented at ISPOR 2023, May 7-10, 2023; Boston, MA, USA.