HSD42

Utilization and Cost of Molecular Diagnostics and Genetic Testing Prescribed by Various Medical Specialties in the United States

Cho-Han Lee,[1,*] Jenny Liu,[2] Pradeep Kumar,[3] Pranshu,[3] John Stahl,[4] Scott Hengst,[4] Dejan Milentijevic[1]

[1]Janssen Medical Affairs, LLC, Titusville, NJ, USA; [2]Clarivate, Toronto, ON, Canada; [3]Clarivate, Bangalore, India; [4]Clarivate, Philadelphia, PA, USA.

*Presenting author.

INTRODUCTION

- Molecular and genetic testing can facilitate early disease detection, prevention, and precision treatment strategies[1]
- Molecular and genetic testing has become more routine and useful in a variety of medical specialties due to an increased understanding of the molecular pathology and genetic mechanisms behind diseases and advancements in genomic technologies that allow for accurate results at a lower cost[2,3]

OBJECTIVE

- To assess current molecular and genetic testing utilization patterns and costs across top prescribing specialties through claims analysis

METHODS

- This was a retrospective observational study using nationally representative US claims data obtained from the Decision Resources Group Real World Evidence Data Repository (Clarivate) between January 1, 2019 and November 8, 2022[4]
 - This database covers 98% of US health plans, including medical and pharmacy claims
- Included patients underwent molecular and genetic testing according to the selected Current Procedural Terminology codes, excluding immunoassay tests and other non-nucleic acid tests, presented in **Table 1**
- Prescribing specialties, defined as 'referring' providers by the PROVIDER_TYPE variable and National Uniform Claim Committee provider taxonomy codes, were included and grouped as shown in **Table 1**, with "other" encompassing all remaining specialties
- Demographics and claims reimbursement dynamics for molecular and genetic testing were explored using basic descriptive statistics (eg, count, percentage, mean, and median)
- Reimbursement endpoints included the number of claims approvals or rejections across the study period as well as out-of-pocket (OOP) and payer (reimbursed) amounts
- Statistical software tools were Microsoft Excel and Python

Table 1. Market Definitions for Molecular and Genetic Testing

Grouping	Code type	Code	Description
Molecular and genetic testing	CPT	81105-81479	Molecular pathology procedures, including genomic sequencing procedures and other molecular multianalyte assays (81410-81471)
		81490-81599	Multianalyte assays with algorithmic analyses
		0001U-0354U	Proprietary laboratory analyses
Specialty groups	Taxonomy codes	101200000X-405300000X	All prescribing providers were grouped into the following specialty groups: • OB/GYN • Hematology and oncology • Family medicine • Pediatrics • Internal medicine • Gastroenterology • Other (all remaining specialties)

CPT, Current Procedural Terminology; OB/GYN, obstetrics and gynecology.

RESULTS

Study Population

- Of 8.3 million patients, 3.2 million (38%) had data on claims approval status and related cost information and were included in this analysis (**Figure 1A**)
- Among the top 6 prescribing specialties, Obstetrics and gynecology (OB/GYN) had the highest volume of claims (5.7 million) during the study period (**Figure 1B**)
- Hematology and oncology and Family medicine had 1.6 million and 1.1 million claims, respectively, whereas the remaining top 3 specialties had markedly lower totals (**Figure 1B**)
- Patient characteristics are shown in **Table 2** by physician specialty

Figure 1. Total patients with remit records (A) and total molecular and genetic testing claims volume by specialty[a] (B).

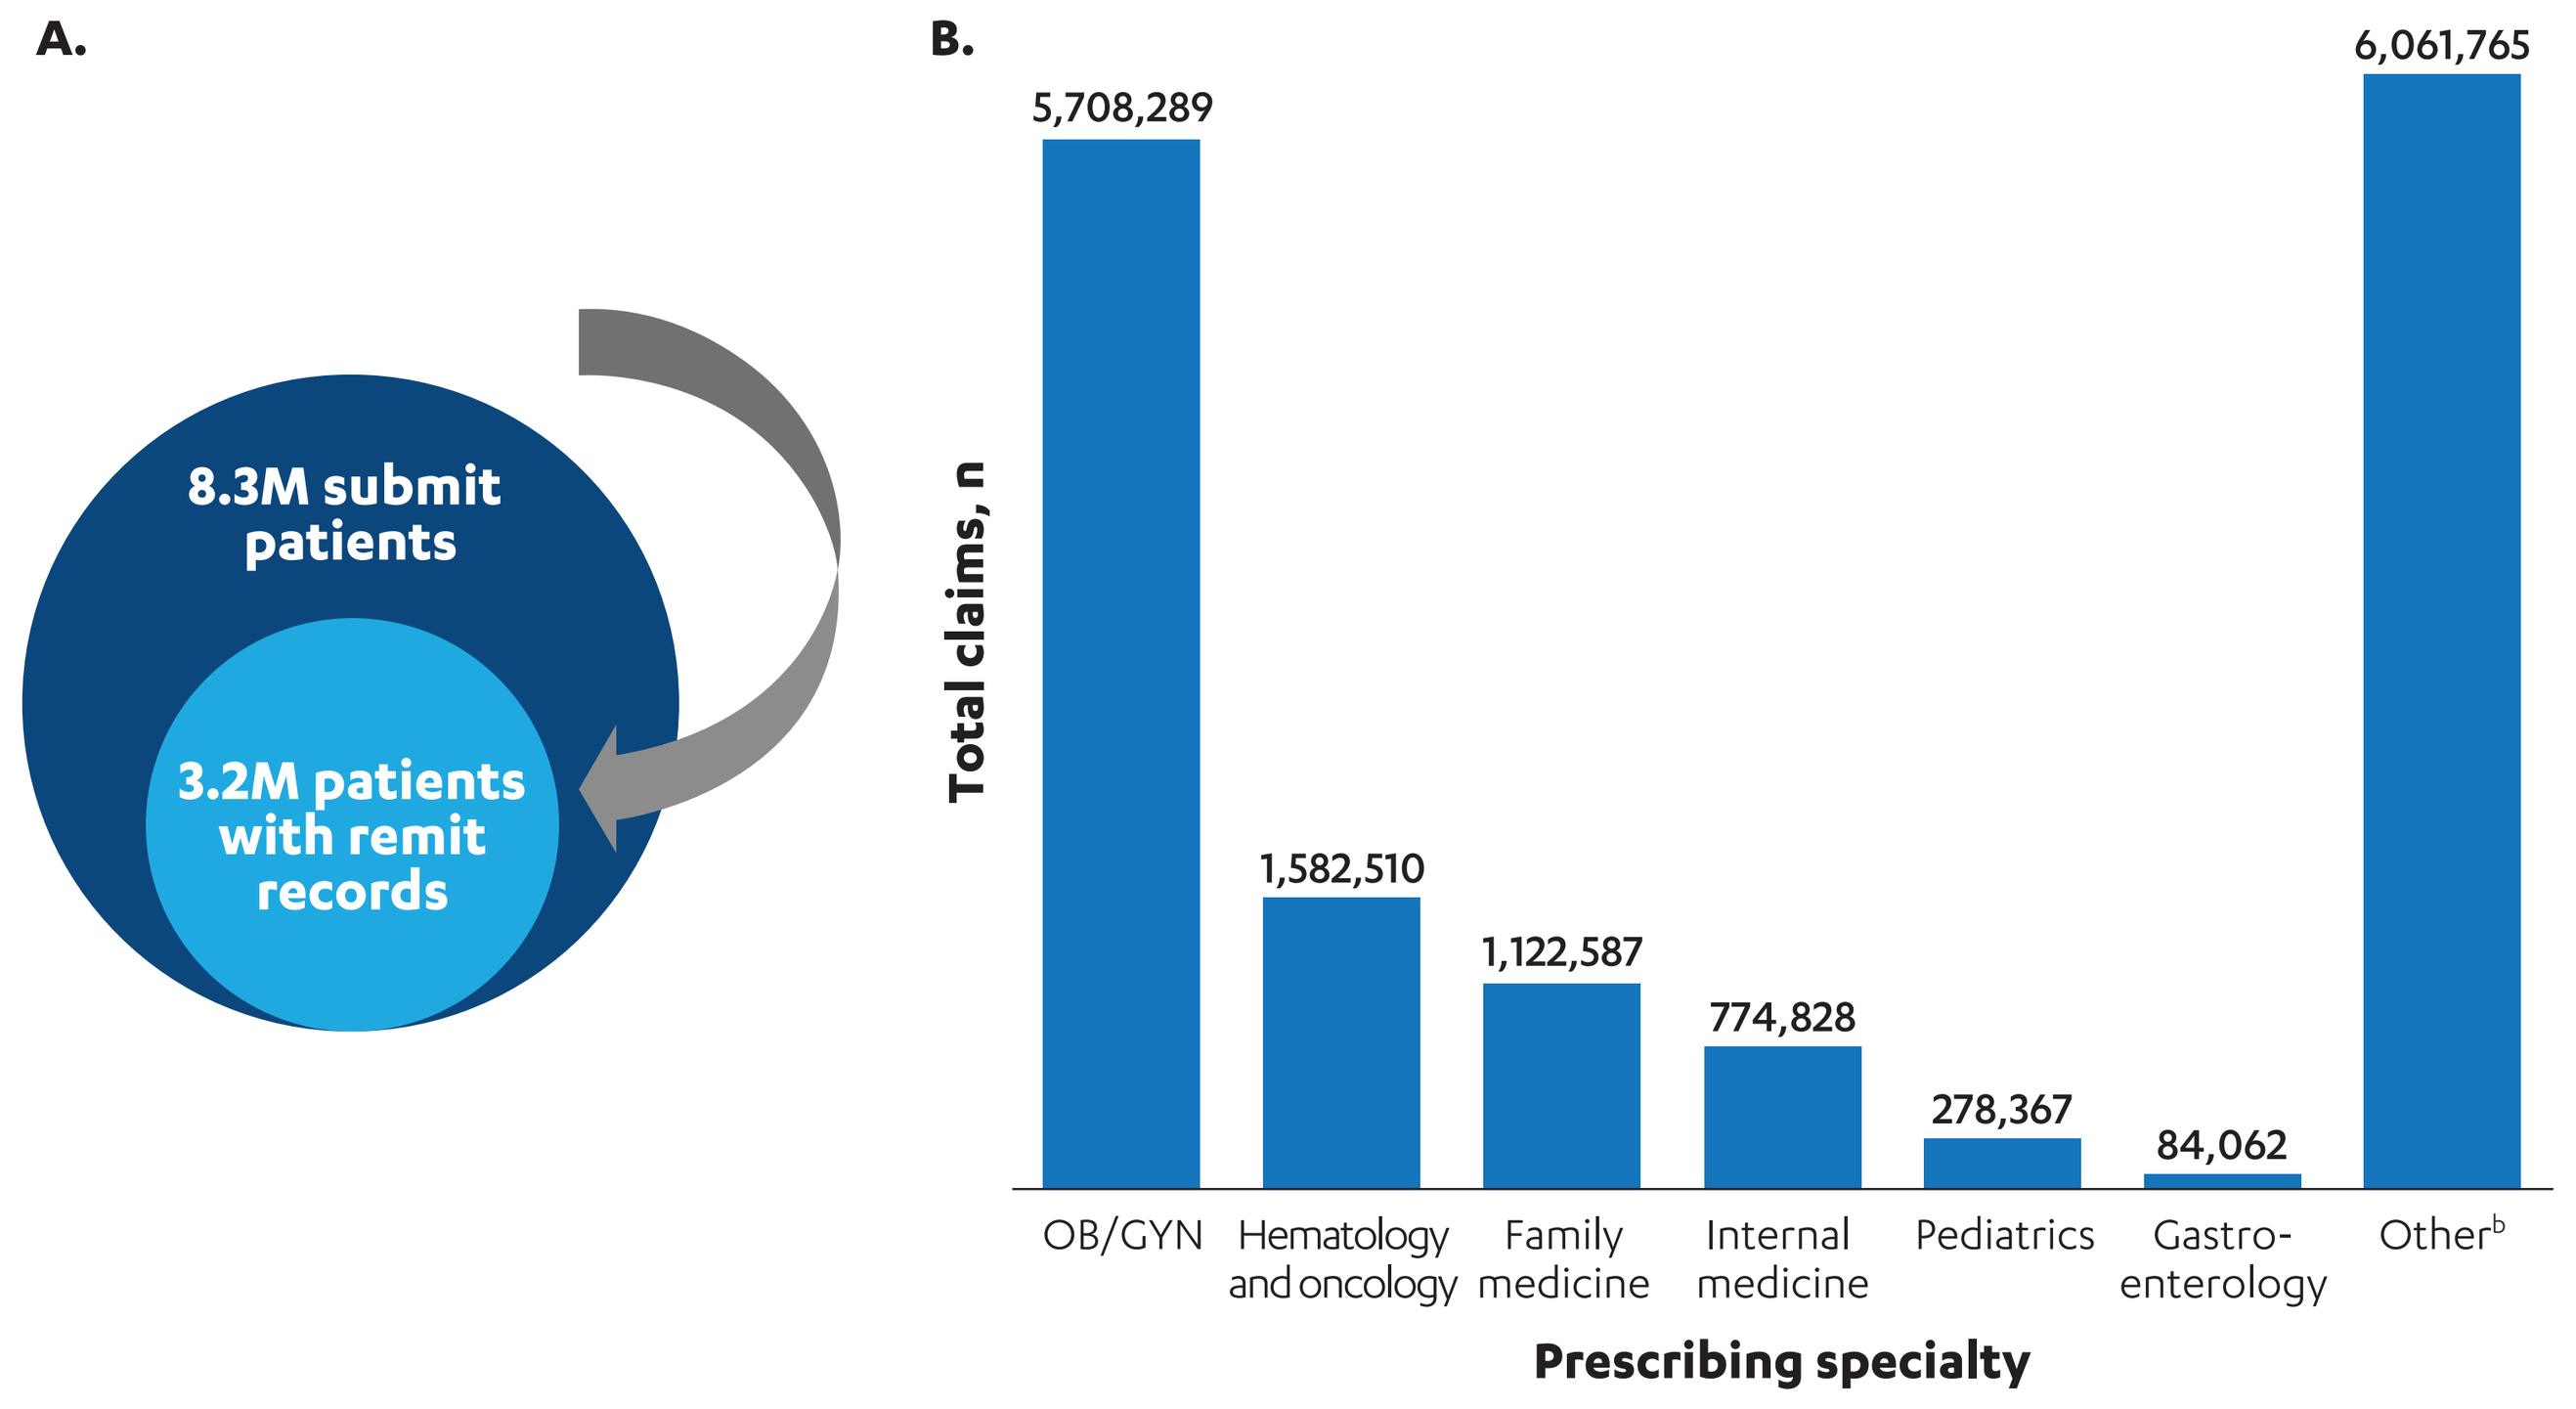
A.
8.3M submit patients
3.2M patients with remit records
B.
Total claims, n
5,708,289
1,582,510
1,122,587
774,828
278,367
84,062
6,061,765
OB/GYN
Hematology and oncology
Family medicine
Internal medicine
Pediatrics
Gastro-enterology
Other[b]
Prescribing specialty

OB/GYN, obstetrics and gynecology.
[a]Data include all approved and rejected claims and show only claims with matching remit records (remit data).
[b]Other includes allied health, null specialty information, and all other provider taxonomy codes that did not rank within the top 6 physician specialty groups.

Table 2. Patient Demographics for Molecular and Genetic Testing by Physician Specialty

Physician specialty	Patients, n	Mean age,[a] y	Median age,[a] y	Female, %
OB/GYN	1,743,833	34	33	90
Family medicine	1,053,347	54	58	59
Hematology and oncology	946,022	63	65	55
Internal medicine	784,355	59	61	57
Pediatrics	644,712	5	0	49
Gastroenterology	137,681	58	60	57
Other[b]	3,387,183	46	45	67

OB/GYN, obstetrics and gynecology.
[a]Patient's age when receiving genetic testing.
[b]Other includes allied health, null specialty information, and all other provider taxonomy codes that did not rank within the top 6 physician specialty groups.

Approval and Rejection of Claims

- Overall, Gastroenterology had the highest claims approval rate at 61% and Pediatrics had the lowest at 29% (**Figure 2**)
- Gastroenterology generally had the highest approval rates across the 4 years, ranging from 52% to 66%; Pediatrics claims experienced the lowest approval rates, although in 2021 there was a marked increase to 37% (**Figure 3**)
- Over 50% of patients were on commercial insurance; patient insurance type patterns were consistent across the top specialties (**Figure 4**)
 - Pediatrics had the highest proportion of Medicaid patients, as it includes eligible children under the Children's Health Insurance Program

Figure 2. Molecular and genetic testing approval and rejection rates by physician specialty.

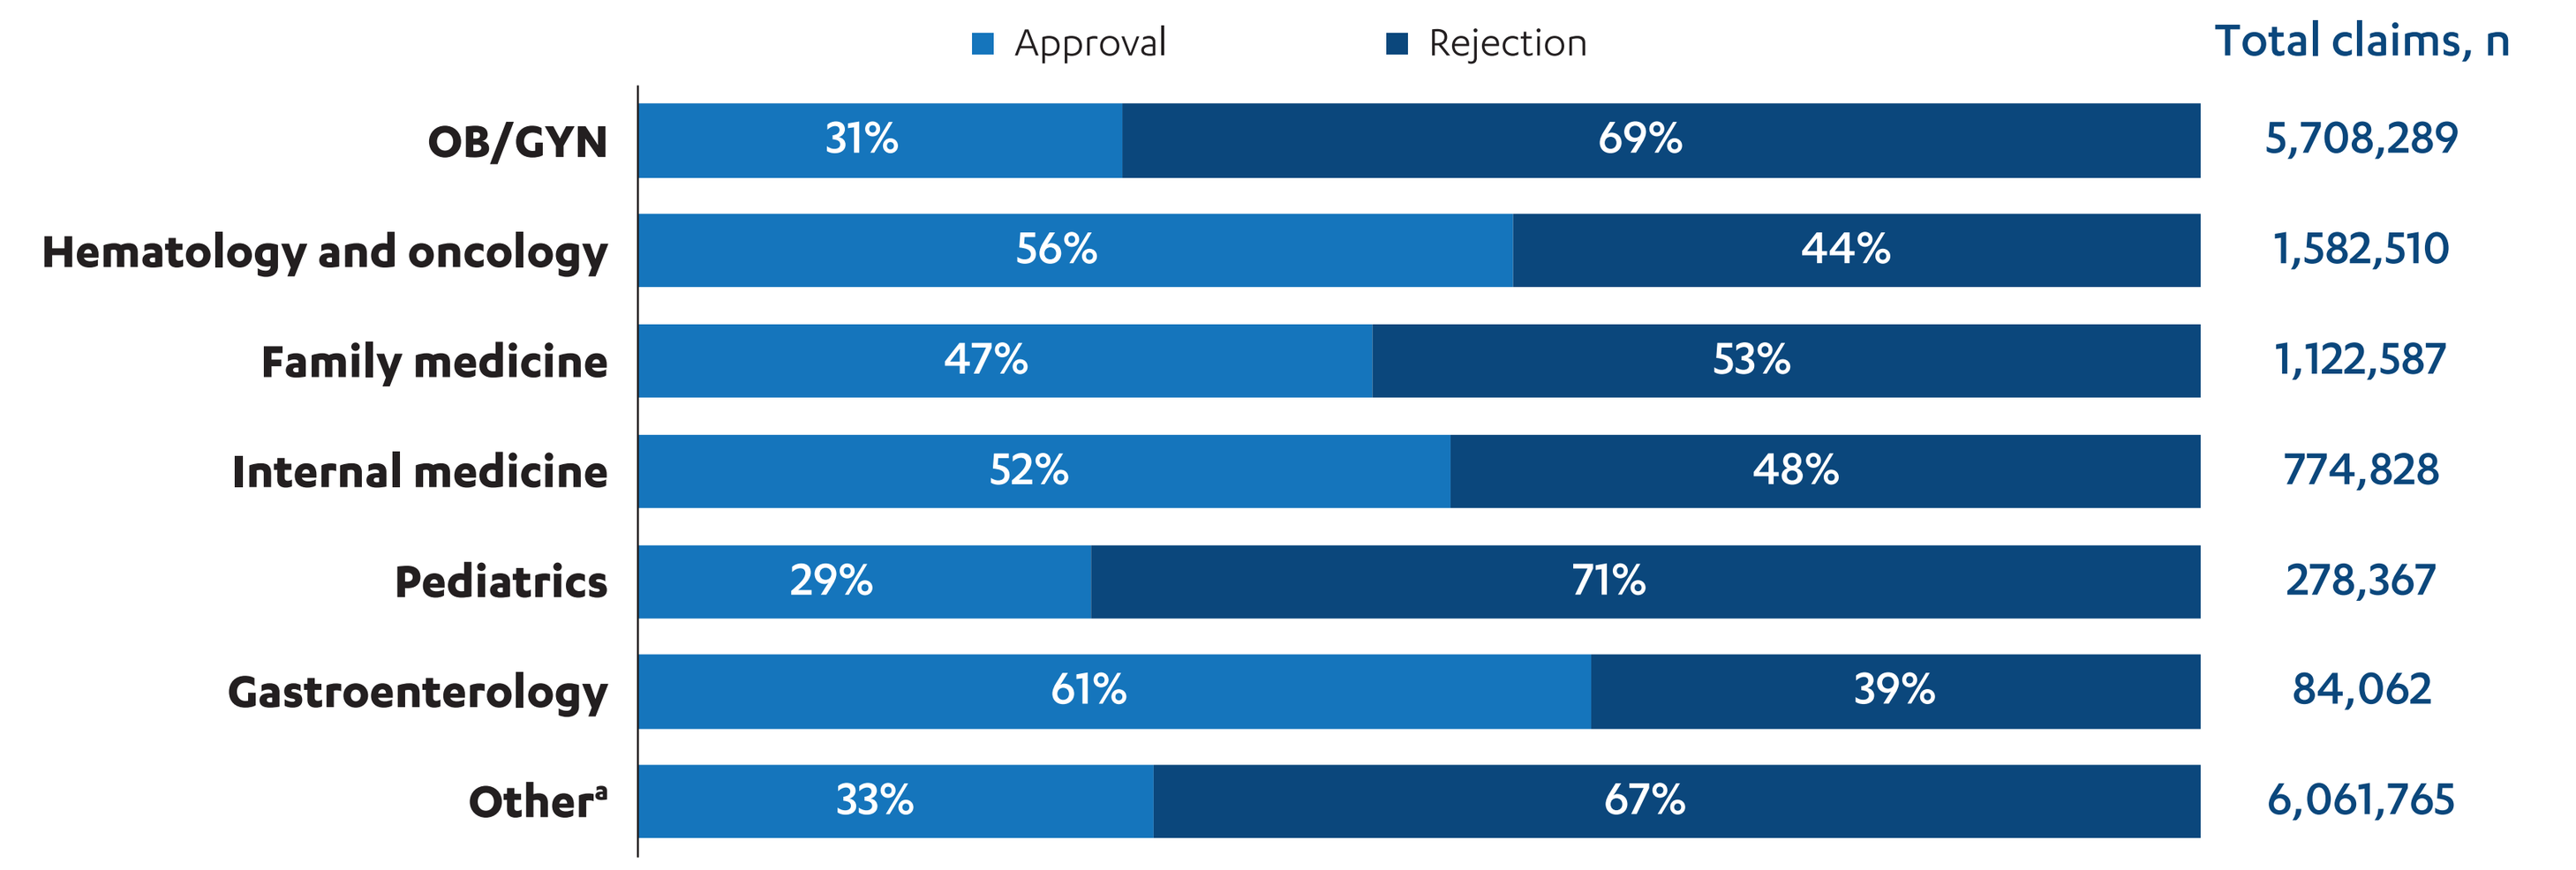
Approval
Rejection
Total claims, n
OB/GYN 31% 69% 5,708,289
Hematology and oncology 56% 44% 1,582,510
Family medicine 47% 53% 1,122,587
Internal medicine 52% 48% 774,828
Pediatrics 29% 71% 278,367
Gastroenterology 61% 39% 84,062
Other[a] 33% 67% 6,061,765

OB/GYN, obstetrics and gynecology.
[a]Other includes allied health, null specialty information, and all other provider taxonomy codes that did not rank within the top 6 physician specialty groups.

Figure 3. Annual molecular and genetic testing claims approval rates by physician specialty.

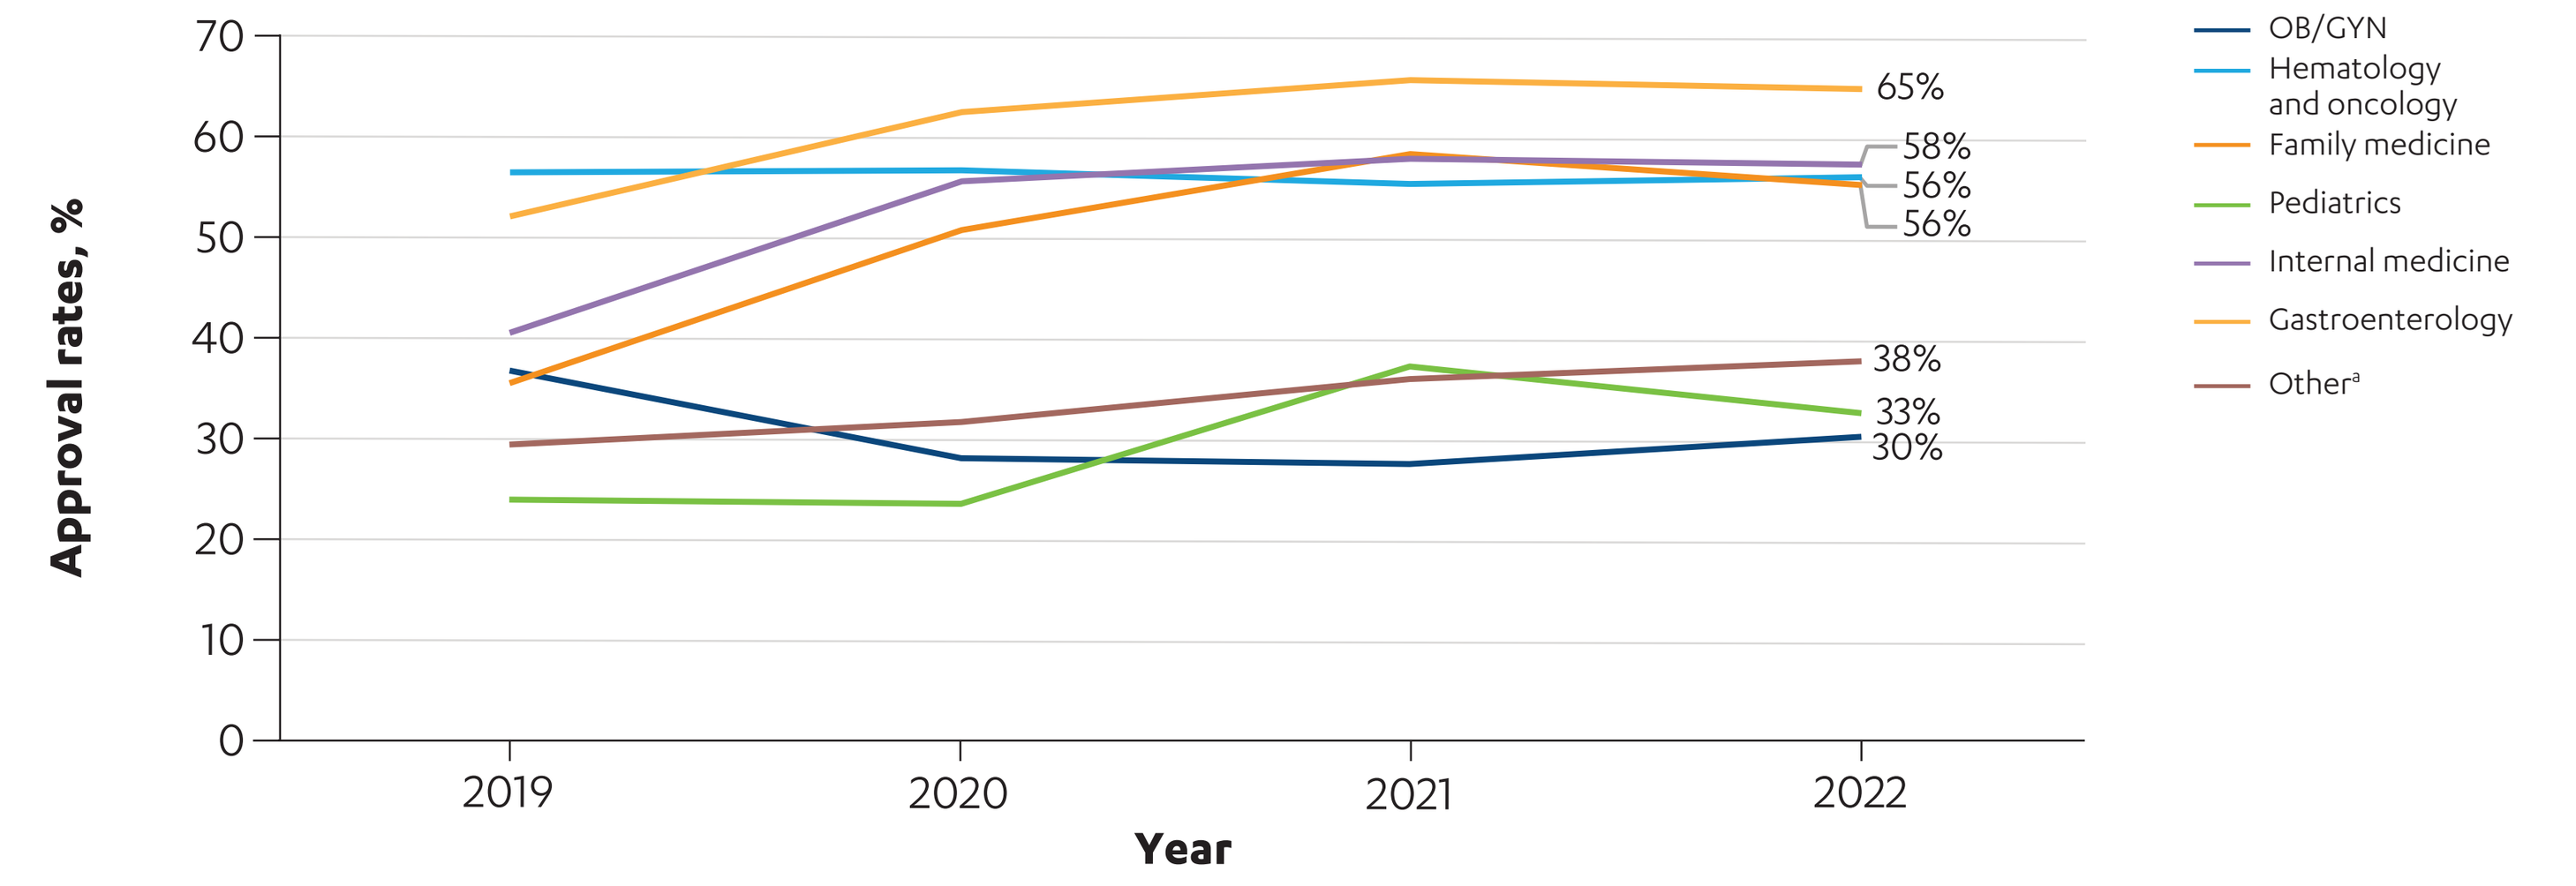
Approval rates, %
0
10
20
30
40
50
60
70
2019
2020
2021
2022
Year
65%
58%
56%
56%
38%
33%
30%
OB/GYN
Hematology and oncology
Family medicine
Pediatrics
Internal medicine
Gastroenterology
Other[a]

OB/GYN, obstetrics and gynecology.
[a]Other includes allied health, null specialty information, and all other provider taxonomy codes that did not rank within the top 6 physician specialty groups.

Figure 4. Patient insurance type by physician specialty.

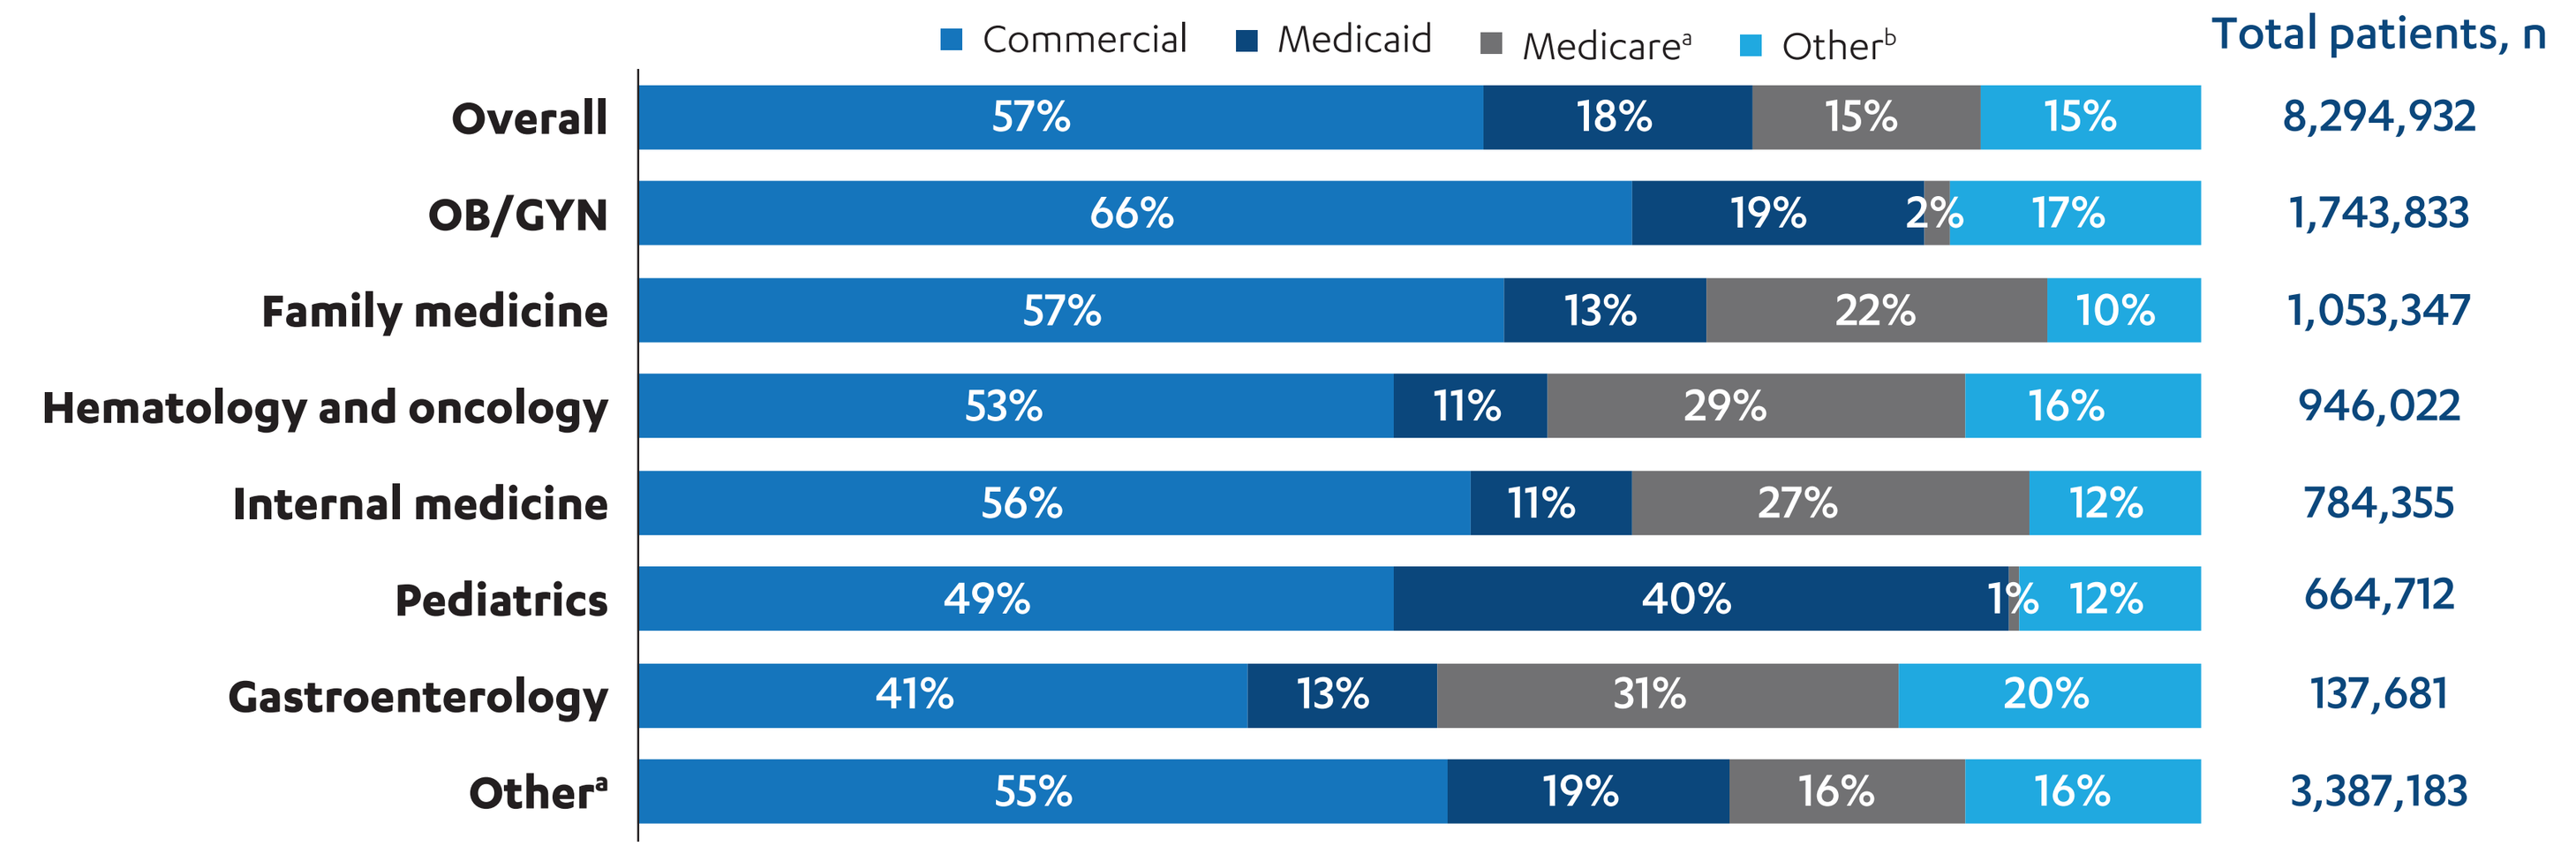
Commercial
Medicaid
Medicare[a]
Other[b]
Total patients, n
Overall 57% 18% 15% 15% 8,294,932
OB/GYN 66% 19% 2% 17% 1,743,833
Family medicine 57% 13% 22% 10% 1,053,347
Hematology and oncology 53% 11% 29% 16% 946,022
Internal medicine 56% 11% 27% 12% 784,355
Pediatrics 49% 40% 1% 12% 664,712
Gastroenterology 41% 13% 31% 20% 137,681
Other[a] 55% 19% 16% 16% 3,387,183

OB/GYN, obstetrics and gynecology.
[a]Medicare also includes dual insurance (ie, Medicare and Medicaid).
[b]Other includes Veterans Affairs health care insurance, other special government insurance types, and unknown payer types.

Patient OOP and Physician Reimbursement Costs

- A total of 5,749,455 paid/approved claims were captured using the study inclusion criteria
- OB/GYN claims showed the highest mean patient OOP amounts ($20) among specialties (**Figure 5A**)
 - Among the highest quartile of claims, patient OOP costs ranged from $50 (Hematology and oncology) to $83 (Pediatrics; **Figure 5B**)
- Approximately 40% of the submitted amount for molecular and genetic testing claims was reimbursed by payers (**Figure 6A** and **B**)
 - Among the 6 specialties evaluated, the highest mean submitted amounts were from Pediatrics ($1109) and Hematology and oncology ($1062); Family medicine ($467) and Internal medicine ($422) had the highest mean payer-reimbursed amounts (**Figure 6A**)

Figure 5. Mean patient OOP costs[a] (A) and top 25% of patient OOP costs[b] (B) for molecular/genetic testing claims by physician specialty.

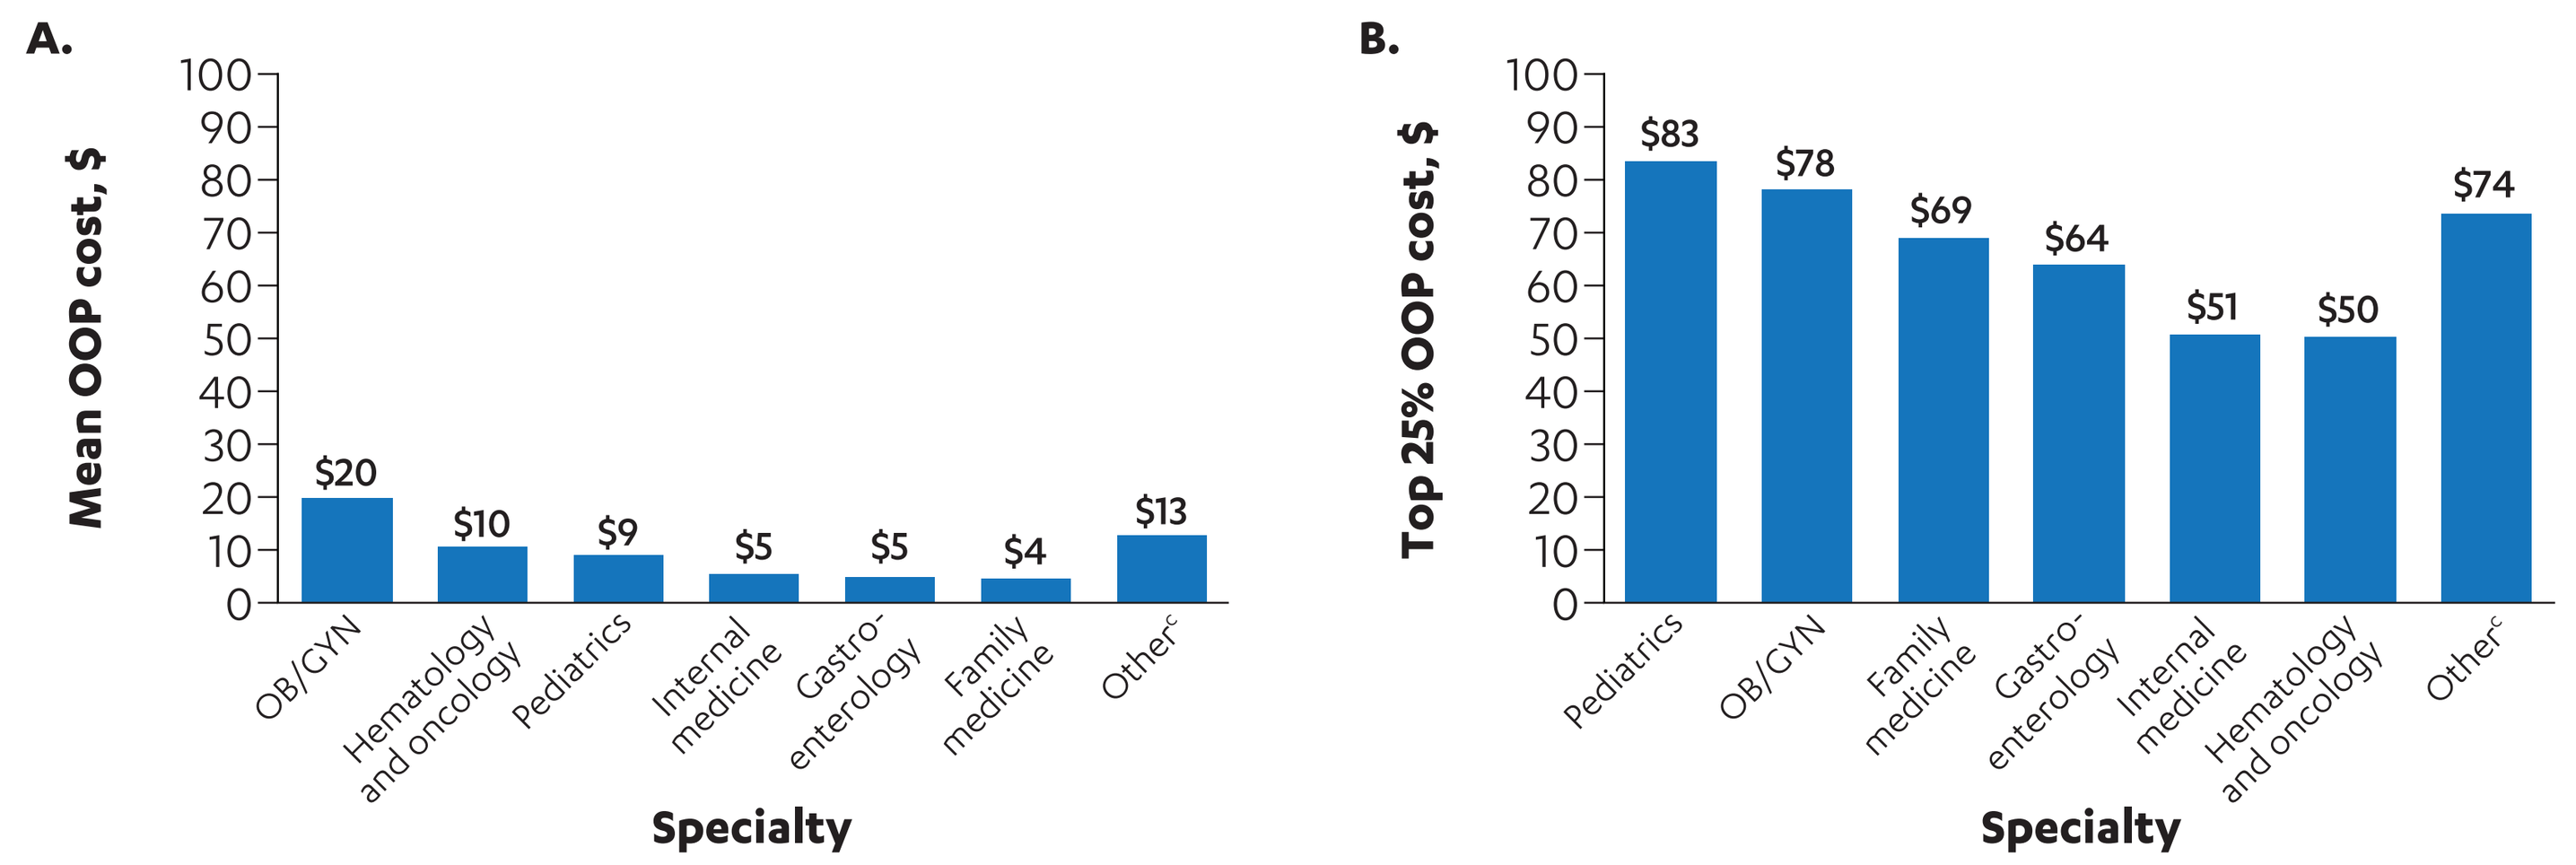
A.
Mean OOP cost, $
$20 OB/GYN
$10 Hematology and oncology
$9 Pediatrics
$5 Internal medicine
$5 Gastro-enterology
$4 Family medicine
$13 Other[c]
Specialty
B.
Top 25% OOP cost, $
$83 Pediatrics
$78 OB/GYN
$69 Family medicine
$64 Gastro-enterology
$51 Internal medicine
$50 Hematology and oncology
$74 Other[c]
Specialty

OB/GYN, obstetrics and gynecology; OOP, out-of-pocket.
[a]Median amounts equal $0 for all specialties due to the large number of $0 claims.
[b]Average OOP cost to the patient was based on the top 25% of patient-paid claims.
[c]Other includes allied health, null specialty information, and all other provider taxonomy codes that did not rank within the top 6 physician specialty groups.

Figure 6. Mean (A) and median (B) physician-submitted and physician-reimbursed amounts for molecular/genetic testing claims by physician specialty.[a]

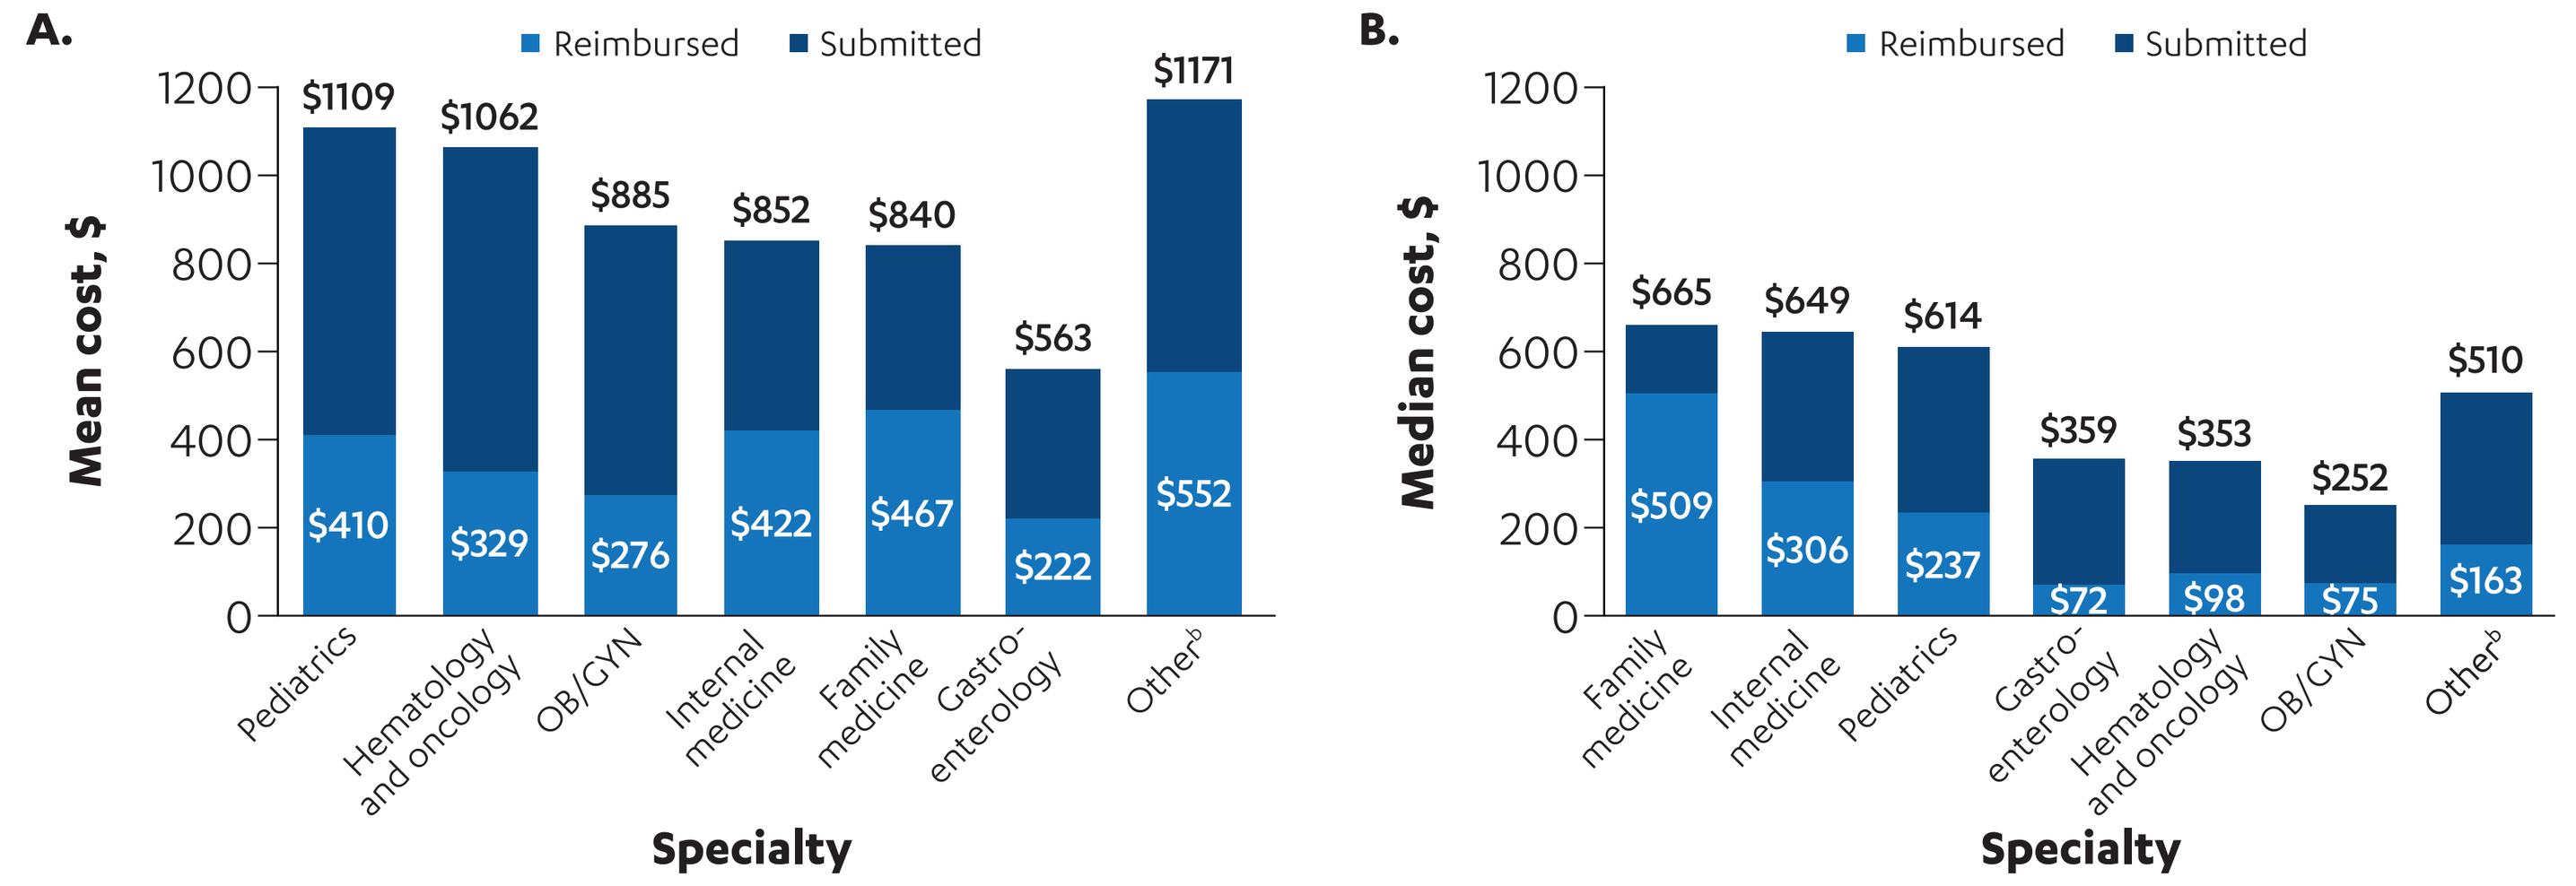
A.
Reimbursed
Submitted
Mean cost, $
Pediatrics $1109 $410
Hematology and oncology $1062 $329
OB/GYN $885 $276
Internal medicine $852 $422
Family medicine $840 $467
Gastro-enterology $563 $222
Other[b] $1171 $552
Specialty
B.
Reimbursed
Submitted
Median cost, $
Family medicine $665 $509
Internal medicine $649 $306
Pediatrics $614 $237
Gastro-enterology $359 $72
Hematology and oncology $353 $98
OB/GYN $252 $75
Other[b] $510 $163
Specialty

OB/GYN, obstetrics and gynecology.
[a]Large differences between mean and median amounts may be due to outliers "skewing" the mean toward higher values.
[b]Other includes allied health, null specialty information, and all other provider taxonomy codes that did not rank within the top 6 physician specialty groups.

LIMITATIONS

- The claims database did not capture free genetic testing kits sponsored by pharmaceutical companies and patient advocacy groups
- The database did not cover all the payers in the United States. Only 38% of the submitted claims were matched with remit records, making 3.2 million patient records eligible for the reimbursement analysis
- The exact names and contents for the molecular and genetic testing are not identifiable in the database
- The patient deductible amounts were $0 for most of the claims; it is unclear whether the $0 copay amount was due to the maximized annual deductible

CONCLUSIONS

- Across the top prescribing specialties in the United States, the approval rates for molecular/genetic testing remained consistent despite the pandemic period
- Given that the range of OOP costs was between $4 and $20 and the range of the average reimbursement was between $222 and $467, molecular/genetic testing costs may be reasonable for many patients and payers
- Improved understanding of the importance of molecular/genetic testing, patients' knowledge of insurance, and clinical guideline recommendations may facilitate better approval rates for tests leading to earlier diagnosis

REFERENCES

1. Kotze MJ, et al. *Crit Rev Clin Lab Sci*. 2015;52(3):120-137.
2. Bajguz D, et al. *J Genet Couns*. 2022;31(3):771-780.
3. Katsanis SH, et al. *Nat Rev Genet*. 2013;14(6):415-426.
4. Decision Resources Group Real World Evidence US Data Repository.

ACKNOWLEDGMENTS

This study was sponsored by Janssen Scientific Affairs, LLC. Medical writing support was provided by David Le, PharmD, of Lumanity Communications Inc., and was funded by Janssen Scientific Affairs, LLC.

DISCLOSURES

C-H Lee: Employee – Janssen Scientific Affairs, LLC; Stock – Johnson & Johnson. **J Liu:** Employee – Clarivate. **P Kumar:** Employee – Clarivate. **Pranshu:** Employee – Clarivate. **J Stahl:** Employee – Clarivate. **S Hengst:** Employee – Clarivate. **D Milentijevic:** Employee – Janssen Scientific Affairs, LLC.

Poster presented at The Professional Society for Health Economics and Outcomes Research (ISPOR) Annual Meeting; May 7-10, 2023; Boston, MA, USA.

PHARMACEUTICAL COMPANIES OF Johnson & Johnson