HSD63

Real-world Data Analysis of Genetic Testing for Inherited Retinal Diseases

Cho-Han Lee,[1,*] Jenny Liu,[2] Pradeep Kumar,[3] Pranshu,[3] John Stahl,[4] Scott Hengst,[4] Dejan Milentijevic[1]

[1]Janssen Medical Affairs, LLC, Titusville, NJ, USA; [2]Clarivate, Toronto, ON, Canada; [3]Clarivate, Bangalore, India; [4]Clarivate, Philadelphia, PA, USA.

*Presenting author.

INTRODUCTION

- Inherited retinal diseases (IRDs), which affect approximately 1 in every 3500 to 4000 people globally, cause progressive vision loss and are the leading cause of disability and blindness in people <60 years of age[1,2]
- Because traditional treatments cannot restore vision or, at least, stop progressive vision loss, most IRDs are considered to be incurable[3]
- Genetic testing may identify vision-associated gene mutations, which could allow patients with IRDs to receive novel gene therapies or enroll in clinical trials
 - The American Academy of Ophthalmology diagnosis guidelines recommend using genetic testing to enable patients to receive treatments, such as voretigene neparvovec-rzyl, and to enroll in clinical trials for IRD[4]

OBJECTIVE

- To understand the current utilization of molecular and genetic testing specific to IRDs, including percent utilization, costs, and approval/rejection rates in the United States

METHODS

- This was a retrospective observational study using nationally representative US claims data obtained from the Decision Resources Group Real World Evidence Data Repository (Clarivate) between January 1, 2019 and November 8, 2022[5]
 - This database covers 98% of US health plans, including medical and pharmacy claims
- The study included patients with ≥2 IRD diagnoses who were undergoing ≥1 of the 14 genetic tests commonly ordered for IRDs (as identified by selected Current Procedural Terminology codes; **Table 1**)
- Demographics and claims reimbursement dynamics for IRD molecular and genetic testing were explored using basic descriptive statistics (eg, count, percentage, mean, and median)
- Statistical software tools were Microsoft Excel and Python

Table 1. Market Definitions for Molecular and Genetic Testing in IRDs

Grouping	Code type	Code	Description
IRD diagnosis	ICD-10/9	H35.5/362.70	Hereditary retinal dystrophy
		H35.50/362.70	Unspecified hereditary retinal dystrophy/ Leber congenital amaurosis
		H35.51/362.73	Vitreoretinal dystrophy
		H35.52/362.74	Pigmentary retinal dystrophy
		H35.53/362.75	Other dystrophies primarily involving the sensory retina
		H35.54/362.76	Dystrophies primarily involving the retinal pigment epithelium
Molecular and genetic testing	CPT	81400-81408	Tier 2 molecular pathology procedures, levels 1 to 9
		81434	Hereditary retinal disorders
		81460	Whole mitochondrial genome
		81465	Whole mitochondrial genome large deletion analysis
		81479	Unlisted molecular pathology (*RPGR* single-gene test included)
		81599	Unlisted multianalyte assay with algorithmic analysis

CPT, Current Procedural Terminology; ICD-10/9, International Classification of Diseases, 10th/9th Revisions; IRD, inherited retinal disease.

RESULTS

Study Population

- 166,781 patients were identified to have ≥2 IRD diagnoses during the study period (**Table 2**)
 - Of these patients, 2577 (~2%) were identified to have undergone ≥1 of the 14 included genetic tests
- Among the 2577 patients with IRDs included in the study, 57% were female and 43% were male; the mean age for IRD diagnosis was 52 years (median, 56 years), and the mean age for molecular and genetic testing was 51 years (median, 57 years)
 - Patients in the study experienced moderate comorbidities, with a median Charlson Comorbidity Index score of 3

Table 2. Patient Attrition

		Counts, n		Attrition, %	
	Description	Patients	Claims	Patients	Claims
STEP 1	Patients undergoing ≥1 of 14 genetic tests from procedure dataset	2,599,630	4,390,868	–	–
STEP 2	Patients with IRD Dx (ICD-10: 6 IRD Dx codes; ICD-9 codes not applicable due to time period filter)	294,000	912,154	–	–
STEP 3	Patients with ≥2 IRD Dx codes	166,781	784,936	56.73	86.05
STEP 4	Overlap steps 1 and 3 (IRD patients taking genetic testing)	2577	4122[a]	1.55	0.53
STEP 5	Patients with approval/rejection details (claims at service-line level: paid, rejected, and reversed claims)	981	4092[b]	38.07	99.27
STEP 6	Patients with approval/rejection details (claims at service-line level: only paid and rejected claims)	978	3828[b]	37.95	92.87

Dx, diagnosis; ICD-9, International Classification of Diseases, 9th Revision; ICD-10, International Classification of Diseases, 10th Revision; IRD, inherited retinal disease.
[a]Claim counts based on Claim ID.
[b]Claim counts based on service-line reimbursements.

Approval and Rejection of Claims

- Among the 3828 total claims evaluated in this study, 31% (n = 1179) of claims for molecular and genetic testing were approved (**Figure 1**)
- Of the 2649 rejected claims, the 2 most common reasons for rejection were lack of product/patient coverage (38%) and physician/administration error (31%; **Figure 2**)

Figure 1. Overall claims approval rates over time (N = 3828).

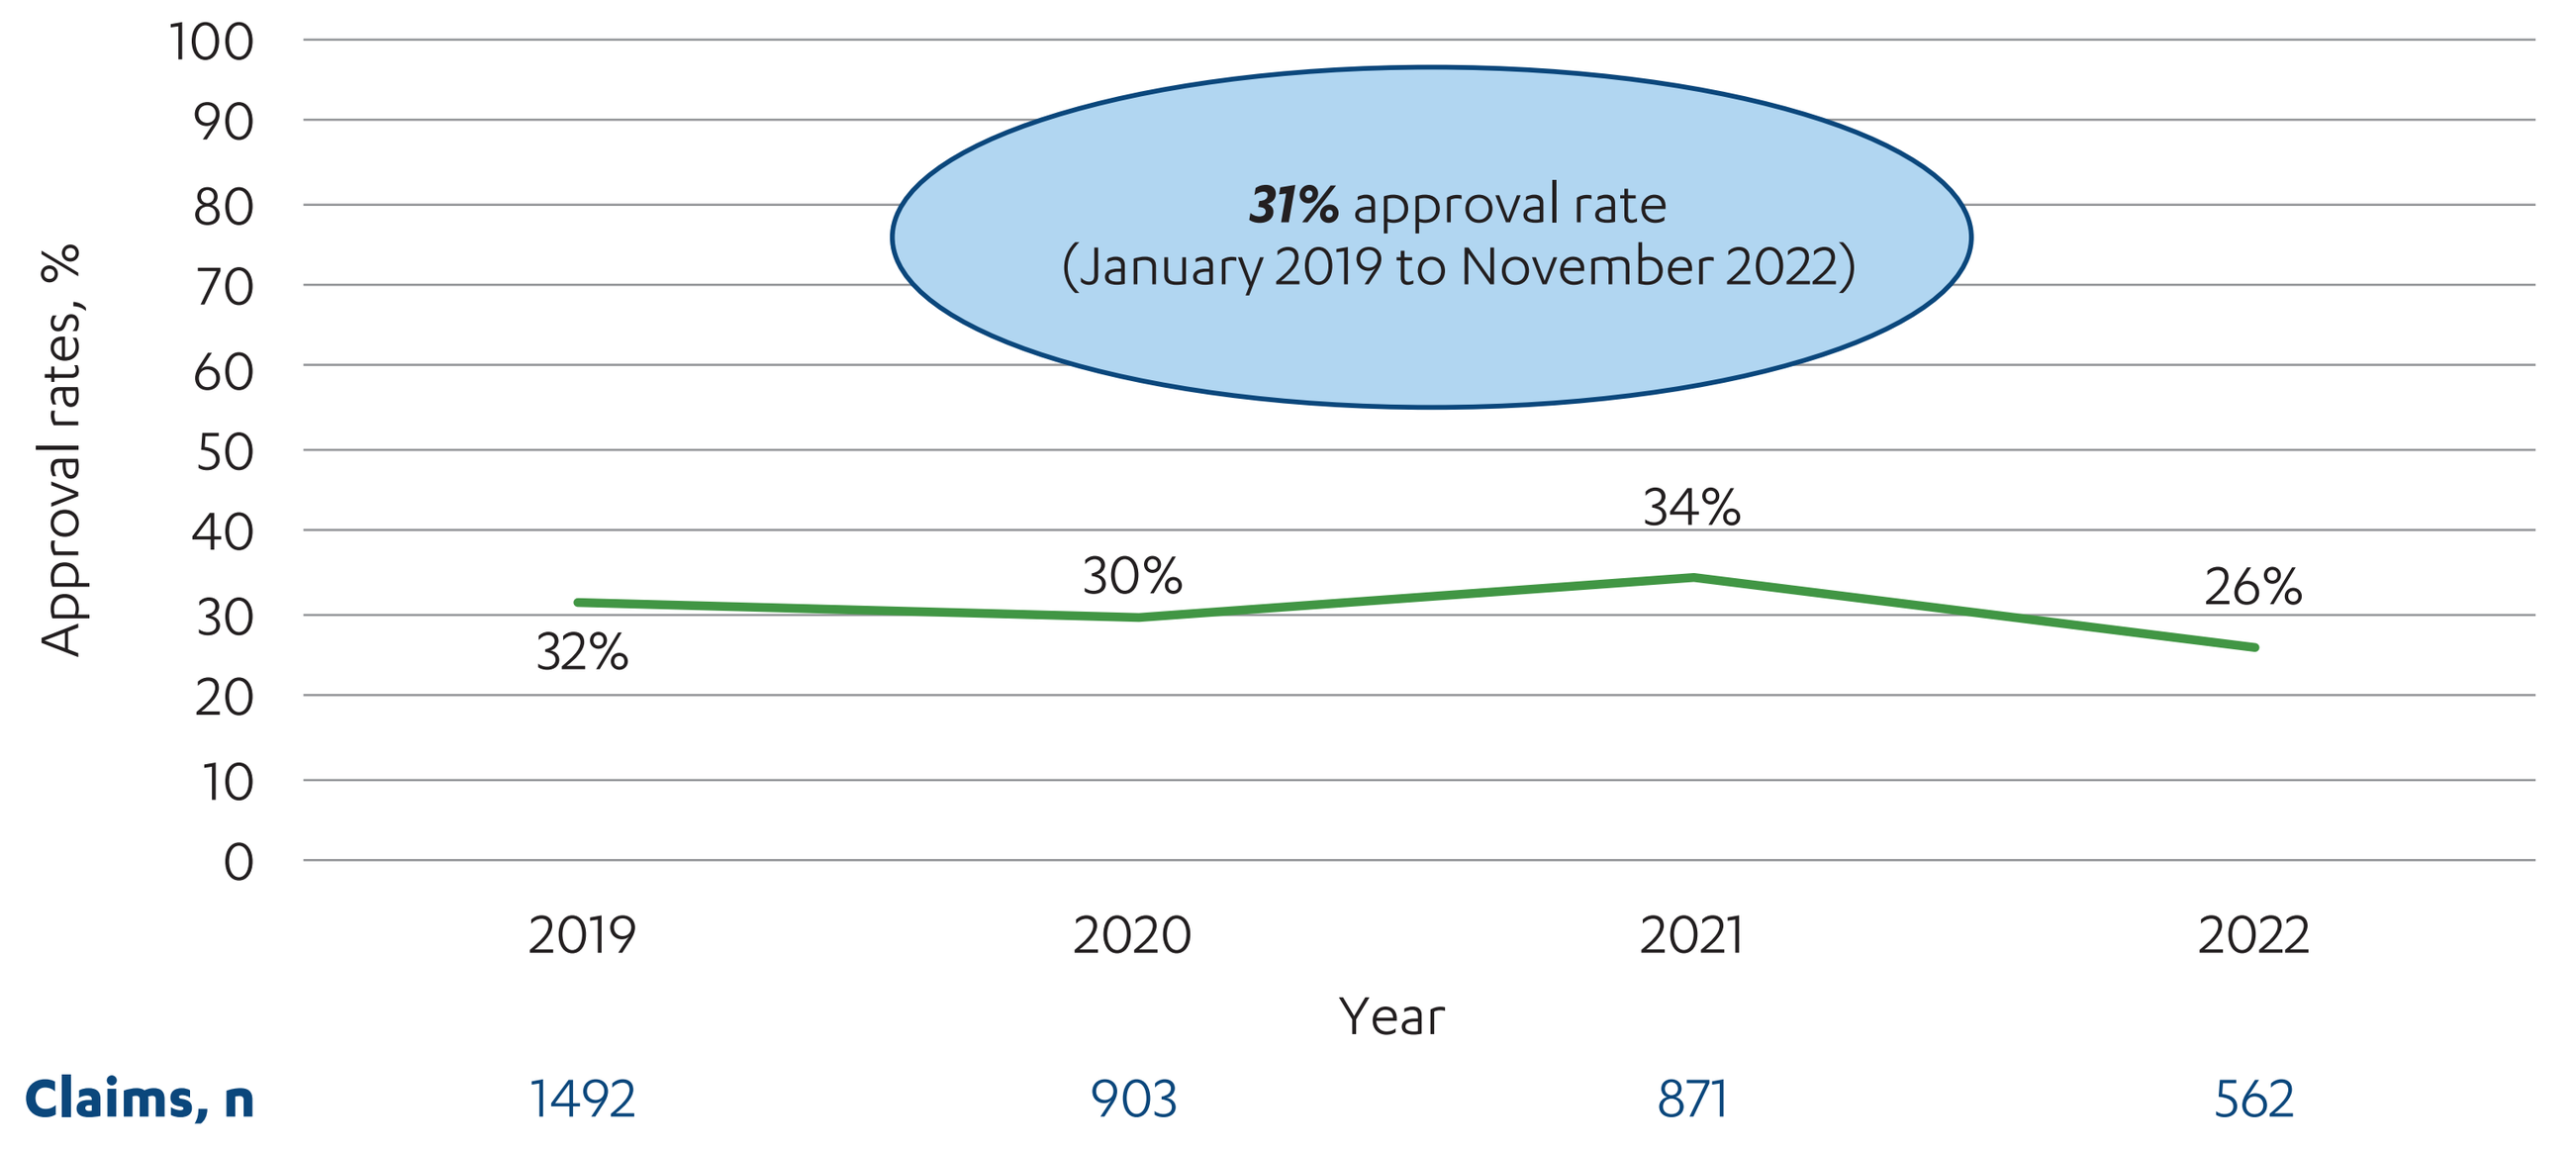
100
90
80
70
60
50
40
30
20
10
0
Approval rates, %
31% approval rate
(January 2019 to November 2022)
32%
30%
34%
26%
2019
2020
2021
2022
Year
Claims, n
1492
903
871
562

Figure 2. Overall reasons for rejection of molecular and genetic testing claims (n = 2649).

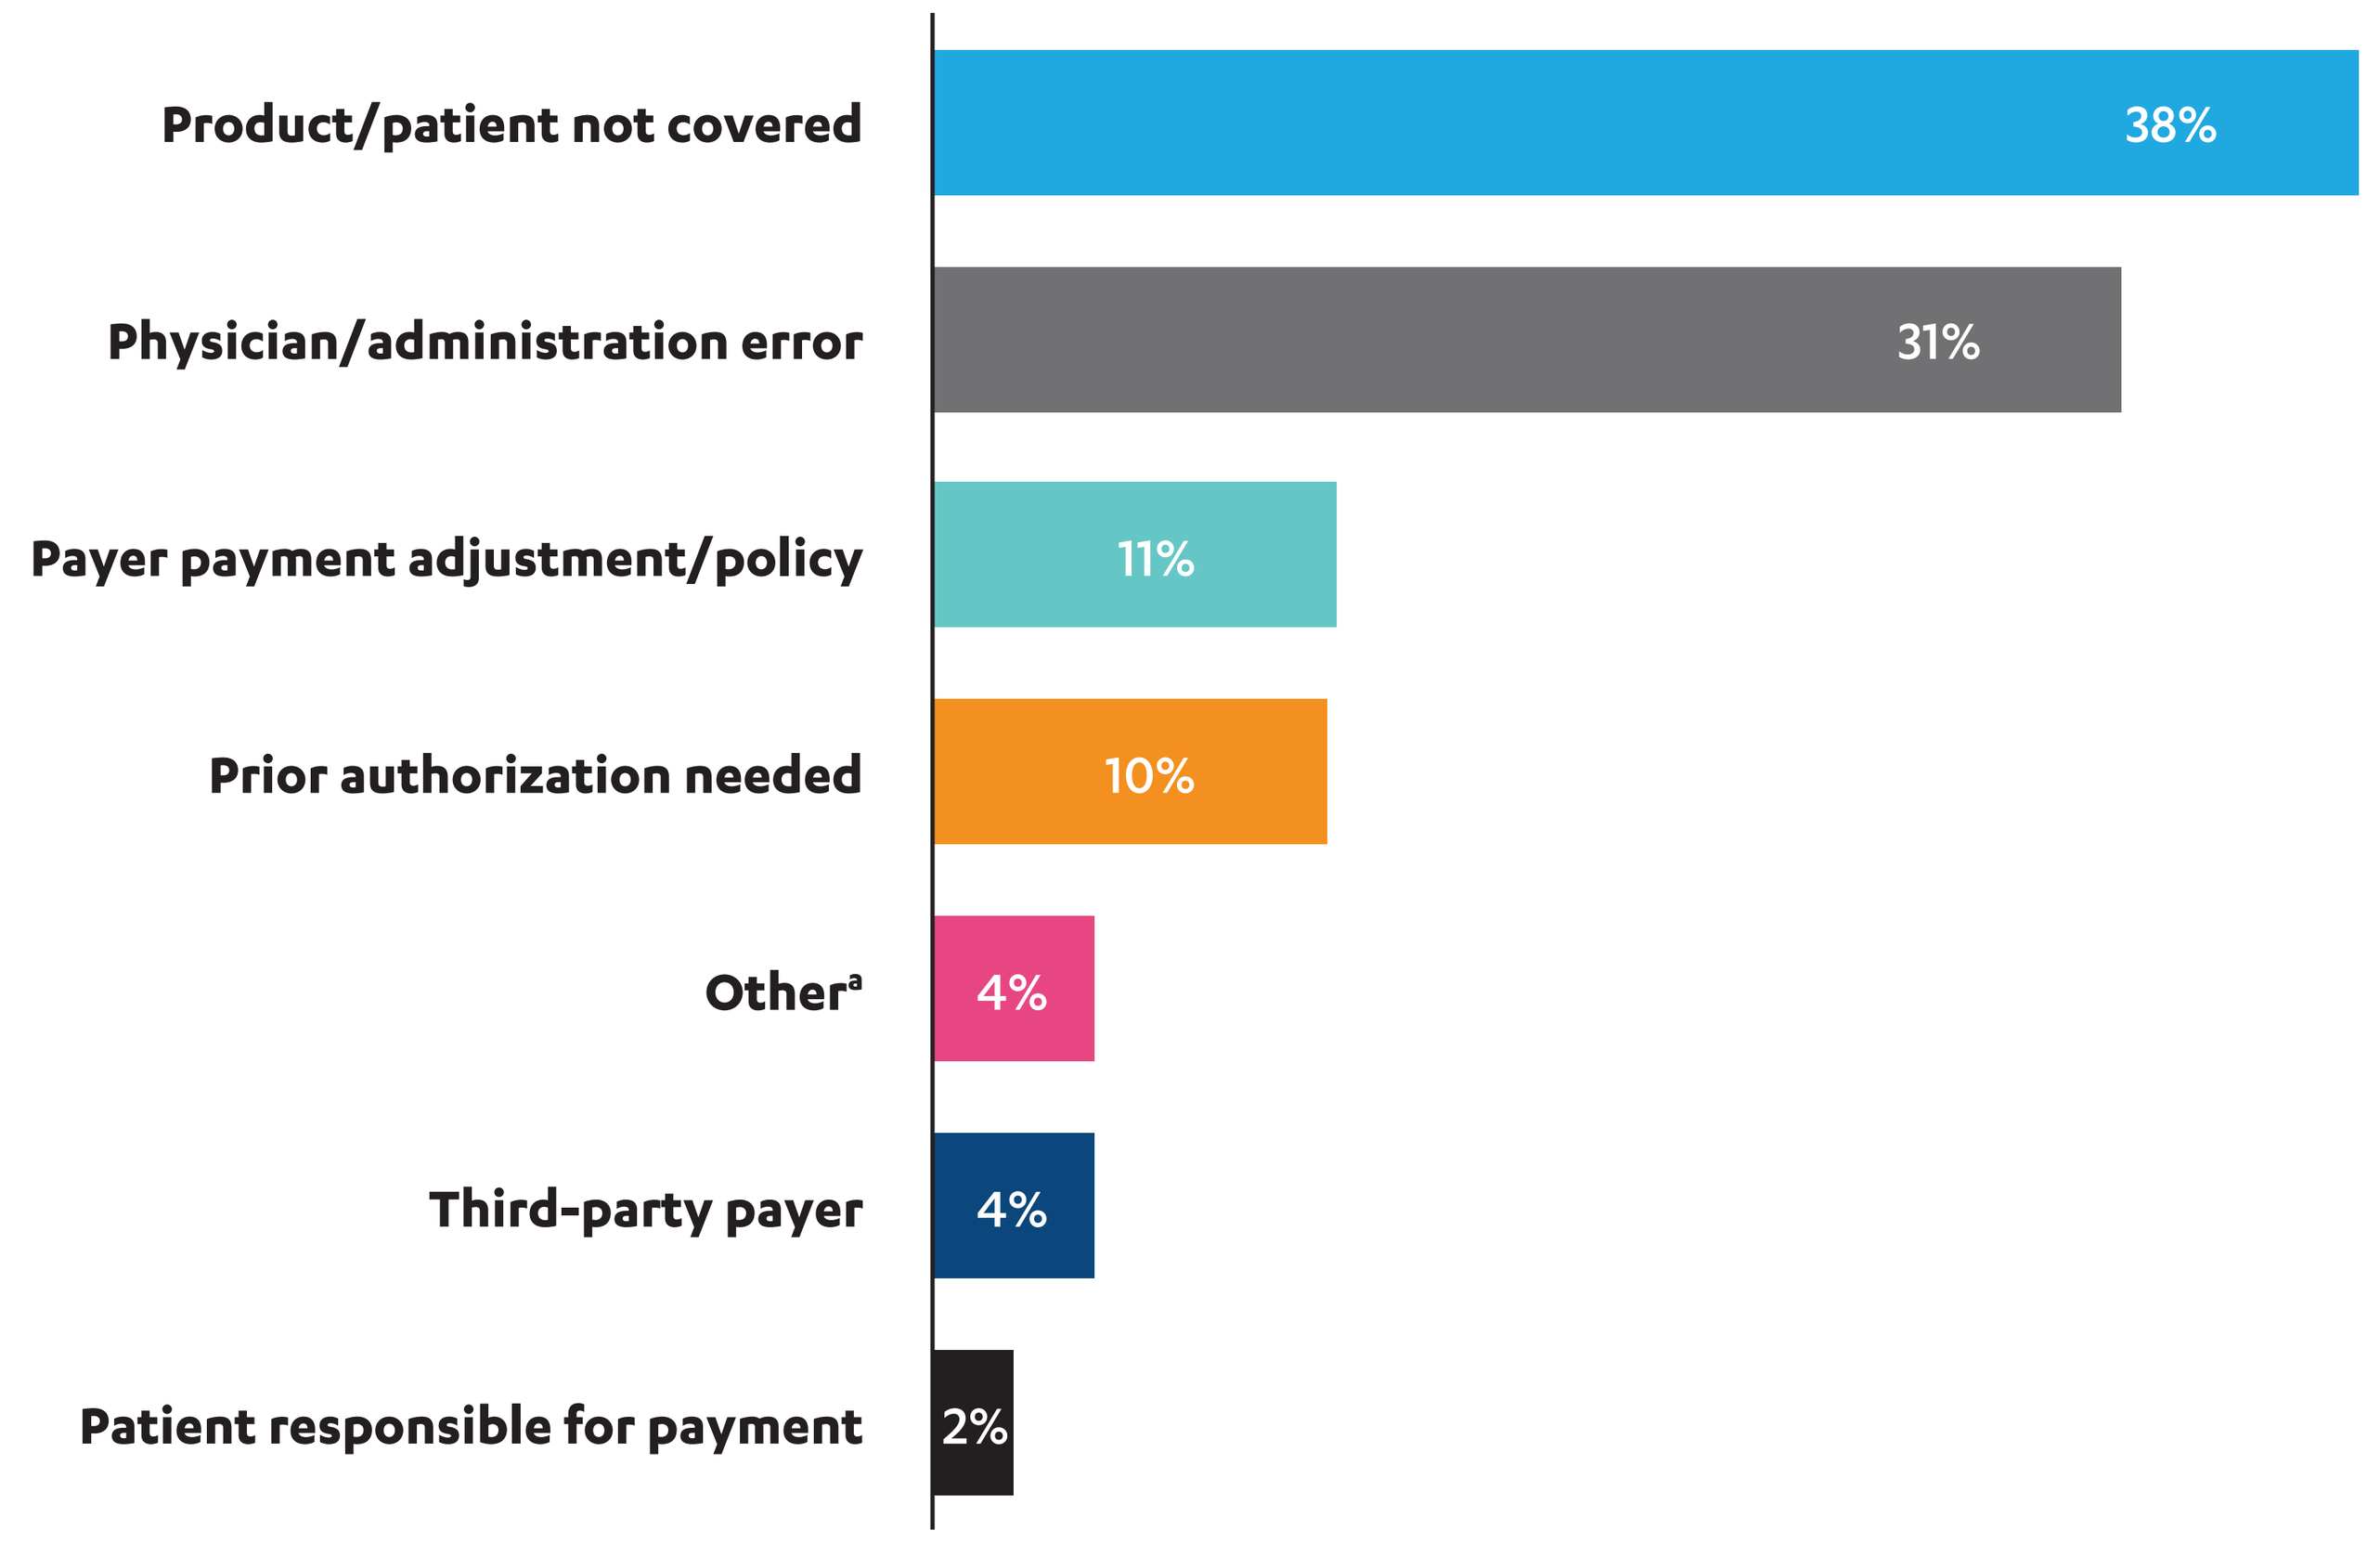
Product/patient not covered
38%
Physician/administration error
31%
Payer payment adjustment/policy
11%
Prior authorization needed
10%
Other[a]
4%
Third-party payer
4%
Patient responsible for payment
2%

[a]Includes rejected claims with reasons recorded as "unknown" (n = 79).

- Molecular and genetic testing claim approval rates were highest for patients covered by Medicare, at 47% overall, whereas claims from patients insured by commercial payers were approved at a lower rate of 22% (**Figure 3**)
 - Claims billed through other insurance types (eg, Veterans Affairs) were less frequently approved, with an 11% approval rate

Figure 3. Approval and rejection of molecular and genetic testing claims by common payer types.

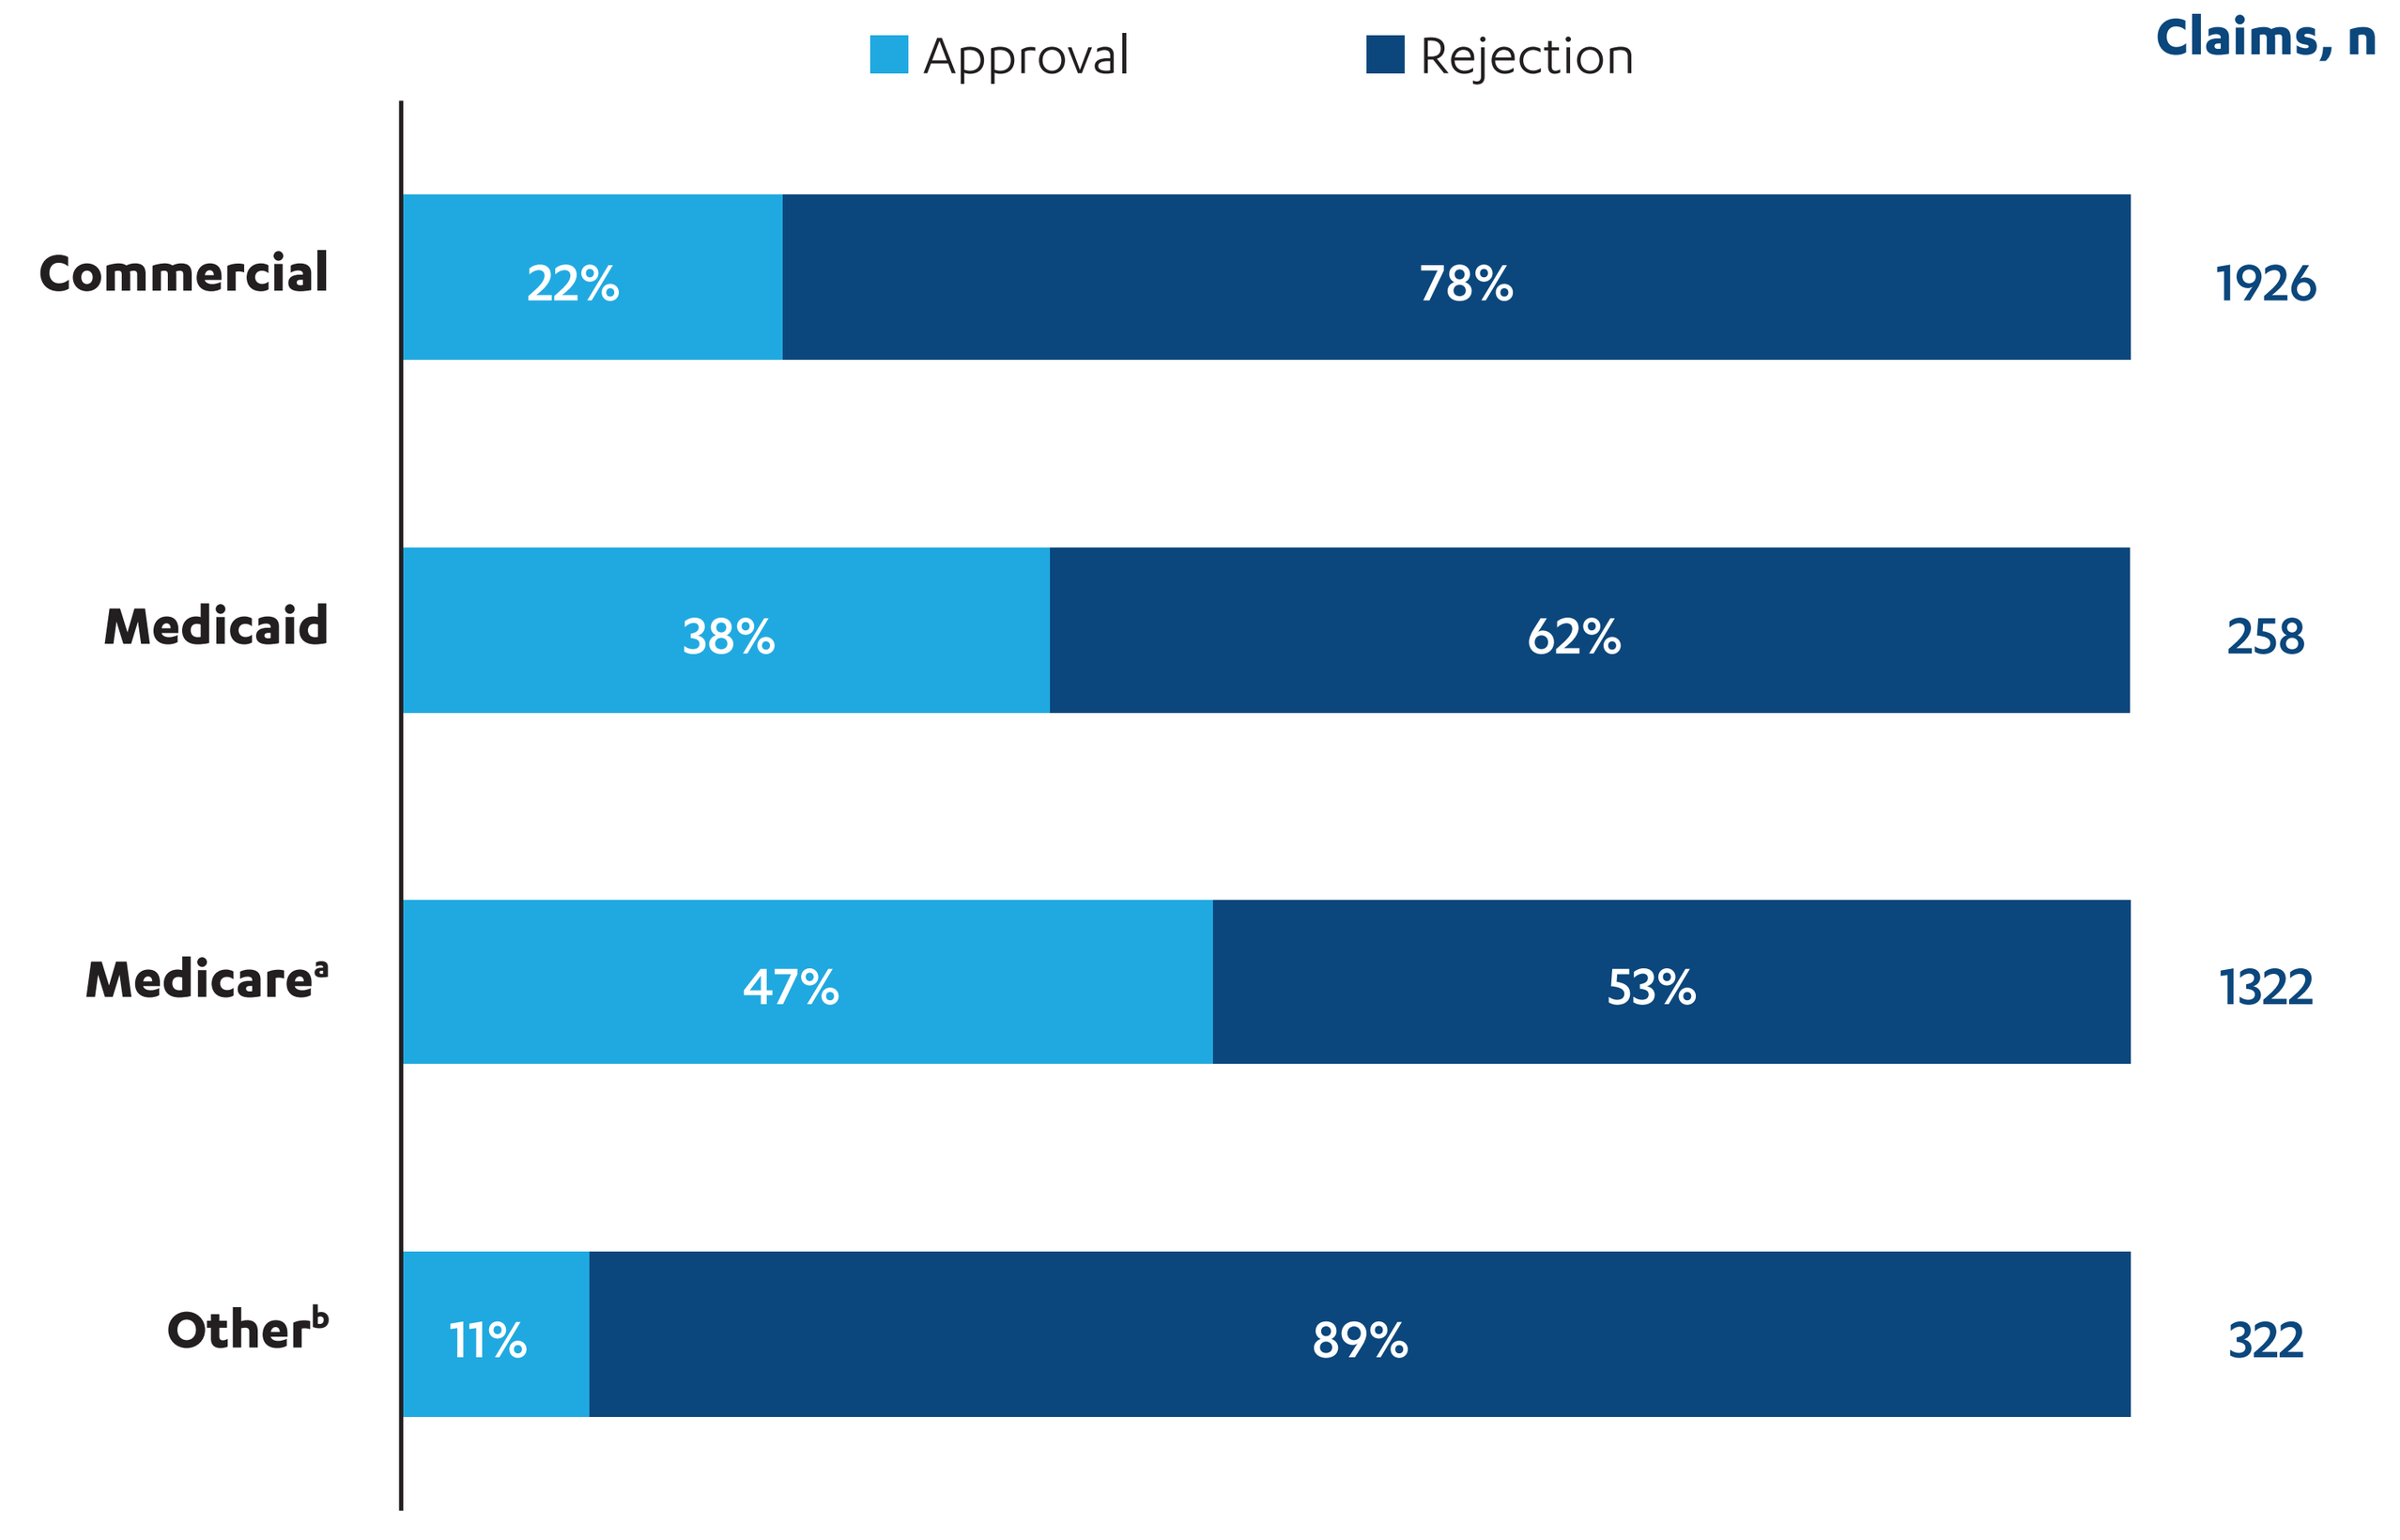
Approval
Rejection
Claims, n
Commercial
22%
78%
1926
Medicaid
38%
62%
258
Medicare[a]
47%
53%
1322
Other[b]
11%
89%
322

[a]Medicare also includes 1 claim from a patient with dual insurance (ie, Medicare and Medicaid).
[b]Other includes Veterans Affairs health care insurance, other special government insurance types, and unknown payer types.

Patient Out-of-Pocket and Physician Reimbursement Costs

- Of the paid claims captured based on the inclusion criteria, over 50% were paid by Medicare at a mean amount of $1578; 35% were paid by commercial payers at a mean amount of $606 (**Figure 4A**)
- The overall mean out-of-pocket (OOP) amount for a patient-paid claim was $11, whereas the mean payer-reimbursed amount was $1091 (**Figure 4B**)
- Within the top quartile of OOP amounts for patient-paid claims, the average OOP amount was $119

Figure 4. Mean (median) reimbursement amounts (A) and patient OOP costs[a] (B) for molecular and genetic testing claims by payer type.

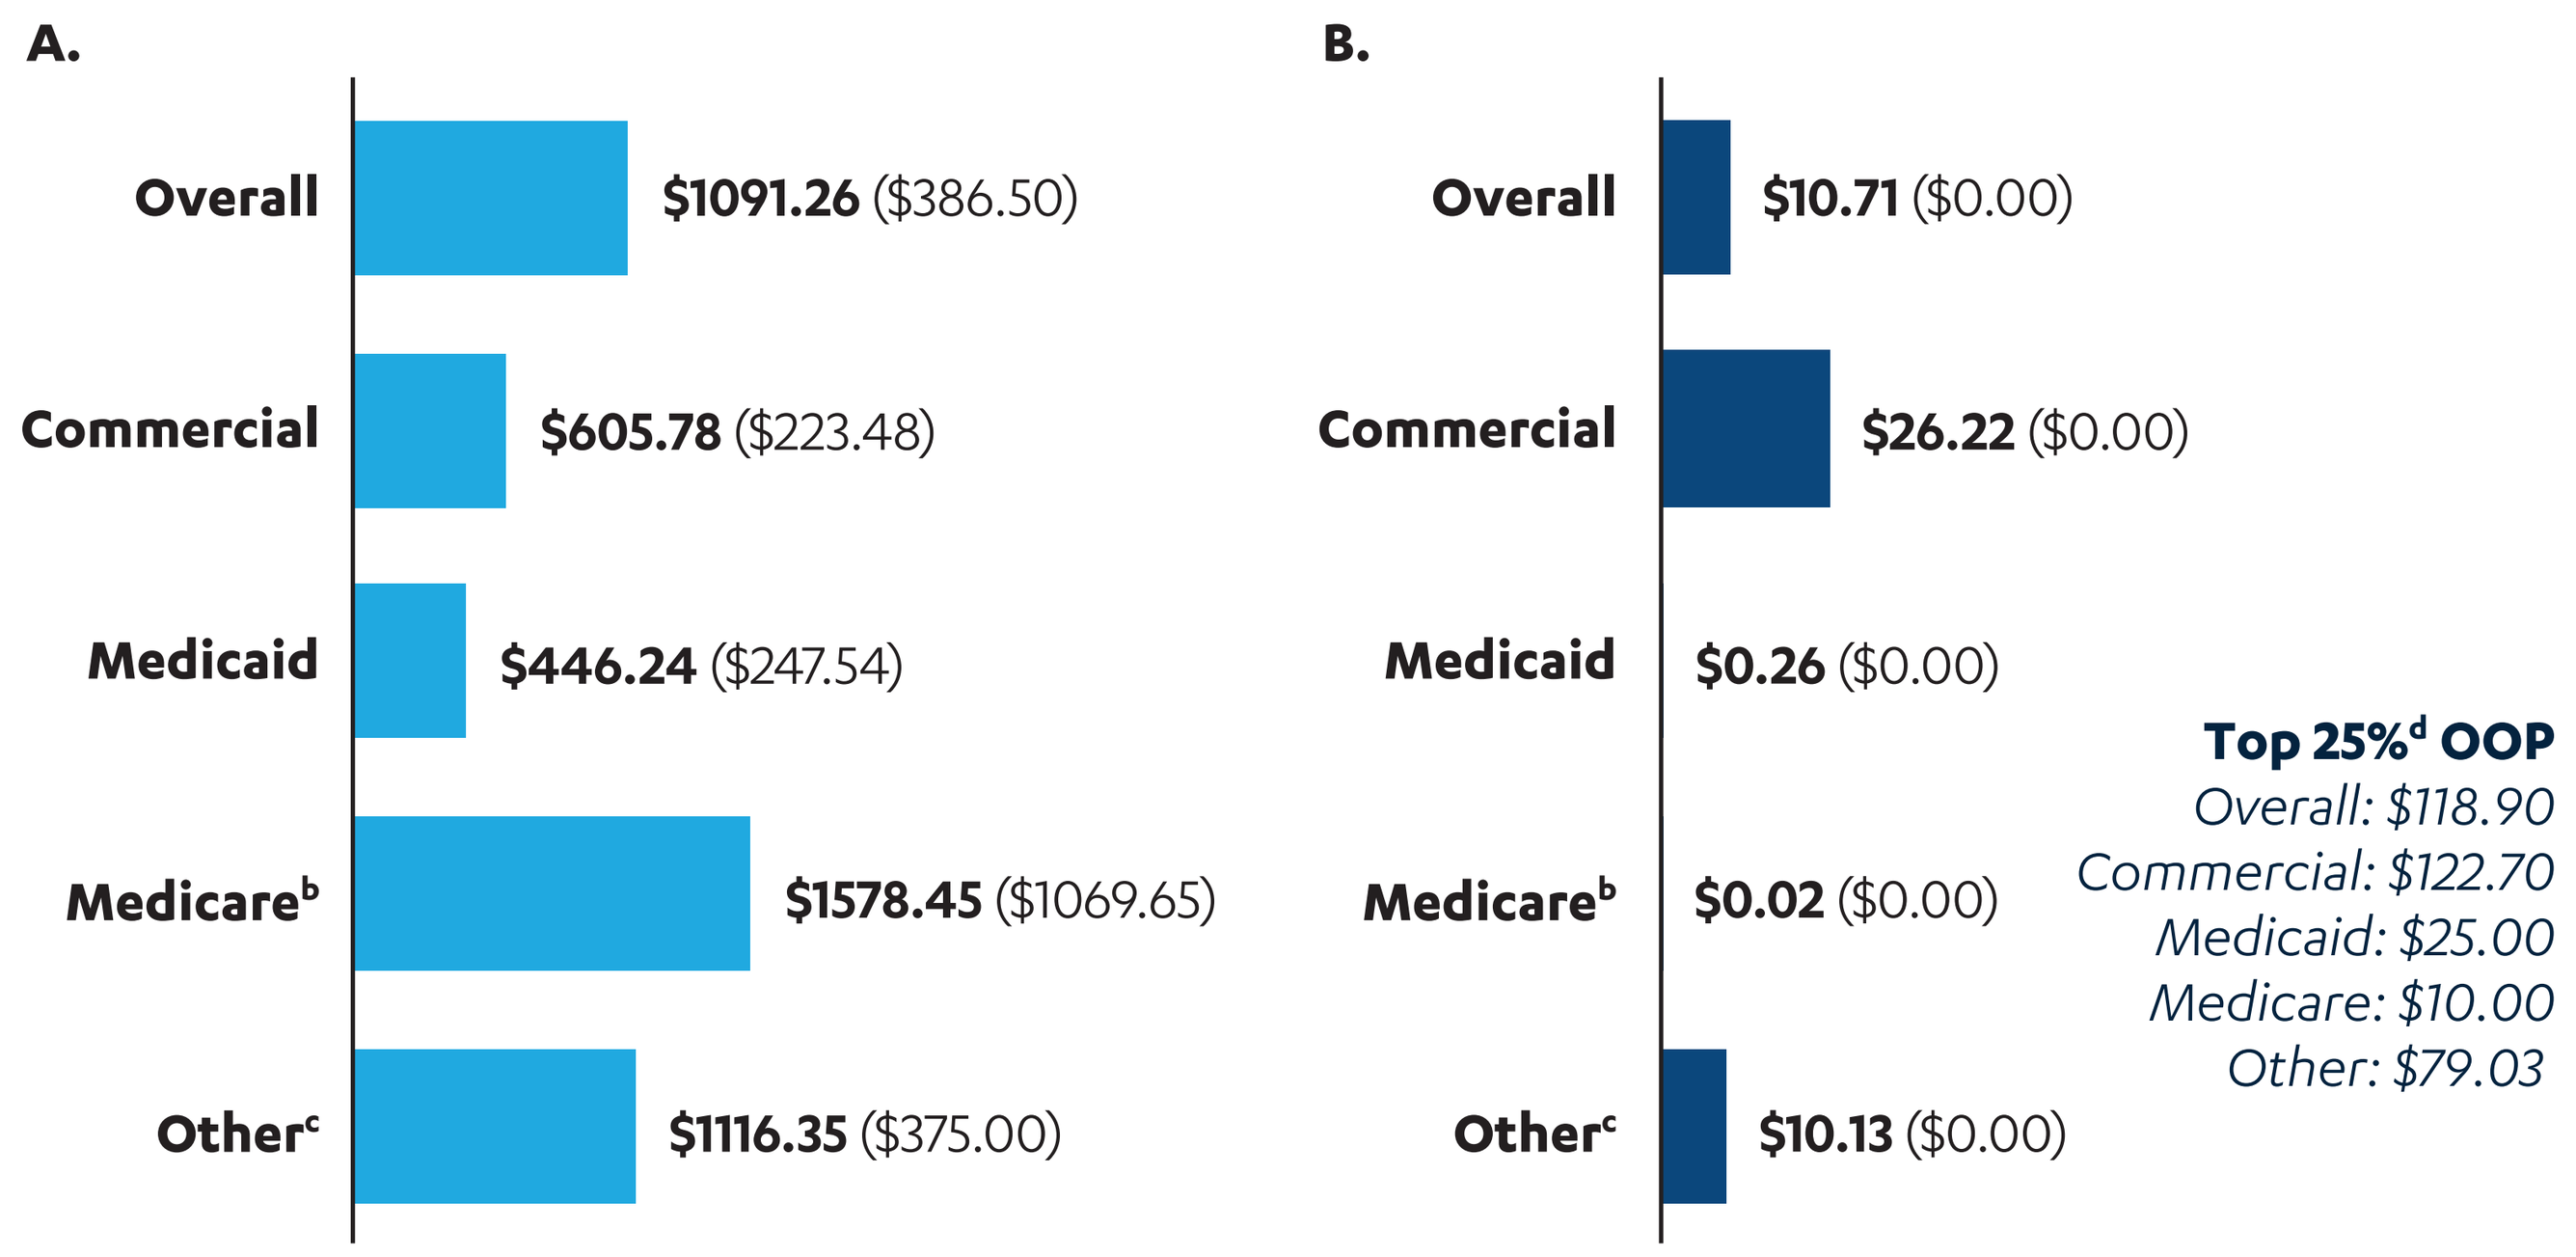
A.
Overall
$1091.26 ($386.50)
Commercial
$605.78 ($223.48)
Medicaid
$446.24 ($247.54)
Medicare[b]
$1578.45 ($1069.65)
Other[c]
$1116.35 ($375.00)
B.
Overall
$10.71 ($0.00)
Commercial
$26.22 ($0.00)
Medicaid
$0.26 ($0.00)
Medicare[b]
$0.02 ($0.00)
Other[c]
$10.13 ($0.00)
Top 25%[d] OOP
Overall: $118.90
Commercial: $122.70
Medicaid: $25.00
Medicare: $10.00
Other: $79.03

OOP, out-of-pocket.
[a]Median amounts are equal to $0.00 for all payer types due to the large number of $0 claims.
[b]Medicare also includes 1 claim from a patient with dual insurance (ie, Medicare and Medicaid).
[c]Other includes Veterans Affairs health care insurance, other special government insurance types, and unknown payer types.
[d]Average OOP cost to patient based on the top 25% of patient-paid claims.

LIMITATIONS

- The claims database did not capture free genetic testing kits sponsored by pharmaceutical companies and patient advocacy groups. Therefore the real-world genetic testing rate in IRDs may be higher than 2%
- The exact names and contents for the molecular and genetic testing are not identifiable in the database
- The patient deductible amount was $0 for most of the claims; it is unclear whether the $0 copay amount was due to the maximized annual deductible

CONCLUSIONS

- Molecular and genetic testing may enable patients to receive treatments, such as voretigene neparvovec-rzyl, or to enroll in clinical trials. However, these real-world data suggest low utilization of molecular and genetic testing by health care professionals, even though the OOP expenditures and cost of testing may be affordable to many patients with IRDs and payers
- Eye care professionals and payers need to be informed of the importance of molecular and genetic testing given the current significant increase in clinical development for patients with IRDs, in which previously no treatments were available

REFERENCES

1. Henderson RH. *Pediatrics and Child Health.* 2020;30(1):19-27.
2. Cross N, et al. *Clin Ophthalmol.* 2022;16:1993-2010.
3. Hu ML, et al. *Clin Exp Optom.* 2021;104(4):444-454.
4. Stone EM, et al. *Ophthalmology.* 2012;119(11):2408-2410.
5. Decision Resources Group Real World Evidence US Data Repository.

ACKNOWLEDGMENTS

This study was sponsored by Janssen Scientific Affairs, LLC. Medical writing support was provided by David Le, PharmD, of Lumanity Communications Inc., and was funded by Janssen Scientific Affairs, LLC.

DISCLOSURES

C-H Lee: Employee – Janssen Scientific Affairs, LLC; Stock – Johnson & Johnson. **J Liu:** Employee – Clarivate. **P Kumar:** Employee – Clarivate. **Pranshu:** Employee – Clarivate. **J Stahl:** Employee – Clarivate. **S Hengst:** Employee – Clarivate. **D Milentijevic:** Employee – Janssen Scientific Affairs, LLC.

Poster presented at The Professional Society for Health Economics and Outcomes Research (ISPOR) Annual Meeting; May 7-10, 2023; Boston, MA, USA.

PHARMACEUTICAL COMPANIES OF Johnson & Johnson