EPH112

Economic impact of rapid treatment initiation of HIV with bictegravir/emtricitabine /tenofovir alafenamide (B/F/TAF) from a Canadian healthcare perspective

Guinan K[1], Mathurin K[1,2], Lachaine J[1,2]

[1]PeriPharm, Montreal, QC, Canada, [2]University of Montreal, Montreal, QC, Canada,

Presented at ISPOR 2023, May 7-10, 2023, Boston Conference & Exhibition Center, Boston, Massachusetts

INTRODUCTION

- Sexually transmitted and blood borne infections, such as the human immunodeficiency virus (**HIV**) remain a significant health concern in Canada. Even if HIV is preventable and treatable in many cases, HIV imposes a significant physical, emotional, and economic cost.[1]
- Antiretroviral therapy (**ART**) is the standard of care to treat HIV.[2] However, evidence suggests that delaying the initiation of ART increases the incidence of HIV by contributing to its transmission.
- Bictegravir/emtricitabine/tenofovir alafenamide (**B/F/TAF**) is a combination of three ARTs that has the attributes to support rapid treatment initiation compared to the actual clinical practice.[3]
- Immediate treatment initiation at diagnosis can help patients achieve and maintain virological suppression earlier in their infection and prevent new HIV infections.[4,5]
- Rapid treatment initiation may also be beneficial for difficult-to-treat patients, such as people who inject drugs (**PWID**), where delays in starting treatment may result in the patient disengaged with care.[5]
- Although multiple benefits have been demonstrated with rapid ART treatment initiation, there is still a delay in initiating the treatment in Canada after HIV diagnosis, thus impacting the disease burden.

OBJECTIVE

- The objective of this study was to assess, from a Canadian perspective, the potential epidemiological and economic impact of rapid initiation of HIV treatment with B/F/TAF compared to current initiation in clinical practice.

METHODS

- A dynamic transmission model for HIV was adapted to the Canadian setting to assess the impact of rapid treatment initiation with B/F/TAF compared to current clinical practice.

Rapid treatment initiation with B/F/TAF (**7 days** from diagnosis to treatment)[6]

VS.

Current clinical practice (**45 days** from diagnosis to treatment)[2]

- A Markov tree was developed. Three key subgroups were considered:
 - Heterosexual men and women (**Het**)
 - Men who have sex with men (**MSM**)
 - PWID

- The prevalent HIV population was divided by health states, each with different risks of transmission, by subgroup (**Figure 1**).

Figure 1. Markov Model Structure

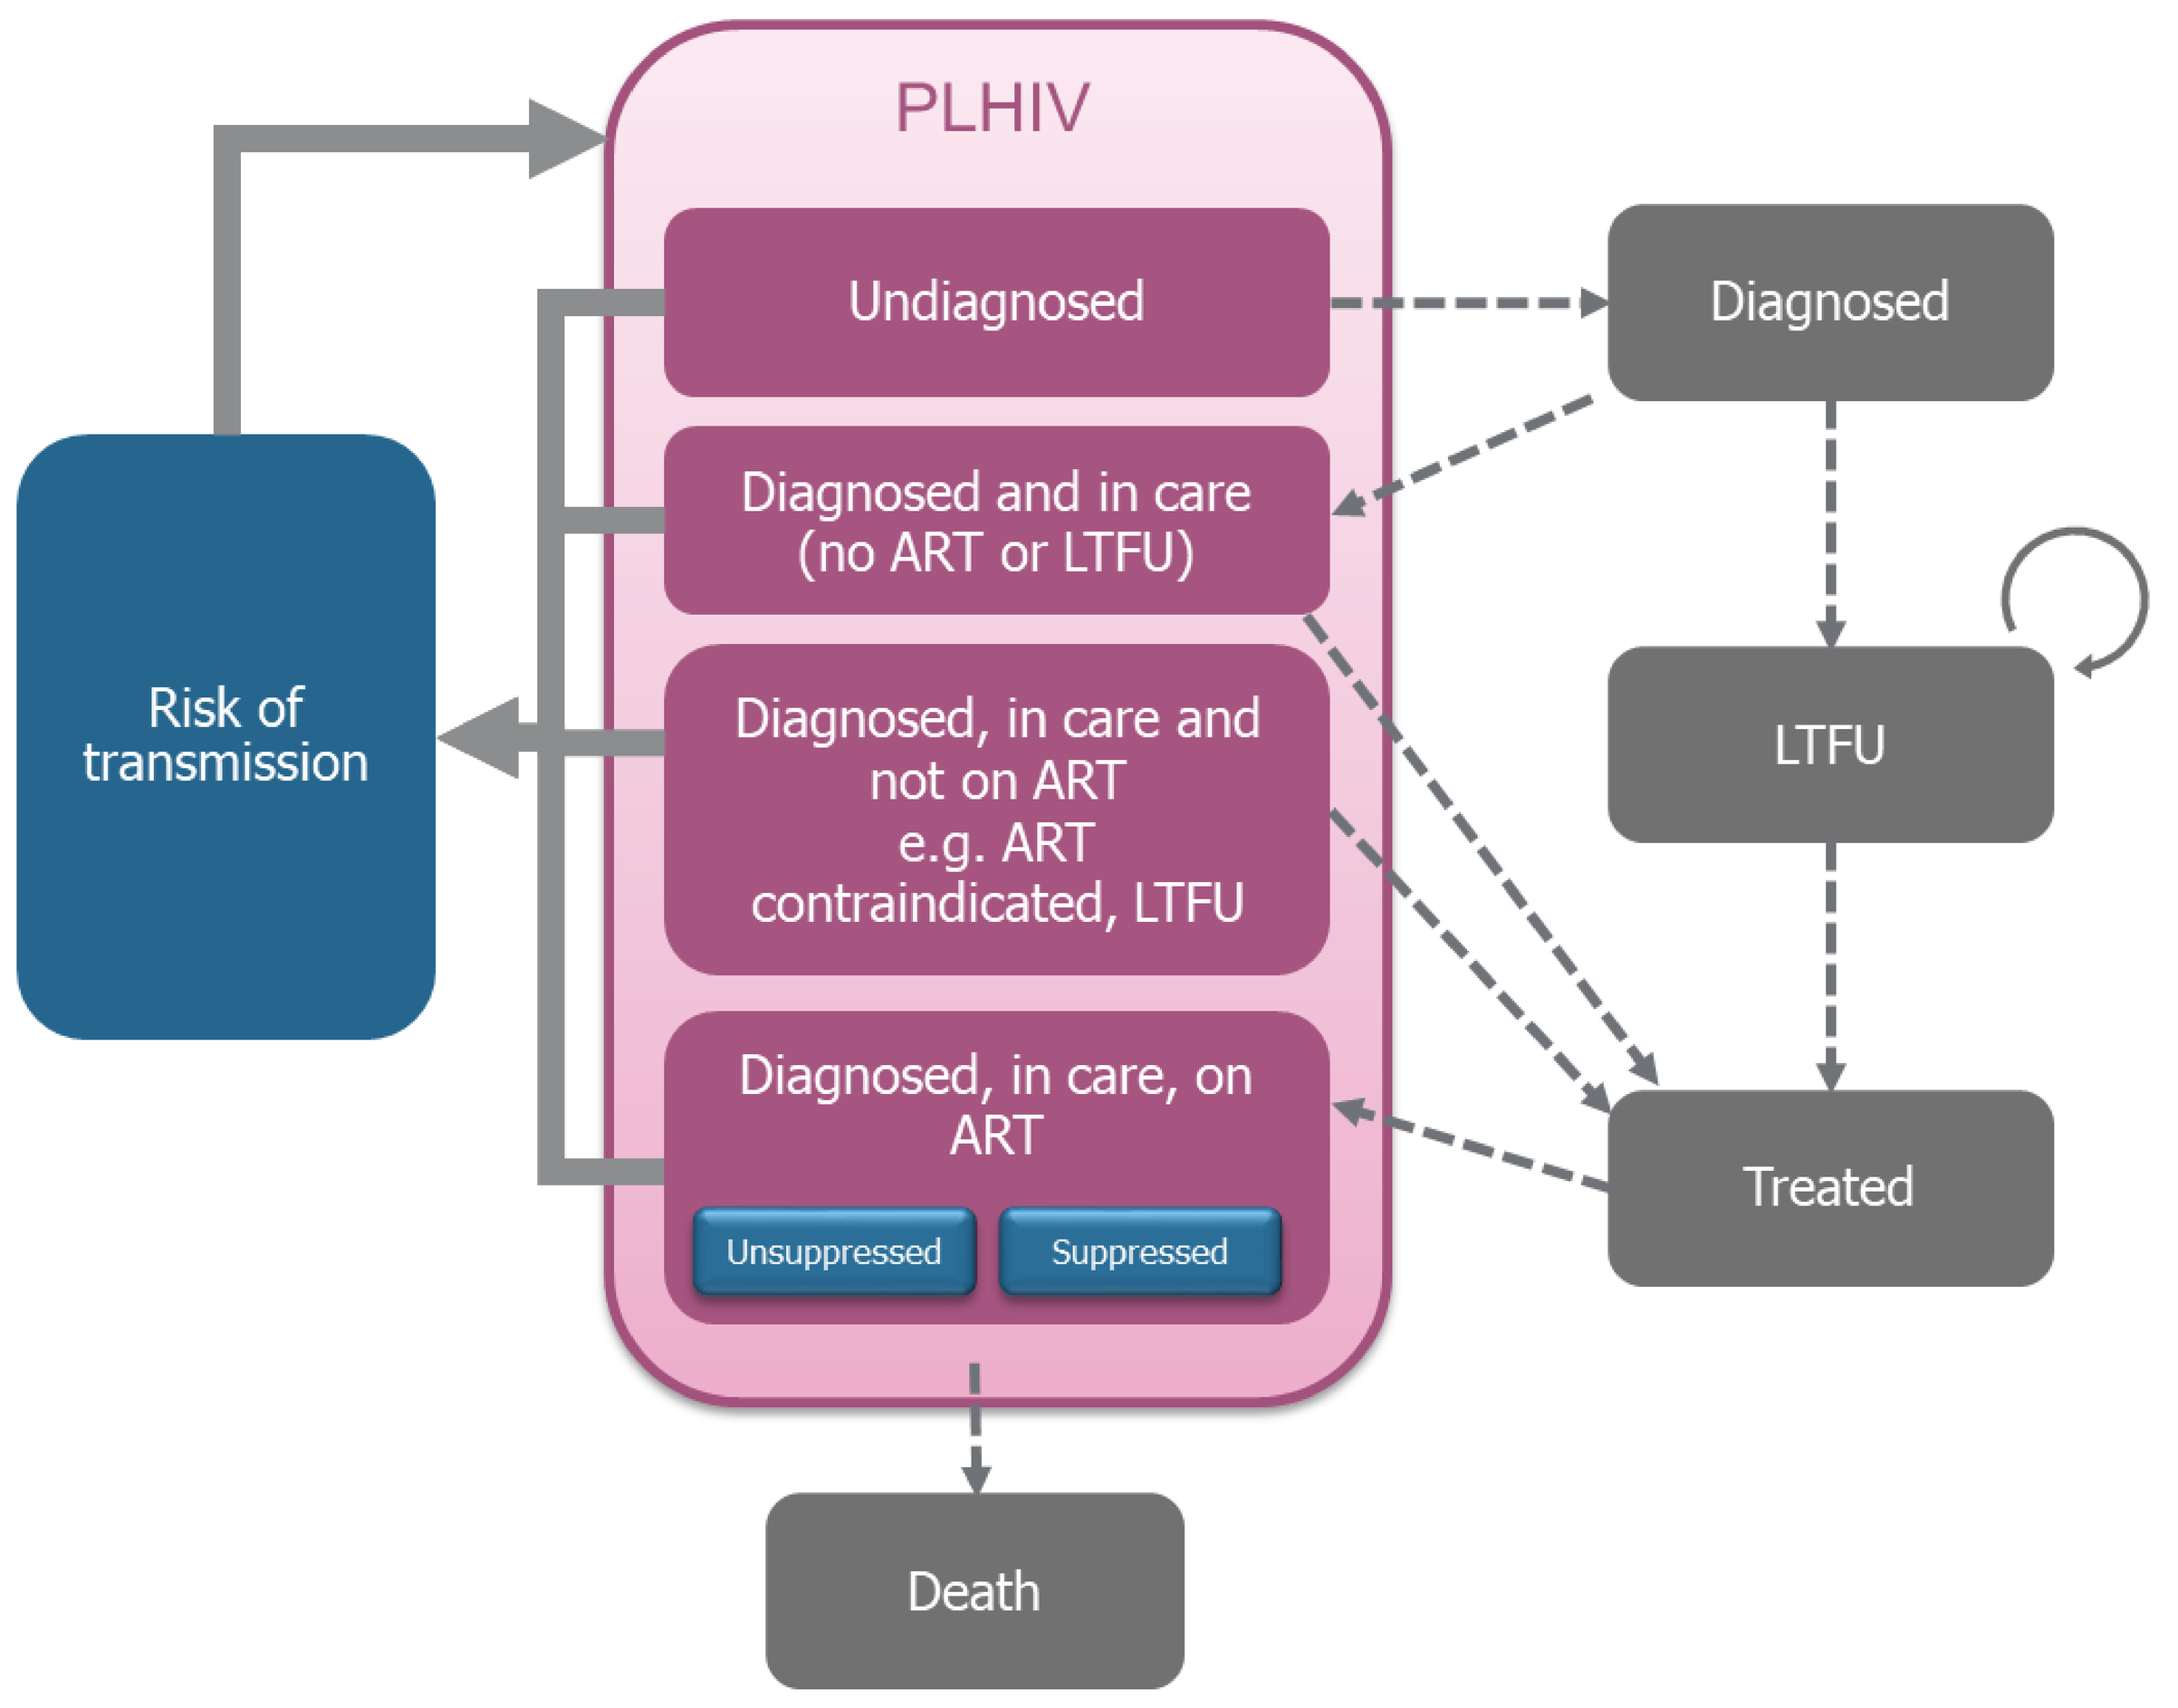
PLHIV
Undiagnosed
Diagnosed and in care (no ART or LTFU)
Diagnosed, in care and not on ART e.g. ART contraindicated, LTFU
Diagnosed, in care, on ART
Unsuppressed
Suppressed
Risk of transmission
Diagnosed
LTFU
Treated
Death

Abbreviations: ART: antiretroviral therapy; **LTFU:** lost to follow-up; **PLHIV:** people living with HIV

- Infectious individuals contribute to the incidence of new infections.
- Lifetime direct health care costs of HIV were applied to infected patients. Productivity costs were added in a scenario analysis.[7]
- Analyses were conducted from a public healthcare perspective for a time horizon of 20 years (year 2020 to 2040).
- The robustness of the results was assessed using one-way sensitivity analyses (**OWSA**).

RESULTS

- The model predicts a decline in incidence, in both strategies, with an average decline of 0.93% and 0.81%, in the rapid treatment initiation with B/F/TAF and current clinical practice, respectively (**Figure 2**).
- Rapid B/F/TAF initiation has the potential to result in 415 fewer Canadian HIV infections over a 20-year period, compared to current clinical practice (**Figure 3**).

Figure 2. Predicted Overall Annual HIV Incidence by Strategy

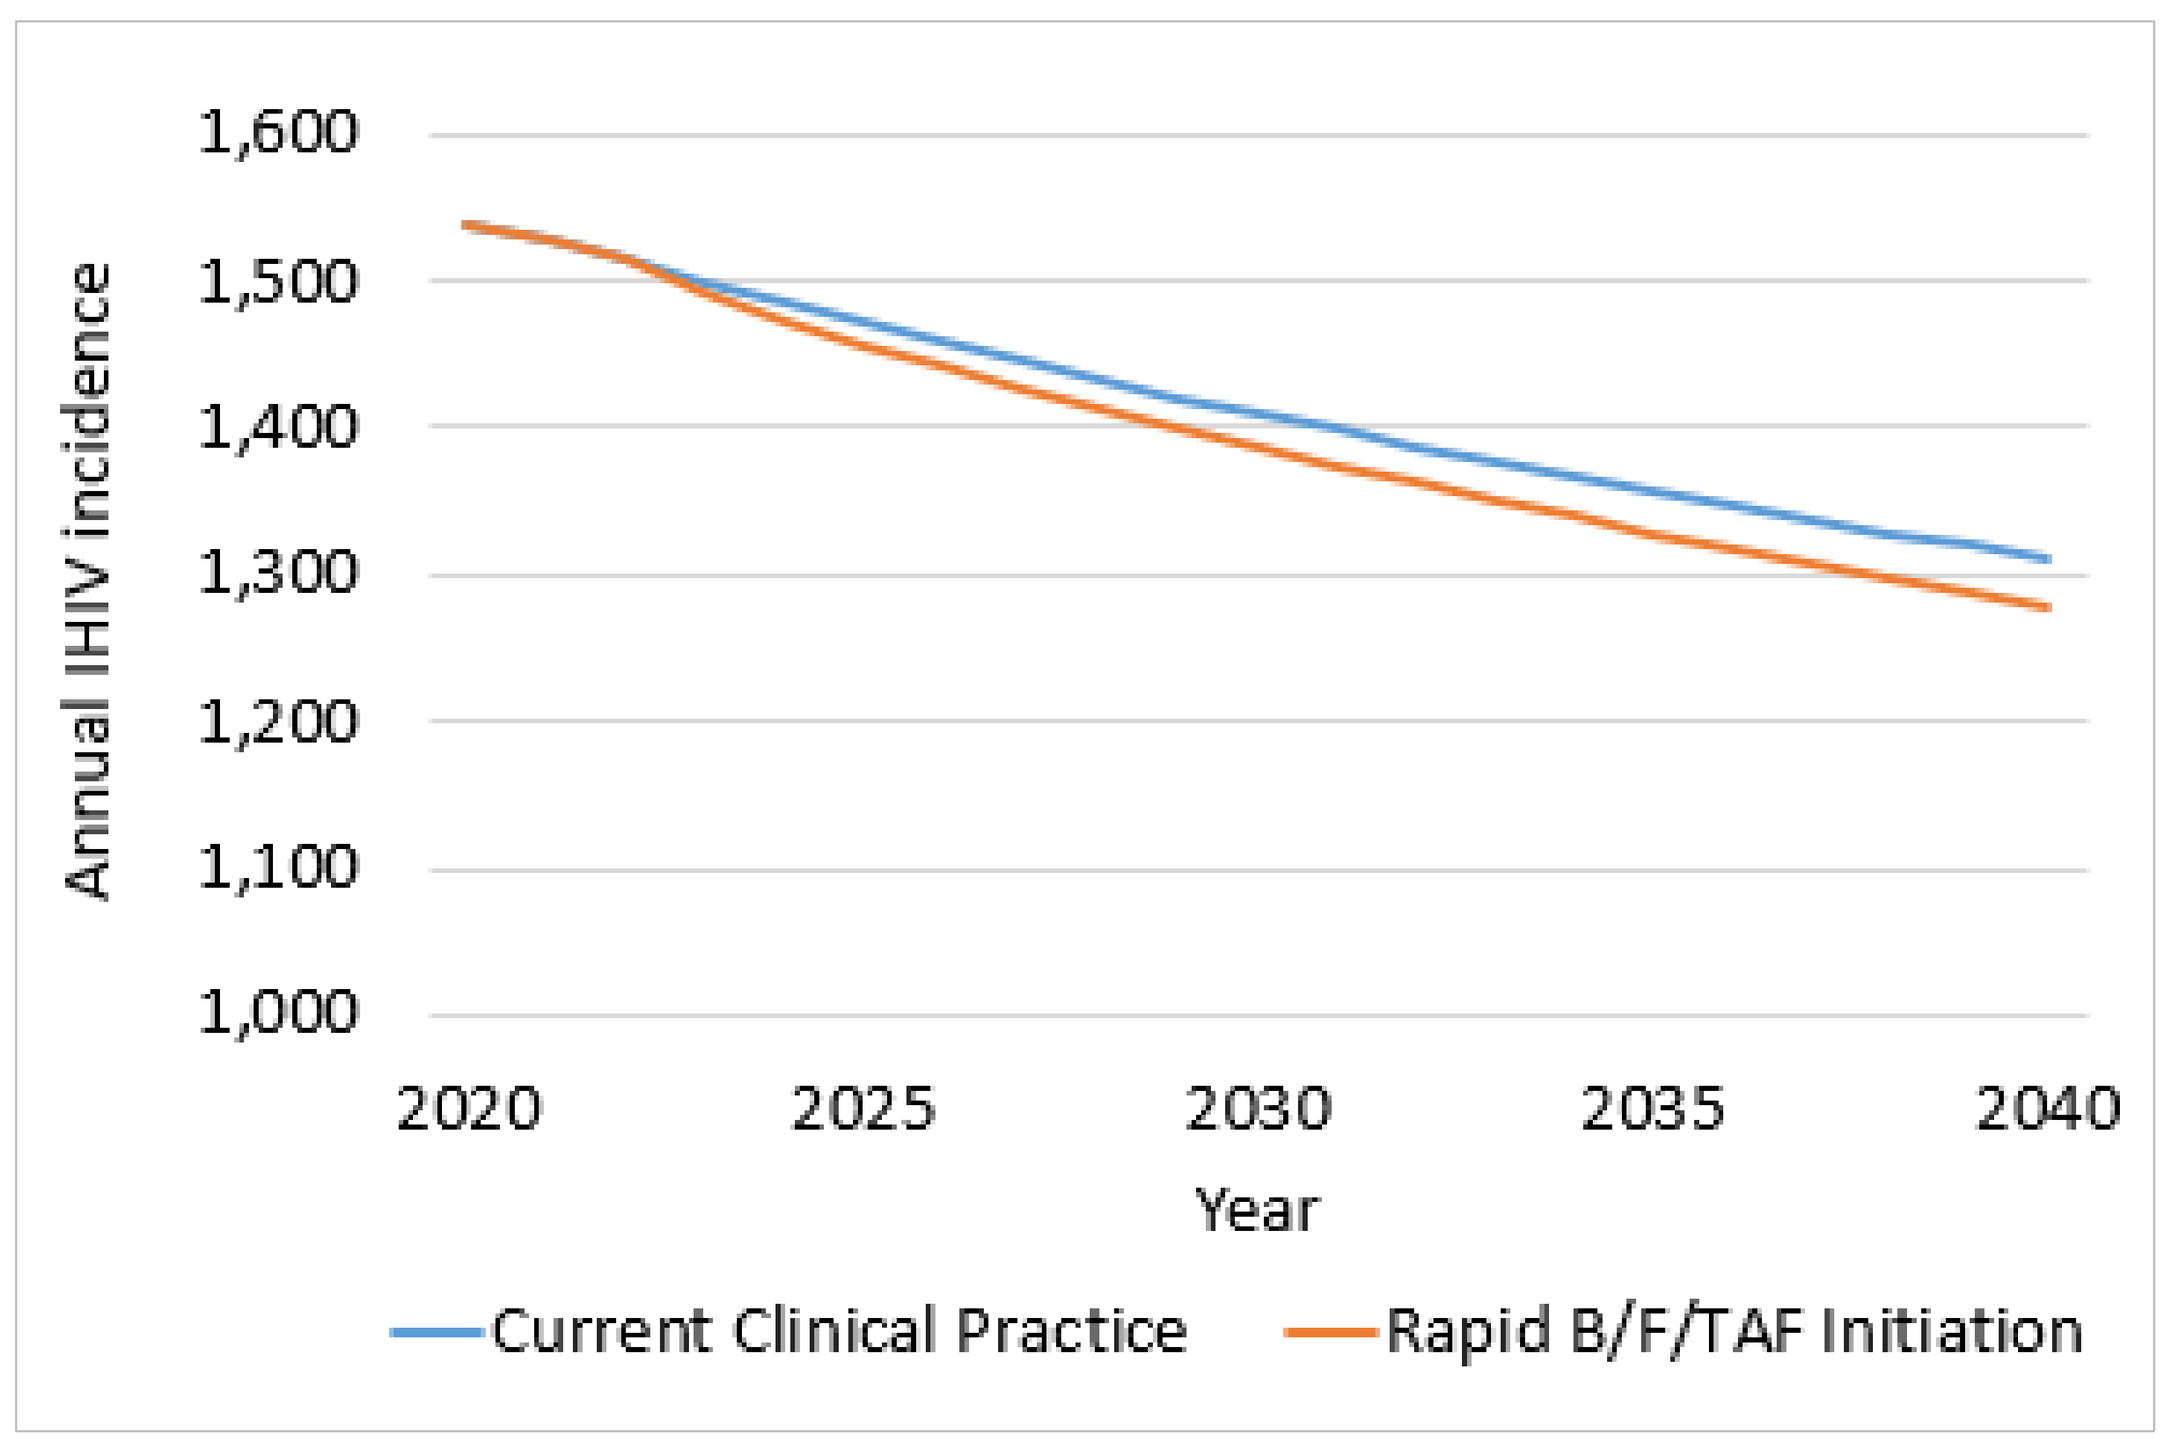
1,600
1,500
1,400
1,300
1,200
1,100
1,000
Annual IHIV incidence
2020
2025
2030
2035
2040
Year
Current Clinical Practice
Rapid B/F/TAF Initiation

Figure 3. Overall Difference in Cumulative Incidence: Current Clinical Practice vs Rapid B/F/TAF Initiation

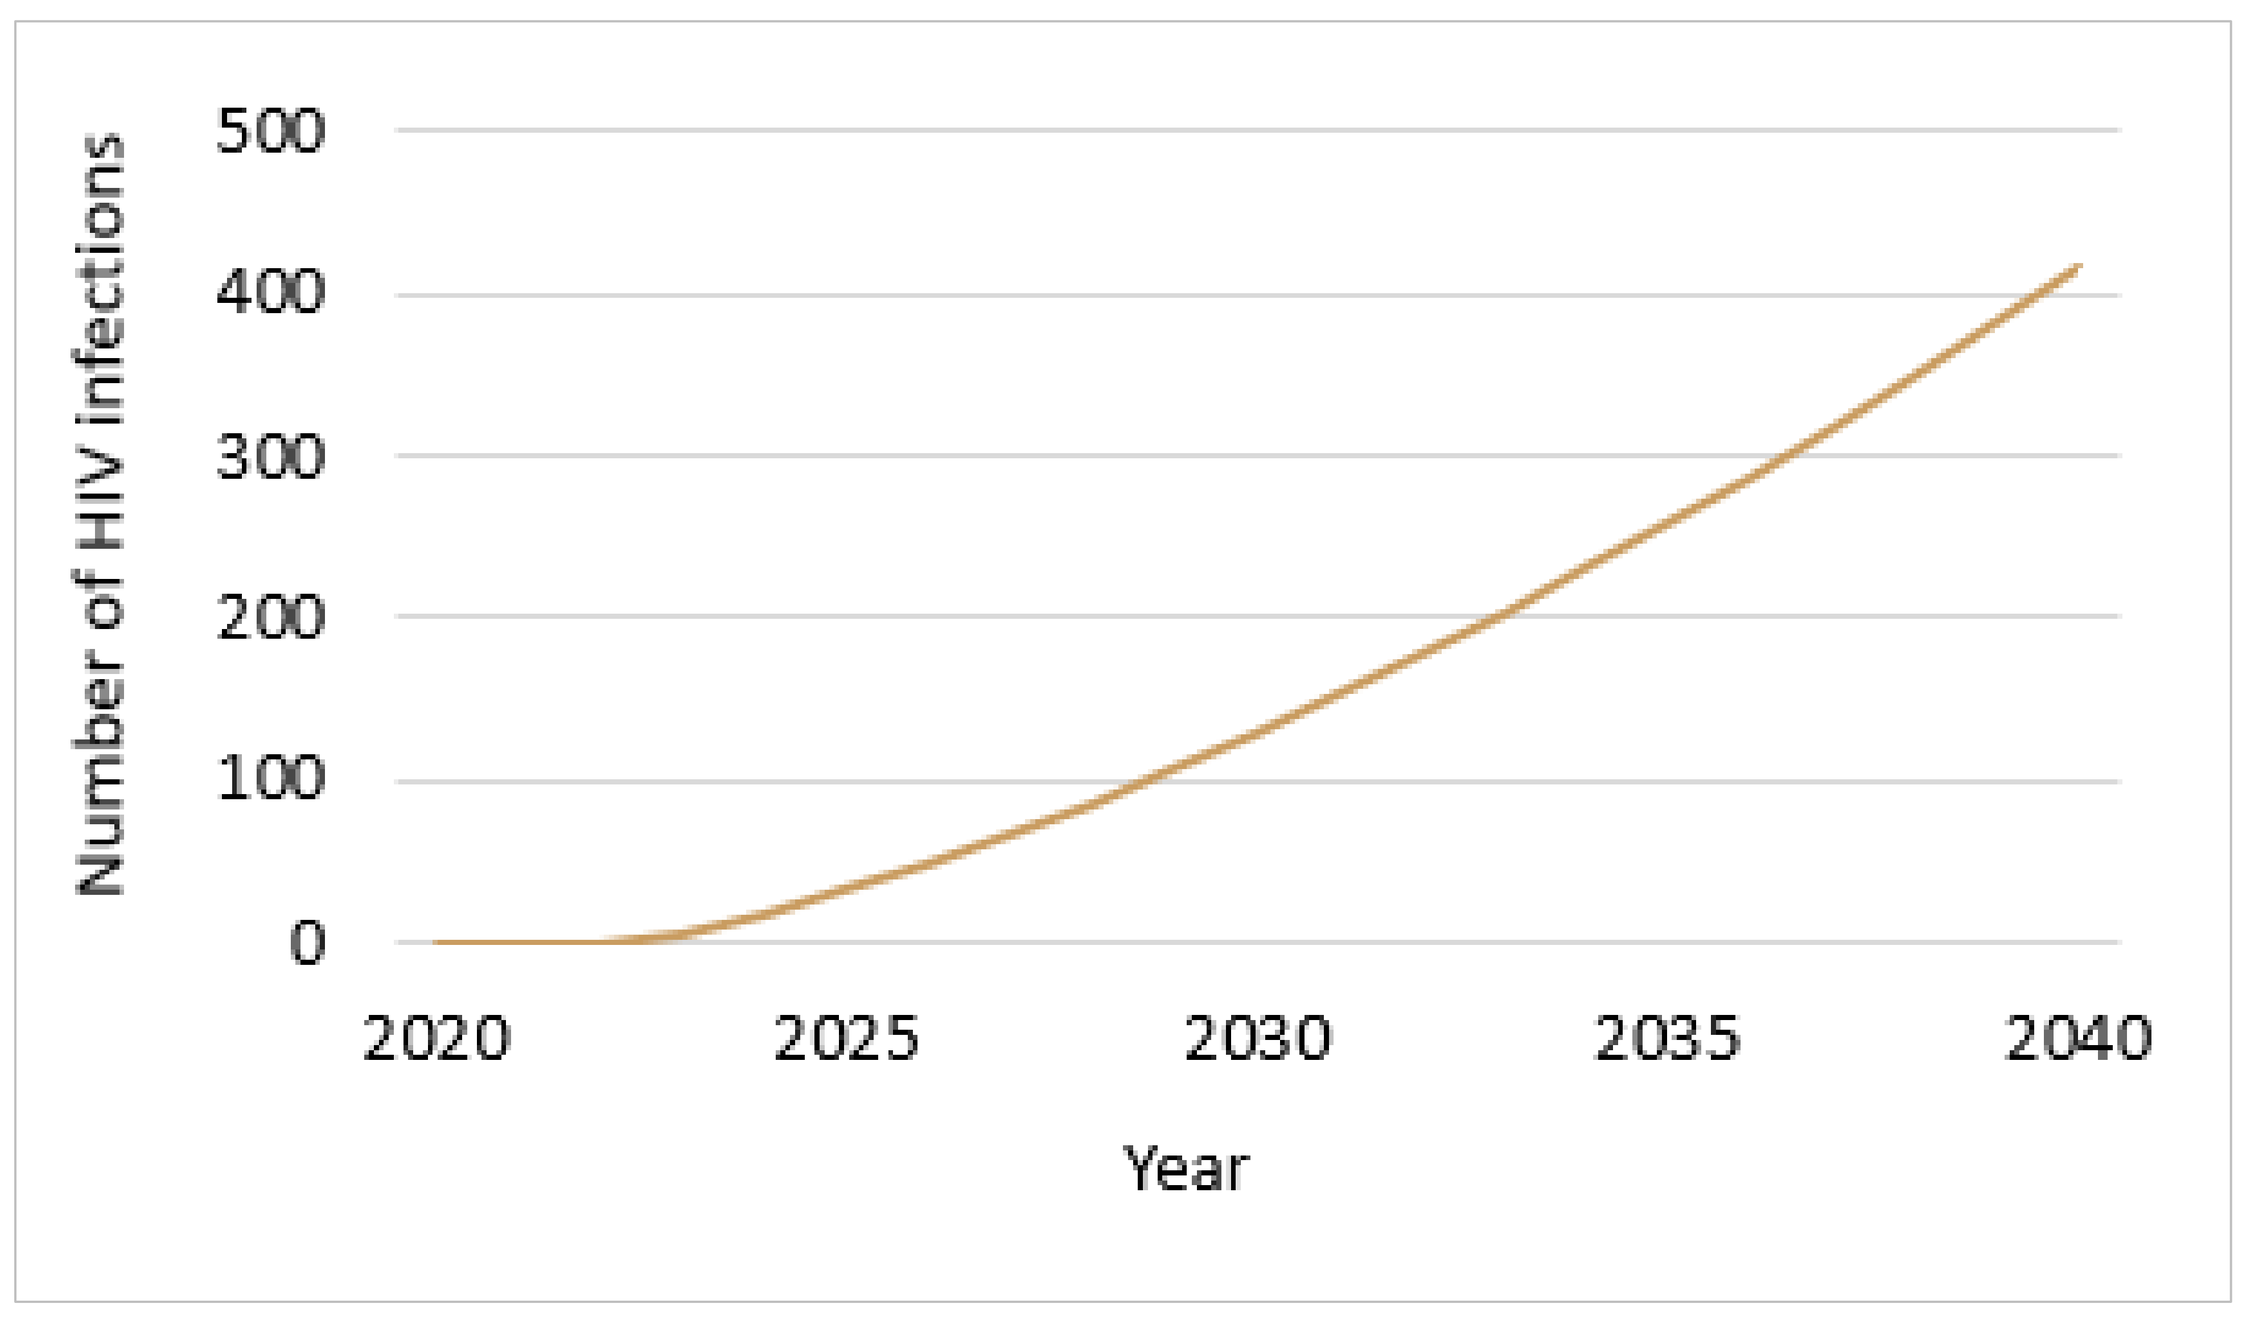
500
400
300
200
100
0
Number of HIV infections
2020
2025
2030
2035
2040
Year

- Nearly half of new HIV infections avoided (42%) were from the MSM, while 33% were from heterosexuals and 25% from PWID (**Figure 4**).
- Of note, all provinces show similar distributions among each subgroup except for the provinces of Alberta, Saskatchewan, and Manitoba. In Alberta and Manitoba, approximately half of the new HIV infections come from the Het subgroup. In contrast, mostly all new HIV infections (82%) in Saskatchewan come from the PWID subgroup, therefore showing social disparities between provinces.
- Over the 20-year projection period, rapid B/F/TAF initiation is expected to result in savings of $139M to the Canadian healthcare system (**Figure 5**); When considering productivity costs, potential savings increased to $510M.

Figure 4. HIV Incidence Avoided, by Subgroup (2020–2040)

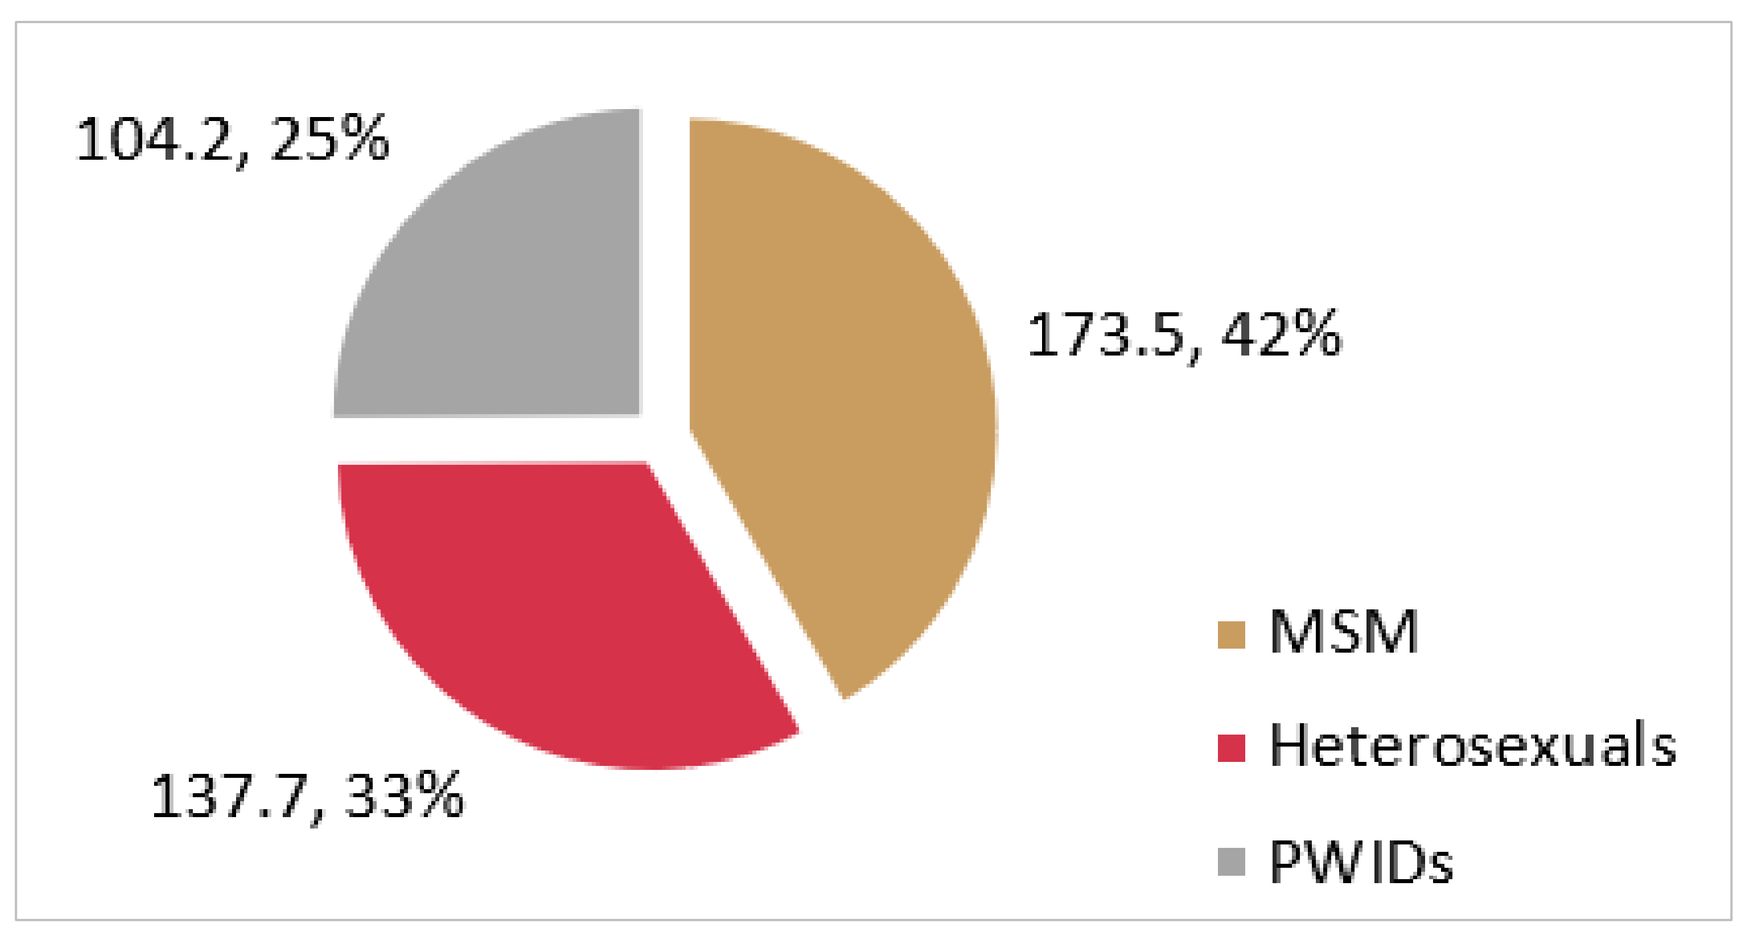
104.2, 25%
173.5, 42%
137.7, 33%
MSM
Heterosexuals
PWIDs

Figure 5. Estimated Savings to the Healthcare Provider from Avoided HIV Infections over 20 years

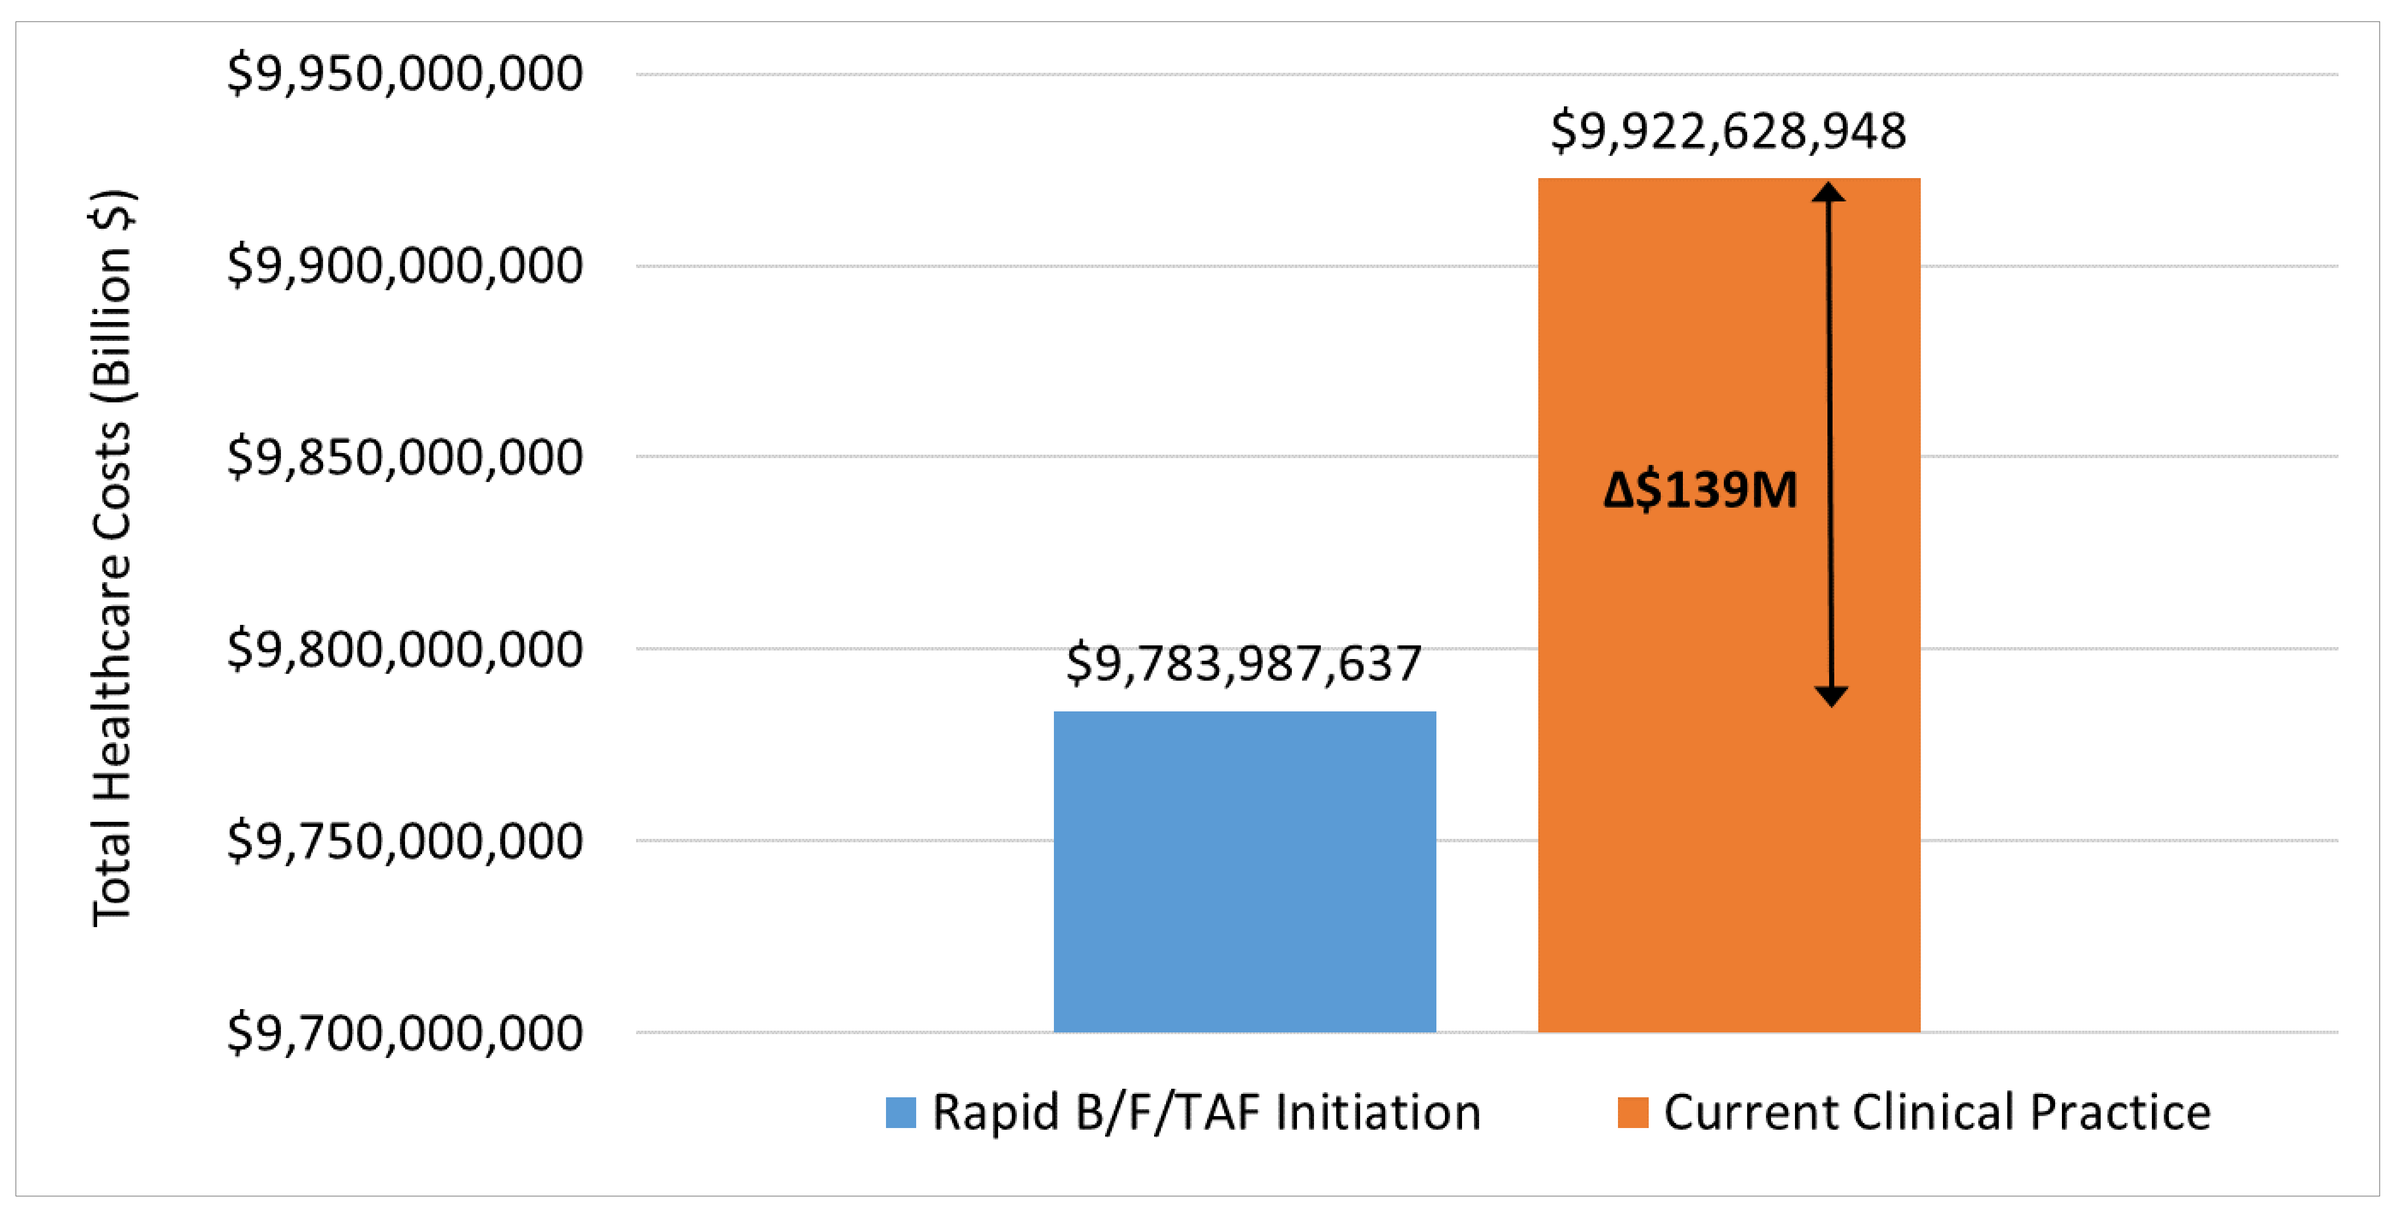
$9,950,000,000
$9,900,000,000
$9,850,000,000
$9,800,000,000
$9,750,000,000
$9,700,000,000
Total Healthcare Costs (Billion $)
$9,922,628,948
$9,783,987,637
Δ$139M
Rapid B/F/TAF Initiation
Current Clinical Practice

- The OWSAs demonstrate that rapid BIKTARVY® initiation may be associated with avoided HIV infections ranging from 325 to 693 and estimated lifetime cost savings ranging from $103 million to $510 million dollars over the 20-year projection period.
- Varying the time to ART initiation by ±7 days in current clinical practice results in savings ranging from $115M to $162M over the 20-year projection period.

CONCLUSIONS

- This study suggests that rapid B/F/TAF initiation represents an advantageous therapeutic strategy to reduce HIV incidence and provide substantial costs savings for the Canadian healthcare system.
- As such, efforts should be made country-wide to reduce the delay in ART initiation following HIV diagnosis.

REFERENCES

1. Estimates of HIV incidence, prevalence and Canada's progress on meeting the 90-90-90 HIV targets, 2020 (2022).
2. World Health Organisation. Guidelines for managing HIV disease and rapid initiation of antiretroviral therapy. 2017
3. Gilead Sciences Canada Inc. Product Monograph: BIKTARVY (Bictegravir/emtricitabine/tenofovir alafenamide tablets). 2021.
4. Cohen MS, Chen YQ, McCauley M, et al. Antiretroviral Therapy for the Prevention of HIV-1 Transmission. *New England Journal of Medicine*. 2016;375(9):830-839. doi:10.1056/NEJMoa1600693
5. Rodger AJ, Cambiano V, Bruun T, et al. Sexual Activity Without Condoms and Risk of HIV Transmission in Serodifferent Couples When the HIV-Positive Partner Is Using Suppressive Antiretroviral Therapy. *JAMA*. 2016;316(2):171-181. doi:10.1001/jama.2016.5148
6. Tattersall T, Tam C, Moore D, et al. Healthcare and treatment experiences among people diagnosed with HIV before and after a province-wide treatment as prevention initiative in British Columbia, Canada. *BMC Public Health*. May 21 2022;22(1):1022. doi:10.1186/s12889-022-13415-2
7. Canadian AIDS Society. The Economic Cost of HIV/AIDS in Canada. 2011

DISCLOSURES

- Jean Lachaine is a partner at PeriPharm, a company that has served as a consultant to Gilead and has received funding from Gilead.
- Jean Lachaine, Kimberly Guinan, and Karine Mathurin, from PeriPharm, have participated in the study conduct, data interpretation and the approval of the abstract.
- The authors would like to thank Mathieu Pelletier for his support in the model development and for the writing of the abstract. Mathieu Pelletier is an employee of PeriPharm and has received funding from PeriPharm for supporting the economic modelling and abstract development.
- No author has received funding for developing the abstract. Gilead participated in the design and provided financial support for the study. Gilead reviewed and approved this publication.