A Retrospective Analysis of Real-World Treatment Patterns in Patients Over Age 64 with Dry Eye Disease Receiving OTX-101 Ophthalmic Solution 0.09%, Cyclosporine Ophthalmic Emulsion 0.05%, or Lifitegrast Ophthalmic Solution 5%

Paul Karpecki,[1] Victoria Barghout,[2] Brad Schenkel,[3] Lynn Huynh,[4] Anamika Khanal,[4] Brittany Mitchell,[3] Mihran Yenikomshian,[4] Enrico Zanardo,[5] Cynthia Matossian[6]

[1]Kentucky Eye Institute, University of Pikeville Kentucky College of Optometry, Lexington, KY, USA; [2]VEB HealthCare, Morristown, NJ, USA; [3]Sun Pharmaceutical Industries, Inc., Princeton, NJ, USA; [4]Analysis Group, Inc., Boston, MA, USA; [5]Analysis Group, Inc., Denver, CO, USA; [6]CM Associates, LLC, New Hope, PA, USA

BACKGROUND

- Dry eye disease (DED) is a multifactorial ocular surface disorder characterized by loss of tear film homeostasis[1]
- In DED, quantitative and/or qualitative tear deficiency leads to tear hyperosmolarity and tear film instability, which induce a self-perpetuating cycle of ocular surface inflammation and damage[2]
- The risk of developing DED increases along with age[3]; therefore, it is important to understand treatment patterns, including persistence, in older patients with DED
- Cequa® (cyclosporine ophthalmic solution 0.09%), Restasis® (cyclosporine ophthalmic emulsion 0.05%), and Xiidra® (lifitegrast ophthalmic solution 5%) are 3 mainstay anti-inflammatory agents indicated for the long-term treatment of DED[4-6]
- Cequa—a novel, nanomicellar cyclosporine A (CsA) solution (**Figure 1**)—was designed to deliver high CsA levels to ocular tissues while minimizing ocular adverse reactions such as burning and redness in order to improve patient tolerability[7]

Figure 1. Nanomicellar formulation of Cequa

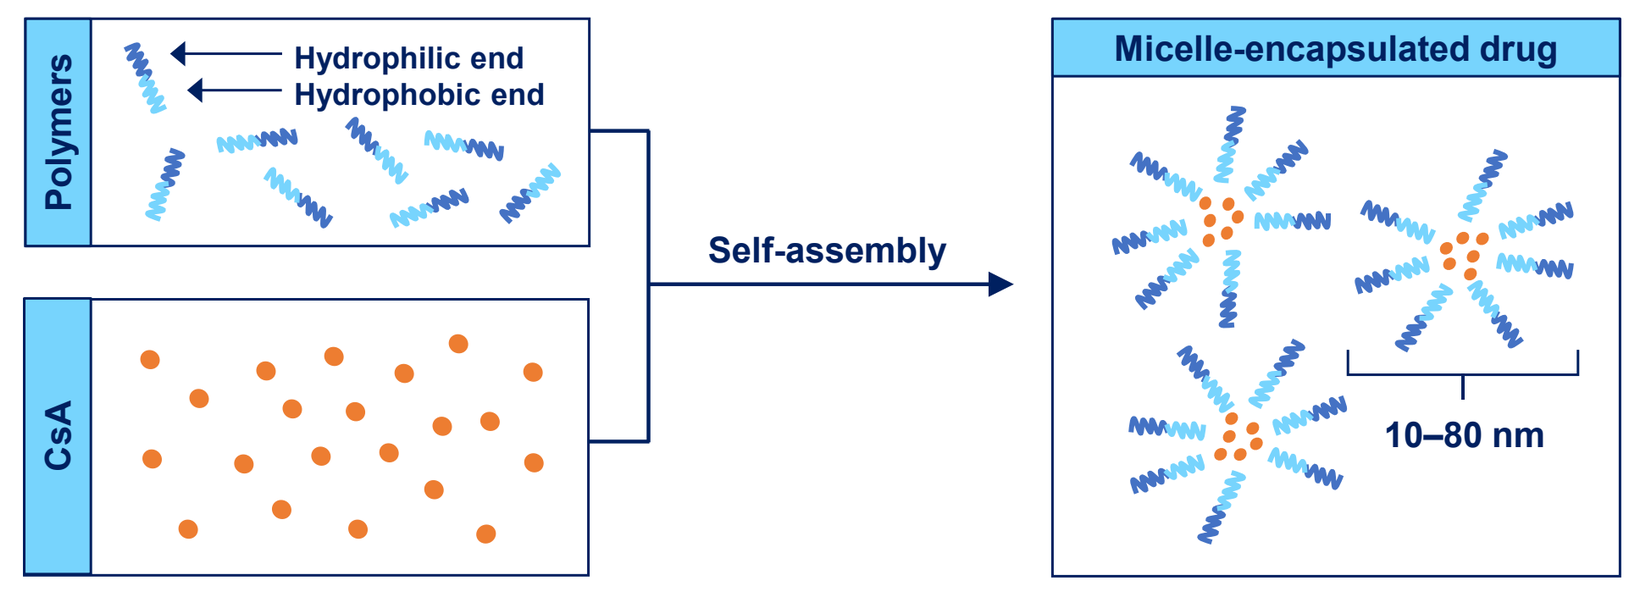
Polymers
Hydrophilic end
Hydrophobic end
CsA
Self-assembly
Micelle-encapsulated drug
10–80 nm

CsA, cyclosporine A.

- While Cequa, Restasis, and Xiidra have demonstrated efficacy and safety in the treatment of DED in multiple clinical trials, limited comparative data exist for these 3 treatments[8]

OBJECTIVE

- To compare treatment patterns in real-world patients with DED, including among patients >64 years of age, who received Cequa, Restasis, or Xiidra

METHODS

Study design

- This was a real-world, retrospective, longitudinal cohort study (**Figure 2**)
- This study utilized data from the Symphony Health Integrated Dataverse (IDV; Symphony Health, Blue Bell, PA) from 07/2019 to 06/2021
 - — The IDV is a nationally representative, provider-based claims database that includes claims submitted to all payer types (eg, commercial plans, Medicare, Medicaid, employer, etc.) and covers approximately 75% of the US population annually

Figure 2. Study design

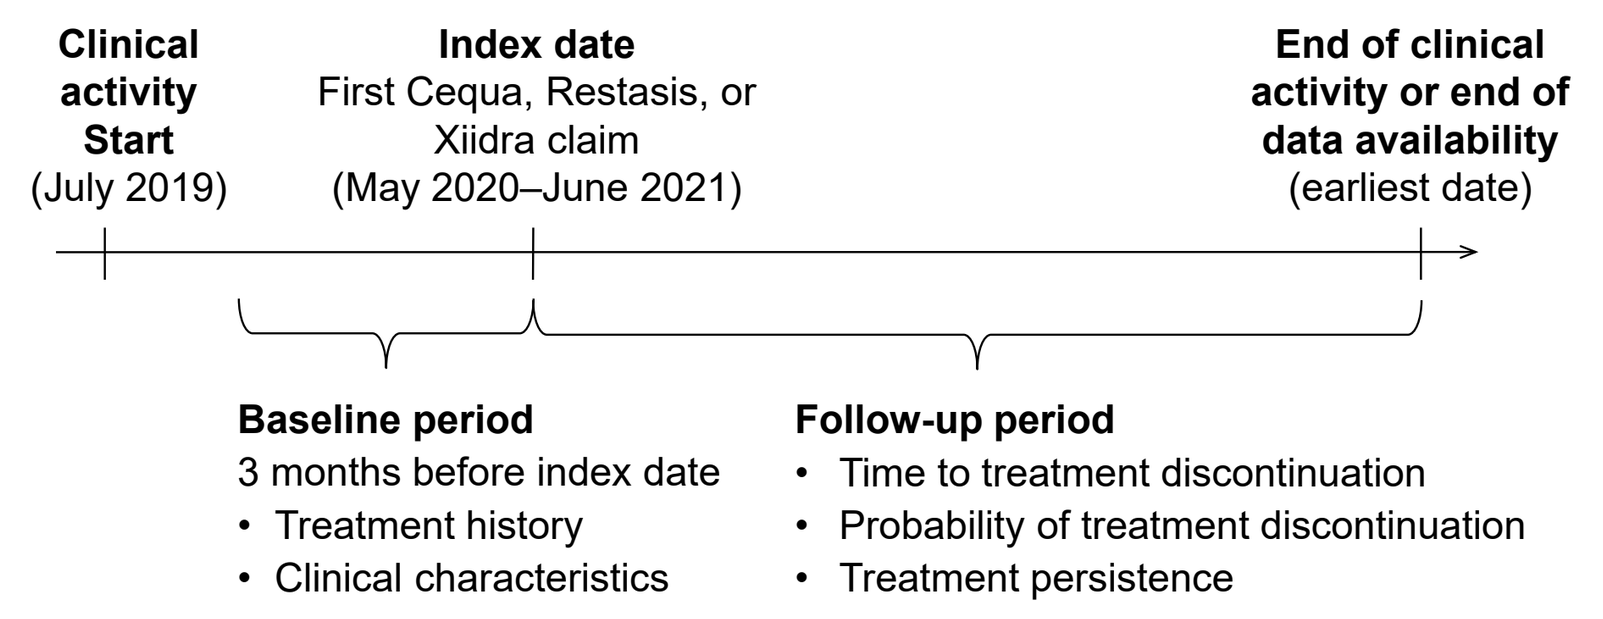
Clinical activity Start
(July 2019)
Index date
First Cequa, Restasis, or Xiidra claim
(May 2020–June 2021)
End of clinical activity or end of data availability
(earliest date)
Baseline period
3 months before index date
• Treatment history
• Clinical characteristics
Follow-up period
• Time to treatment discontinuation
• Probability of treatment discontinuation
• Treatment persistence

Study population

- Key eligibility criteria
 - — Patients are at least 18 years of age at the index date
 - — Patients have ≥1 diagnosis of DED from 07/2019 to 06/2021
 - — The first claim for Cequa, Restasis, or Xiidra is between 05/2020 and 06/2021
 - — There are ≥1 additional claims for index treatment within 4 months after the index date
 - — There are ≥2 claims before or on the index date
 - — There is evidence of clinical activity in the baseline period and within 1 year after the index date (or until end of follow-up, if end of follow-up is <1 year after the index date)
- The dataset included all patients with a Cequa claim, and randomly sampled patients with Restasis or Xiidra claims selected 2:1 relative to Cequa patients
- Patients were sorted into 3 cohorts based on treatment received at the index date: Cequa (first Cequa claim 05/2020 to 06/2021), Restasis (not on Cequa with a Restasis claim before a Xiidra claim), and Xiidra (not on Cequa with a Xiidra claim before a Restasis claim)

Assessments

- Endpoints included time to treatment discontinuation, probability of treatment discontinuation, and treatment persistence for Cequa, Restasis, and Xiidra
 - — Treatment discontinuation was defined as a period of >120 days between prescription claims, or between the last prescription claim and the end of continuous clinical activity or end of data availability
 - — Time to treatment discontinuation was defined as the time from treatment initiation to the onset of discontinuation
 - — Treatment persistence was defined as the percentage of patients on each treatment at various time intervals after the index date
- Analyses first compared Cequa vs Restasis, and then Cequa vs Xiidra

Statistical analysis

- Patient demographics, disease, and clinical characteristics and treatment pattern variables were summarized as means, standard deviations, interquartile range, and medians for continuous variables, and as frequencies and percentages for categorical variables
- Kaplan-Meier (KM) analysis and a log rank test were used to examine time to treatment discontinuation
 - — KM curves were prepared for subgroups of interest, including patients >64 years (the median age of the study population at index date) and ≤64 years of age, and medians and corresponding 95% confidence intervals (CIs) were calculated
- A logistic model assessed the association between index treatment and treatment discontinuation
 - — Both unadjusted and adjusted odds ratios, 95% CIs, and *P*-values were reported
 - — Statistically significant associations were determined based on *P*-values <0.05

RESULTS

Patient demographics

- A total of 7,102 patients met eligibility criteria: 1,846 in the Cequa cohort, 2,248 in the Restasis cohort, and 3,008 in the Xiidra cohort; of all patients, 3,344 were >64 years of age (**Table 1**)

Table 1. Patient demographics and baseline characteristics

	Cequa n = 1,846	Restasis n = 2,248	Xiidra n = 3,008
Age at index date, years			
Mean ± SD	60.6 ± 13.0	64.6 ± 11.9	62.4 ± 11.9
Median (IQR)	62.0 (53.0, 71.0)	66.0 (58.0, 74.0)	63.0 (55.0, 72.0)
>64, n (%)	761 (41.2)	1,268 (56.4)	1,315 (43.7)
Sex, n (%)			
Female	1,539 (83.4)	1,935 (86.1)	2,555 (84.9)
Male	307 (16.6)	313 (13.9)	453 (15.1)
CCI, mean ± SD	0.7 ± 1.4	1.2 ± 1.8	1.0 ± 1.7
Type of insurance plan, n (%)			
Medicare	621 (33.6)	1,244 (55.3)	1,173 (39.0)
Commercial	603 (32.7)	589 (26.2)	766 (25.5)
Medicaid	232 (12.6)	268 (11.9)	316 (10.5)
Employer	155 (8.4)	196 (8.7)	394 (13.1)
Other	256 (13.9)	338 (15.0)	520 (17.3)
Unknown	183 (9.9)	203 (9.0)	413 (13.7)
Follow-up period, months			
Mean ± SD	6.8 ± 3.3	7.6 ± 3.5	6.8 ± 3.4
Median (IQR)	6.9 (4.2, 9.3)	7.8 (4.7, 10.5)	6.4 (3.9, 9.6)

CCI, Charlson Comorbidity Index; IQR, interquartile range; SD, standard deviation.

Time to treatment discontinuation

- In patients >64 years of age, the time to treatment discontinuation for those receiving Cequa was significantly longer than those receiving Restasis (*P* = 0.002; **Figure 3A**)
 - — Median time to discontinuation was 275 days for Cequa vs 208 days for Restasis
- The median >64 year old patient receiving Cequa stayed on treatment a numerically longer time (275 days) than the median >64 year old patient receiving Xiidra (269 days), but the difference in KM time to discontinuation was not statistically significant (*P* = 0.624; **Figure 3B**)

Figure 3A and 3B. Kaplan-Meier survival curve for time to treatment discontinuation in patients >64 years of age for A) Cequa vs Restasis and B) Cequa vs Xiidra

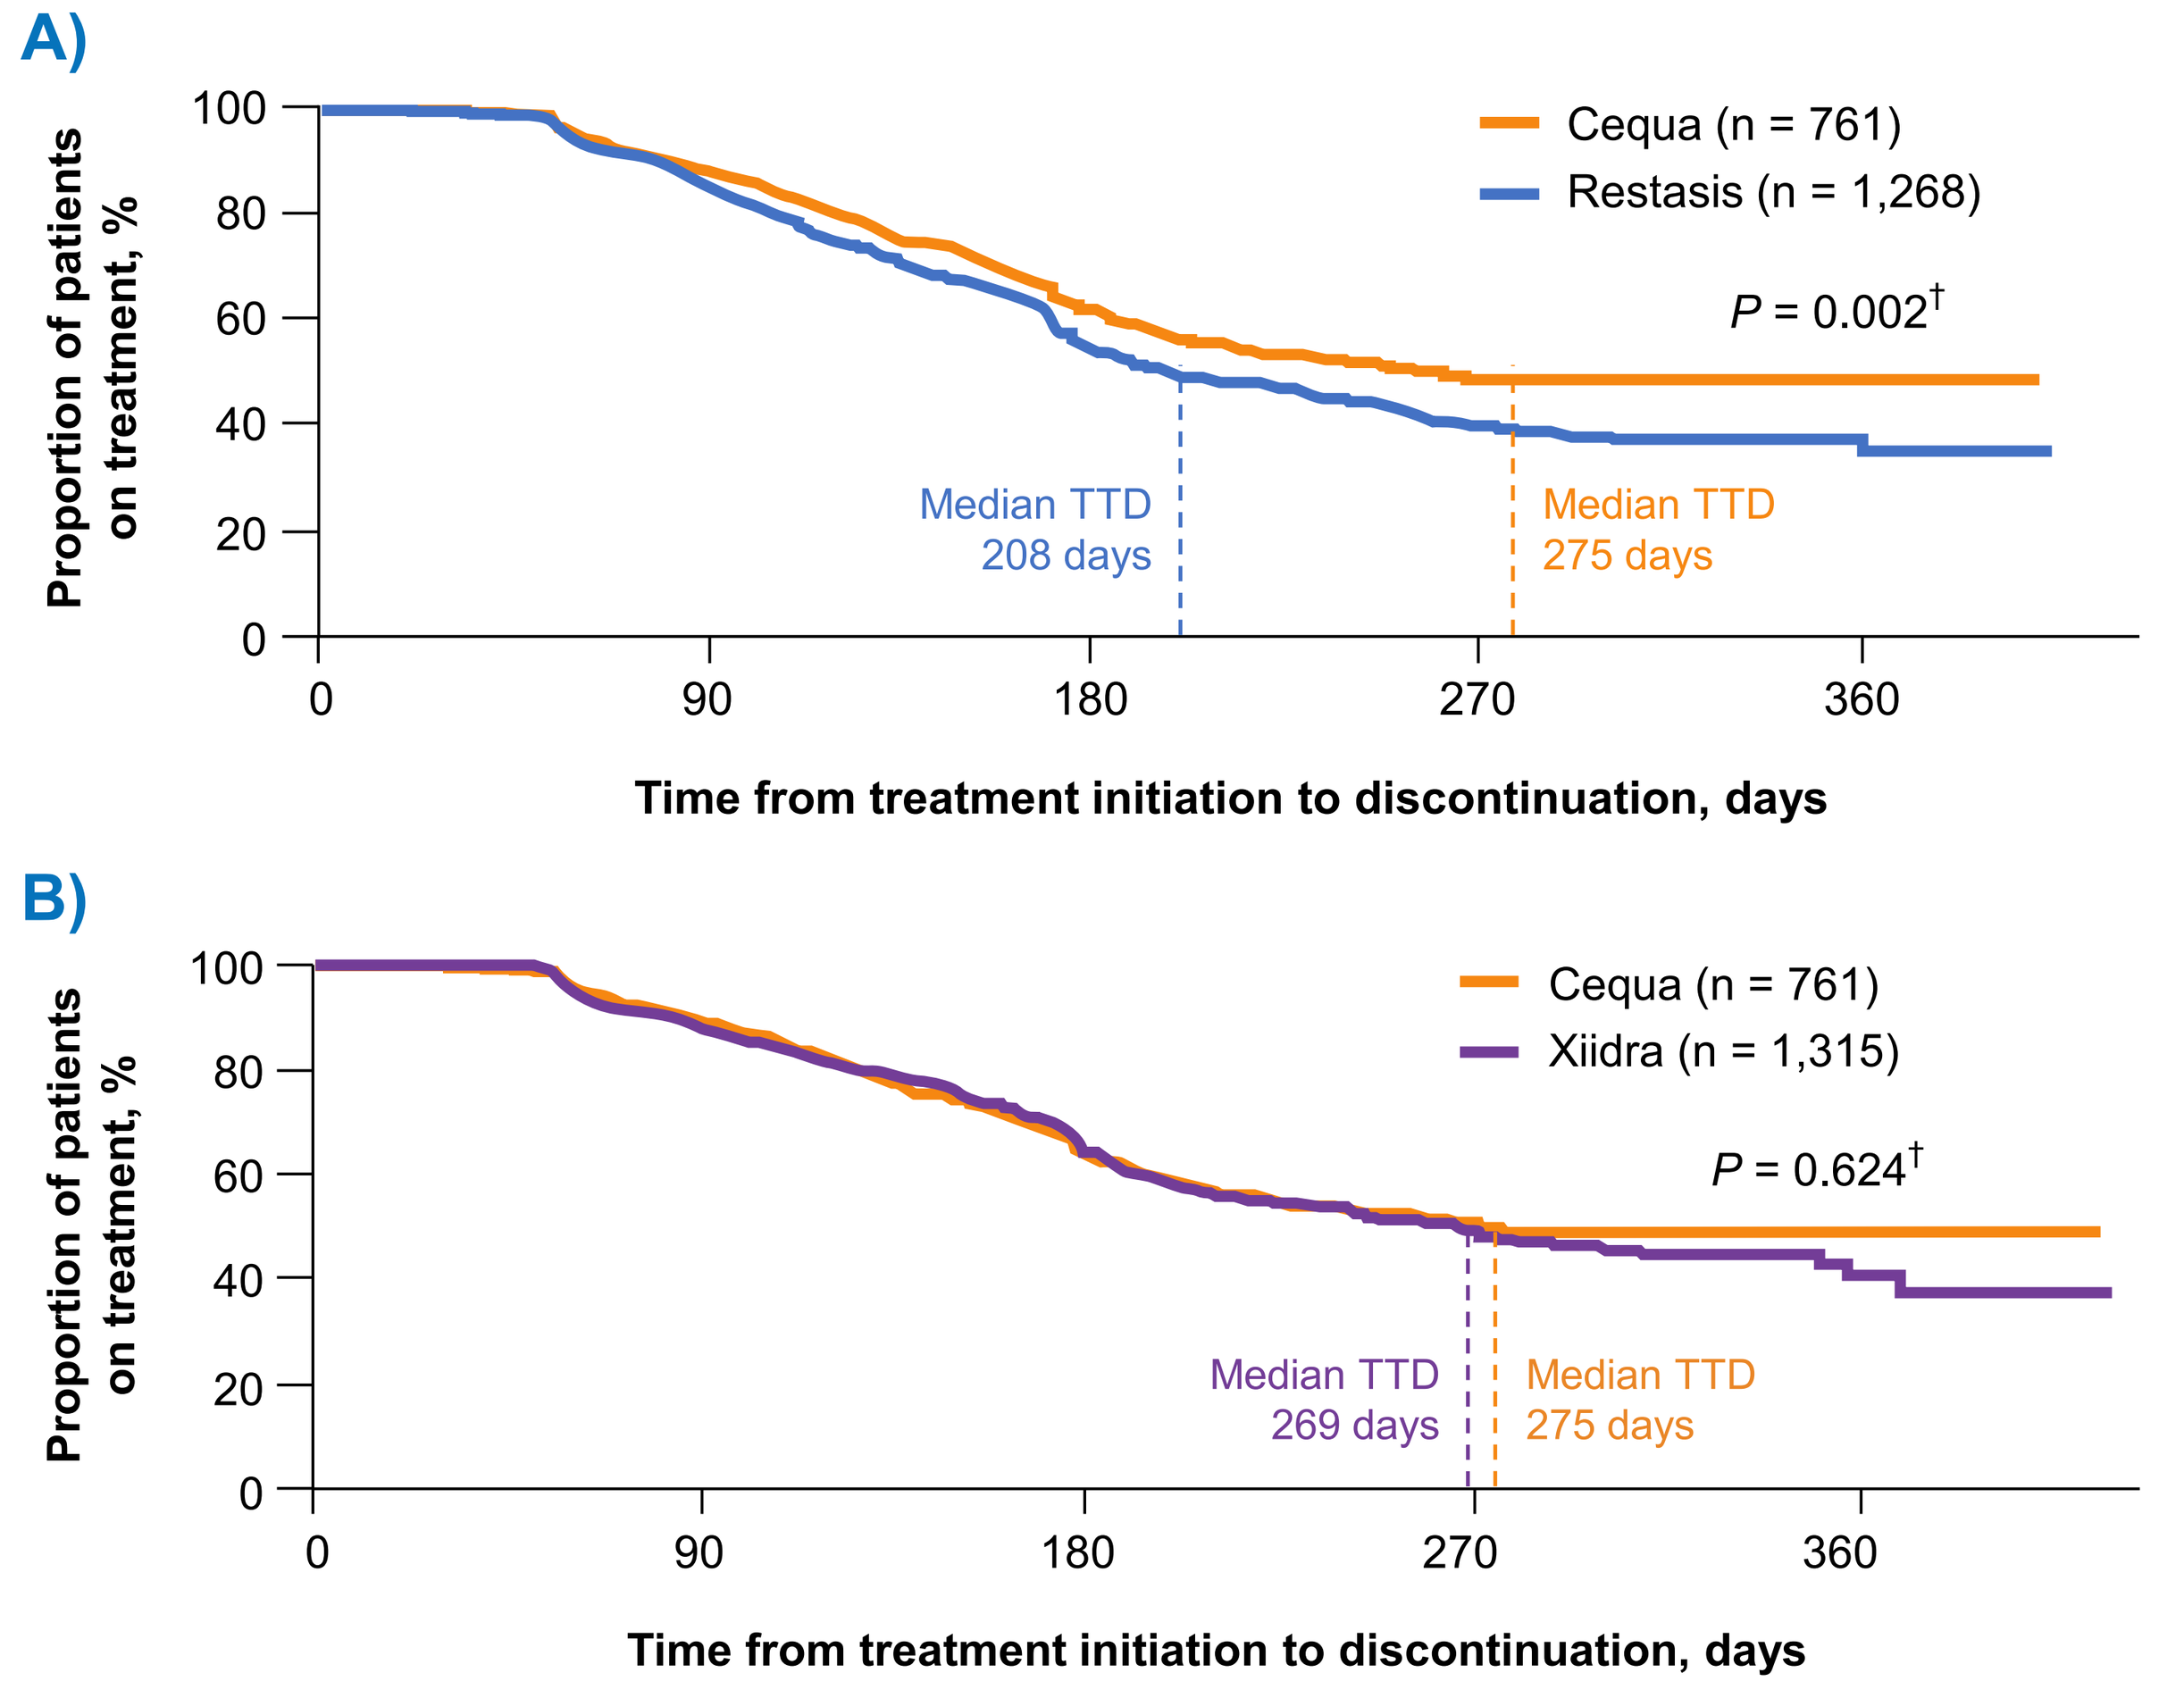
A)
Proportion of patients on treatment, %
Cequa (n = 761)
Restasis (n = 1,268)
P = 0.002†
Median TTD 208 days
Median TTD 275 days
Time from treatment initiation to discontinuation, days
B)
Proportion of patients on treatment, %
Cequa (n = 761)
Xiidra (n = 1,315)
P = 0.624†
Median TTD 269 days
Median TTD 275 days
Time from treatment initiation to discontinuation, days

†*P*-value refers to the difference between treatments in survival curves based on the log-rank test.
TTD, time to treatment discontinuation.

- Time to treatment discontinuation was significantly longer for patients receiving Cequa vs those receiving Restasis (*P* = 0.033; **Figure 4A**) and median time to treatment discontinuation was numerically longer for Cequa vs Xiidra, but the KM curves of time to treatment discontinuation were not statistically different (*P* = 0.825; **Figure 4B**)
 - — Median time to treatment discontinuation was 354 days for Cequa vs 241 days for Restasis

Figure 4A and 4B. Kaplan-Meier survival curve for time to treatment discontinuation for A) Cequa vs Restasis and B) Cequa vs Xiidra

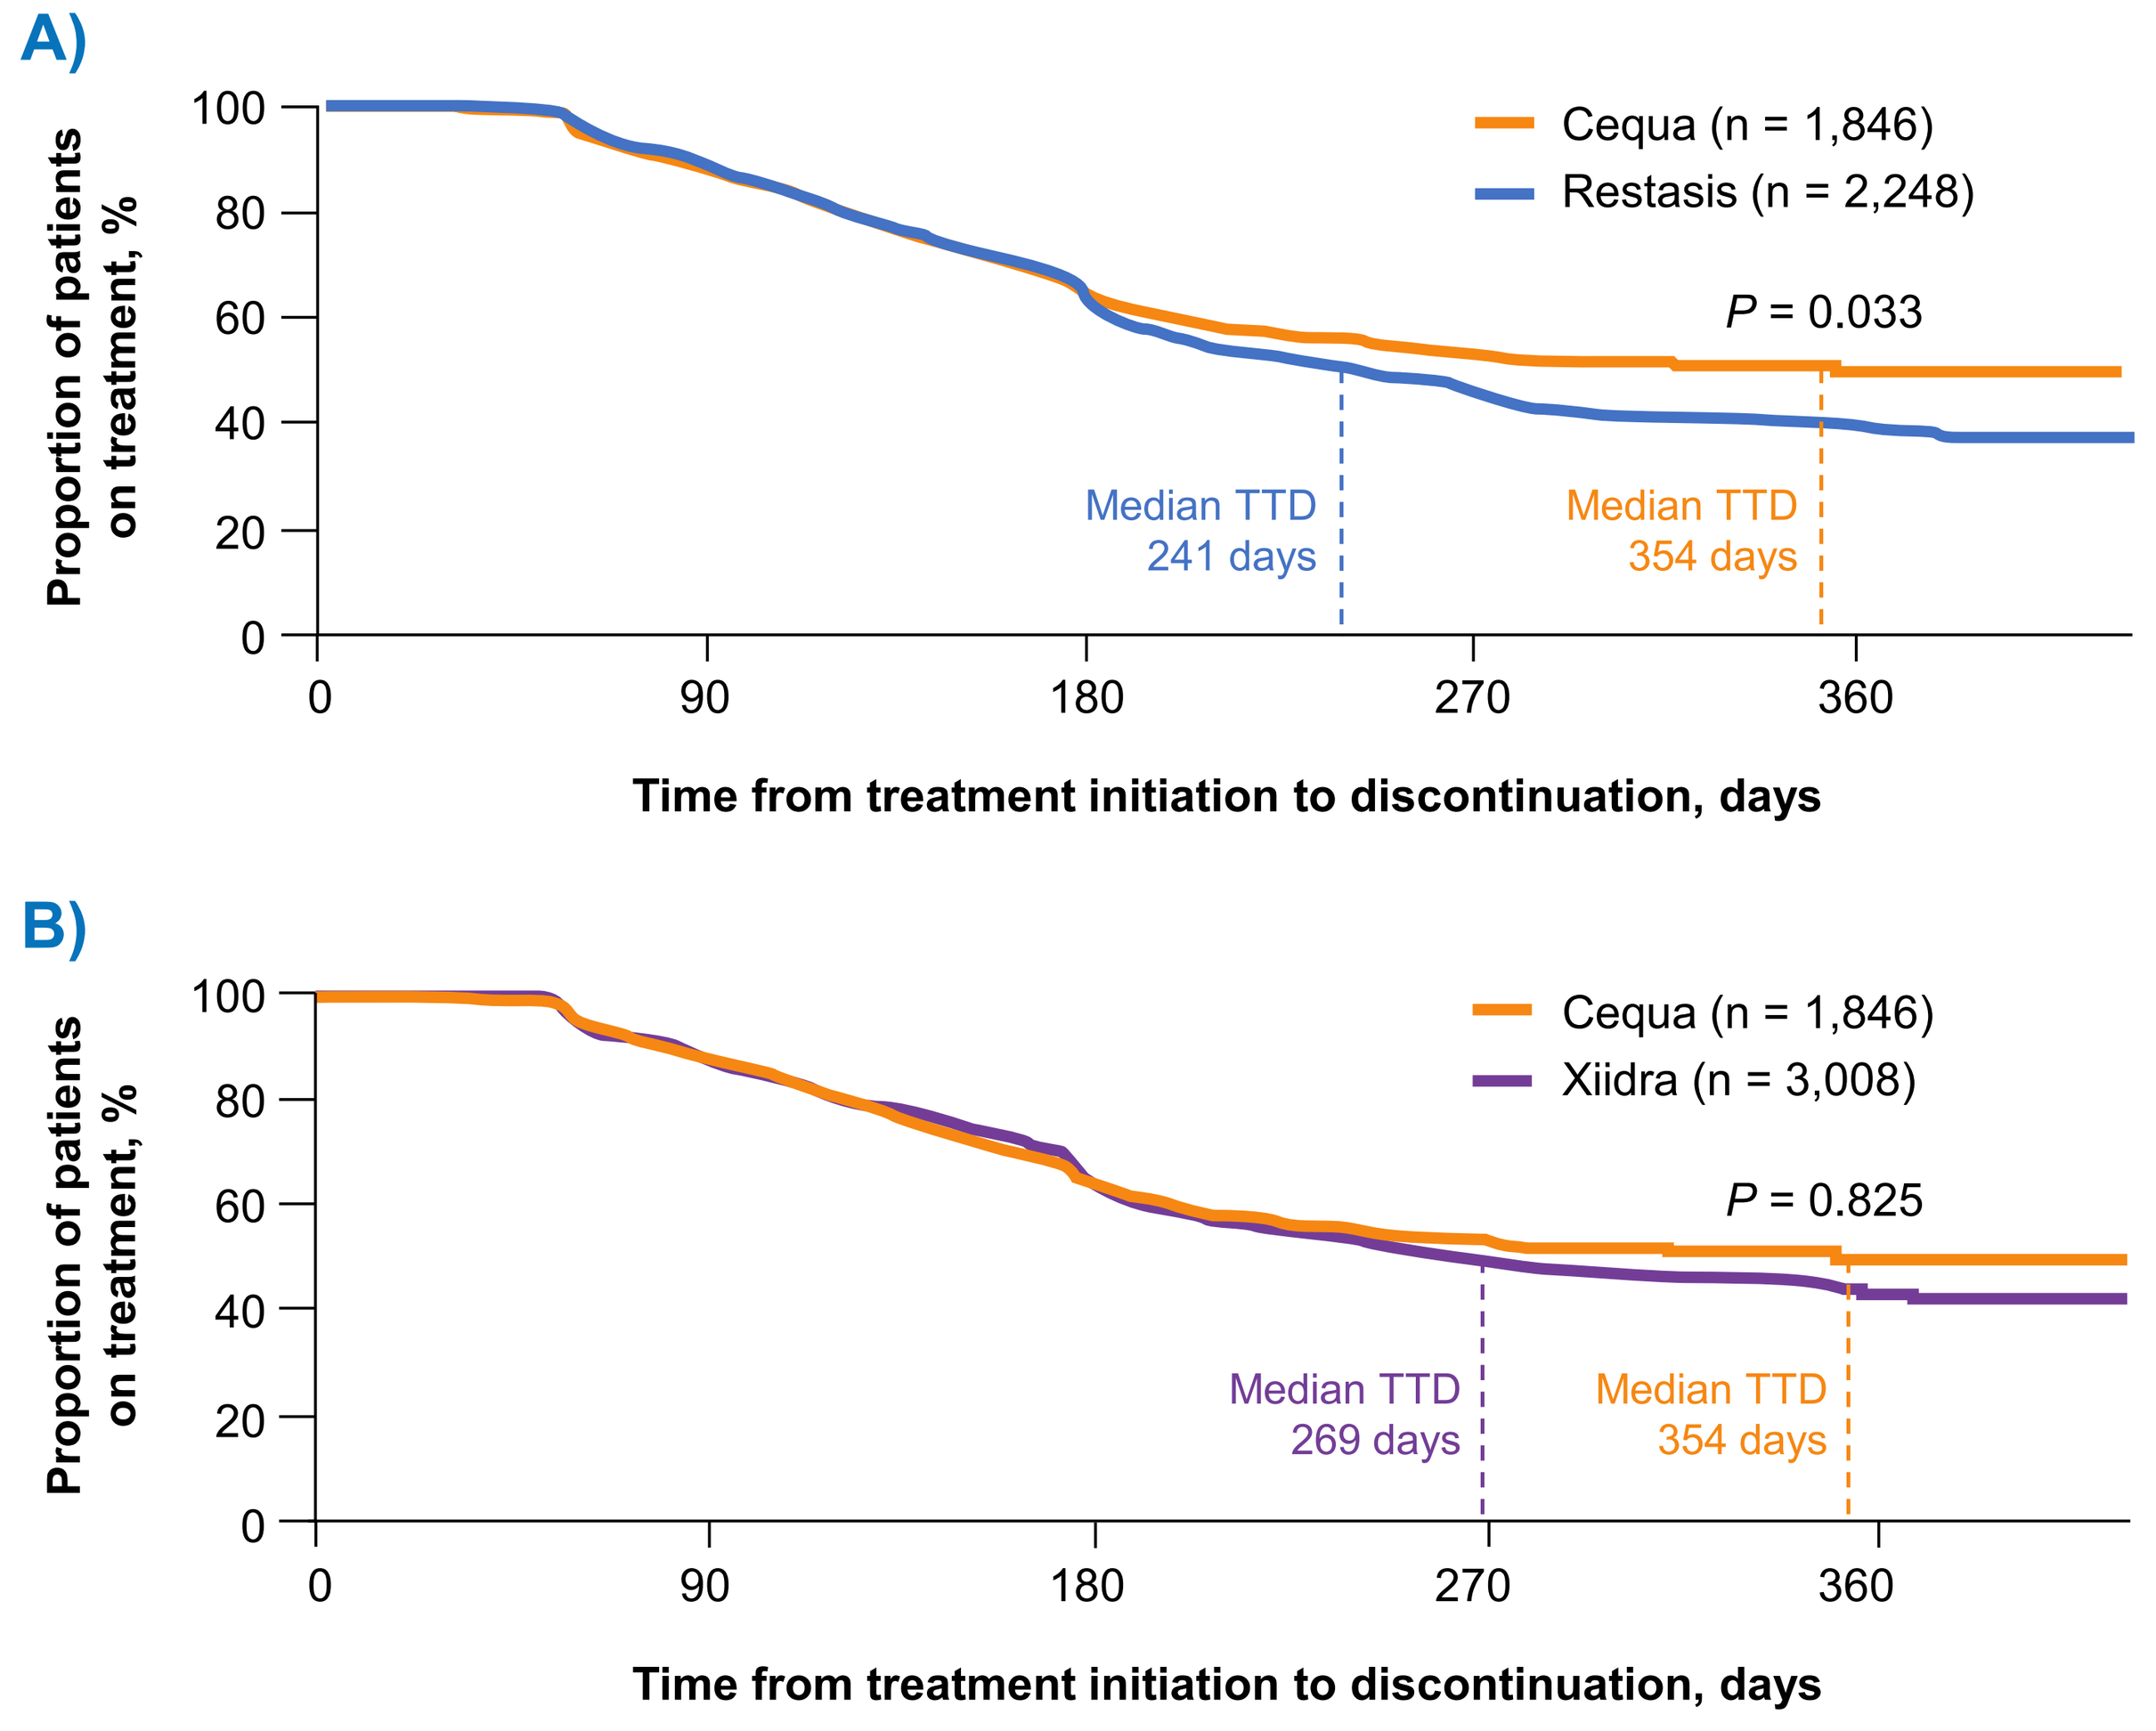
A)
Proportion of patients on treatment, %
Cequa (n = 1,846)
Restasis (n = 2,248)
P = 0.033
Median TTD 241 days
Median TTD 354 days
Time from treatment initiation to discontinuation, days
B)
Proportion of patients on treatment, %
Cequa (n = 1,846)
Xiidra (n = 3,008)
P = 0.825
Median TTD 269 days
Median TTD 354 days
Time from treatment initiation to discontinuation, days

TTD, time to treatment discontinuation.

Probability of treatment discontinuation

- Overall, patients >64 years of age receiving Cequa or Restasis were 27% more likely to discontinue treatment than patients ≤64 years of age, after adjusting for index treatment, Charlson Comorbidity Index (CCI), eye-related comorbidities, and insurance
- Patients on Restasis were 35% more likely to discontinue treatment than patients on Cequa after adjusting for age, CCI, insurance, and eye-related comorbidities (*P* <0.001; **Table 2**)
- For patients receiving Cequa or Xiidra, there was no significant difference in the probability of treatment discontinuation for patients >64 years of age vs ≤64 years of age (*P* = 0.118)

Table 2. Logistic regression model of the probability of treatment discontinuation, Cequa vs Restasis

	Estimated OR	95% CI	*P*-value
Unadjusted model			
Treatment			
Restasis (ref Cequa)	1.41	(1.24, 1.61)	<0.001*
Adjusted model			
Treatment			
Restasis (ref Cequa)	1.35	(1.16, 1.57)	<0.001*
Age, years			
>64 (ref ≤64)	1.27	(1.07, 1.51)	0.007*
CCI	0.98	(0.94, 1.02)	0.343
Eye-related comorbidities			
Yes (ref no)	0.90	(0.76, 1.08)	0.262
Insurance (ref commercial)			
Employer	0.87	(0.66, 1.15)	0.336
Medicaid	0.62	(0.47, 0.82)	0.001*
Medicare	0.94	(0.78, 1.12)	0.478

*Indicates statistical significance as based on *P*-value <0.05.
CCI, Charlson Comorbidity Index; CI, confidence interval; OR, odds ratio; ref, reference.

- Cequa and Xiidra patients had similar odds of treatment discontinuation, both in an unadjusted model and after adjusting for age, CCI, insurance, and eye-related comorbidities (**Table 3**)

Table 3. Logistic regression model of the probability of treatment discontinuation, Cequa vs Xiidra

	Estimated OR	95% CI	*P*-value
Unadjusted model			
Treatment			
Xiidra (ref Cequa)	1.01	(0.89, 1.14)	0.917
Adjusted model			
Treatment			
Xiidra (ref Cequa)	0.97	(0.84, 1.12)	0.718
Age, years			
>64 (ref ≤64)	1.15	(0.97, 1.37)	0.118
CCI	0.99	(0.95, 1.04)	0.788
Eye-related comorbidities			
Yes (ref no)	0.85	(0.72, 1.01)	0.067
Insurance (ref commercial)			
Employer	0.88	(0.69, 1.10)	0.259
Medicaid	0.79	(0.62, 1.01)	0.060
Medicare	0.79	(0.66, 0.96)	0.015*

*Indicates statistical significance as based on *P*-value <0.05.
CCI, Charlson Comorbidity Index; CI, confidence interval; OR, odds ratio; ref, reference.

Treatment persistence

- At 360 days after index date, 49.8% of Cequa patients remained on treatment vs 39.4% of Restasis patients (*P* = 0.036) and 44.0% of Xiidra patients (*P* = 0.854; **Figure 5**)

Figure 5. Proportion of patients remaining on treatment after index date

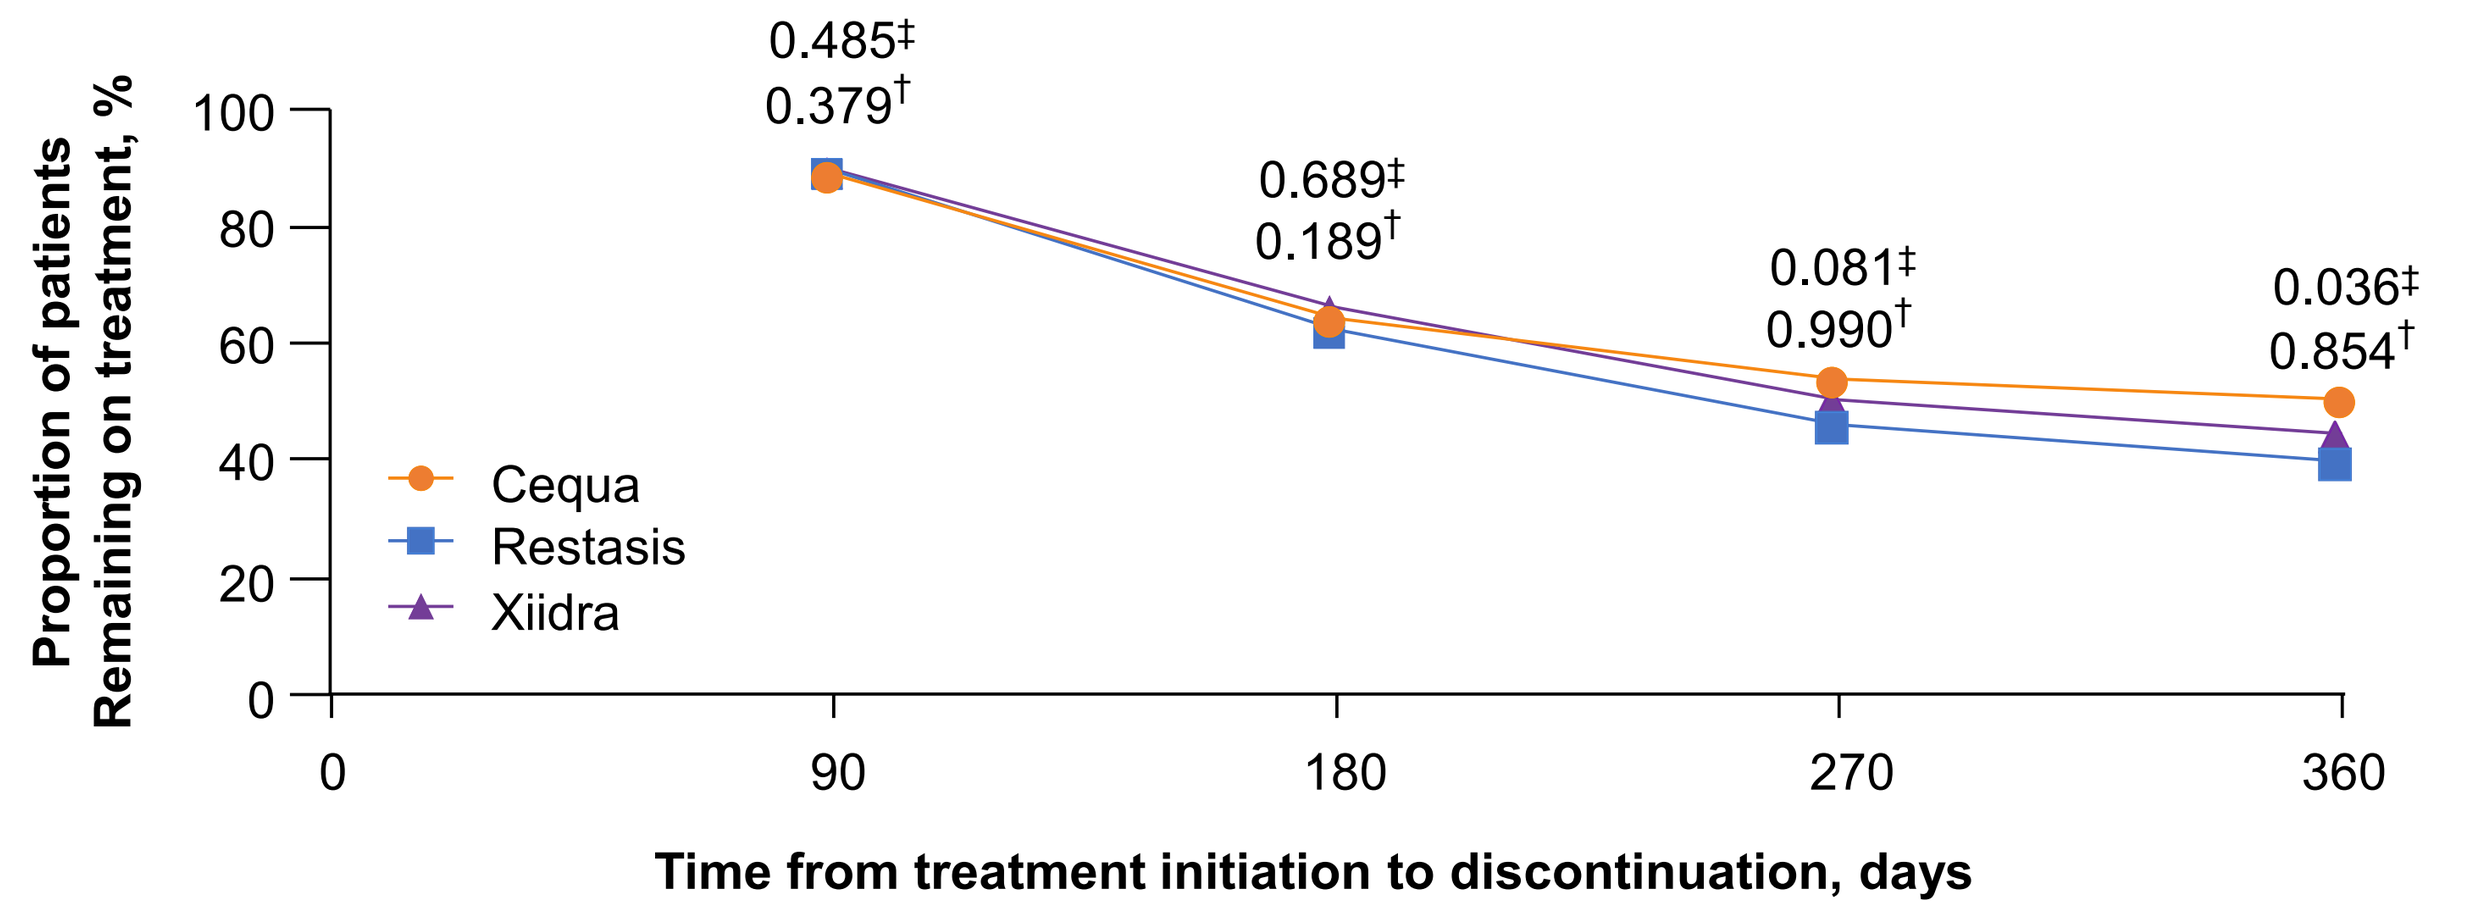
Proportion of patients Remaining on treatment, %
0.485‡
0.379†
0.689‡
0.189†
0.081‡
0.990†
0.036‡
0.854†
Cequa
Restasis
Xiidra
Time from treatment initiation to discontinuation, days

‡*P*-value for analysis of Cequa vs Restasis. Day 360 *P*-value is significant as based on significance level <0.05.
†*P*-value for analysis of Cequa vs Xiidra

CONCLUSIONS

- Patients >64 years of age were significantly more likely to discontinue DED treatment than those ≤64 years. Additionally, patients >64 years receiving Cequa remained on treatment longer than patients >64 years receiving Restasis, and time to treatment discontinuation was significantly different between Cequa and Restasis patients
 - — The time on treatment was similar for Cequa and Xiidra patients >64 years
- Patients taking Cequa remained on treatment longer and were significantly less likely to discontinue treatment than those taking Restasis; differences in time to treatment discontinuation between Cequa and Restasis patients were significant
- Patients on Cequa showed numerically greater time on treatment and persistence than patients on Xiidra, though these results were not statistically significant
- These findings are important in that they highlight real-world treatment pattern differences for patients with DED who are receiving Cequa, Restasis, or Xiidra

REFERENCES

1) Craig JP, et al. *Ocul Surf.* 2017;15(3):276–283. **2)** Bron AJ, et al. *Ocul Surf.* 2017;15(3):438–510. **3)** Stapleton F, et al. *Ocul Surf.* 2017;15(3):334–365. **4)** Sun Pharmaceutical Industries, Inc. CEQUA® (cyclosporine ophthalmic solution) 0.09% Full Prescribing Information. 2022. **5)** Allergan, Inc. Restasis® (cyclosporine ophthalmic emulsion) 0.05% Full Prescribing Information. 2017. **6)** Novartis Pharmaceuticals Corporation. Xiidra® (lifitegrast ophthalmic solution) Full Prescribing Information. 2020. **7)** Cholkar K, et al. *Transl Vis Sci Technol.* 2015;4(3):1. **8)** Shen Lee B, et al. *Ophthalmol Ther.* 2022;11(4):1333-1369.

ACKNOWLEDGMENTS

Medical writing and editorial support were provided by Jennifer Masucci, VMD, of AlphaBioCom, a Red Nucleus company, and were funded by Sun Pharma.

DISCLOSURES

PK reports consultant fees from Alcon Labs; Aldeyra; Allergan/AbbVie; Azura Pharmaceuticals; Bausch + Lomb; BioTissue; Bruno Pharmaceuticals; Cambium Pharmaceuticals; Dompé; Eyedetec; Imprimis; Johnson & Johnson Vision; Kala Pharmaceuticals; Keplr Vision; Kiora; Konan Medical; Mallinckrodt; Neurolens; Novartis; Oasis Medical; Ocuphire; Oculus; OcuMedic; OcuSoft; Oyster Point; RegenerEyes; Science Based Health; Sight Sciences; Silk Technologies; Sun Pharma; Surface Pharmaceuticals; Tarsus Medical; TearClear; Visant Medical; and Vital Tears. **VB** is Managing Director of VEB HealthCare, LLC, which receives funding from Novartis; BMS; Sun Pharma; and Taiho. **BS** and **BM** are employees of Sun Pharma. **LH, AK, MY,** and **EZ** are employees of the Analysis Group, Inc. **CM** has received consultant fees from Aerie Pharmaceuticals; Alcon; Bausch + Lomb; Bruder Healthcare; Checked Up; EyePoint; Johnson & Johnson; Kala Pharmaceuticals; Lacriscience; Lenstec; Lumenis; Novartis; Ocular Science; Ocular Therapeutix; Olympic; Quidel; Sun Pharma; TearLab; TissueTech; and Zeiss; and personal fees from Veterinarian Recommended Solutions.