A New View on Quantification of Disease Modification: Two Case Studies from Parkinson's Disease

Milana Ivkovic[1], Anders Gustavsson[2,3], Teresa León[1], Julie Hviid Hahn-Pedersen[1], Lars Lau Raket[4,5]

[1]Novo Nordisk A/S, [2]Division of Neurogeriatrics, Department of Neurobiology, Care Sciences and Society, Karolinska Institutet, Sweden, [3]Quantify Research, Sweden, [4]Clinical Memory Research Unit, Department of Clinical Sciences, Lund University, Sweden, [5]Formerly, Novo Nordisk A/S

Background

Therapies that slow disease progression are a major unmet need in neurodegenerative diseases, and researchers continue to seek disease-modifying treatments (DMTs), such as in Parkinson's disease (PD). Evidence on the effect of DMTs includes both impact on the underlying pathophysiology of the disease and impact on clinical outcomes.

The clinical treatment effect is typically reported as a difference in outcome measures, between the group of treated patients and the patients in the control group at a certain point in time, typically the end of study.

This representation of the clinical treatment effect has several limitations:

1. It can be difficult to interpret, especially by patients, because the treatment difference is typically a number on a point scale where point values are arbitrarily anchored. These scales are typically only used in a research setting, so prescribers and patients may not be familiar with these constructs at all.
2. It might not capture the underlying mode through which the treatment modifies the disease progression – through delaying or slowing it. That could also, potentially, lead to difficulties when extrapolating the effect beyond the trial time horizon, especially if there is much variation between the treatment effect estimates at different timepoints (when measuring at different visits).
3. It may overestimate uncertainty, if only using data from two time points (start and end), omitting relevant information in between.

Objectives

Here we aim to explore an interpretable quantification of time delay (of worsening of clinical symptoms) of two PD therapies which were investigated for potential of disease modification in early-stage PD, based on published trial results[1,2].

- rasagiline (approved therapy, irreversible MAO-B selective inhibitor)
- prasinezumab (investigational anti-α-Synuclein antibody)

Methods

Published summary data were retrieved from two delayed-start drug trials investigating their potential of disease modification in early-stage PD: ADAGIO (rasagiline, 36+36 weeks) and PASADENA (prasinezumab, 52+52 weeks).

Based on the published trial results, the point estimates of the mean time-delay were approximated by visually estimating the horizontal difference for a fixed point on the Y-axis observed in the early and delayed-start parts of both trials.

- The Unified Parkinson's Disease Rating Scale (UPDRS) total score (ADAGIO) and Movement Disorder Society UPDRS part III score (PASADENA) were used.
- Estimates were presented for the half-way visit (start of the delayed-start parts) and at the end of study.

Infobox: Delayed-start trial design

The delayed-start cohort design was used to evaluate the disease-modifying effect of treatments.

- Patients were randomly assigned to receive active treatment at different times.
- The treatment period was divided into a placebo-controlled period and a delayed-start period.
- During the placebo-controlled period, some patients received active treatment while others received placebo.
- In the delayed-start period, patients who received the active treatment during the placebo-controlled period continued the treatment, while those who received the placebo were switched to the active treatment.
- If the treatment difference observed during the placebo-controlled period is preserved (at least in part) during the delayed-start period, it suggests a consistent and lasting effect.

Patients in the ADAGIO trial who received rasagiline during the placebo-controlled period remained on active treatment (early-start) throughout the study period (36+36 weeks). In contrast, patients receiving placebo during the initial placebo-controlled period were switched to rasagiline after 36 weeks (delayed-start). A similar design was implemented in the PASADENA trial, where the placebo-controlled period lasted for 52 weeks comparing placebo to prasinezumab, followed by the delayed-start period comparing early vs. delayed prasinezumab for an additional 52 weeks.

- ADAGIO showed contradictory results for different doses of rasagiline (1 mg/day vs. 2 mg/day) and PASADENA failed to meet its primary endpoint.
- Despite these mixed results, both trials showed some promise for disease modification.

Results

Graphical estimation of mean time delay showed an alternative representation of treatment effects (figures modified from [1,2]).

Lower dose rasagiline (1 mg/day) results suggested a persistent time delay of approx. 24 weeks comparing end of early treatment and delayed-start cohorts, consistent with a potential disease modifying effect.

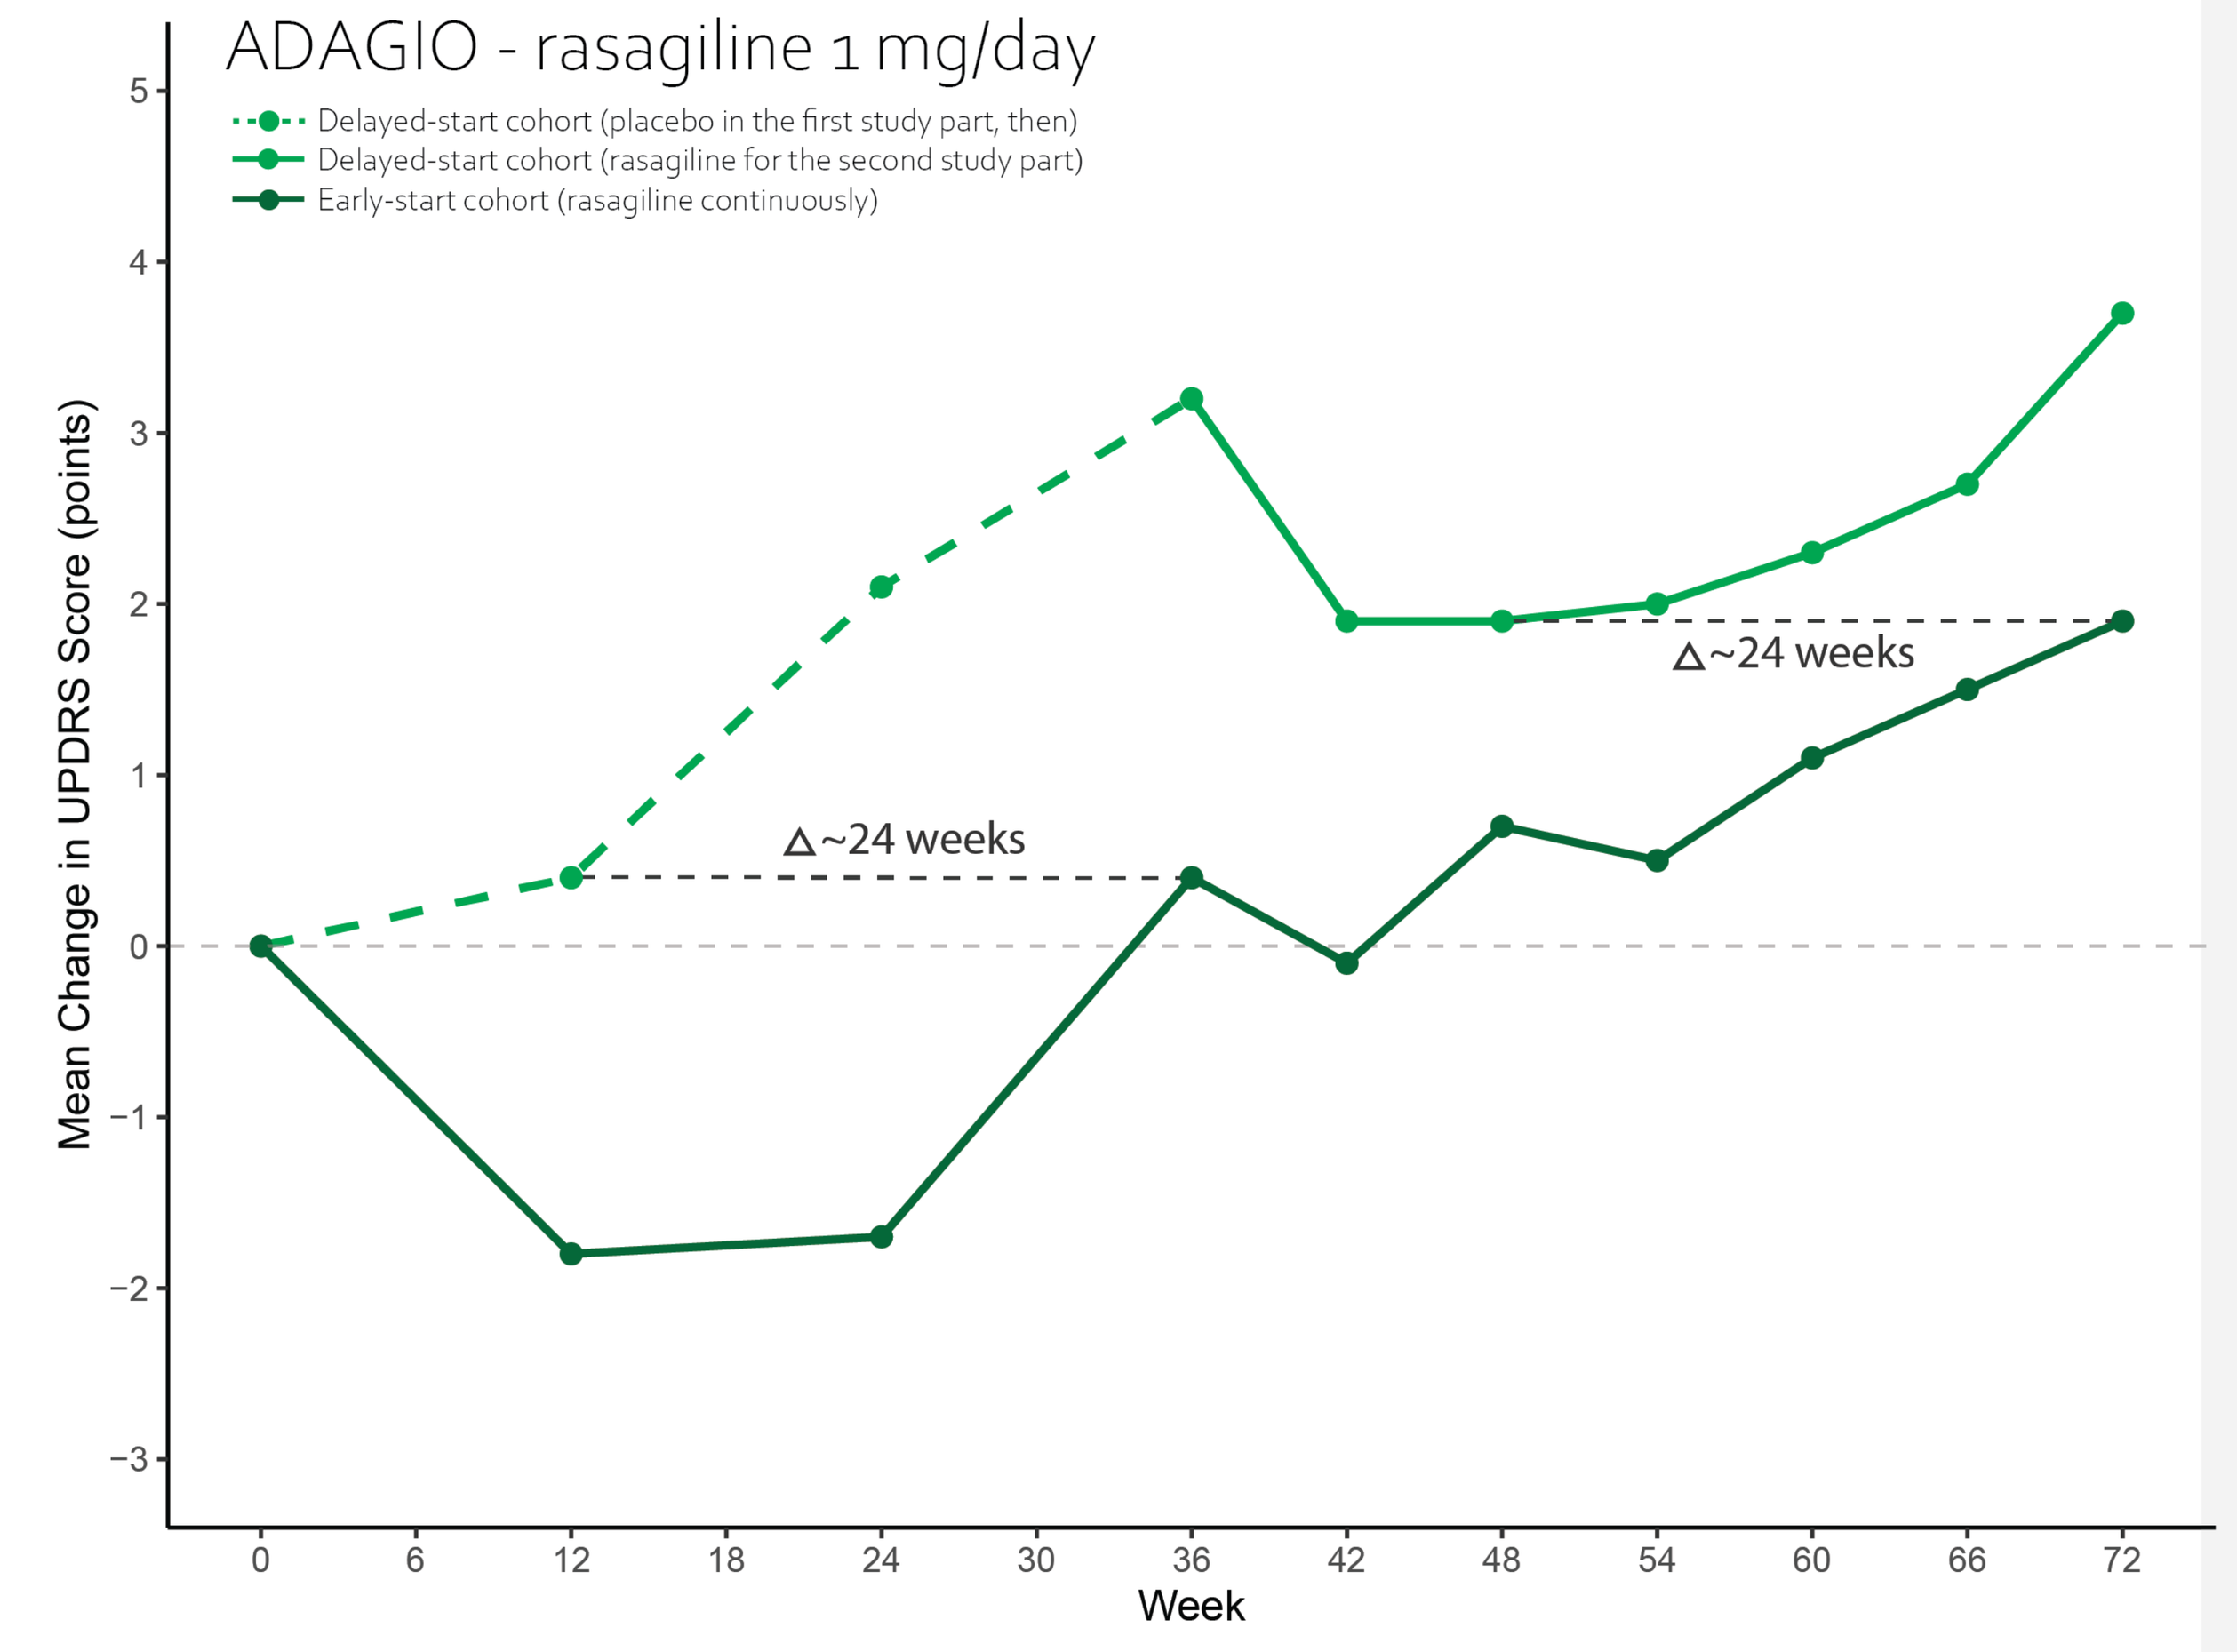
ADAGIO - rasagiline 1 mg/day
Delayed-start cohort (placebo in the first study part, then)
Delayed-start cohort (rasagiline for the second study part)
Early-start cohort (rasagiline continuously)
Mean Change in UPDRS Score (points)
Week
△~24 weeks
△~24 weeks

Higher dose rasagiline (2 mg/day) results showed no persistent effect.

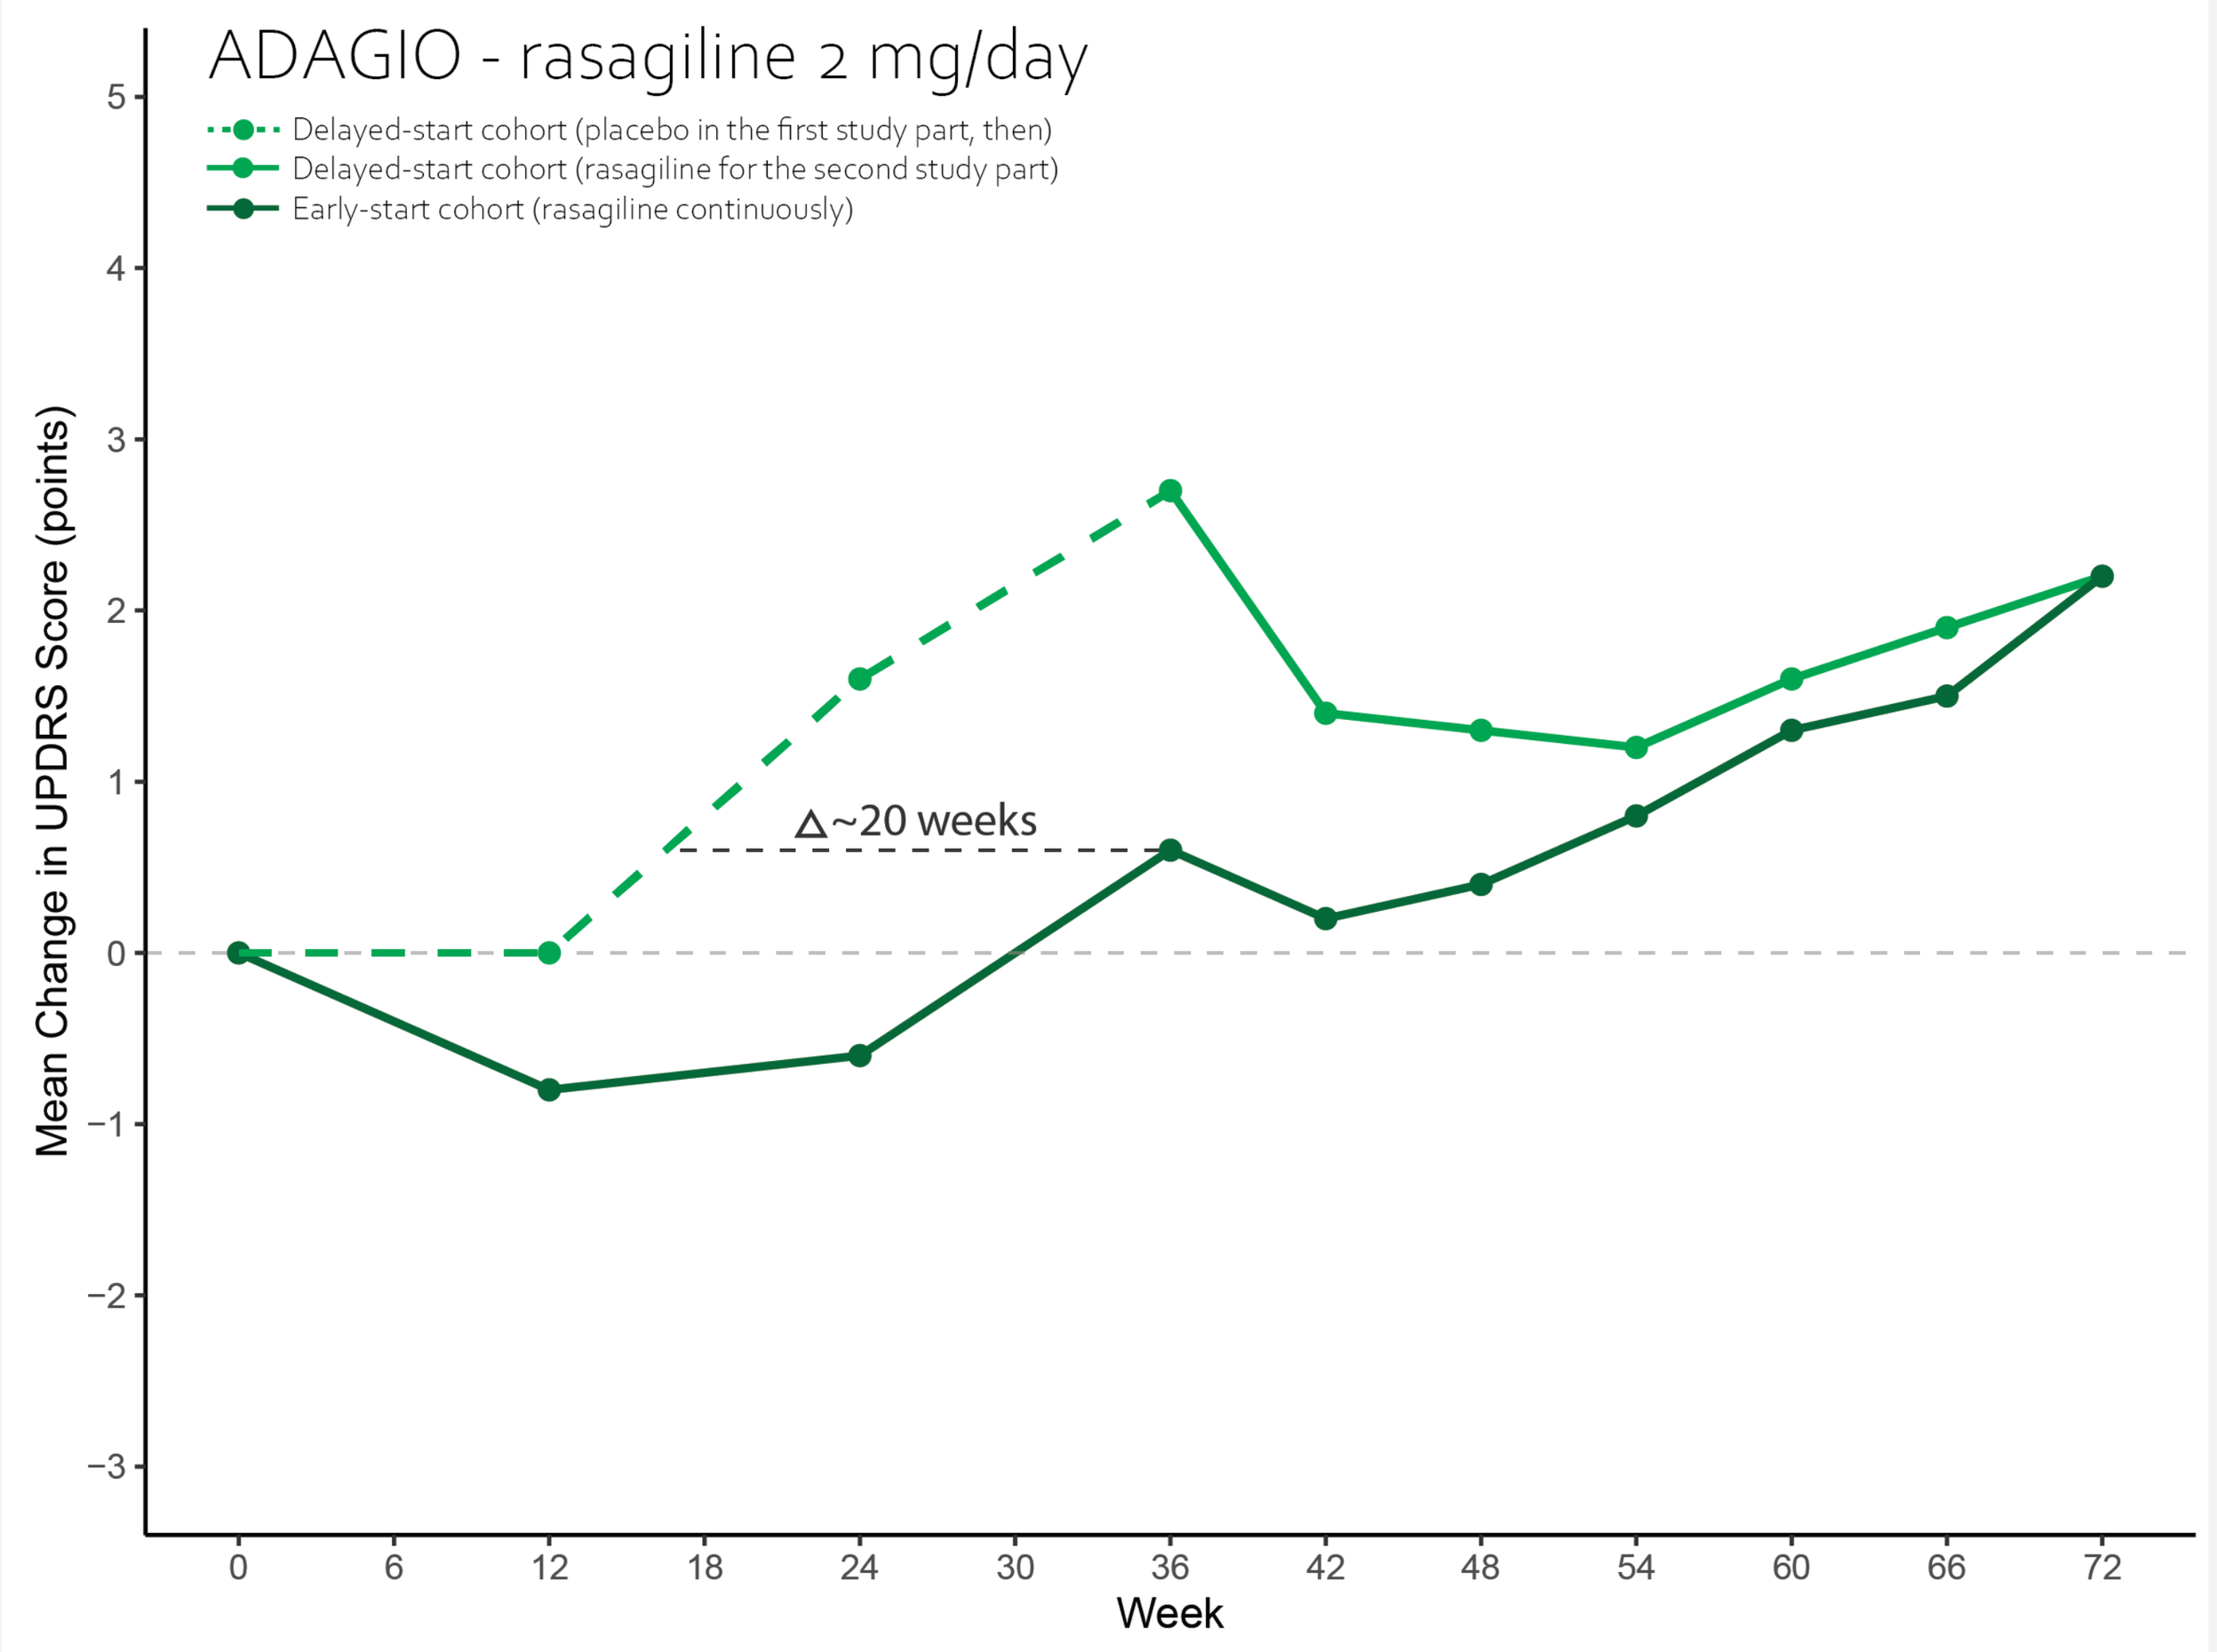
ADAGIO - rasagiline 2 mg/day
Delayed-start cohort (placebo in the first study part, then)
Delayed-start cohort (rasagiline for the second study part)
Early-start cohort (rasagiline continuously)
Mean Change in UPDRS Score (points)
Week
△~20 weeks

For prasinezumab an estimated time delay of approx. 20 weeks after one year, also persisted after two years.

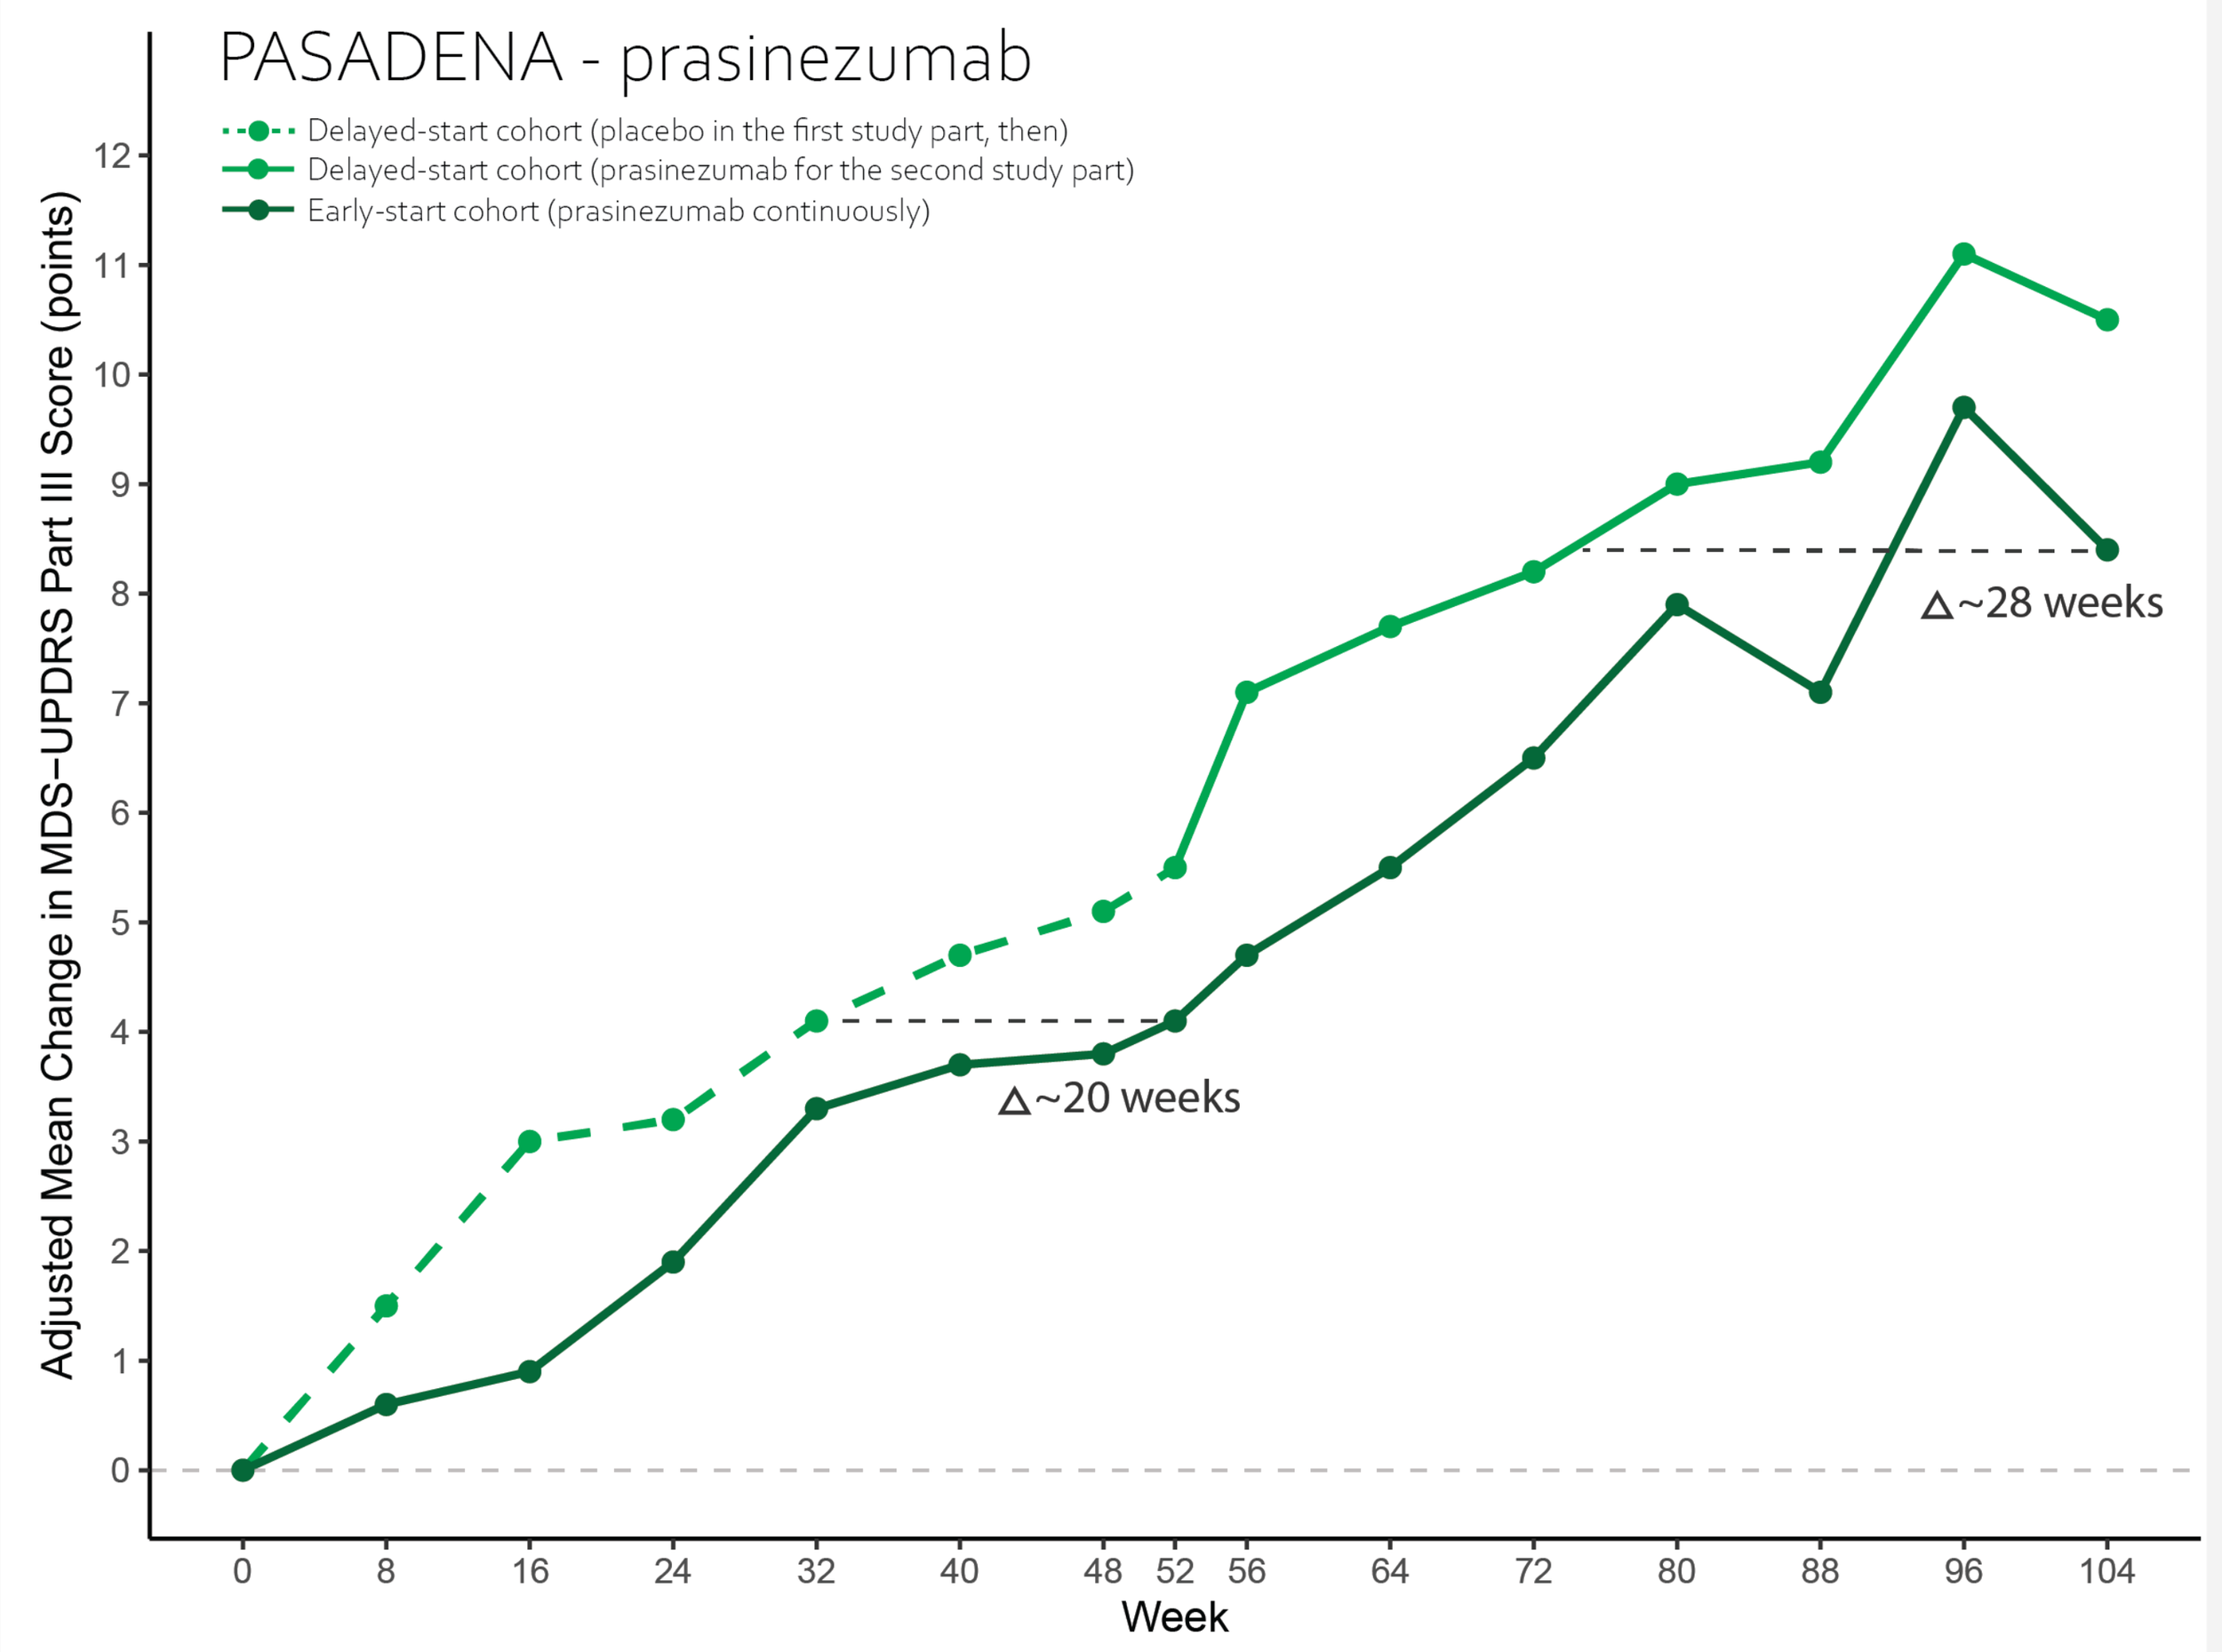
PASADENA - prasinezumab
Delayed-start cohort (placebo in the first study part, then)
Delayed-start cohort (prasinezumab for the second study part)
Early-start cohort (prasinezumab continuously)
Adjusted Mean Change in MDS−UPDRS Part III Score (points)
Week
△~20 weeks
△~28 weeks

The original reporting from clinical trials (difference in mean change from baseline) can be compared to the alternative reporting (mean time delay). The time delay may be easier to interpret.

		rasagiline 1 mg/day		rasagiline 2 mg/day	
Time point	**Cohorts**	**Difference in mean change from baseline UPDRS total score (original reporting)**	**Mean time delay (alternative reporting)**	**Difference in mean change from baseline UPDRS total score (original reporting)**	**Mean time-delay (alternative reporting)**
Week 36	Early-start rasagiline vs. placebo	2.9	24 weeks	1.0	20 weeks
Week 72	Early-start rasagiline vs. delayed-start rasagiline	1.7	24 weeks	0.0	0 weeks

		prazinezumab	
Time point	**Cohorts**	**Difference in mean change from baseline UPDRS part-III score (original reporting)**	**Mean time-delay (alternative reporting)**
Week 52	Early-start prasinezumab vs. placebo	1.3	20 weeks
Week 104	Early-start prasinezumab vs. delayed-start prasinezumab	2	28 weeks

Conclusions

Estimation of time delays can add to the understanding of potential disease modifying effects:

1. It offers an alternative representation of treatment effects which may be easier to interpret and more meaningful to patients. For instance, a time delay of 24 weeks after 36 weeks of treatment might provide for an easier interpretation of the study results than the corresponding 2.9 difference in mean change from baseline measured by the UPDRS total score.
2. Sustained delays over time comparing early vs. delayed start can support hypotheses of a clinically relevant disease modifying effect, even when conventional analysis using treatment effects measured as points on the outcome scale would find the difference to be very small and its clinical relevance questionable.

This analysis is limited by the use of published summary data.

- We approximated the estimates by visually inspecting the published outcome plots, so slight differences from the actual results are to be expected.
- We limited the analysis to the point estimates of the average change from baseline and did not account for uncertainty.

With patient level data available, one could get precise estimates of time delays by utilizing repeated measures methods, such as the progression model for repeated measures[3], and quantify disease-modifying aspects of treatments.

Abbreviations

DMT, disease-modifying treatments; PD, Parkinson's disease; UPDRS, Unified Parkinson's Disease Rating Scale

References

1. Olanow, C. Warren, et al. A double-blind, delayed-start trial of rasagiline in Parkinson's disease. New England Journal of Medicine 361.13 (2009): 1268-1278.
2. Pagano, Gennaro, et al. Trial of prasinezumab in early-stage Parkinson's disease. New England Journal of Medicine 387.5 (2022): 421-432.
3. Raket LL. Progression models for repeated measures: Estimating novel treatment effects in progressive diseases. Statistics in medicine 41.28 (2022):5537-5557.

CO21 Monday May 8th 2023

9:45AM - 1:15PM

International Society for Pharmacoeconomics and Outcomes Research, Boston, MA, USA.