Budget Impact Analysis of Adopting Lanreotide in the Treatment of Acromegaly and GEP-NET at Public Hospitals in Saudi Arabia.

Alnajjar F[1], Raef H[2], Alharbi B[3], Mahrous M[3], Almalki M[1], Alrumaih A[3], Kamas N[4], Alharbi M[5], Magdy H[6], Emam A[7]

King Fahad Medical City, Riyadh, Saudi Arabia, [2] King Faisal Specialist and Research Center, Riyadh, Saudi Arabia, [3] Ministry of Defense and Aviation (MODA) Hospital, Saudi Arabia, [4] King Fahd Specialist Hospital, Dammam, Saudi Arabia, [5] HEPA Solutions, Saudi Arabia, [6] Ipsen, Middle East, Turkey & Africa, [7] Ipsen, Saudi Arabia

For further information, please send your question(s) to amro.nasr@ipsen.com

EE168

Copies of this poster obtained through QR Code are for personal use only and may not be reproduced without written permission from the authors.

Background

- Acromegaly is a rare disease that affects the pituitary gland, and thereby causes the production of high levels of growth hormone (GH). According to recent data, prevalence of acromegaly globally is 5.9 per 100,000 people per year[1]. A retrospective study in Saudi Arabia (SA) found that 195 cases were identified during the past 25 years[2].
- Gastroenteropancreatic neuroendocrine tumors (GEP-NET) often affect cells that secrete hormones thereby resulting in hypersecretion of these hormones[3]. According to recent studies, the GEP-NET incidence rate is 3.56 per 100,000[4]. A retrospective study in Saudi Arabia (SA) found that 72 cases were identified during between 2010 and 2016[6].
- Octreotide (OCT) and lanreotide (LAN) are long-acting somatostatin analogs approved for acromegaly and GEP-NET[5] in Saudi Arabia.
- From the perspective of public hospitals in Saudi Arabia, OCT is the only option available as a drug formulary in many public hospitals that can be used for patients with acromegaly and GEP-NET.
- The differences between LAN and OCT are shown in Table 1.

Table 1: Characteristics of LAN and OCT

	OCT	LAN
Route of Administration	Intramuscular(IM)	Deep Subcutaneous(SC)
Dose Frequency	Every 4 weeks	Every 4 weeks
Sold As (Formulation)*	Powder/Solvent to be mixed into suspension	Pre-filled ready-to-use injection
Price per Unit	20mg (NUPCO Price)** 30mg (NUPCO Price)**	90mg (NUPCO Price) 120mg (NUPCO Price)

**The available formulation on the formulary of public hospitals in Saudi Arabia.* **National Unified Procurement Company(NUPCO)

Objective

To evaluate the budget impact of switching patients with acromegaly or GEP-NET on the formulary from octreotide to lanreotide from the perspective of public hospital payers in Saudi Arabia in a 3-year horizon.

Methods

- A model was developed to estimate the budgetary impact of adopting LAN as a treatment option for patients with acromegaly or GEP-NET.
- The model was adapted in accordance with the local treatment practice based on cost data retrieved from the National Unified Procurement Company (NUPCO) and validated by local experts.
- The model calculated the annual budget impact of introducing LAN in the current Saudi market for two scenarios:
 - ➢ Current scenario: use of OCT for 300 patients per year for the approved indications.
 - ➢ Projected scenario: LAN for 300 patients per year for the approved indications.
- The model estimated 300 patients would be eligible for treatment each year for each scenario at five major public hospitals and calculated the annual budget over a 3-year horizon.

Methods

300 Patients with Acromegaly or GEP-NET

Payer Perspective in 3 Years

300 Patients with OCT vs. 300 Patients with LAN
Drug cost + Care costs Drug cost + Care costs

Direct Drug Acquisition Costs and Care Costs

Total Costs for Each Scenario

Difference in Total Costs

Model Structure

- Medical resources and experts' assumptions were taken into consideration as follows:
 - ➢ Number of annual hospital visits was estimated to be 12 per year for LAN and OCT patients (including administration and follow-up visits).
 - ➢ Since LAN is given deep SC and can be injected independently by the patient or caregiver as per the Saudi label, local experts 300 patients total of 50% only will be receiving the treatment self-administration.
 - ➢ 150 patients treated with LAR needs 1800 visits/per year but 150 patients treated with LAN needs only 900 visits per year.
- One-way sensitivity analysis was done for factors of drug cost, maximum value, and minimum value for each product in total costs per patient per year with assumptions of incremental of (±10%).

Results

- Over a 3-year horizon, the introduction of LAN is predicted to result in a budget reduction. The introduction of LAN is predicted to result in a budget reduction of 21.2% per year, leading to potential cumulative cost savings of $2.87 million for 300 treated patients over a 3-year horizon (Figure 1).
- Self-administration of LAN (estimated proportion of 150 patients per year) would save medical expenses.
- Based on the assumption that 50% of patients could self-administer with LAN each patient would need six visits (follow-up visits) per year instead of 12, saving 5 hours of nurse time annually per patient as time spent preparing and administrating drug (Figure 2).
- The results from one-way sensitivity analysis showed consistent cost savings upon the parameter variation. The number of units per year for LAN had the highest impact on the overall budget impact (Table 2).

Table 2: One-way Sensitivity Analysis

	Total Costs LAN	Total Costs OCT	Change in Total Cost
Base Case	$ 11,901	$ 15,098	$ -4,007
Direct drug cost of LAN (+10%)	$ 13,058	$ 15,098	$ -2,040
Direct drug cost of LAN (-10%)	$ 10,744	$ 15,098	$ -4,354
Direct drug cost of OCT (+10%)	$ 11,901	$ 16,535	$ -4,634
Direct drug cost of OCT (-10%)	$ 11,901	$ 13,660	$ -1,759

Results

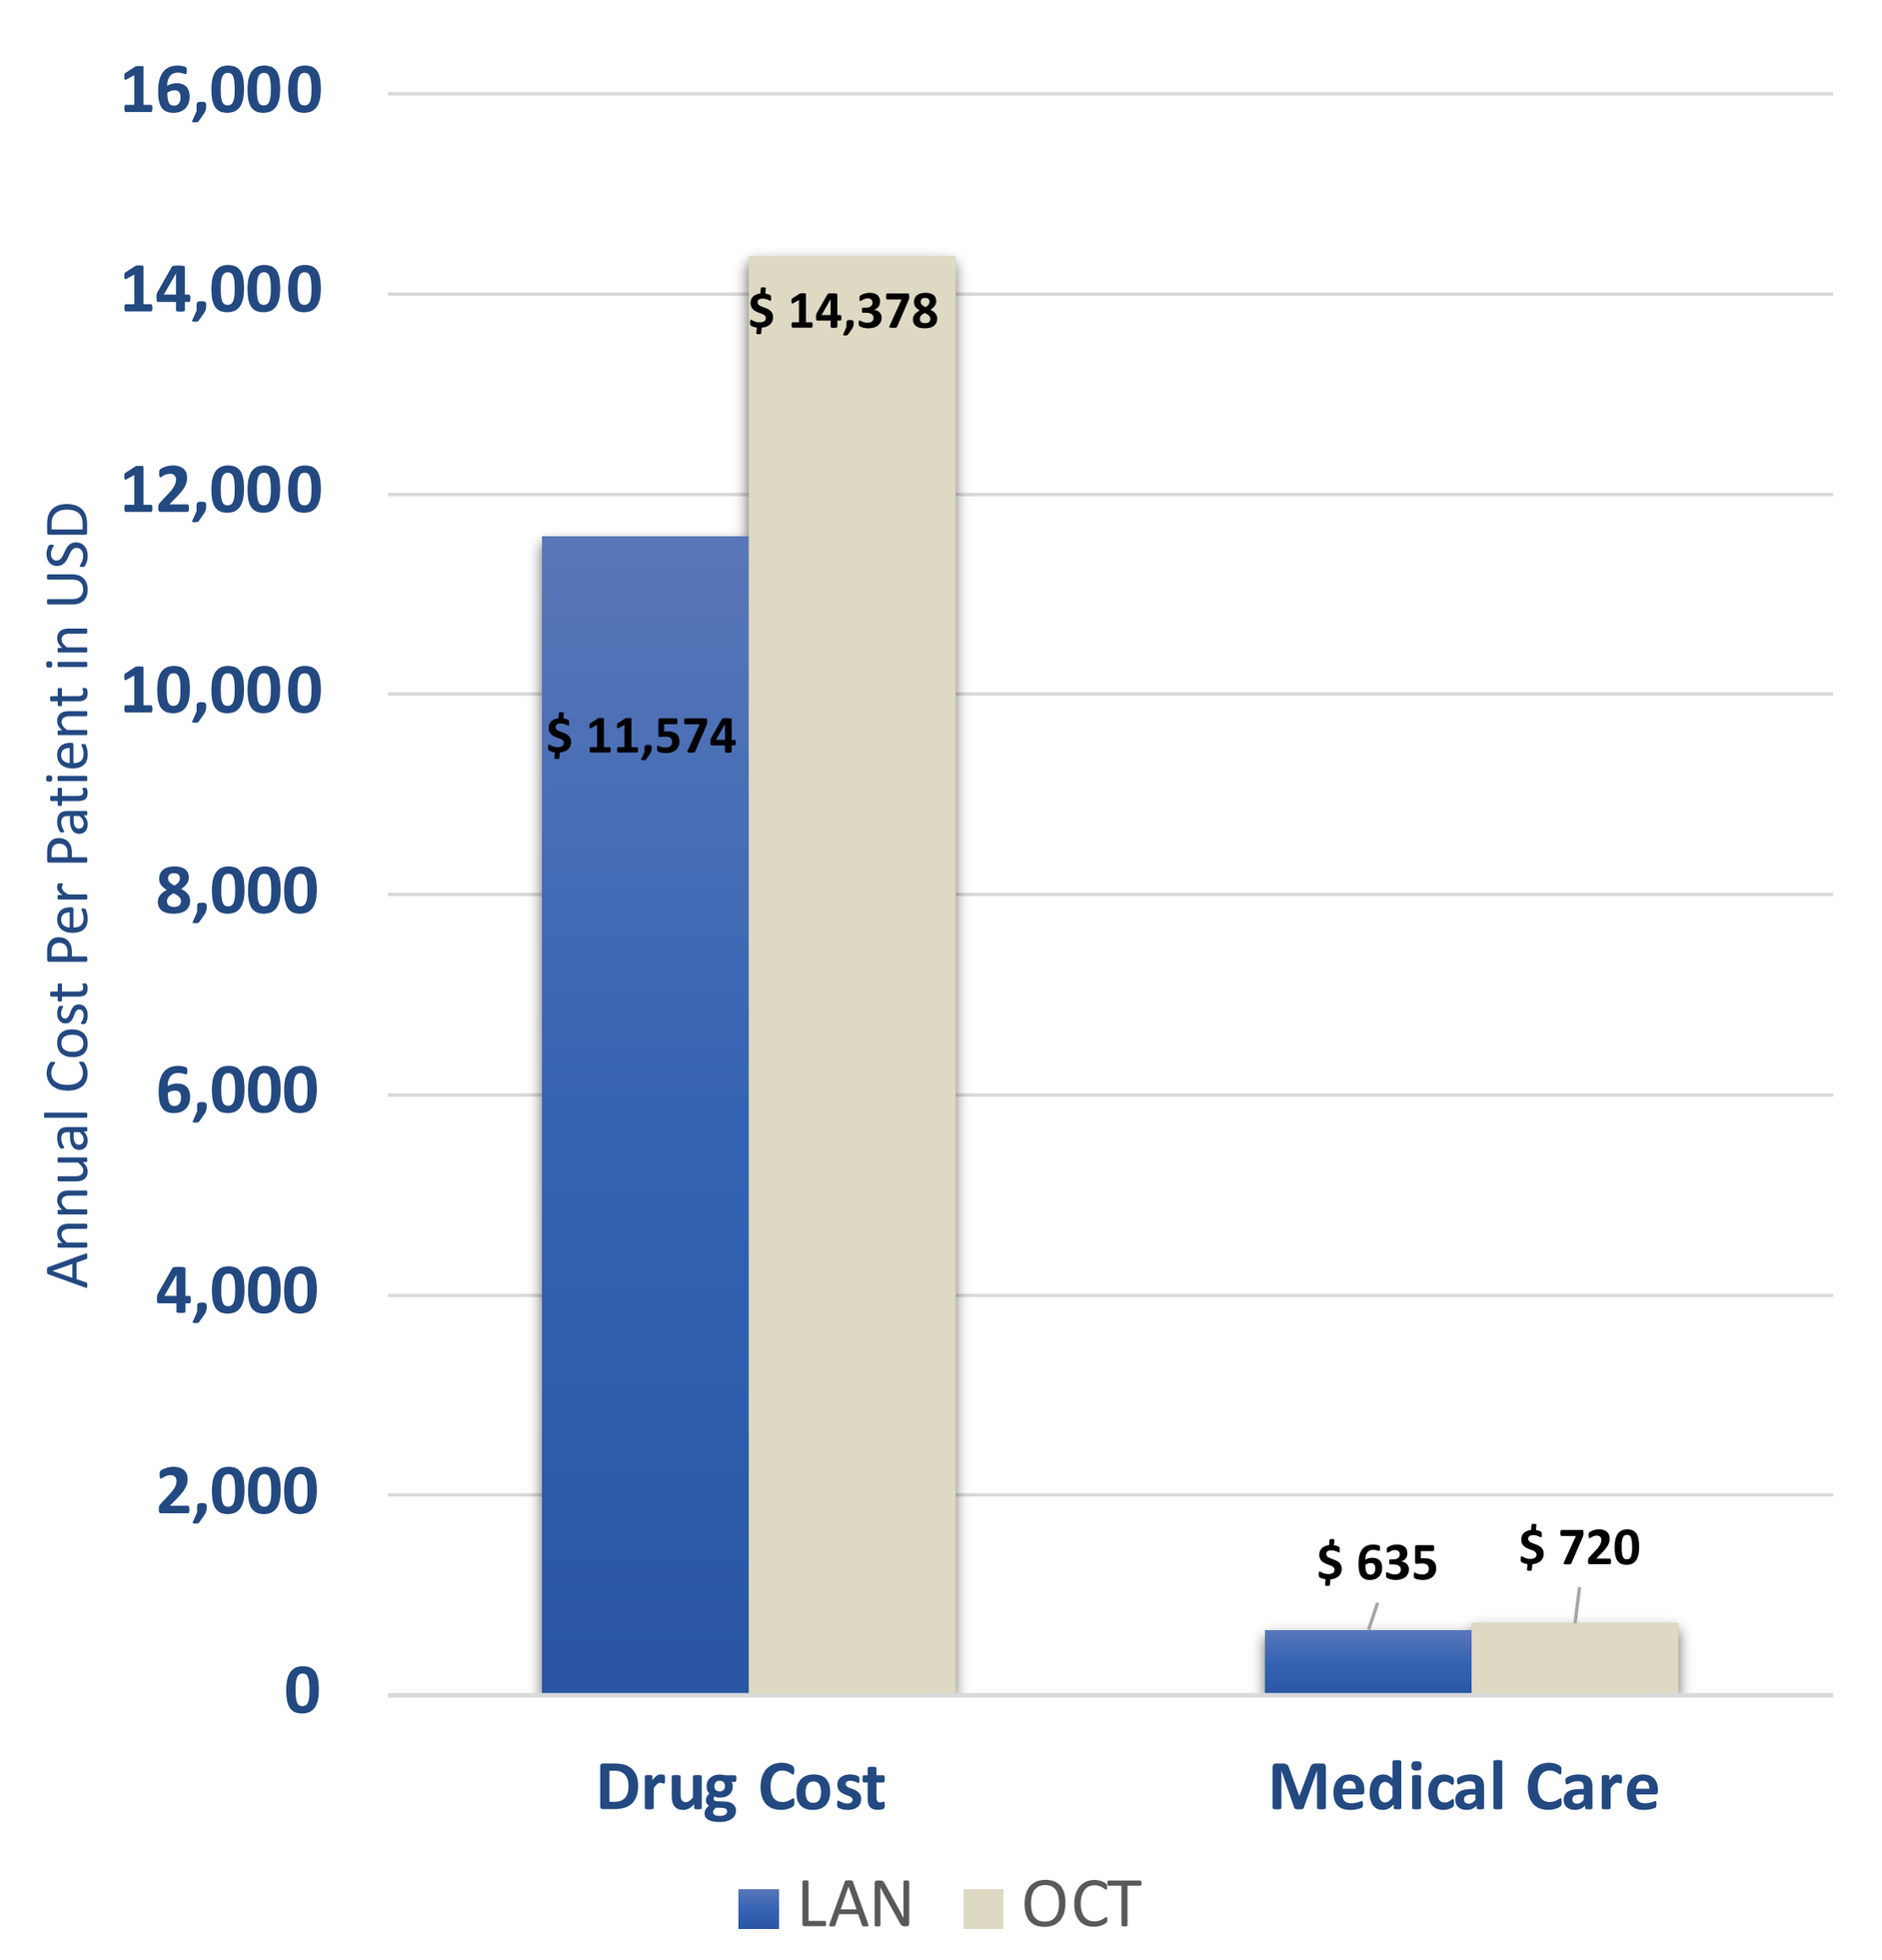
16,000
14,000
12,000
10,000
8,000
6,000
4,000
2,000
0
Annual Cost Per Patient in USD
$ 14,378
$ 11,574
$ 635
$ 720
Drug Cost
Medical Care
LAN
OCT

- Medical Care includes administration, nursing, and primary care/clinic visits
- This is the annual cost yearly per patient. The first is coming from the direct acquisition cost (NUPCO price) the second from the care cost services per year and comes from hospital data and expert feedback.

Figure 1: Annual Total Costs Per Patient in USD

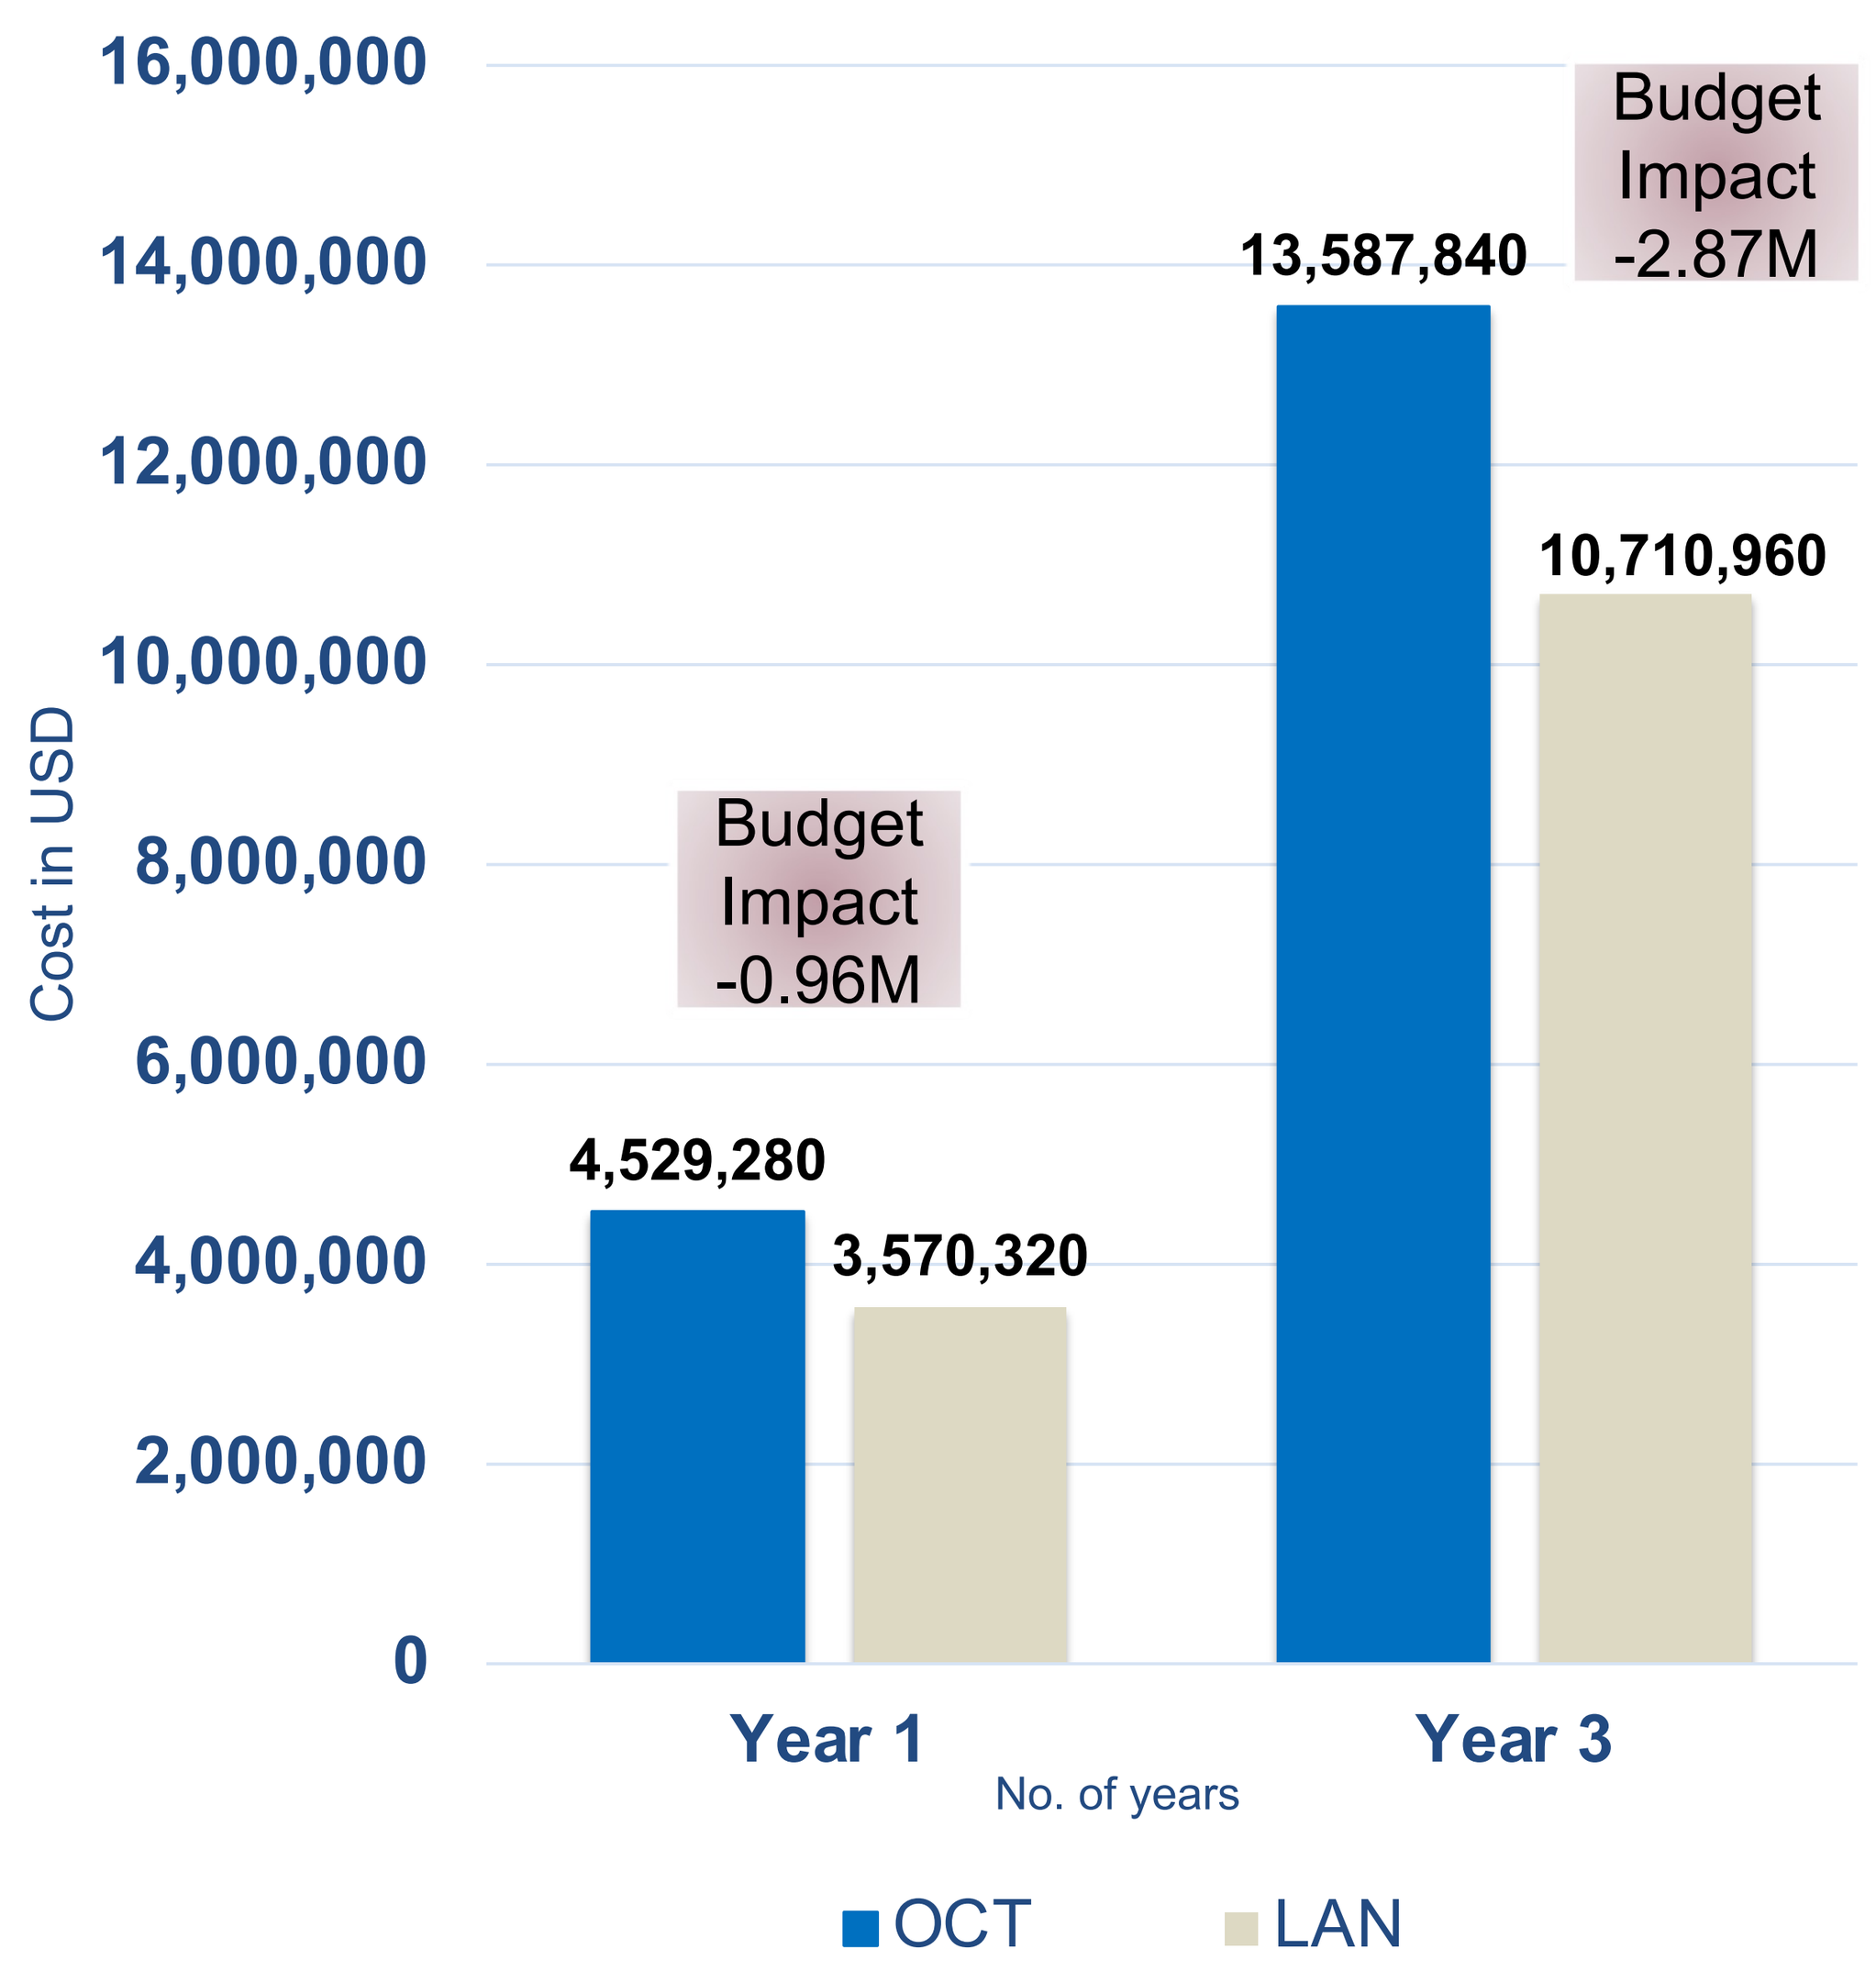
16,000,000
14,000,000
12,000,000
10,000,000
8,000,000
6,000,000
4,000,000
2,000,000
0
Cost in USD
Budget Impact -2.87M
13,587,840
10,710,960
Budget Impact -0.96M
4,529,280
3,570,320
Year 1
Year 3
No. of years
OCT
LAN

Figure 2: First-year cost and cumulative 3-year Cost in USD per 300 Patients.

Conclusions

- The impact analysis showed that adding LAN to the formulary and replacing OCT would result in a cumulative cost saving of $ 2.87 million in the KSA public sector budget over the 3-year horizon.
- The cost savings are primarily driven by lower drug acquisition and reduction in care services cost due to the option of self-administration.
- Furthermore, adding LAN to the drug formulary may help to improve the patient's compliance with the emphasis that 50% of patients will self-inject LAN, reducing the number of hospital visits and hours nurses spend with each patient.

References

1.Crisafulli, S., Luxi, N., Sultana, J., Fontana, A., Spagnolo, F., Giuffrida, G., Ferraù, F., Gianfrilli, D., Cozzolino, A., Cristina De Martino, M., Gatto, F., Barone-Adesi, F., Cannavò, S., & Trifirò, G. (2021). Global epidemiology of acromegaly: a systematic review and meta-analysis, *European Journal of Endocrinology*,*185*(2), 251-263. Retrieved Jan 14, 2023, from https://eje.bioscientifica.com/view/journals/eje/185/2/EJE-21-0216.xml
2.AlMalki MH, Ahmad MM, Buhary BM, Aljawair R, Alyamani A, Alhozali A, Alshahrani A, Alzahrani S, Nasser T, Alzahrani W, Raef H, Aldawish M, Elkhzaimy A. Clinical features and therapeutic outcomes of patients with acromegaly in Saudi Arabia: a retrospective analysis. Hormones (Athens). 2020 Sep;19(3):377-383. doi: 10.1007/s42000-020-00191-0. Epub 2020 May 9. PMID: 32388630.
3.Ershadinia N, Tritos NA. Diagnosis and Treatment of Acromegaly: An Update. Mayo Clin Proc. 2022 Feb;97(2):333-346. doi: 10.1016/j.mayocp.2021.11.007. PMID: 35120696.
4.Das S, Dasari A. Epidemiology, Incidence, and Prevalence of Neuroendocrine Neoplasms: Are There Global Differences? Curr Oncol Rep. 2021 Mar 14;23(4):43. doi: 10.1007/s11912-021- 01029-7. PMID: 33719003; PMCID: PMC8118193.
5.Díez M, Teulé A, Salazar R. Gastroenteropancreatic neuroendocrine tumors: diagnosis and treatment. Ann Gastroenterol. 2013;26(1):29-36. PMID: 24714698; PMCID: PMC3959515.
6.Bazarbashi, S., Aseafan, M., Elgazzar, T. *et al.* Characteristics and treatment results of patients with gastroenteropancreatic neuroendocrine tumors in a tertiary care centre. *BMC Endocr Disord* **23**, 74 (2023). https://doi.org/10.1186/s12902-023-01326-1

Author Contributions: Substantial contributions to study conception/design, or acquisition/analysis/interpretation of data: all authors. Drafting of the publication, or revising it critically for important intellectual content: all authors. Final approval of the publication: all authors.

Acknowledgement: The authors thank Ipsen team and all contributors for their commitment and dedication to the goal of evaluating the economic project.

Disclosure: This study was conducted by HEPA and funded by Ipsen

Medical Writing Support: The authors thank Sondus, HEPA Solutions Company, for providing medical writing support, which was industry sponsored in accordance with Good Publication Practice guidelines (GPP 2022).

Presented at the [ISPOR, Boston, USA] | Virtual format | [05-2023]

This study was sponsored by Ipsen