EE402

Cost-effectiveness Analysis of Pasireotide Long-Acting Release as a Second Line Treatment for Adult Acromegaly Patients

Paiva, Helder, PhD[1]; Teloian, Diana, MA[2]; Tichy, Eszter, MSc[2]; Schmidt, Fabian, MSc[3]; Kalinowska, Agnieszka, MSc[4]

[1]Recordati Rare Diseases, São Paulo, Brazil; [2]Evidera, Inc, Budapest, Hungary; [3]Recordati Rare Diseases, Puteaux, France; [4]MAHTA INTL, Warsaw, Poland

Objective

This study aims to evaluate the cost-effectiveness of pasireotide versus other second-line pharmacological treatments in the Brazilian public payer setting for adults with acromegaly for whom surgery is not an option or has not been curative and who are inadequately controlled with another somatostatin analogue.

Background

- Acromegaly is a rare hormonal disorder caused by excessive levels of growth hormone (GH) and insulin-like growth factor 1 (IGF-1).[1]
- Patients with acromegaly also develop disease-associated comorbidities, including diabetes, hypertension, cardiomyopathy, colon cancer, and sleep apnea[2-4] that impact quality of life[5] and cause excess mortality.[1,5]
- The primary treatment option for acromegaly is surgery. However, postoperative remission rate is 66%.[6]
- In case of persistent disease after surgery, medical therapy is recommended. First-generation long-acting release (LAR) somatostatin receptor ligands (SRL) (octreotide, lanreotide) and cabergoline (for patients with modest elevations of IGF-1 and mild signs and symptoms of acromegaly) are recommended as first-line treatments.[6,7]
- Recommended second-line treatment options are first-generation SRLs, pasireotide[6] (an injectable second-generation SRL with high binding affinity for human somatostatin receptor subtypes 1, 2, 3, and 5) that impacts GH and IGF-1, and pegvisomant-based regimens (monotherapy and combination with an SRL)[6] that target only IGF-1 normalization.[8]
- Patients may achieve full control (FC), partial control (PC), or remain in no control (NC) state in terms of biomarker levels as defined in **Table 1**. (Note that updated treatment practice guidelines published in 2014 recommend a more stringent level of GH for FC, <1 μg/l.[6] However, to be consistent with all efficacy data sources, the model applies a 2.5 μg/l GH cut-off. Treatment effects based on a 1.0 μg/l GH level were assessed in the scenario).

Table 1. Acromegaly control definitions

Disease control	Biochemical criteria option 1: GH and IGF-1	Biochemical criteria option 2: IGF-1
Full Control	GH < 2.5 μg/l and IGF-1 ≤ ULN[11,21,35]	IGF-1 ≤ ULN[11,35]
Partial Control	Not full control, at least 50% decrease in GH and IGF-1[7]	Not full control, at least 50% decrease in IGF-1[7]
No Control	Not full control, not partial control[11,21,35]	Not full control, not partial control[11,35]

GH = growth hormone; IGF-1 = insulin-like growth factor 1; ULN = upper limit of normal

Methods

- A hybrid model combining a **six-month decision-tree** (initial) phase followed by a lifetime Markov phase with **six-month cycles** was designed to show the health and economic outcomes of pasireotide.
- All patients **enter the model with NC of acromegaly**, have failed first-generation SRLs in the previous line, and start receiving a base dose (40mg) of pasireotide or another second-line comparator.
- Disease control is evaluated after three months (first assessment) and six months (second assessment) from second-line therapy initiation during the decision-tree phase.
- Depending on the treatment effects at the assessments, patients enter the Markov phase continuing second-line treatment or transitioning to a subsequent line as described in **Figure 1**.

Figure 1. Model Structure

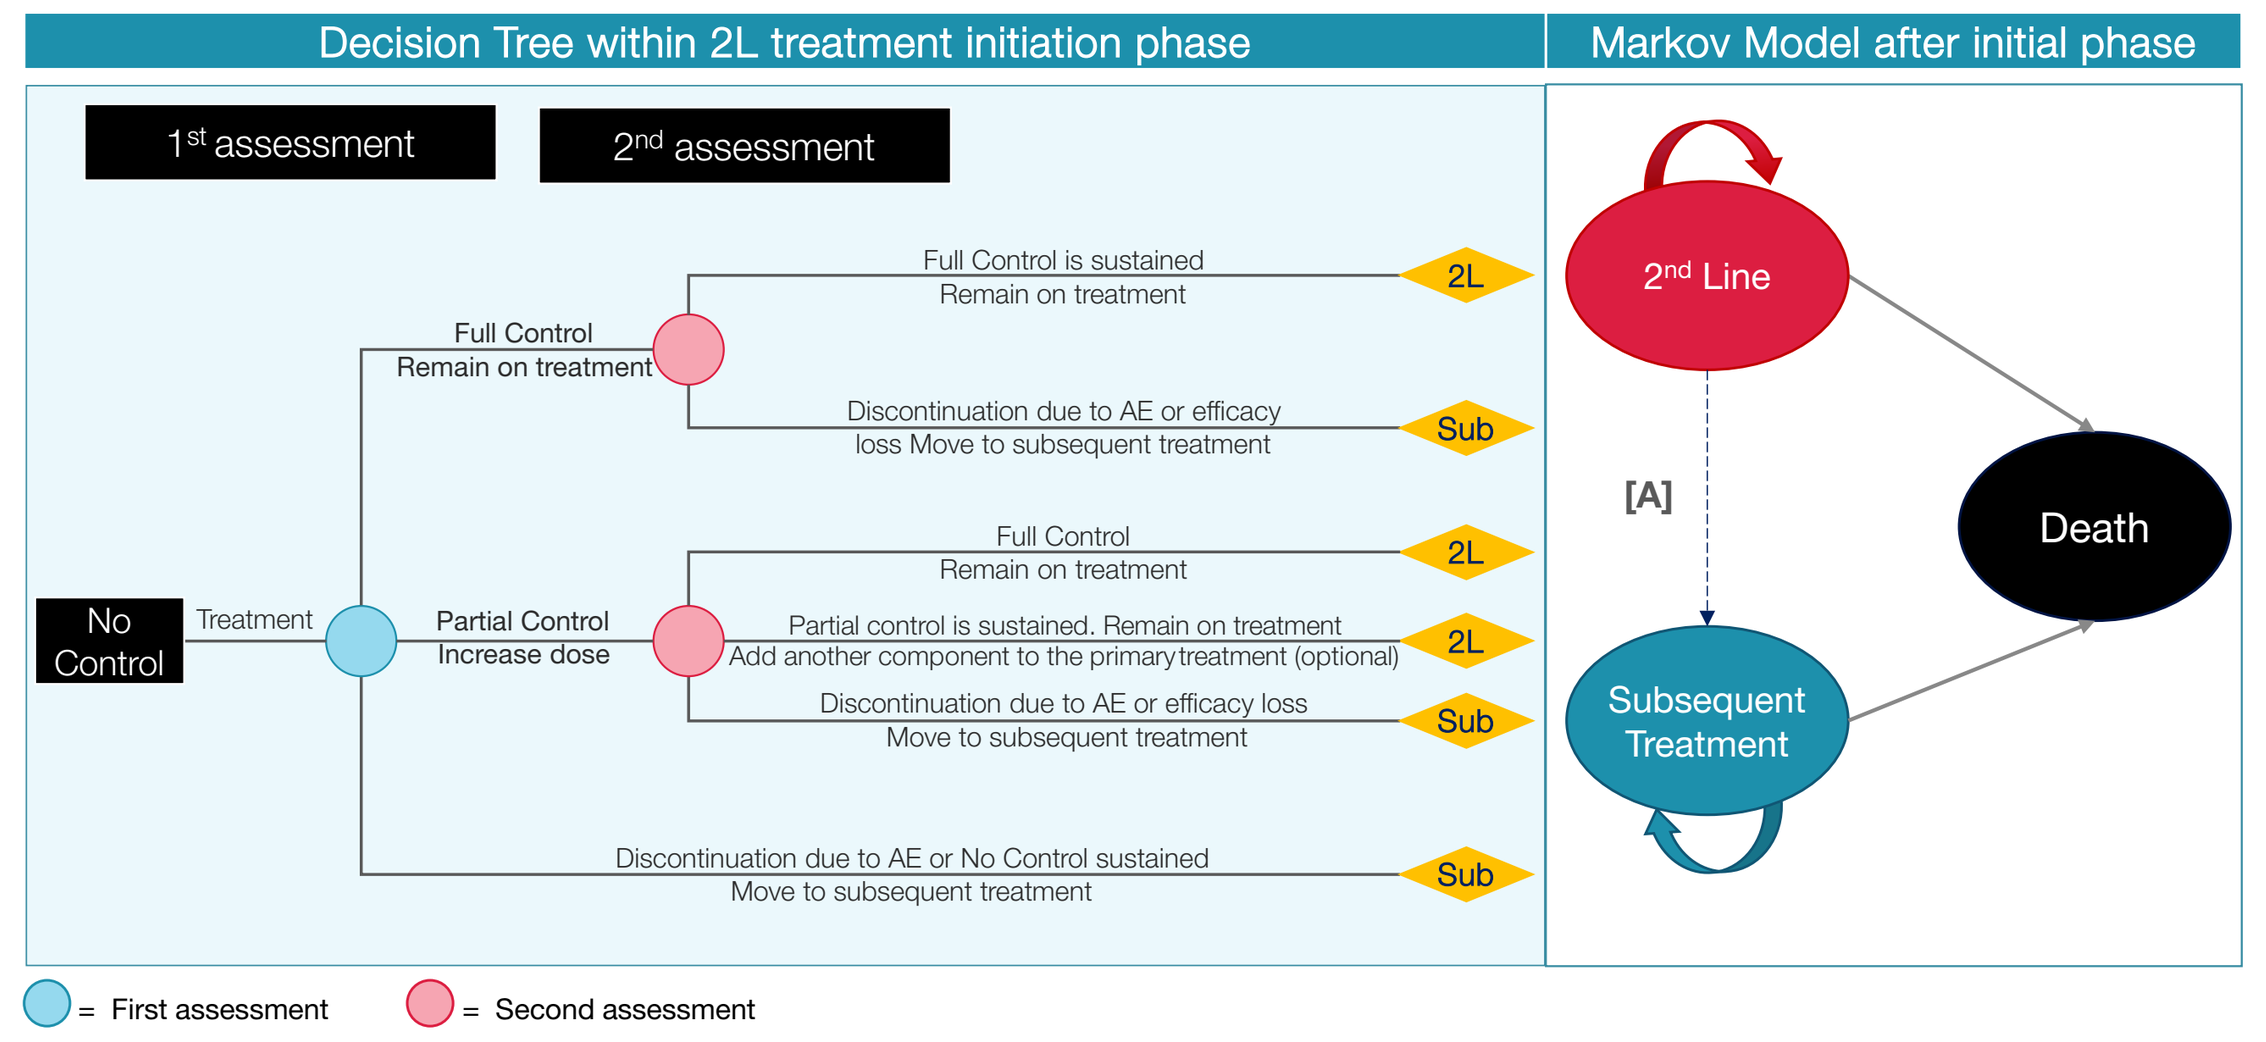
Decision Tree within 2L treatment initiation phase
Markov Model after initial phase
1st assessment
2nd assessment
Full Control is sustained
Remain on treatment
Full Control
Remain on treatment
Discontinuation due to AE or efficacy
loss Move to subsequent treatment
Full Control
Remain on treatment
No Control
Treatment
Partial Control
Increase dose
Partial control is sustained. Remain on treatment
Add another component to the primary treatment (optional)
Discontinuation due to AE or efficacy loss
Move to subsequent treatment
Discontinuation due to AE or No Control sustained
Move to subsequent treatment
2L
Sub
2nd Line
[A]
Death
Subsequent Treatment
= First assessment
= Second assessment

2L = second line; AE = adverse event; Sub = subsequent treatment;
2nd line in Markov model includes those with full and partial control. Subsequent treatment state includes patients with full, partial and no control.
[A] transition: patients on 2nd line of therapy can switch to subsequent treatment in case of response loss
Note: *Path [A] transition probability will be assumed to be 0% in base case*

Methods (Cont'd)

- Patients remain on second-line treatment until death if they achieve FC or PC during the six-month decision tree (initial) phase, whereas patients with uncontrolled disease move to a subsequent line. During the initial phase, patients may also move to a subsequent line due to loss of efficacy or treatment discontinuation due to serious adverse events (AE).
- Patients who achieve PC during first assessment are administered an increased dose of active treatment to achieve better control.
- In the Markov phase, patients can move to a subsequent line if they lose efficacy to second-line treatment. In the base case it is assumed that no patients lose efficacy throughout lifetime. In the scenario analysis, the probability of treatment effect loss for patients treated with pasireotide was tested (based on the discontinuation rate from PAOLA study).[9]
- The subsequent line consists of lifetime medical treatments or a combination of radiotherapy (RT) and an SRL. If a patient achieves biochemical control with RT + SRL they stop any acromegaly treatment. Otherwise, if RT + SRL did not have a successful result, they continue receiving the same SRL for lifetime after RT duration is over. Considering available acromegaly treatments options in Brazil the model assumes that half of the patients will get octreotide + RT and half lanreotide + RT in a subsequent line.
- The model implements a lifetime horizon. Costs and benefits are discounted at a rate of 5% per year.[10]
- In the base case, the model applies GH and IGF-1 as biochemical markers for treatment response and evaluates the cost-effectiveness of pasireotide against maximal doses of lanreotide and octreotide.
- To assess pegvisomant-based treatments that target only normalization of IGF-1 concentration, the model incorporates a scenario where efficacy is defined as IGF-1 normalization only, since pegvisomant is not a recommended treatment in the Brazilian acromegaly treatment guidelines.

Model Inputs

Clinical Inputs

- Transition probabilities for pasireotide and SRLs were based on the PAOLA[11] clinical trial. When alternative biochemical criteria (disease control biomarker: IGF-1) were considered in a scenario, the model applied odds ratios from a network meta-analysis[12] to derive transition probabilities for pegvisomant-inclusive treatments. Transition probabilities are summarized in **Table 2** and **Table 3**.
- During the first 6 months from treatment initiation patients may discontinue treatment due to adverse events, with 3-month probability of 0.67% for octreotide,[21] 2.62% for pasireotide (average of 40mg and 60mg dose discontinuation),[11] and 10.56% for lanreotide.[14] Pegvisomant based treatments had 5.53-10.56% treatment discontinuation probability.[14,13]
- In subsequent line with the combination of RT and medical treatment, the duration of therapy was assumed to be 10 years, and the proportion of patients experiencing control is 60%.[15]

Table 2. Transition probabilities (GH and IGF-1). Source: PAOLA study[11]

Treatment	First Assessment (at 3 months)			Second assessment (at 6 months)	
	Achieve FC	Achieve PC	Remain in NC*	PC achieve FC with increased dose	Remain in PC
Pasireotide	8.01%	11.99%	80.00%	100.00%	0.00%
Octreotide max dose	0.00%	0.00%	100.00%	NA	NA
Lanreotide max dose	0.00%	0.00%	100.00%	NA	NA

*All no control patients move to subsequent line after first assessment
FC = full control; NA = not applicable; NC = no control; PC = partial control

Table 3. Transition probabilities (IGF-1). Source for pegvisomant-based treatments – ITC,[12] remainder - PAOLA[11]

Treatment	First Assessment (at 3 months)			Second assessment (at 6 months)	
	Achieve FC	Achieve PC	Remain in NC*	PC achieve FC with increased dose	Remain in PC
Pasireotide	13.18%	12.98%	73.85%	100.00%	0.00%
Octreotide max dose	0.00%	0.00%	100.00%	NA	NA
Lanreotide max dose	0.00%	0.00%	100.00%	NA	NA
Pegvisomant	6.74%	17.43%	75.83%	100.00%	0.00%
Pegvisomant + Octreotide	15.66%	12.94%	71.40%	100.00%	0.00%
Pegvisomant + Lanreotide	15.66%	12.94%	71.40%	100.00%	0.00%

*All no control patients move to a subsequent line after the first assessment
FC = full control; NA = not applicable; NC = no control; PC = partial control

Mortality

- The association between acromegaly and its risk of mortality relative to general population is captured through standardized mortality ratios (SMR) for each health state. The model assumes mortality of FC patients with acromegaly to be equal to the **general population mortality**,[16] while the SMR of active disease patients is greater than one. The SMR for **NC patients (2.40)** was extracted from a published study.[16] The **PC** SMR was estimated as the arithmetic average of the SMR of patients with FC and with NC **(1.70); subsequent treatment** SMR was assumed to be equal to the SMR of acromegaly patients who underwent radiotherapy **(2.10)**.[17]

Health-related Quality of Life

- Patients' quality of life was captured through (1) health-state utility; (2) treatment-related AE disutility; and (3) comorbidity-related disutilities per health state.
- Health state utilities for FC and NC health states were based on Liu 2018,[18] the subsequent treatment utility was derived from Kyriakakis 2017[19] and the PC utility was assumed to be the average of FC and PC values.
- The comorbidity distribution depends on biological control criteria and was derived from published studies.[2-4] The model considers only cardiovascular (hypertension, arrythmia, cardiomyopathy), endocrine (diabetes) and respiratory (sleep apnea) comorbidities. Comorbidity distributions were available for FC and NC patients; assumptions were made to estimate values for partial control and subsequent treatment health states.

Table 4. Health State Utilities

		Comorbidity Disutility[5,20]	
Health State	Health State Utilities	Base case (GH and IGF-1)	Scenario (IGF-1)
Full Control	0.7500	- 0.1156	- 0.1222
Partial Control	0.6400	- 0.1470	- 0.1513
No Control	0.5300	- 0.1784	- 0.1804
Subsequent Treatment	0.5500	- 0.1407	- 0.1455

GH = growth hormone; IGF-1 = insulin-like growth factor 1

- For each intervention, the model employed grade 3–4 AEs that occurred in >2% of patients[21-25] for any treatment. AE disutilities were extracted from the published literature.[5,20,26] AE disutilities for the modeled treatments ranged between 0.0002 and 0.0035. Subsequent treatment utility was decremented for hypopituitarism disutility[5] (–0.1172) for the duration of RT (10 years) as it is a common toxicity.

Dosing and Drug Acquisition Cost Inputs

- Lanreotide, octreotide, and cabergoline prices[27] are exempted from tax, while the pasireotide price[28] includes 17% tax as it is not a reimbursed product in Brazil.

Table 5. Dosing and Drug Acquisition Cost Inputs

Treatment	Base Dose	Unit cost	Monthly cost	Increased Dose (applied to PC patients)	Unit cost	Monthly cost
Pasireotide	40mg/28 days	R$161.73/mg	R$7,033	60mg/28 days	R$107.82/mg	R$7,033
Octreotide max dose	40mg/28 days	R$118.35/mg	R$5,146	40mg/28 days	R$118.35/mg	R$5,146
Lanreotide max dose	120mg/28 days	R$17.07/mg	R$2,226	120mg/28 days	R$17.07/mg	R$2,226
Additional comparator evaluated in the IGF-1 scenario						
Pegvisomant	13mg daily	R$25.39/mg	R$10,045	20mg daily	R$25.39/mg	R$15,454
Pegvisomant + Octreotide	Pegvisomant 15mg daily	R$25.39/mg	R$15,451	Pegvisomant 20mg daily	R$25.39/mg	R$19,314
	Octreotide 30mg/28 days	R$118.35/mg		Octreotide 30mg/28 days	R$118.35/mg	
Pegvisomant + Lanreotide	Pegvisomant 15mg daily	R$25.39/mg	R$13,817	Pegvisomant 20mg daily	R$25.39/mg	R$17,681
	Lanreotide 120mg/28 days	R$17.07/mg		Lanreotide 120mg/28 days	R$17.07/mg	

IGF-1 = insulin-like growth factor 1; PC = partial control
Note: dose of octreotide monotherapy was based on the Brazilian acromegaly guideline[29]
Note: Base dose of pegvisomant was based on average baseline data of patients treated with pegvisomant excluding data on patients with 0mg dose in ACROSTUDY.[30]
Note: Pegvisomant is up-titrated over time until maximum licensed dose of 30mg/day is reached.[30] The dose increase per half year (in pegvisomant monotherapy 0.31mg/day and in pegvisomant combination therapy 0.89mg/day) was estimated based on ACROSTUDY.[31]

Other Costs

- Based on the treatment guidelines and recommendations for acromegaly, the model applies regular monitoring. Unit costs for the procedures were derived from istema de Gerenciamento da Tabela de Procedimentos, Medicamentos e OPM do SUS (SIGTAP)[32] and frequencies of resources were based on published recommendations.[6,33-35]
- AE management costs were derived from the published nationwide study on healthcare costs in Brazil[36] and Brazil's official medicines and procedures cost from Ministry of Health database (SIGTAP[32])
- Comorbidity costs per health state were derived from published study Zhao et al 2020.[36]
- Subsequent treatment cost for the first 10 years was estimated as average of RT + maximal-dose octreotide and RT + maximal-dose lanreotide: R$5,083 per month (considering RT cost of R$1,397 per month; includes R$4,168 radiotherapy cost[32] divided by 3, assuming radiotherapy is implemented every 90 days, and R$23.08, cost for hospitalization for the radiotherapy treatment[32]).
- Patients who do not achieve FC with RT + SRL after 10 years, incur monthly R$3,686 medical treatment cost (average of octreotide and lanreotide costs) for lifetime.
- All costs in the model are in 2022 Brazilian reals.

Results

- Compared to maximal doses of SRLs, pasireotide was associated with higher incremental quality-adjusted life years (QALY) (0.95), resulting in an incremental cost-effectiveness ratio (ICER) of R$150,051 and R$159,143 per QALY (**Table 6**) vs. maximum doses of octreotide and lanreotide, respectively.

Table 6. Model Results. Base Case—GH and IGF-1 is Biochemical Control Criteria

Health Outcomes	Pasireotide	Octreotide max dose	Lanreotide max dose
Total LYs	14.3989	14.1117	14.1117
Full Control	2.8883	0.0000	0.0000
Partial Control	0.0288	0.0000	0.0000
No Control	0.2500	0.2500	0.2500
Subsequent Treatment	11.2318	13.8617	13.8617
Total QALYs	5.7756	4.8292	4.8283
Full Control	1.8319	0.0000	0.0000
Partial Control	0.0142	0.0000	0.0000
No Control	0.0878	0.0878	0.0870
Subsequent Treatment	3.8417	4.7413	4.7413
Costs	Pasireotide	Octreotide max dose	Lanreotide max dose
Total Costs	R$775,849	R$633,834	R$625,098
Drug acquisition	R$737,497	R$595,718	R$586,958
Administration	R$0	R$0	R$0
Monitoring	R$7,042	R$6,165	R$6,165
Comorbidity	R$30,679	R$31,200	R$31,200
AE management	R$632	R$752	R$776
Incremental Results		Octreotide max dose	Lanreotide max dose
Incremental LYs		0.2871	0.2871
Incremental QALYs		0.9464	0.9473
Incremental Costs		R$142,015	R$150,751
ICER per LY		R$494,572	R$524,996
ICER per QALY		R$150,051	R$159,143

AE = adverse event; ICER = incremental cost-effectiveness ratio; LY = life year; QALY = quality-adjusted life year

- The deterministic sensitivity analysis (DSA) demonstrated that the model results were most sensitive to variation of FC and subsequent treatment utilities and doses of pasireotide (**Figure 2**).
- The probabilistic sensitivity analysis (PSA) analysis showed that in 100% of iterations pasireotide was more costly and more effective than SRL comparators (**Figure 3**).

Figure 2. Tornado Diagrams of One-way Sensitivity Analysis

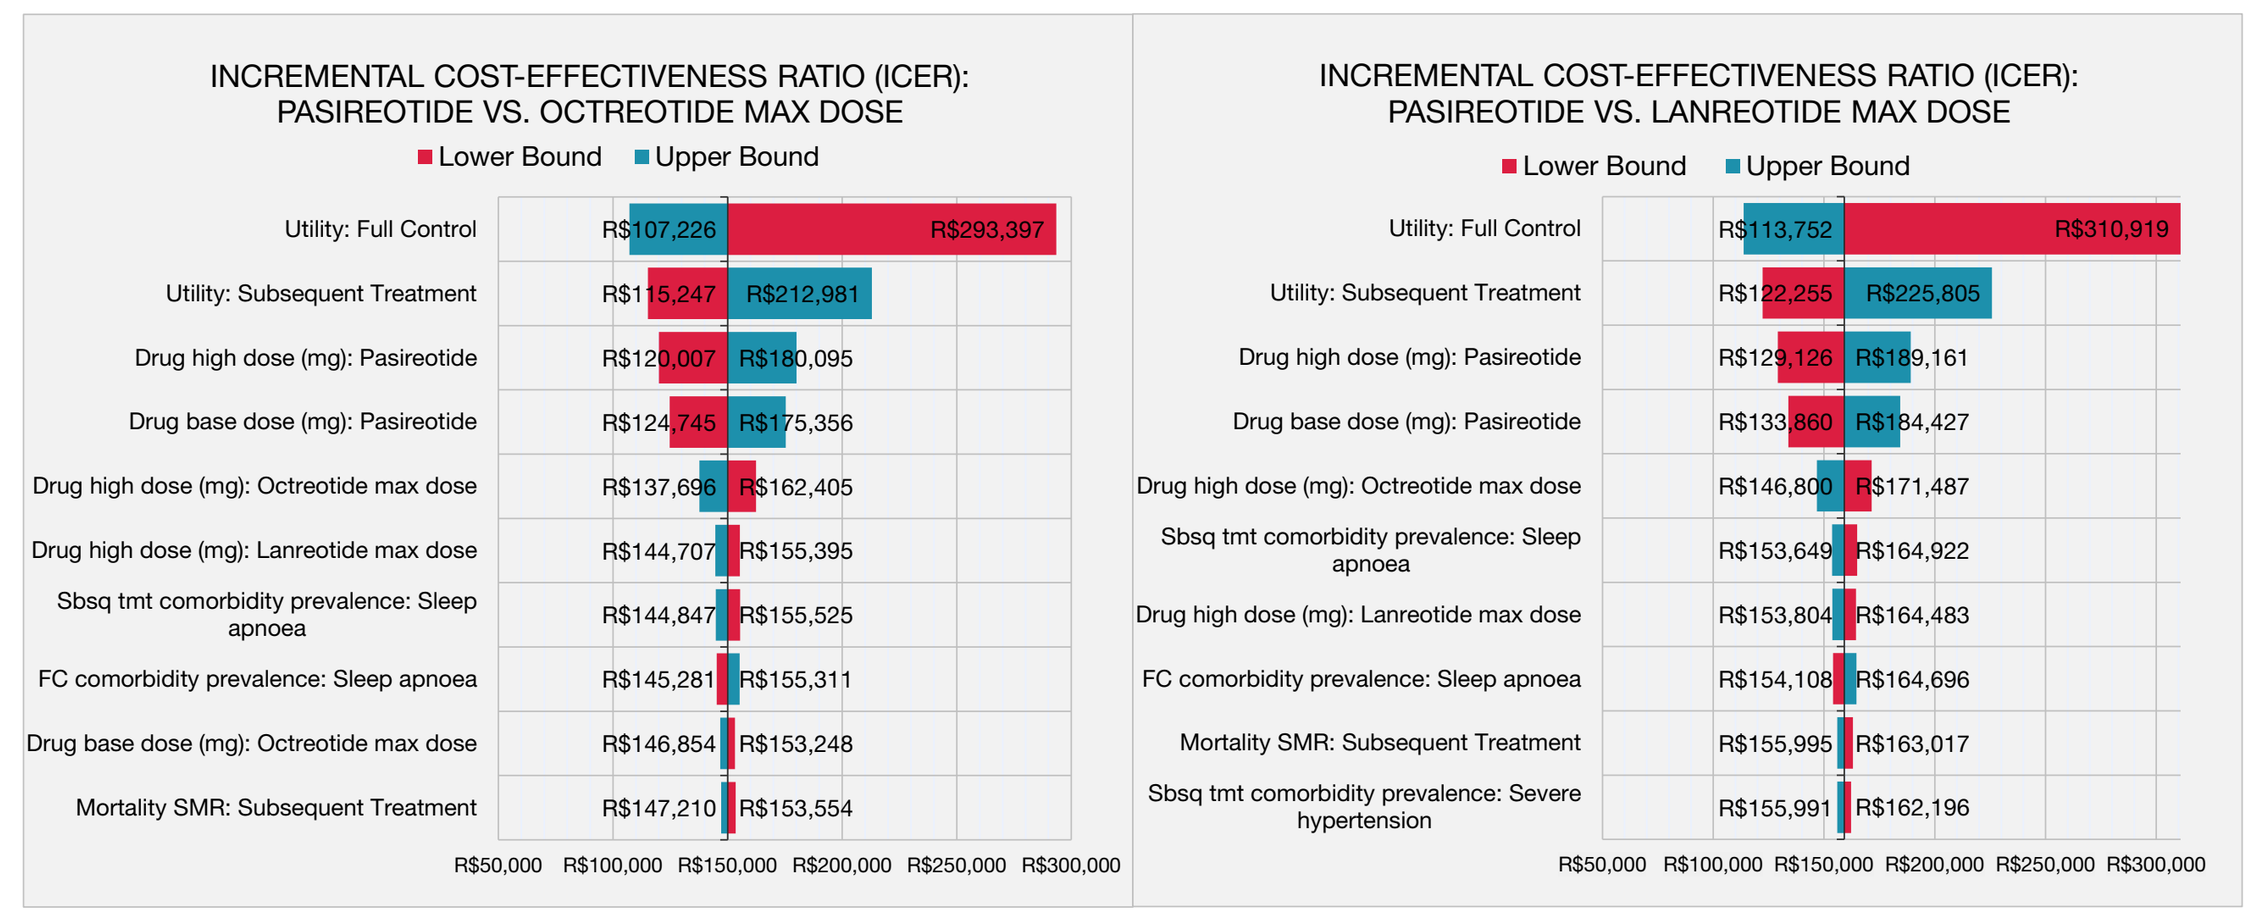
INCREMENTAL COST-EFFECTIVENESS RATIO (ICER): PASIREOTIDE VS. OCTREOTIDE MAX DOSE
Lower Bound
Upper Bound
Utility: Full Control R$107,226 R$293,397
Utility: Subsequent Treatment R$115,247 R$212,981
Drug high dose (mg): Pasireotide R$120,007 R$180,095
Drug base dose (mg): Pasireotide R$124,745 R$175,356
Drug high dose (mg): Octreotide max dose R$137,696 R$162,405
Drug high dose (mg): Lanreotide max dose R$144,707 R$155,395
Sbsq tmt comorbidity prevalence: Sleep apnoea R$144,847 R$155,525
FC comorbidity prevalence: Sleep apnoea R$145,281 R$155,311
Drug base dose (mg): Octreotide max dose R$146,854 R$153,248
Mortality SMR: Subsequent Treatment R$147,210 R$153,554
R$50,000 R$100,000 R$150,000 R$200,000 R$250,000 R$300,000
INCREMENTAL COST-EFFECTIVENESS RATIO (ICER): PASIREOTIDE VS. LANREOTIDE MAX DOSE
Lower Bound
Upper Bound
Utility: Full Control R$113,752 R$310,919
Utility: Subsequent Treatment R$122,255 R$225,805
Drug high dose (mg): Pasireotide R$129,126 R$189,161
Drug base dose (mg): Pasireotide R$133,860 R$184,427
Drug high dose (mg): Octreotide max dose R$146,800 R$171,487
Sbsq tmt comorbidity prevalence: Sleep apnoea R$153,649 R$164,922
Drug high dose (mg): Lanreotide max dose R$153,804 R$164,483
FC comorbidity prevalence: Sleep apnoea R$154,108 R$164,696
Mortality SMR: Subsequent Treatment R$155,995 R$163,017
Sbsq tmt comorbidity prevalence: Severe hypertension R$155,991 R$162,196
R$50,000 R$100,000 R$150,000 R$200,000 R$250,000 R$300,000

FC = full control; SMR = standardized mortality ratio; Sbsq = subsequent; Tmt = treatment

Figure 3. Cost-effectiveness Scatterplots

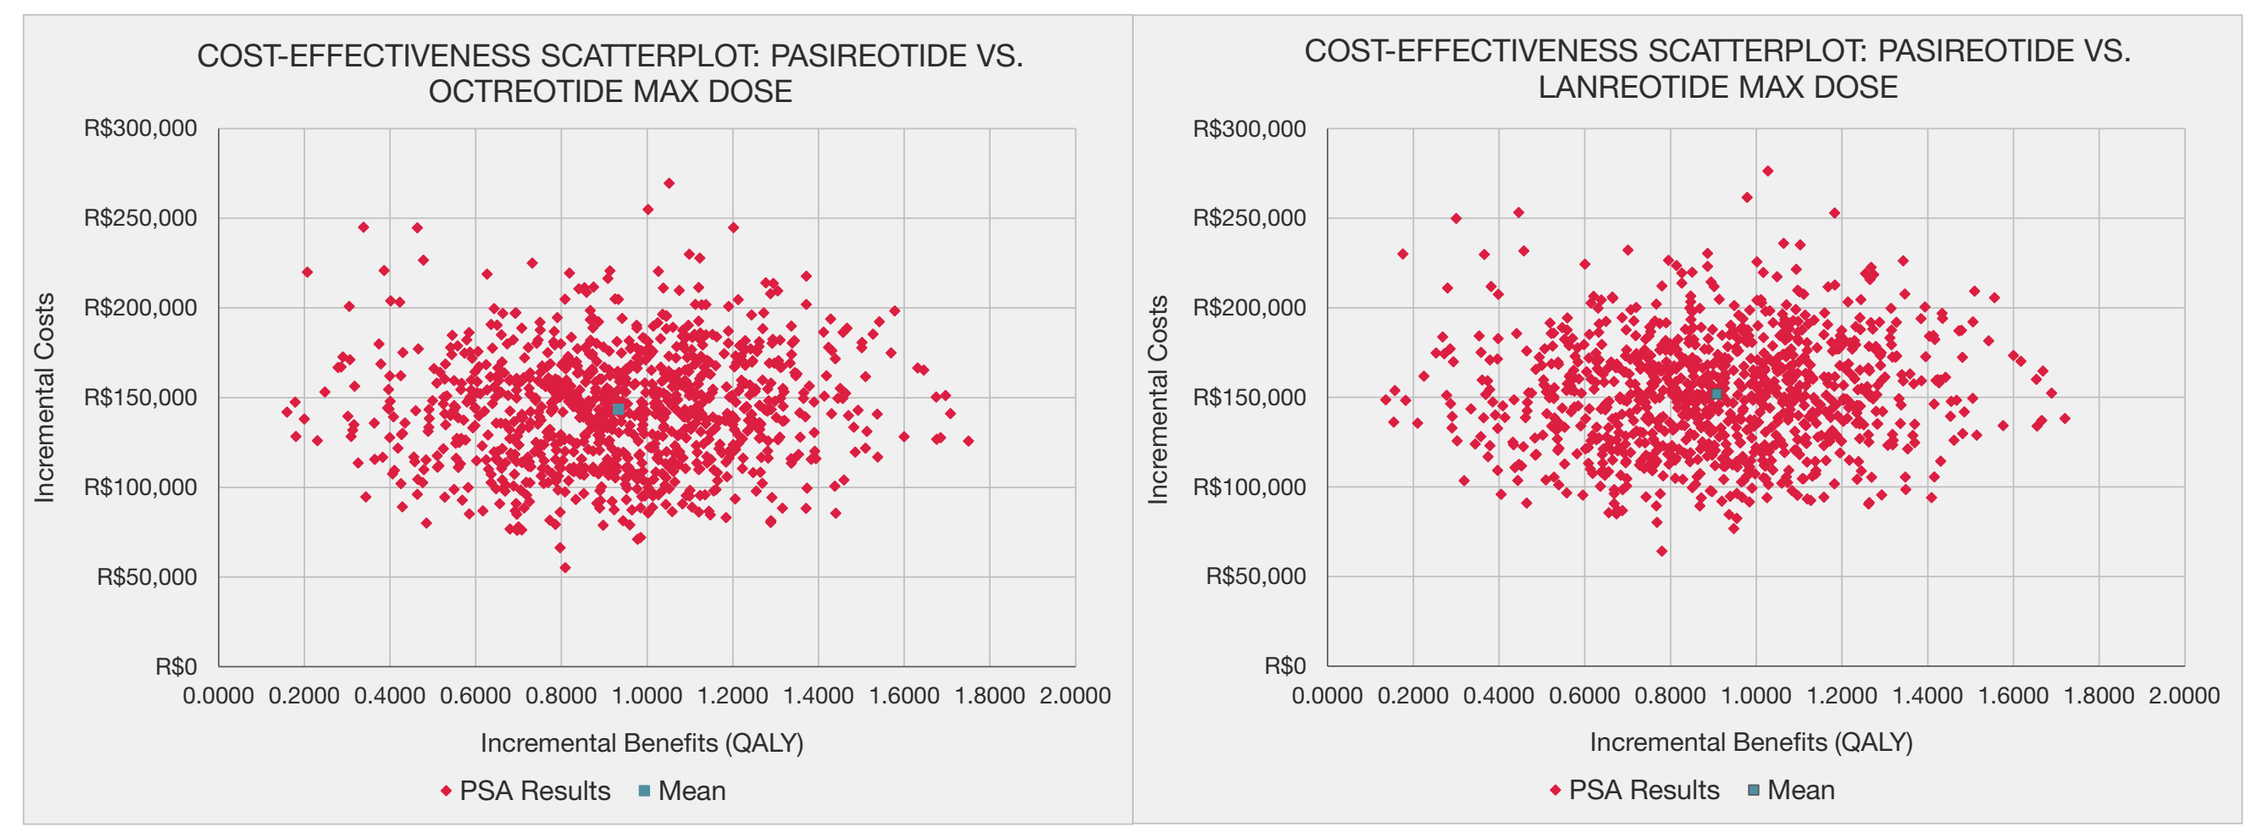
COST-EFFECTIVENESS SCATTERPLOT: PASIREOTIDE VS. OCTREOTIDE MAX DOSE
COST-EFFECTIVENESS SCATTERPLOT: PASIREOTIDE VS. LANREOTIDE MAX DOSE
Incremental Costs
R$300,000
R$250,000
R$200,000
R$150,000
R$100,000
R$50,000
R$0
0.0000 0.2000 0.4000 0.6000 0.8000 1.0000 1.2000 1.4000 1.6000 1.8000 2.0000
Incremental Benefits (QALY)
PSA Results
Mean

PSA = probabilistic sensitivity analysis; QALY = quality-adjusted life year

- In a scenario with IGF-1 normalization as the only biochemical control criterion, pasireotide was dominant versus pegvisomant and pegvisomant + lanreotide, but less costly (–R$855,724) and less effective (–0.036 QALYs) versus pegvisomant + octreotide (**Table 7**). The results versus SRLs were comparable to the base case.
- The results of the scenario assuming that some patients can lose the effect of second-line treatment (despite FC or PC) are similar to the base case. Pasireotide was associated with higher incremental QALYs (0.74), resulting in ICER of R$116,620 and R$128,358 per QALY vs. maximum doses of octreotide and lanreotide, respectively.
- A scenario that applied efficacy data with the 1.0 μg/l GH cut-off from an extension to the PAOLA study[9] resulted in R$158,105 and R$178,893 cost per QALY vs. octreotide and lanreotide, respectively.

Table 7. Model Results. Scenario - IGF-1 is Biochemical Control Criteria

Incremental Results vs. Pasireotide	Octreotide max dose	Lanreotide max dose	Pegvisomant	Pegvisomant + Octreotide	Pegvisomant + Lanreotide
Incremental LYs	0.3757	0.3757	0.0494	-0.0130	0.0289
Incremental QALYs	1.2298	1.2307	0.1688	-0.0357	0.1005
Incremental Costs	R$184,029	R$192,765	-R$440,543	-R$855,724	-R$673,342
ICER per LY	R$489,774	R$513,024	PAS is dominant	PAS is less costly, less effective	PAS is dominant
ICER per QALY	R$149,637	R$156,636	PAS is dominant	PAS is less costly, less effective	PAS is dominant

AE = adverse event; ICER = incremental cost-effectiveness ratio; LY = life year; PAS = pasireotide; QALY = quality-adjusted life year

Conclusions

- **The CEM population comprised patients who were already treated with maximum dose of first-generation SRLs (octreotide and lanreotide) and remained with uncontrolled disease. Treatment with pasireotide resulted in improved health outcomes (LYs and QALYs) and increased cost compared to monotherapy treatments with maximal doses of octreotide and lanreotide.**
- **The high burden of disease reflected in a high standardized mortality ratio relative to the general population and comorbidities that are prevalent among patients with acromegaly reveal the importance of addressing medical needs of patients with uncontrolled disease. Clinical trial outcomes show that pasireotide provides significant health benefits to these patients. With limited options for second-line medical treatment in Brazil, the reimbursement of pasireotide should be considered cost-effective.**

References

1. Melmed S. *J Clin Invest.* 2009;119(11):3189-3202.
2. Carmichael JD, et al. *BMC Endocr Disord.* 2017;17(1):15.
3. Colao A, et al. *Eur J Endocrinol.* 2011;165(5):713-721.
4. Davi MV, et al. *Eur J Endocrinol.* 2008;159(5):533-540.
5. Whittington MD, et al. *Growth Horm IGF Res.* 2021;59:101389.
6. Katznelson L, et al. *J Clin Endocrinol Metab.* 2014;99(11):3933-3951.
7. Melmed S, et al. *Nat Rev Endocrinol.* 2018;14(9):552-561.
8. Giustina A, et al. *J Endocrinol Invest.* 2017;40(6):577-589.
9. Colao A, et al. *Eur J Endocrinol.* 2020 Jun;182(6):583. doi: 10.1530/EJE-19-0762. PMID: 32217809; PMCID: PMC7222286.
10. Ministério da Saúde. https://bvsms.saude.gov.br/bvs/publicacoes/diretrizes_metodologicas_diretriz_avaliacao_economica.pdf. Accessed May, 2022.
11. Gadelha MR, et al. *Lancet Diabetes Endocrinol.* 2014;2(11):875-884.
12. IMS Health. Data on File: Indirect treatment comparison. 2014.
13. Trainer PJ, et al. *Clin Endocrinol (Oxf).* 2009;71(4):549-557.
14. Andries M, et al. *Clin Endocrinol (Oxf).* 2008;68(3):473-480.
15. Jenkins PJ, et al. *J Clin Endocrinol Metab.* 2006;91(4):1239-1245.
16. Bolfi F, et al. *Eur J Endocrinol.* 2018;179(1):59-71.
17. Sherlock M, et al. *J Clin Endocrinol Metab.* 2009;94(11):4216-4223.
18. Liu S, et al. *J Investig Med.* 2018;66(3):653-660.
19. Kyriakakis N, et al. *Clin Endocrinol (Oxf).* 2017;86(6):806-815.
20. Shiroiwa T, et al. *Value Health.* 2021;24(8):1193-1202.
21. Pasireotide. Study CSOM230C2305. Full Clinical Study Report. 2012. After crossover data. Data on File
22. An Z, et al. *BMC Endocr Disord.* 2020;20(1):57.
23. Colao A, et al. *Endokrynol Pol.* 2019;70(4):305-312.
24. Leonart LP, et al. *Endocrine.* 2019;63(1):18-26.
25. Trainer PJ, et al. *N Engl J Med.* 2000;342(16):1171-1177.
26. Harris S, et al. *Can J Diabetes.* 2014;38(1):45-52.
27. Drug unit prices. https://portaldatransparencia.gov.br/licitacoes/consulta. Accessed May, 2022.
28. Agência Nacional de Vigilância Sanitária (ANVISA). http://antigo.anvisa.gov.br/en/consulta-lista-de-preco-de-medicamento. Accessed May 18, 2022.
29. Ministério da Saúde. https://bvsms.saude.gov.br/bvs/publicacoes/protocolo_clinico_diretrizes_terapeuticas_acromegalia.pdf. Accessed May 18, 2022.
30. Fleseriu M, et al. *Eur J Endocrinol.* 2021;185(4):525-538.
31. Strasburger CJ, et al. *Eur J Endocrinol.* 2018;178(4):321-329.
32. Ministério da Saúde. http://sigtap.datasus.gov.br/tabela-unificada/app/sec/inicio.jsp. Accessed February 8, 2022.
33. Katznelson L, et al. *Endocr Pract.* 2011;17 Suppl 4:1-44.
34. Melmed S, et al. *Pituitary.* 2013;16(3):294-302.
35. Silverstein JM. *Endocr Connect.* 2015;4(4):R59-67.
36. Zhao Q, et al. *Innovation (Camb).* 2020;1(1):100013.

Disclosures and Acknowledgments

Disclosures: Study funded by Recordati Rare Diseases. DT, and ET are employees of Evidera, a business unit of ThermoFisher, which has received payments from Recordati Rare Diseases for services related to this poster. HP and FS are employees and shareholders of Recordati Rare Diseases. AK is an employee of MAHTA INTL.

Acknowledgements: The authors would like to thank Michael Grossi and Kawthar Nakayima of Evidera, a business unit of ThermoFisher, for their editorial and graphic design contributions.

Presented at ISPOR US • May 7–10, 2023 • Boston, MA, US

Funding provided by Recordati Rare Diseases