Meaningful Change Thresholds in Patient-reported Outcome Scores for Patients With Colorectal Cancer — A Systematic Literature Review

PCR117

Ziyan Chen[1], Julien Taieb[2], Marko Rehn[3], Christian Eichinger[4], Istvan Majer[5]

[1]Amgen Inc., Tampa, FL, USA; [2]Georges Pompidou European Hospital, Paris, France; [3]Amgen Inc, Thousand Oaks, CA, USA; [4]Oxford PharmaGenesis, Oxford, UK; [5]Amgen (Europe) GmbH, Rotkreuz, Switzerland

BACKGROUND

- Worldwide, colorectal cancer (CRC) is the third most common type of cancer and a leading cause of cancer-related death[1]
- Kirsten rat sarcoma viral oncogene homolog (KRAS) glycine 12 to cysteine (p.G12C) mutation has been identified as a putative oncogenic driver in several types of solid tumor. The KRAS p.G12C mutation is estimated to occur in approximately 3% of patients with CRC[2-3]
- The prognostic significance of KRAS p.G12C status in mCRC is not established with certainty. Some studies suggest that KRAS p.G12C is associated with decreased survival compared with other KRAS mutations or KRAS wild-type disease,[2-6] while others have shown that it is not an independent prognostic factor of patient outcome[7-9]
- In the ongoing phase 3 CodebreaK 300 study, the efficacy and safety of sotorasib and panitumumab is compared with investigator's choice (trifluridine/tipiracil, or regorafenib) in previously treated mCRC patients with KRAS p.G12C mutation. Patients' health-related quality of life (HRQoL) is assessed by several instruments including the European Organisation for Research and Treatment of Cancer Core 30-item Quality of Life Questionnaire (EORTC QLQ-C30), Brief Pain Inventory (BPI), Brief Fatigue Inventory (BFI), overall symptom bother as measured by the Functional Assessment of Cancer Therapy General (FACT-G) questionnaire, EuroQol's EQ-5D 5-level instrument, and the Patient Global Impression of Change (PGIC) scale

OBJECTIVE

- Data on HRQoL evidence in patients with CRC are scarce. This systematic literature review (SLR) aimed to identify thresholds used to evaluate meaningful changes in patient-reported outcome (PRO) scores at the item, domain, or total score level in patients with CRC

METHODS

- Data sources and search strategy: Electronic searches were performed on the Ovid® platform in 3 databases (Embase, MEDLINE, and the Cochrane Library) covering full publications published since 2001 and conference abstracts since 2020. In addition, hand searches were performed to identify evidence from clinical trial registries and proceedings of major clinical (e.g., American Society of Clinical Oncology) and health economic congresses (e.g., International Society of Pharmacoeconomics and Outcomes Research)
- Citation Screening: Titles and abstracts of identified publications were screened against the predefined eligibility criteria by a single reviewer. Congress abstracts were screened by a single reviewer
- Full Text Review: Full texts of publications considered relevant at title and abstract screening were reviewed against the same eligibility criteria by a single reviewer
- Data Extraction and quality assessment: One person extracted data from included articles and a second person checked each data point for accuracy. The quality of the included studies was assessed by a single reviewer using validated tools

Table 1. Study Eligibility Criteria

PICOT	Inclusion Criteria	Exclusion Criteria
Population	Adult patients with CRC	N/A
Interventions/comparators	Any	N/A
Outcomes	Metrics of meaningful change for • within-patient • within-group, and • between-group differences. Thresholds used at the item, domain, or total score level for PRO instruments	Studies not mentioning a metric of clinically meaningful change or an indication that MID was investigated
Study types	Interventional studies Observational/RWE studies	Preclinical studies, case series and reports, conference abstracts
Time horizon	Published since 2001 for database searches	N/A

CRC: colorectal cancer; MID: minimally important difference; N/A: not applicable; PICOT: patient, intervention, comparator, outcomes, time horizon; PRO: patient-reported outcomes; RWE: real-world evidence

RESULTS

Study selection

- Figure 1 presents the PRISMA flow diagram. In total, 70 studies met the inclusion/exclusion criteria (21 interventional studies, 49 observational studies)
 - In total, 21 publications analyzed PRO data that included patients with mCRC
 - KRAS mutation status was reported in 2 publications; no study reported results stratified by KRAS mutation status and/or disease stage

Figure 1. PRISMA Flow Diagram

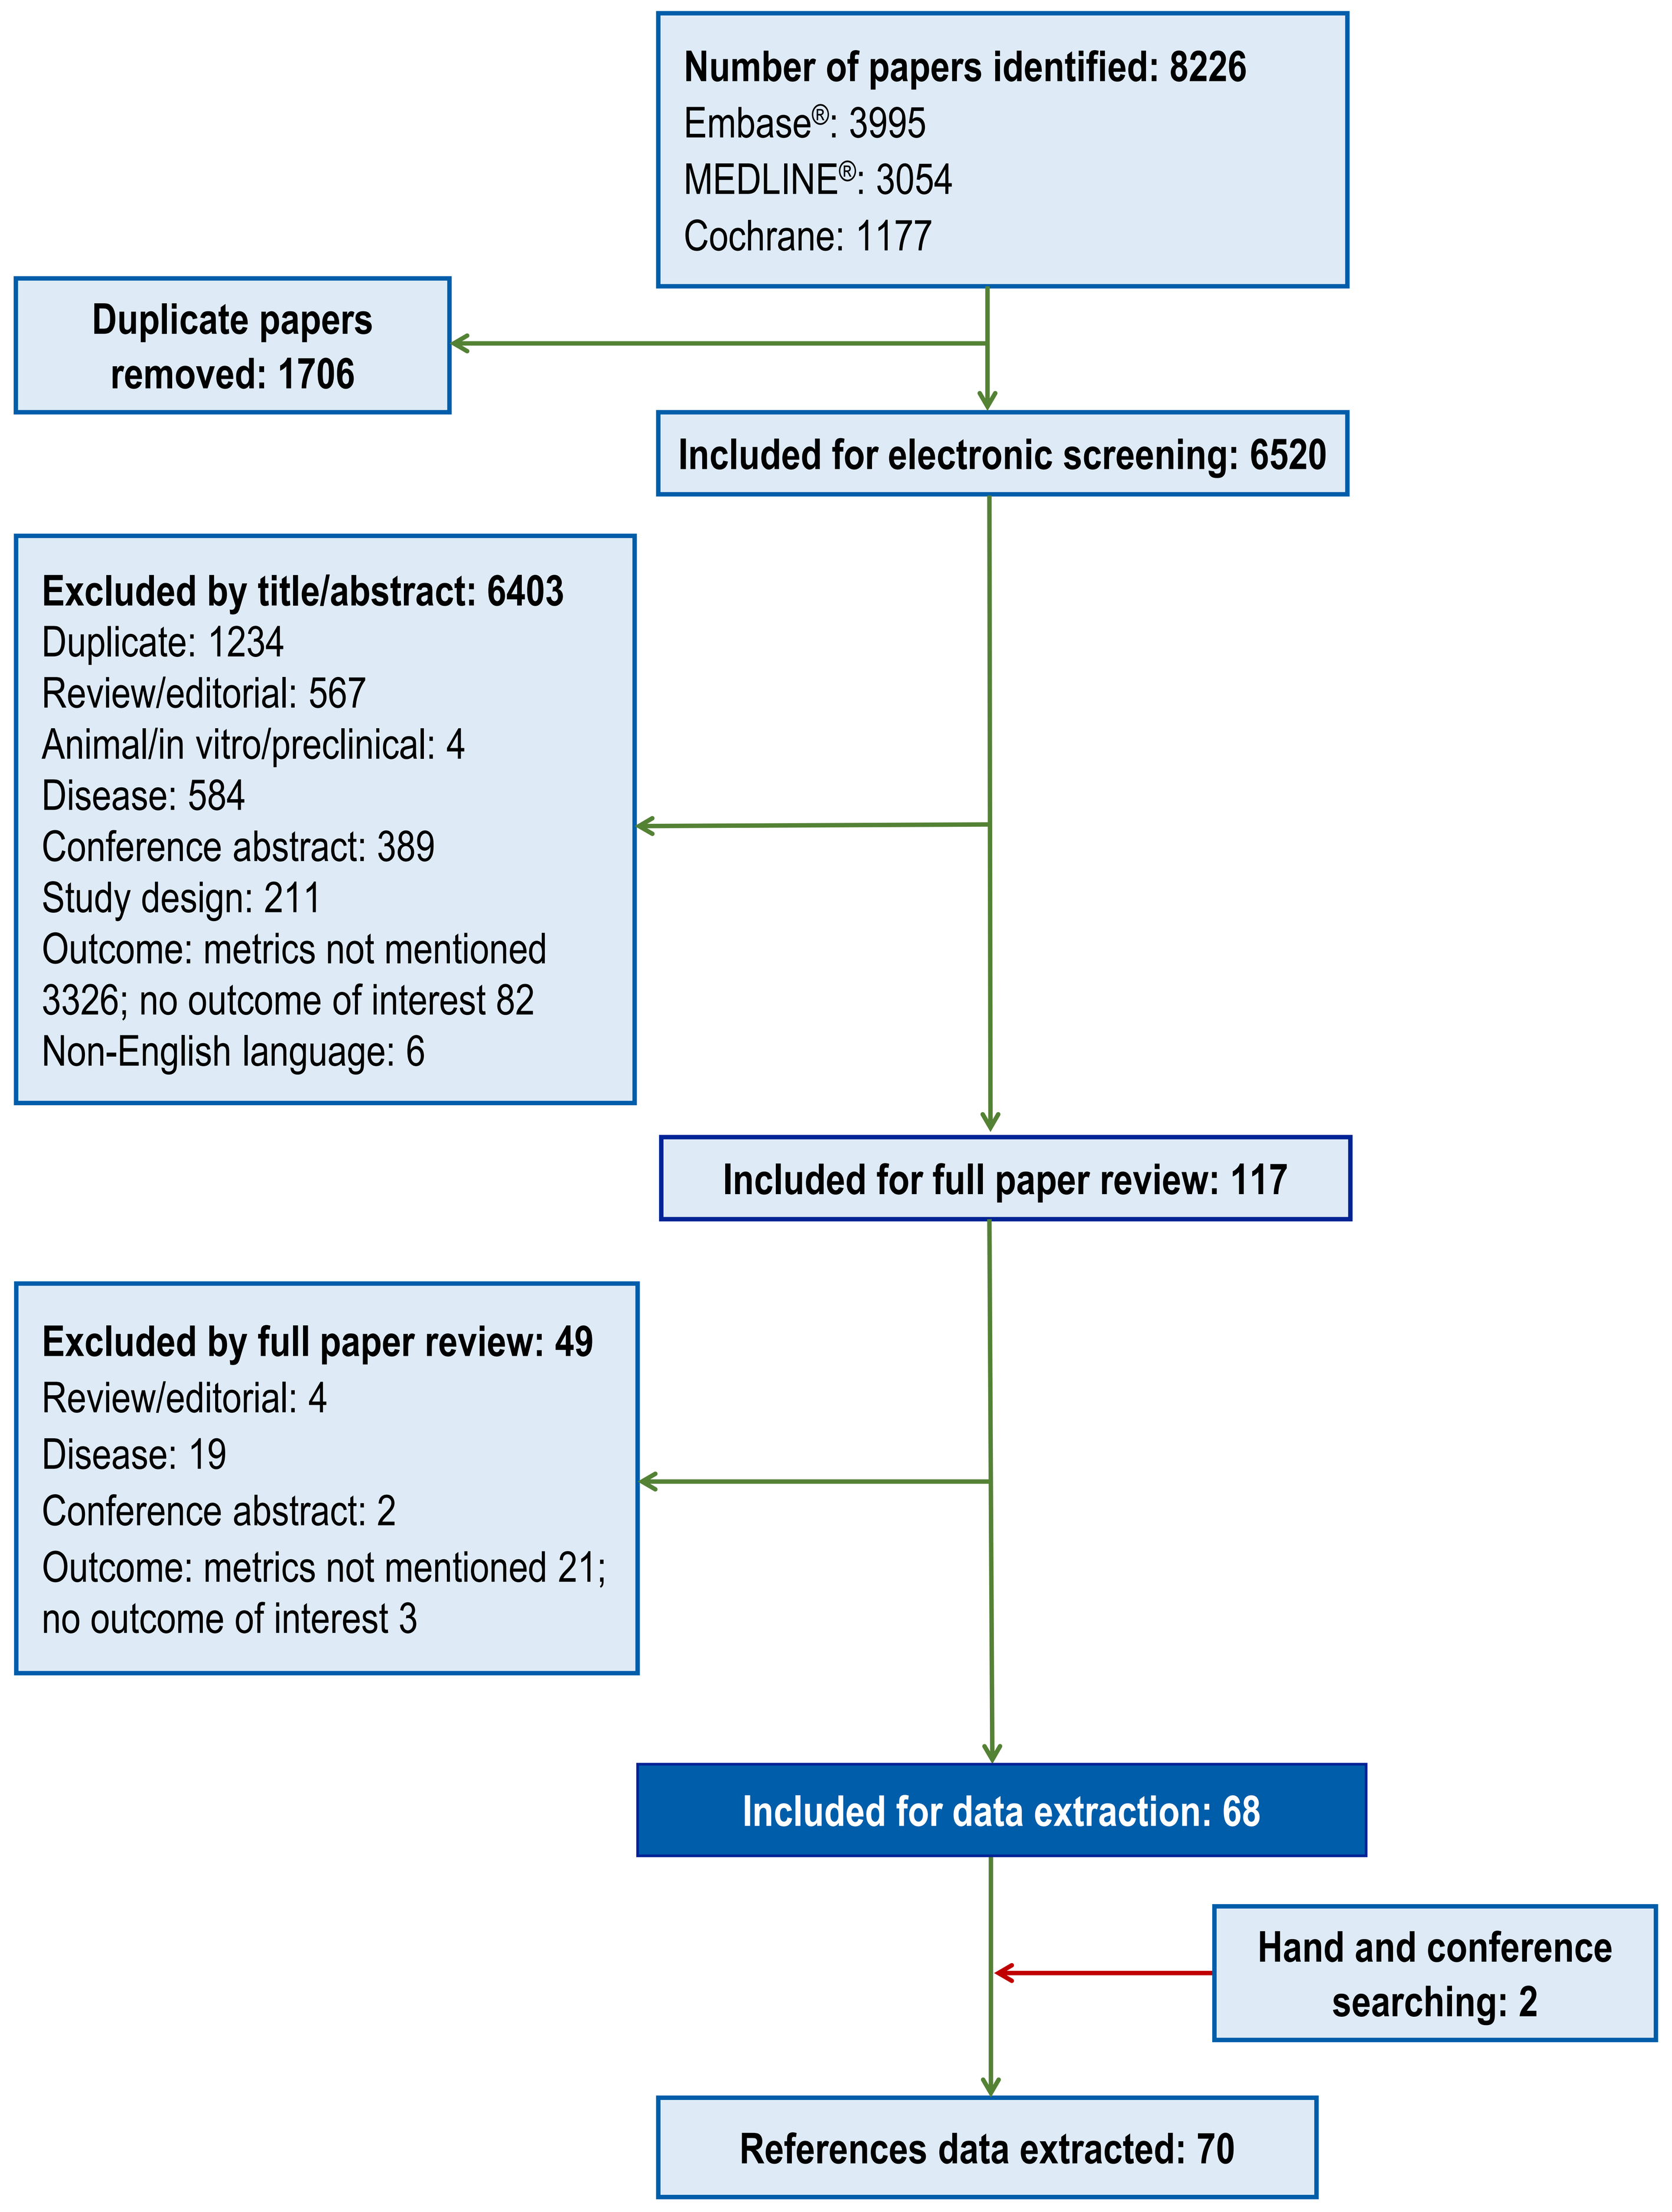

Number of papers identified: 8226
Embase®: 3995
MEDLINE®: 3054
Cochrane: 1177
Duplicate papers removed: 1706
Included for electronic screening: 6520
Excluded by title/abstract: 6403
Duplicate: 1234
Review/editorial: 567
Animal/in vitro/preclinical: 4
Disease: 584
Conference abstract: 389
Study design: 211
Outcome: metrics not mentioned 3326; no outcome of interest 82
Non-English language: 6
Included for full paper review: 117
Excluded by full paper review: 49
Review/editorial: 4
Disease: 19
Conference abstract: 2
Outcome: metrics not mentioned 21; no outcome of interest 3
Included for data extraction: 68
Hand and conference searching: 2
References data extracted: 70

PRISMA: Preferred Reporting Items for Systematic reviews and Meta-Analyses

PRO instruments used

- Based on the identified literature, the EORTC quality of life questionnaires were most often used in studies reporting meaningful changes in PRO scores
 - EORTC QLQ Core 30-item questionnaire (C30) in 34 studies
 - EORTC QLQ Colorectal Cancer 38-item module (CR38) in 6 studies
 - EORTC QLQ Colorectal Cancer 29-item module (CR29) in 2 studies
 - EORTC QLQ Chemotherapy-Induced Peripheral Neuropathy 20-item module (CIPN20) in 4 studies
 - EORTC QLQ Colorectal Liver Metastases 21-item module (LMC21) in 2 studies
- The FACT questionnaires were frequently used although less often than the EORTC QLQ-C30 questionnaire
 - FACT-General (FACT-G) in 7 publications
 - FACT-Colorectal (FACT-C) in 11 publications
 - FACT Colorectal Cancer Symptom Index 9-item scale (FCSI-9) in 2 publications
- The use of EuroQol's Questionnaire 5 dimensions (EQ-5D) instruments were reported in 9 publications
- Outcomes from the PGI-C were reported in 1 publication
- Table 2 presents the frequency of the most often used PRO instruments by study type for studies including all CRC and mCRC patients

Table 2. Most Frequently Used PRO Instruments in the Included Studies by Stage of the Disease and Study Type

Instrument	All CRC			mCRC		
	Total	Interventional	Observational	Total	Interventional	Observational
EORTC QLQ-C30	34	14	20	16	11	5
EQ-5D	9	5	4	5	4	1
FACT-C	11	3	8	0	0	0

CRC: colorectal cancer; EORTC QLQ-C30: European Organisation for Research and Treatment of Cancer Core 30-item Quality of Life Questionnaire; EQ-5D: EuroQoL Questionnaire 5 dimensions; FACT-C: Functional Assessment of Cancer Therapy-Colorectal; mCRC, metastatic colorectal cancer; PRO: patient-reported outcome.

Overview of applied thresholds

- The most often (35%) reported threshold for EORTC instruments was 10 points for all modules, domains, and subscales
- The reported range of thresholds was 5-9 points for the FACT-C total score and 1–3 points for subscales. The range of thresholds was 3-6 points for the FACT-G total score and 2 points for subscales.
- Reported thresholds for the EQ-5D instruments was 5.48–12 points on the visual analog scale (VAS) and 0.06–0.09 on the utility index value
- Table 3 summarizes the range of reported minimally important difference (MID) values for the most frequently used PRO instruments
- Thresholds were reported from four studies that analyzed PRO data in time-to-deterioration analyses

Table 3. Range of Minimal Important Difference Values for the Most Frequently Reported PRO Instruments in Patients with mCRC

	Possible score	MID range across all studies	MID range across interventional studies	MID range across observational studies
EORTC QLQ-C30	0–100	Domains and symptom scales: 5–10 points	Domains and symptom scales: 5–10 points	Domains and symptom scales: 5–10 points
FACT-C	0–108	Total score: 5–8 points TOI-C: 4–7 points Subscales: 2–3 points	N/A	Total score: 5–8 points TOI-C: 4–7 points Subscales: 2–3 points
EQ-5D VAS	0–100	5.48–12 points	5.48–12 points	7 points
EQ-5D utility value	<0–1	0.06–0.09 points	0.06–0.09 points	0.074 points

EORTC QLQ-C30: European Organisation for Research and Treatment of Cancer Core 30-item Quality of Life Questionnaire; EQ-5D: European Quality of Life Questionnaire 5 dimensions; FACT-C: Functional Assessment of Cancer Therapy-Colorectal; mCRC, metastatic colorectal cancer; MID: minimally important difference; N/A: not applicable; TOI-C: Trial Outcome Index-Colorectal; PRO: patient-reported outcome; VAS: visual analog scale.

KEY FINDINGS

- In total, 70 studies were identified. Among those, 21 studies included patients with mCRC
- EORTC QLQ-C30, FACT-C, EQ-5D, and FACT-G were the most often reported instruments in the identified literature. Time until deterioration was reported in 4 publications
- MID thresholds for PRO scores established previously (not specifically for patients with CRC) were widely used as reference values in interventional and observational studies that included patients with CRC and mCRC
- Across the identified publications for mCRC, the thresholds ranged from 5 to 10 points for EORTC QLQ-C30, 5 to 8 points for the FACT-C total score, 5.48 to 12 points for the EQ-5D VAS, 0.06 to 0.9 for EQ-5D utility index value
- No thresholds were identified for BFI and BPI questionnaires and no publications reporting data for patients with KRAS mutations were identified
- The identified MID thresholds may be used to evaluate and compare HRQoL outcomes in patients with mCRC included in clinical trials. However, more research is needed to evaluate specific thresholds for patients with mCRC and KRAS mutations

Disclosures:

ZC, MR, and IM are employees of Amgen and hold Amgen stocks. CE was paid consultant to conduct the SLR. JT has provided consultancy for Amgen.

This study was supported by Amgen Inc. Medical writing and editorial support was provided by Tim Harrison.

Contact Information:

Ziyan Chen zchen03@amgen.com

References:

1. WHO. 2020 statistics. https://www.who.int/en/news-room/fact-sheets/detail/cancer (accessed May 26, 2021)
2. Fakih M, Tu H, Hsu H et al. *Oncologist*. 2022 Aug 5;27(8):663-674.
3. Hil H, Shivani A, Archana B et al. *Journal of Clinical Oncology.* 2023 41:4_suppl, 41-41
4. Modest DP, Ricard I, Heinemann V et al. *Ann Oncol*. 2016;27:1746-1753.
5. Henry JT, Coker O, Chowdhury S. *JCO Precis Oncol*. 2021;5:613-621.
6. Jones RP, Sutton PA, Evans JP et al. *Br J Cancer*. 2017;116:923-929.
7. Lavacchi D, Fancelli S, Roviello G, et al. *Front Oncol.* 2022 Nov 29;12:1055019.
8. Osterlund E, Ristimäki A, Kytölä S, et al. *Front Oncol*. 2022 Feb 16;12:826073.
9. Lee JK, Sivakumar S, Schrock AB, et al. *NPJ Precis Oncol*. 2022 Dec 9;6(1):91.

© 2023

International Society for Pharmacoeconomics and Outcomes; Boston, MA; May 7-10, 2023