Patient Versus Caregiver and Clinician Reports of Cognitive Difficulties in Patients with Schizophrenia Switching to Long-Acting Injectable Antipsychotic Aripiprazole Lauroxil: A Post Hoc Analysis

Mark G A Opler,[1] Amy Claxton,[2*] James McGrory,[2] Sabina Gasper,[2] Meihua Wang,[2] Sergey Yagoda[2]

[1]WCG MedAvante-ProPhase, Inc., New York, NY, USA; [2]Alkermes, Inc., Waltham, MA, USA

*AC was an employee of Alkermes, Inc., at the time of the study

INTRODUCTION

- Impaired cognition is associated with poor overall functionality in people with schizophrenia[1,2] and is considered an area of unmet treatment need[3]
- Individuals with schizophrenia may have deficits across multiple cognitive domains,[4,5] and these impairments may be exacerbated by side effects associated with antipsychotic medication[6,7]
- When assessing cognition, it is not clear whether patients, clinicians, and caregivers make similar judgments regarding the level of impairment present and which assessments are most related to real-world functioning
- The New York Assessment of Adverse Cognitive Effects of Neuropsychiatric Treatment (NY-AACENT) scale[8] was designed to evaluate the presence and severity of adverse cognitive effects in patients treated with medications, including antipsychotic agents
 - The NY-AACENT scale includes 3 components—a patient form, a clinician form, and a caregiver form—for assessing cognitive impairment in patients
- The NY-AACENT scale was administered in a phase 4 study that enrolled patients with schizophrenia who switched to the long-acting injectable (LAI) antipsychotic aripiprazole lauroxil (AL; Aristada)[9]

OBJECTIVES

- To examine patient, clinician, and caregiver reports of cognitive impairment in patients with schizophrenia switching to AL and to assess the level of agreement between reporters

METHODS

Study Design and Assessments

- This was a post hoc analysis from a 6-month, open-label, prospective, phase 4 study in patients with clinically stable schizophrenia who switched from their prior LAI to AL[9]
- Patients were administered AL (441, 662, or 882 mg monthly or 882 mg every 6 weeks) for up to 6 months
 - The initial AL dose and any subsequent dose adjustments were made according to the investigator's clinical judgment
 - Most commonly prescribed adjunctive psychiatric medications (including ongoing oral antipsychotics) were permitted[9]
- The NY-AACENT scale was administered at baseline (screening visit) and at month 6 or early termination
 - The NY-AACENT scale was completed by patients, their caregivers, and clinicians (study investigators) at each time point
 - The NY-AACENT scale includes 7 domains: Working Memory, Attention/Vigilance, Verbal Learning/Memory, Visual Learning/Memory, Reasoning and Problem Solving, Speed of Processing, and Social Cognition
 - Cognitive difficulty in each domain was rated as not present, mild, moderate, severe, or extreme over the past week

Study Population

- Adult outpatients aged 18–65 years with a diagnosis of schizophrenia according to *Diagnostic and Statistical Manual of Mental Disorders*, Fifth Edition,[10] criteria were eligible for study participation
- Additional eligibility criteria included:
 - Clinical stability, defined as no hospitalizations for acute psychiatric exacerbation within the 2 months prior to screening
 - Brief Psychiatric Rating Scale (BPRS) score between 30 and 45 at screening
 - At least 3 doses of paliperidone palmitate or risperidone LAI before screening, with no antipsychotic medication regimen change in the 4 weeks before study day 1

Statistical Analysis

- The proportions of patients, clinicians, and caregivers who endorsed each categorical response (not present, mild, moderate, severe, extreme) were summarized by individual NY-AACENT domains at baseline and at last assessment
- The level of agreement between groups in ratings of cognitive difficulty was evaluated using weighted kappa coefficients for each NY-AACENT domain at baseline and at last assessment
 - Kappa coefficient values were interpreted as follows: ≤0, no agreement; 0.01–0.20, slight agreement; 0.21–0.40, fair agreement; 0.41–0.60, moderate agreement; 0.61–0.80, substantial agreement; 0.81–1.00, almost perfect agreement[11]

RESULTS

Study Population

- Fifty-one patients were enrolled (**Table 1**); 50 patients were included in this analysis, and 39 completed the NY-AACENT scale at baseline and last assessment

Table 1. Demographics and Baseline Characteristics

Category	All Patients[a] N=50
Age, mean (SD), years	40.4 (11.8)
Sex, male, n (%)	37 (74.0)
Race, n (%)	
Black or African American	25 (50.0)
White	21 (42.0)
Asian	3 (6.0)
Other	1 (2.0)
Body mass index, mean (SD), kg/m²	31.6 (6.6)
CGI-S, mean (SD), kg/m²	3.9 (0.6)
BPRS Total Score, mean (SD)	37.6 (5.8)

[a]N=50, patients included in the post hoc analysis only.
BPRS, Brief Psychiatric Rating Scale; CGI-S, Clinical Global Impressions-Severity.

BPRS Total Score Change Over Time

- Patients were clinically stable, with a mean (SD) baseline BPRS total score of 37.6 (5.8)
- Mean BPRS total scores remained stable over 6 months of AL treatment (**Figure 1**)

Figure 1. Mean (SE) BPRS Total Score Over Time,[a] (N=50)

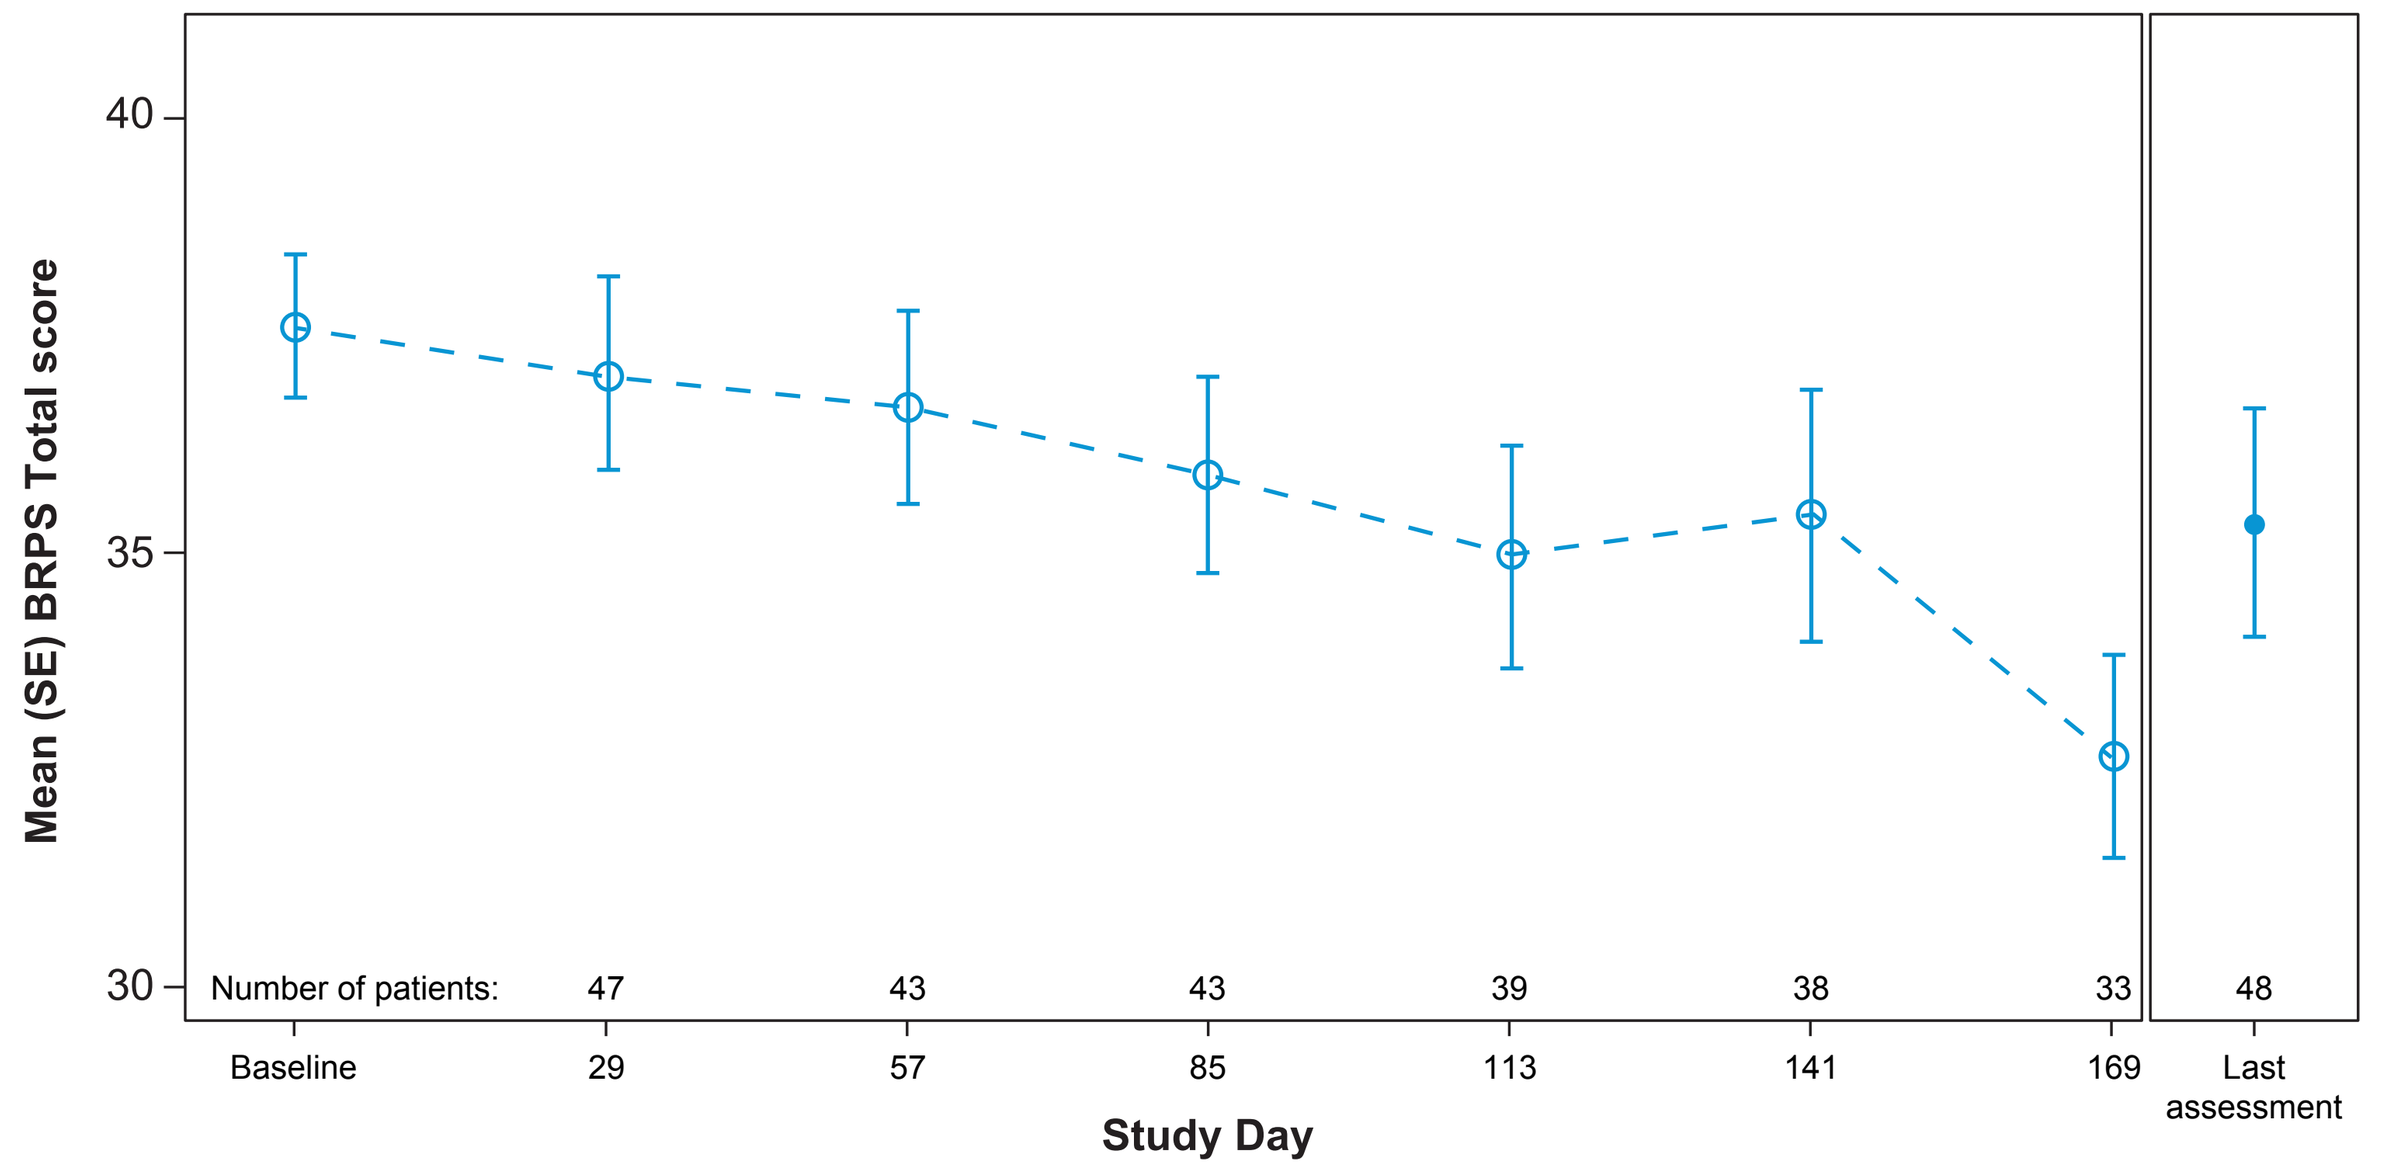
Mean (SE) BRPS Total score
40
35
30
Number of patients: 47 43 43 39 38 33 48
Baseline
29
57
85
113
141
169
Last assessment
Study Day

[a]Based on observed data.
BPRS, Brief Psychiatric Rating Scale; SE, standard error.

- Findings from the Clinical Global Impressions–Severity (CGI-S) scores over time (data not shown) were similar to those from the BPRS scores

NY-AACENT Domain Scores

- At baseline, cognitive difficulties were most commonly reported as 'not present' or 'mild' across all NY-AACENT domains by patients, clinicians, and caregivers; however, there was substantial variation across domains (**Figure 2A–G**)
- There was a shift in the distribution of responses toward 'not present'/'mild' at last assessment for all reporters and across all domains, as illustrated by the shift from blue to green bars in **Figure 2A–G**

Figure 2. NY-AACENT Domain Scores by Patients, Clinicians, and Caregivers; Baseline and Last Assessment

A. Working Memory Impairment

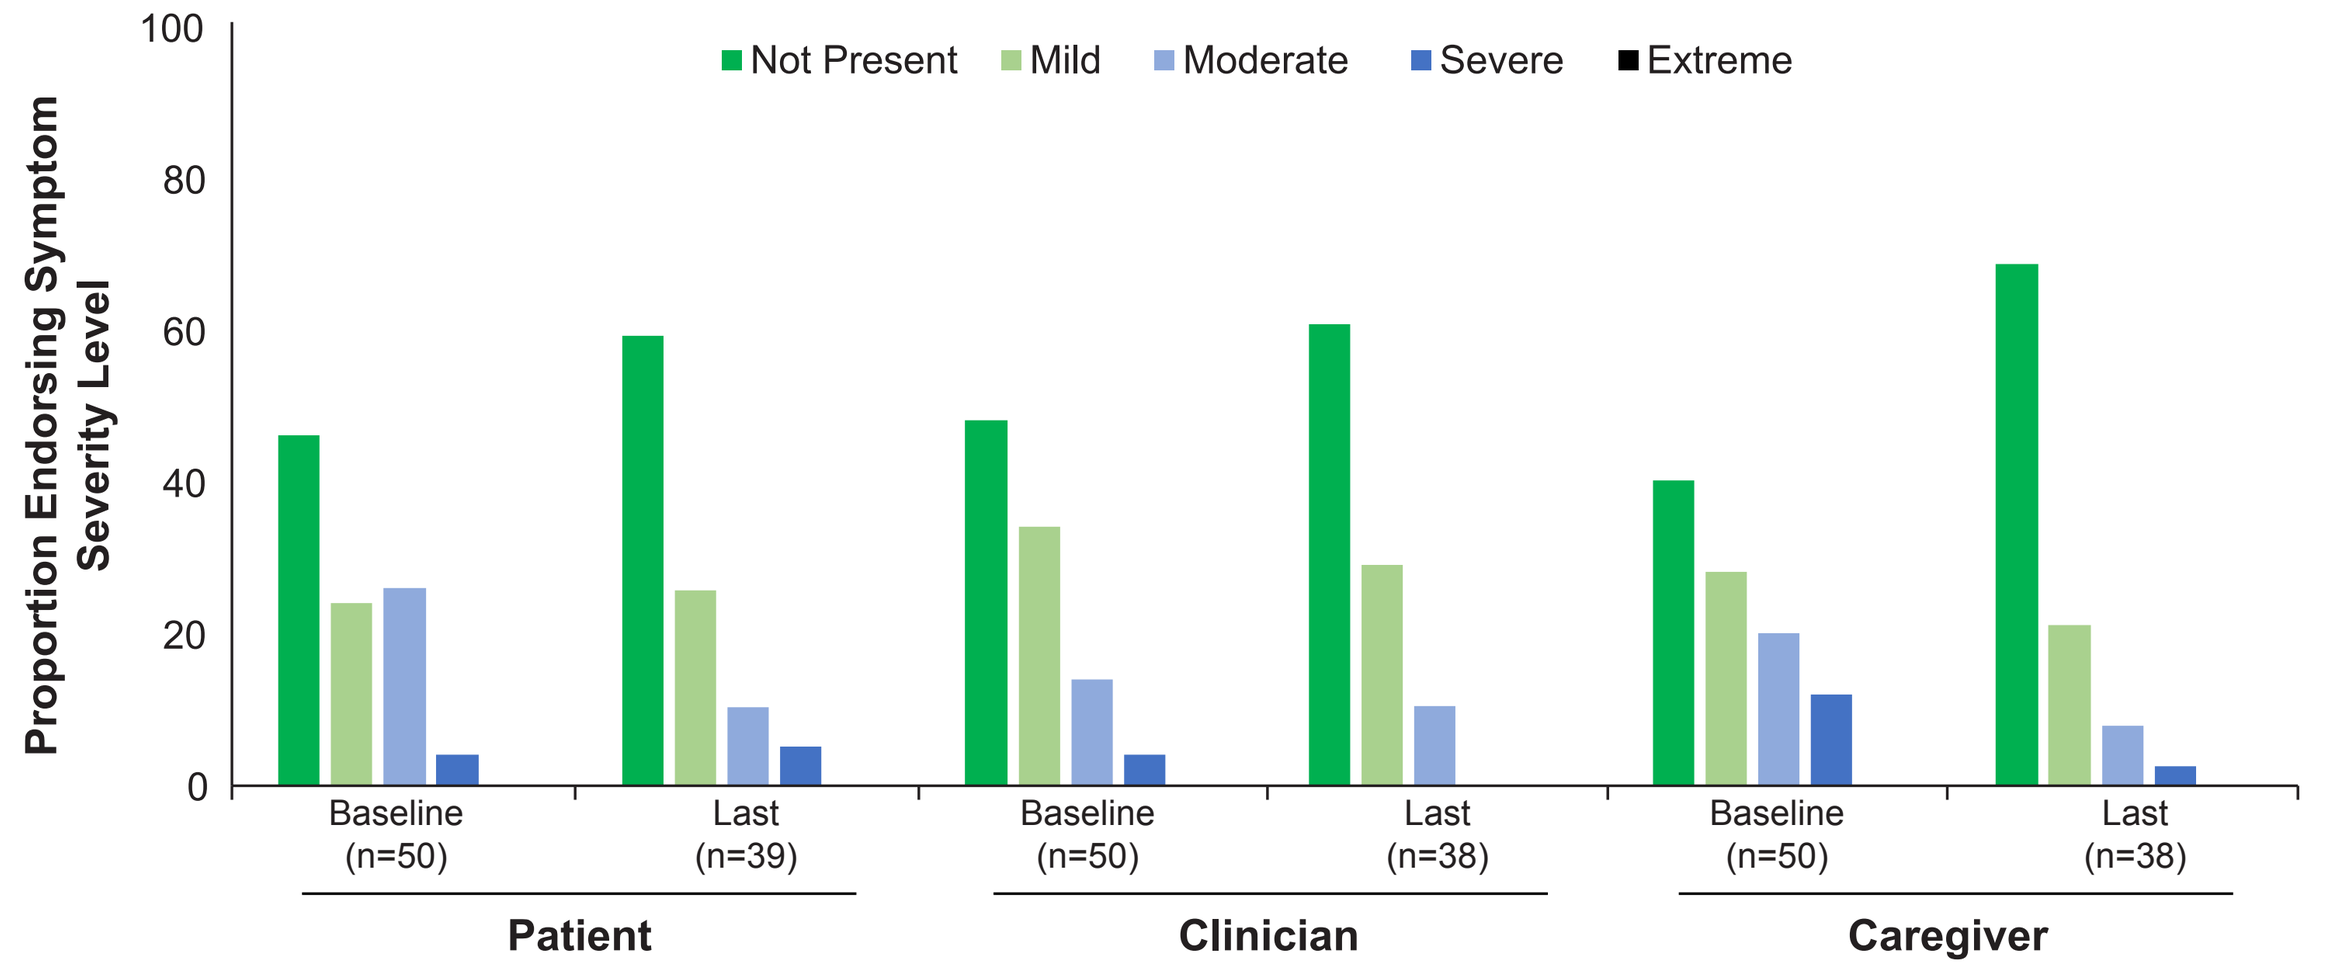
Not Present
Mild
Moderate
Severe
Extreme
Proportion Endorsing Symptom Severity Level
Baseline (n=50)
Last (n=39)
Baseline (n=50)
Last (n=38)
Baseline (n=50)
Last (n=38)
Patient
Clinician
Caregiver

B. Attention/Vigilance Impairment

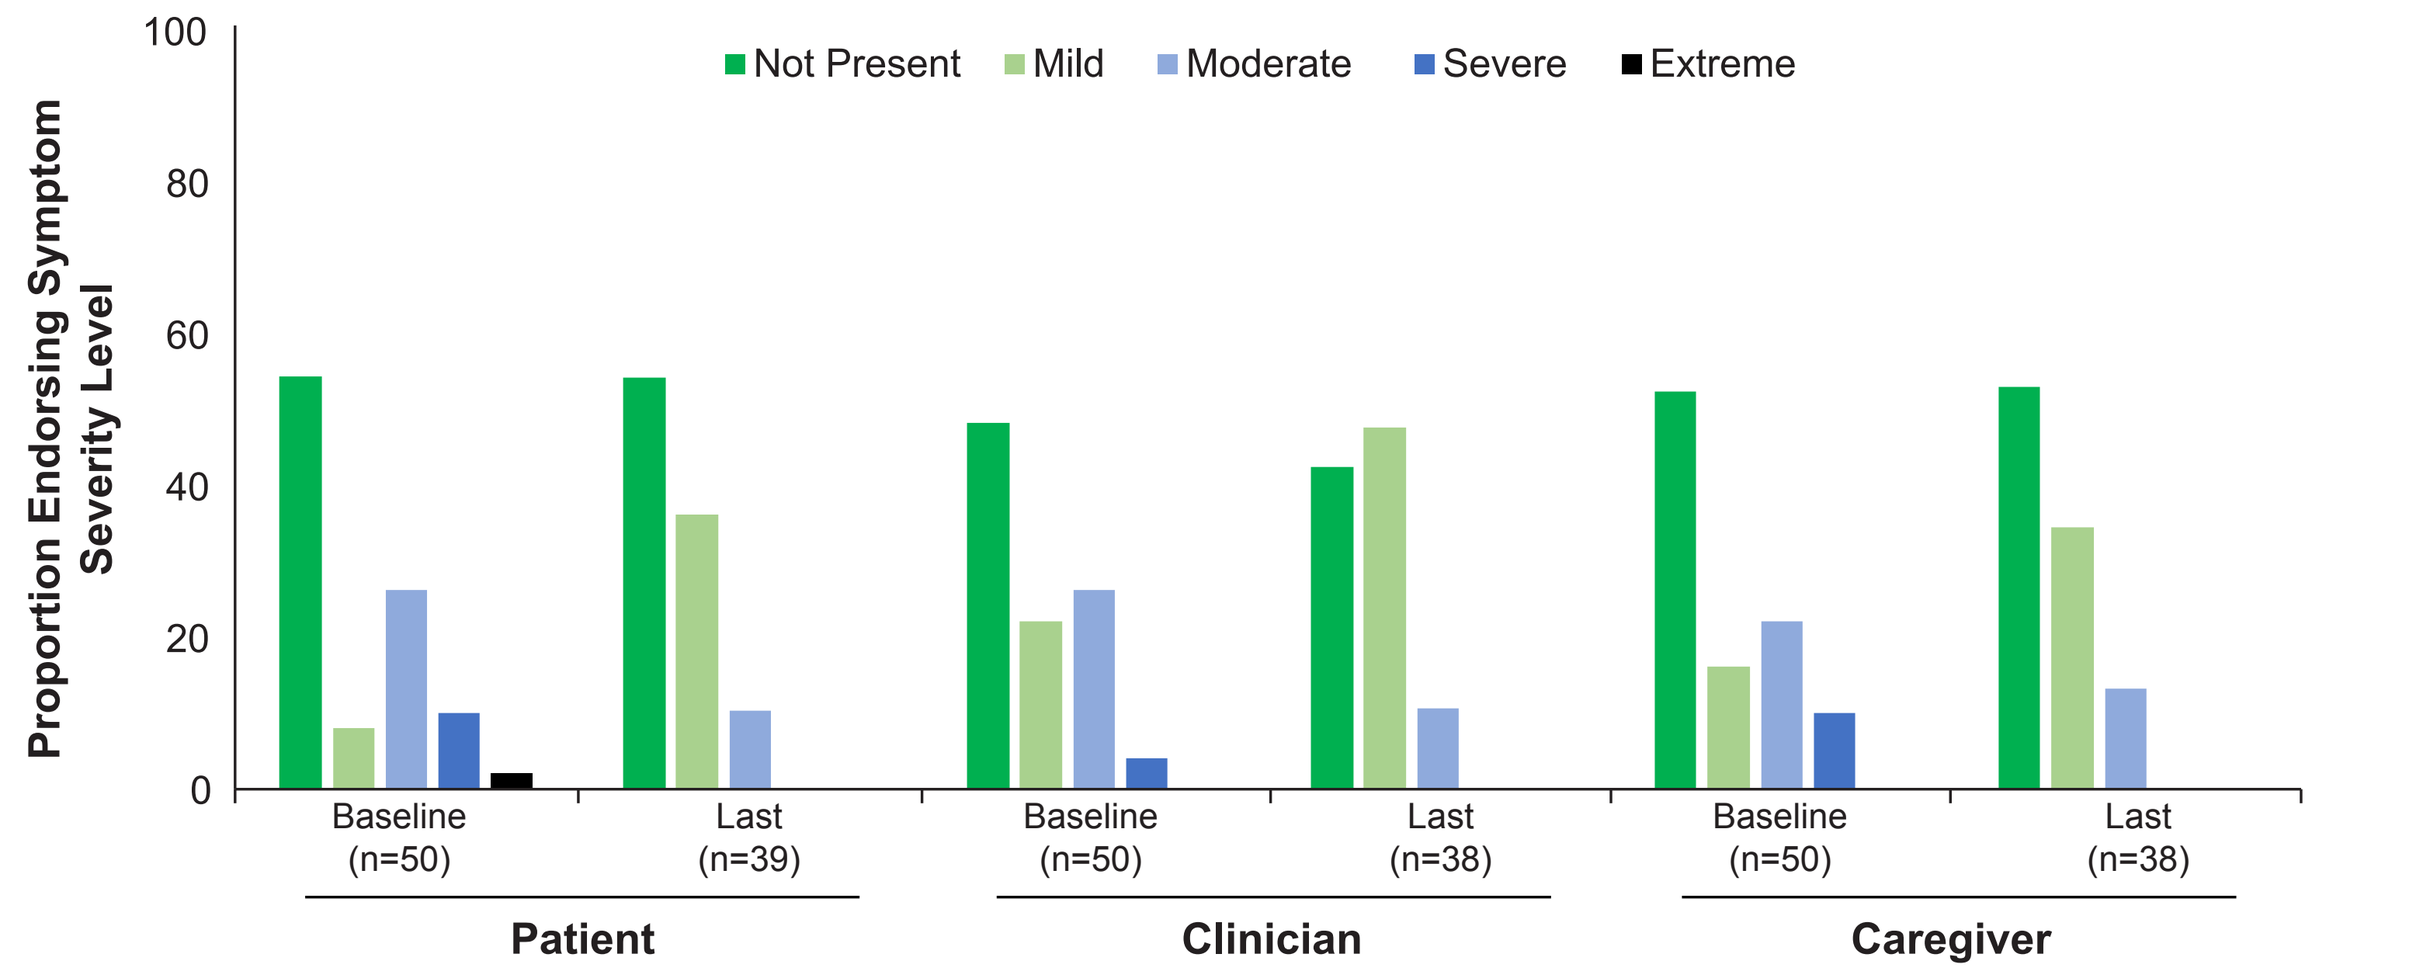
Not Present
Mild
Moderate
Severe
Extreme
Proportion Endorsing Symptom Severity Level
Baseline (n=50)
Last (n=39)
Baseline (n=50)
Last (n=38)
Baseline (n=50)
Last (n=38)
Patient
Clinician
Caregiver

C. Verbal Learning/Memory Impairment

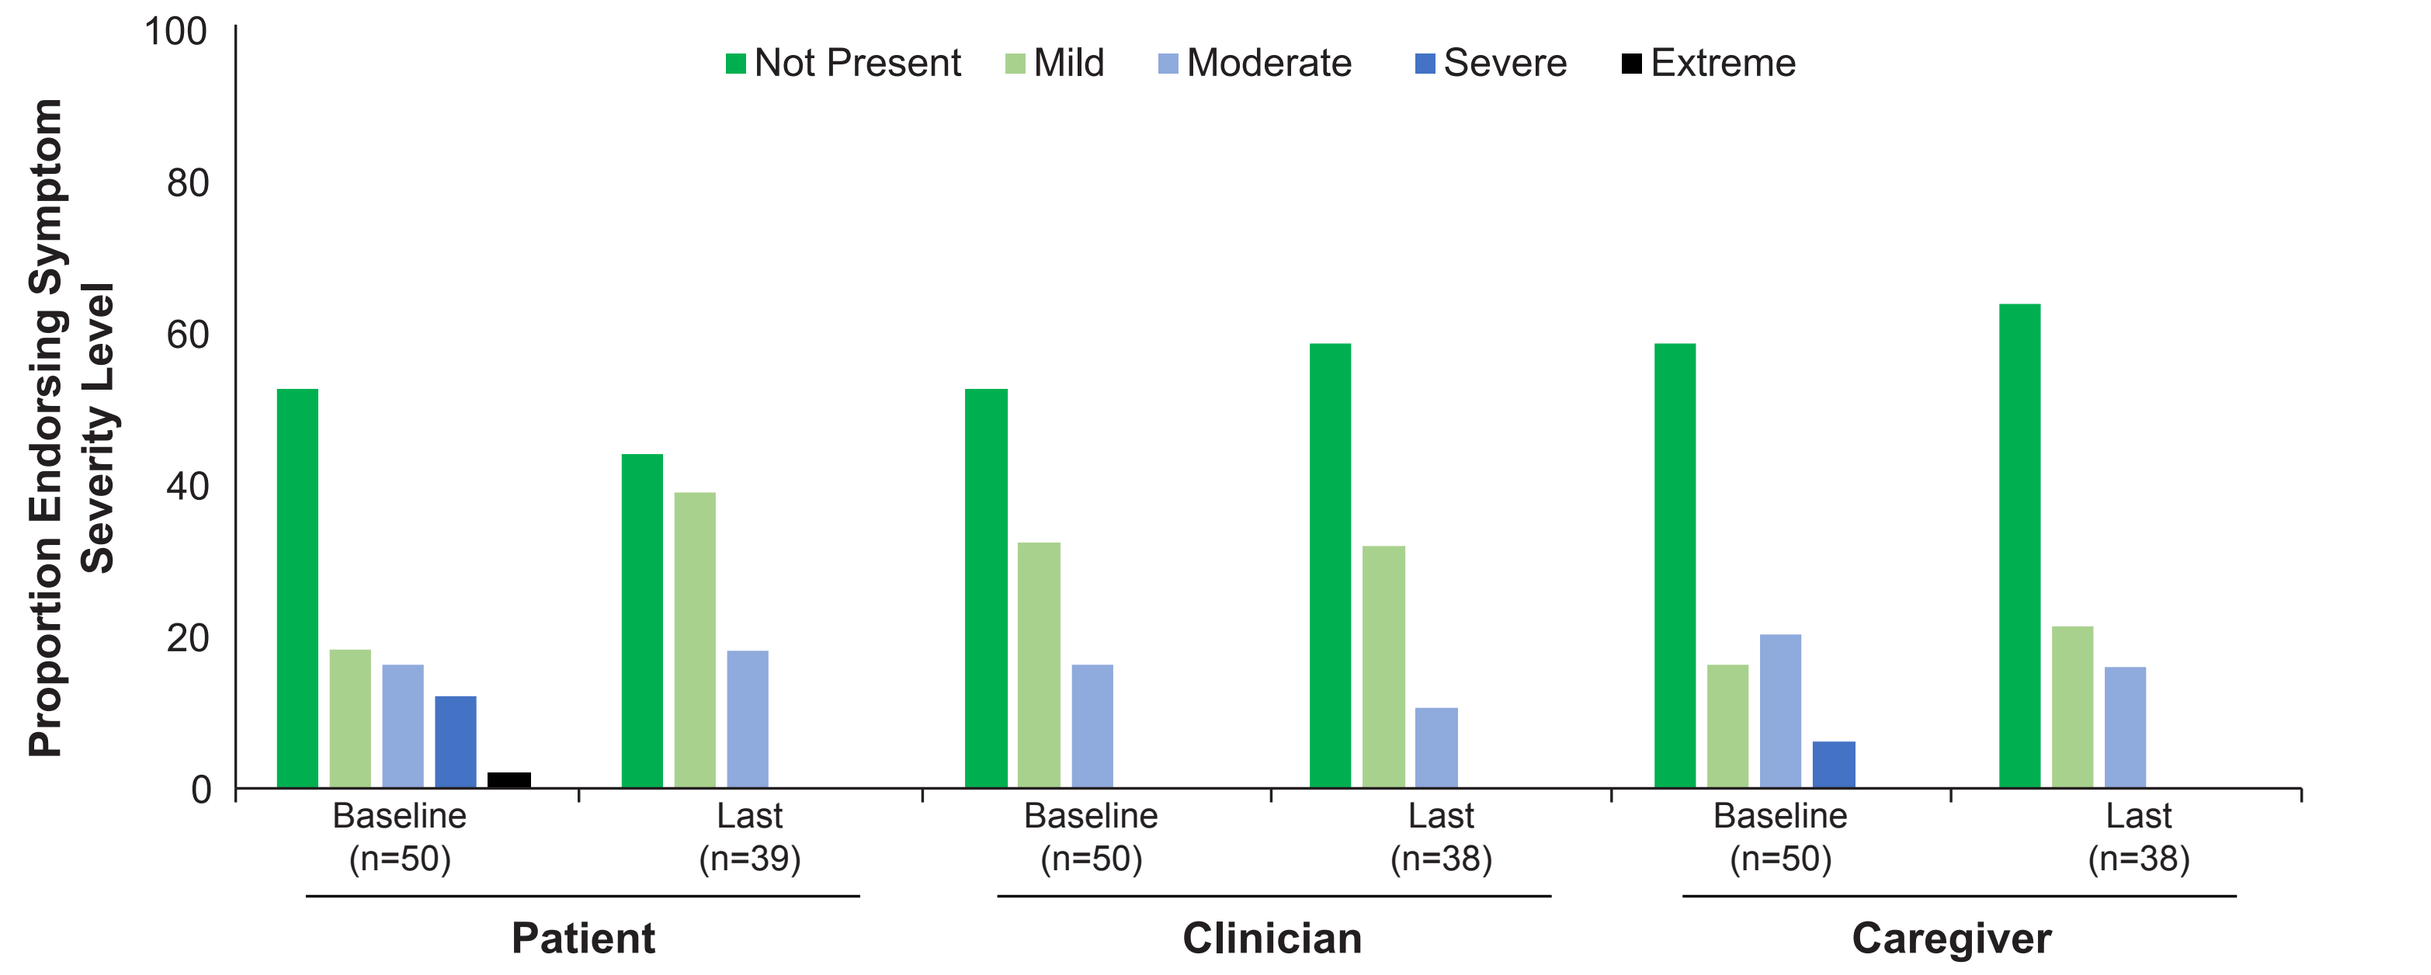
Not Present
Mild
Moderate
Severe
Extreme
Proportion Endorsing Symptom Severity Level
Baseline (n=50)
Last (n=39)
Baseline (n=50)
Last (n=38)
Baseline (n=50)
Last (n=38)
Patient
Clinician
Caregiver

D. Visual Learning/Memory Impairment

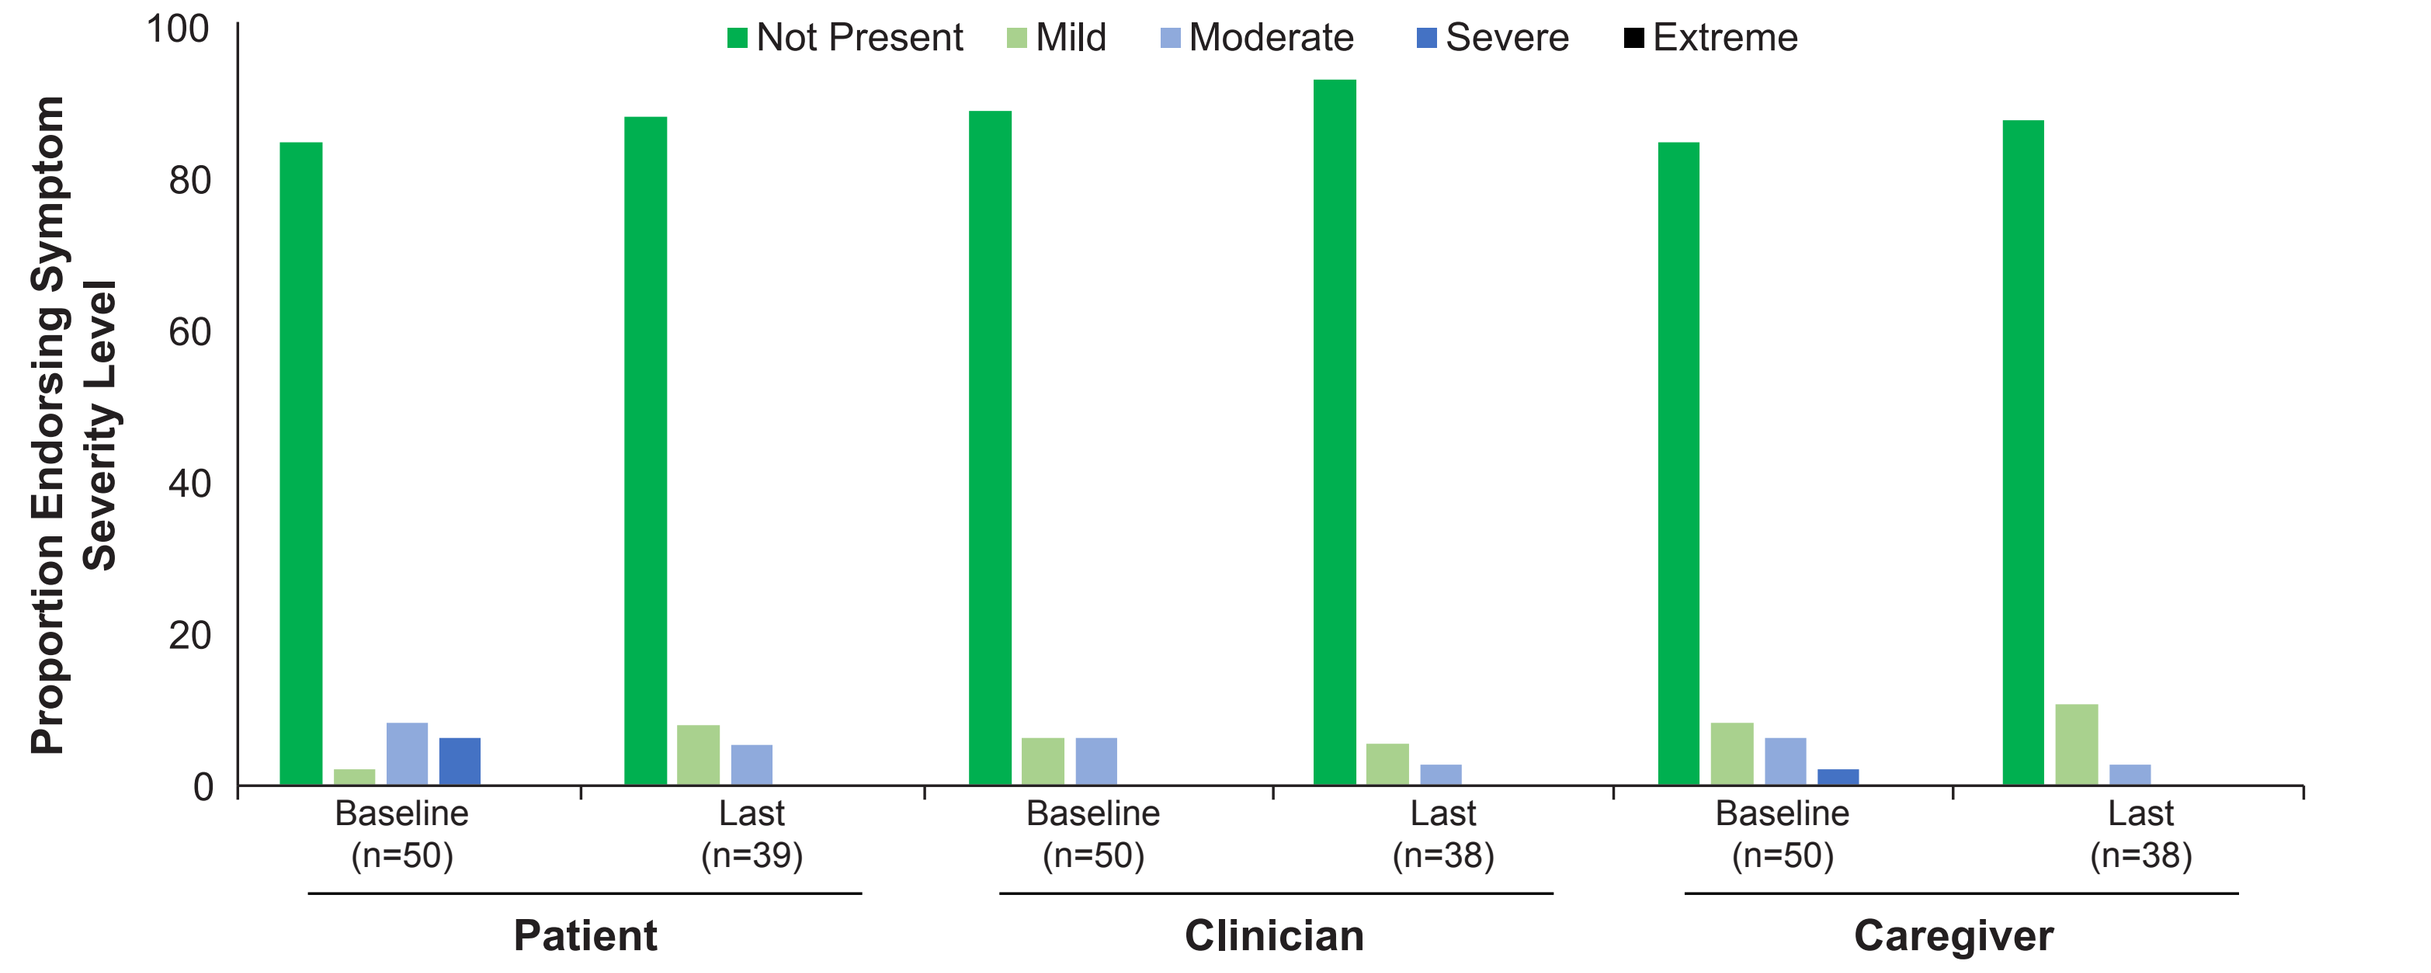
Not Present
Mild
Moderate
Severe
Extreme
Proportion Endorsing Symptom Severity Level
Baseline (n=50)
Last (n=39)
Baseline (n=50)
Last (n=38)
Baseline (n=50)
Last (n=38)
Patient
Clinician
Caregiver

E. Reasoning and Problem-Solving Impairment

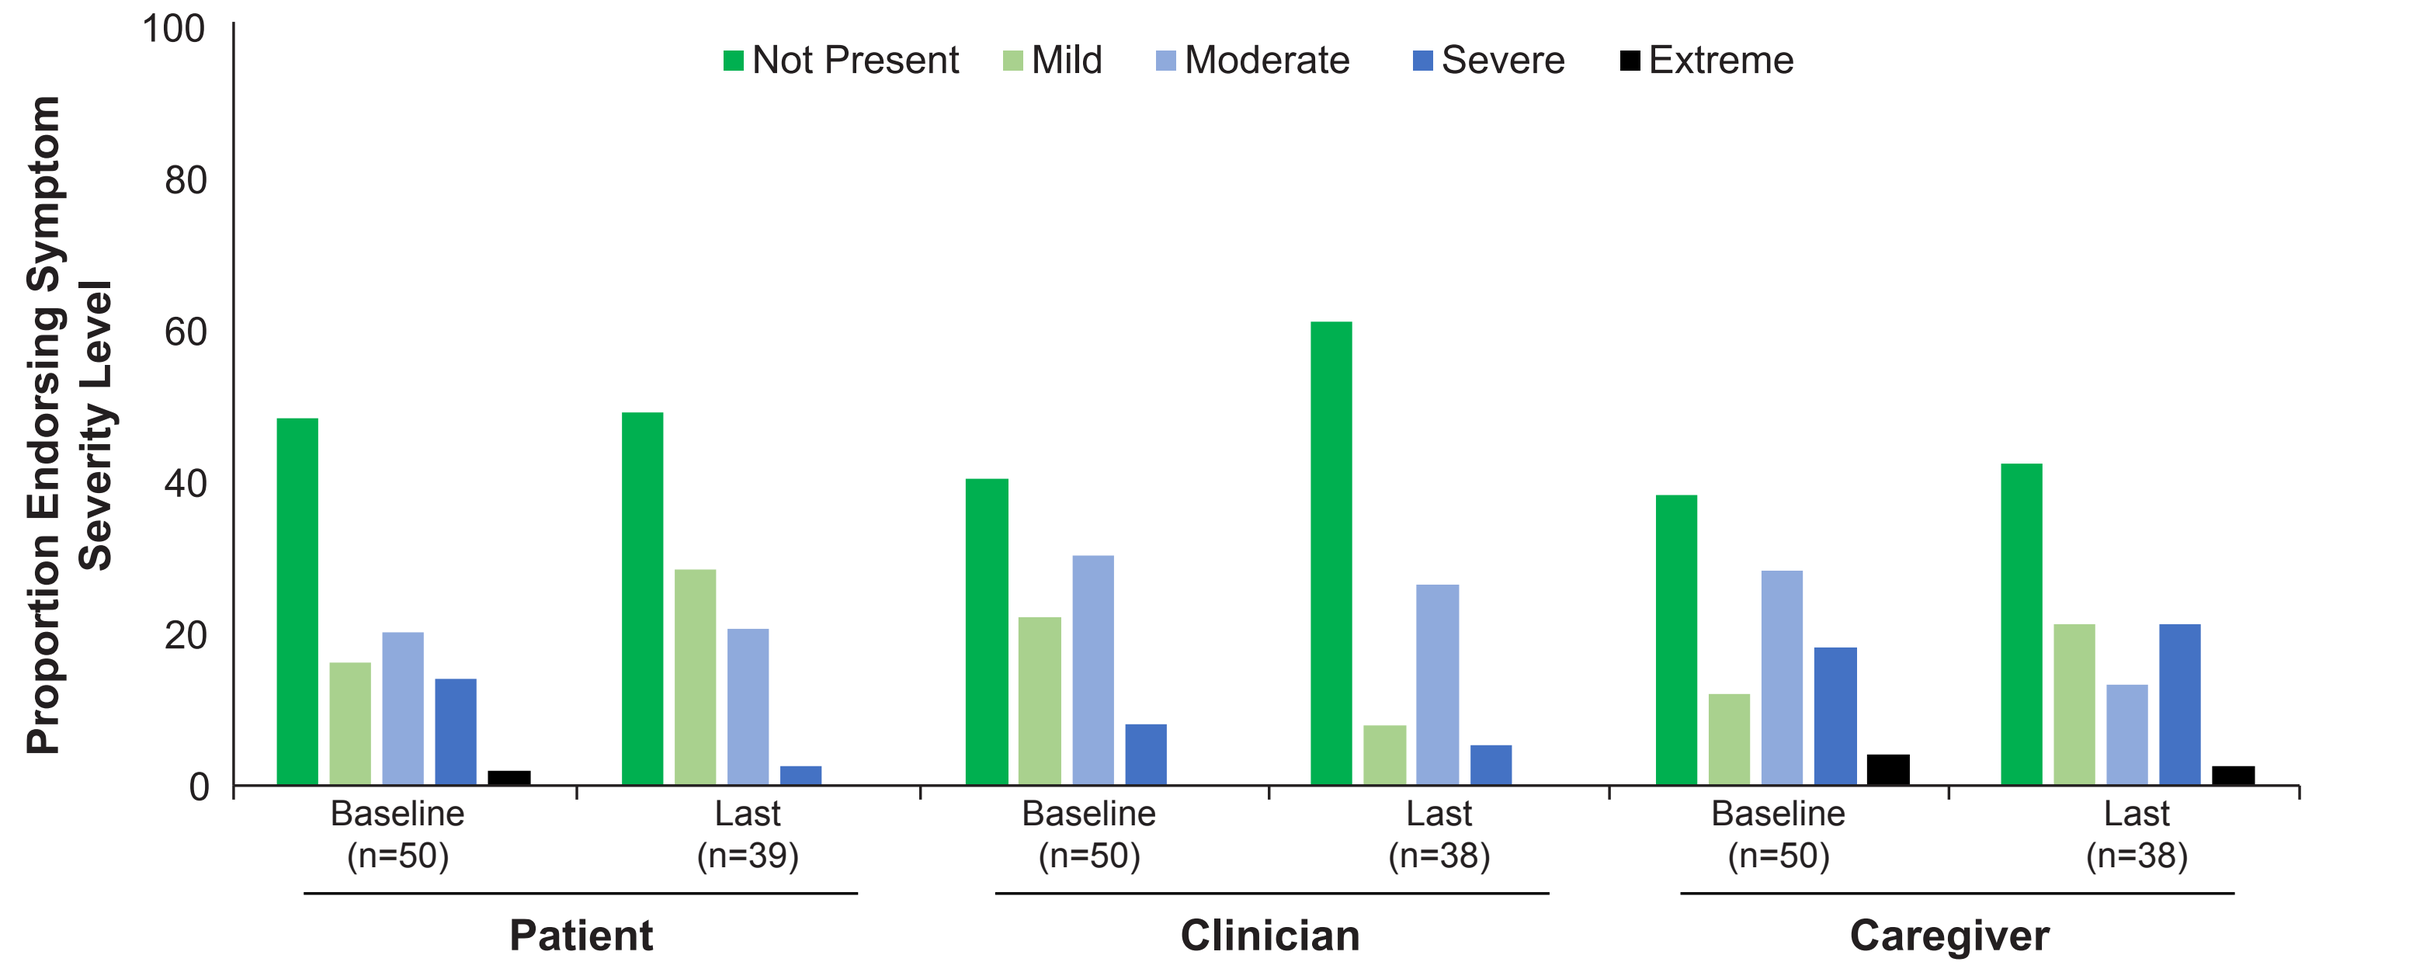
Not Present
Mild
Moderate
Severe
Extreme
Proportion Endorsing Symptom Severity Level
Baseline (n=50)
Last (n=39)
Baseline (n=50)
Last (n=38)
Baseline (n=50)
Last (n=38)
Patient
Clinician
Caregiver

F. Speed of Processing Impairment

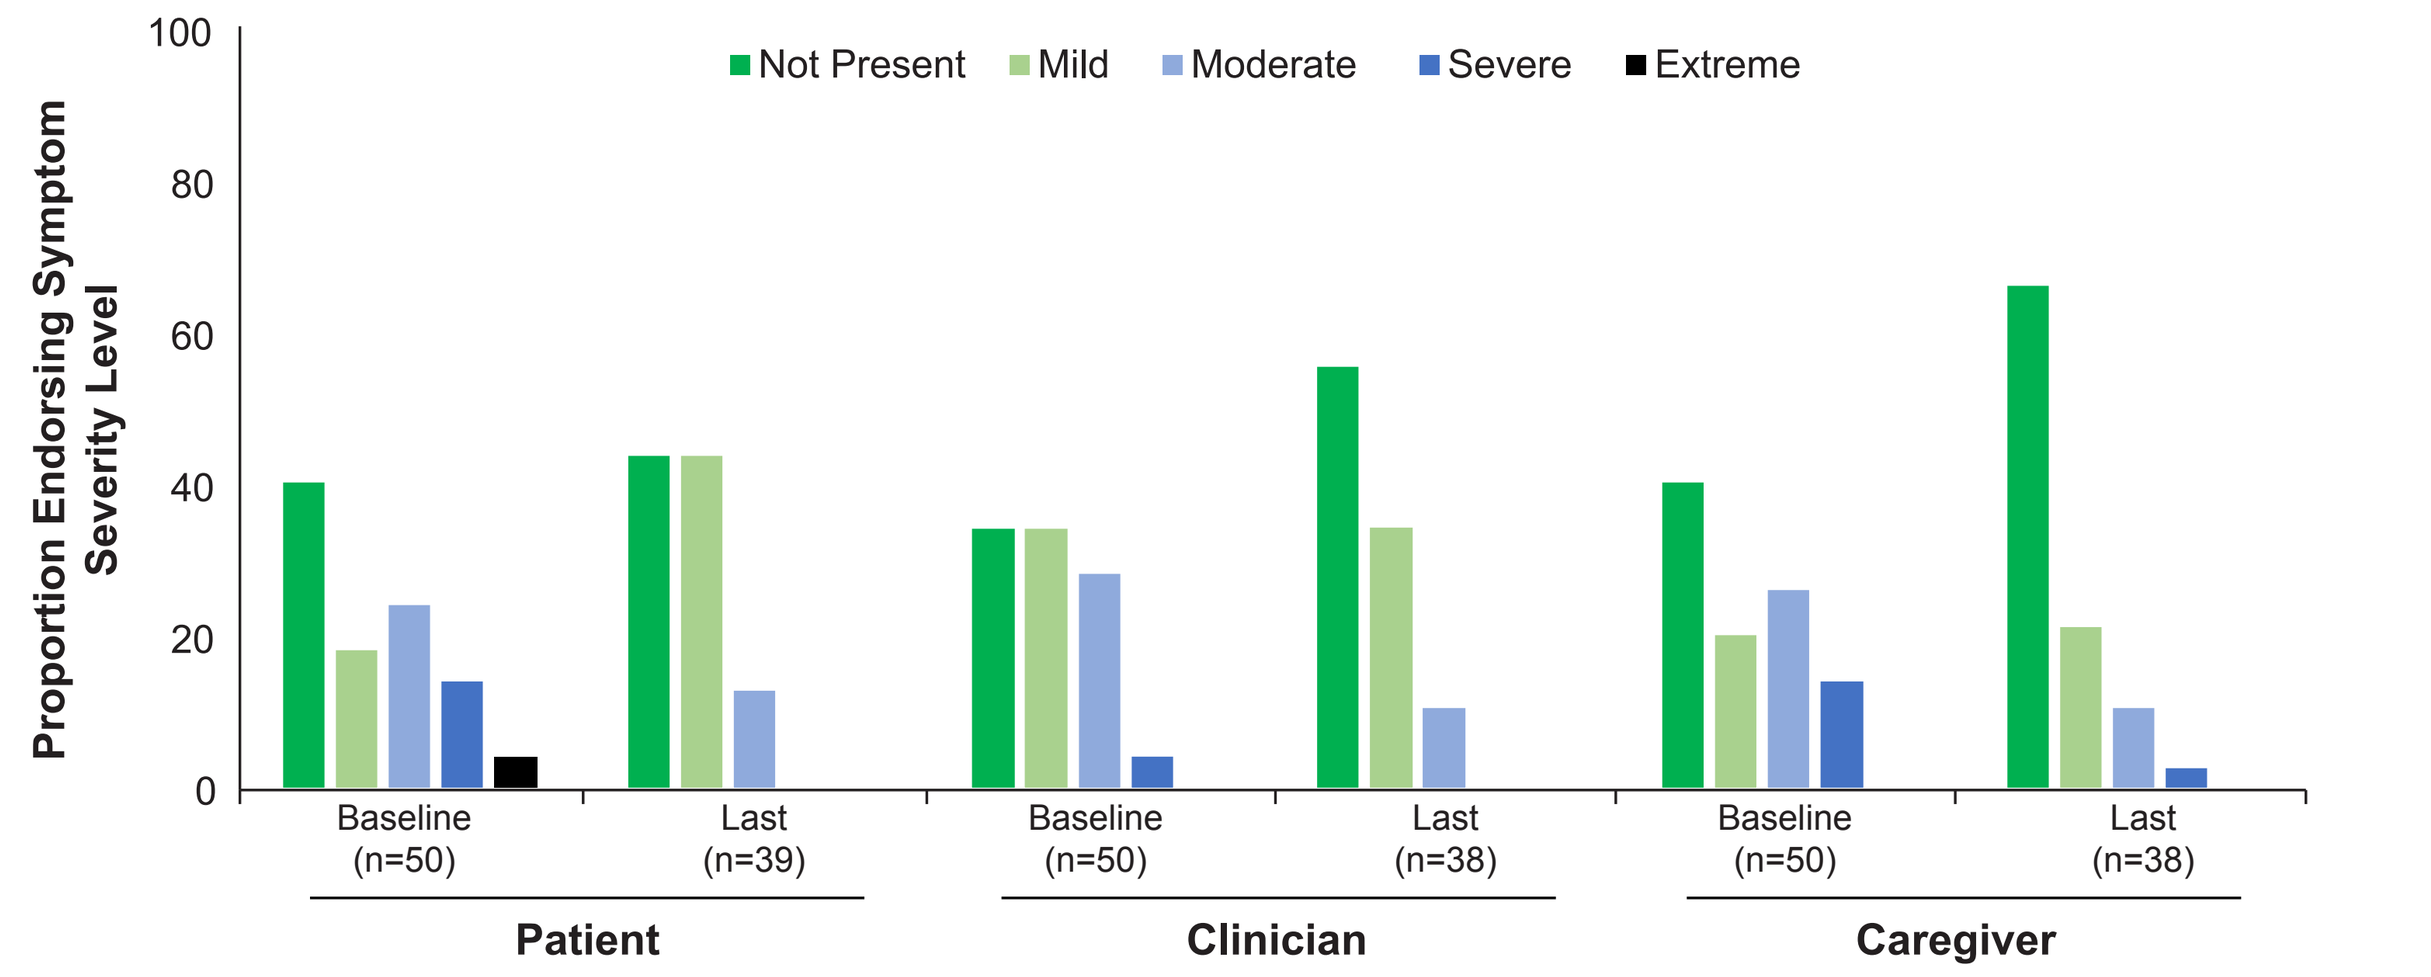
Not Present
Mild
Moderate
Severe
Extreme
Proportion Endorsing Symptom Severity Level
Baseline (n=50)
Last (n=39)
Baseline (n=50)
Last (n=38)
Baseline (n=50)
Last (n=38)
Patient
Clinician
Caregiver

G. Social Cognition Impairment

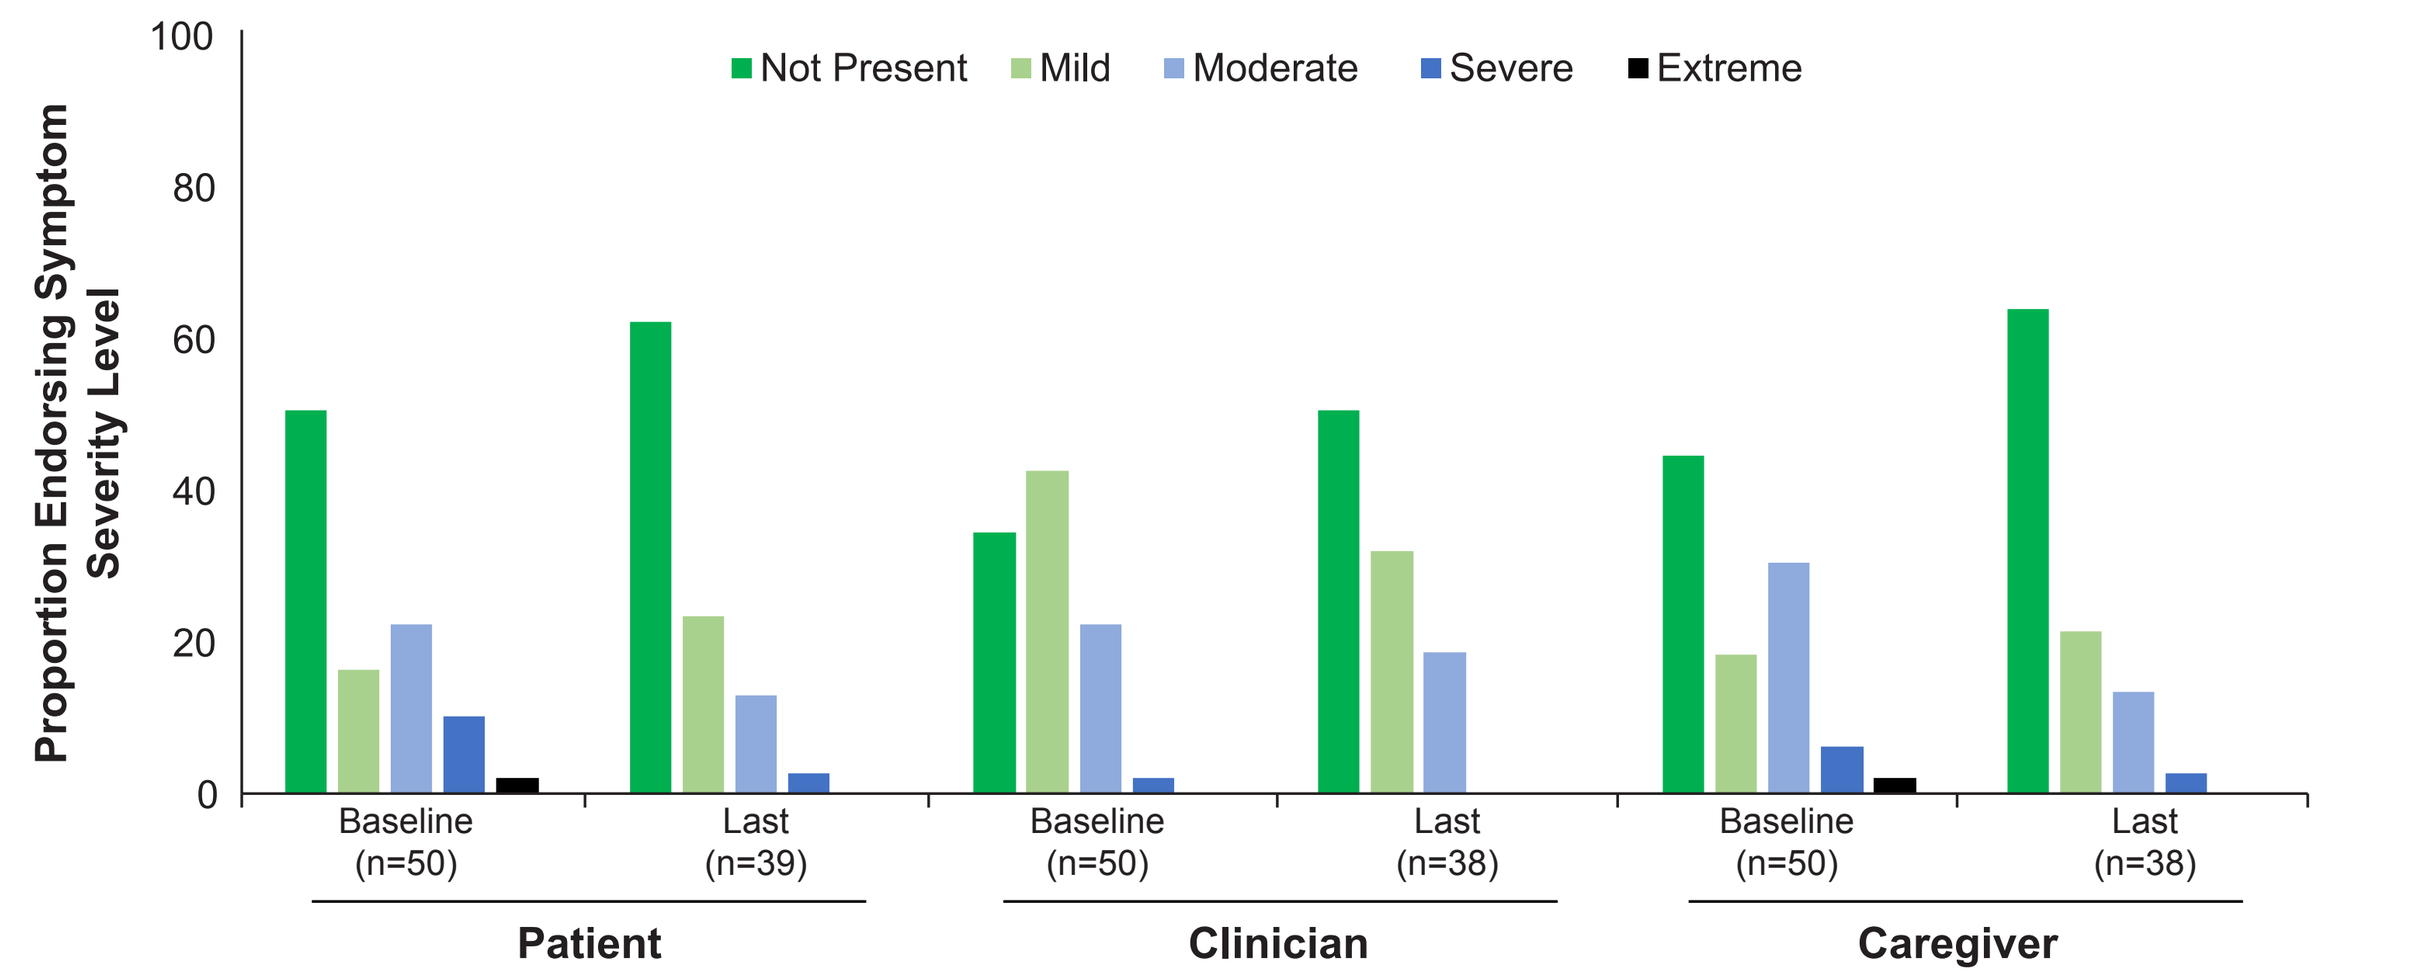
Not Present
Mild
Moderate
Severe
Extreme
Proportion Endorsing Symptom Severity Level
Baseline (n=50)
Last (n=39)
Baseline (n=50)
Last (n=38)
Baseline (n=50)
Last (n=38)
Patient
Clinician
Caregiver

Level of Agreement Between Groups

- Level of agreement between patient, clinician, and caregiver scores varied considerably across NY-AACENT domains (**Table 2**)
- Weighted kappa coefficients indicated fair to substantial agreement between patients and clinicians across most domains at last assessment[12]
 - Level of agreement between patients and clinicians was lower for Speed of Processing and Social Cognition at baseline and last assessment

Table 2. Level of Agreement, Weighted Kappa Coefficient

slight agreement (0.01–0.20) | fair agreement (0.21–0.40) | moderate agreement (0.41–0.60) | substantial agreement (0.61–0.80)

	Patient–Clinician Agreement K (95% CI)	Patient–Caregiver Agreement K (95% CI)	Clinician–Caregiver Agreement K (95% CI)
Baseline			
Working Memory	0.44 (0.24, 0.64)	0.25 (0.05, 0.46)	0.49 (0.31, 0.67)
Attention/Vigilance	0.43 (0.24, 0.62)	0.32 (0.10, 0.53)	0.59 (0.42, 0.75)
Verbal Learning/Memory	0.47 (0.29, 0.65)	0.35 (0.13, 0.57)	0.35 (0.16, 0.54)
Visual Learning/Memory	0.55 (0.25, 0.84)	0.60 (0.33, 0.86)	0.39 (0.02, 0.76)
Reasoning and Problem Solving	0.37 (0.19, 0.56)	0.41 (0.21, 0.62)	0.46 (0.28, 0.63)
Speed of Processing	0.14 (−0.06, 0.33)	0.26 (0.04, 0.48)	0.42 (0.22, 0.62)
Social Cognition	0.22 (0.03, 0.42)	0.31 (0.09, 0.52)	0.37 (0.19, 0.55)
Last assessment			
Working Memory	0.56 (0.36, 0.76)	0.49 (0.23, 0.75)	0.69 (0.51, 0.87)
Attention/Vigilance	0.43 (0.19, 0.68)	0.33 (0.06, 0.60)	0.45 (0.19, 0.70)
Verbal Learning/Memory	0.45 (0.21, 0.68)	0.21 (−0.05, 0.47)	0.48 (0.23, 0.73)
Visual Learning/Memory	0.64 (0.20, 1.00)	0.29 (−0.06, 0.64)	0.13 (−0.18, 0.44)
Reasoning and Problem Solving	0.41 (0.20, 0.62)	0.50 (0.32, 0.67)	0.64 (0.51, 0.78)
Speed of Processing	0.37 (0.13, 0.62)	0.22 (−0.02, 0.45)	0.34 (0.09, 0.59)
Social Cognition	0.32 (0.04, 0.59)	0.07 (−0.21, 0.35)	0.57 (0.36, 0.77)

LIMITATIONS

- This was a post hoc descriptive analysis based on available clinical trial data; the trial was not designed and powered to assess the agreement between patient, clinician, and caregiver responses on the NY-AACENT scale
- This was an open-label study in which all patients were treated with the LAI antipsychotic AL; the lack of a treatment comparison limits the ability to interpret the findings for the NY-AACENT scale
- Because the study population was limited to those who met inclusion and exclusion criteria, these results may not generalize to all patients with schizophrenia and who are treated with antipsychotics
- The NY-AACENT was designed to evaluate cognitive impairment due to treatment, not to evaluate overall fluctuations in status
- In this trial, cognition was not measured objectively. The NY-AACENT is based on subjective impression rather than performance-based testing and is therefore potentially prone to various subjective biases
 - Patients, clinicians, and caregivers may value aspects of cognition differently

CONCLUSIONS

- In this post hoc analysis of clinically stable patients with schizophrenia who switched to treatment with aripiprazole lauroxil, most patients, clinicians, and caregivers reported that cognitive difficulties were 'not present' or 'mild' across the 7 NY-AACENT domains at baseline
 - At baseline, clinicians more often reported cognitive impairment was 'not present' or 'mild' compared with patients for all NY-AACENT domains except Reasoning and Problem Solving
 - After 6 months of open-label AL treatment, there was a shift in the distribution of responses toward 'not present'/'mild' at last assessment for all reporters and across all domains
- The level of agreement on the magnitude of patient cognitive impairment was generally greater between clinicians and caregivers and between patients and clinicians than between patients and caregivers; however, the domains of greatest agreement varied
 - Agreement increased between baseline and 6 months across most cognitive domains, with patient-clinician agreement improving in all domains except for Verbal Learning and Memory
- These results suggest that up to 6 months of open-label treatment with AL was not associated with adverse effects on cognitive functioning in patients with schizophrenia who switched from their prior LAI antipsychotic medication
- Further analysis is needed to examine associations between domains with low patient–clinician agreement and functioning in patients with schizophrenia treated with LAI antipsychotics

REFERENCES

1. Bowie CR, et al. *Am J Psychiatry.* 2006;163:418-25. **2.** Nuechterlein KH, et al. *J Clin Psychiatry.* 2014;75 Suppl 2:25-9. **3.** Gould F, et al. *Neuropsychology.* 2015;29:675-82. **4.** Hoff AL, et al. *Am J Psychiatry.* 1999;156:1336-41. **5.** Keefe RS, et al. *Neuropsychopharmacology.* 2006;31:2033-46. **6.** Eum S, et al. *Schizophr Res.* 2017;190:129-35. **7.** Ang MS, et al. *J Clin Psychopharmacol.* 2017;37:651-6. **8.** Correll C, et al. *Schizophr Bull.* 2019;45(suppl 2):S221. **9.** Miller BJ, et al. *Schizophr Res.* 2019;208:44-8. **10.** American Psychiatric Association. Diagnostic and Statistical Manual of Mental Disorders, DSM-5. Washington, DC: American Psychiatric Publishing; 2013. **11.** McHugh ML. *Biochem Med (Zagreb).* 2012;22:276-82. **12.** Landis JR, Koch GG. *Biometrics.* 1977;33:159-74.

AUTHOR DISCLOSURES

Mark G A Opler is an employee and shareholder of WCG MedAvante-ProPhase, Inc. Amy Claxton was an employee of Alkermes, Inc., at the time of the study and may own stock/options in the company. James McGrory, Sabine Gasper, Meihua Wang, and Sergey Yagoda are employees of Alkermes, Inc., and may own stock/options in the company.

ACKNOWLEDGMENTS

This study was sponsored by Alkermes, Inc. Medical writing and editorial support were provided by Peloton Advantage, LLC, an OPEN Health company, and funded by Alkermes, Inc.

Copies of this poster obtained through this QR (Quick Response) code are for personal use only and may not be reproduced without permission of Alkermes, Inc. For permission, contact USMedInfo@Alkermes.com.

Alkermes

Supported by funding from Alkermes, Inc.