The Relative Value of Anti-Obesity Medications Compared to Similar Therapies

Nina Kim[1], Joaquin Estrada[2], Isabella Chow[2], Aleksandrina Ruseva[1], Abhilasha Ramasamy[1], Chakkarin Burudpakdee[1], Christopher Blanchette[1]

https://sciencehub.novonordisk.com/ispor2022/Kim.html?cid=qr-6230407241

Obesity represents the highest economic burden

Annual Economic Burden in the US, $ billion (2020 USD)

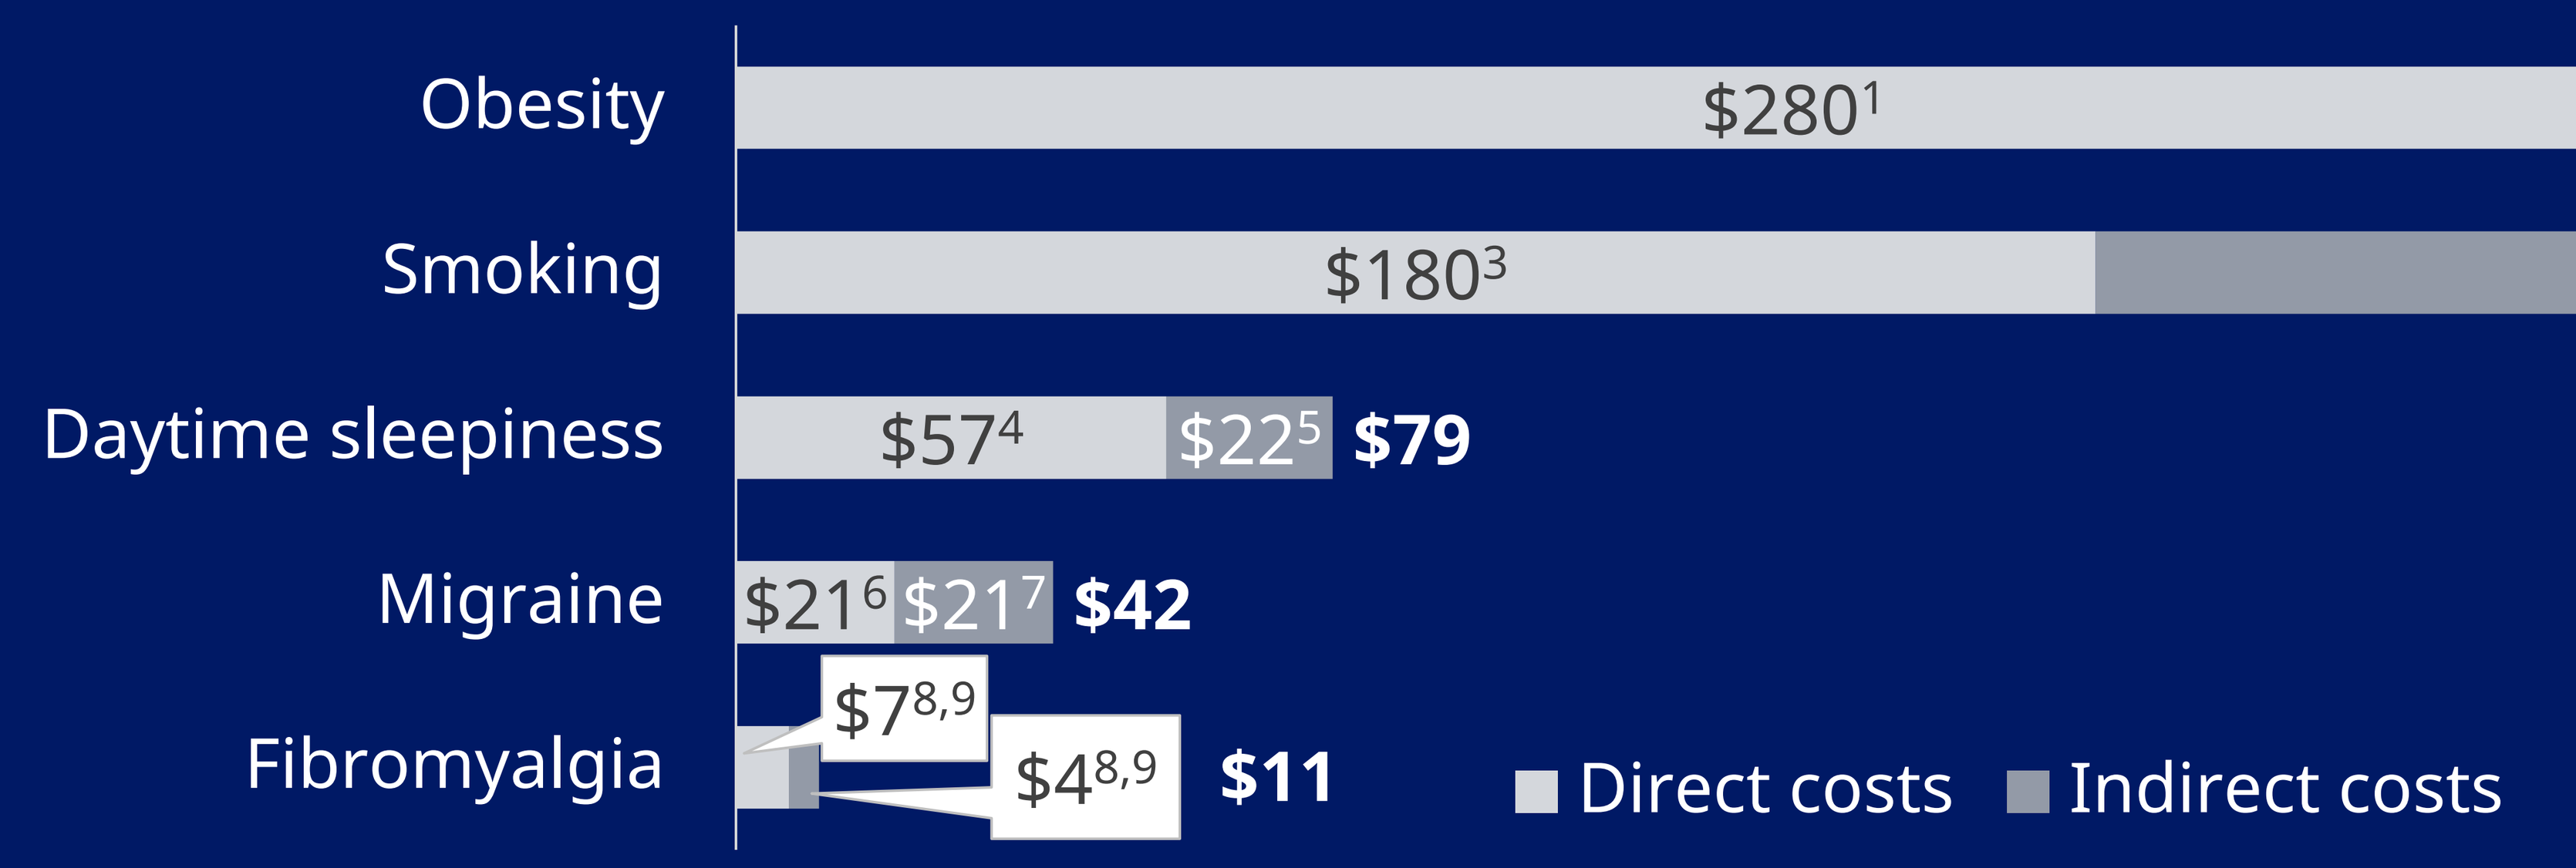
Obesity
$280[1]
$396[2]
$676
Smoking
$180[3]
$165[3]
$345
Daytime sleepiness
$57[4]
$22[5]
$79
Migraine
$21[6]
$21[7]
$42
Fibromyalgia
$7[8,9]
$4[8,9]
$11
Direct costs
Indirect costs

Notes: Direct costs are medical and pharmacy costs attributable to the burden of the disease. Indirect costs are costs not directly associated to treatment but are produced by the disease (e.g., productivity loss).

Background and Aims

- Access/coverage for anti-obesity medications (AOMs) is limited compared to treatments for other similar chronic diseases, despite high disease burden associated with obesity.
- We aimed to assess the relative value of AOMs through analysis and comparison of cost-benefit and/or clinical benefit for covered medications across selected therapeutic areas.

Methods

- An overview of the methodology is described in **Table 1**.
 - The study was conducted in three different phases (1) a grey literature review to identify therapeutic areas similar to obesity and potential analogues, (2) selection and prioritization of therapeutic areas for the targeted literature review (TLR), (3) a TLR on the selected therapeutic areas to identify literature for inclusion in the analysis.
- The TLR was performed to identify clinical and economic outcomes for the selected therapeutic areas.
- Parameters extracted and evaluated included direct and indirect medical costs, cost drivers, and associated comorbidities.

Table 1: Therapeutic area analogue identification flow chart

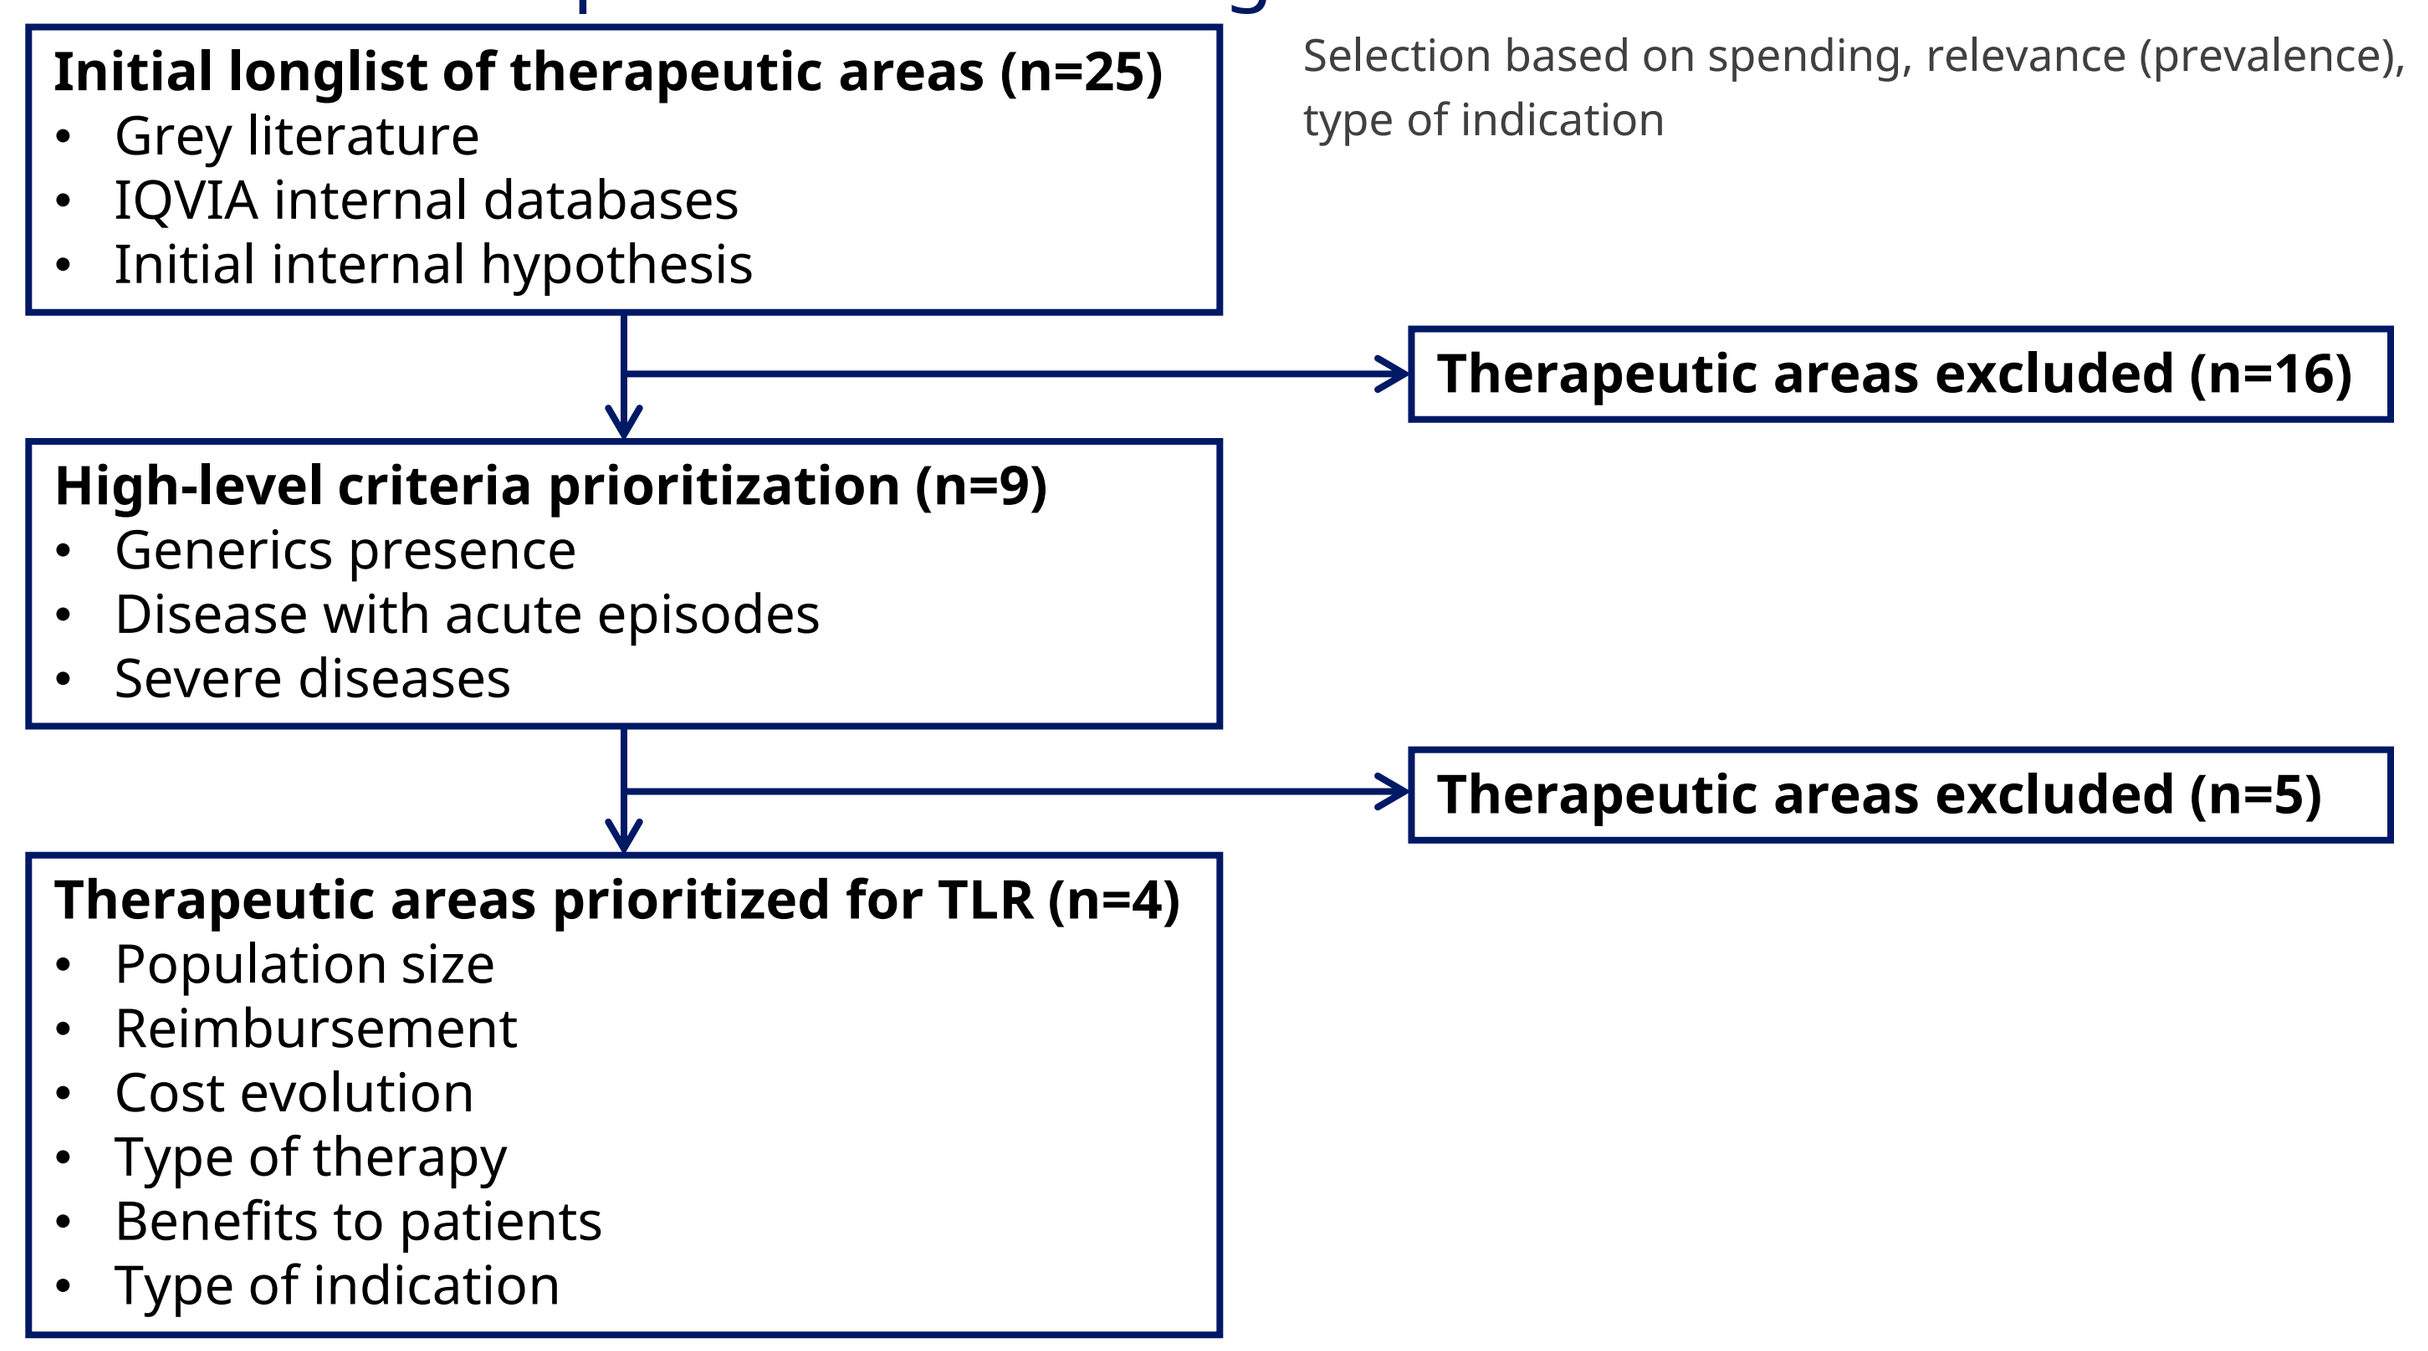
Initial longlist of therapeutic areas (n=25)
• Grey literature
• IQVIA internal databases
• Initial internal hypothesis
Selection based on spending, relevance (prevalence), type of indication
Therapeutic areas excluded (n=16)
High-level criteria prioritization (n=9)
• Generics presence
• Disease with acute episodes
• Severe diseases
Therapeutic areas excluded (n=5)
Therapeutic areas prioritized for TLR (n=4)
• Population size
• Reimbursement
• Cost evolution
• Type of therapy
• Benefits to patients
• Type of indication

Affiliations: [1]Novo Nordisk, Inc., Plainsboro, NJ, USA; [2]IQVIA, Inc., San Francisco, CA, USA

This study was sponsored by Novo Nordisk, Inc,. The authors acknowledge the medical writing assistance of Elizabeth Tanner and Rebecca Hahn of KJT Group, Inc. (Rochester, NY, USA).

Key results

- Four therapeutic area comparators and their pharmacotherapy analogues were identified (smoking cessation, varenicline; daytime sleepiness, modafinil; migraine, erenumab; and fibromyalgia, pregabalin), selected based on similarity to obesity and AOMs across important parameters of value (e.g., US prevalence, reimbursement history, type of therapies).
- The TLR identified 2,956 papers. Results were screened for relevant data for extraction and analysis resulting in 89 publications.
- Obesity and smoking represented the highest economic burden with $676 and $345 billion in yearly direct and indirect medical costs, respectively (**Result Banner**).
- Weight loss resulted in a reduction of $2,586 in direct medical costs per patient per year (PPPY), which is higher than the cost reduction associated with varenicline ($930 PPPY), modafinil ($1,045 PPPY), or erenumab ($468 PPPY); pregabalin utilization showed an increase of $924 PPPY (**Figure 1**). Main drivers of cost savings were reductions in outpatient, inpatient, and emergency room costs.
- Obesity was associated with more comorbidities (17) than comparators (migraine=9, smoking=8, daytime sleepiness=5, fibromyalgia=2) (**Table 2**).
- Smoking and obesity were associated with the costliest comorbidities of cardiovascular disease, stroke, and cancer.
- To understand the annual productivity loss due to disease, we multiplied annual average absenteeism per person by estimated prevalence in an average mid-size company for each condition. The annual workday loss was highest for obesity and daytime sleepiness, with an annual workday loss of >520 days for an average employer, for an annual productivity loss of approximately $130,000 for each condition (**Table 3**).

Presented at ISPOR 2022 conference, May 15-18, 2022.

Disclosures: NK, AR, AR, CB, and CB, were employees and/or shareholders of Novo Nordisk, Inc. at the time research was conducted. JE and IC are employees of IQVIA, Inc., which received funding to conduct the research study on which the poster is based.

Figure 1: Real-world annual medical cost reduction by treatment area

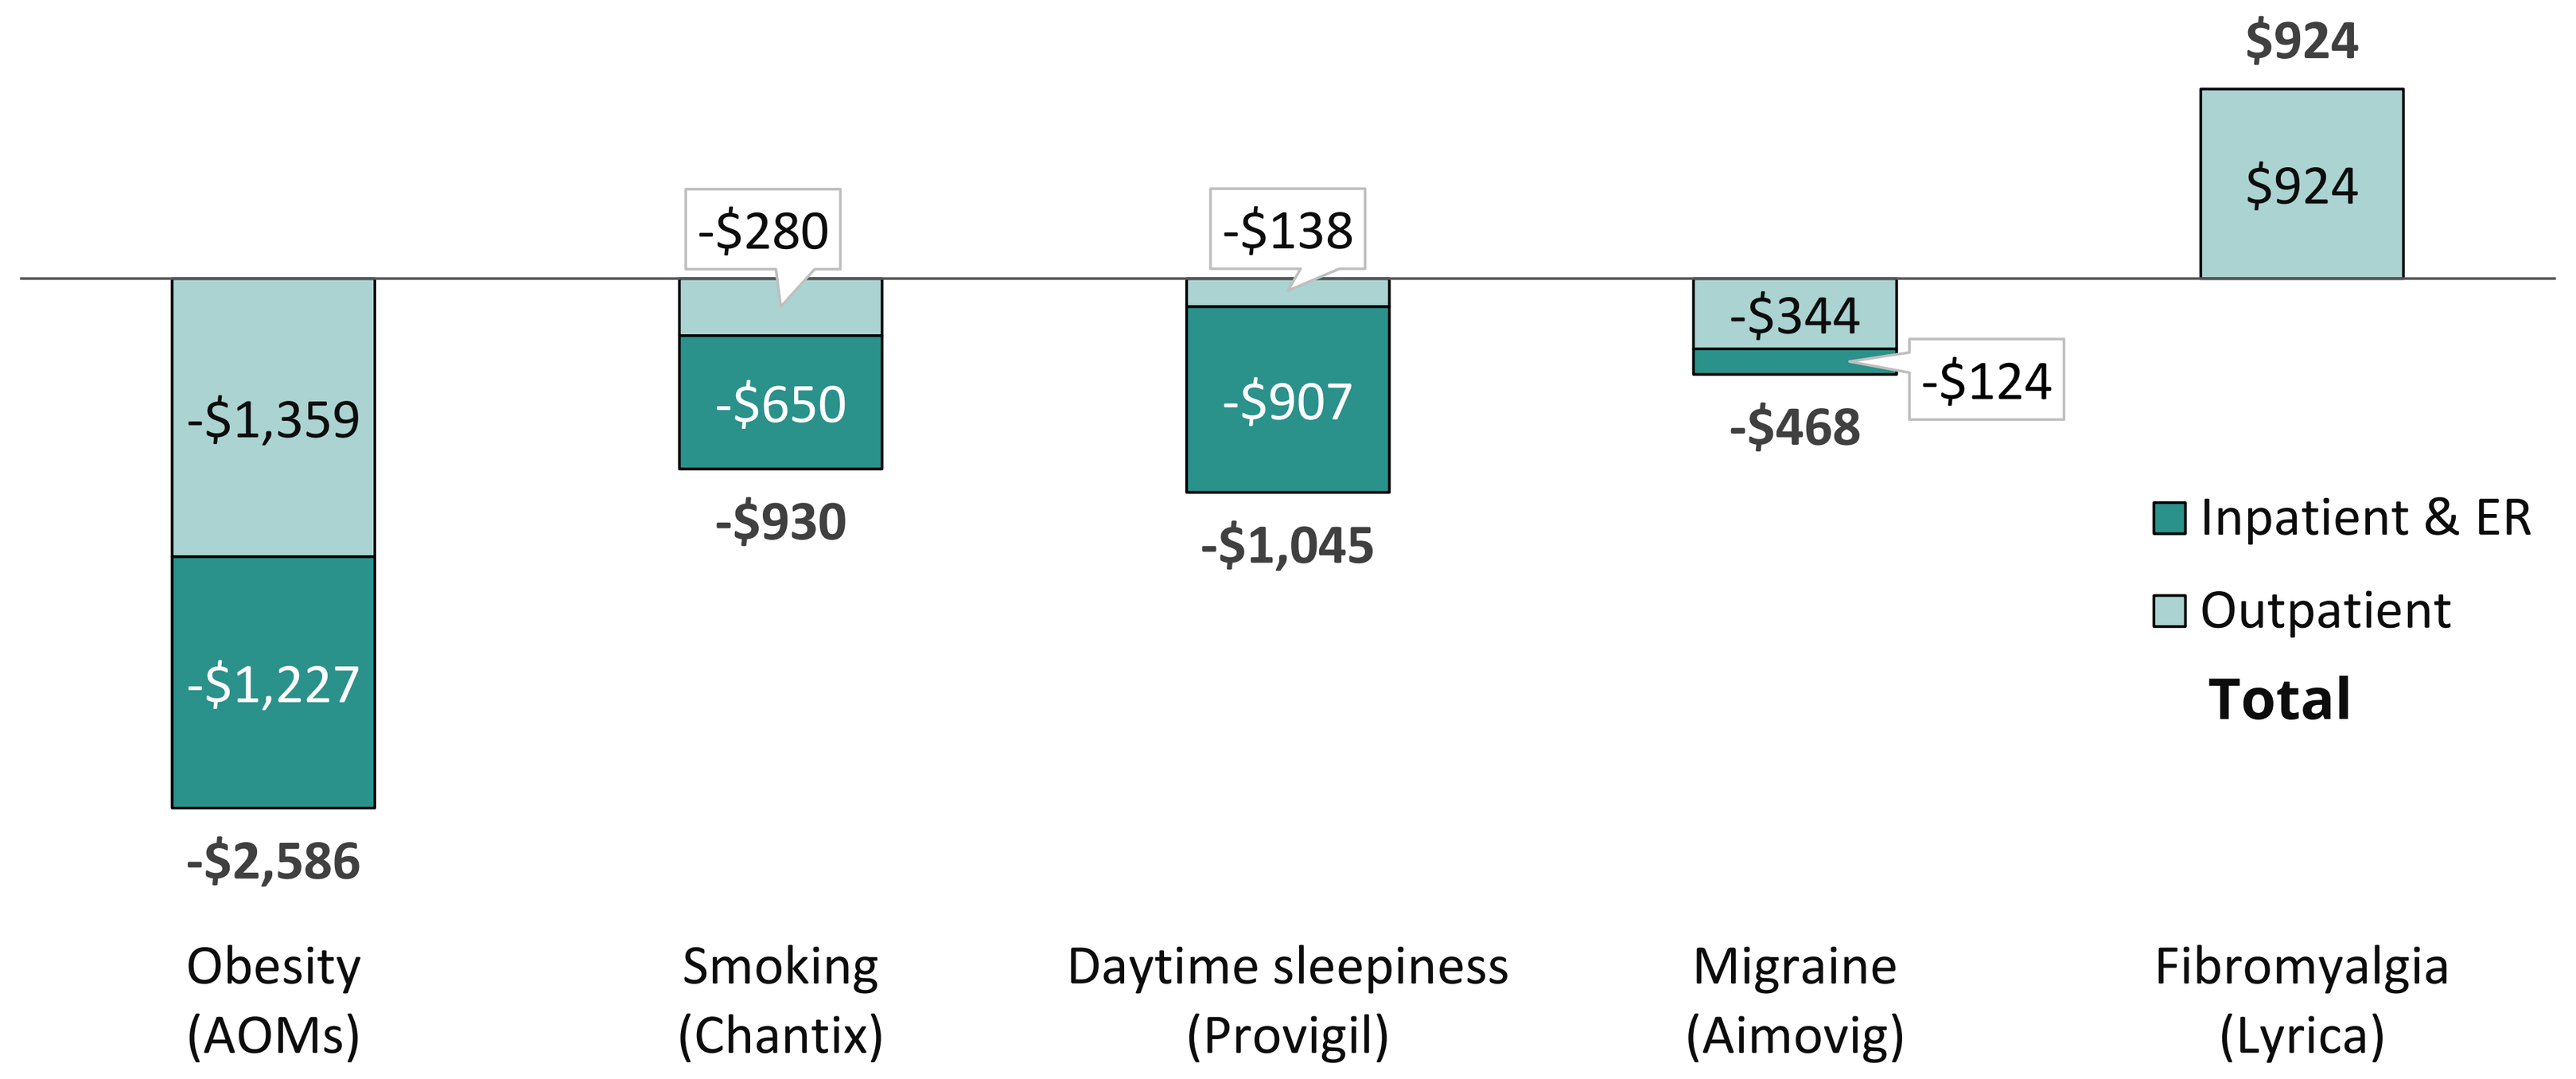
$924
$924
-$280
-$138
-$1,359
-$650
-$907
-$344
-$124
-$468
-$930
-$1,045
-$1,227
-$2,586
Inpatient & ER
Outpatient
Total
Obesity (AOMs)
Smoking (Chantix)
Daytime sleepiness (Provigil)
Migraine (Aimovig)
Fibromyalgia (Lyrica)

AOMs, anti-obesity medications; ER, emergency room

Table 2: Most common comorbidities and financial impact for each condition

	Obesity	Smoking	Excessive Sleepiness	Migraine	Fibromyalgia
Cardiovascular Disease	$$$	$$$			
Cancer	$$$	$$$			
Stroke	$$$	$$$		$$$	
Pulmonary embolism	$$$	$$$			
Kidney failure	$$				
Sleep apnea	$$	$$	$$		
Coronary artery disease	$$				
Respiratory	$$	$$	$$		
Osteoarthritis	$$			$$	
Pain	$$		$$	$$	$$
Non-alcoholic fatty liver disease (NAFLD)	$				
Dyslipidemia	$				
gastroesophageal reflux disease (GERD)	$				
Depression	$	$	$	$	
Hypertension	$			$	$
Urinary stress	$				
Diabetes	$				
Anxiety		$	$	$	
Endometriosis					
Epilepsy				$	
Irritable bowel syndrome (IBS)				$	
Osteoporosis				$$	

Note: The number of $ correlates with the cost of the comorbidity relative to each other: light blue and $=low, medium blue and $$=moderate, dark blue and $$$=high.

Table 3: Annual workday loss incremental to the disease, for a mid-sized company

Therapeutic area	**Annual additional absenteeism** (per person per year)	**Prevalence in an average mid-size company** (number of employees)	**Annual workday loss** (days)	**Annual productivity loss due to absenteeism** (USD)
Obesity	3.0 days[10]	175	525	$129,000
Smoking	2.3 days[11]	70	161	$40,000
Daytime sleepiness	4.6 days[12]	115	529	$130,000
Migraine	1.7 days[13]	85	145	$36,000
Fibromyalgia	13 days[14]	20	260	$93,000

Notes: Baseline of annual average absenteeism is 2.34 days in workers with normal weight.[1] Annual workday loss was calculated using the assumption of 500 employees for an average mid-size employer and reported prevalence rates for each condition. Productivity loss to absenteeism assumes the US national average hourly wage, US Bureau of Labor and Statistics 2021.

Conclusions and Implications

- Obesity had the highest economic burden and was associated with greatest number of comorbidities.
- Obesity was associated with the highest number of costly comorbidities and treatment of obesity had the largest medical cost savings.
- AOMs provide high relative value compared to selected analogues.
- Despite this, AOMs are not currently covered by most health plans. AOM coverage and utilization may reduce the economic burden associated with obesity.

References:
[1]Cawley J, et al. J Manag Care Spec Pharm. Mar 2021;27(3):354-366. doi:10.18553/jmcp.2021.20410
[2]Dee A, et al. BMC Res Notes. 2014;7:242-242. doi:10.1186/1756-0500-7-242
[3]Lee LJ, et al. Adv Ther. Feb 2019;36(2):365-380. doi:10.1007/s12325-018-0858-y
[4]Knauert M, et al. World J Otorhinolaryngol Head Neck Surg. 2015;1(1):17-27. doi:10.1016/j.wjorl.2015.08.001
[5]Sassani A, et al. Sleep. May 1 2004;27(3):453-8. doi:10.1093/sleep/27.3.453
[6]Tepper SJ, et al. The Journal of Headache and Pain. 2021/04/19 2021;22(1):27. doi:10.1186/s10194-021-01238-2
[7]Yucel A, et al. Am J Manag Care. Dec 1 2020;26(12):e403-e408. doi:10.37765/ajmc.2020.88547
[8]Frech FQ, et al. Am J Pharm Benefits. 2017;9(6):200-207.
[9]Walitt B, et al. PloS one. 2015;10(9):e0138024-e0138024. doi:10.1371/journal.pone.0138024
[10]Cawley J, et al. J Occup Environ Med. Jul 1 2021;63(7):565-573. doi:10.1097/jom.0000000000002198
[11]Halpern MT, et al. Tob Control. 2001;10(3):233-238. doi:10.1136/tc.10.3.233
[12]Reynolds AC, et al. Sleep Health. Oct 2017;3(5):357-361. doi:10.1016/j.sleh.2017.06.003
[13]Michel P, et al. Cephalalgia. Jun 1999;19(5):503-10. doi:10.1046/j.1468-2982.1999.019005503.x
[14]Chandran A, et al. J Manag Care Pharm. Jul-Aug 2012;18(6):415-26. doi:10.18553/jmcp.2012.18.6.415