The Budget Impact of Zanubrutinib for the Treatment of Waldenström's Macroglobulinemia in the United States

Sizhu Liu[1], Keri Yang[1], David Campbell[2], Kristen Migliaccio-Walle[2], Boxiong Tang[1]

[1] BeiGene Ltd., Emeryville, CA; [2] Curta, Seattle, WA

Background

- Waldenström's macroglobulinemia (WM) is a rare B-cell malignancy of which Bruton's tyrosine kinase (BTK) plays a critical role in B-cell receptor signaling[1,2]
- Standards of care commonly used in the United States (US) include ibrutinib (+/- rituximab), bendamustine + rituximab (BR), bortezomib + dexamethasone + rituximab (BDR), and rituximab monotherapy (R-mono) for the treatment of WM
- Zanubrutinib, a highly selective, next-generation BTK inhibitor, was approved for the treatment of adult patients with WM in the US on August 31, 2021, expanding treatment options for patients with WM[3]
- There are few published studies evaluating the economic impact of treatments for WM and none have evaluated the impact of access to zanubrutinib

Objective

- To estimate the budget impact of providing access to zanubrutinib for the treatment of adult patients with WM from the US healthcare payer perspective

Methods

Structure

- An Excel-based budget impact model (BIM) with a 3-year time horizon was developed to estimate the economic impact of providing adult patients with WM access to zanubrutinib within a hypothetical 1-million-member US health plan (**Figure 1**)
- The target patient population included adult patients with WM in a US health plan who are treatment naïve or relapsed/refractory to treatment
- A scenario without access to zanubrutinib where patients could receive treatment with ibrutinib (+/- rituximab), BR, BDR, R-mono was compared to a scenario with access to zanubrutinib

Inputs and Assumptions

- The model assumed a hypothetical plan size of 1 million members of which 16.5% were covered by Medicare based on the proportion of the US population ≥65 years old[4]
- WM incidence rates were 0.2 per 100,000 for patients <65 years and 2.5 per 100,000 for patients ≥65 years[5]
- Among the WM incident population, the Lymphoma and Leukemia Society estimates that 87.5% are diagnosed and treated[6]
- Further, of those diagnosed and treated, an estimated 81.6% have relapsed/refractory disease based on enrollment from the ASPEN trial[7]

Treatment Costs and Duration

- Cost inputs included drug acquisition costs, drug administration costs, drug monitoring costs, and adverse event management costs (**Table 1**)
- Drug acquisition costs in 2021 US$ were calculated using wholesale acquisition cost, duration of treatment, and dosing quantities and frequencies from prescribing information and trial publications[7-12]

Monitoring Costs

- Clinical monitoring and testing were selected based on prescribing information or trial publications for each comparator
- Clinical monitoring or testing frequencies were based on assumptions while costs were calculated from the Centers for Medicare & Medicaid Services fee schedules or the National Fee Analyzer[13,14]

Adverse Event Management Costs

- Adverse events were selected based upon their clinical relevance to WM and modeled treatments
- Adverse event management costs were estimated based on rates of grade 3 or higher adverse events from prescribing information or trial publications, and duration of treatment[7,9,10,15,16]
- Cost per adverse event were based on Healthcare Cost and Utilization Project (HCUP) costs and were inflated to 2021 US dollars using the medical care component of the Consumer Price Index[17,18]

Market Dynamics

- Market shares in the scenario without access to zanubrutinib were calculated from available market insights[19]
- In the scenario with access to zanubrutinib, it is assumed uptake is 10% in year 1, followed by an increase of 5% in year 2 and year 3
- Uptake of zanubrutinib was assumed to come from all other treatment comparators proportionally

Figure 1. Budget Impact Model Structure

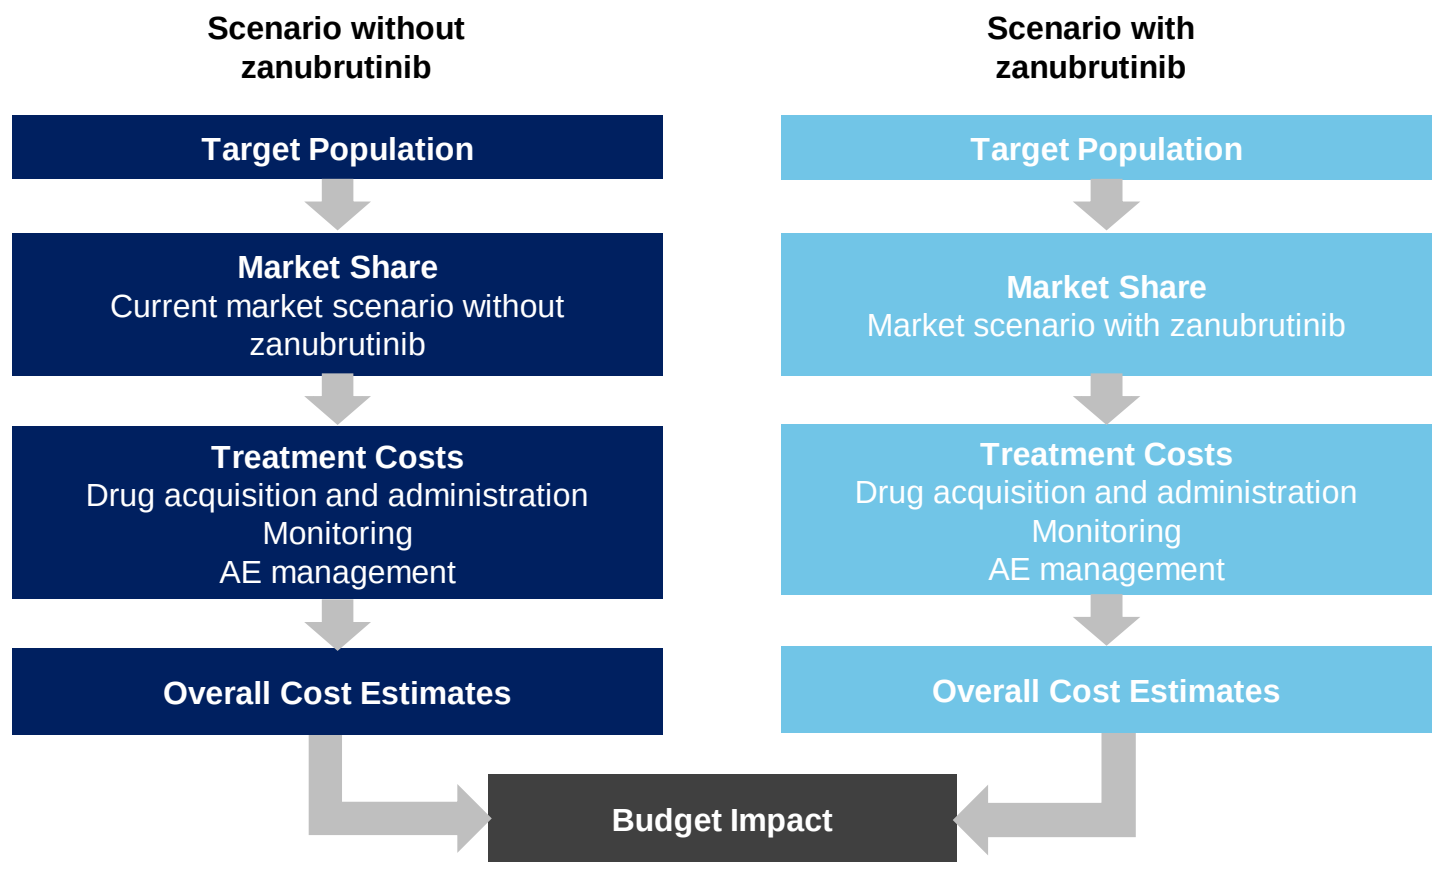
Scenario without zanubrutinib
Scenario with zanubrutinib
Target Population
Market Share
Current market scenario without zanubrutinib
Market scenario with zanubrutinib
Treatment Costs
Drug acquisition and administration
Monitoring
AE management
Overall Cost Estimates
Budget Impact

Abbreviations: AE – adverse event.

Table 1. Model Inputs for Treatment-Related Costs

Regimen	Zanubrutinib	Ibrutinib	BR	BDR	R-Mono
Drug Cost per Month	$13,693	$15,119	$5,464	$5,731	$6,623
Cost per Administration (Medicare/Commercial)	$0 / $0	$0 / $0	$214 / $632	$284 / $848	$144 / $417
Monitoring Cost per Month (Medicare/ Commercial)	$160 / $421	$160 / $421	$170 / $470	$170 / $470	$170 / $470
AE Management Cost per Month (Medicare/ Commercial)	$588 / $614	$507 / $525	$284 / $270	$357 / $354	$336 / $306

Abbreviations: AE – adverse events; BDR – bortezomib + dexamethasone + rituximab; BR – bendamustine + rituximab; R – rituximab.

Outputs

- The model reported outputs as the incremental total budget impact, incremental per-member-per-year (PMPY) and per-member-per-month (PMPM) budget impact, and per-treated-member-per-year (PTMPY) and per-treated-member-per-month (PTMPM) budget impact for the health plan (**Figure 2**)

Sensitivity Analysis

- A one-way sensitivity analysis was completed by varying each input by ±10% to determine which variables had the greatest impact on the results (**Figure 3**)

Results

Figure 2. Incremental Budget Impact by Year

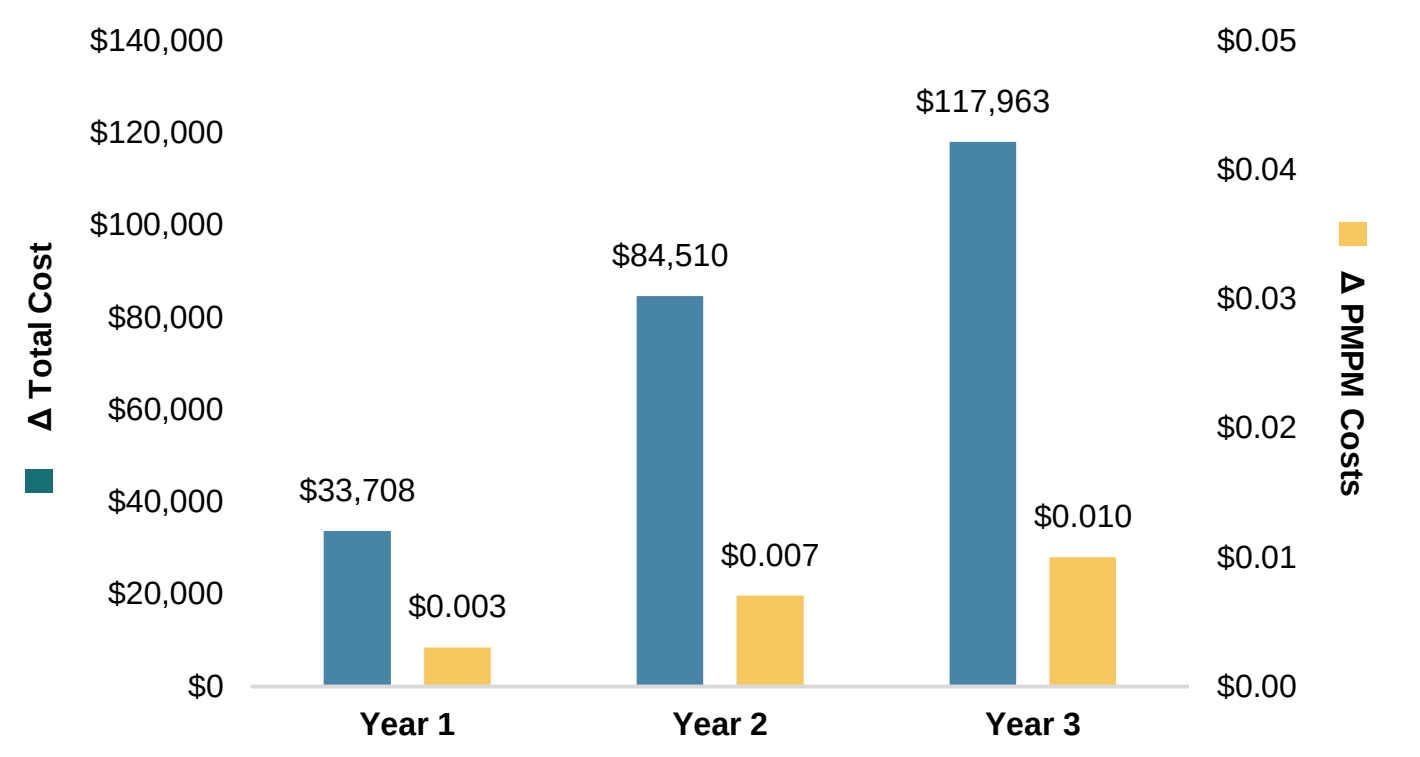
$140,000
$120,000
$100,000
$80,000
$60,000
$40,000
$20,000
$0
Δ Total Cost
$33,708
$84,510
$117,963
$0.003
$0.007
$0.010
Year 1
Year 2
Year 3
$0.05
$0.04
$0.03
$0.02
$0.01
$0.00
Δ PMPM Costs

Abbreviations: Δ – change; PMPM – per member per month

Results

Budget Impact

- In a 1-million-member payer plan, 5 patients with WM were estimated to be diagnosed and treated
- In the scenario without zanubrutinib, total cost results are $546,668 in year 1, $700,607 in year 2, and $704,360 in year 3
- In the scenario with access to zanubrutinib, total cost results are $580,375 in year 1, $785,117 in year 2, and $822,322 in year 3
- Over three years, the incremental budget impact of providing patient access to zanubrutinib was $33,708 ($0.003 PMPM) in year 1, $84,510 ($0.007 PMPM) in year 2, and $117,963 ($0.010 PMPM) in year 3 (**Figure 2**)

Sensitivity Analysis

- Results of the budget impact model were most sensitive to changes in the acquisition costs of zanubrutinib, ibrutinib + R, and BR (**Figure 3**)

Figure 3. One-Way Sensitivity Analysis for Total Budget Impact in Year 1

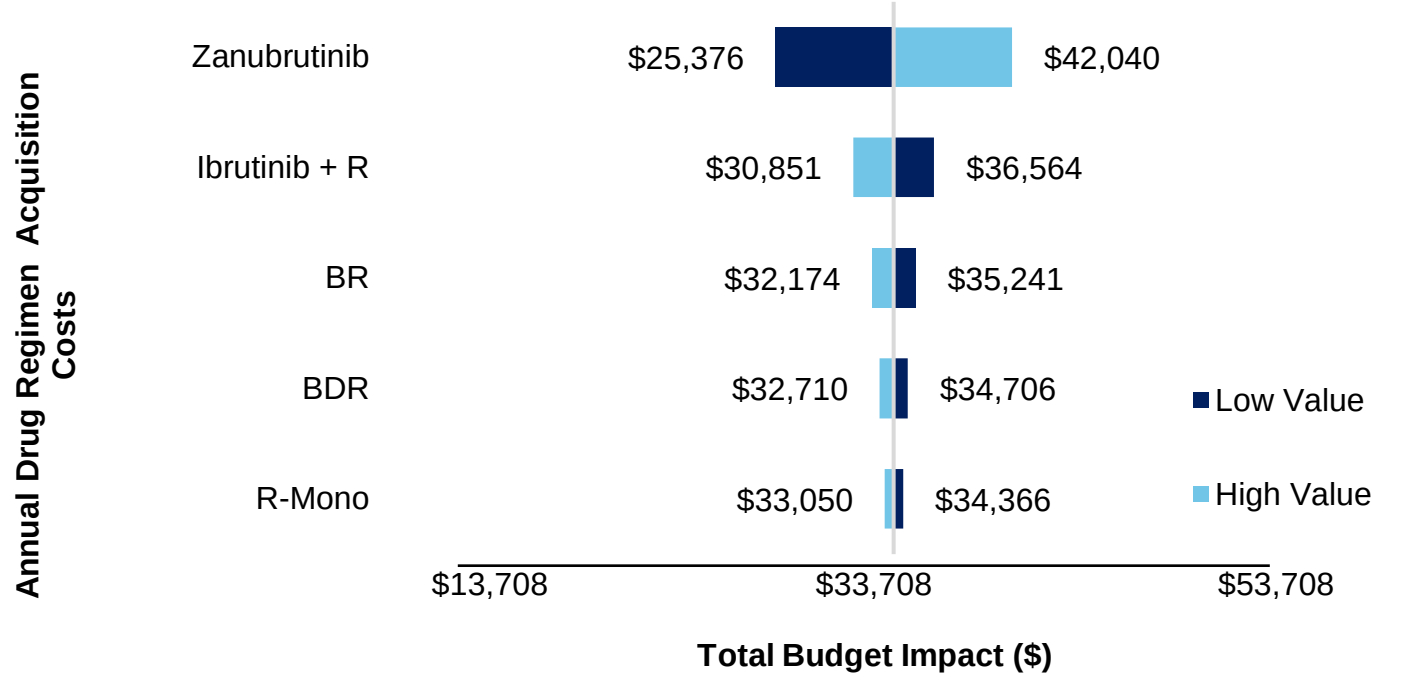
Annual Drug Regimen Acquisition Costs
Zanubrutinib
$25,376
$42,040
Ibrutinib + R
$30,851
$36,564
BR
$32,174
$35,241
BDR
$32,710
$34,706
R-Mono
$33,050
$34,366
Low Value
High Value
$13,708
$33,708
$53,708
Total Budget Impact ($)

Abbreviations: BR – bendamustine + rituximab; BDR – bortezomib + dexamethasone + rituximab; R – rituximab.

Limitations

- The population size and incidence of WM were assumed to remain constant over the modeled time horizon
 - This is not expected to have a large impact due to the relatively brief time horizon
- Incident patients were assumed to initiate treatment at the beginning of the year in which they were diagnosed and treated for the median duration reported in trial publications or prescribing information, which may not reflect real-world trends
- Default medication doses are based on recommended dosing and administration of their respective product labels, which may not reflect real-world treatment patterns, adherence, or persistence
- The model utilized inputs and assumptions to estimate budget impacts, and the generalizability to specific health plans with different treatment patterns or costs may be limited

Conclusion

- Zanubrutinib offers an important therapeutic option for the treatment of adult patients with WM
- Providing access to zanubrutinib for the treatment of patients with WM showed minimal budget impact on a US health plan over a three-year time horizon

Presented at the ISPOR 2022, Washington, DC, May 15–18,2022

References: 1. Owen RG, et al. *Semin Oncol*. 2003;30:110-115. 2. Pal Singh S, et al. *Mol Cancer*. 2018;17(1):57. 3. US FDA. FDA approves zanubrutinib for Waldenström's macroglobulinemia. Available at: https://www.fda.gov/drugs/resources-information-approved-drugs/fda-approves-zanubrutinib-waldenstroms-macroglobulinemia. Accessed April 3, 2022. 4. US Census. Available at: https://www.census.gov/data.html. Accessed July 22, 2021. 5. SEER cancer statistics review. Table 19.26 All lymphoid neoplasms with detailed non-Hodgkin lymphoma subtypes: all races, both sexes, 2008-2017. 6. Leukemia & lymphoma society. Waldenström Macroglobulinemia Facts. January 2012. Available at: https://www.lls.org/sites/default/files/_file_assets/waldenstrommacroglobulinemia.pdf. Accessed July 22, 2021. 7. Tam CS, et al. *Blood*. 2020;136(18):2038-2050. 8. RED BOOK® Online. IBM Watson. Accessed 26 August 2021. 9. Dimopoulos MA, et al. *N Engl J Med*. 2018;378(25):2399-2410. 10. Rummel MJ, et al. *Lancet*. 2013;381(9873):1203-1210. 11. Gavriatopoulou M, et al. *Blood*. 2017;129(4):456-459. 12. Paludo J, et al. *Br J Haematol*. 2017;179(1):98-105. 13. CMS 2021 Clinical Diagnostic Laboratory Fee Schedule. Available at: https://www.cms.gov/medicaremedicare-fee-service-paymentclinicallabfeeschedclinical-laboratory-fee-schedule-files/21clabq3. Accessed July 2, 2021. 14. National Fee Analyzer. Optum360; 2020. 15. Dimopoulos MA, et al. *Blood*. 2013;122(19):3276-3282. 16. Treon SP, et al. *Br J Haematol*. 2011;154(3):357-62. 17. Agency for Healthcare Research and Quality. Healthcare Cost and Utilization Project (HCUP). 2015. https://hcupnet.ahrq.gov. Accessed August 26, 2021. 18. United States Bureau of Labor Statistics. CPI for All Urban Consumers (CPI-U) for Medical Care 2000 to 2021. Available at: https://www.bls.gov/cpi/data.htm. Accessed July 28, 2021. 19. BeiGene data on file. WM market shares 2021.