Disease Burden and Treatment Satisfaction Among Patients with Chronic Inflammatory Demyelinating Polyneuropathy: A Quantitative Patient Survey

Charles Minor[1]; Mary Mulrooney[2]; Natalia Hakimi-Hawken[3]; Catherine Coulouvrat[4]; Jennifer Fine[4]; Daniel Shaffer[2]; Matthew O'Hara[2]

[1]Sanofi, Cambridge, MA, USA; [2]Trinity Life Sciences, Waltham, MA, USA; [3]Sanofi, Amsterdam, Netherlands; [4]Sanofi, Paris, France

Poster Number: PCR267

Copies of this poster obtained through QuickResponse (QR) Code are for personal use only

Introduction

- Chronic inflammatory demyelinating polyneuropathy (CIDP) is a rare, immune-mediated neurological disorder with an estimated global prevalence ranging from 0.7 to 10.3 cases per 100,000 across the literature[1-3]
- The heterogenous presentation of CIDP, together with overlapping characteristics, similar to other polyneuropathies, can make differential diagnosis of CIDP difficult[2,4]
- Despite available treatments (e.g., intravenous immunoglobulin [IVIG]), CIDP can present a substantial burden for patients due to their side effects and infusion requirements that can reduce independence and negatively impact quality of life (QoL)[2,5]

Objectives

- The objectives of this study were to capture patients' experience and satisfaction with currently available standard of care CIDP treatments (e.g., IG therapy, steroids, immunosuppressants, or rituximab) and the impact of CIDP on QoL

Methodology

- This was a cross sectional, observational study in adult patients diagnosed with CIDP in the United States (US)
- Patients were recruited through the Guillain-Barré Syndrome/CIDP Foundation International patient advocacy organization and an independent online patient research panel; data were collected between from August to November 2022
- A web-enabled survey was used to collect quantitative information on self-reported CIDP treatment experience and satisfaction across key treatment attributes (e.g., efficacy, durability of treatment response, accessibility, dosing frequency, ease and route of administration [RoA], and QoL)

Results

Patient Demographics

- In total, a representative sample of 71 patients completed the quantitative survey; among them, the average age was 52 years, 79% were female, and 86% were white (**Table 1**)

Table 1. Demographics and Characteristics

Characteristic	All patients(N=71)
Age in years, average (SD)	52 (12)
Female sex, n (%)	56 (79)
Race or Ethnicity, n (%)[a]	
White	61 (86)
Hispanic or Latinx	6 (8)
Black	4 (6)
Asian	4 (6)
Middle Eastern	1 (1)
Native American	1 (1)
Time since diagnosis in years, average (SD)	6 (4)
Diagnosing physician, n (%)[b]	
Neurologist	59 (83)
Neuromuscular specialist	9 (13)
Other[c]	3 (4)
Current disease severity, n (%)	
Mild	29 (41)
Moderate	33 (46)
Severe	9 (13)
Change in disease severity since diagnosis, n (%)	
Improved	25 (35)
Worsened	16 (22)
Both improved and worsened	26 (37)
Remained the same	4 (6)
Receiving caregiver support, n (%)	
Yes	32 (45)
No	39 (55)

Abbreviation: CIDP = chronic inflammatory demyelinating polyneuropathy. [a]Since patients could select more than one response, values may not add up to 100%. [b]Primary care physician, pulmonologist, and psychiatrist were omitted from the table as no patient selected them as their diagnosing physician. [c]'Other' responses included infectious disease specialist (n=2) and immunologist and neurologist (n=1).

CIDP Treatment Experience

- Most patients were currently receiving IVIG (n=53, 75%), followed by subcutaneous IG (n=17, 24%; **Table 2**)
- Over half of patients (n=38, 54%) surveyed reported a lack of response to prior therapy or residual disability despite prior treatments

Table 2. CIDP Treatment Experience

Characteristic	All patients (N=71)
Primary treating physician for CIDP, n (%)[a]	
Neurologist	58 (82)
Neuromuscular specialist	5 (11)
Psychiatrist	1 (1)
Other[b]	4 (6)
Current chronic treatment for CIDP, n (%)[c]	
IVIG	53 (75)
SCIG	17 (24)
Oral steroids	12 (17)
IV steroids	12 (17)
Immunosuppressants	9 (13)
Rituximab	6 (8)
Plasmapheresis/plasma exchange	5 (7)
None	4 (6)
Other[d]	6 (8)
First treatment for CIDP, n (%)	
IVIG	50 (76)
Oral steroids	14 (20)
IV steroids	12 (17)
SCIG	7 (10)
Immunosuppressants	5 (7)
Rituximab	2 (3)
Well-controlled on IVIG/SCIG, n (%)[e]	
Yes (IG responders)	33 (46)
No (refractory to IG)	38 (54)

Abbreviations: CIDP = chronic inflammatory demyelinating polyneuropathy; IG = immunoglobulin; IVIG = IV immunoglobulin; SCIG = subcutaneous immunoglobulin. [a]Primary care physician and pulmonologist were omitted from the table as no patient selected them as their primary CIDP-treating physician. [b]'Other' responses included immunologist (n=2), physiologist (n=1), and immunologist and neurologist (n=1). [c]Patients could select more than one response. [d]'Other' responses included gabapentin (n=2), neurontin, tramadol (n=1), chemo (cytotoxin) (n=1), cymbalta and gabapentin (n=1), cell cept (n=1). [e]Patients were characterized as 'refractory to IG' if they had received IG treatment in the past that did not work or perform as expected, or were currently receiving IG treatment but were looking for alternative chronic treatments due to lack of controlled symptoms or disease progression; patients were characterized as 'IG Responders' if they were currently receiving and well-controlled on IG treatment, were not looking for alternative chronic treatments, and had not received IG treatment in the past.

Results

Satisfaction With Current CIDP Treatment

- Less than half of patients rated high satisfaction (≥6 points on 1-7 scale) with any attribute of their current CIDP treatment, including only 34% and 30% of patients satisfied with the efficacy and durability of treatment response, respectively (**Figure 1**)

Figure 1. CIDP Treatment Attribute Importance and Satisfaction with Current Treatment Attributes

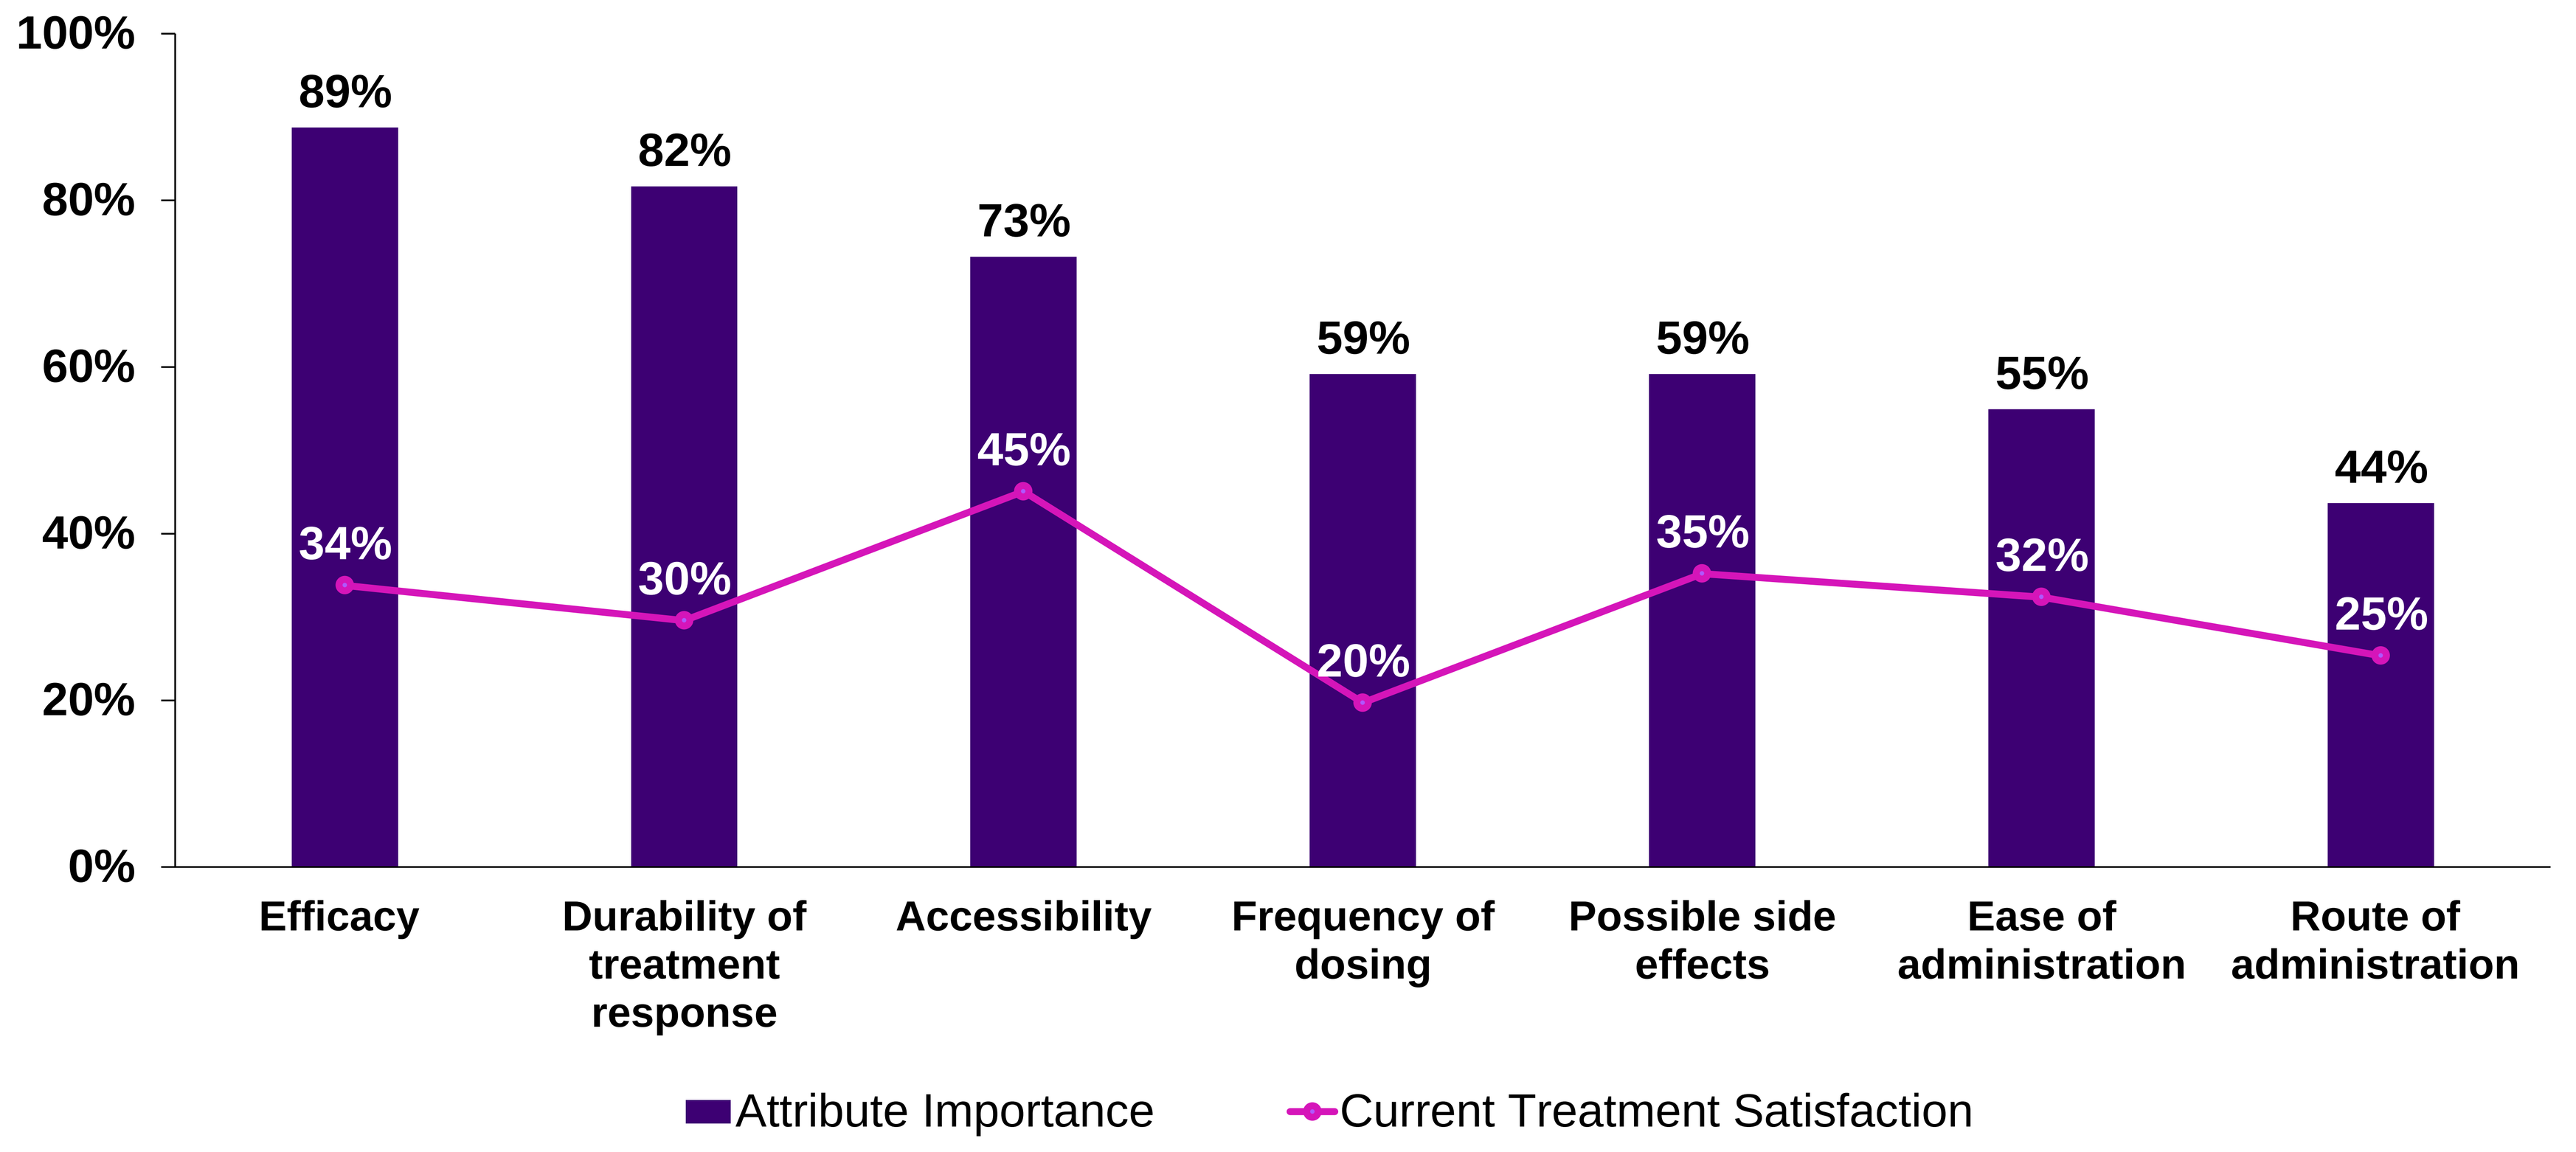
100%
80%
60%
40%
20%
0%
89%
82%
73%
59%
59%
55%
44%
34%
30%
45%
20%
35%
32%
25%
Efficacy
Durability of treatment response
Accessibility
Frequency of dosing
Possible side effects
Ease of administration
Route of administration
Attribute Importance
Current Treatment Satisfaction

Abbreviation: CIDP = chronic inflammatory demyelinating polyneuropathy.

Effect of CIDP on Quality of Life

- Two-thirds of patients surveyed (n=48, 68%) reported that CIDP had a strong to significant impact on their QoL (≥6 points; **Figure 2A**)
- Across activities of daily living, ambulation was the most affected by CIDP (n=57, 80% indicated at least some impact; ≥3 points), followed by personal hygiene (n=46, 65%), dressing (n=41, 58%), continence (n=33, 46%), toileting (n=31, 44%), and feeding (n=27, 38%; **Figure 2B**)

Figure 2. Effect of CIDP on Quality of Life and Ability to Perform Activities of Daily Living

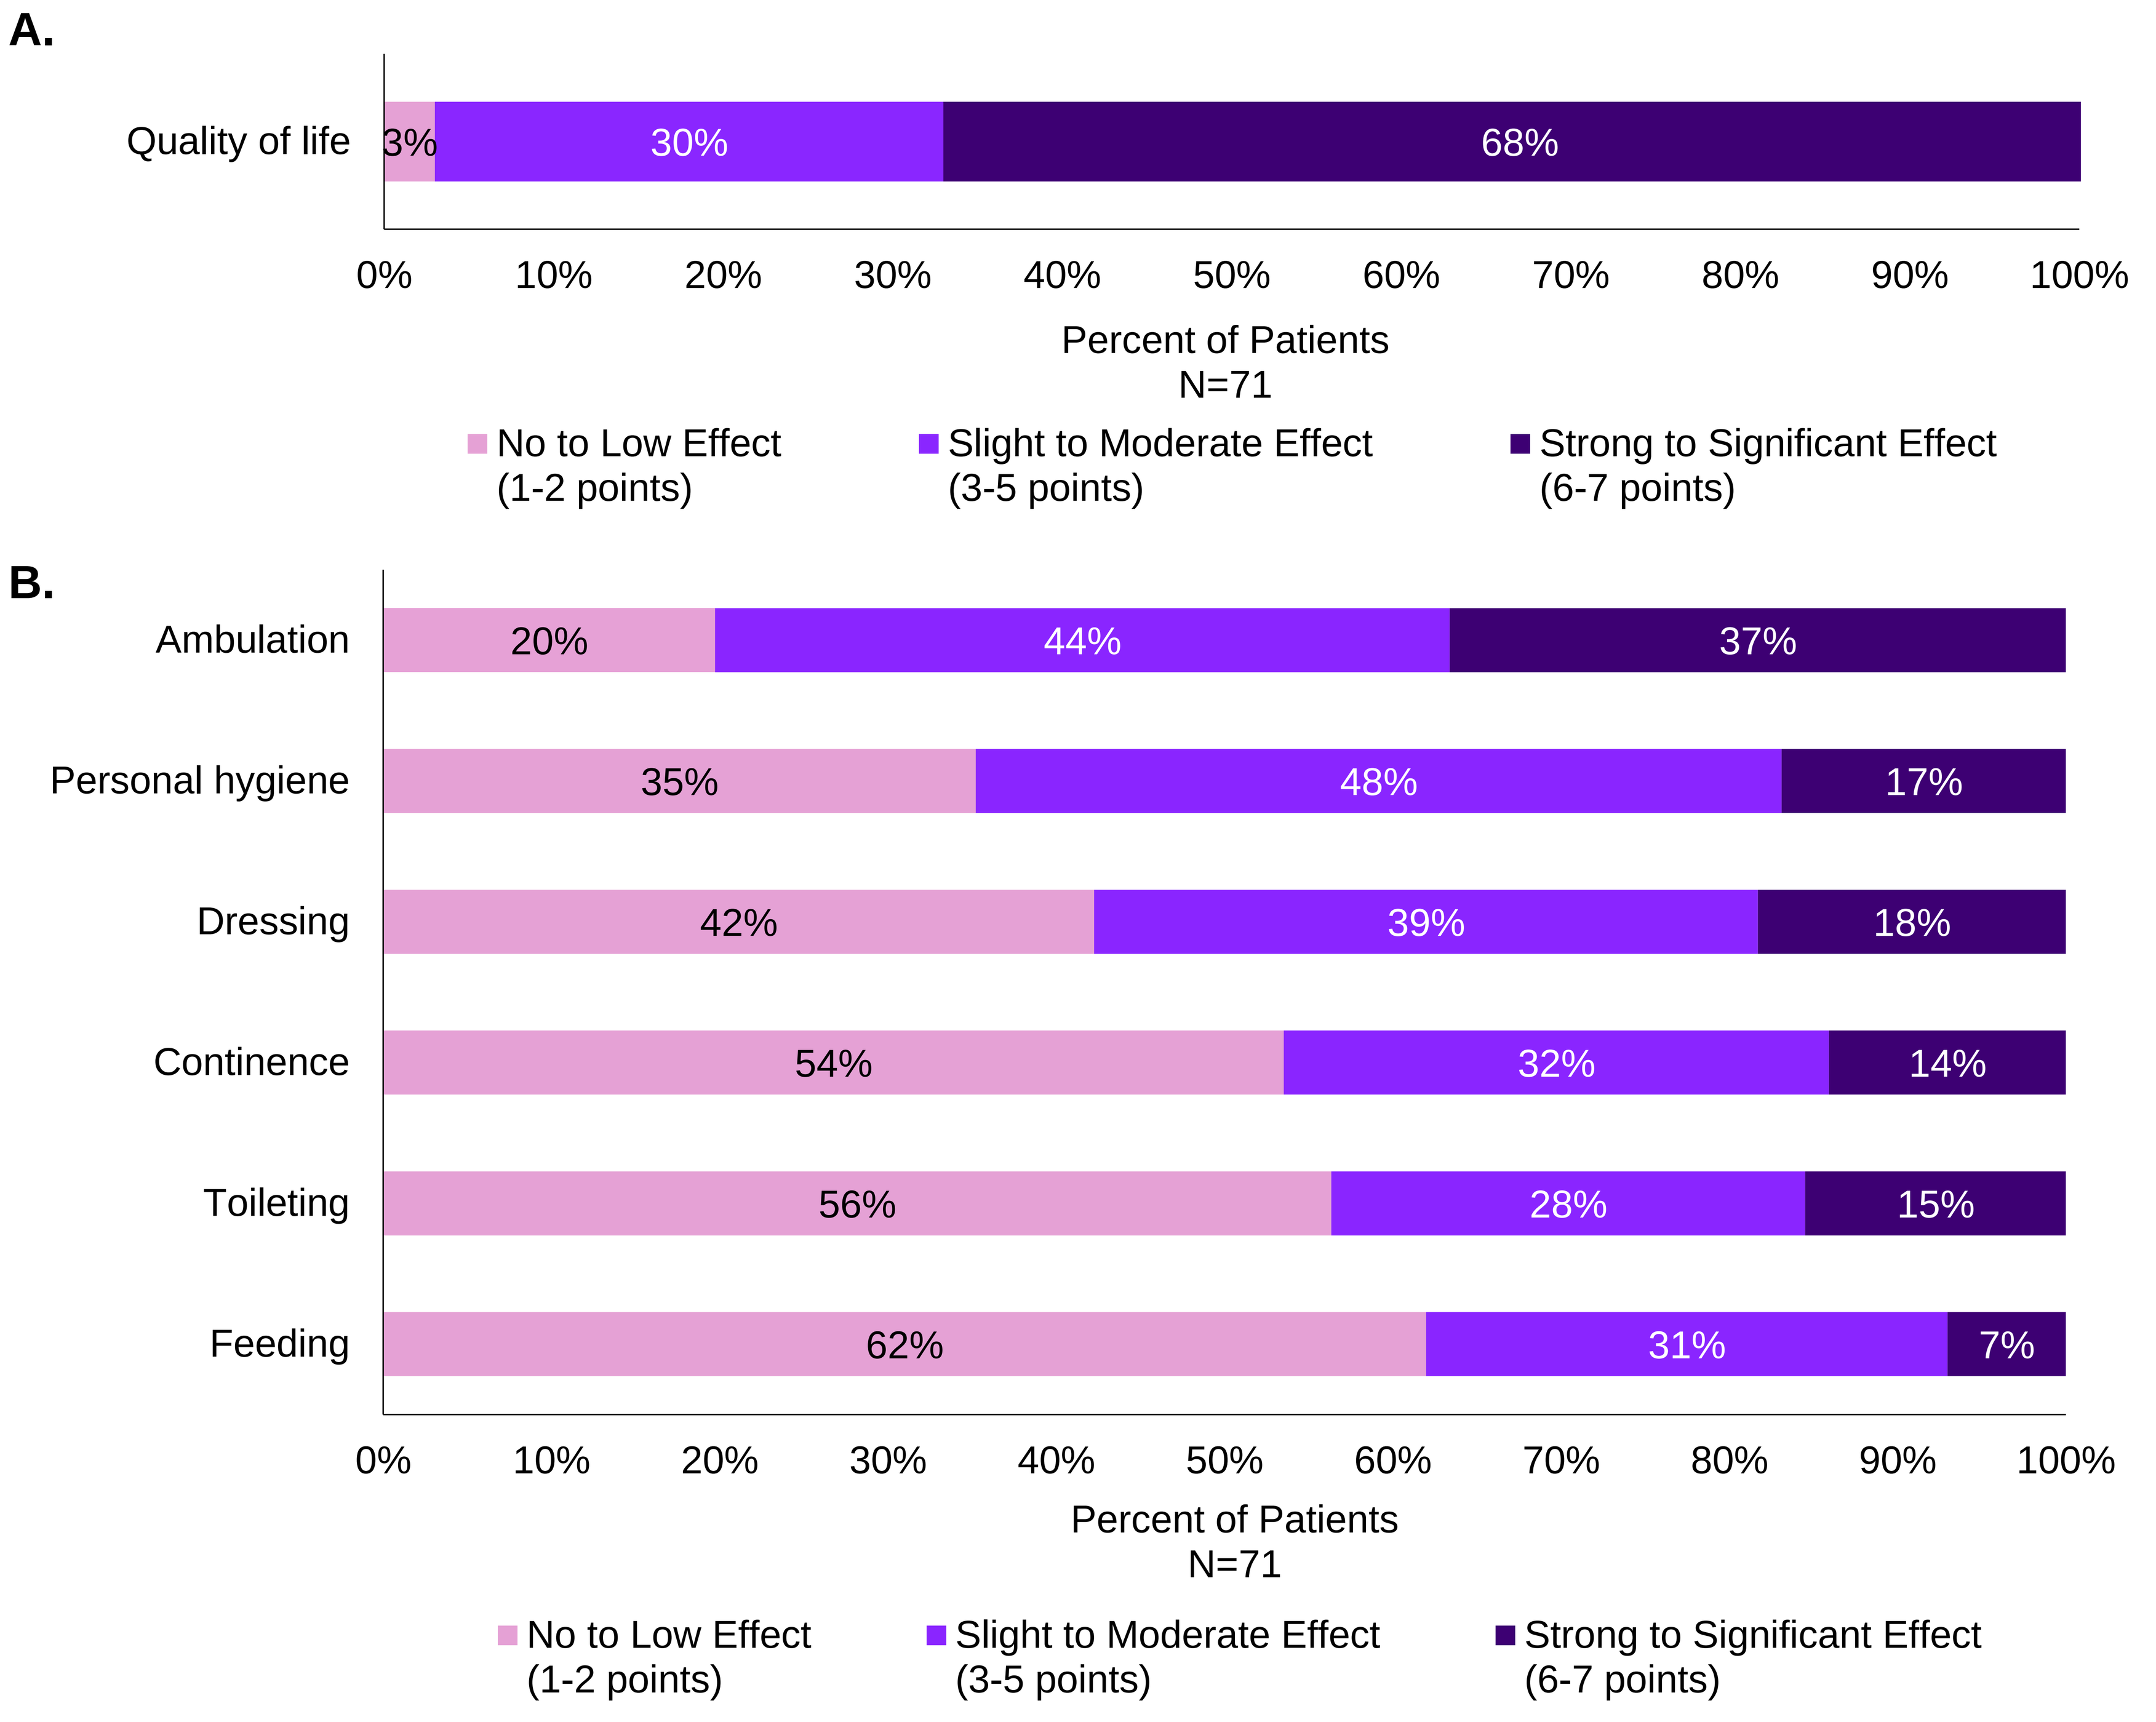
A.
Quality of life
3%
30%
68%
0%
10%
20%
30%
40%
50%
60%
70%
80%
90%
100%
Percent of Patients
N=71
No to Low Effect (1-2 points)
Slight to Moderate Effect (3-5 points)
Strong to Significant Effect (6-7 points)
B.
Ambulation
20%
44%
37%
Personal hygiene
35%
48%
17%
Dressing
42%
39%
18%
Continence
54%
32%
14%
Toileting
56%
28%
15%
Feeding
62%
31%
7%
0%
10%
20%
30%
40%
50%
60%
70%
80%
90%
100%
Percent of Patients
N=71
No to Low Effect (1-2 points)
Slight to Moderate Effect (3-5 points)
Strong to Significant Effect (6-7 points)

Abbreviations: CIDP = chronic inflammatory demyelinating polyneuropathy;

Conclusions

- **Findings from this study underscore the considerable unmet needs and burden of CIDP in the US, including moderate to low satisfaction with current treatments, and the strong negative effect on QoL**
- **Additional research is needed to understand how future therapies can alleviate the disease and management burden of CIDP**

REFERENCES

1. Saperstein DS, Katz JS, Amato AA, Barohn RJ. Clinical spectrum of chronic acquired demyelinating polyneuropathies. *Muscle Nerve*. 2001;24(3):311-324.
2. Van den Bergh PY, Hadden RD, Bouche P, et al. European Federation of Neurological Societies/Peripheral Nerve Society guideline on management of chronic inflammatory demyelinating polyradiculoneuropathy: report of a joint task force of the European Federation of Neurological Societies and the Peripheral Nerve Society - first revision [published correction appears in *Eur J Neurol*. 2011 May;18(5):796]. *Eur J Neurol*. 2010;17(3):356-363.
3. Broers MC, Bunschoten C, Nieboer D, Lingsma HF, Jacobs BC. Incidence and Prevalence of Chronic Inflammatory Demyelinating Polyradiculoneuropathy: A Systematic Review and Meta-Analysis. *Neuroepidemiology*. 2019;52(3-4):161-172.
4. Eftimov F, Lucke IM, Querol LA, Rajabally YA, Verhamme C. Diagnostic challenges in chronic inflammatory demyelinating polyradiculoneuropathy. Brain. 2020;143(11):3214-3224.
5. Allen JA, Gelinas DF, Freimer M, Runken MC, Wolfe GI. Immunoglobulin administration for the treatment of CIDP: IVIG or SCIG?. *J Neurol Sci*. 2020;408:116497.

FUNDING DISCLOSURES

This study was funded by Sanofi US Services Inc.

ACKNOWLEDGEMENT

The authors would like to thank all the patient groups and organizations that responded to the survey.

CONFLICT OF INTERESTS

CM, NHH, CC, and JF are employees of Sanofi and hold stock or stock options of Sanofi.

International Society for Pharmacoeconomics and Outcomes Research (ISPOR)-EU 2024 (November 17-20, 2024), Barcelona, Spain