Budget impact of Tezepelumab for the treatment of Severe Asthma in the Brazilian Private Healthcare System

Poster Code: EE28

AUTHORS:

Diegoli H[1], Dias C[2], Furtado D[3]

[1]Academia VBHC, São Paulo, Brazil; [2]AstraZeneca do Brasil, Cotia, Brazil; [3]Unimed Nova Friburgo

OBJECTIVES:

Asthma, a chronic inflammatory condition of the airways, affects about 20 million people in Brazil, with 5%–10% of cases classified as severe, leading to significant morbidity and high healthcare utilization. Although with a smaller number of patients, severe asthma represents the majority of healthcare resource utilization in asthma. Biologic drugs have transformed the management of severe asthma. In Brazil, current medicines are available as omalizumab for severe allergic asthma (SAA), mepolizumab and benralizumab for severe eosinophilic asthma (SEA) and dupilumab for for T2-high asthma or oral corticosteroid-dependent asthma. However, unmet needs persist in patients with severe asthma. The PATHFINDER clinical program has demonstrated the effectiveness and safety of tezepelumab for severe asthma, irrespective of phenotype or biomarker. This study aims to estimate the budget impact (BI) of incorporating tezepelumab into the Brazilian supplementary health system.

METHODS:

The Budget Impact (BI) analysis was conducted over a 5-year period from the perspective of the private healthcare system. Eligible patients included those with uncontrolled severe asthma, encompassing both severe eosinophilic asthma (SEA) and severe allergic asthma (SAA). The total population was defined by epidemiological parameters, including the prevalence of severe asthma, the proportion of uncontrolled disease despite the use of inhaled corticosteroids and Long- Acting Beta-Agonists (IC + LABA), and the population covered by the Brazilian supplementary healthcare system. Acquisition costs for tezepelumab and its comparators were sourced from Brazil's Medicines Market Regulation Chamber (CMED), inclusive of taxes. The projected market share of treatments was determined based on market research. A deterministic sensitivity analysis was performed, varying the parameters by ±20%. Additionally, different scenarios, including variations in the rate of incorporation and market share, were estimated. The study adhered to the Brazilian Guidelines for Budget Impact Analyses.

Table 1. Drug costs utilized in the model.

Drug	Price (BRL)	First year		Subsequent years	
		Doses	Annual cost (BRL)	Doses	Annual cost (BRL)
Tezepelumab	9,428	13	122,558	13	122,558
Dupilumab	4,714	27	127,272	26	122,559
Omalizumab	Variable	Variable		Variable	
Benralizumab	14,994	8	119,951	6,5	97,460
Mepolizumab	9,456	13	122,934	13	122,934

Table 2. Omalizumab costs utilized in the model.

Dose (vials)	Proportion of patients
150mg/Q4W (1)	11.5%
300mg/Q4W (2)	23.1%
450mg/Q4W (3)	22.4%
600mg/Q4W (4)	15.0%
375mg/Q2W (5)	7.7%
450mg/Q2W (6)	11.7%
525mg/Q2W (7)	4.7%
600mg/Q2W (8)	4.0%
Cost per vial	R$ 2,738.63
Average 4-weeks cost	R$ 9.817,99
Average cost per year	R$ 127,633.85

Table 3. Number of eligible patients for treatment with tezepelumab.

Year	Eligible patients		
	SAA	SEA	Total
1	3680	2453	6133
2	3710	2474	6184
3	3739	2493	6232
4	3766	2511	6276
5	3791	2527	6318

SEA: severe eosinophilic asthma; SAA: severe allergic asthma.

Figure 2. Deterministic sensitivity analysis of the 5-year budget impact with the incoporation of tezepelumab.

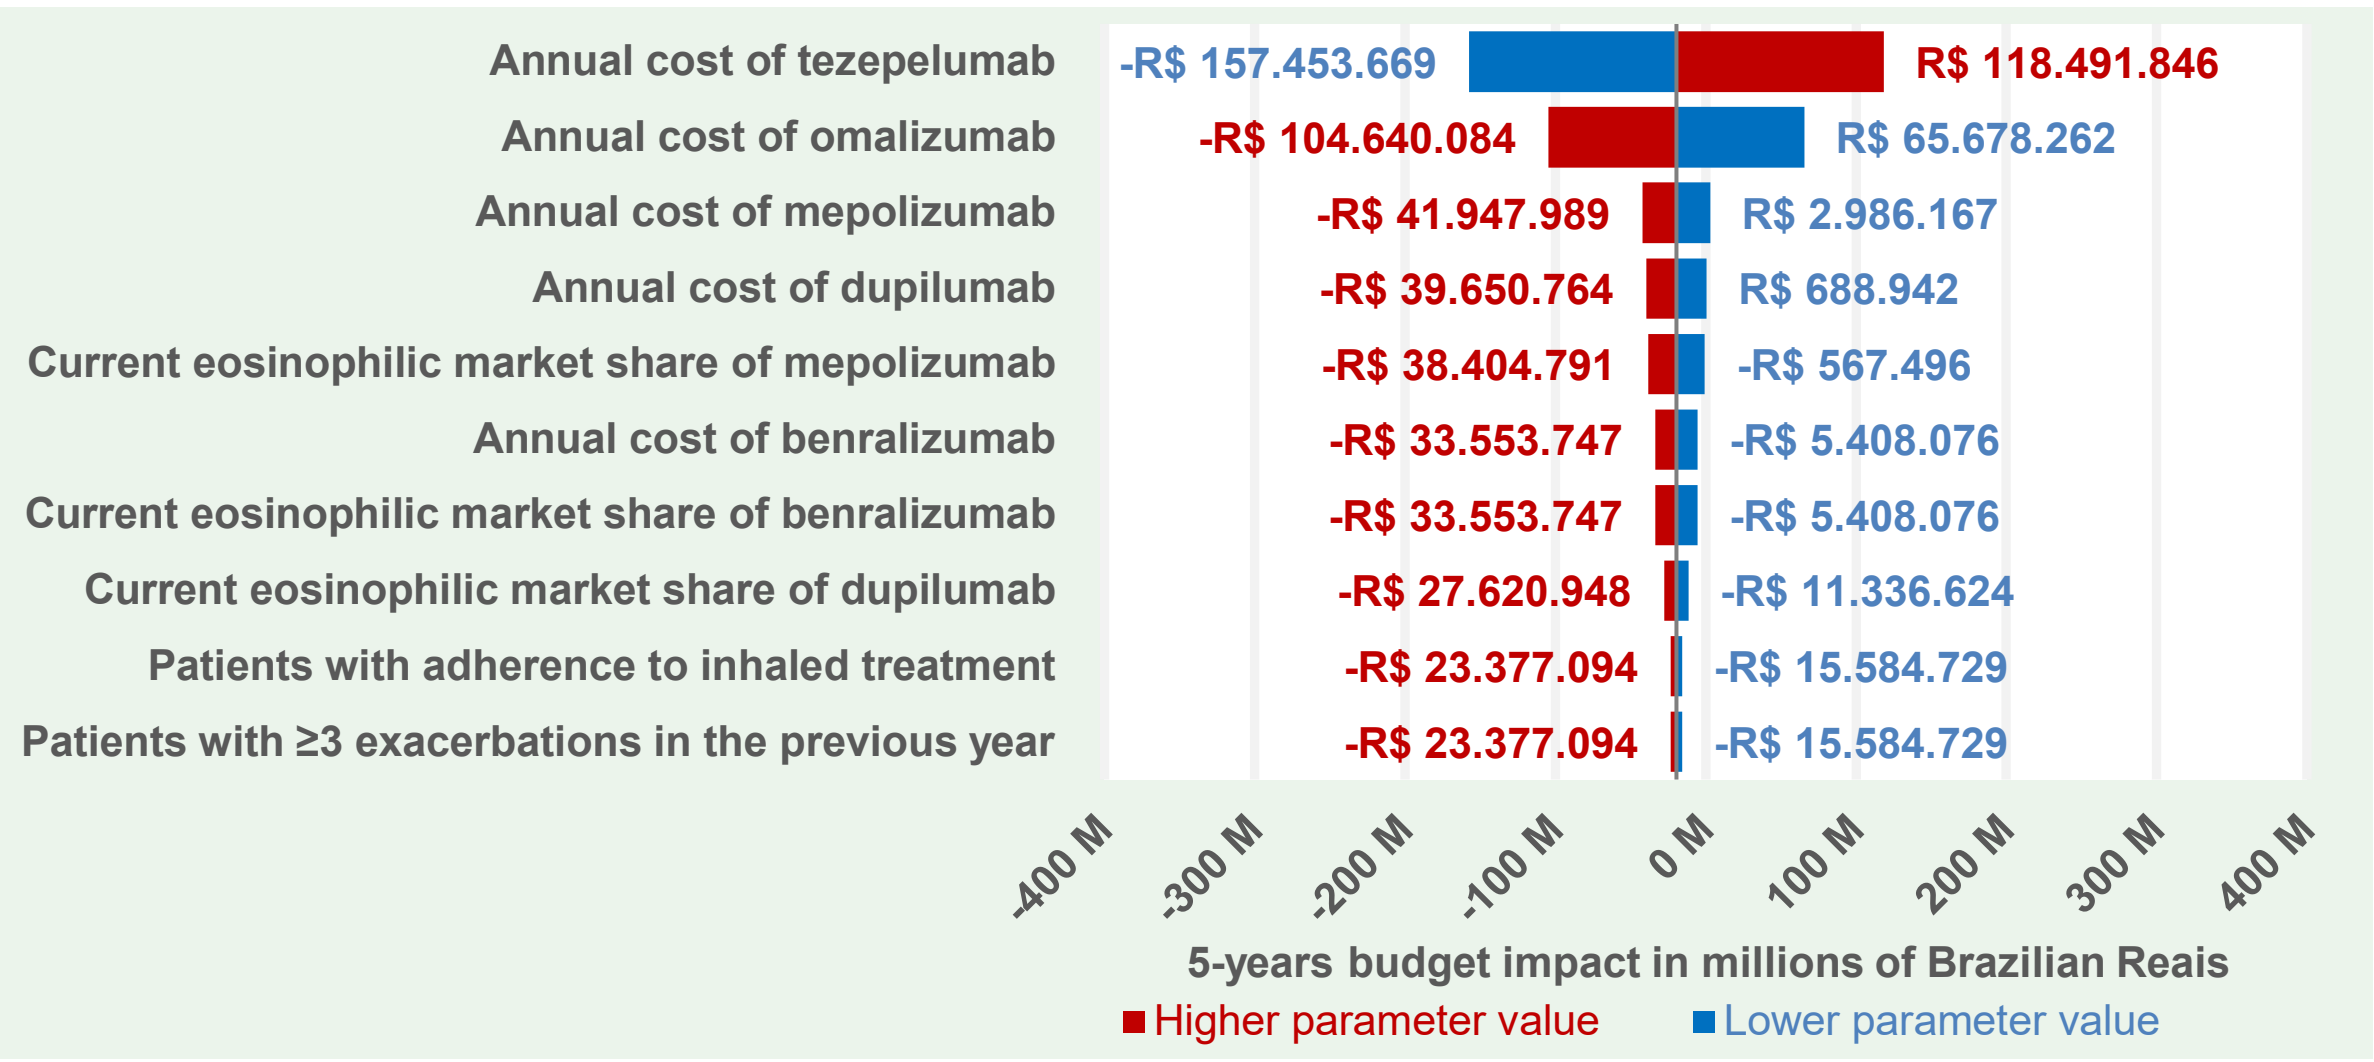
Annual cost of tezepelumab
-R$ 157.453.669
R$ 118.491.846
Annual cost of omalizumab
-R$ 104.640.084
R$ 65.678.262
Annual cost of mepolizumab
-R$ 41.947.989
R$ 2.986.167
Annual cost of dupilumab
-R$ 39.650.764
R$ 688.942
Current eosinophilic market share of mepolizumab
-R$ 38.404.791
-R$ 567.496
Annual cost of benralizumab
-R$ 33.553.747
-R$ 5.408.076
Current eosinophilic market share of benralizumab
-R$ 33.553.747
-R$ 5.408.076
Current eosinophilic market share of dupilumab
-R$ 27.620.948
-R$ 11.336.624
Patients with adherence to inhaled treatment
-R$ 23.377.094
-R$ 15.584.729
Patients with ≥3 exacerbations in the previous year
-R$ 23.377.094
-R$ 15.584.729
-400 M
-300 M
-200 M
-100 M
0 M
100 M
200 M
300 M
400 M
5-years budget impact in millions of Brazilian Reais
Higher parameter value
Lower parameter value

Figure 1. Five-year budget impact: base case without tezepelumab incorporation, base case with incorporation, and scenarios with faster and slower incorporation rates.

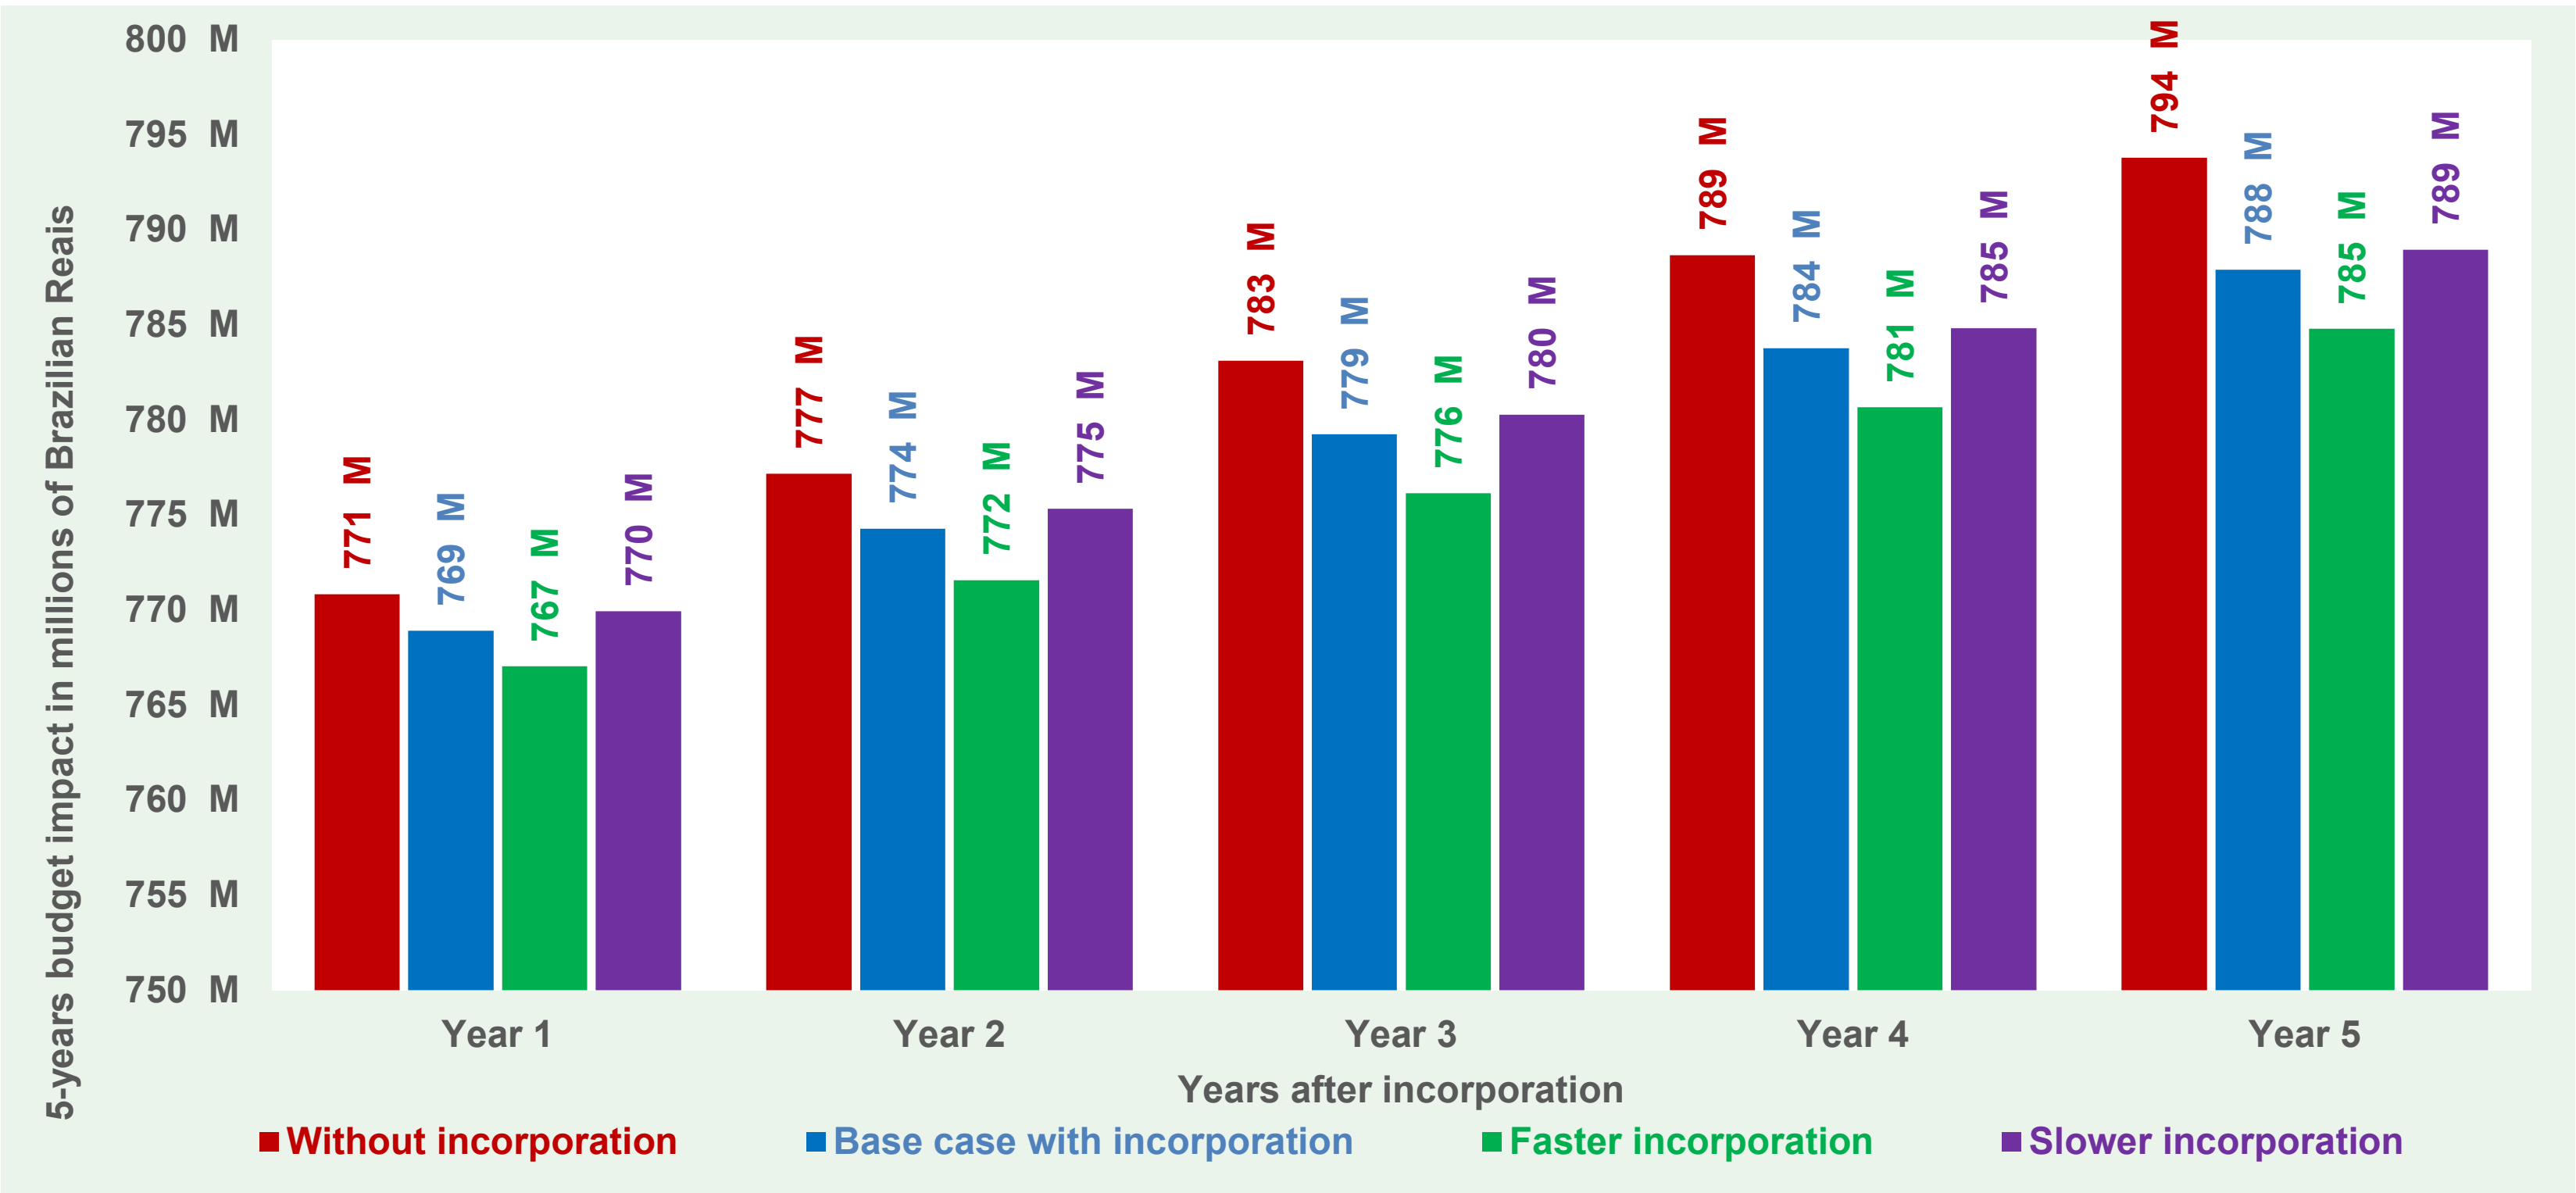
5-years budget impact in millions of Brazilian Reais
800 M
795 M
790 M
785 M
780 M
775 M
770 M
765 M
760 M
755 M
750 M
771 M
769 M
767 M
770 M
777 M
774 M
772 M
775 M
783 M
779 M
776 M
780 M
789 M
784 M
781 M
785 M
794 M
788 M
785 M
789 M
Year 1
Year 2
Year 3
Year 4
Year 5
Years after incorporation
Without incorporation
Base case with incorporation
Faster incorporation
Slower incorporation

RESULTS:

The estimated eligible population increased from 6,133 to 6,318 over 5 years. In the base case scenario, the incorporation of Tezepelumab resulted in 491 to 1,769 patients receiving the treatment over 5 years, with a cost reduction of -BRL 19,480,911 over the period.The cost reduction was greater in SAA (-BRL 18,888,169) than in SEA (-BRL 592,742). In sensitivity analyses, the cost of tezepelumab followed by cost of omalizumab had the highest sensitivity. A more rapid incorporation (920 to 2,691 patients) resulted in a total cost reduction of -BRL 33,385,321. The cost analysis revealed that the expenses for tezepelumab and dupilumab remain constant in the subsequent years following the first year of treatment. Among all the biologic medications evaluated, benralizumab was considered the least expensive medication over the years, followed by tezepelumab.

CONCLUSION:

This analysis underscores the importance of considering tezepelumab as a treatment option within the Brazilian supplementary health system, given its association with a substantial reduction in medical costs. The findings highlight not only the clinical benefits of tezepelumabe but also its potential to reduce overall healthcare expenditures.