Incidence and prevalence of progressive pulmonary fibrosis (PPF) other than idiopathic pulmonary fibrosis: a systematic literature review and meta-analysis

EPH261

Golchin N,[1] Lesperance T,[1] Scheuring J,[1] Wan V,[2] Hofer K,[2] Collet JP,[2] Elpers B,[1*] Patel A[1]

[1]Bristol Myers Squibb, Princeton, New Jersey, USA; [2]Evidinno Outcomes Research Inc., Vancouver, British Columbia, Canada

*At the time of the analysis

Introduction

- Among patients with interstitial lung disease (ILD), the development of progressive pulmonary fibrosis (PPF) is associated with a severe prognosis
 - PPF is associated with well-known risk factors (e.g., rheumatoid arthritis, sarcoidosis) or the evolution of idiopathic pulmonary fibrosis (IPF)
- Established criteria for non-IPF PPF have only recently been published in clinical practice guidelines[1]; clear epidemiological information is scarce

Objectives

- To estimate the incidence and prevalence of non-IPF PPF within adults globally and across regions

Methods

Systematic literature review

- Methods adhered to standard methodologies for conducting and reporting of systematic literature reviews[2,3,4]
 - Study eligibility criteria were defined using the CoCoPop framework (Condition, Context, Population)[3]
 - Included were English-language studies reporting incidence or prevalence of non-IPF PPF in adults (≥18 years) within the general population of any country
- MEDLINE®, Embase, and Cochrane Database of Systematic Reviews were searched from Jan 1, 2000, to Nov 7, 2023 via OvidSP using pre-defined search strategies
 - Bibliographies of relevant literature reviews were also searched, as were abstracts from the American Thoracic Society, British Thoracic Society, European Respiratory Society, Canadian Society of Respiratory Therapists, American College of Rheumatology, and European Congress of Rheumatology (2021 – 2023)
- Quality of the included studies was assessed via the Joanna Briggs Institute checklist for prevalence studies[5]

Statistical analysis

- Where necessary, data provided by included studies were used to calculate denominators, numerators, and 95% confidence intervals (CIs)
 - In studies reporting annual incidence/prevalence, a weighted average was calculated over study period; point estimates were analyzed as reported
 - Adjusted rates were preferred over crude rates
 - For studies that reported incidence or prevalence by year or sex, values were averaged according to population size for each year or sample size for each sex to produce a single rate per study
- Meta-analysis using the DerSimonian-Laird random-effects model, a version of the inverse variance method[2,6]
- Pooled weighted random-effects incidence and prevalence estimates were generated using the metafor R package[7]

Results

Study Selection

- 3,823 records were identified from databases (**Figure 1**); 6 studies were included in the review[8-13]
 - One study was only available as a conference abstract[13]; authors reported incidence of acute interstitial pneumonitis, and total sample size was not reported; therefore, this study was excluded from the meta-analysis
- Five studies were ultimately included in the meta-analysis[8-12]

Figure 1. PRISMA diagram of study selection

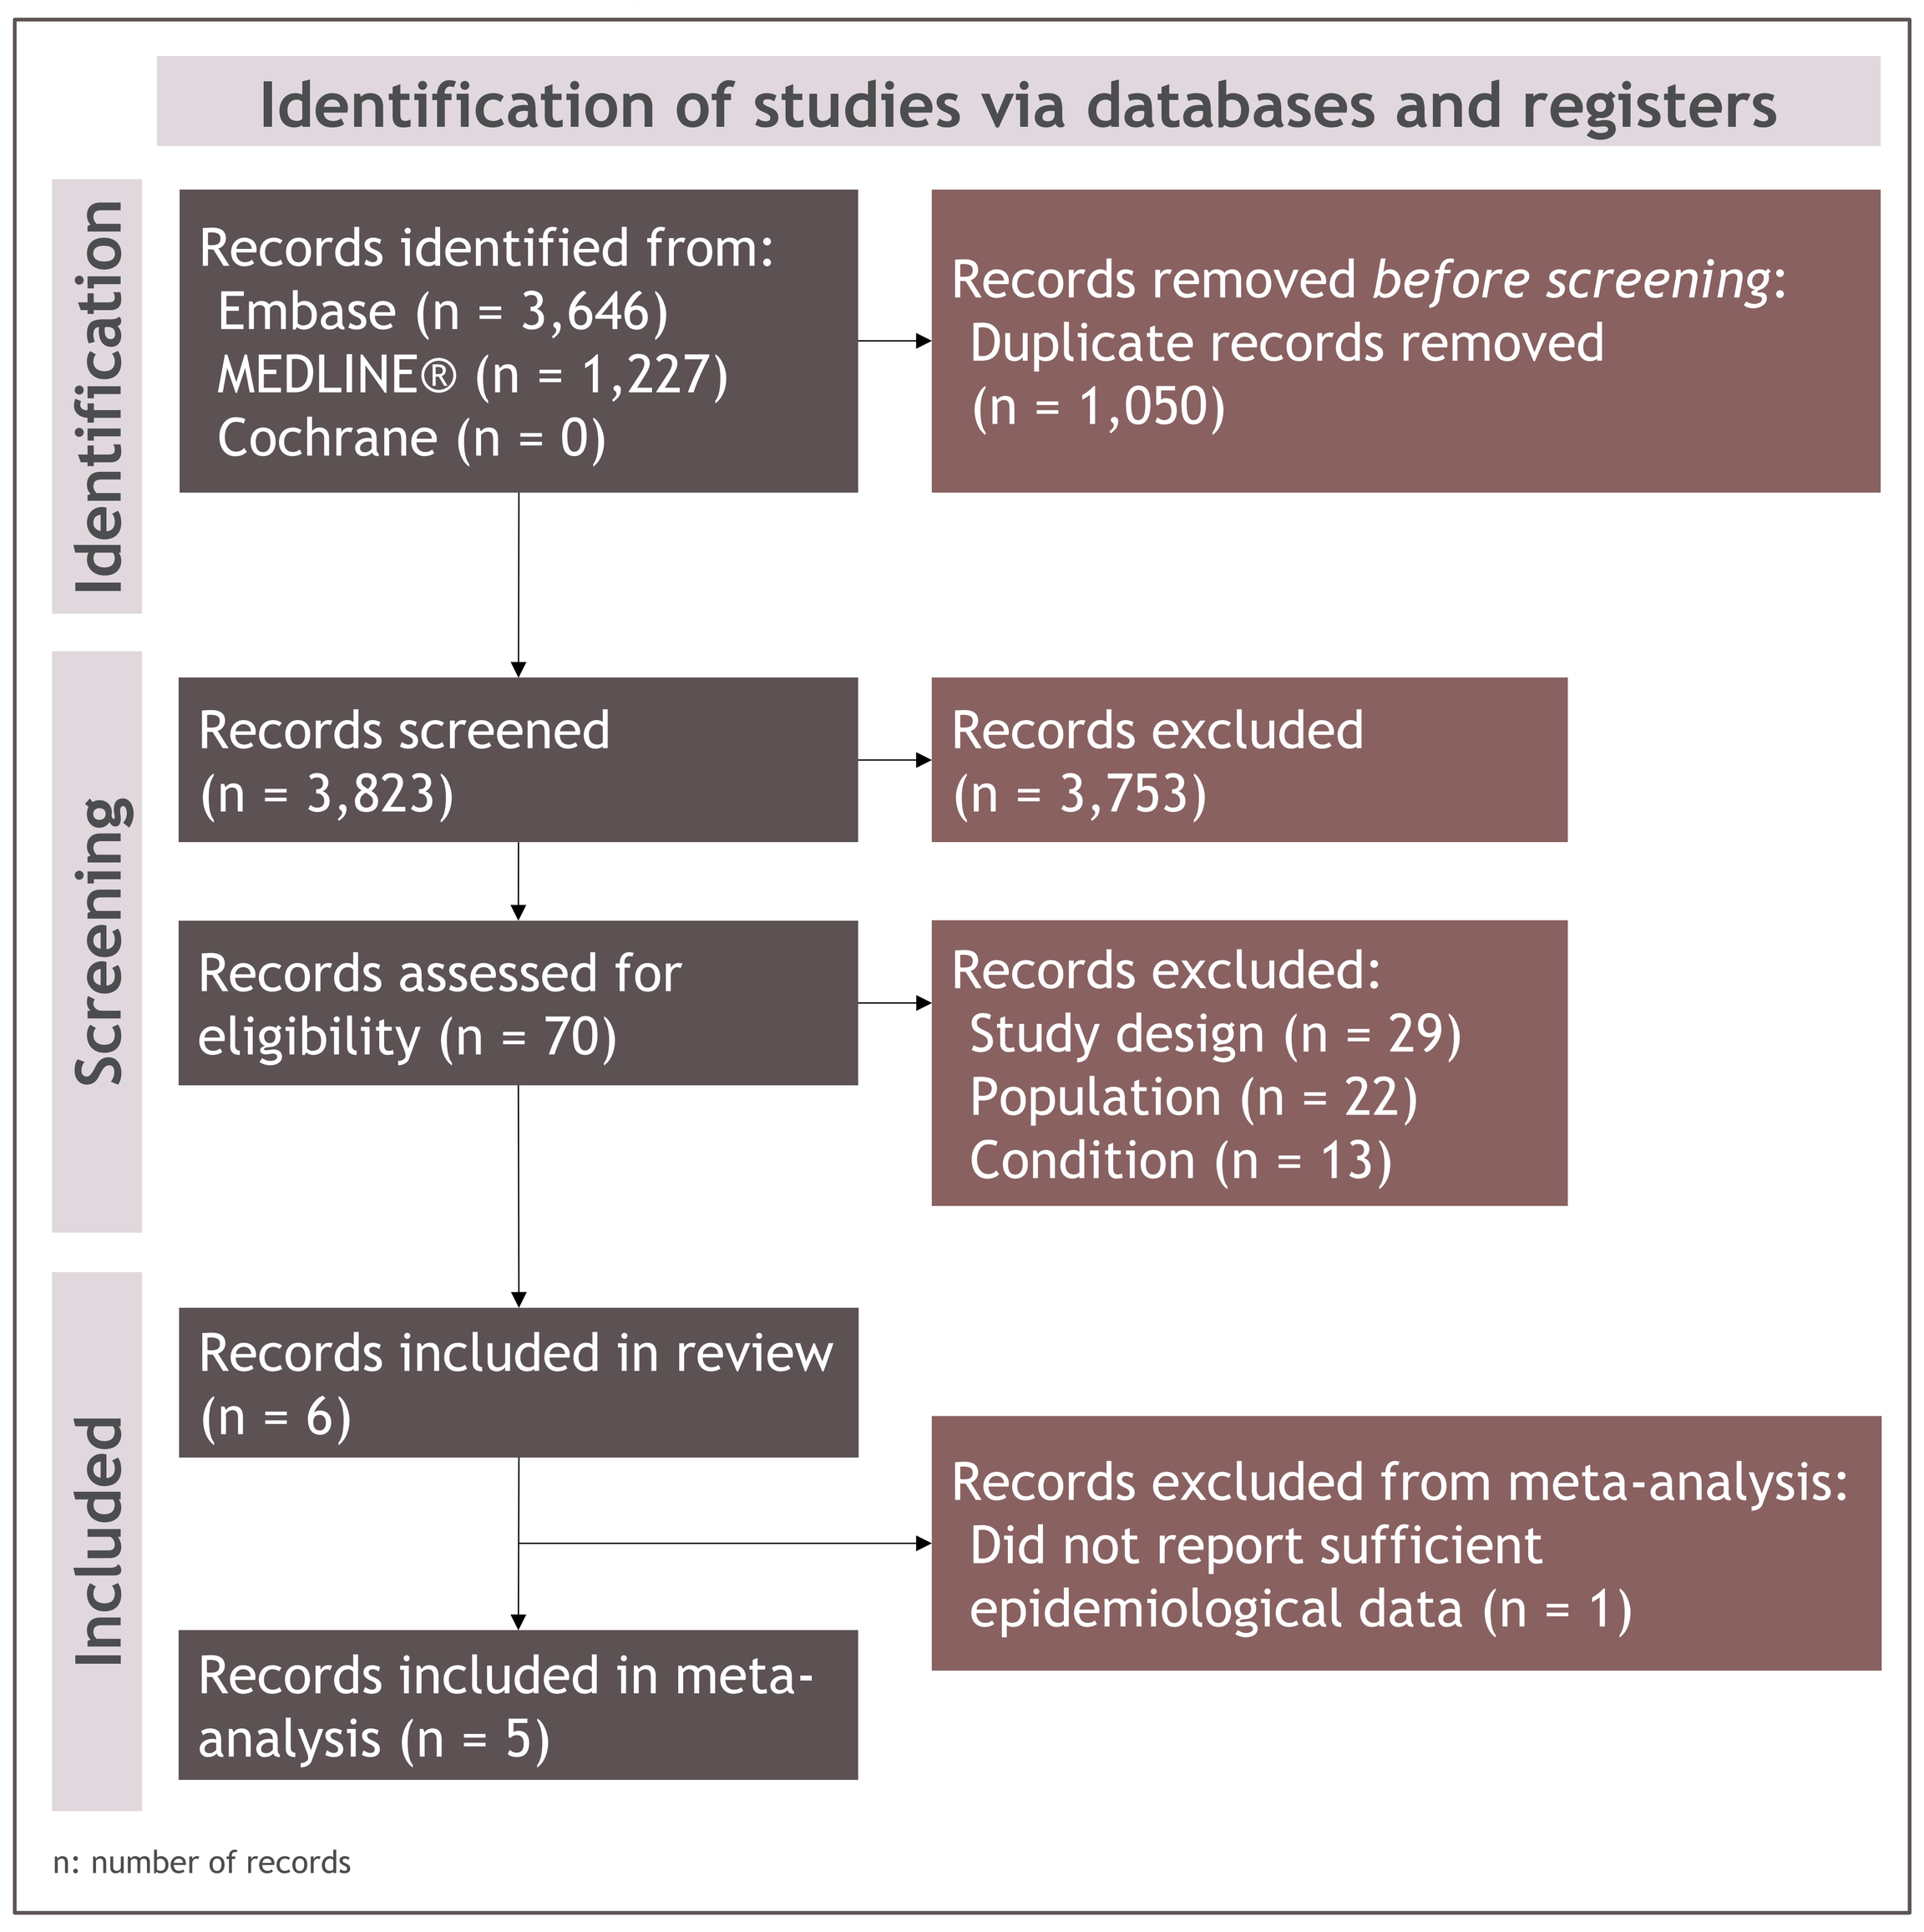

Identification of studies via databases and registers
Identification
Screening
Included
Records identified from: Embase (n = 3,646) MEDLINE® (n = 1,227) Cochrane (n = 0)
Records removed *before screening*: Duplicate records removed (n = 1,050)
Records screened (n = 3,823)
Records excluded (n = 3,753)
Records assessed for eligibility (n = 70)
Records excluded: Study design (n = 29) Population (n = 22) Condition (n = 13)
Records included in review (n = 6)
Records excluded from meta-analysis: Did not report sufficient epidemiological data (n = 1)
Records included in meta-analysis (n = 5)
n: number of records

Population characteristics

- Studies reported on adults; mean age range: 68-69 years[9,10]
- % males ranged from 47-52% (median: 51.9%)[9-11]
- Disease contexts for development of ILD commonly included autoimmune-related ILD, hypersensitivity pneumonitis, ILD due to external factors, and sarcoidosis

Study characteristics and quality appraisal

- Studies included in the meta-analysis were retrospective and conducted between 2010 and 2019 (**Table 2**)
- Two studies were conducted in the US,[11,12] two in Europe,[8,10] one in South Korea[9]
- Study period ranged from 4[11,12] to 8[9] years (median: 5 years)
- Population size ranged from about 3 million[12] to 65 million[10]
- Source populations (denominators) were representative of the general population in three studies,[8-10] and sub-populations from large administrative claims databases in two studies[11,12]
 - Olson et al. (2021)[11] used insurance database while Singer et al. (2022)[12] used both private insurance and Medicare Advantage (Part D)
- Studies were generally well designed, with noted transparency on decisions regarding the algorithms to select patients or detect fibrosis progression
 - However, most algorithms were not validated and may have identified patients that did not have PPF (i.e., false positives), thereby inflating prevalence or incidence rates

Table 2. Characteristics of studies included in meta-analysis

Author (year)	Study design	Study country: Data source	Time period	Source population size
Hilberg (2022)[8]	Retro-spective	Belgium: Leuven Univ. Hosp., Ghent Univ. Hosp., Liege Univ. Hosp. Centre; Denmark: Lillebaelt Hosp.; Finland: Turku Hosp.; Greece: Heraklion University Hosp., University Hosp. of Larissa, General Hosp. of Thessaloniki, Athens Medical Centre; Norway: Oslo Univ. Hosp.; Portugal: Coimbra Hosp. and Univ. Centre, São João Univ. Hosp. Centre, Vila Nova de Gaia/Espinho Hosp. Centre, Beatriz Ângelo Hosp.	2014-2018	Not reported
Joung (2023)[9]	Retro-spective	South Korea: Korean Health Insurance Review and Assessment (HIRA) database	2011-2018	97% of the South Korean population: 50,796,898
Nasser (2021)[10]	Retro-spective	France: Système National des Données de Santé	2010-2016	98.8% of the French population of over 66 million people
Olson (2021)[11]	Retro-spective	United States: IBM MarketScan Research Databases	2012-2015	37,565,644
Singer (2022)[12]	Retro-spective	United States: Medicare; Optum Research Database	2016-2019	Medicare: 2,936,729; Optum: 6,009,363

Discussion

- First meta-analysis of non-IPF PPF incidence and prevalence
- Global pooled estimates suggest non-IPF PPF is rare, with wide variation across countries; highest estimates in the US
- US estimates for incidence (27.6 per 100,000) and prevalence (57.8–60.7 per 100,000) were significantly higher than those from Europe and South Korea
 - US Medicare data may have overestimated incidence and prevalence due to reduced population sampled, whereas studies from France and South Korea used nearly complete national population data, resulting in more reliable estimates
 - Hilberg et al. (2022)[8] used hospital data and clinician-validated cases across six European countries; however, estimates across regions showed variability due to differences in database quality, population selection, and catchment areas. Underestimated figures were observed in Portugal and Greece due to participation of multiple hospitals with imprecise reference populations and likely missing true cases
- Lack of specific ICD codes for non-IPF PPF led to reliance on algorithms and claims databases, resulting in potential misclassification and variability across studies
 - Errors in ICD code entry and updates, and variability in study algorithms contributed to inconsistency in reported estimates
 - There is need for better validated algorithms and specific ICD codes to improve the accuracy of PPF diagnosis and reporting in epidemiological studies

Conclusion

- PPF is a complex form of ILD; various risk factors affect progression, and pooled global estimates suggest it is rare
- This study showed a higher incidence and prevalence of non-IPF PPF in the US vs. other countries at the population level; however, evidence to support this are limited
- A contributing factor to this finding may be related to overestimation of incidence and prevalence caused by selected denominators from claims databases that were not representative of the general populations of the US
- Contributing factors to the wide variability in study estimates of non-IPF PPF across regions may include:
 i. Use of non-clinically validated algorithms to identify PPF from claims databases;
 ii. Variation in types of denominators or data sources;
 iii. IPF cases were not strictly excluded in one US study[11]
- Further standardization in the definition of non-IPF PPF is required for accurate estimates of incidence and prevalence

Pooled global incidence of non-IPF PPF was 10.9 per 100,000 persons (95% CI: 1.3, 20.5); pooled global prevalence was 37.0 per 100,000 persons (95% CI: 23.6, 50.5)

Figure 2. Forest plot of incidence of non-IPF PPF by region

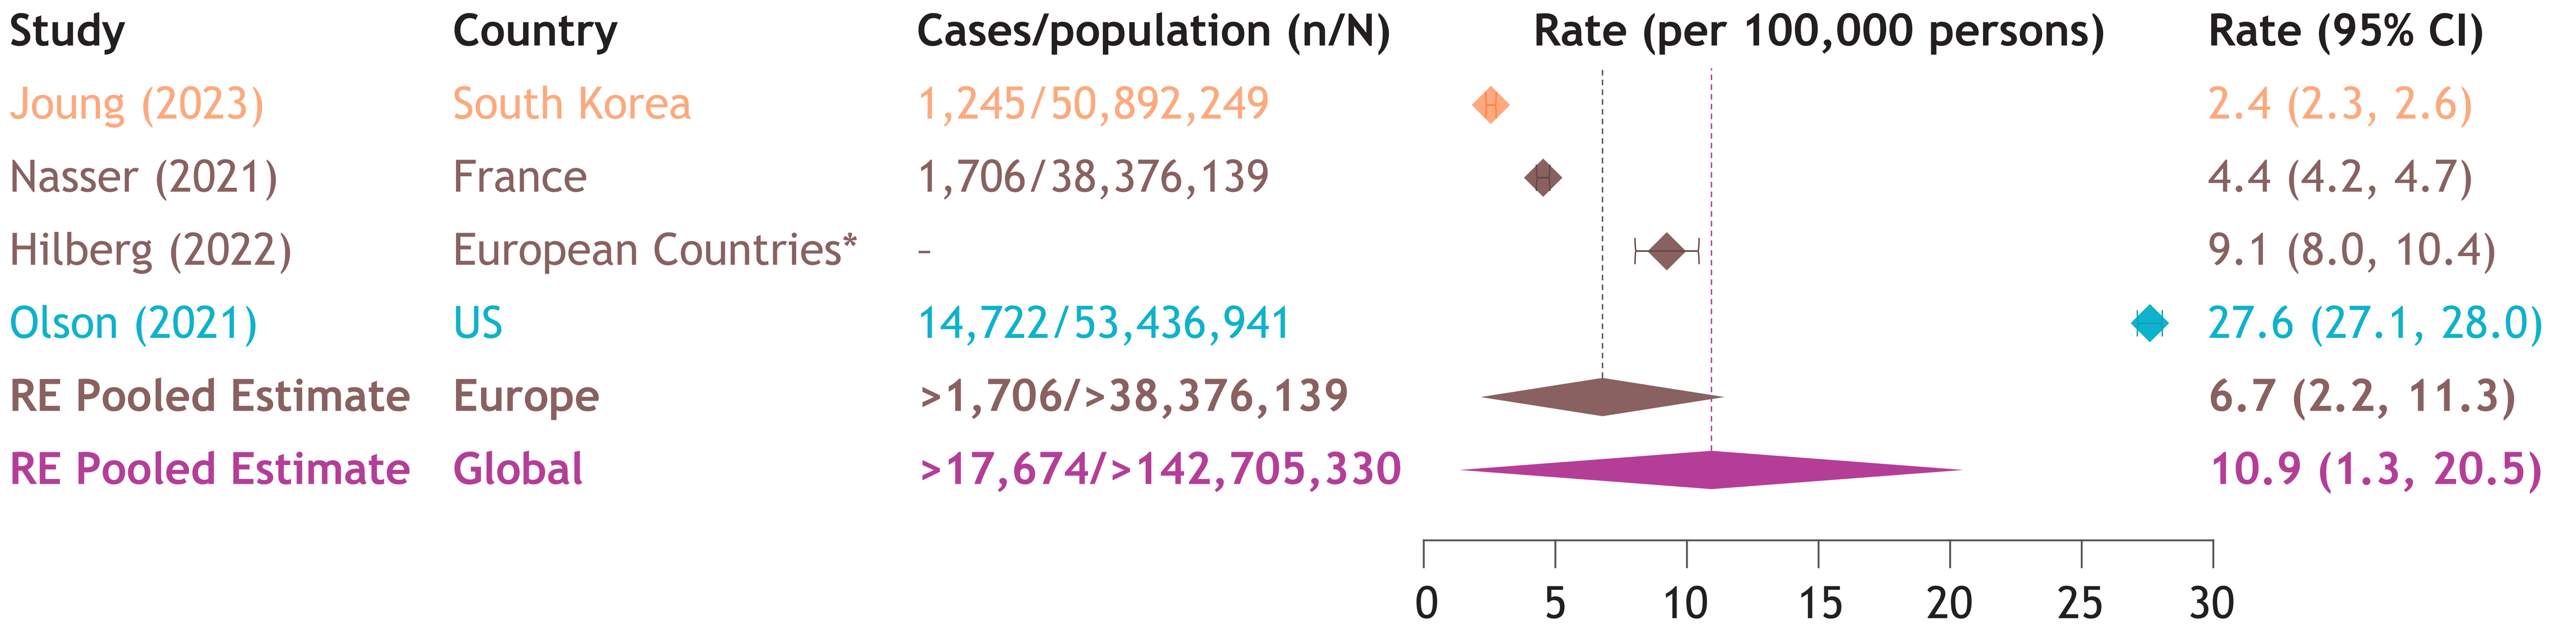

Study | Country | Cases/population (n/N) | Rate (per 100,000 persons) | Rate (95% CI)
Joung (2023) | South Korea | 1,245/50,892,249 | 2.4 (2.3, 2.6)
Nasser (2021) | France | 1,706/38,376,139 | 4.4 (4.2, 4.7)
Hilberg (2022) | European Countries* | - | 9.1 (8.0, 10.4)
Olson (2021) | US | 14,722/53,436,941 | 27.6 (27.1, 28.0)
RE Pooled Estimate | Europe | >1,706/>38,376,139 | 6.7 (2.2, 11.3)
RE Pooled Estimate | Global | >17,674/>142,705,330 | 10.9 (1.3, 20.5)
0 5 10 15 20 25 30

Figure 3. Forest plot of prevalence of non-IPF PPF by region

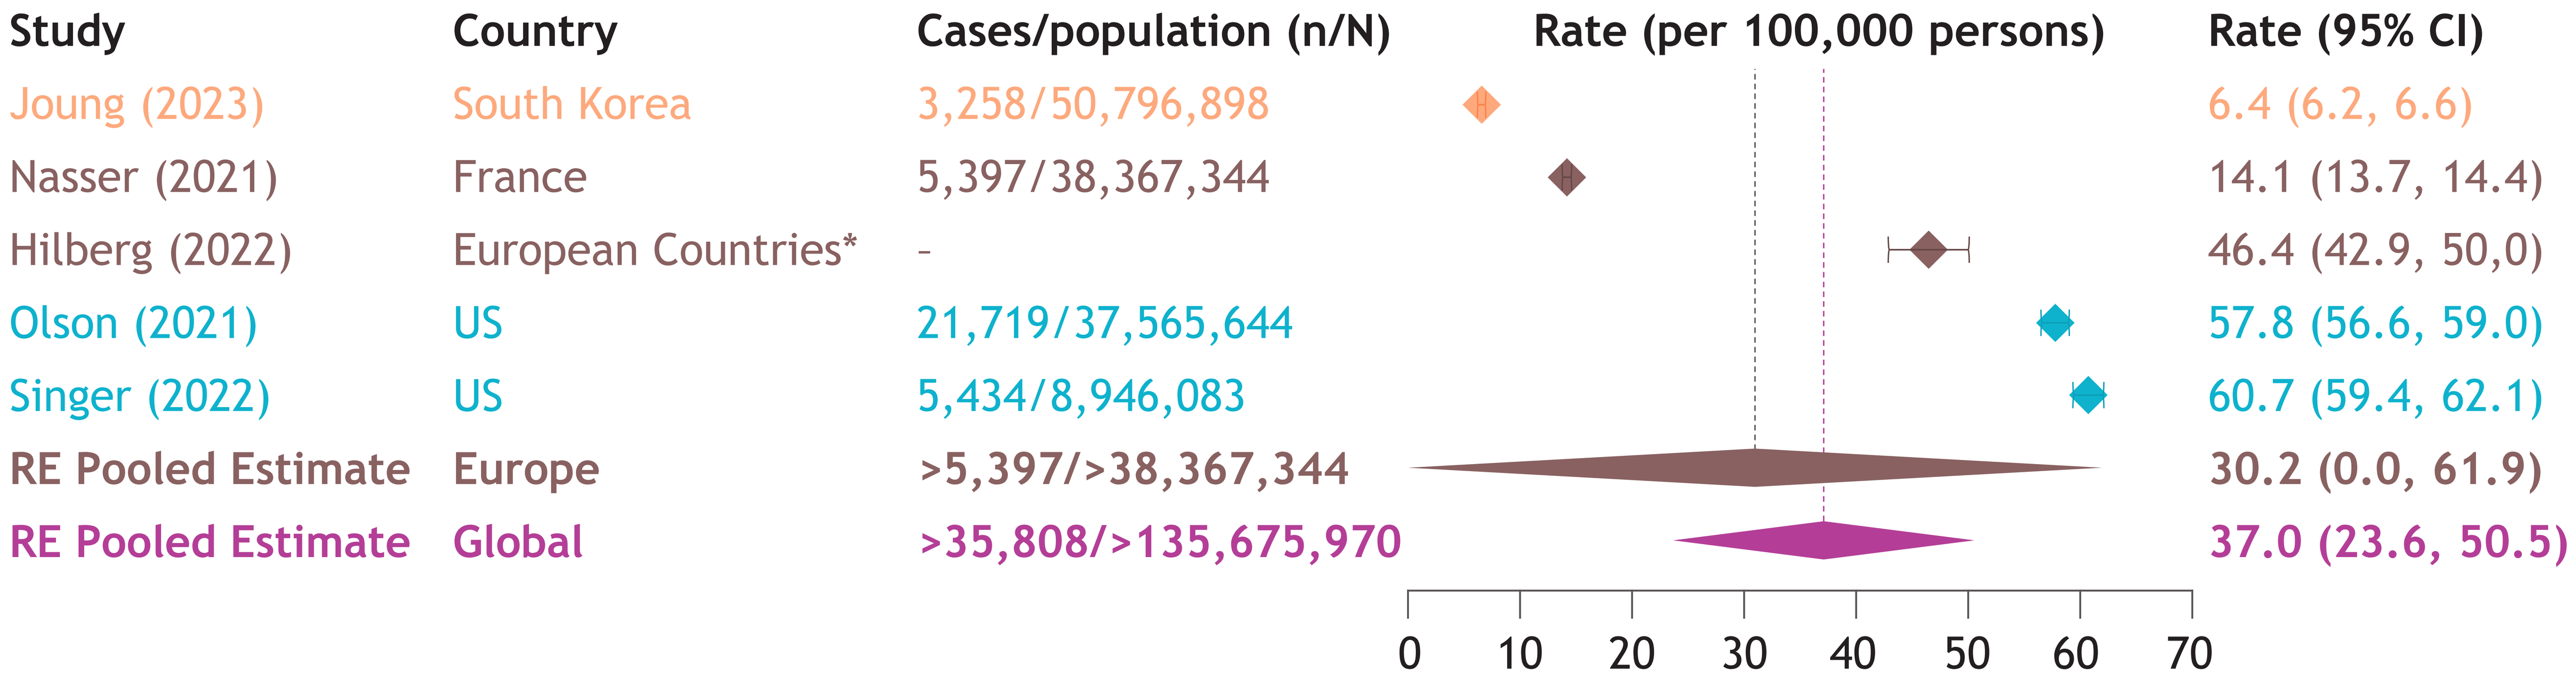

Study | Country | Cases/population (n/N) | Rate (per 100,000 persons) | Rate (95% CI)
Joung (2023) | South Korea | 3,258/50,796,898 | 6.4 (6.2, 6.6)
Nasser (2021) | France | 5,397/38,367,344 | 14.1 (13.7, 14.4)
Hilberg (2022) | European Countries* | - | 46.4 (42.9, 50,0)
Olson (2021) | US | 21,719/37,565,644 | 57.8 (56.6, 59.0)
Singer (2022) | US | 5,434/8,946,083 | 60.7 (59.4, 62.1)
RE Pooled Estimate | Europe | >5,397/>38,367,344 | 30.2 (0.0, 61.9)
RE Pooled Estimate | Global | >35,808/>135,675,970 | 37.0 (23.6, 50.5)
0 10 20 30 40 50 60 70

*Belgium, Denmark, Finland, Greece, Norway, and Portugal. - : Not reported; CI: Confidence interval; Non-IPF: Non-idiopathic pulmonary fibrosis; PPF: Progressive pulmonary fibrosis; RE: Random effects; US: United States

Incidence of non-IPF PPF

- Study from Asia[9] reported a lower annual incidence rate (2.4 [95% CI: 2.3, 2.6]) than the pooled rate across two European studies[8,10] (6.7 [2.2, 11.3]; **Figure 2**)
- Annual incidence of 27.6 (95% CI: 27.1, 28.0) from the US study[11] was higher than the pooled incidence (5.2 [3.2, 7.2]; data not shown) across 3 studies from other countries[8-10]

Prevalence of non-IPF PPF

- Pooled prevalence across two US studies[11,12] (59.3 [95% CI: 56.4, 62.1]; data not shown) was higher than that for studies from Europe[8,10] (30.2 [0.0, 61.9]; **Figure 3**)
 - Wide 95% CI of the European pooled prevalence due to large difference of reported prevalence estimates between pooled studies[8,10]

References

1. Raghu G, et al. *Am J Respir Crit Care Med* 2022;205(9):e18–e47.
2. Higgins JPT, et al. (editors). Cochrane Handbook for Systematic Reviews of Interventions. 2nd Edition. Chichester (UK): John Wiley & Sons, 2019.
3. Munn Z, et al. *Int J Evid Based Healthc* 2015;13(3):147-153.
4. Page MJ, et al. *BMJ* 2021;372:n71. doi: 10.1136/bmj.n71.
5. Munn Z, et al. *Int J Health Policy Manag* 2014;3(3):123.
6. DerSimonian R, et al. *Control Clin Trials* 1986;7(3):177-188.
7. Viechtbauer W. *J Stat Softw* 2010;36(3):1-48.
8. Hilberg O, et al. *ERJ Open Res* 2022;8(1). doi: 10.1183/23120541.00597-2021.
9. Joung KI, et al. *BMC Pulm Med* 2023;23(1):98.
10. Nasser M, et al. *Respir Res* 2021;22(1):162.
11. Olson AL, et al. *Adv Ther* 2021;38(7):4100-4114.
12. Singer D, et al. *Ann Am Thorac Soc* 2022;19(7):1112-1121.
13. Alaws H, et al. *Chest* 2023;164(4):A3131

Conflicts of Interest

- Negar Golchin, Tamara Lesperance, Julia Scheuring, Brandon Elpers*, and Aditya Patel are employees and/or shareholders of Bristol Myers Squibb. *At the time of the analysis
- Victoria Wan, Kimberly Hofer, and Jean-Paul Collet are employees of Evidinno Outcomes Research Inc. (Vancouver, BC, Canada), which was contracted by Bristol Myers Squibb to conduct this study

Presented at ISPOR Europe 2024; November 17-20, 2024; Barcelona, Spain

Email: khofer@evidinno.com

Copies of this poster are for personal use only and may not be reproduced without written permission of the authors.