HPR41

Biosimilars: The Key to Lowering Access Barriers to Omalizumab Across Europe

Minyoung Jang[1] , Seungjae Yi[1] Taek Kwon[1]

[1]Celltrion Inc., Market Access, Incheon, Korea, Rep. of South

Introduction & Objectives

- This study examines possibilities facilitating access to omalizumab, an anti-IgE antibody treatment used for allergic asthma (AA), chronic spontaneous urticaria (CSU), and chronic rhinosinusitis with nasal polyps (CRSwNP).
- **High treatment costs, stringent reimbursement policies, and complex clinical eligibility requirements** such as the need for multiple diagnostic tests, **limit patient access to omalizumab.**
- This study evaluates cost savings with the introduction of biosimilars and assesses the potential of the biosimilars to mitigate access barriers.

Materials & Methods

- A review of reimbursement guidelines for omalizumab was conducted across European countries, focusing on national health coverage policies and clinical eligibilities. We also conducted in-depth interviews with physicians and healthcare professionals to investigate physician perceptions to omalizumab access.
- A cost analysis was conducted to estimate savings under a scenario where the biosimilar was priced at 35% discount from originator. The starting market share is projected at 20%, with an anticipated annual growth rate of 10%. The model incorporated IQVIA data.

Figure 1. Projected Annual Savings on Omalizumab: Comparison of Originator and Biosimilar (2023 - 2028)

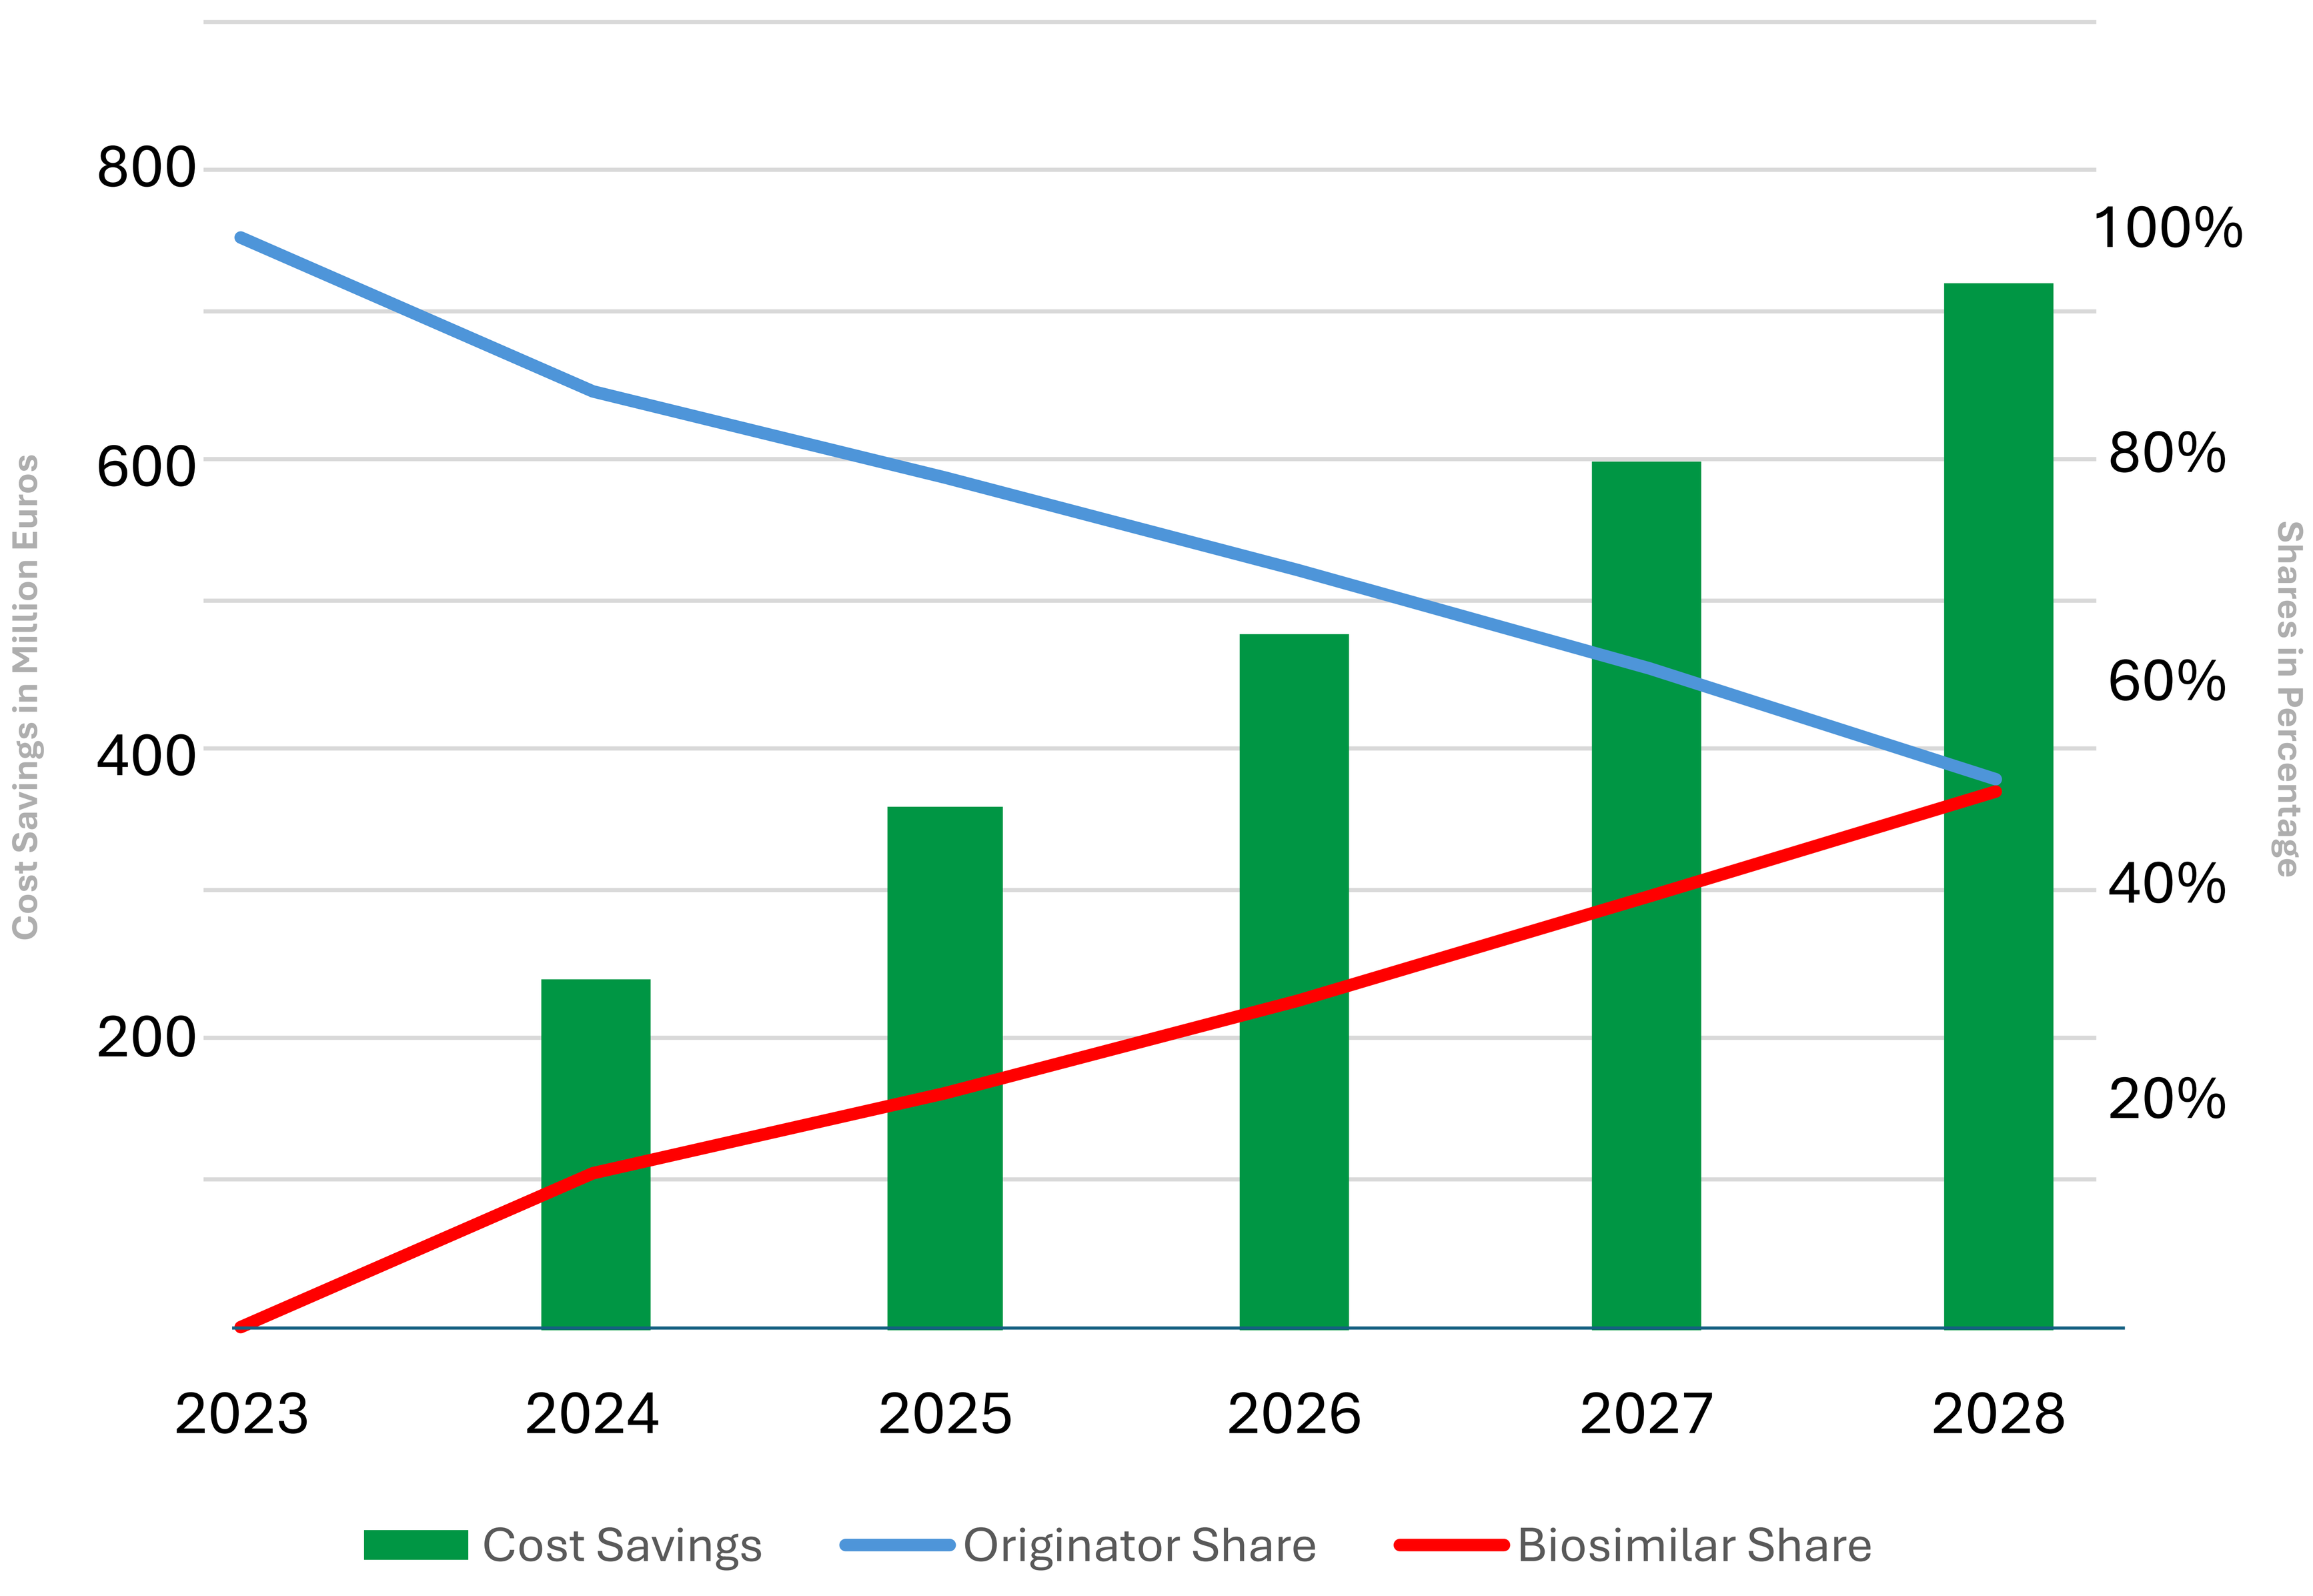
800
600
400
200
Cost Savings in Million Euros
100%
80%
60%
40%
20%
Shares in Percentage
2023
2024
2025
2026
2027
2028
Cost Savings
Originator Share
Biosimilar Share

Results

- **Country-level disparities in reimbursement to omalizumab were identified across European countries**, with notably limited access for CRSwNP due to a lack of reimbursement. CRSwNP is not reimbursed in France, Spain, and the UK, and is subject to strict guidelines in Germany and Italy
- Interviewees reported that strict initiation criteria restrict access to omalizumab for AA and CSU. Patients are often required to experience multiple severe exacerbations and rely heavily on steroids before qualifying. **Treatment initiation is often delayed because it requires approval from medical or payor boards**.
- In some regions, lengthy reimbursement processes can delay patient access to treatments. Reimbursement decisions often require extended approval procedures.
- **Interviewees highlighted that affordability is crucial for improving patient access.** Biosimilars could play a pivotal role in expanding reimbursement for conditions like CRSwNP, potentially aiding patients who currently lack access to biologic treatments.
- Interviewees also mentioned use of omalizumab for conditions such as food allergies. While the US FDA has approved omalizumab for food allergy treatment, in the EU, patients must seek it as an off-label option, which poses a barrier to optimal clinical outcome.
- According to our cost model, **increased uptake of biosimilars could significantly impact spending, with projections showing a reduction in the cost** of the originator drug **from $1.97 billion in 2023 to $1.56 billion by 2028.**

Conclusion

- **The study highlights a widespread need to improve patient access to omalizumab across Europe, emphasizing significant disparities in reimbursement and eligibility criteria.**
- **Introducing biosimilars for omalizumab could enhance patient access and reduce disparities by lowering treatment costs.**