EE599

Estimating the Potential Lifetime Health and Economic Impact of V116 on Pneumococcal Disease in Mexico

Urrego-Reyes J[1]; Yi Z[2]; Parellada C[3]; Hirata L[3]; Cossrow N[2]; Johnson KD[2]; Owusu-Edusei K[2]

[1]MSD Colombia, Bogota, Colombia
[2]Merck & Co., Inc., Rahway, NJ, USA
[3]MSD Brazil, São Paulo, SP, Brazil

Background

- *Streptococcus pneumoniae (S. pneumoniae)* is a gram-positive bacterium that can cause invasive pneumococcal disease (IPD) and non-bacteremic pneumococcal pneumonia (NBPP), particularly in young children and older adults. The risk of pneumococcal disease (PD) increases with age and certain medical conditions, making these populations especially vulnerable[1,2]
- Currently, Mexico's national immunization program includes pneumococcal vaccination for young children with PCV13 (2+1 schedule) and adults aged ≥60 years, offering PCV13 for healthy adults and a sequential regimen (PCV13+PPSV23) for high-risk patients[3]
- Despite availability of immunization schedules, adult vaccination coverage rates (VCR) remain <25%[4], resulting in a significant burden of PD. In addition, new serotypes not included in current vaccines are emerging as an important cause of IPD and NBPP[5]
- V116 is a novel 21-valent PCV, specifically designed for adults. It contains **21** serotypes: **3, 6A, 7F, 8, 9N, 10A, 11A, 12F, 15A, 15C,16F, 17F, 19A, 20A, 22F, 23A, 23B, 24F, 31, 33F,** and **35B**. Eight **(15A, 15C, 16F, 23A, 23B, 24F, 31, 35B)** are unique serotypes not covered in any currently licensed vaccines

Objective

- To estimate the potential health and economic impact of vaccination with V116 versus PCV20 on pneumococcal disease in adults aged ≥60 years in Mexico

Methods

- A multi-cohort Markov model was adapted to estimate the lifetime (until death or turned 100 years old) cases of IPD, post meningitis sequelae (PMS), NBPP, IPD and NBPP related deaths, and the associated direct medical costs among the Mexican adults aged ≥60 years vaccinated with V116, PCV20 and PCV13 (**Figure 1**)
- The study population was stratified into three risk groups using national health survey data: low-risk (LR), at-risk (AR), and high-risk (HR) adults[6,7]
- The model assumed the same serotype-specific vaccine effectiveness and vaccination coverage rate of 60%. The waning of the effectiveness was assumed to remain stable for the first 5 years and then linearly declines to zero in the following 10 years[5]
- IPD serotype distribution data were obtained from GIVEBPVac reports (2018-2021 period)[8] and NBPP serotype distribution was assumed to be the same as for IPD
- Epidemiological and cost data were obtained from literature and national databases
- Economic outcomes were discounted at 5.0% annually and undiscounted clinical cases were reported. Costs were expressed in 2024 US Dollars (USD) using an exchange rate of 1 USD = 19.89 Mexican Pesos (Oct 23rd, 2024)
- Base case results were summarized for V116 and PCV20, and PCV13. A second scenario for V116 and PCV20 versus PCV13+PPSV23 in AR and HR populations was performed
- One-way sensitivity analysis (OWSA) was conducted to assess the robustness of the results and to identify the most influential parameters on the total direct medical costs saved by V116 when compared to PCV20

Figure 1. A schematic diagram depicting the structure of the state-transition Markov model

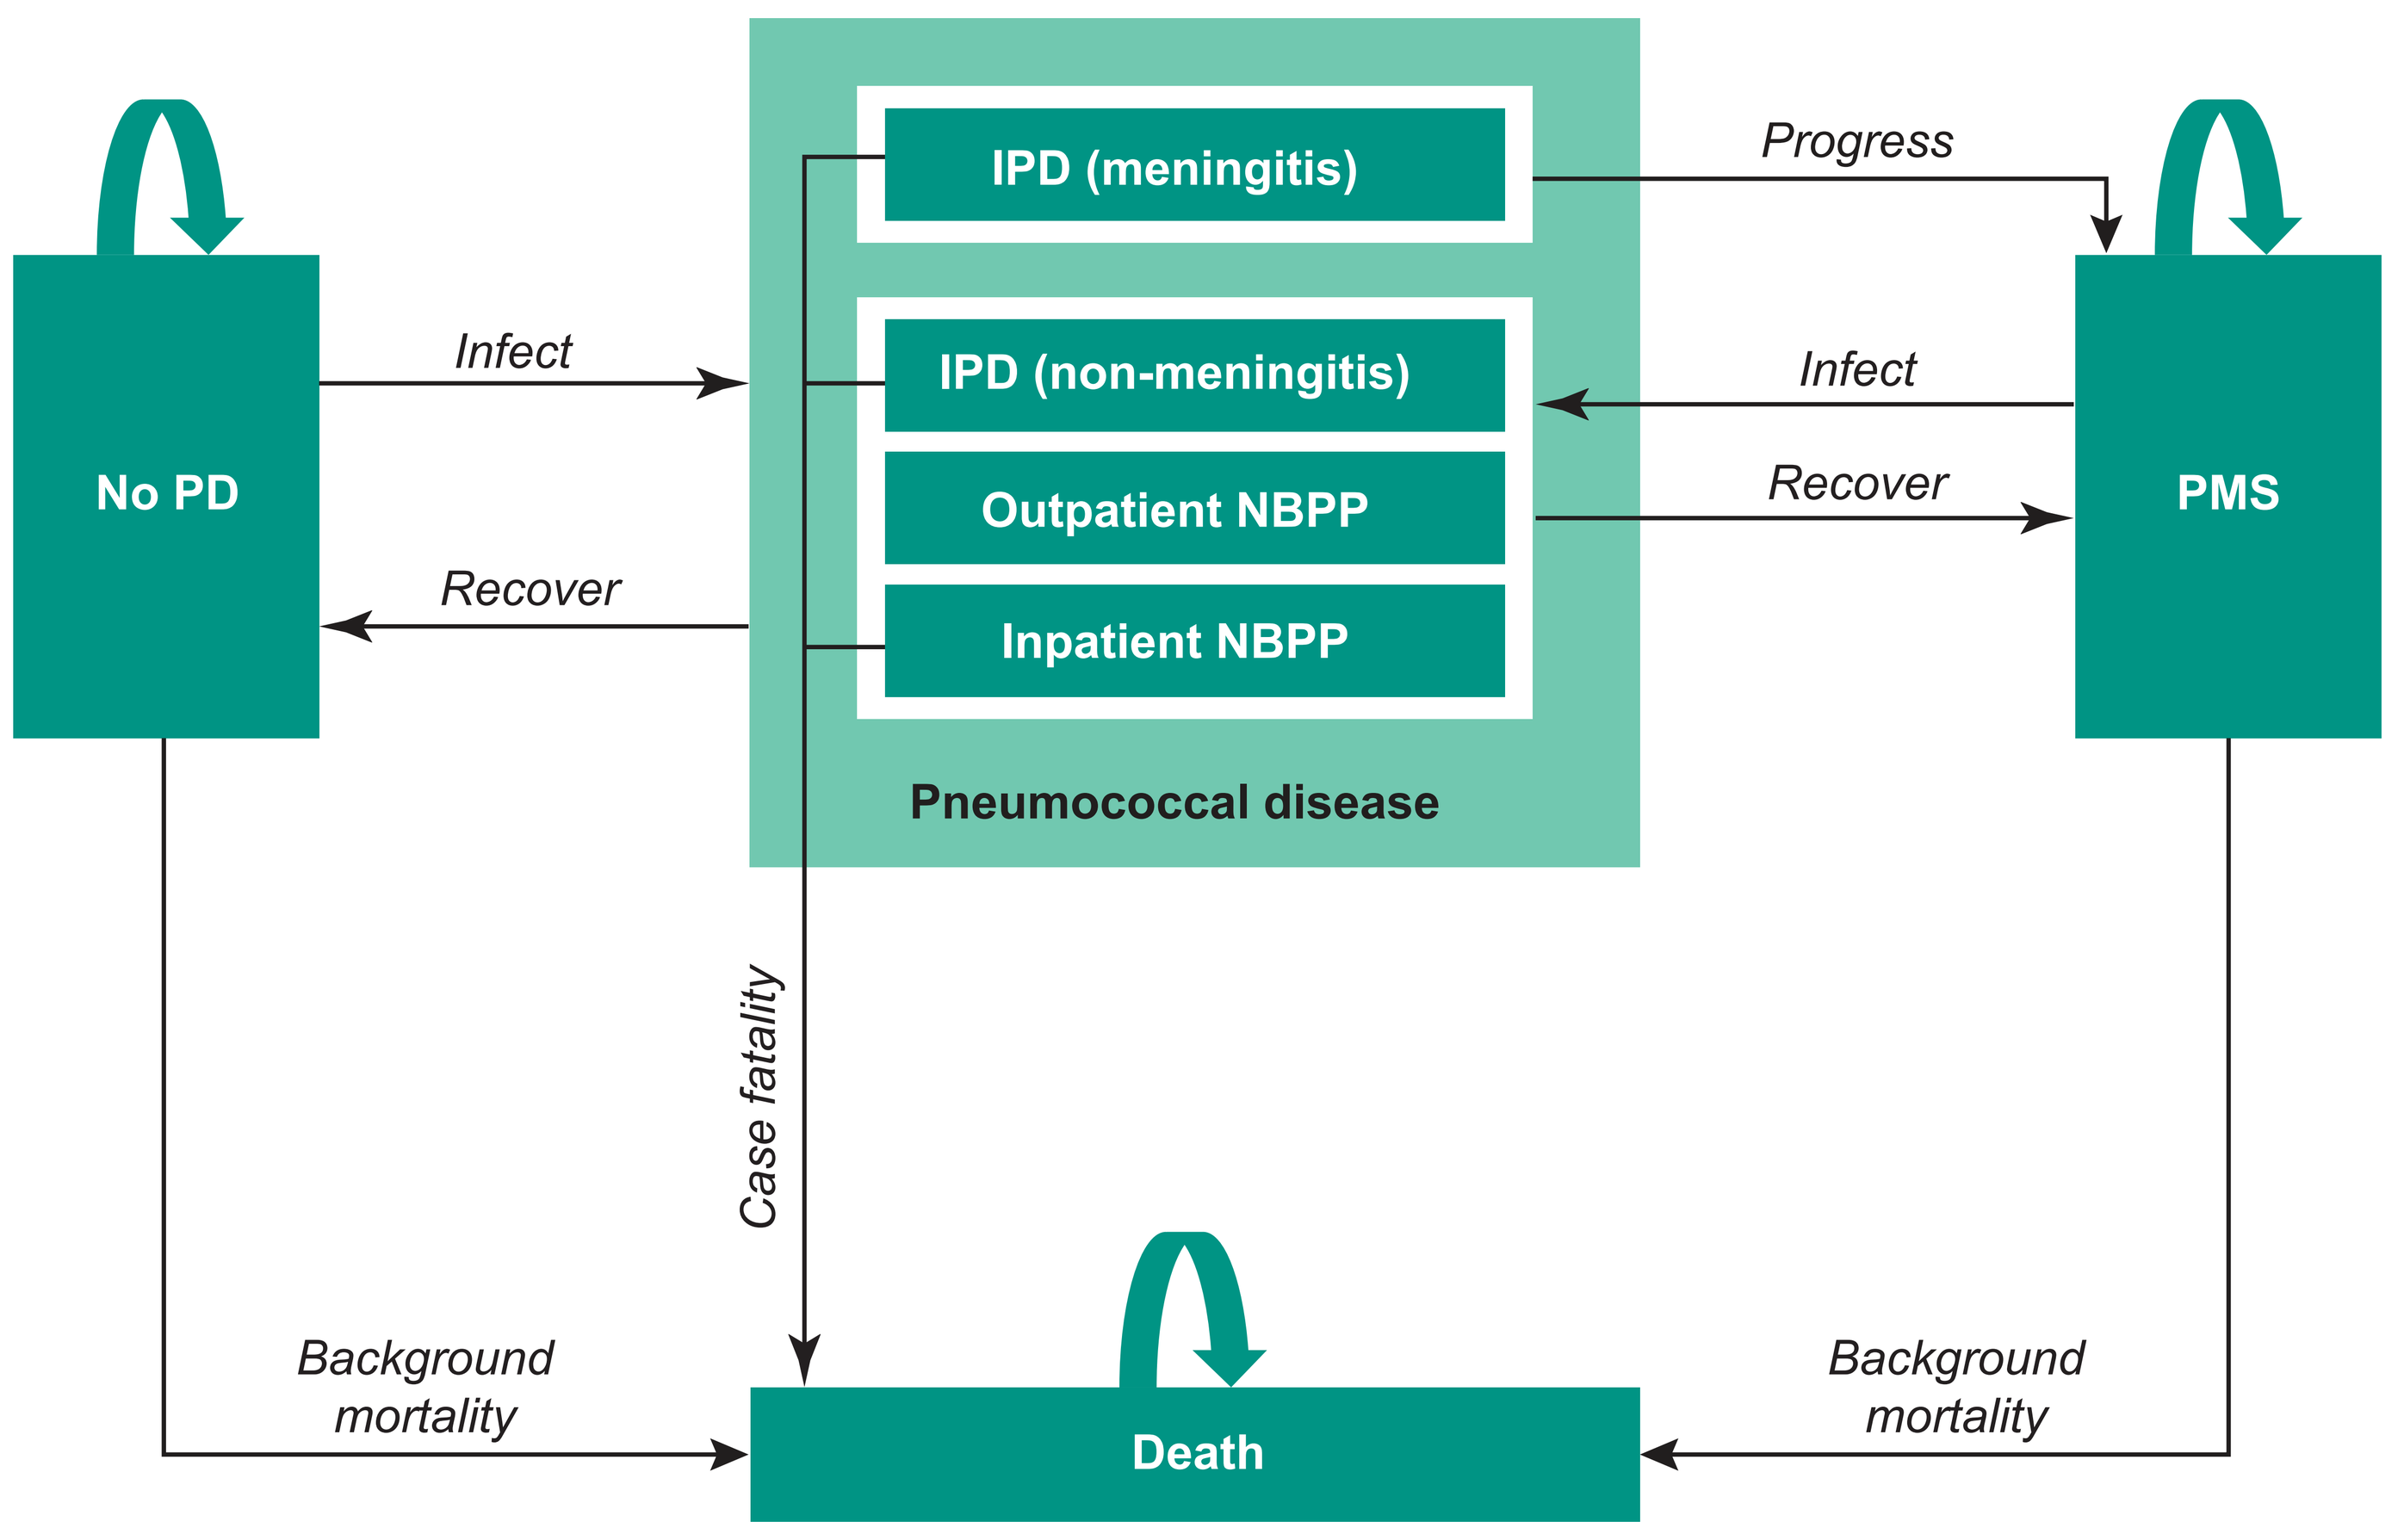
No PD
Infect
Recover
IPD (meningitis)
IPD (non-meningitis)
Outpatient NBPP
Inpatient NBPP
Pneumococcal disease
Progress
Infect
Recover
PMS
Case fatality
Background mortality
Death
Background mortality

PD, pneumococcal disease; IPD, invasive pneumococcal disease; NBPP, non-bacteremic pneumococcal pneumonia; PMS, post-meningitis sequelae

Results

- In adults aged ≥60 years and compared to PCV13, V116 prevented an additional 3,434 IPD cases and 1,841 IPD-related deaths, 26,838 inpatient and 10,835 outpatient NBPP cases, and 7,461 NBPP-related deaths. This represented more than twice the IPD cases and related deaths, and NBPP (inpatient and outpatient) cases and related deaths prevented by PCV20. Specifically, PCV20 averted 1,675 IPD cases and 898 related deaths, 13,085 inpatient and 5,284 outpatient NBPP cases, and 3,638 NBPP-related deaths compared to PCV13 (**Table 1**)
- In terms of total direct costs, V116 and PCV20 saved an additional USD 141.3 million and USD 68.9 million, respectively, compared to PCV13. Moreover, V116 saved USD 72.4 million more than PCV20 (**Table 1**)
- In AR and HR populations, V116 prevented an additional 1,530 IPD cases and 820 related deaths, 19,135 NBPP cases (inpatient + outpatient), and 3,712 related deaths compared to PCV13+PPSV23. These results are more than 5 times the IPD cases and deaths, and almost 3 times the NBPP cases and deaths prevented by PCV20 (**Table 2**)
- Furthermore, V116 and PCV20 saved an additional USD 65 million and USD 21 million, respectively, compared to PCV13+PPSV23 (**Table 2**)
- The OWSA consistently showed that V116 resulted in greater direct medical costs savings compared to PCV20 (**Figure 2**)

Table 1. Estimated lifetime clinical and economic outcomes of V116 and PCV20 compared to PCV13 in adults aged ≥60 years

	V116	PCV20	PCV13	Cases/costs averted, compared to PCV13		Additional reduction by V116, compared to PCV20	
				By V116	By PCV20	Number	X-fold
Clinical Outcomes (Undiscounted)							
IPD	38,047	39,807	41,482	3,434	1,675	1,759	2.05
PMS	1,030	1,077	1,123	93	45	48	2.05
NBPP-IP	832,347	846,101	859,185	26,838	13,085	13,754	2.05
NBPP-OP	212,027	217,578	222,862	10,835	5,284	5,551	2.05
IPD deaths	20,393	21,336	22,234	1,841	898	943	2.05
NBPP deaths	231,392	235,216	238,854	7,461	3,638	3,824	2.05
Economic Outcomes (discounted) in 2024 USD							
Total direct costs	**$2,456,613,966**	**$2,529,021,488**	**$2,597,927,790**	**$141,313,824**	**$68,906,302**	**$72,407,522**	**2.05**
IPD	$154,560,477	$164,169,728	$173,317,583	$18,757,107	$9,147,855	$9,609,252	2.05
PMS	$18,717,105	$19,964,016	$21,150,626	$2,433,521	$1,186,610	$1,246,911	2.05
NBPP-IP	$1,789,544,437	$1,834,671,828	$1,877,608,461	$88,064,024	$42,936,633	$45,127,391	2.05
NBPP-OP	$493,791,947	$510,215,915	$525,851,120	$32,059,172	$15,635,205	$16,423,968	2.05

V116, 21-valent pneumococcal conjugate vaccine; PCV20, 20-valent pneumococcal conjugate vaccine; PCV13, 13-valent pneumococcal conjugate vaccine; IPD, invasive pneumococcal disease; PMS, post-meningitis sequelae; NBPP-IP, non-bacteremic pneumococcal pneumonia inpatient; NBPP-OP, non-bacteremic pneumococcal pneumonia outpatient.

Table 2. Estimated lifetime clinical and economic outcomes of V116 and PCV20 compared to PCV13+PPSV23 in adults aged ≥60 years at-risk and high-risk

	V116	PCV20	PCV13+PPSV23	Cases/costs averted, compared to PCV13+PPSV23		Additional reduction by V116, compared to PCV20	
				By V116	By PCV20	Number	X-fold
Clinical Outcomes (Undiscounted)							
IPD	29,767	31,034	31,297	1,530	263	1,267	5.82
PMS	806	840	847	41	7	34	5.86
NBPP-IP	654,606	663,287	667,956	13,350	4,669	8,681	2.86
NBPP-OP	173,755	177,551	179,540	5,785	1,989	3,796	2.91
IPD deaths	15,955	16,634	16,775	820	141	679	5.82
NBPP deaths	181,980	184,394	185,692	3,712	1,298	2,414	2.86
Economic Outcomes (discounted) in 2024 USD							
Total direct costs	**$1,903,670,763**	**$1,948,239,499**	**$1,969,501,536**	**$65,830,773**	**$21,262,037**	**$44,568,736**	**3.10**
IPD	$102,768,797	$108,322,519	$109,399,302	$6,630,506	$1,076,783	$5,553,722	6.16
PMS	$14,754,790	$15,654,121	$15,817,084	$1,062,293	$162,962	$899,331	6.52
NBPP-IP	$1,411,553,081	$1,439,765,674	$1,454,610,983	$43,057,902	$14,845,308	$28,212,593	2.90
NBPP-OP	$374,594,095	$384,497,184	$389,674,167	$15,080,073	$5,176,983	$9,903,089	2.91

V116, 21-valent pneumococcal conjugate vaccine; PCV20, 20-valent pneumococcal conjugate vaccine; PCV13, 13-valent pneumococcal conjugate vaccine; PPSV23, Pneumococcal polysaccharide vaccine 23; IPD, invasive pneumococcal disease; PMS, post-meningitis sequelae; NBPP-IP, non-bacteremic pneumococcal pneumonia inpatient; NBPP-OP, non-bacteremic pneumococcal pneumonia outpatient.

Figure 2. Sensitivity analysis – estimated lifetime direct medical cost for adults aged ≥60 years saved by V116, compared with PCV20 (2024 USD); discounted

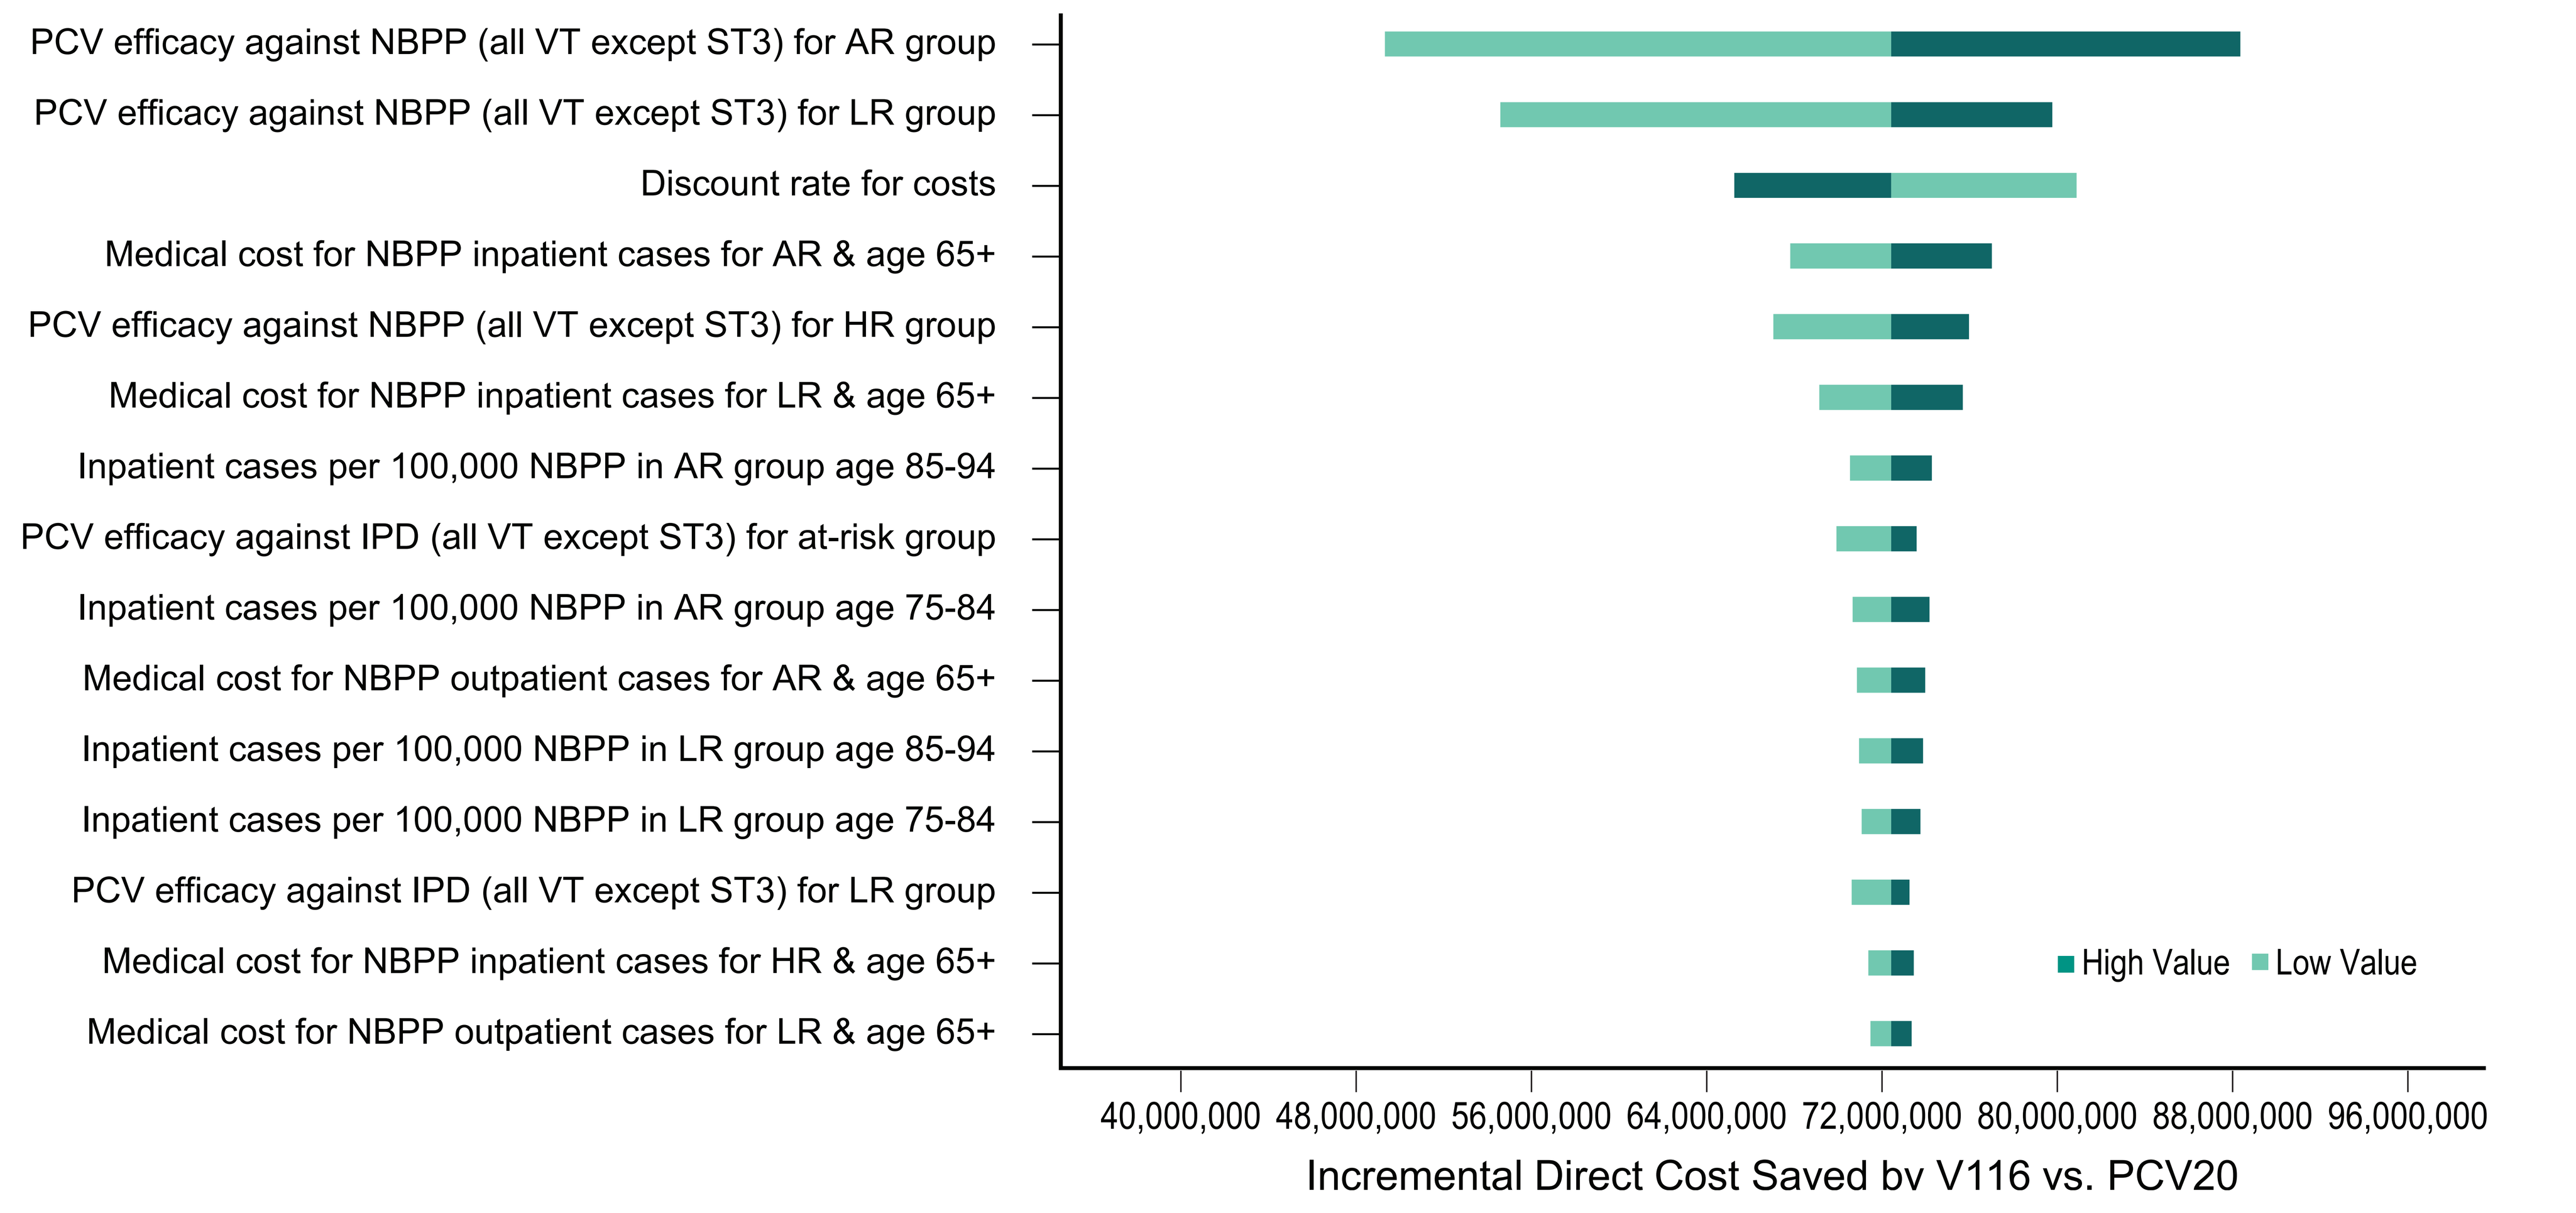
PCV efficacy against NBPP (all VT except ST3) for AR group
PCV efficacy against NBPP (all VT except ST3) for LR group
Discount rate for costs
Medical cost for NBPP inpatient cases for AR & age 65+
PCV efficacy against NBPP (all VT except ST3) for HR group
Medical cost for NBPP inpatient cases for LR & age 65+
Inpatient cases per 100,000 NBPP in AR group age 85-94
PCV efficacy against IPD (all VT except ST3) for at-risk group
Inpatient cases per 100,000 NBPP in AR group age 75-84
Medical cost for NBPP outpatient cases for AR & age 65+
Inpatient cases per 100,000 NBPP in LR group age 85-94
Inpatient cases per 100,000 NBPP in LR group age 75-84
PCV efficacy against IPD (all VT except ST3) for LR group
Medical cost for NBPP inpatient cases for HR & age 65+
Medical cost for NBPP outpatient cases for LR & age 65+
High Value
Low Value
40,000,000
48,000,000
56,000,000
64,000,000
72,000,000
80,000,000
88,000,000
96,000,000
Incremental Direct Cost Saved bv V116 vs. PCV20

PCV, pneumococcal conjugate vaccine; NBPP, non-bacteremic pneumococcal pneumonia; AR, at-risk; IPD, invasive pneumococcal disease; VT, vaccine type; LR, low-risk; HR, high-risk.

Discussion and Conclusion

- In comparison to PCV13, V116 significantly reduced the health and economic burden associated with PD in older adults in Mexico, achieving more than double the impact of PCV20. Among AR and HR populations, V116 showed a markedly greater effect compared to PCV20, particularly in reducing IPD by more than five times
- These findings should be considered alongside Mexico's current immunization policies, vaccination coverage, and prevalent disease-causing serotypes. Analysis of 2018-2022 IPD reports[8] revealed that the 8 unique serotypes included in V116 accounted for approximately 21% of cases. The overall coverage for IPD-causing serotypes was 57% for PCV20 and 77% for V116
- Previous studies in Mexico have highlighted a substantial burden of PD in older adults, with high case-fatality rates for pneumonia and bacteremia[9]
- Based on the current epidemiological situation, the inclusion of V116 in the NIP could significantly reduce the health and economic burden of PD in adults aged ≥60 years in Mexico, particularly in comparison to PCV20

References

1. US Centers for Disease Control and Prevention. ABCs Surveillance Report: Streptococcus pneumoniae, 2019. Available on: https://www.cdc.gov/abcs/downloads/SPN_Surveillance_Report_2019.pdf. Accessed on October 9, 2024.
2. US Centers for Disease Control and Prevention. Active Bacterial Core surveillance. Available on: https://www.cdc.gov/abcs/. Accessed on October 9, 2024.
3. World Health Organization. Vaccination schedule for Pneumococcal disease, 2024. https://immunizationdata.who.int/global/wiise-detail-page/vaccination-schedule-for-pneumococcal disease?ISO_3_CODE=MEX&TARGETPOP_GENERAL=ADULTS+CATCHUP_C+RISKGROUPS. Accessed on October 11, 2024.
4. Mongua-Rodríguez N, Ferreira-Guerrero E, Delgado-Sánchez G, et al. Vacunación en adultos y adultos mayores en México. *Salud Publica Mex*. 2023;65:s146-s152. Published 2023 Jun 13. doi:10.21149/14786.
5. Stoecker C. Economic assessment PCV15 & PCV20. Presented at ACIP. June 15, 2021. https://www.cdc.gov/vaccines/acip/meetings/downloads/slides 2021 06/03 Pneumococcal Stoecker 508.pdf.
6. Shamah-Levy T, Romero-Martínez M, Barrientos-Gutiérrez T, et al. Encuesta Nacional de Salud y Nutrición 2021 sobre Covid-19. Resultados nacionales. Cuernavaca, México: Instituto Nacional de Salud Pública, 2022.
7. Basto-Abreu A, López-Olmedo N, Rojas-Martínez R, et al. Prevalencia de prediabetes y diabetes en México: Ensanut 2022. *Salud Publica Mex*. 2023;65:s163-s168. Published 2023 Jun 13. doi:10.21149/14832.
8. Grupo Interinstitucional para la Vigilancia de Enfermedades Bacterianas Prevenibles por Vacunación (GIVEBPVac). Available on: https://insp.mx/lineas-de-investigacion/medicamentos-en-salud-publica/sireva.html.
9. Urrego-Reyes J, Cornejo DP, Hirata L, et al. Burden of clinical syndrome associated with pneumococcal disease in Mexico: A 2019 perspective. Poster presented at ISPPD-13; Cape Town, South Africa; March 17 20, 2024.

Disclosures

Juan Urrego-Reyes is an MSD Colombia employee. Cintia Parellada and Luciana Hirata are MSD Brazil employees. Zinan Yi, Nicole Cossrow, Kelly Johnson and Kwame Owusu-Edusei are employees of Merck Sharp & Dohme LLC, a subsidiary of Merck & Co., Inc., Rahway, NJ, USA. All may hold stock or stock options in Merck & Co., Inc., Rahway, NJ, USA. V116 was developed by Merck & Co., Inc., Rahway, NJ, USA.

Corresponding author

Juan Urrego-Reyes; email: juan.camilo.urrego.reyes@merck.com

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from the Congress or the author of this poster.

https://bit.ly/48vpZHG

Presented at ISPOR Europe; Barcelona, Spain; 17-20 November 2024.

Copyright © 2024 Merck & Co., Inc., Rahway, NJ, USA and its affiliates. All rights reserved.