Model-Projected Survival and Lifetime Clinical Outcomes of Exagamglogene Autotemcel (Exa-cel) in Patients With Transfusion-Dependent Beta-Thalassemia in Canada

Chuka Udeze[1], Michael Gargano[1], Lisa Kaindl[2], Sarah Kane[2], Allen Zhou[2], Siddharth Jain[1], Andrea Lopez[1]

[1.] *Vertex Pharmaceuticals Incorporated, Boston, MA, USA;* [2.] *Eversana, Burlington, ON, Canada*

Poster EE383

BACKGROUND

- β-thalassemia is a rare hereditary hemoglobinopathy characterized by reduced or absent β-globin production; transfusion-dependent β-thalassemia (TDT) is the most severe form of the disease[1-3]
 - Patients with TDT require regular red blood cell transfusions (RBCTs) for survival[1,2]
- In Canada, standard of care (SOC) for TDT includes regular RBCTs and iron chelation therapy
- Exagamglogene autotemcel (exa-cel) is a cellular product consisting of autologous CD34+ hematopoietic stem and progenitor cells (HSPCs) modified by non-viral, *ex vivo* CRISPR/Cas9 that reduces erythroid-specific expression of *BCL11A*, reactivating synthesis of fetal hemoglobin (HbF); increases in HbF levels ameliorate the severity of β-thalassemia and therefore has the potential to eliminate the need for RBCTs, providing a functional cure for patients with TDT[4]
 - In the pivotal Phase 3 trial CLIMB THAL-111 (January 2023 data cut), 91% (32/35) of participants treated with exa-cel in the primary efficacy set met the primary endpoint TI12 (defined as a weighted average hemoglobin level of ≥9 grams per deciliter without RBCT for at least 12 consecutive months)
 - Of the three participants that did not achieve TI12, one participant had a relative reduction in annualized RBCT volume of 84% and 2 participants stopped RBCT at 14.5 months and 12.2 months after exa-cel infusion and have been transfusion-free for 7.3 months and 4.0 months, respectively, starting 60 days after the final transfusion
- Based on the positive results from the CLIMB THAL-111 trial, Health Canada granted marketing authorization for exa-cel for the treatment of patients aged ≥12 years with TDT in September 2024

OBJECTIVE

- To assess the potential survival and lifetime clinical outcomes of exa-cel versus SOC in Canada for patients with TDT

METHODS

Model Overview

- A Markov cohort model was developed to project survival and clinical outcomes of patients with TDT treated with exa-cel versus SOC in Canada over a lifetime horizon
- The model includes transfusion status (i.e., transfusion independent [TI], transfusion reduced [TR], and transfusion dependent [TD]) and death as health states; transfusion status is assumed to impact patients' iron levels (serum ferritin, liver iron, and cardiac iron), which impacts the development of TDT-related complications (**Figure 1**)
- At model start, all patients are assumed to be TD (i.e., receiving regular RBCTs and having non-normal iron levels)
- Patients treated with SOC are assumed to maintain the baseline transfusion status (TD), RBCT frequency, and non-normal iron levels throughout the model horizon
- The model has three phases for exa-cel-treated patients: a treatment phase, an iron-normalization/change phase, and a post-iron normalization phase (remainder of patient's lifetime)
- Patients treated with exa-cel can potentially achieve TI or TR at the end of the treatment phase
 - Patients who achieve TI or TR are assumed to remain in that health state for the remainder of the model time horizon
- Patients' iron levels change based on their transfusion status; the model assumes that the change in iron level categories (normal, low, medium, high) occurs at a constant rate over the duration of the iron normalization/change phase. A patient's iron level category in a given cycle determines the risk of developing a TDT-related complication
 - Patients who achieve TI achieve iron normalization (i.e., achieve recommended iron levels per UK Thalassaemia Society Standards) at the end of the iron normalization period and are assumed to be at no further risk of TDT-related complications
 - Patients who achieve TR experience a reduction in their baseline iron levels (e.g., reduce from high to medium, or medium to low), but do not achieve iron normalization
 - Patients who remain TD remain at baseline iron levels
- Mortality risk is estimated based on transfusion status, the presence of complications, and the occurrence of other transplantation-related events

Figure 1. Schematic of TDT Model Structure

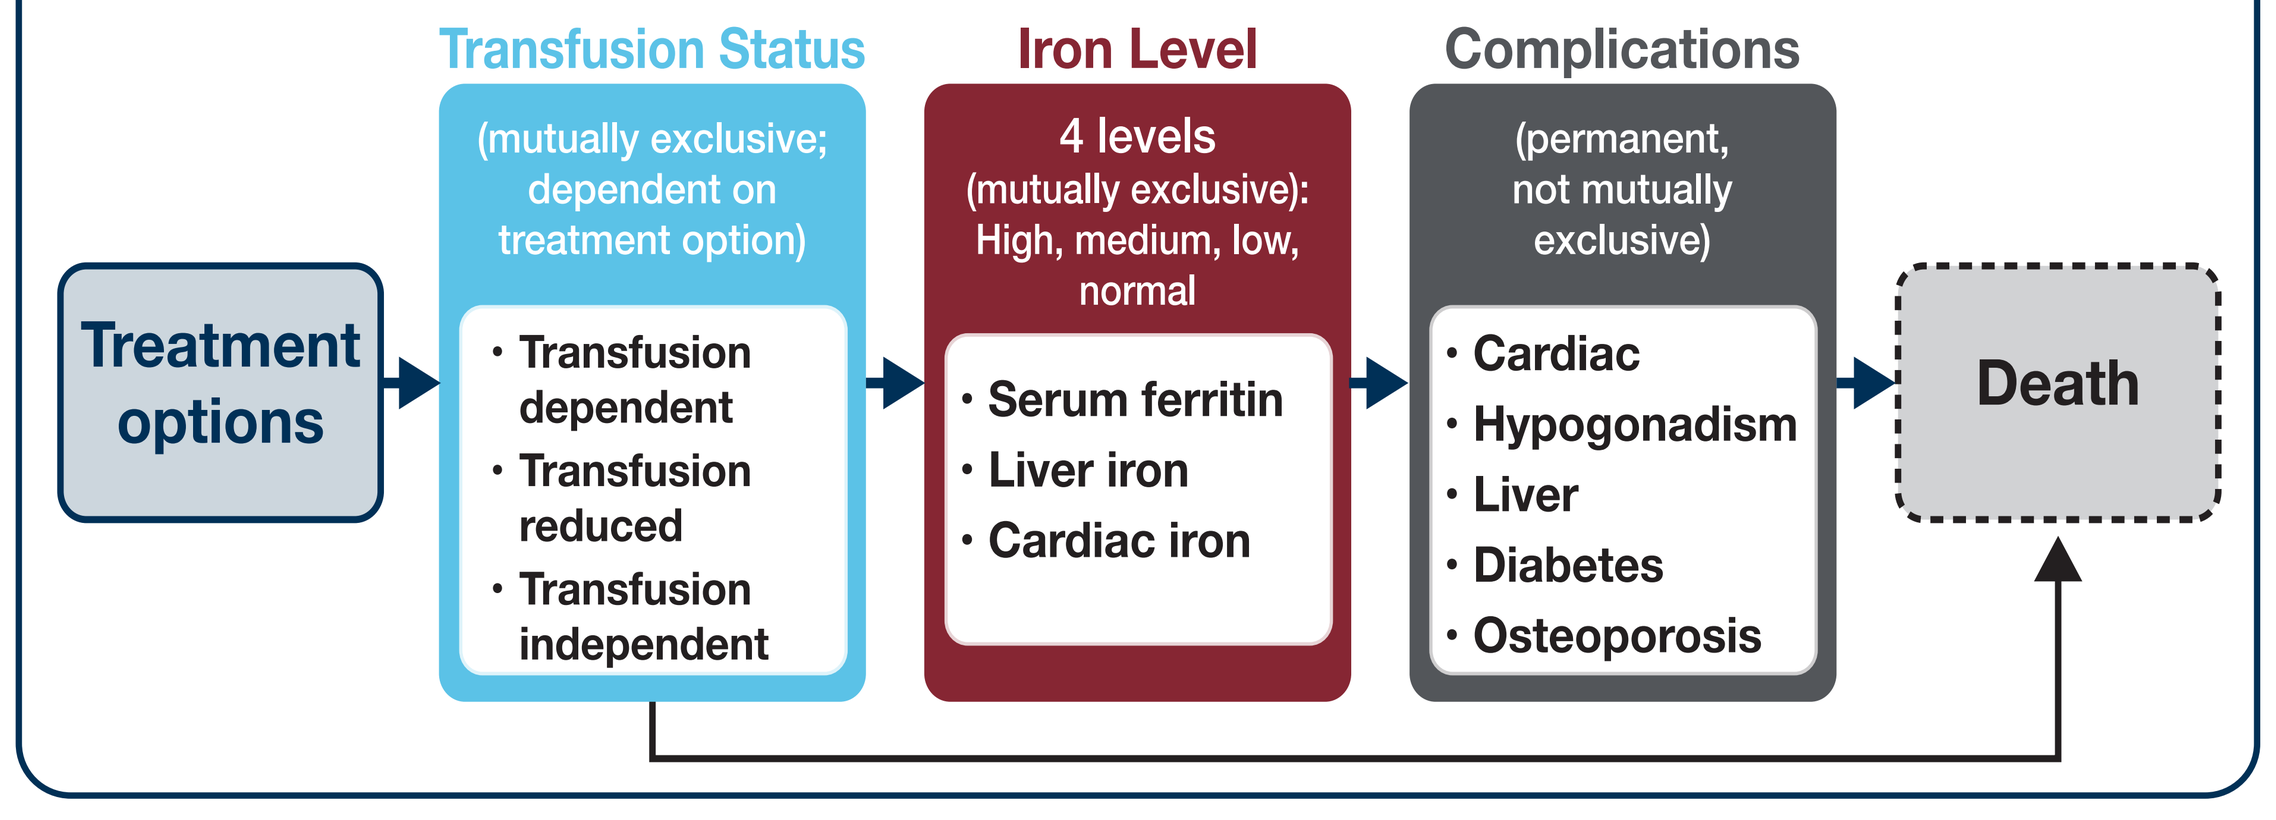
Transfusion Status
Iron Level
Complications
Treatment options
(mutually exclusive; dependent on treatment option)
• Transfusion dependent
• Transfusion reduced
• Transfusion independent
4 levels (mutually exclusive): High, medium, low, normal
• Serum ferritin
• Liver iron
• Cardiac iron
(permanent, not mutually exclusive)
• Cardiac
• Hypogonadism
• Liver
• Diabetes
• Osteoporosis
Death

Data Sources and Model Inputs

- A cohort of patients with TDT was modeled from baseline – the cohort had a mean age of 21.1 years and required an average of 17.2 RBCTs per year, based on the baseline characteristics of participants enrolled in the CLIMB THAL-111 Phase 3 trial[4]
- Distributions of baseline iron levels were derived from published literature[5]
 - Serum ferritin: Low (≤1,000 ng/ml): 23.0%; Moderate (1,000-2,500 ng/ml): 38.8%; High (>2,500 ng/ml): 38.2%
 - Cardiac iron: Low (>20 ms): 88.2%; Moderate (10-20 ms): 11.8%; High (<10 ms): 0.0%
 - Liver iron: Low (<7 mg/g): 60.5%; Moderate (7-15 mg/g): 23.5%; High (≥15 mg/g): 16.0%
- Exa-cel clinical efficacy was informed by the pre-specified interim analysis of the CLIMB THAL-111 clinical trial published in Locatelli et al. 2024 *N Engl J Med*[4] (data as of January 2023; median follow-up duration: 20.4 months; range: 2.1 – 48.1 months)
 - 91% of patients treated with exa-cel were assumed to transition to the TI health state at the end of the treatment phase, given 32 of 35 patients treated with exa-cel in the primary efficacy set had met the primary endpoint TI12
 - The remaining 9% of patients treated with exa-cel were assumed to transition to the TR health state at the end of the treatment phase, with an average reduction in transfusions of 87.2% from baseline, based on the clinical outcomes of the remaining 3 trial patients who did not achieve TI12 at the point of the pre-specified interim analysis in CLIMB THAL-111
- The duration of the treatment phase was assumed to be 1 year and the duration of the time to iron normalization/change was assumed to be 5 years
- Model inputs for baseline prevalence of TDT-related complications were based on CLIMB THAL-111; the risks of developing TDT-related complications, based on iron levels and transfusion status, were derived from published literature (**Table 1**)
 - Patients treated with exa-cel who achieve TI and normal iron levels were assumed to be at no further risk of developing complications
- Transfusion status-dependent mortality was considered as standardized mortality ratios (SMRs) applied to the age- and gender-specific mortality rates in the Canadian general population:
 - Consistent with other economic analyses[6], patients in the TD health state were assumed to have a 3.9-fold increased risk of mortality vs the general population[7]
 - Patients in the TI health state were assumed to have a 25% increased risk of mortality vs the general population to account for the impact of previous TDT and use of myeloablative conditioning as part of the exa-cel treatment process
 - Patients in the TR health state were assumed to have a 2.6-fold increased risk of mortality, estimated as the mid-point of TI and TD
 - Patients who developed cardiac complications had an increased annual mortality risk of 13%[6,8] and patients with diabetes were assumed to have a 1.5-fold increased risk of mortality[9]

Table 1. Risk of Developing TDT-Related Complications

Complication	Risk of Developing Complication	Source
Cardiac complications	Annual risk by cardiac iron content level: • Low: 1.1% • Moderate: 1.9% • High: 4.0%	Pepe et al. 2018[10]
Liver complications	Annual risk by liver iron content level: • Low: 0.0% • Moderate: 0.0% • High: 8.5%	Assumption; Angelucci et al. 2002[11]
Diabetes **Hypogonadism**	Annual risk equation based on age, serum ferritin levels and cardiac levels	Ang et al. 201412
Osteoporosis	Age-specific monthly incidence rate in the general population with an increased risk associated with TD and TR	Hippisley-Cox et al. 2009[13]; Li et al. 2023[14]

Model Outcomes

- The following outcomes were projected by the model:
 - Mean life expectancy
 - Mean number of RBCTs over patient lifetime
 - Proportion of patients developing complications
- All outcomes are undiscounted

Scenario Analyses

- Model parameters were varied to examine the impact of alternative inputs on clinical outcomes, including:
 - *Population:* A cohort with mean age of 12 years with no baseline complications, based on the minimum age requirements for CLIMB THAL-111 trial eligibility
 - *Alternative trial data cut:* Utilization of CLIMB THAL-111/131 April 2023 data cut (follow-up duration up to 51.1 months) to inform exa-cel clinical efficacy in model (TI: 92.9% [based on 39/42 achieving TI12]; TR: 7.1%, with 89.6% reduction in RBCT [based on reduction of remaining 3 patients])[4]
 - *Alternative efficacy definition & alternative trial data cut:* Assume that patients that have stopped RBCT for ≥60 days by the time of the April 2023 data cut (as opposed to TI12 endpoint in base case) are in the TI health state (TI: 100% based on April 2023 data cut of CLIMB THAL-111/131)[4]

RESULTS

Base Case Results

- Over a lifetime horizon, patients treated with exa-cel had a substantial increase in survival of 18.1 years compared to SOC (**Table 2**)
 - The mean predicted survival (i.e., age at death) of patients receiving exa-cel was 67.4 years vs. 49.3 years for patients treated with SOC
- Patients treated with exa-cel received ~458 fewer RBCTs over the lifetime horizon compared to SOC (exa-cel: 26.4 vs. SOC: 484.5) (**Table 2**)
- Further, the lifetime burden of complications of TDT was projected to be substantially lower in patients treated with exa-cel than in those receiving SOC (**Figure 3**)

Figure 2. Projected Survival for Patients with TDT

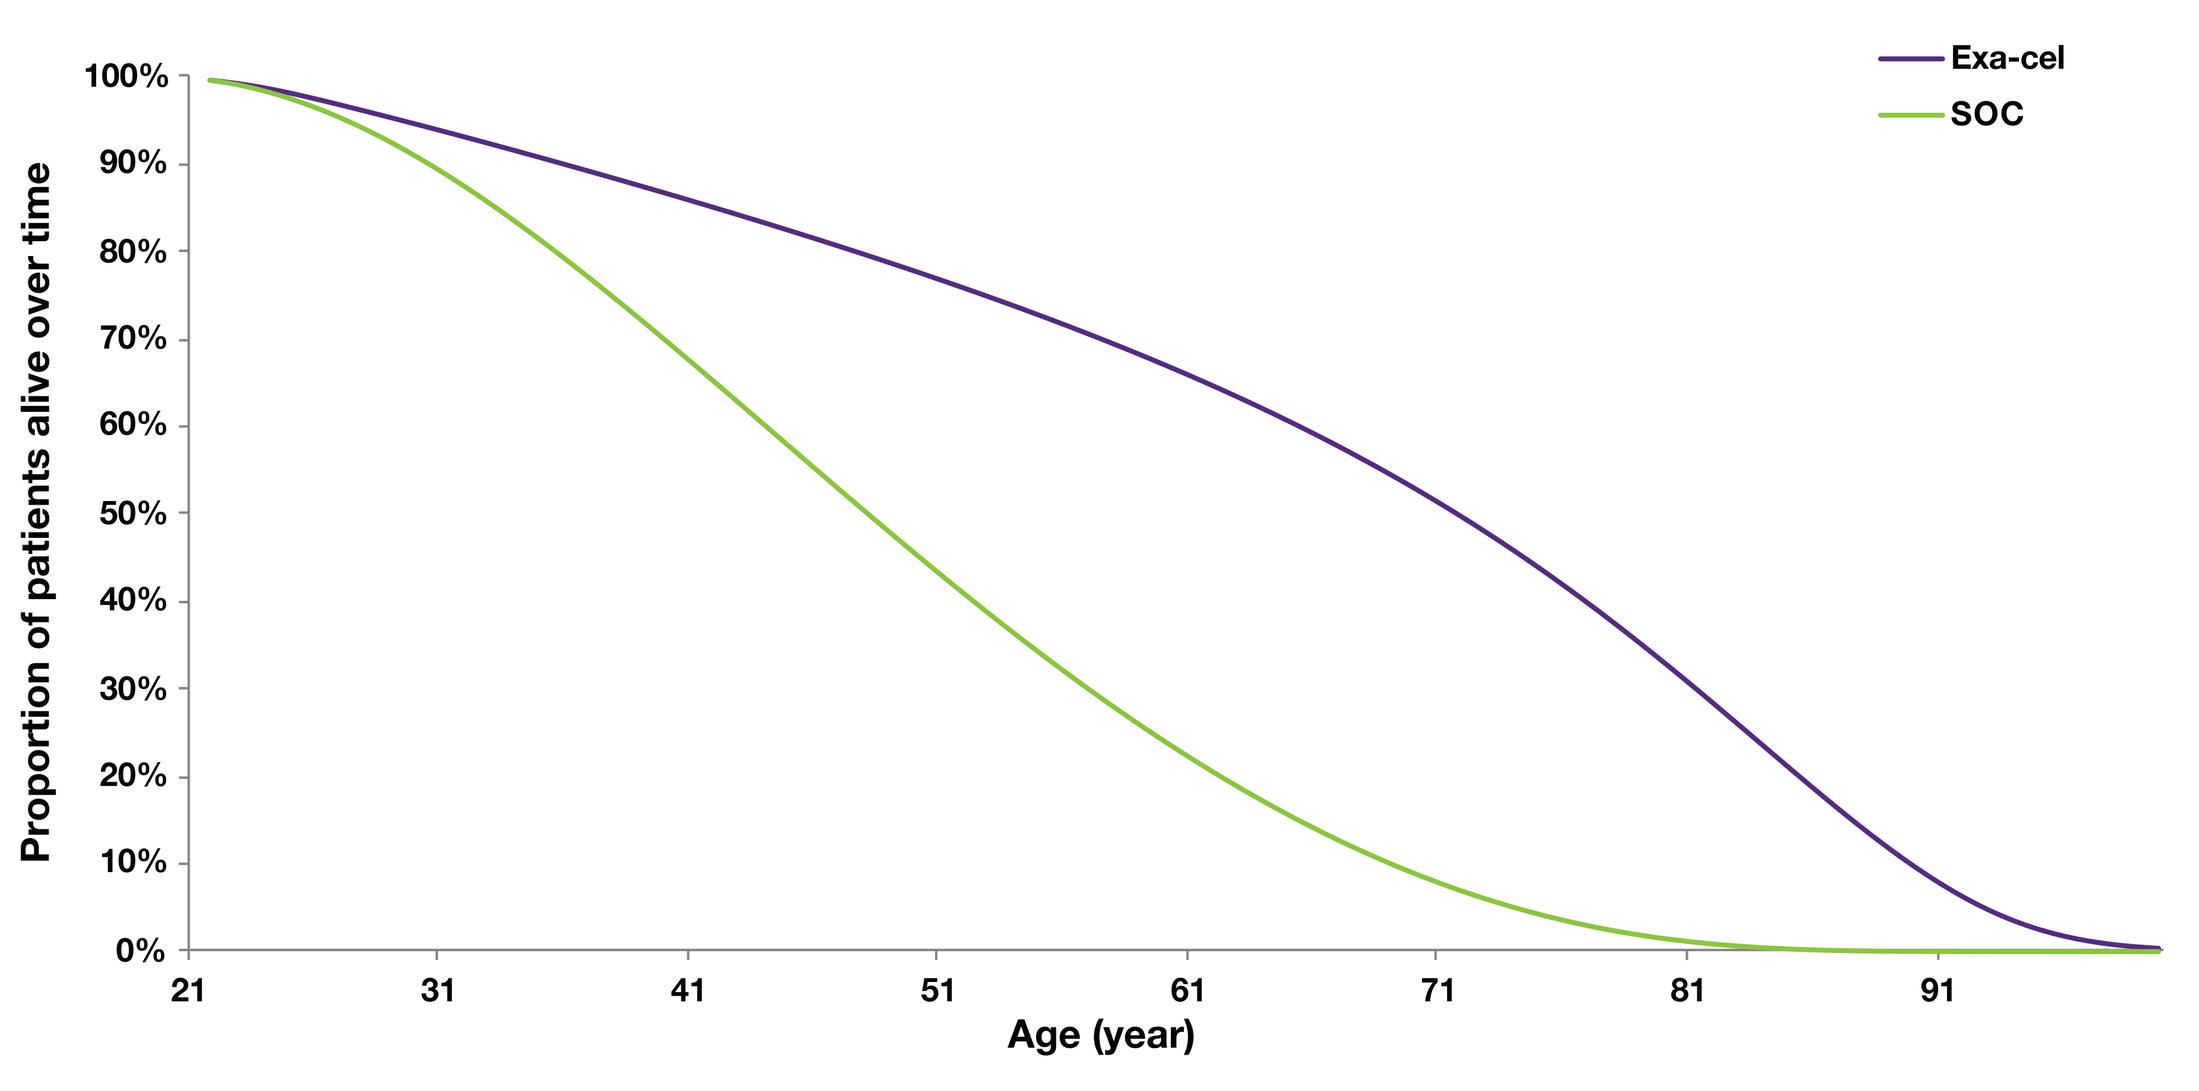
Exa-cel
SOC
Proportion of patients alive over time
100%
90%
80%
70%
60%
50%
40%
30%
20%
10%
0%
21
31
41
51
61
71
81
91
Age (year)

Table 2. Projected Survival and Number of RBCTs

Outcome	Exa-cel	SOC	Δ, Exa-cel vs SOC
Survival			
Mean life years (from model start)	46.3	28.2	18.1
Mean age of death (years)	67.4	49.3	18.1
Mean number of RBCTs over a lifetime			
RBCTs	26.4	484.5	-458.1

Figure 3. Proportion of Patients with TDT-Related Complications Over a Lifetime Horizon

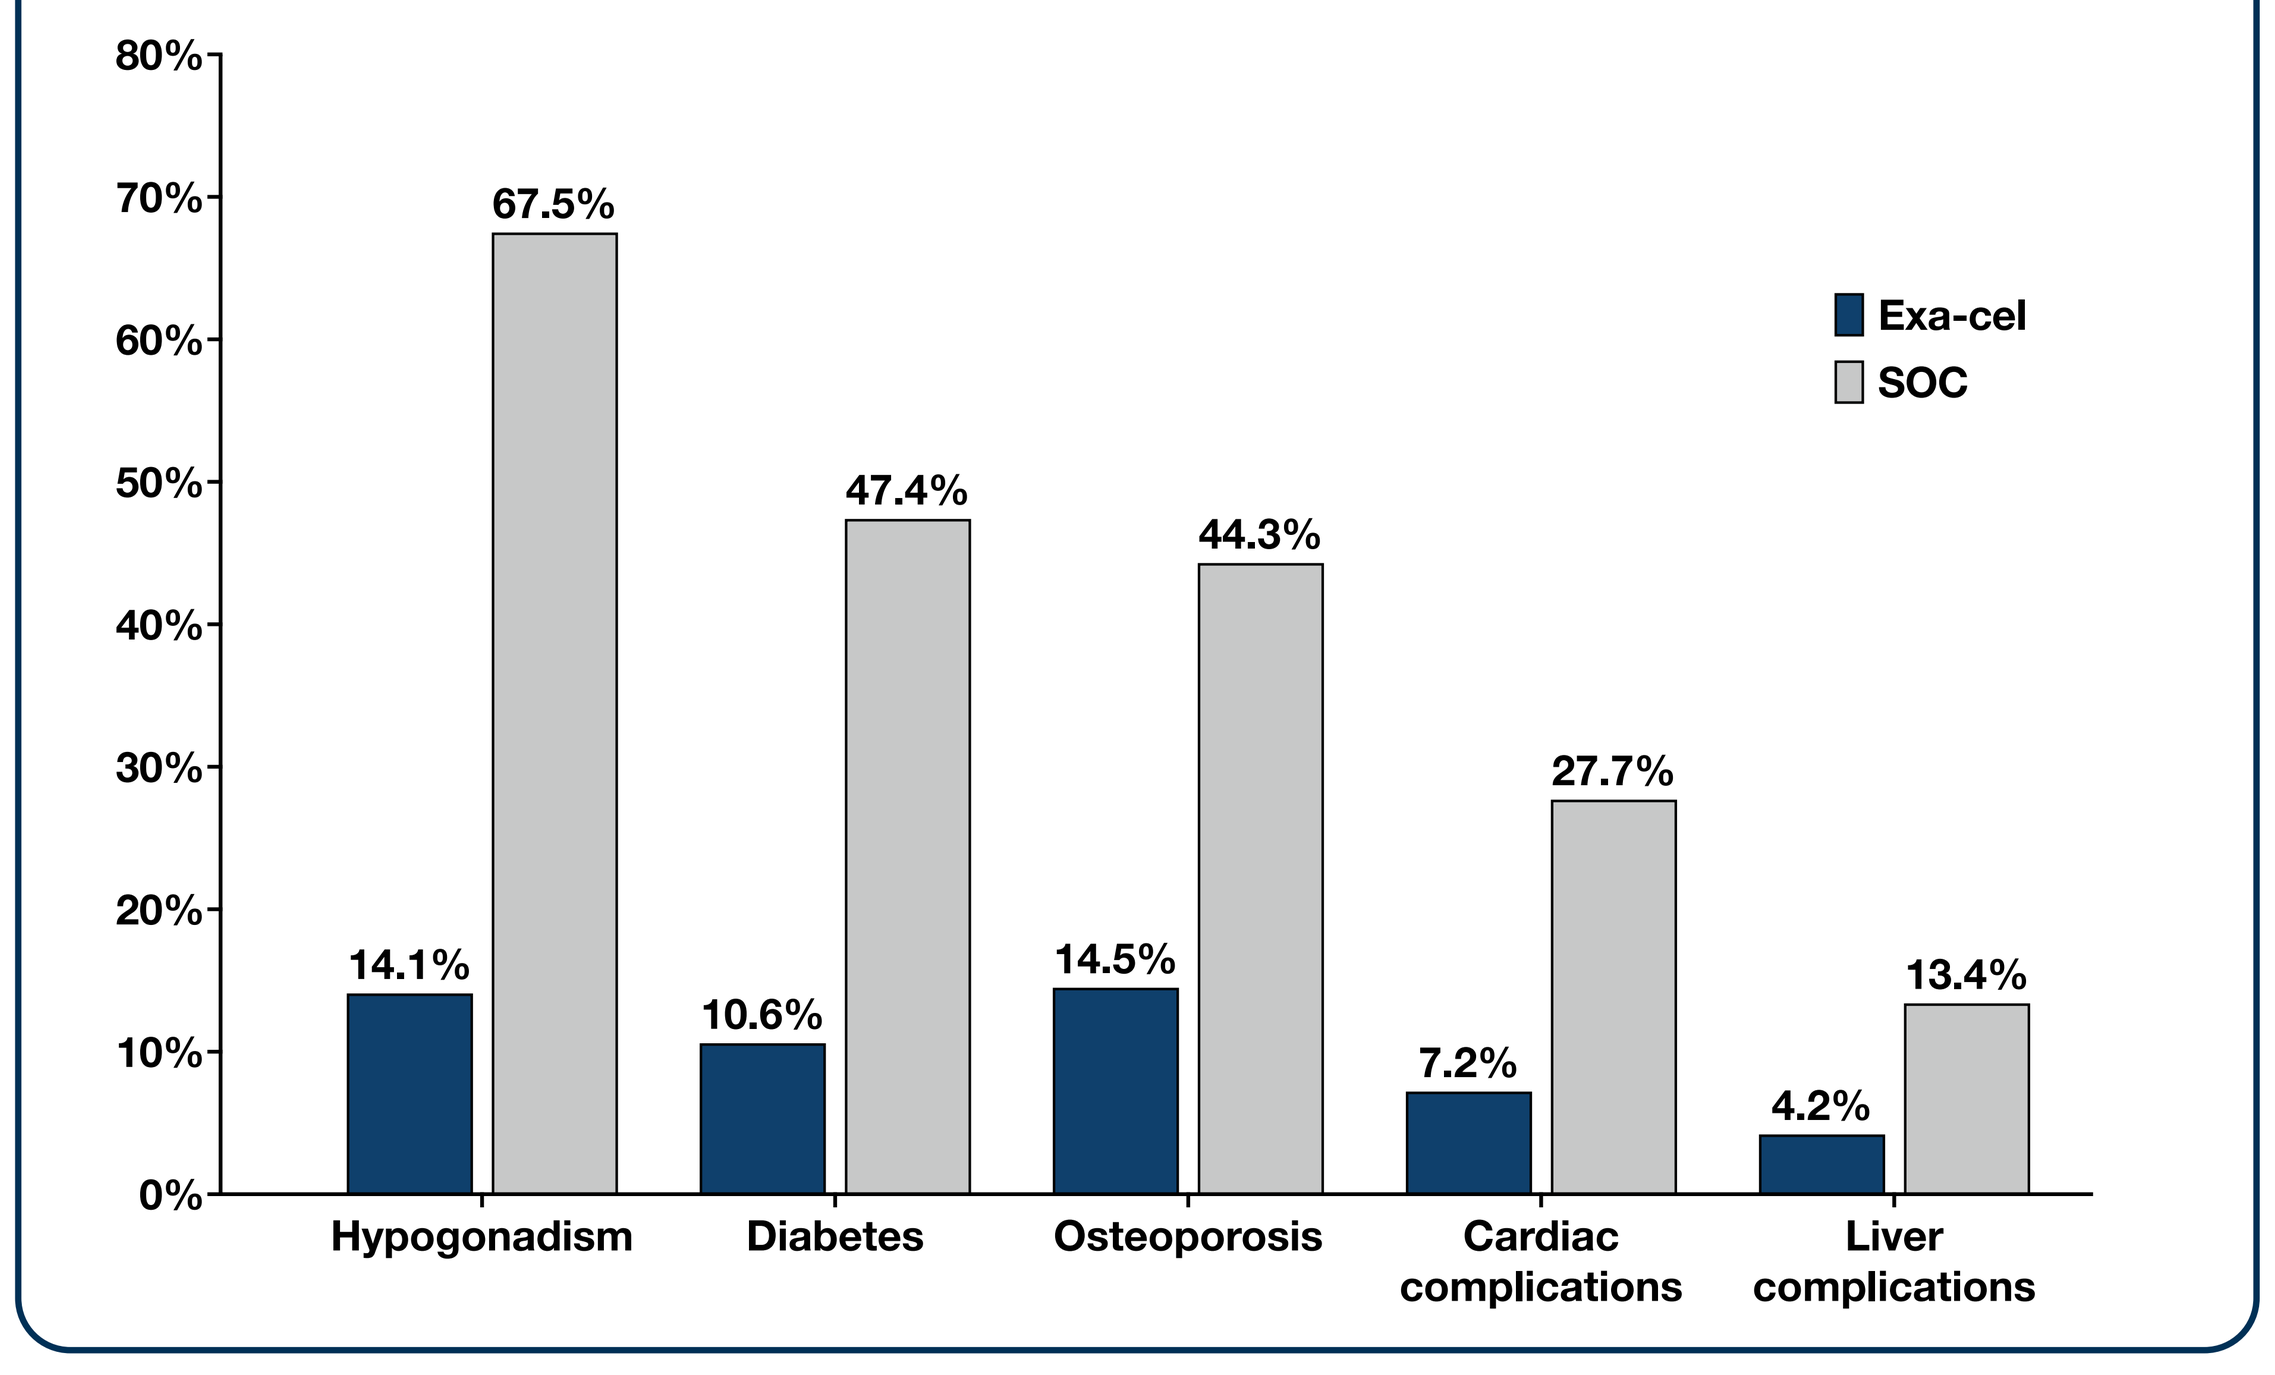
80%
70%
60%
50%
40%
30%
20%
10%
0%
Exa-cel
SOC
14.1%
67.5%
10.6%
47.4%
14.5%
44.3%
7.2%
27.7%
4.2%
13.4%
Hypogonadism
Diabetes
Osteoporosis
Cardiac complications
Liver complications

Scenario Analyses Results

- Patients treated with exa-cel had an increase in survival ranging from 18.1 to 21.4 years compared to SOC, across the various scenarios analyzed (**Table 3**)

Table 3. Scenario Analysis Results

Outcome	Incremental mean LYs (Exa-cel vs SOC)	Incremental mean number of RBCTs (Exa-cel vs SOC)
Base case	18.1	-458.1
Age 12 years	21.7	-488.4
Alternative trial data cut (April 2023)	18.4	-454.1
Alternative efficacy assumption & alternative trial data cut (April 2023)	21.0	-459.8

LY: life years

Limitations

- Healthcare decision analytic models based solely on transfusions and iron levels could oversimplify the complexity of TDT pathophysiology given the impact of anemia and ineffective erythropoiesis
- Complication risks were based on published literature using historical data on patients with TDT from the UK and Europe; additional contemporary Canadian-specific complication risks may more accurately reflect the modeled population
- As a simplifying assumption, the modeled cohort receiving SOC was assumed to maintain initial iron levels and frequency of RBCTs throughout the lifetime horizon
- The model did not estimate the impact of luspatercept on projected outcomes; previous literature suggests including this therapy as part of SOC treatment provides modest improvements in clinical outcomes to patients with TDT[1]
- Lifetime clinical efficacy inputs for exa-cel were based on up to 48.1 months of clinical data; however, given the mechanism of action for exa-cel, treatment durability is expected to be lifelong
- The base case assumption that the iron normalization/change period lasts 5 years was conservative and consistent with previous TDT health technology assessment assumptions[6,15]

CONCLUSIONS

- **Model projections suggest that exa-cel could substantially improve survival and lower the prevalence of TDT-related complications, and reduce disease burden in patients with TDT in Canada compared to treatment with SOC**

Author Disclosures

LK, SK, and **AZ** are employees of Eversana. **CU, MG, SJ,** and **AL** are employees of Vertex Pharmaceuticals Incorporated and may hold stock or stock options in the company.

Acknowledgments

Editorial coordination and support was provided by Nathan Blow, PhD, under the guidance of the authors, and graphic design support was provided by Jonathan Kirk, MS. **NB** and **JK** are employees of Vertex Pharmaceuticals Incorporated, and may own stock or stock options in the company.

References

1. Lal A, Wong T, Keel S, Pagano M, Chung J, Kamdar A, Rao L, Ikeda A, Puthenveetil G, Shah S, Yu J, Vichinsky E. The transfusion management of beta thalassemia in the United States. *Transfusion.* 2021 Oct;61(10):3027-3039. doi: 10.1111/trf.16640. Epub 2021 Aug 28. PMID: 34453453; PMCID: PMC9292563.
2. Muncie HL Jr, Campbell J. Alpha and beta thalassemia. *Am Fam Physician*. 2009 Aug 15;80(4):339-44. PMID: 19678601.
3. Taher AT, Musallam KM, Cappellini MD. β-Thalassemias. *N Engl J Med.* 2021 Feb 25;384(8):727-743. doi: 10.1056/NEJMra2021838. PMID: 33626255.
4. Locatelli F, Lang P, Wall D, Meisel R, Corbacioglu S, Li AM, de la Fuente J, Shah AJ, Carpenter B, Kwiatkowski JL, Mapara M, Liem RI, Cappellini MD, Algeri M, Kattamis A, Sheth S, Grupp S, Handgretinger R, Kohli P, Shi D, Ross L, Bobruff Y, Simard C, Zhang L, Morrow PK, Hobbs WE, Frangoul H; CLIMB THAL-111 Study Group. Exagamglogene Autotemcel for Transfusion-Dependent β-Thalassemia. *N Engl J Med*. 2024 May 9;390(18):1663-1676. doi: 10.1056/NEJMoa2309673. Epub 2024 Apr 24. PMID: 38657265.
5. Shah F, Telfer P, Velangi M, Pancham S, Wynn R, Pollard S, Chalmers E, Kell J, Carter AM, Hickey J, Paramore C, Jobanputra M, Ryan K. Routine management, healthcare resource use and patient and carer-reported outcomes of patients with transfusion-dependent β-thalassaemia in the United Kingdom: A mixed methods observational study. *Eur J Haem.* 2021 Sep 8;2(4):738-749. doi: 10.1002/jha2.282. PMID: 35845207; PMCID: PMC9175788.
6. NICE. Betibeglogene autotemcel for treating transfusion-dependent beta-thalassaemia [ID968]. National Institute of Health and Care Excellence. Updated 2 December 2021. Accessed 12 October 2022. https://www.nice.org.uk/guidance/gid-ta10334/documents/committee-papers
7. Delea TE, Sofrygin O, Thomas SK, Baladi JF, Phatak PD, Coates TD. Cost effectiveness of once-daily oral chelation therapy with deferasirox versus infusional deferoxamine in transfusion-dependent thalassaemia patients: US healthcare system perspective. *Pharmacoeconomics.* 2007;25(4):329-42.
8. Kremastinos DT, Tsetsos GA, Tsiapras DP, Karavolias GK, Ladis VA, Kattamis CA. Heart failure in beta thalassemia: a 5-year follow-up study. *Am J Med*. 2001 Oct 1;111(5):349-54. doi: 10.1016/s0002-9343(01)00879-8. PMID: 11583636.
9. Heald A, Stedman M, Robinson A, Davies M, Livingston M, Alshames R, Moreno G, Gadsby R, Rayman G, Gibson M, de Lusignan S, Whyte M. Mortality Rate Associated with Diabetes: Outcomes From a General Practice Level Analysis in England Using the Royal College of General Practitioners (RCGP) Database Indicate Stability Over a 15 Year Period. *Diabetes Ther*. 2022 Mar;13(3):505-516. doi: 10.1007/s13300-022-01215-1. Epub 2022 Feb 21. PMID: 35187627; PMCID: PMC8934837.
10. Pepe A, Meloni A, Rossi G, Midiri M, Missere M, Valeri G, Sorrentino F, D'Ascola DG, Spasiano A, Filosa A, Cuccia L, Dello Iacono N, Forni G, Caruso V, Maggio A, Pitrolo L, Peluso A, De Marchi D, Positano V, Wood JC. Prediction of cardiac complications for thalassemia major in the widespread cardiac magnetic resonance era: a prospective multicentre study by a multi-parametric approach. *Eur Heart J Cardiovasc Imaging*. 2018 Mar 1;19(3):299-309. doi: 10.1093/ehjci/jex012. PMID: 28200076.
11. Angelucci E, Muretto P, Nicolucci A, Baronciani D, Erer B, Gaziev J, Ripalti M, Sodani P, Tomassoni S, Visani G, Lucarelli G. Effects of iron overload and hepatitis C virus positivity in determining progression of liver fibrosis in thalassemia following bone marrow transplantation. *Blood.* 2002 Jul 1;100(1):17-21. doi: 10.1182/blood.v100.1.17. PMID: 12070002.
12. Ang AL, Tzoulis P, Prescott E, Davis BA, Barnard M, Shah FT. History of myocardial iron loading is a strong risk factor for diabetes mellitus and hypogonadism in adults with β thalassemia major. *Eur J Haematol.* 2014;92(3):229-236.
13. Hippisley-Cox J, Coupland C. Predicting risk of osteoporotic fracture in men and women in England and Wales: prospective derivation and validation of QFractureScores. *BMJ.* 2009 Nov 19;339:b4229. doi: 10.1136/bmj.b4229. PMID: 19926696; PMCID: PMC2779855.
14. Li N, Udeze C, Ly NF, Ingleby FC, Fleming SD, Conner S, Howard J, Shah F. Mortality and Clinical Complications Among Patients With Transfusion-Dependent β-Thalassaemia in England. Presented at 63rd Annual Meeting of the British Society for Hematology. April 2023.
15. Beaudoin FL, Richardson M, Synnott PG, Lancaster V, Fluetsch N, HerceHagiwara B, Campbell JD, Pearson SD, Rind DM. Betibeglogene Autotemcel for Beta Thalassemia: Effectiveness and Value. Institute for Clinical and Economic Review. Updated 2 June, 2022. https://icer.org/wp-content/uploads/2021/11/ICER_Beta-Thalassemia_Evidence-Report_060222-1.pdf

Presented at ISPOR EUROPE, Barcelona, Spain, 17-20 November 2024

Sponsored by Vertex Pharmaceuticals Incorporated