CO90

Matching-Adjusted Indirect Comparison of Efficacy and Safety for Lebrikizumab, Dupilumab and Tralokinumab in patients with moderate-to-severe atopic dermatitis not adequately controlled or non-eligible for cyclosporine

Ezzedine, K.[1,2]; Caillet, G.[3]; Joubert, J.M.[3]; Rand, K[4]; Estévez-Carrillo, A.[4]; Akmaz, B.[3]; Solé-Feu, L.[3]

[1]EA 7379 EpidermE, Université Paris-Est Créteil (UPEC), Créteil, France, [2]Department of Dermatology, Henri Mondor University Hospital, AP-HP, Créteil, France, [3] Almirall S.A.S/ Almiral S.A, Barcelona, Spain, [4] Maths in Health, Klimmen, Netherlands

Sponsored by Almirall S.A.

ISPOR Europe 2024; Barcelona, Spain; November 17-20, 2024

OBJECTIVE

- This study aims to compare the efficacy and safety at week 16 of Lebrikizumab, Dupilumab, and Tralokinumab, administered biweekly in combination with low- to mid-potency topical corticosteroids (TCS), during the induction period in patients not adequately controlled or non-eligible for cyclosporine.

CONCLUSION

- Adjusting for unequal distributions of effect modifiers between trials of patients with moderate to severe AD not adequately controlled or non-eligible for cyclosporine, Lebrikizumab demonstrates no statistically significant differences in efficacy compared to Dupilumab in terms of EASI 75 and IGA 0/1 at week 16.
- Lebrikizumab is significantly superior to Tralokinumab for EASI 75 and shows no statistically significant difference in IGA 0/1.
- Rates of adverse events were similar between Lebrikizumab and both Dupilumab and Tralokinumab.

BACKGROUND

- Atopic dermatitis (AD) is a chronic, relapsing, heterogeneous skin disease characterized by symptoms such as intense itch, sleep disturbances, and skin pain, which negatively impacts patients' sleep, daily activities, and social relationships.
- Ciclosporin A (CsA) is a potent immunosuppressant widely used to manage AD and is the only classic systemic therapy approved for severe AD in Europe. However, it is not effective in all patients and its use is limited by side-effects.
- Lebrikizumab, Dupilumab, and Tralokinumab are monoclonal antibodies that have demonstrated efficacy and safety in clinical trials of moderate-to-severe AD patients with an inadequate response to CsA, or non-eligible for cyclosporine. In clinical practice, these monoclonal antibodies may be used in combination with low- to mid-potency topical corticosteroids (TCS).
- In the absence of head-to-head trials comparing Lebrikizumab, Dupilumab and Tralokinumab, Indirect Treatment Comparison (ITC) can be used to estimate the relative efficacy of these treatments.

METHODS

Data Sources

- **Lebrikizumab**: individual patient data (IPD) from the adult modified intention-to-treat subsample of patients were available from the ADvantage trial (NCT05149313).
- **Dupilumab**: aggregate data from the LIBERTY AD CAFÉ trial (NCT02755649) were extracted from Bruin-Weller et al. [1]
- **Tralokinumab:** aggregate data from the ECZTRA 7 trial (NCT03761537) were extracted from Gutermuth et al. [2]

Matching-Adjusted Indirect Comparison (MAIC)

- Bucher and MAIC are both ITC methodologies. Unlike Bucher, MAIC is specifically designed to reduce bias by adjusting for differences in the distribution of effect modifiers between trials being compared. This methodology is accepted by major Health Technology Assessments (HTA) organisations in Europe, including NICE. The NICE Technical Support Document 18 was followed in this analysis.
- Given the presence of placebo arms in all three clinical trials under consideration, an anchored MAIC was considered.
- Key outcomes of interest include the proportion of patients achieving a 75% improvement in the Eczema Area Severity Index (EASI-75) and an Investigator's Global Assessment score of 0 or 1 (IGA 0/1) at week 16, as well as the overall rate of adverse events.
- Potential effect modifiers were identified and informed by two approaches: recommendations from the literature and analysis of the IPD from the ADvantage trial. Outcomes at week 16 served as dependent variables in logistic regressions, with baseline scores, treatment allocation and their interactions as predictors.

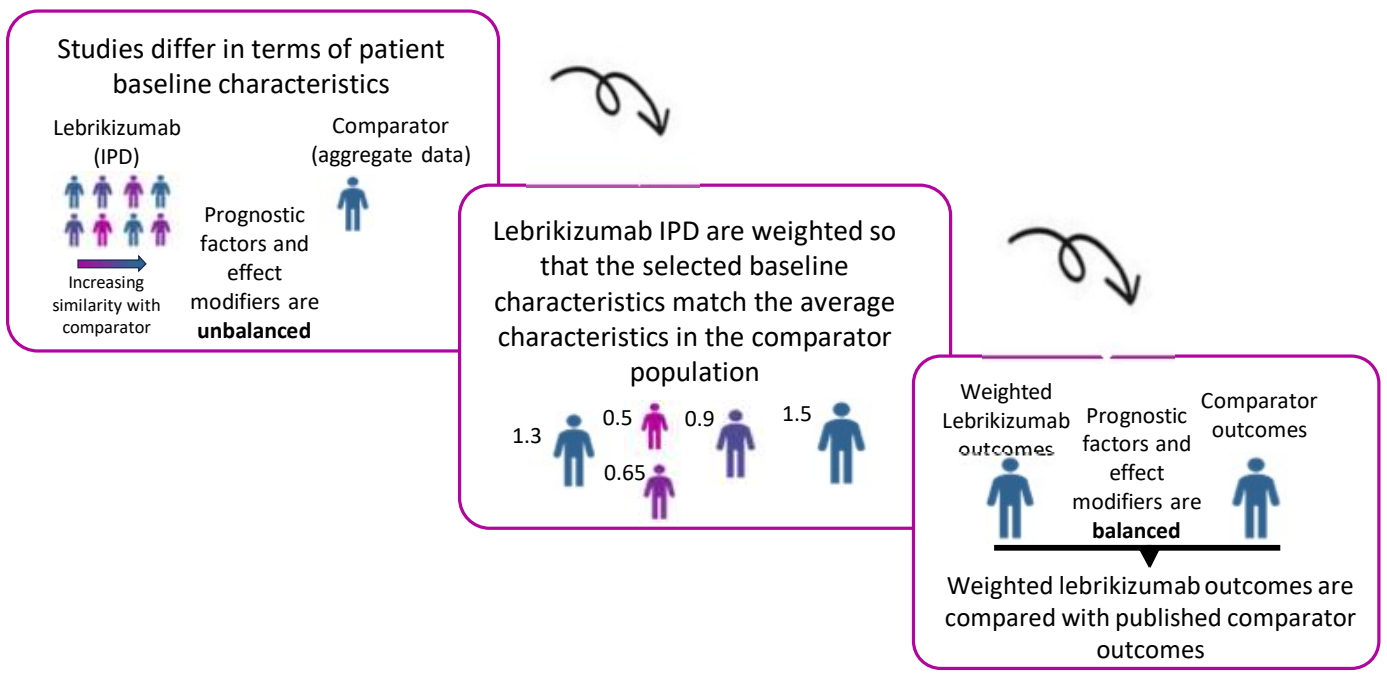
Studies differ in terms of patient baseline characteristics
Lebrikizumab (IPD)
Comparator (aggregate data)
Prognostic factors and effect modifiers are **unbalanced**
Increasing similarity with comparator
Lebrikizumab IPD are weighted so that the selected baseline characteristics match the average characteristics in the comparator population
1.3
0.5
0.65
0.9
1.5
Weighted Lebrikizumab outcomes
Prognostic factors and effect modifiers are **balanced**
Comparator outcomes
Weighted lebrikizumab outcomes are compared with published comparator outcomes

- The ADvantage trial was restricted to patients who matched the eligibility criteria of LIBERTY AD CAFÉ and ECZTRA 7, focusing on the adult-modified intention-to-treat population and excluding adolescents.
- IPD from the ADvantage trial was used to match the baseline characteristics of LIBERTY AD CAFÉ and ECZTRA 7 trials through using propensity score re-weighting. Separate re-weighting processes were conducted for each comparison.
 - In our base case analysis, matching variables were baseline scores on the EASI and affected body surface area. Sensitivity analyses were conducted to test various combinations of matching variables.
 - The quality of the re-weighting process was assessed through the effective sample size (ESS) and the distribution of the weights.
 - The baseline characteristics of the re-weighted ADvantage trial were compared to those of LIBERTY AD CAFÉ and ECZTRA 7 to confirm homogeneity between the populations.
- Risk ratios for EASI-75, IGA 0/1, and adverse events (AE) were estimated through re-weighted rates.

RESULTS

Propensity score re-weighting

- 292 out of 331 participants from the ADvantage study matched the eligibility criteria of the LIBERTY AD CAFÉ and ECZTRA 7 trials.
- Logistic regression analyses on the ADvantage trial IPD identified EASI score and percentage of body surface area (BSA) at baseline as key effect modifiers for EASI 75 and IGA 0/1 outcomes at week 16.
- Matching for EASI and %BSA baseline scores reduced the effective sample size from 292 to 207 (matched to Dupilumab) and 226 (matched to Tralokinumab). The distribution of the weights was acceptable and did not highlight any extreme individuals.
- The distribution of effect modifiers was balanced between the ADvantage and LIBERTY AD CAFÉ/ ECZTRA 7 after reweighing. The base case matching effect modifiers are highlighted in bold.

	Pre-matching ADvantage	LIBERTY AD CAFÉ	Post-matching ADvantage (Dupi)	ECZTRA 7	Post-matching ADvantage (Tralo)
Age, mean (SD)	36.42 (13.98)	35.43 (11.62)	36.17 (14.41)	38.2 (13.14)	36.31 (14.35)
Male, n (%)	151 (52%)	165 (60%)	137.76 (55%)	133 (62%)	138.15 (57%)
AD duration, mean (SD)	26.31 (14.48)	26.19 (8.92)	26.89 (14.6)	30.4 (12.33)	26.71 (14.83)
BSA, mean (SD)	44.08 (18.88)	**53.28 (17.87)**	**53.28 (17.87)**	**55.55 (19.23)**	**55.55 (19.23)**
EASI, mean (SD)	27.49 (9.8)	**30.41 (9.45)**	**30.41 (9.45)**	**33.1 (10.38)**	**33.1 (10.38)**
DLQI, mean (SD)	15.67 (7.13)	16.19 (5.08)	16.31 (7.15)	13.85 (7.64)	16.58 (7.2)
POEM, mean (SD)	20.65 (5.81)	21.93 (4.17)	20.66 (5.86)	19.2 (6.1)	21.12 (5.67)
SCORAD, mean (SD)	64.11 (11.69)	69.66 (9.54)	66.09 (11.58)	67.8 (12.08)	68.24 (11.7)
CsA, n (%)	155 (53%)	207 (75%)	140.93 (56%)	141 (66%)	135.16 (56%)

Matching-Adjusted Indirect Comparison (MAIC)

The MAIC risk ratio (RR) for patients administered Lebrikizumab compared to Dupilumab at week 16 was 1.076 (95% CI: 0.662, 1.750) for EASI-75, 0.867 (95% CI: 0.397, 1.892) for IGA 0/1, and 1.052 (95% CI: 0.834, 1.328) for overall adverse events.

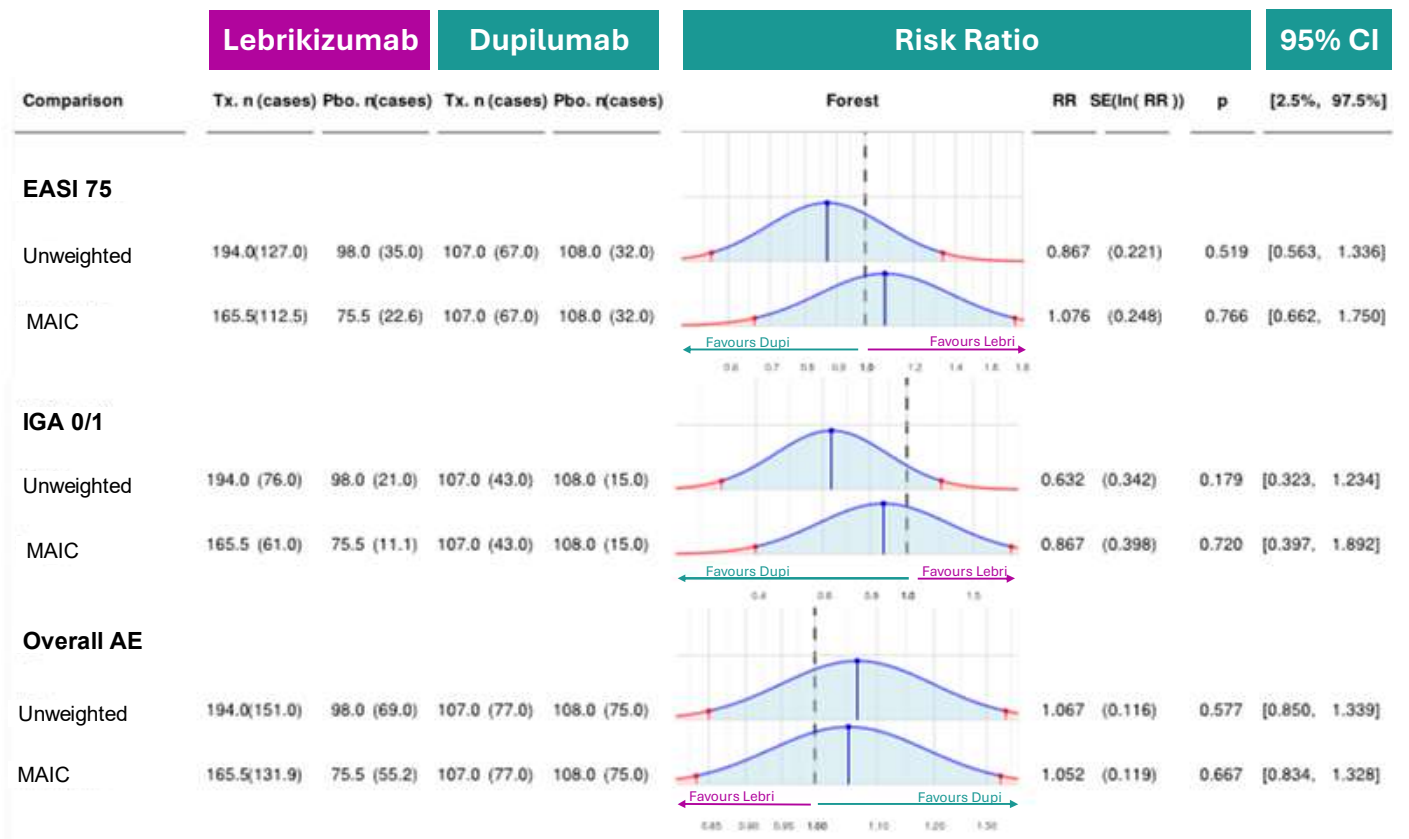

Comparison	Lebrikizumab Tx. n (cases)	Lebrikizumab Pbo. n(cases)	Dupilumab Tx. n (cases)	Dupilumab Pbo. n(cases)	RR	SE(ln(RR))	p	95% CI [2.5%, 97.5%]
EASI 75								
Unweighted	194.0(127.0)	98.0 (35.0)	107.0 (67.0)	108.0 (32.0)	0.867	(0.221)	0.519	[0.563, 1.336]
MAIC	165.5(112.5)	75.5 (22.6)	107.0 (67.0)	108.0 (32.0)	1.076	(0.248)	0.766	[0.662, 1.750]
IGA 0/1								
Unweighted	194.0 (76.0)	98.0 (21.0)	107.0 (43.0)	108.0 (15.0)	0.632	(0.342)	0.179	[0.323, 1.234]
MAIC	165.5 (61.0)	75.5 (11.1)	107.0 (43.0)	108.0 (15.0)	0.867	(0.398)	0.720	[0.397, 1.892]
Overall AE								
Unweighted	194.0(151.0)	98.0 (69.0)	107.0 (77.0)	108.0 (75.0)	1.067	(0.116)	0.577	[0.850, 1.339]
MAIC	165.5(131.9)	75.5 (55.2)	107.0 (77.0)	108.0 (75.0)	1.052	(0.119)	0.667	[0.834, 1.328]

Forest
Favours Dupi
Favours Lebri
Favours Dupi
Favours Lebri
Favours Lebri
Favours Dupi

The MAIC RR for patients treated with Lebrikizumab compared to Tralokinumab was 1.739 (95% CI: 1.160, 2.606) for EASI-75, 1.535 (95% CI: 0.818, 2.878) for IGA 0/1, and 1.147 (95% CI: 0.939, 1.401) for overall adverse events.

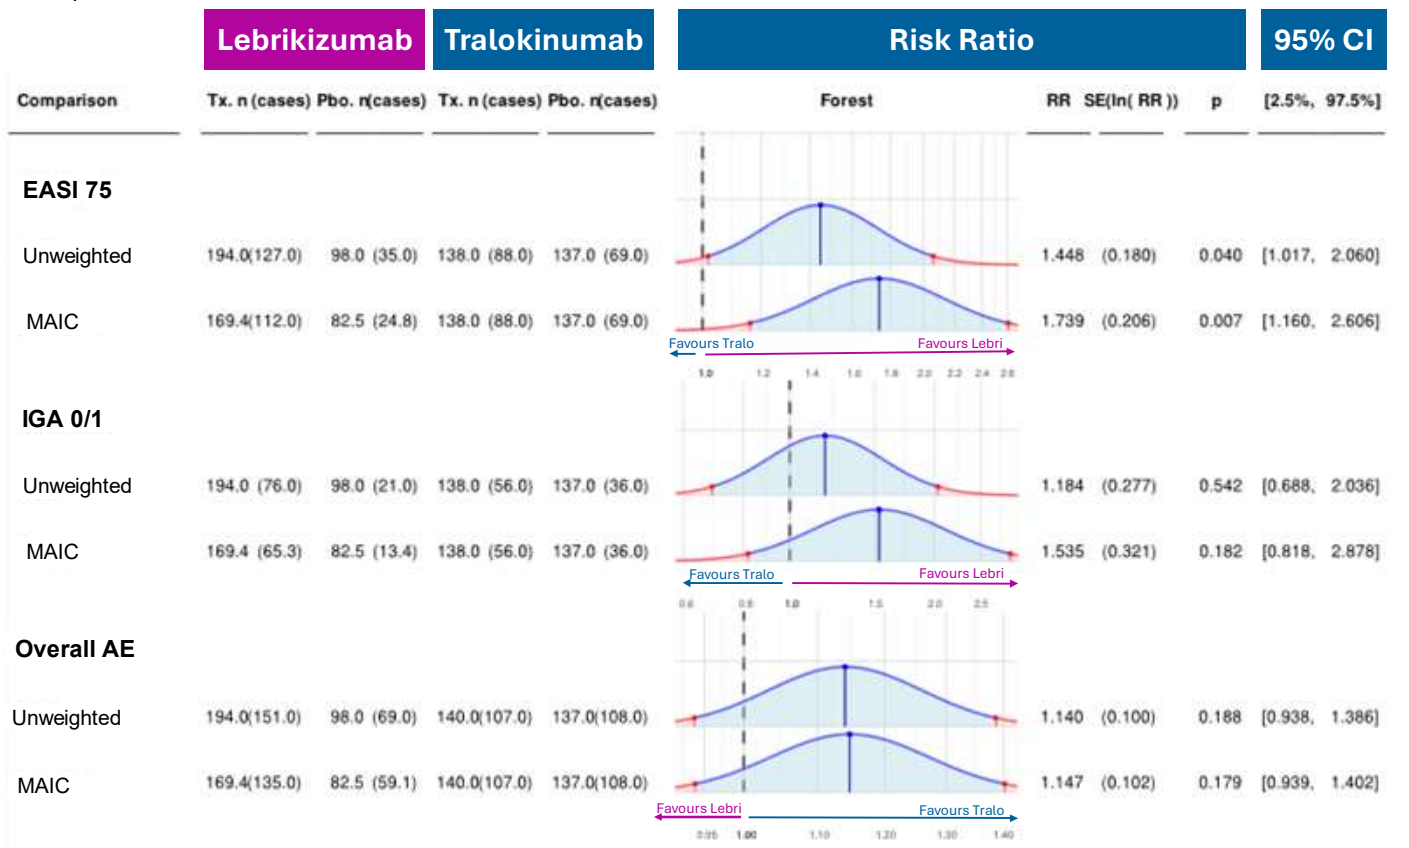

Comparison	Lebrikizumab Tx. n (cases)	Lebrikizumab Pbo. n(cases)	Tralokinumab Tx. n (cases)	Tralokinumab Pbo. n(cases)	RR	SE(ln(RR))	p	95% CI [2.5%, 97.5%]
EASI 75								
Unweighted	194.0(127.0)	98.0 (35.0)	138.0 (88.0)	137.0 (69.0)	1.448	(0.180)	0.040	[1.017, 2.060]
MAIC	169.4(112.0)	82.5 (24.8)	138.0 (88.0)	137.0 (69.0)	1.739	(0.206)	0.007	[1.160, 2.606]
IGA 0/1								
Unweighted	194.0 (76.0)	98.0 (21.0)	138.0 (56.0)	137.0 (36.0)	1.184	(0.277)	0.542	[0.688, 2.036]
MAIC	169.4 (65.3)	82.5 (13.4)	138.0 (56.0)	137.0 (36.0)	1.535	(0.321)	0.182	[0.818, 2.878]
Overall AE								
Unweighted	194.0(151.0)	98.0 (69.0)	140.0(107.0)	137.0(108.0)	1.140	(0.100)	0.188	[0.938, 1.386]
MAIC	169.4(135.0)	82.5 (59.1)	140.0(107.0)	137.0(108.0)	1.147	(0.102)	0.179	[0.939, 1.402]

Forest
Favours Tralo
Favours Lebri
Favours Tralo
Favours Lebri
Favours Lebri
Favours Tralo

DISCLOSURES

Ezzedine, K.: is a professor at Université Paris-Est Créteil (UPEC) and affiliated with EA 7379 EpidermE ; **Caillet, G. and Joubert, J.M. :** are employees of Almirall S.A.S/Almirall S.A; **Rand, K** : is a principal and partner at Maths in Health B.V., a Dutch HEOR and stats consultancy that does work for clients in pharma, government, and education**; Estévez-Carrillo, A.** is an employee of Maths in Health B.V., **Akmaz, B. and Solé-Feu, L.** are employees of Almirall S.A.S/Almirall S.A.

This analysis was funded by Almirall S.A. Copyright © 2024 Almirall , S. A. All rights reserved.

REFERENCES

[1] de Bruin-Weller M, et al. Br J Dermatol. 2018;178(5):1083-1101.
[2] Gutermuth J, et al. Br J Dermatol. 2022;186(3):440-452.
[3] Phillippo DM, et al. NICE DSU technical support document 18: methods for population-adjusted indirect comparisons in submissions to NICE.