Rheumatoid arthritis, cost-effectiveness analysis of biosimilar tocilizumab in Spain

POSTER EE376

Fernando Pérez-Ruiz[1], Carlos Crespo-Diz[2], Carlos Crespo[3,4], Mónica Cerezales[5], Marcelo Alejandro Guigini[6], José Ignacio Peinado-Fabregat[6], Joan Antoni Schoenenberger-Arnaiz[7,8], Mónica Climente[9].

[1]Division of Rheumatology Cruces University Hospital, Barakaldo, Spain. [2]Department of Pharmacy, Health Area of Pontevedra and O Salnés/Pontevedra University Hospital Complex, Spain. [3]Axentiva Solutions S.L., Barcelona, Spain. [4] Department G.M. Statistics, Universidad de Barcelona, Barcelona, Spain [5]Axentiva Solutions S.L., Oviedo, Spain. [6]Fresenius Kabi Spain, S.A.U., Barcelona, Spain. [7]Pharmacy Service of the Arnau de Vilanova University Hospital, Lleida, Spain. [8]Biomedical Research Institute of Lleida (IRBLleida), Lleida, Spain. [9]Department of Pharmacy Doctor Peset University Hospital, Valencia, Spain.

INTRODUCTION

Rheumatoid arthritis (RA) is a chronic inflammatory disease that requires complex management due to its high morbidity. Biosimilars have emerged as effective and more affordable therapeutic alternatives. Clinical guidelines recommend adjusting treatment based on disease activity and comorbidities, aiming for remission or low disease activity. In severe cases, biologics are considered when other treatments have not been effective. Recently, the biosimilar tocilizumab was approved in Spain as a new option to improve the management of patients with high RA activity.

Objective:
To analyse the cost-effectiveness of biosimilar tocilizumab and other available biological or targeted synthetic DMARDs (disease modifying antirheumatic drugs) versus biosimilar infliximab for the treatment of patients suffering moderate or severe rheumatoid arthritis in Spain.

METHODS

A Markov model including the following health states: remission of the disease, low activity, moderate activity, high activity, and death; with a lifetime horizon and from the healthcare system perspective. The model included *abatacept, adalimumab, baricitinib, certolizumab, etanercept, filgotinib, golimumab, infliximab, rituximab, sarilumab,* reference and biosimilar *tocilizumab (bsTCZ), tofacitinib*, and *upadacitinib*, with different administration methods considered for each drug, as specified in Table 1.

Efficacy measured by DiseaseActivity Score (DAS) 28 was retrieved from meta-analysis and additional data from clinical trials; Spanish pharmacological costs were obtained from a hospital pharmacy nationwide database, and ancillary healthcare costs from regional published tariffs for medical resources. Deterministic and probabilistic analysis were run, and the incremental cost-effectiveness ratio (ICER) for the cost per percentage of remission and cost per quality-adjusted life year (QALY) gained were calculated.

RESULTS

The lifetime cost (Figure 1) of *bsTCZ* was the cheapest of all the therapeutic alternatives (€183,741) vs *certolizumab* being the most expensive (€201,972). Percentage of remission (Figure 2) and QALYs (Figure 3) showed better results for *adalimumab, certolizumab*, reference *tocilizumab*, *bsTCZ*, *sarilumab*, *tofacitinib, upadacitinib, baricitinib, filgotinib*, and *abatacept* than *infliximab*. ICERs showed *bsTCZ* as the only therapeutic alternative superior against *infliximab*, being dominant in both cases.

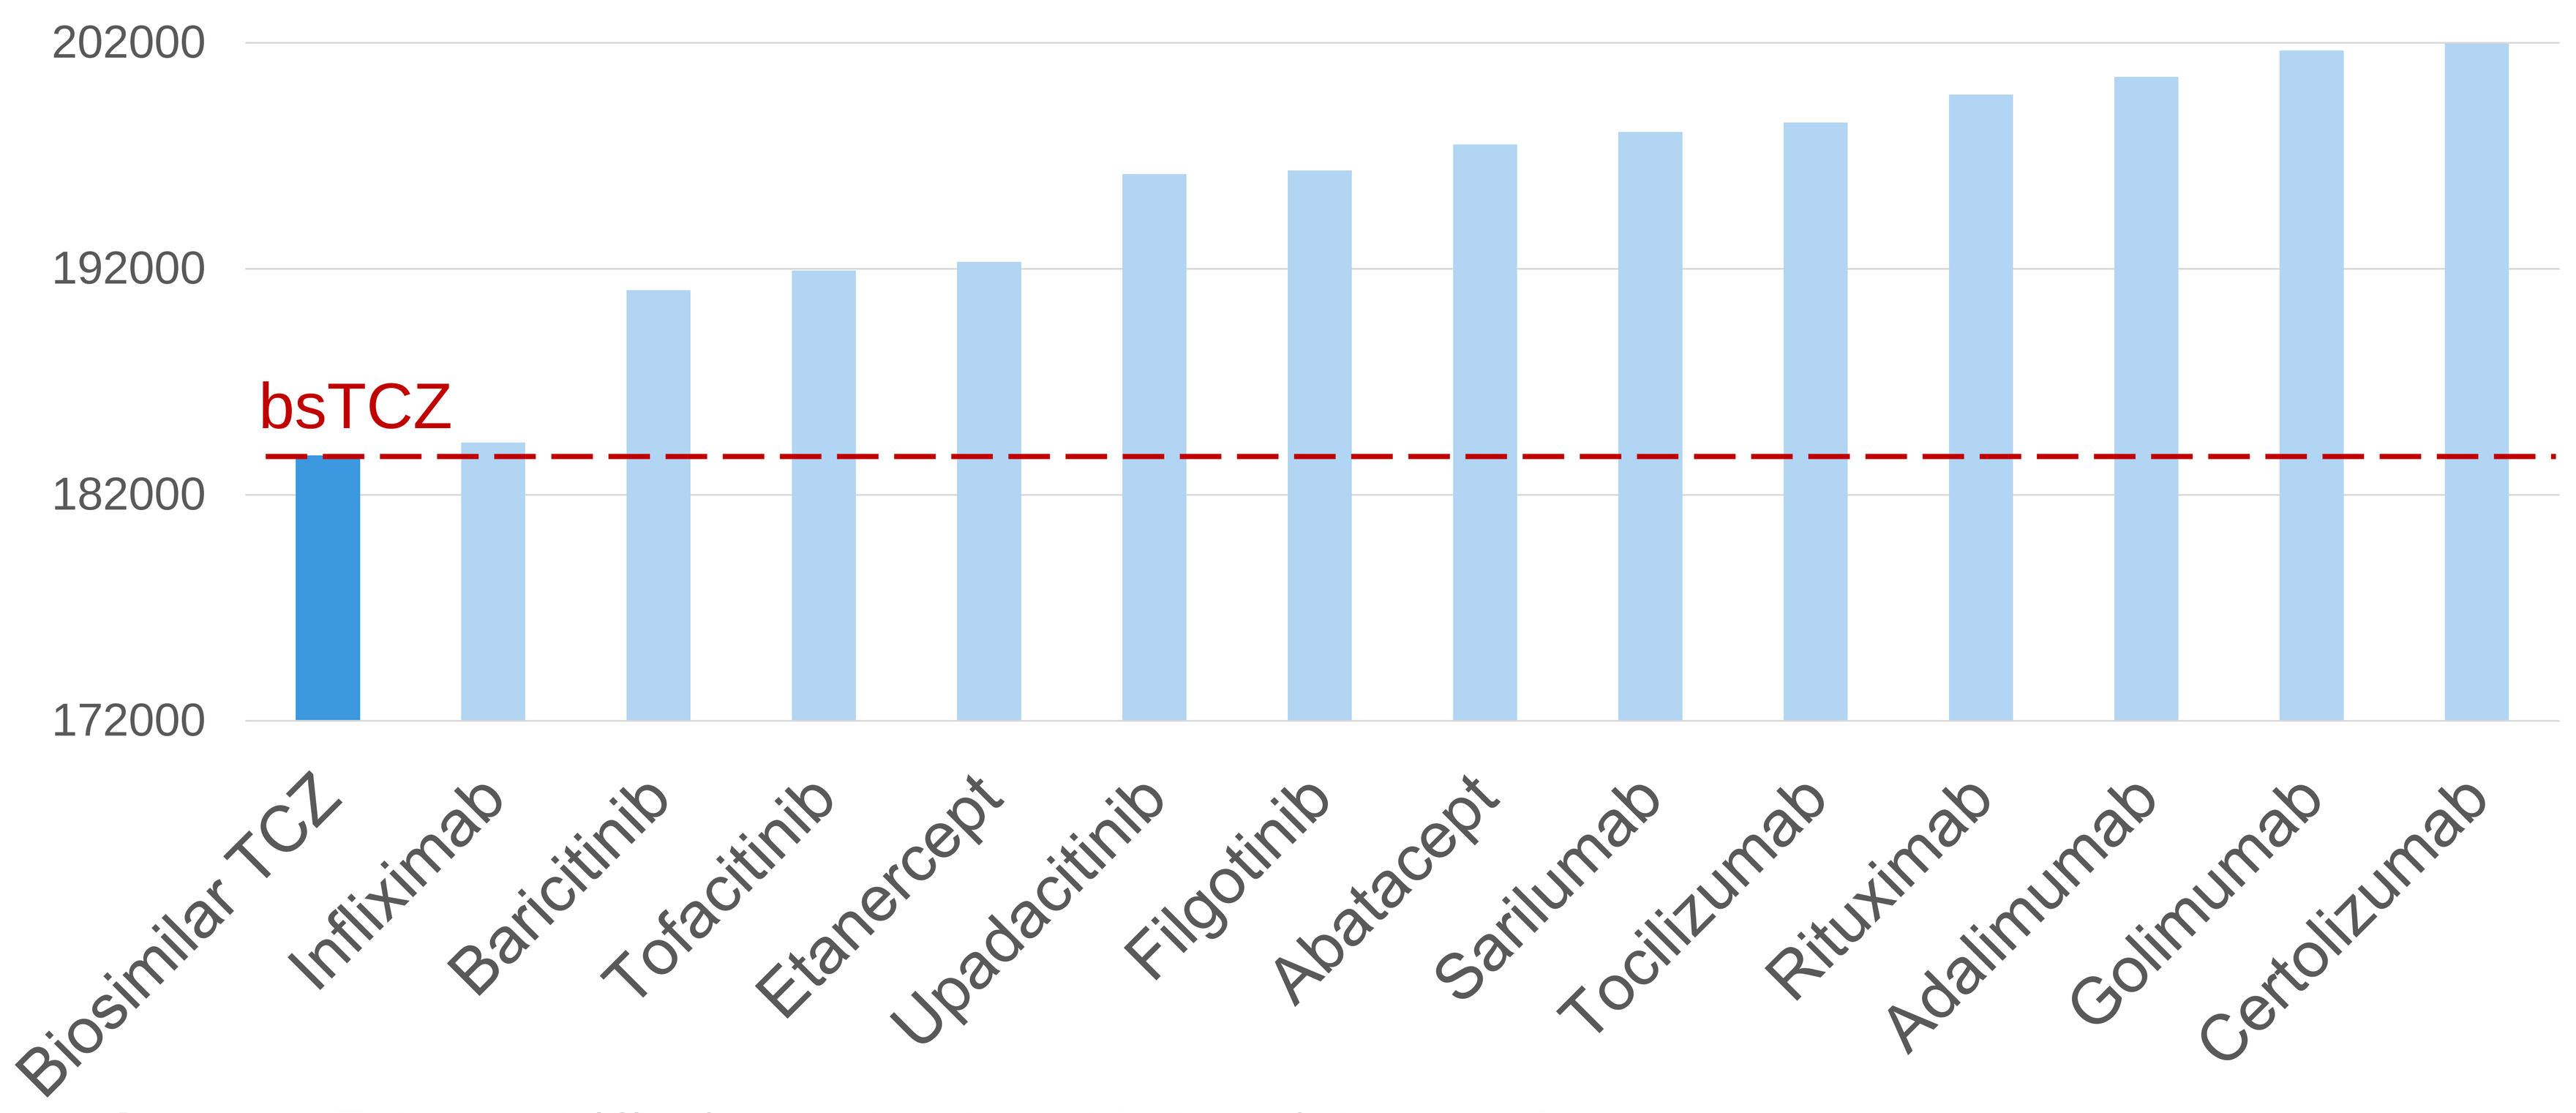
202000
192000
182000
172000
bsTCZ
Biosimilar TCZ
Infliximab
Baricitinib
Tofacitinib
Etanercept
Upadacitinib
Filgotinib
Abatacept
Sarilumab
Tocilizumab
Rituximab
Adalimumab
Golimumab
Certolizumab

Figure 1. *Total cost (€) of each treatment in the lifetime horizon.*

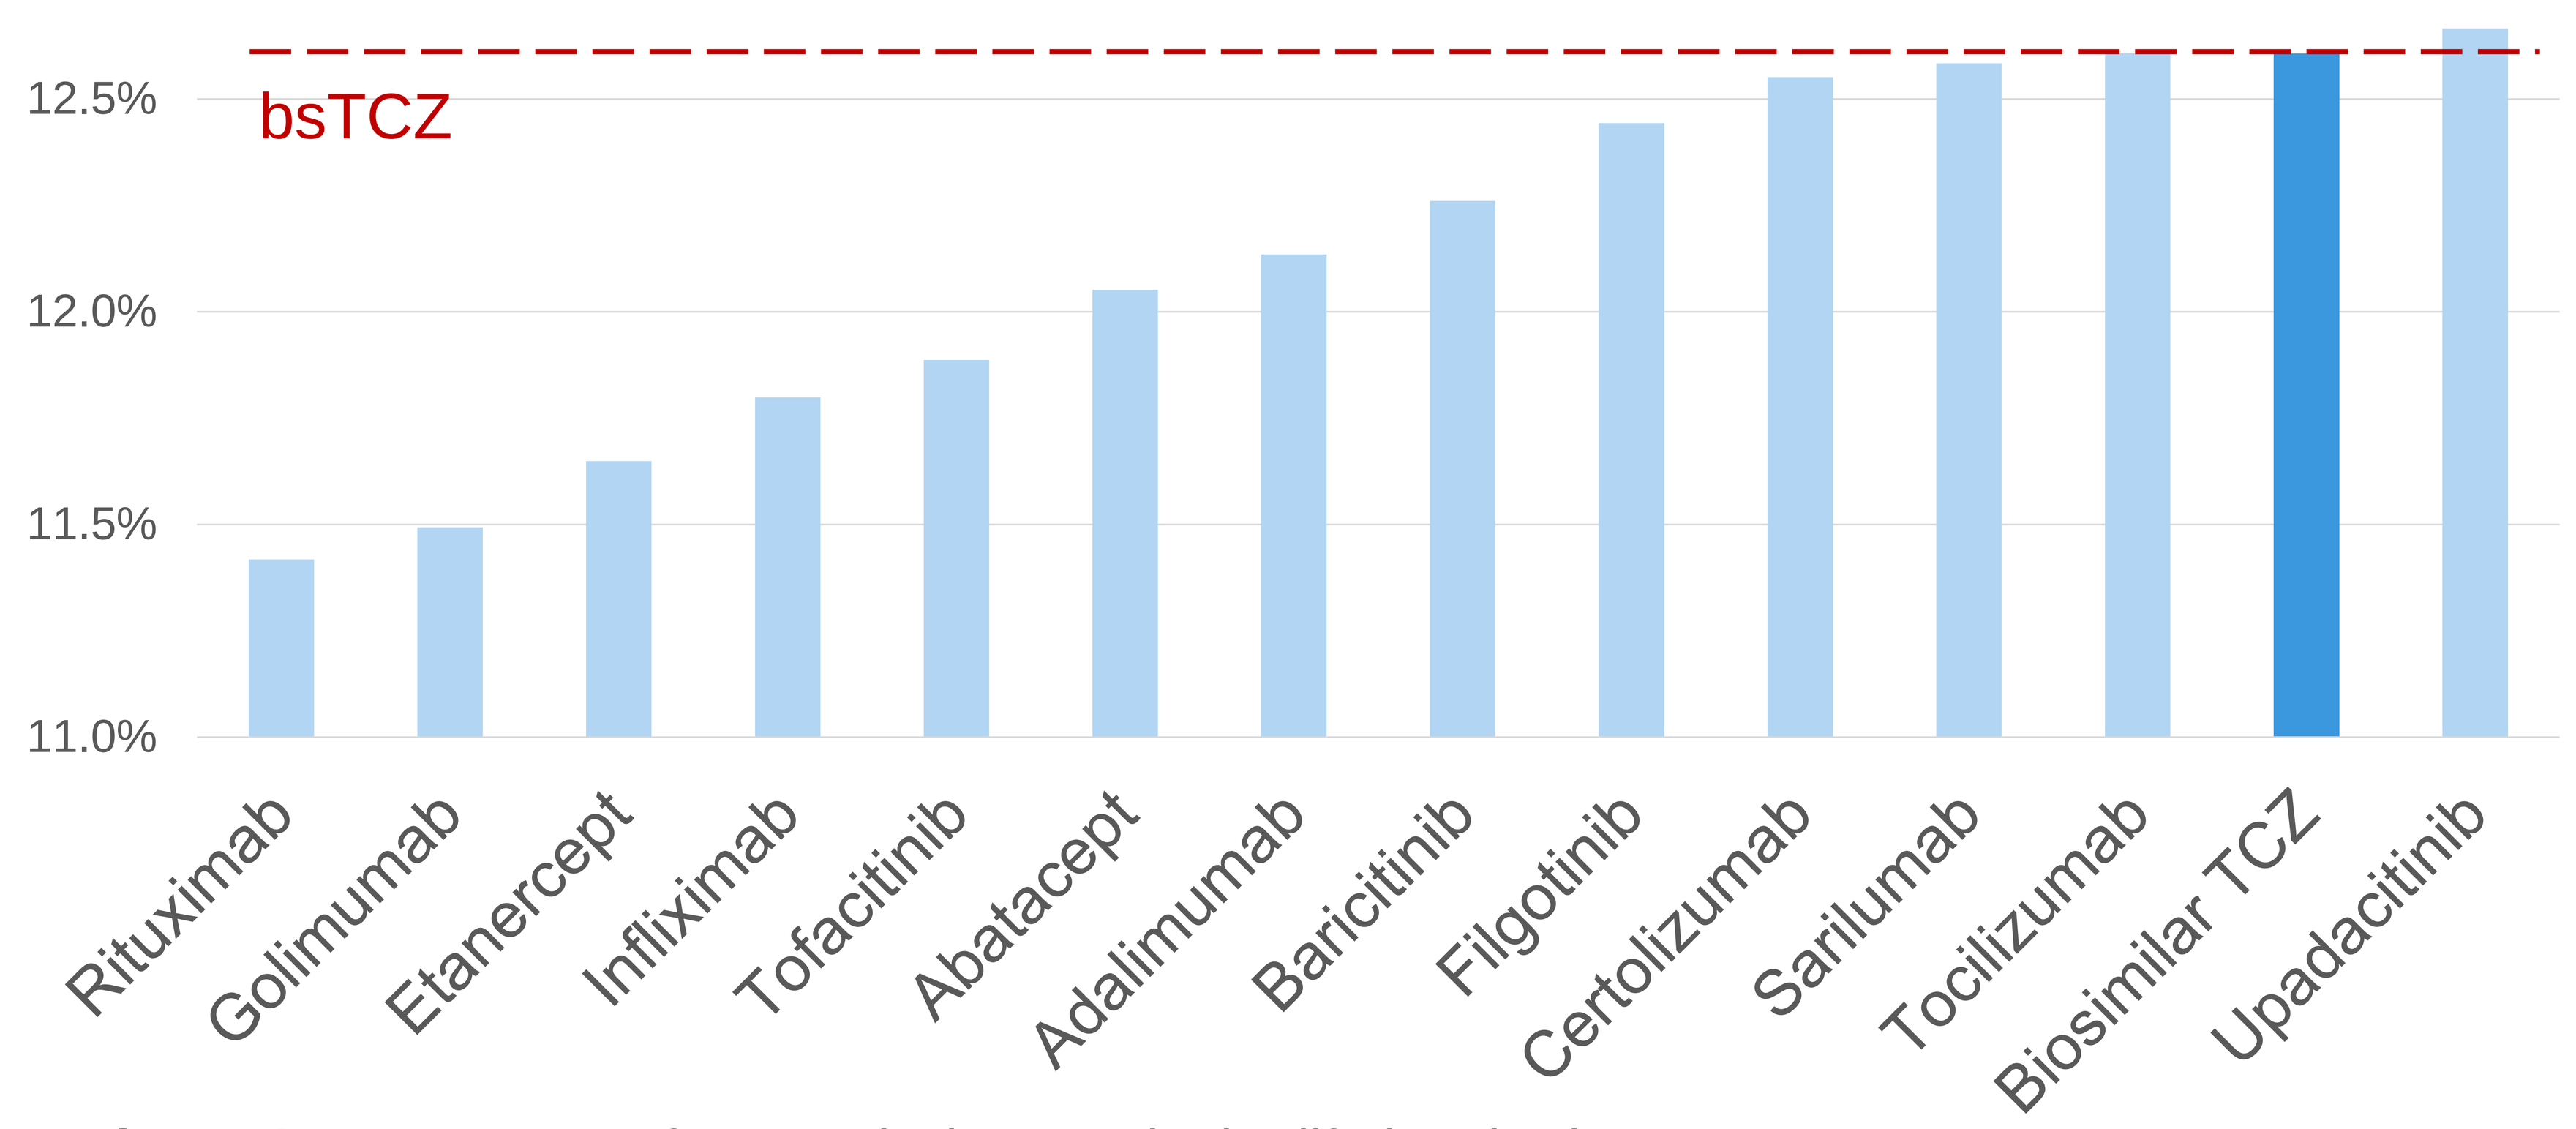
12.5%
12.0%
11.5%
11.0%
bsTCZ
Rituximab
Golimumab
Etanercept
Infliximab
Tofacitinib
Abatacept
Adalimumab
Baricitinib
Filgotinib
Certolizumab
Sarilumab
Tocilizumab
Biosimilar TCZ
Upadacitinib

Figure 2. *Percentage of RA remission rate in the lifetime horizon.*

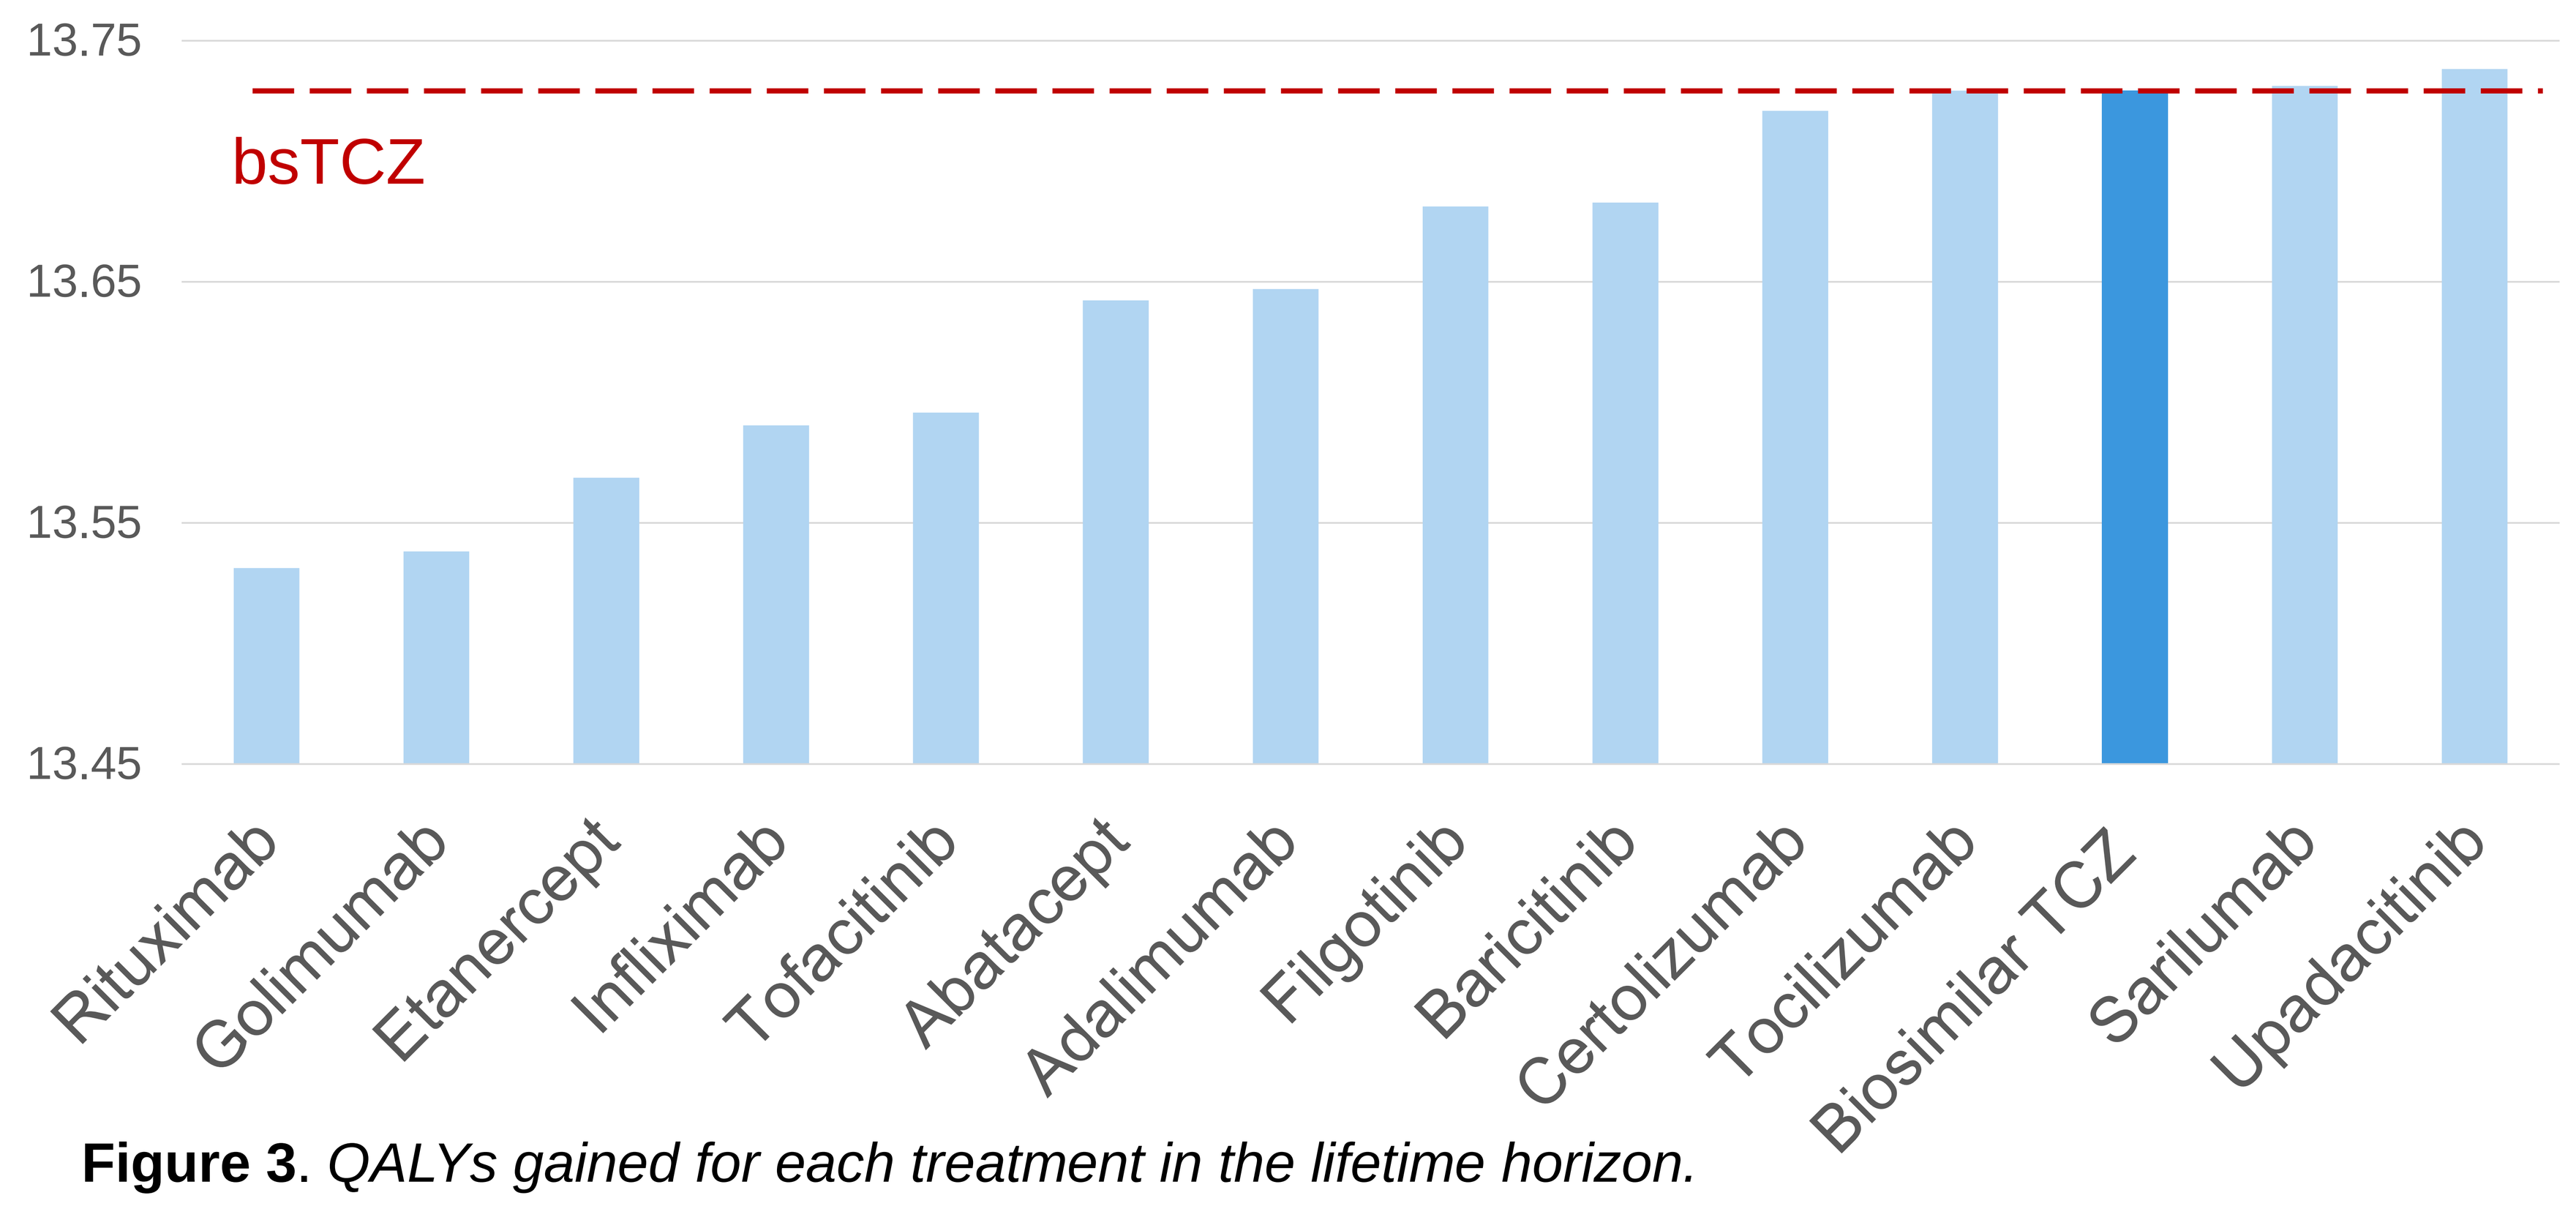
13.75
13.65
13.55
13.45
bsTCZ
Rituximab
Golimumab
Etanercept
Infliximab
Tofacitinib
Abatacept
Adalimumab
Filgotinib
Baricitinib
Certolizumab
Tocilizumab
Biosimilar TCZ
Sarilumab
Upadacitinib

Figure 3. *QALYs gained for each treatment in the lifetime horizon.*

Table 1. *ICER of biosimilar TCZ versus comparators for the treatment of patients with RA in Spain.*

	ICER (€/pRemision) vs infliximab	ICER (€/QALY) vs infliximab
Abatacept (SC)	5.217.561	254.462
Adalimumab (SC)	4.811.243	285.965
Baricitinib (O)	1.460.763	72.953
Certolizumab (SC)	2.344.634	135.293
Etanercept (SC)	Dominated	Dominated
Filgotinib (O)	1.865.703	132.436
Golimumab (SC)	Dominated	Dominated
Rituximab (IV)	Dominated	Dominated
Sarilumab (SC)	1.750.850	97.651
Biosimilar Tocilizumab (SC)	**Dominant**	**Dominant**
Reference Tocilizumab (SC)	1.750.920	101.957
Tofacitinib (O)	8.660.165	1.429.819
Upadacitinib (O)	1.369.146	80.336

IV: intravenous administration; O: oral administration; pRemission: percentage of patients in remission; ICER: incremental cost-effectiveness ratio; SC: subcutaneous administration

Figure 4 shows the cost-effectiveness plane of *bsTCZ* vs. all comparators, representing the 1,000 simulations run. Points on the x-axis indicate changes in QALYs, with positive values favoring *bsTCZ*. Points on the y-axis show cost differences, with most falling in the negative range, indicating lower costs for *bsTCZ*, except when compared to infliximab.

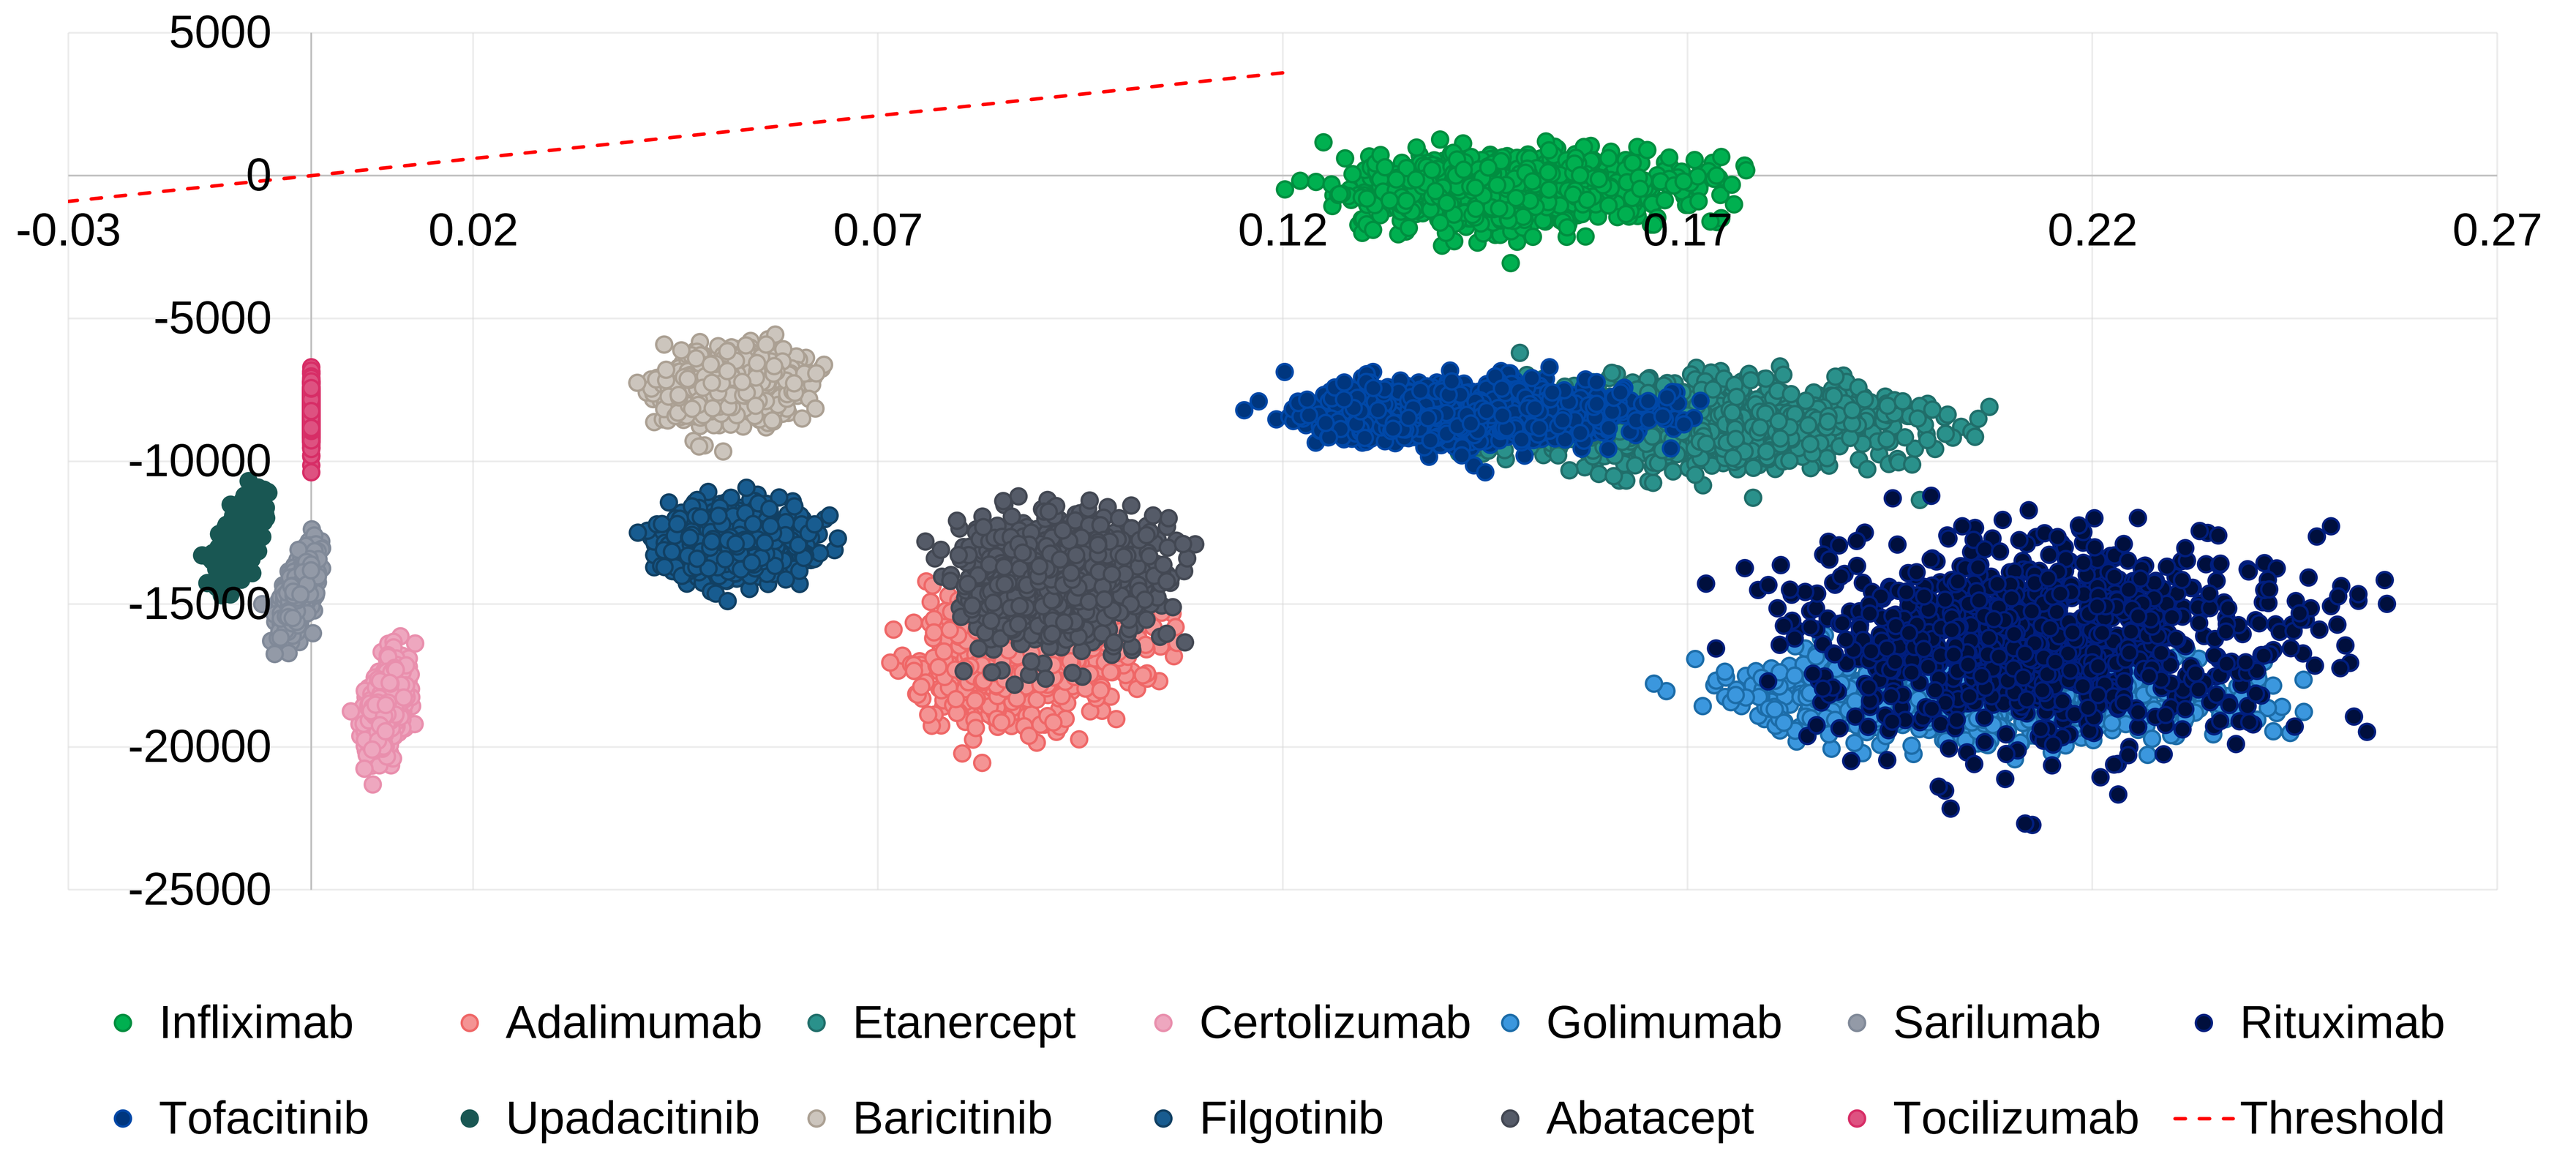
5000
0
-5000
-10000
-15000
-20000
-25000
-0.03
0.02
0.07
0.12
0.17
0.22
0.27
Infliximab
Adalimumab
Etanercept
Certolizumab
Golimumab
Sarilumab
Rituximab
Tofacitinib
Upadacitinib
Baricitinib
Filgotinib
Abatacept
Tocilizumab
Threshold

Figure 4. *Cost-effectiveness plane of the bsTCZ vs. all comparators.*

The deterministic sensitivity analysis identified transition probabilities between disease states and long-term costs of *bsTCZ* as the most influential factors, but across all scenarios, *bsTCZ* remained the **most cost-effective treatment option**.

The probabilistic sensitivity analysis demonstrated that *bsTCZ* was consistently either dominant or cost-effective in all comparisons, with a **100% probability of being cost-effective** at the 22,000€ and 30,000€ thresholds commonly used in Spain.

CONCLUSION

Biosimilar tocilizumab has proven to be a cost-effective alternative for the treatment of rheumatoid arthritis patients, even against the least expensive available biological or targeted synthetic DMARD comparator in Spain in a lifetime-horizon perspective.

FRESENIUS KABI
caring for life

ISPOR 2024, 17-20th November

Carlos Crespo, PhD
ccrespo@axentiva.com