Cost-Effectiveness of Nirmatrelvir/Ritonavir in COVID-19 Patients at High-Risk For Progression in Spain

EE377

Peral C[1], Azanza JR[2], Del Castillo JG[3], Ferrando R[4], Molero JM[5], Soriano A[6], Ariznavarreta A[7], Garí C[7], de Lossada A[1], López-Gómez V[1]

1. Pfizer S.L.U, Madrid, Spain; 2. Clínica Universitaria de Navarra, Pamplona, Spain; 3. Hospital Clínico Universitario de San Carlos, Madrid, Spain; 4. Hospital General Universitario de Castellón, Castellón de la Plana, España; 5. Centro de Salud San Andrés, Madrid, Spain; 6. Hospital Clínic de Barcelona, Barcelona, Spain; 7. Outcomes' 10 (a ProductLife Group Company), Castellón, Spain

INTRODUCTION

- COVID-19 was declared a public health emergency in late 2019[1]. Although vaccines were crucial in combating the virus, new variants and declining vaccination rates have contributed to ongoing cases. As of August 2024, Spain's COVID-19 positivity rate was 37.3 cases per 100,000 inhabitants, with a hospitalization rate of 1.4 per 100,000[2]. These figures highlight the need for additional therapeutic measures, including antiviral treatments like nirmatrelvir/ritonavir (NMV/r)[3].

OBJECTIVES

To evaluate the cost-effectiveness of NMV/r for the treatment of adults with COVID-19 not requiring supplemental oxygen with high-risk factors for developing severe COVID-19, compared to no treatment, from the Spanish National Health System perspective.

METHODS

Design

- A cost-effectiveness model was developed using a hybrid model composed of a decision tree for the first year (acute phase) followed by a Markov model for a lifetime horizon (long-term outcomes; Figure 1)[4-5].
- High-risk patients were defined as those over 65 years of age[6-7] and those with underlying medical conditions such as hypertension, heart or lung disease, diabetes, obesity, or cancer[1]. Data associated with patients over 70 years of age were assumed for modelling the high-risk population.
- For each treatment arm, 1,000 high-risk patients entered the decision tree, segmented into two categories (Figure 1)[8]:
 - Hospitalised: the level of care and hospital mortality associated with COVID-19 were considered.
 - Outpatients: the duration of symptoms was considered, and the absence of mortality was assumed.
- Survivors of the acute phase entered in a two-state Markov model (alive or dead) with annual cycles[8-9].

Figure 1. Model structure (decision tree and Markov model)

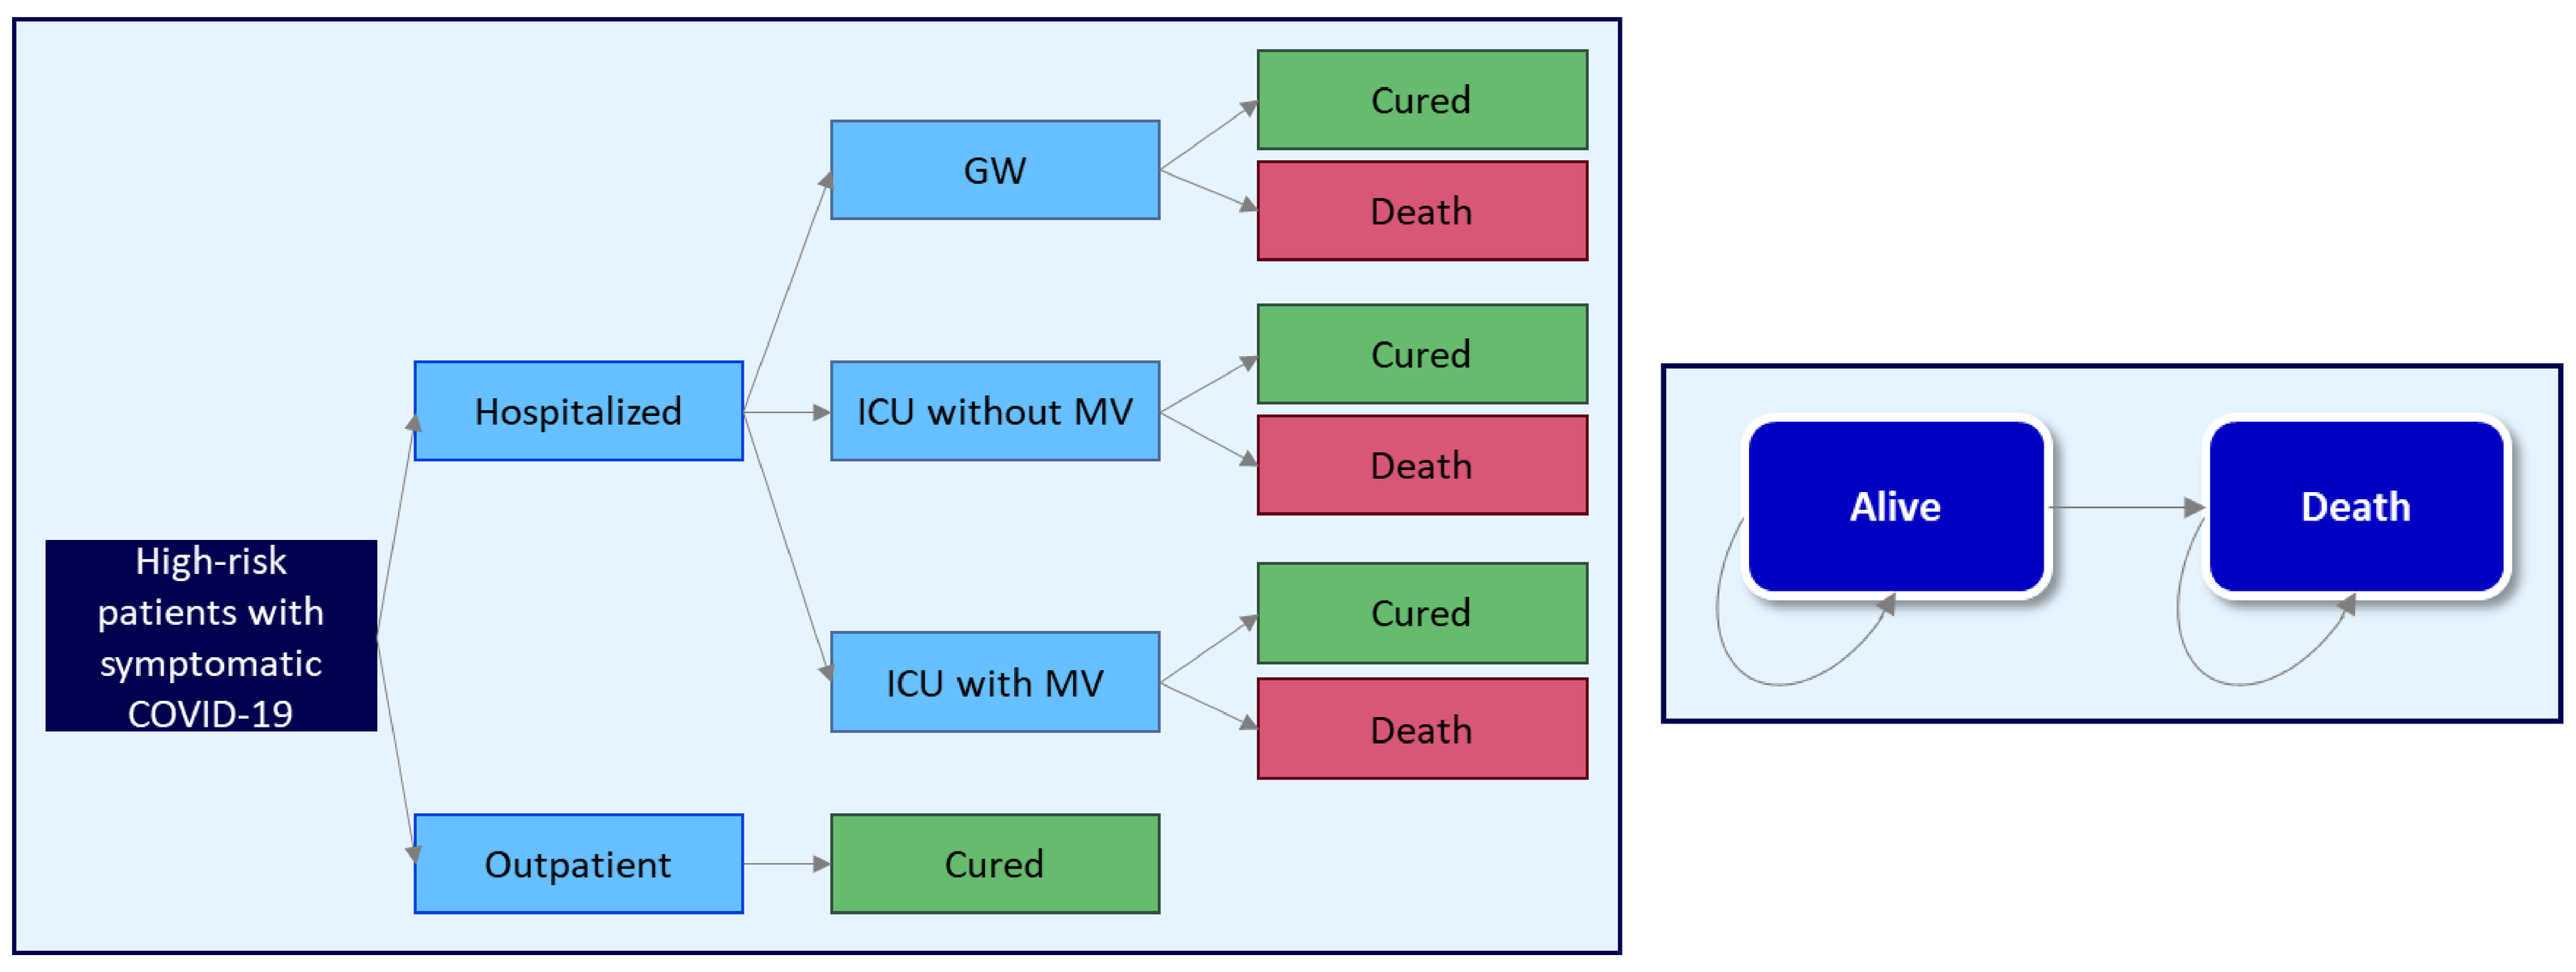
High-risk patients with symptomatic COVID-19
Hospitalized
Outpatient
GW
ICU without MV
ICU with MV
Cured
Death
Cured
Death
Cured
Death
Cured
Alive
Death

GW: general ward; ICU: intensive care unit; MV: mechanical ventilation

Parameters

- All data inputs were validated by a panel of five Spanish experts (four clinicians and one hospital pharmacist) to ensure that the study is consistent with current Spanish clinical practice.
- Assumptions and parameters were based on nationally published data and predominantly derived from publications on the Omicron variant (predominant variant at the time of the study)[2].
- Key efficacy inputs included: risk reductions in the proportion of hospitalisations, deaths and duration of symptoms. Efficacy of NMV/r was derived from the EPIC-HR clinical trial (Table 1)[10].
- Given that the primary objective of vaccination is to mitigate the severity and mortality associated with COVID-19 infection, the vaccination status of patients was also considered[11].

Table 1. Clinical inputs

Parameter	Base case value	References
Treatment efficacy		
Reduction in infection duration	20.0%	Hammond J, et al.[8, 10, 12]
Risk reduction of hospitalisations or deaths	85.8%	
Hospital length of stay/symptom days		
GW	8.6	Specialised Care Register[13]
ICU without MV	12.4	
ICU with MV	7.8	
Symptom days (unvaccinated)	8.3	Menni C, et al.[14]
Symptom days (vaccinated)	4.4	
Lifetime analysis		
Age	68.8	Peláez A, et al.[15]
Increased mortality risk in year after MV discharge	1.33	Lone NI, et al.[16]
Mortality		
Non-hospitalised	0.0%	Expert consensus[9]
Hospitalised patients (unvaccinated)	3.9%	National Centre for Epidemiology[17-19]
Decision tree distribution (proportion)		
Vaccinated	58.1%	Ministry of Health[11]
Hospitalised (unvaccinated)	27.8%	National Centre for Epidemiology[19]
Hospitalised (vaccinated)	6.4%	
ICU	3.0%	
MV	46.4%	Portmann L, et al.[20]

GW: general ward; ICU: intensive care unit; MV: mechanical ventilation

- To capture the quality of life, the model applied utility and disutility values associated with hospitalisations and disease symptoms (Table 2)[21-22].

Table 2. Utilities and disutilities

Parameter	Base case value	References
Baseline utility	0.836	Janssen MF, et al.[21]
Disutility for hospitalised – GW	-0.640	Goswami H, et al.[22]
Disutility for hospitalised – ICU without MV	-0.570	
Disutility for hospitalised – ICU with MV	-0.836	
Disutility for symptom day	-0.290	
Annual disutility post ICU-MV (first year)	-0.13	Sheinson D, et al.[23]
Annual disutility post ICU-MV (years 2 to 5)	-0.04	

GW: general ward; ICU: intensive care unit; MV: mechanical ventilation

- Drug acquisition cost and resource use associated with diagnosis, prescribing and initial follow-up were considered for all patients. Hospitalisation costs for the acute phase were accounted for inpatients. Lifetime average healthcare costs per person were considered until death. All costs were presented in 2024 euros (€) (Table 3)[24-26].

Table 3. Cost inputs

Parameter	Base case value	Source
NMV/r list price*	€832.5	BotPlus[26]
Practitioner's office	€56.36	eSalud[25]
GW - cost per day	€916.1	
ICU without MV - cost per day	€1,310.1	
ICU with MV- cost per day	€1,622.5	
Post-discharge costs- cost per case	€113.3	
Annual healthcare spending per capita	€2,001.0	Ministry of Health[24]

* the rebate established in Spanish Royal Decree-Law 8/2010 was applied (7.5%)[26]; NMV/r: nirmatrelvir/ritonavir; GW: general ward; ICU: intensive care unit; MV: mechanical ventilation

- The model reported outputs including total costs, clinical benefits (outpatient symptom days, number of hospitalisations, ICU admissions, and deaths), and quality-adjusted life-years (QALYs). Both costs and outcomes were discounted at a rate of 3% per year[27].
- The analysis was expressed as incremental cost-effectiveness ratio (ICER): incremental cost per QALY gained (willingness-to-pay [WTP] threshold: €25,000/QALY[28]). One way sensitivity analysis (OWSA) and probabilistic sensitivity analysis (PSA) were performed.

RESULTS

Cost-effectiveness

- NMV/r was found to be a dominant strategy compared with no treatment, resulting in a cost reduction of €169.69 per patient and a QALYs' increase of 0.05 (Table 4).
- NMV/r reduced outpatient symptom days by 0.205 and decreased the number of hospitalisations (0.022 vs 0.154 hospitalisations per patient treated with NMV/r and no treatment, respectively) (Table 4).

Table 4. Base-case and clinical benefit results per patient

	Outcomes	NMV/r	No treatment	Difference
Base-case results	Costs	€28,495.29	€28,664.99	-€169.69
	QALYs	11.40	11.35	0.05
	ICER	Dominant		
Clinical benefit results	Outpatient symptom days	4.698	4.903	-0.205
	Hospitalisations	0.022	0.154	-0.132
	ICU admissions	0.001	0.005	-0.004
	Deaths	0.000	0.005	-0.005

NMV/r: nirmatrelvir/ritonavir; ICER: incremental cost-effectiveness ratio; QALYs: quality-adjusted life-years; ICU: intensive care unit

- Sensitivity analysis confirmed the robustness of the results. Variations in the parameters analysed in the OWSA did not affect the dominance results.
- The PSA showed that NMV/r was dominant or cost-effective in 100% of simulations (67.7% dominant and 32.3% cost-effective), considering the WTP threshold of €25,000 per QALY gained (Figure 2).

Figure 2. Probabilistic sensitivity analysis scatterplot

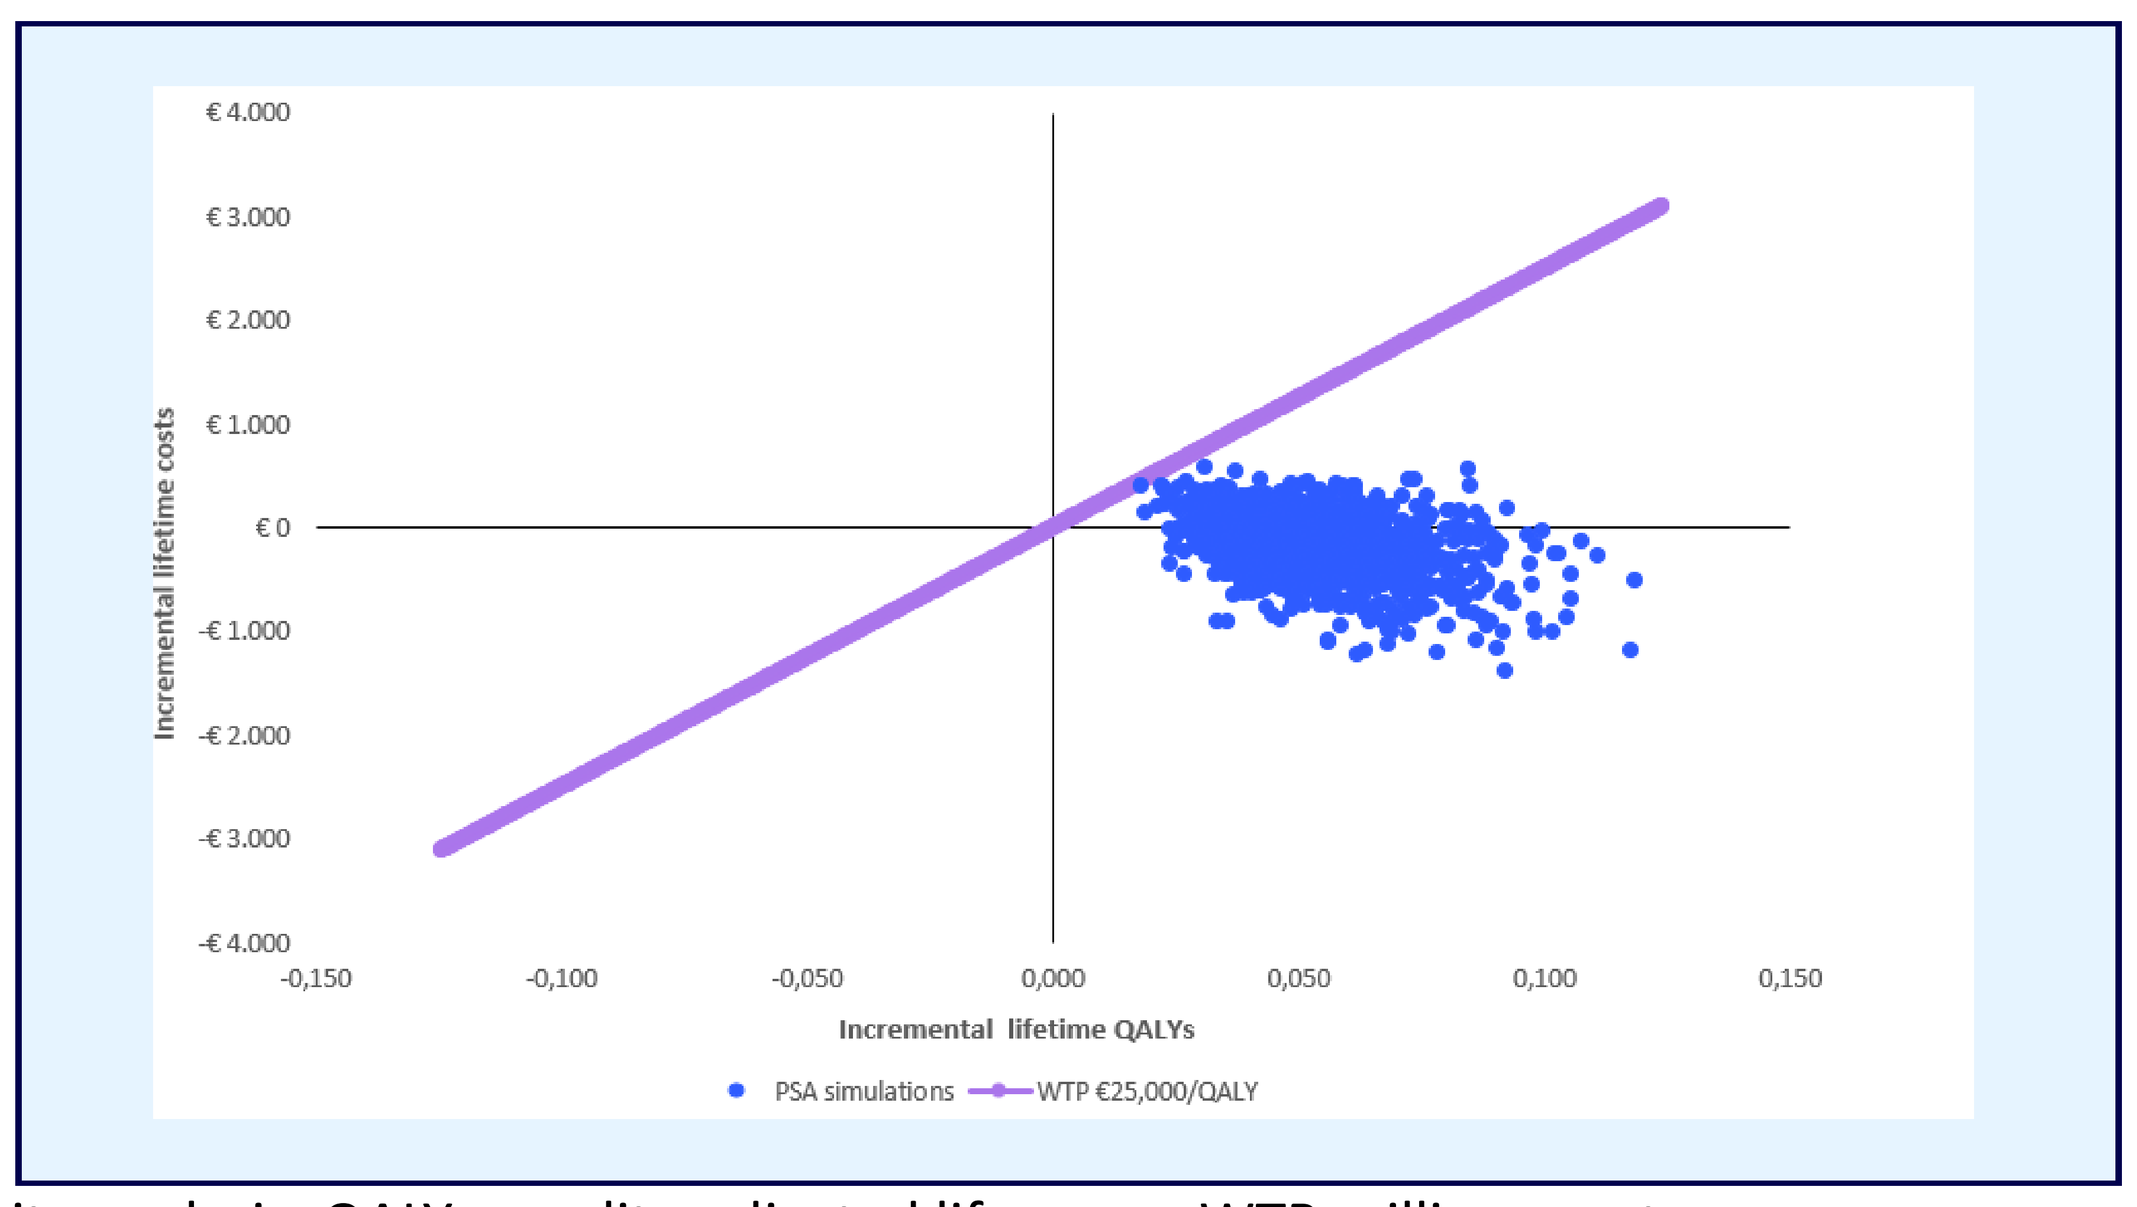
€ 4.000
€ 3.000
€ 2.000
€ 1.000
€ 0
-€ 1.000
-€ 2.000
-€ 3.000
-€ 4.000
Incremental lifetime costs
-0,150
-0,100
-0,050
0,000
0,050
0,100
0,150
Incremental lifetime QALYs
PSA simulations
WTP €25,000/QALY

PSA, probabilistic sensitivity analysis; QALYs, quality-adjusted life-years; WTP, willingness-to-pay

CONCLUSIONS

NMV/r is a dominant option compared to no treatment in high-risk adult patients with symptomatic COVID-19 not requiring supplemental oxygen in Spain.

REFERENCES

1. WHO. Coronavirus disease (COVID-19); 2. Vigilancia centinela de Infección Respiratoria Aguda: gripe, COVID-19 y VRS; 3. Sanyaolu A, et al. Ther Adv Vaccines Immunother. 2022;10:25151355221097559; 4. Carlson J, et al. Value in Health. 2024;27(2):164-72; 5. Kalogeropoulou M, et al. Value in Health. 2023;26(12):S340-S1; 6. Candel FJ, Et al. Viruses. 2023;15(7); 7. Recomendaciones SEIMC para el manejo clínico de pacientes con COVID-19; 8. AEMPS. Ficha técnica Paxlovid; 9. All data inputs and assumptions were validated by an expert panel of five Spanish healthcare professionals (four clinicians and one hospital pharmacist) ; 10. Hammond J, et al. N Engl J Med. 2022;386(15):1397-408; 11. Gestión integral de la vacunación COVID-19 (2023-2024). Ministry of Health; 12. Hammond J, et al. Open Forum Infectious Diseases. 2022;9(Supplement_2); 13. RAE-CMBD. Desde 2016 en adelante; 14. Menni C. et al. The Lancet. 2022;399(10335):1618-24; 15. Peláez A, et al. Med Clin (Barc). 2024;162(11):523-31; 16. Lone NI, et al. Am J Respir Crit Care Med. 2016;194(2):198-208; 17. Efectividad de la campaña de vacunación frente a COVID-19 en España en otoño de 2022; 18. Gestión integral de la vacunación COVID-19 (2020-2023); 19. Situación de COVID-19 en España; 20. Portmann L, et al. JAMA Netw Open. 2023;6(2):e2255599; 21. Janssen MF, et al. Eur J Health Econ. 2019;20(2):205-16; 22. Goswami H, et al. Pharmacoeconomics. 2022;40(7):699-714; 23. Sheinson D, et al. Ther. 2021;38(4):1811-31; 24. Gasto sanitario público, Ministry of health; 25. Gisbert R, et al. Barcelona: Oblikue Consulting, S.L; 26. Bot Plus Web; 27. Guía de evaluación económica e impacto presupuestario en los informes de evaluación de medicamentos. Grupo Génesis; 28. Sacristán JA, et al. Gac Sanit. 2020;34(2):189–93.

DISCLOSURE

This study was sponsored by Pfizer S.L.U. CP, AdL and VLG are employees of Pfizer S.L.U. AA, and CG are employees of Outcomes'10 S.L. which received funding from Pfizer S.L.U. for the analysis.

Pfizer

ISPOR Europe 2024
17-20 November | Barcelona, Spain

OUTCOMES10
A PRODUCTLIFEGROUP COMPANY