Impact of Patient Recall and Reporting Intervals on Real-World Attack Reductions After Berotralstat Initiation in Hereditary Angioedema Patients without C1-Inhibitor Deficiency

PCR233

Lorena Lopez-Gonzalez,[1] Sean D. MacKnight,[2] François Laliberté,[2] Robert C. Schell,[3] Sandra Nestler-Parr,[1] Meri LiVecchi,[1] Patrick Gillard[1]

[1]BioCryst Pharmaceuticals, Inc., Durham, NC, USA; [2]Groupe d'Analyse, Ltée, Montreal, QC, Canada; [3]Analysis Group, Inc., Menlo Park, CA, USA

BACKGROUND

- Hereditary angioedema (HAE) is a rare disease characterized by painful, recurrent, and potentially life-threatening attacks of swelling of the skin and mucous membranes.[1]
- Some patients with HAE experience bradykinin-mediated swelling like HAE-C1-INH despite having normal C1-INH level and function (HAE-nl-C1-INH).[2]
- Prior studies have evaluated the real-world effectiveness of berotralstat, a targeted, once-daily oral medication for the prevention of HAE attacks,[3] on attack rate reductions over 90-day follow-up intervals and using a 90-day baseline recall period.
- To demonstrate the robustness of attack rate reduction findings, this study evaluated attack rate reductions by using a 30-day baseline recall period in addition to the 90-day baseline recall period among patients without C1-inhibitor deficiency (HAE-nl-C1-INH).

METHODS

- This retrospective, real-world study used data from Optime Specialty Pharmacy, the sole dispenser of berotralstat in the US, from December 15, 2020, to January 8, 2024.
- The follow-up period spanned from the index date *(first berotralstat dispensing date)* to the last berotralstat dispensing date during the study period; no patient assessment data were collected after the last berotralstat dispensing (**Figure 1**).

Figure 1. Retrospective Pre–Post Study Design

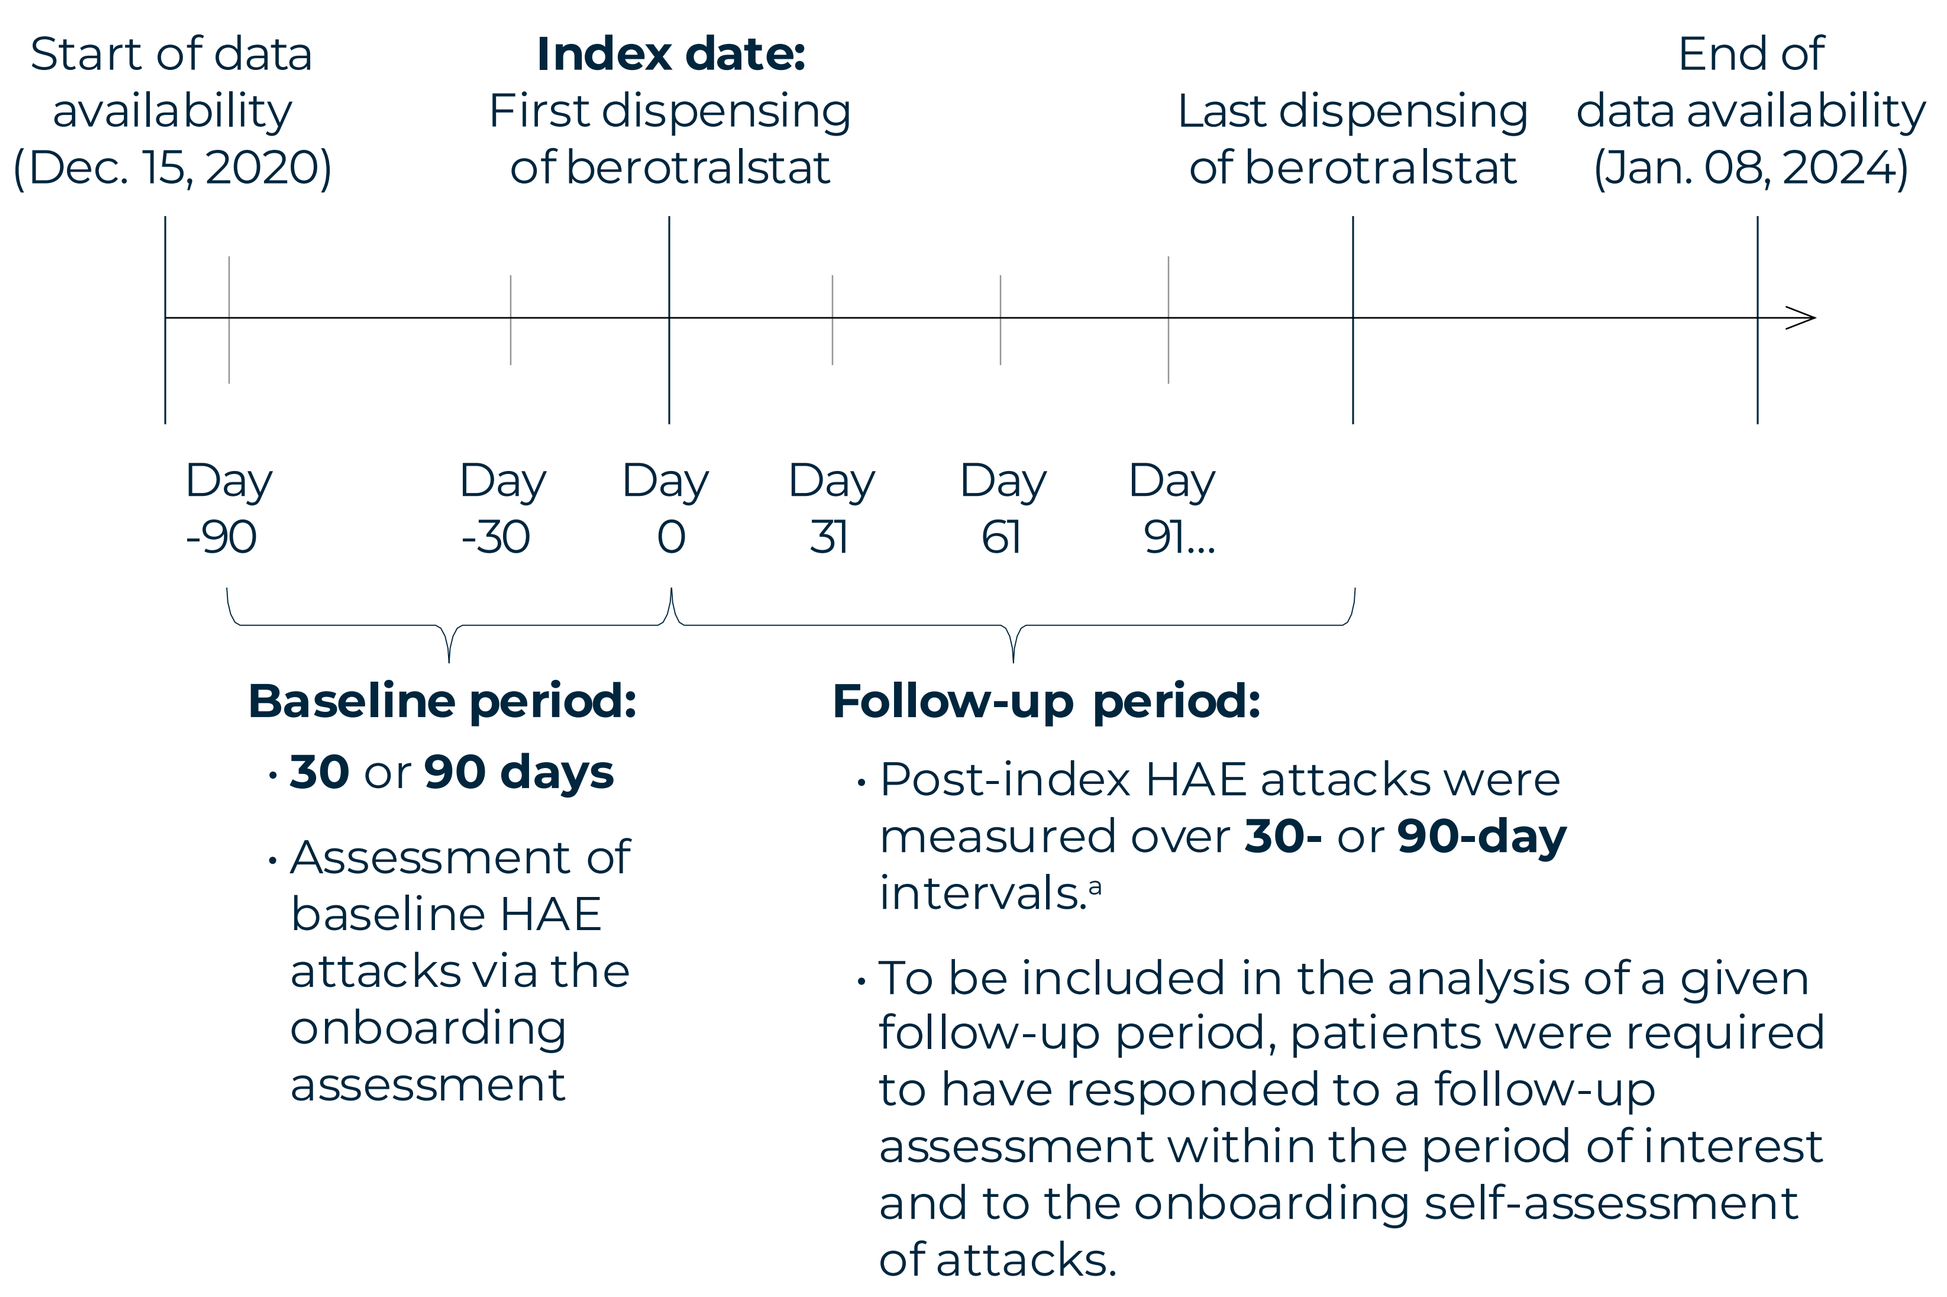
Start of data availability (Dec. 15, 2020)
Index date: First dispensing of berotralstat
Last dispensing of berotralstat
End of data availability (Jan. 08, 2024)
Day -90
Day -30
Day 0
Day 31
Day 61
Day 91...
Baseline period:
• 30 or 90 days
• Assessment of baseline HAE attacks via the onboarding assessment
Follow-up period:
• Post-index HAE attacks were measured over 30- or 90-day intervals.a
• To be included in the analysis of a given follow-up period, patients were required to have responded to a follow-up assessment within the period of interest and to the onboarding self-assessment of attacks.

HAE, hereditary angioedema. [a]To be included in the analysis of the first 30- or 90-day interval, patients were required to have ≥30 or ≥90 days of follow-up, respectively; to be included in the analysis of the second 30-day or 90-day interval, patients were required to have ≥60 or ≥180 days of follow-up; and so on.

Study Outcomes

- Patient-reported HAE attacks were collected at berotralstat initiation and at each refill.
- Mean and median monthly HAE attack rates were reported at 30- and 90-day baseline periods and in 30- and 90-day follow-up intervals.
 - The maximum rate of HAE attacks that patients could experience was assumed to be 1 attack per 2 days.
 - Baseline HAE attack rates were calculated based on the 30-day attack rate and 90-day attack rate (divided by 3 to yield a 30-day attack rate), respectively, from the onboarding assessment.
 - In follow-up, the number of reported HAE attacks was the numerator, and the denominator was the minimum of (a) the time from the previous berotralstat shipment date, and (b) 30 days.

Statistical Analysis

- Mean monthly rates of HAE attacks at baseline (30 or 90 days) and in the follow-up period (segmented into fixed 30- and 90-day intervals) were compared using mean differences, 95% confidence intervals (CIs), and *p*-values from generalized estimating equations (GEE) linear regression models with robust standard errors.

RESULTS

- The study population consisted of 353 patients with ≥2 berotralstat dispensings, HAE-nl-C1-INH type classification based on laboratory measurements, and ≥1 self-assessment of HAE attacks in baseline and follow-up.
- Mean age was 48 years, most patients were female (78.5%), most patients visited an allergist/immunologist (88.4%), and nearly half of patients resided in the South; the mean follow-up period was 398 days (**Table 1**).
- Patients had significantly lower HAE attack rates while on berotralstat during each 30-day follow-up interval (1.68–2.53 attacks/month) versus baseline (5.23–5.63 attacks/month) and during each 90-day follow-up interval (1.63–2.04 attacks/month) versus baseline (4.30–4.66 attacks/month) (**Figure 2**).
- Mean monthly attack rate reductions (95% CI) using 30-day follow-up intervals and a 30-day baseline recall period were -3.75 (-4.43, -3.06) at 12 months (i.e., 331– 360-day interval) and -3.26 (-4.21, -2.32) at 18 months (i.e., 511–540-day interval; both $p<0.001$) (**Figure 3**).
- Using 90-day follow-up intervals and a 90-day baseline recall period, mean monthly attack rates reduced by -2.91 (-3.47, -2.35) at 12 months (i.e., 271–360-day interval) and -2.53 (-3.18, -1.87) at 18 months (i.e., 451–540-day interval; both $p<0.001$) (**Figure 3**).

Table 1. Demographics and Clinical Characteristics

Characteristics	Patients (N=353)
Follow-up period, mean ± SD [median], days	398 ± 301 [320]
Demographics	
Age, mean ± SD [median], years	48.1 ± 16.8 [49]
Female, n (%)	277 (78.5)
Patient weight, mean ± SD [median], kg	84 ± 22 [82]
Region, n (%)	
South	168 (47.6)
Midwest	83 (23.5)
West	54 (15.3)
Northeast	42 (11.9)
Unknown	6 (1.7)
Healthcare practitioner specialty, n (%)	
Allergy/Immunology	312 (88.4)
Nurse practitioner	18 (5.1)
Other	23 (6.5)

SD, standard deviation.

Figure 2. Mean and Median Hereditary Angioedema Attack Rates Before and After Berotralstat Initiation

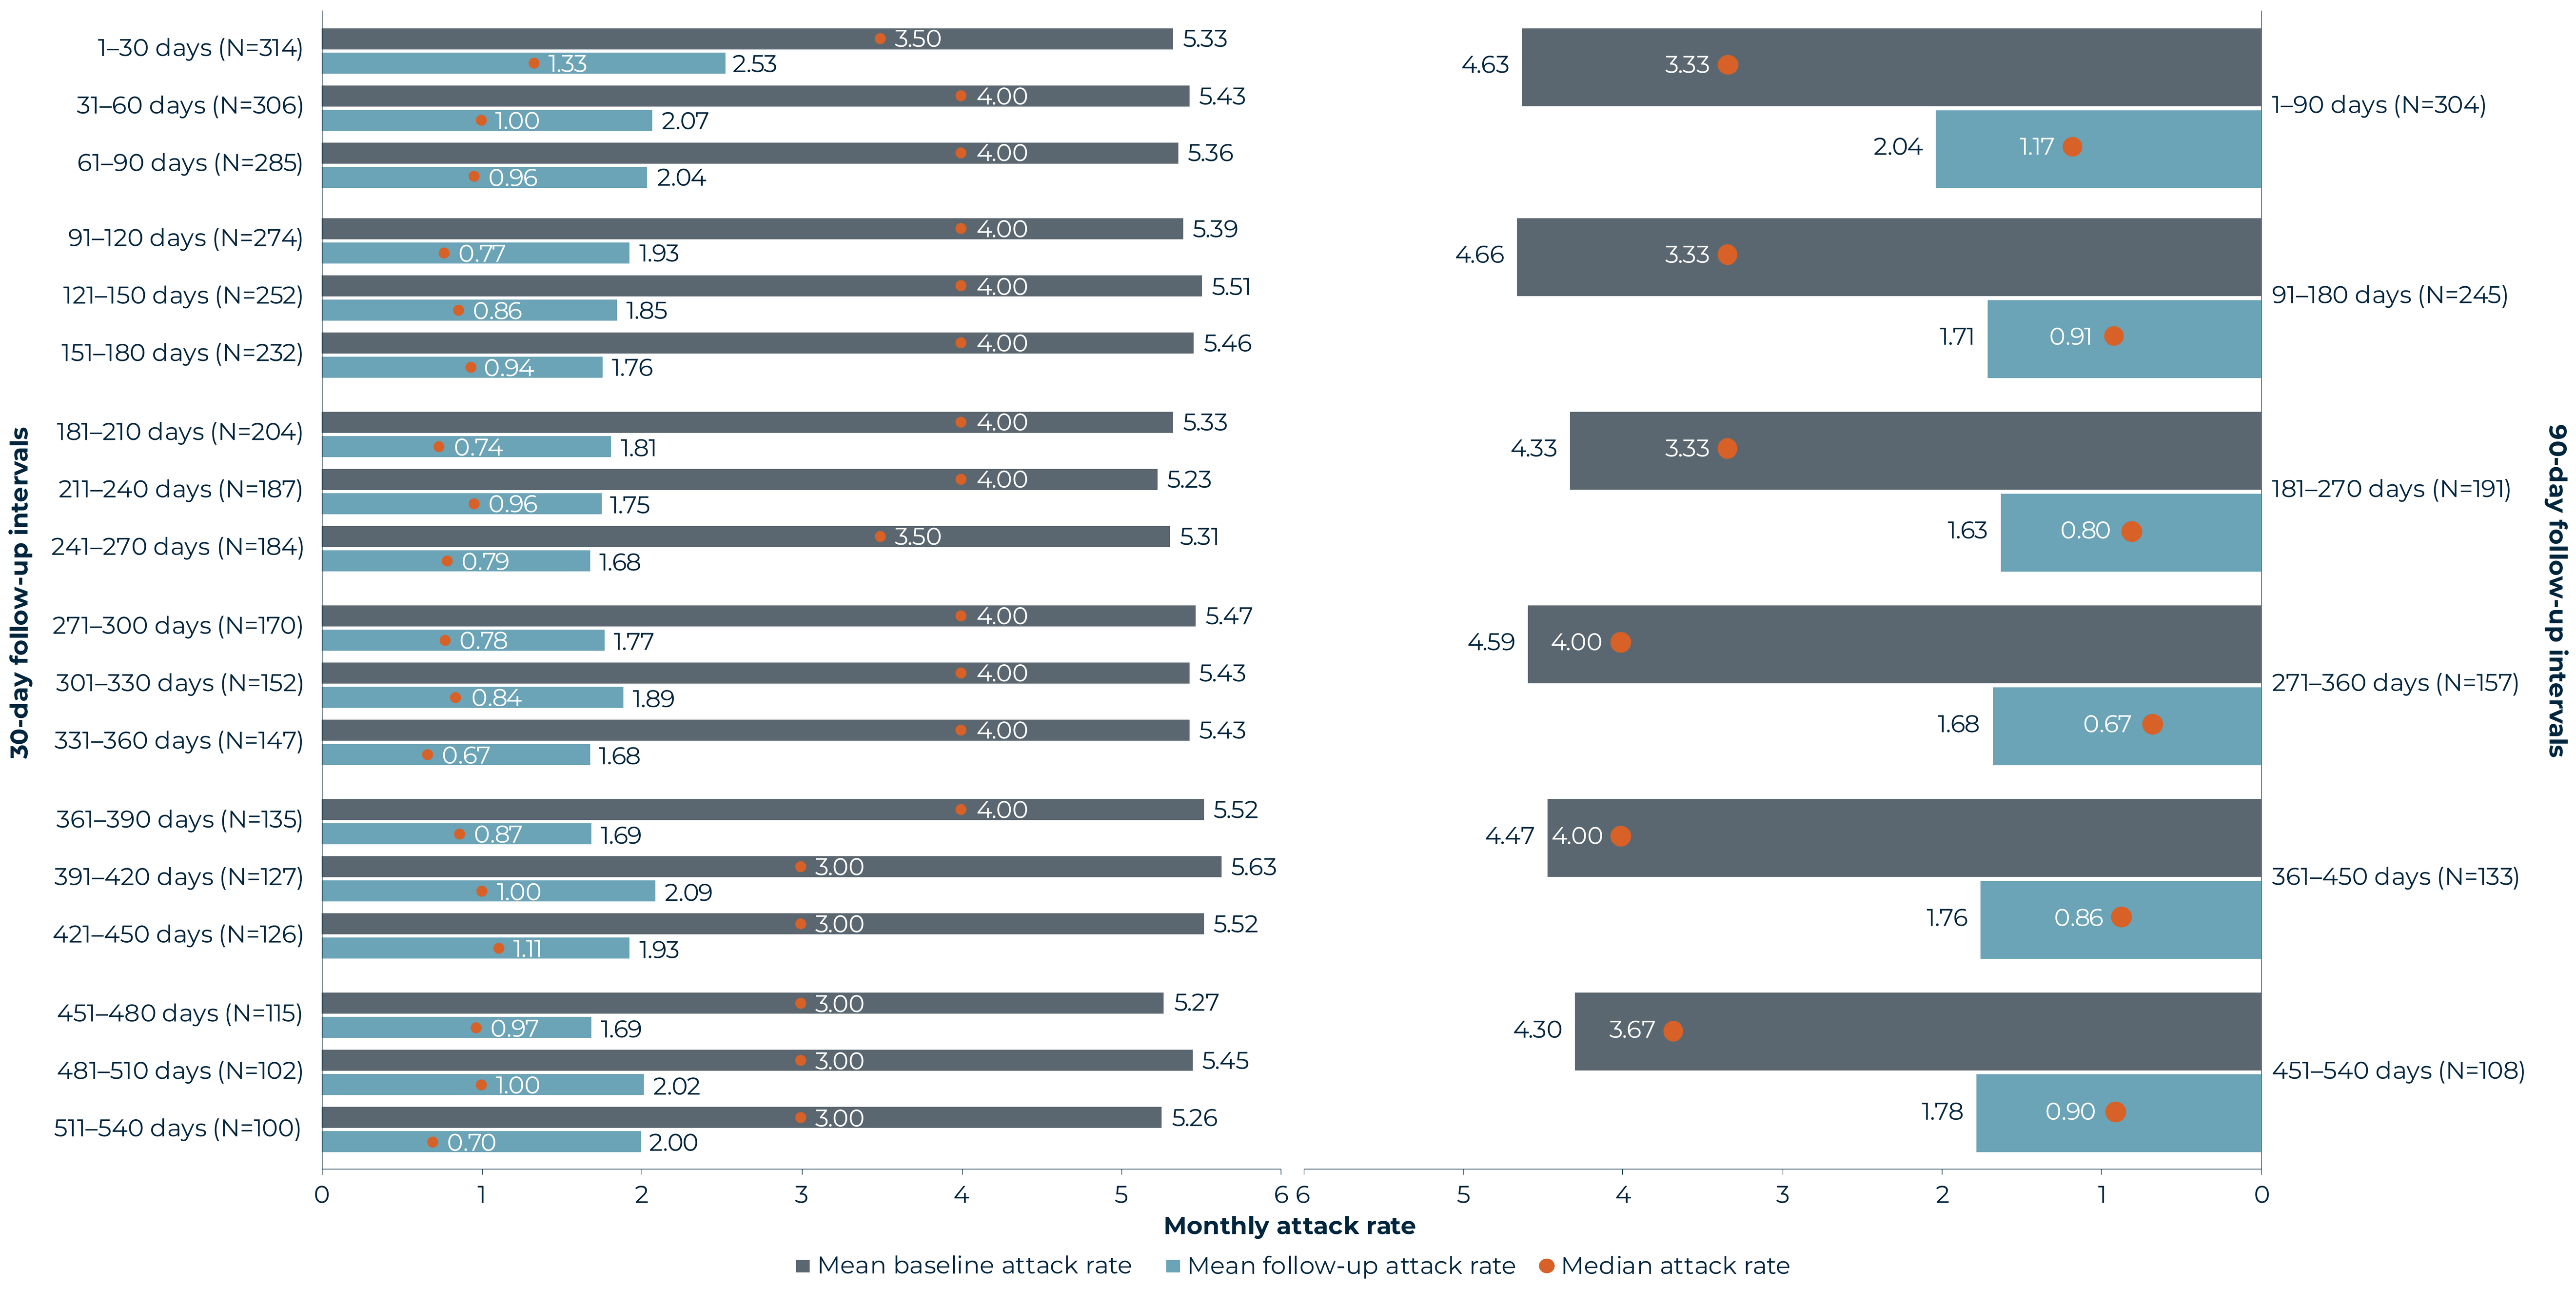
30-day follow-up intervals
1–30 days (N=314) 3.50 5.33 1.33 2.53
31–60 days (N=306) 4.00 5.43 1.00 2.07
61–90 days (N=285) 4.00 5.36 0.96 2.04
91–120 days (N=274) 4.00 5.39 0.77 1.93
121–150 days (N=252) 4.00 5.51 0.86 1.85
151–180 days (N=232) 4.00 5.46 0.94 1.76
181–210 days (N=204) 4.00 5.33 0.74 1.81
211–240 days (N=187) 4.00 5.23 0.96 1.75
241–270 days (N=184) 3.50 5.31 0.79 1.68
271–300 days (N=170) 4.00 5.47 0.78 1.77
301–330 days (N=152) 4.00 5.43 0.84 1.89
331–360 days (N=147) 4.00 5.43 0.67 1.68
361–390 days (N=135) 4.00 5.52 0.87 1.69
391–420 days (N=127) 3.00 5.63 1.00 2.09
421–450 days (N=126) 3.00 5.52 1.11 1.93
451–480 days (N=115) 3.00 5.27 0.97 1.69
481–510 days (N=102) 3.00 5.45 1.00 2.02
511–540 days (N=100) 3.00 5.26 0.70 2.00
90-day follow-up intervals
1–90 days (N=304) 4.63 3.33 2.04 1.17
91–180 days (N=245) 4.66 3.33 1.71 0.91
181–270 days (N=191) 4.33 3.33 1.63 0.80
271–360 days (N=157) 4.59 4.00 1.68 0.67
361–450 days (N=133) 4.47 4.00 1.76 0.86
451–540 days (N=108) 4.30 3.67 1.78 0.90
0 1 2 3 4 5 6
6 5 4 3 2 1 0
Monthly attack rate
Mean baseline attack rate
Mean follow-up attack rate
Median attack rate

Figure 3. Reductions in Hereditary Angioedema Attack Rates After Berotralstat Initiation

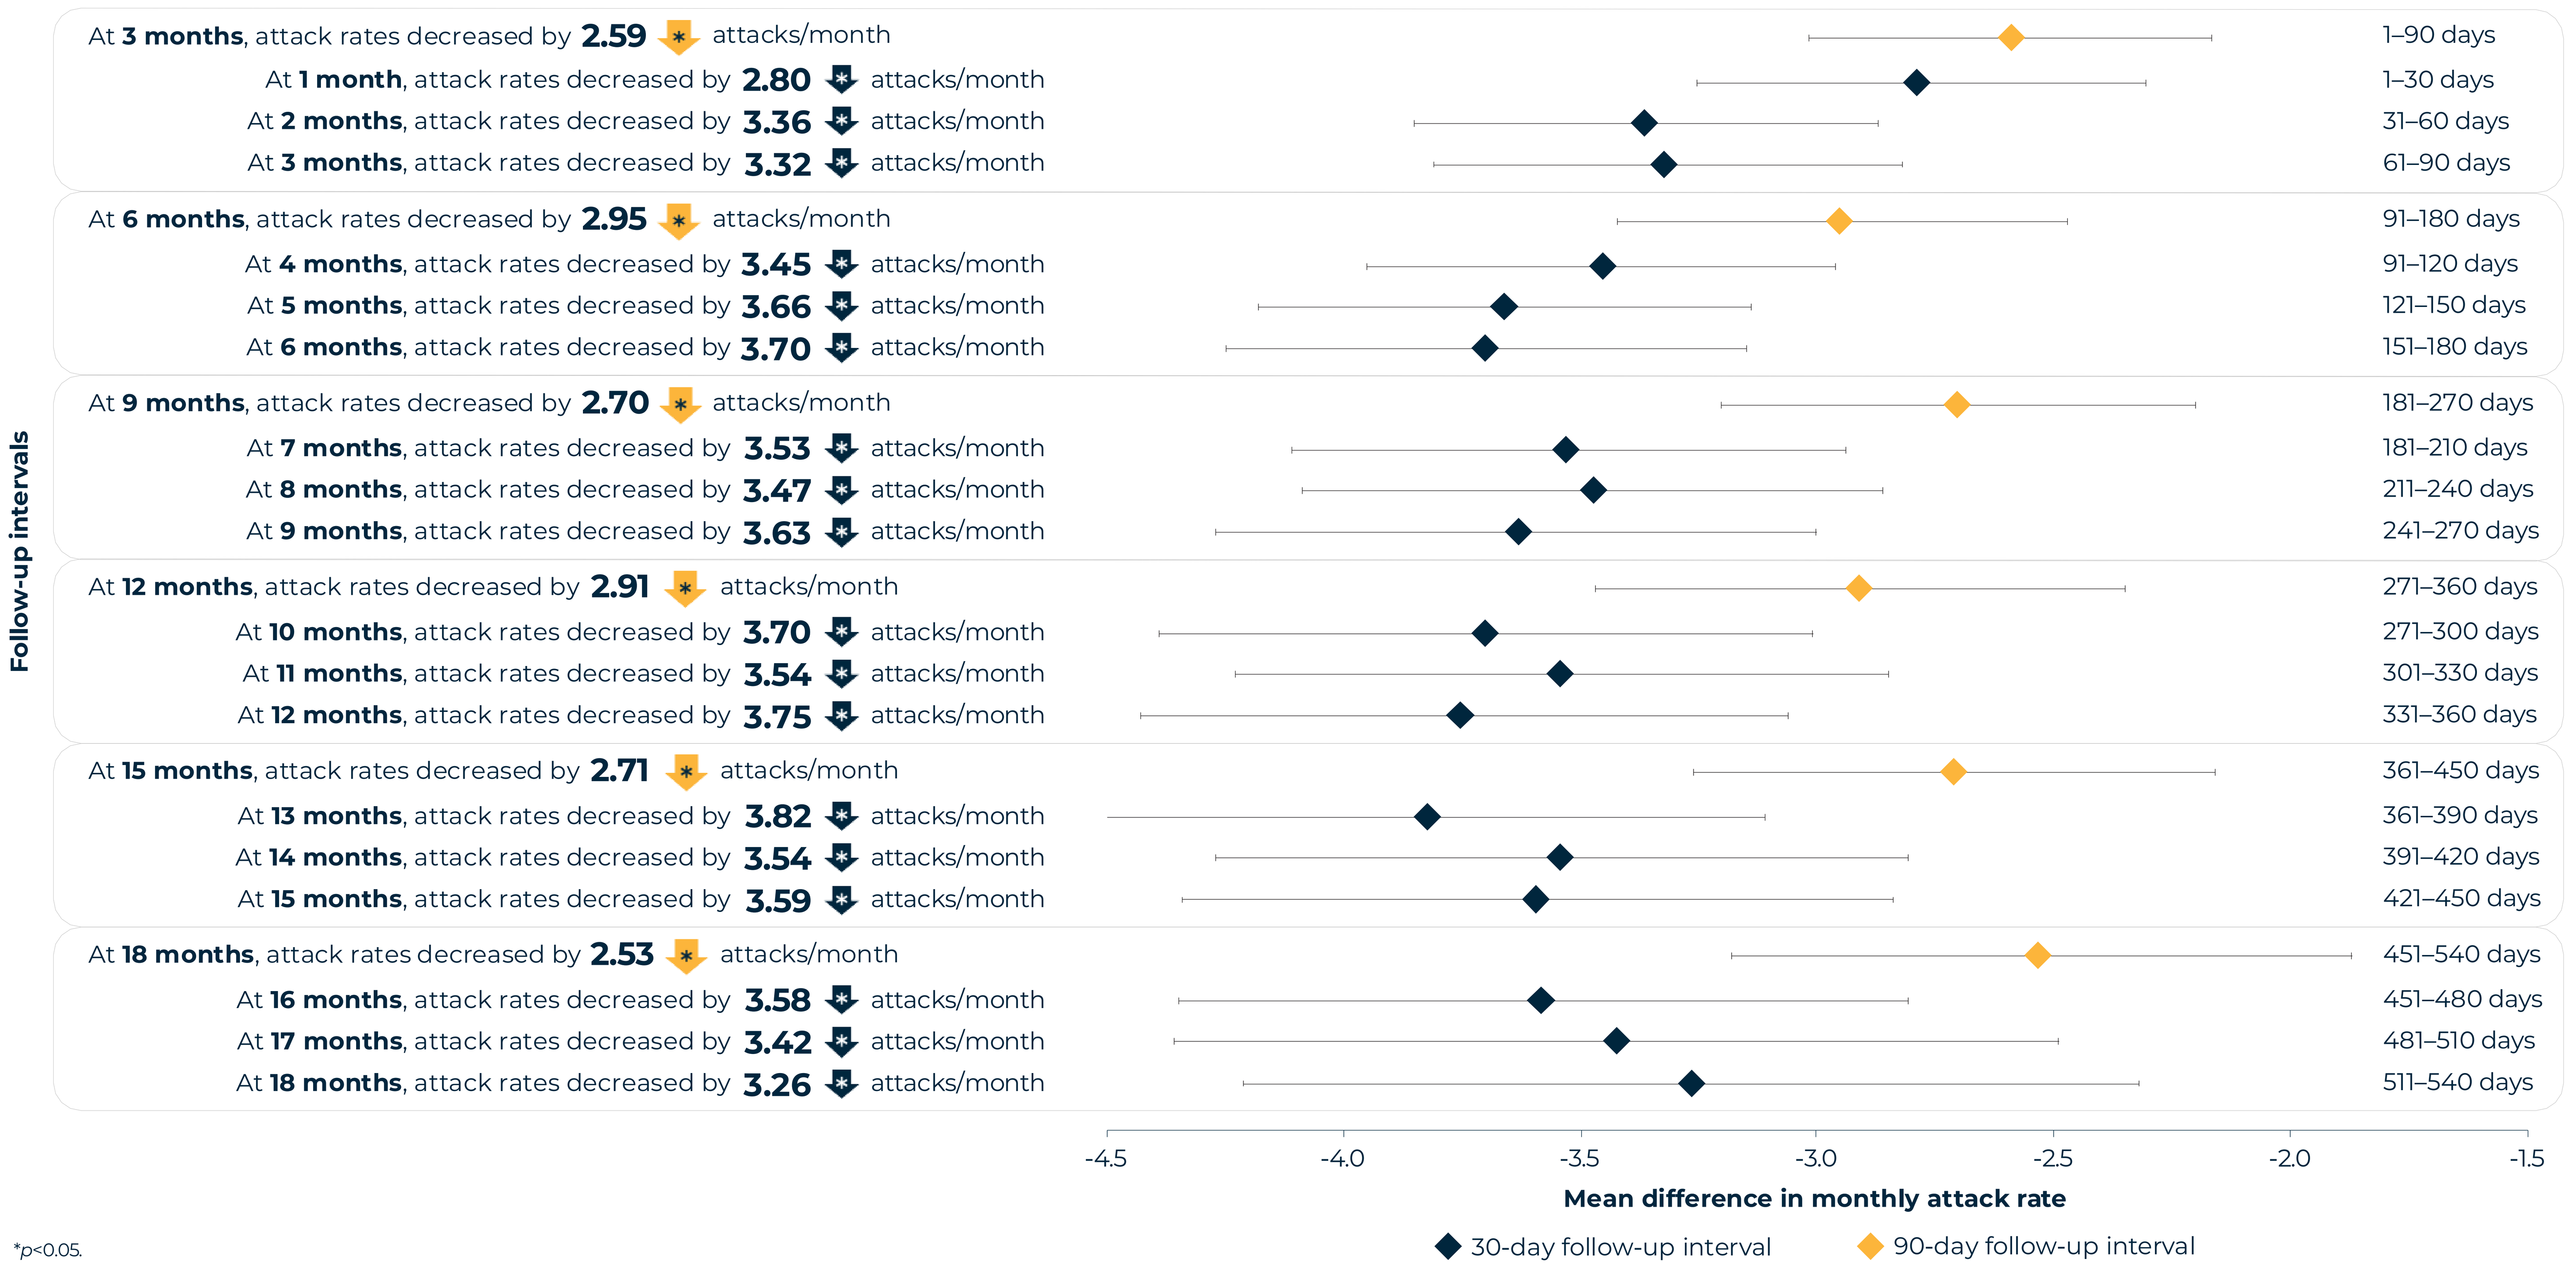
Follow-up intervals
At 3 months, attack rates decreased by 2.59 attacks/month 1–90 days
At 1 month, attack rates decreased by 2.80 attacks/month 1–30 days
At 2 months, attack rates decreased by 3.36 attacks/month 31–60 days
At 3 months, attack rates decreased by 3.32 attacks/month 61–90 days
At 6 months, attack rates decreased by 2.95 attacks/month 91–180 days
At 4 months, attack rates decreased by 3.45 attacks/month 91–120 days
At 5 months, attack rates decreased by 3.66 attacks/month 121–150 days
At 6 months, attack rates decreased by 3.70 attacks/month 151–180 days
At 9 months, attack rates decreased by 2.70 attacks/month 181–270 days
At 7 months, attack rates decreased by 3.53 attacks/month 181–210 days
At 8 months, attack rates decreased by 3.47 attacks/month 211–240 days
At 9 months, attack rates decreased by 3.63 attacks/month 241–270 days
At 12 months, attack rates decreased by 2.91 attacks/month 271–360 days
At 10 months, attack rates decreased by 3.70 attacks/month 271–300 days
At 11 months, attack rates decreased by 3.54 attacks/month 301–330 days
At 12 months, attack rates decreased by 3.75 attacks/month 331–360 days
At 15 months, attack rates decreased by 2.71 attacks/month 361–450 days
At 13 months, attack rates decreased by 3.82 attacks/month 361–390 days
At 14 months, attack rates decreased by 3.54 attacks/month 391–420 days
At 15 months, attack rates decreased by 3.59 attacks/month 421–450 days
At 18 months, attack rates decreased by 2.53 attacks/month 451–540 days
At 16 months, attack rates decreased by 3.58 attacks/month 451–480 days
At 17 months, attack rates decreased by 3.42 attacks/month 481–510 days
At 18 months, attack rates decreased by 3.26 attacks/month 511–540 days
-4.5 -4.0 -3.5 -3.0 -2.5 -2.0 -1.5
Mean difference in monthly attack rate
30-day follow-up interval
90-day follow-up interval

*$p<0.05$.

Limitations

- Patients may have interpreted the definition and distinction between HAE attacks differently, as they were self-reported.
- The presence of a berotralstat dispensing in the data does not indicate that the medication was consumed or that it was taken as prescribed.

CONCLUSIONS

1. Berotralstat was associated with statistically significant and sustained reductions in HAE attack rates through 18 months following berotralstat initiation among patients without C1-inhibitor deficiency in the US.
2. This study observed statistically significant, sustained reductions in HAE attack rates after berotralstat initiation, regardless of the baseline timeframe of measurement (30-day or 90-day recall period) or follow-up timeframe of measurement (30-day or 90-day intervals). Higher baseline attack rates and larger attack reductions were observed when using 30-day recall periods, potentially due to better recall accuracy.

REFERENCES

1. Betschel S, et al. *Allergy, Asthma, & Clin Immunol.* 2019;15(1): 1-29. **2.** Busse PJ and Christiansen SC. *N Engl J Med.* 2020;382(12): 1136-1148. **3.** Berotralstat [package insert]. Durham, NC: November 2023.

FUNDING

This study was funded by BioCryst Pharmaceuticals, Inc.

ACKNOWLEDGMENTS

Editorial support was provided by Cristina Martinez, MA, of Groupe d'Analyse, Ltée, Montreal, QC, Canada, and Ramya Ramasubramanian, PhD, of Analysis Group, Inc, Los Angeles, CA, USA, funded by BioCryst Pharmaceuticals, Inc.

Presented at the International Society for Pharmacoeconomics and Outcomes Research (ISPOR) Europe 2024 • November 17–20, 2024 • Barcelona, Spain