Molecular epidemiology of *ROS1* alterations in advanced non-small cell lung cancer: temporal patterns of testing and positivity across Europe between 2018 and 2022

EPH236

Paul Baas,[1] Yong Yuan,[2] Caroline Rault,[3] Yasemin Karanis,[4] Minouk J. Schoemaker,[5] Alan Calleja,[4] Robin E.J. Munro,[4] John R. Penrod,[2*] Adam Lee,[6] Gabrielle Emanuel,[6] Jens Benn Sørensen[7]

[1]Netherlands Cancer Institute, Amsterdam, the Netherlands; [2]Bristol Myers Squibb, Princeton, NJ, USA; [3]Data Gnosis, Rennes, France; [4]Real World Solutions, IQVIA, London, UK; [5]Real World Solutions, IQVIA, Amsterdam, the Netherlands; [6]Bristol Myers Squibb, Uxbridge, UK; [7]Rigshospitalet, Copenhagen University Hospital, Copenhagen, Denmark

*At the time of the analysis.

Background

- Up to two-thirds of patients with advanced or metastatic non-small cell lung cancer (NSCLC) carry targetable oncology drivers[1]
- International guidelines (eg, European Society for Medical Oncology [ESMO]) recommend testing for these mutations/alterations[2-4]
- Rates of *ROS1* positivity among tested patients range from ~0.5%-3%[1,5-7]; however, real-world data indicate testing rates vary (~20%-65%) depending on the tumor histology, country, and availability of *ROS1*-targeted therapy[5,8,9]
- ESMO guidelines recommend testing for *ROS1* alterations in patients with advanced or metastatic non-squamous NSCLC at diagnosis (not recommended for confirmed squamous cell histology) and first-line therapy for patients with *ROS1*-positive (*ROS1*+) advanced NSCLC with crizotinib, entrectinib, or repotrectinib (repotrectinib is not currently approved by the European Medicines Agency [EMA])[4]
- Timing and type of decision regarding reimbursement of *ROS1*-targeted therapy varies considerably across European countries (**Table 1**)
 - The introduction of national recommendations for *ROS1* testing also varies by country, eg, France, 2012; Germany, 2015; Italy, not available at time of analysis (July 2023); Spain, 2020; England, 2018; Sweden, 2014[9,10]
- As such, there is a need to describe the variability observed between international guidelines and regional variations in the reimbursement of *ROS1*-targeted therapy and the associated *ROS1* testing and positivity rates
- Here, we describe temporal patterns of *ROS1* testing and associated *ROS1* positivity rates in patients with advanced NSCLC across several European countries between 2018-2022 as part of the I-O Optimise international collaborative research initiative[11]

Table 1. Reimbursement decision dates and outcomes for *ROS1* inhibitors crizotinib and entrectinib

Product	HAS[a]	G-BA[b]	AIFA	AEMPS	NICE[c]	TLV
Crizotinib	May 2020 (reimbursed in second line)	Mar 2017 No added benefit			Jul 2018 (CDF and MEA)	
Entrectinib	Jul 2021 SMR: insufficient (not reimbursed)	Feb 2021 No added benefit		May 2022 (non-funding as monotherapy treatment)	Aug 2020	May 2021

Key: No HTA; Positive; Restricted reimbursement; Negative HTA outcome; Negative reimbursement decision

[a]HAS issues a benefit rating or structured medication review based on a product's medical benefit, which determines the reimbursement level. [b]In Germany, products are reimbursed per the EMA label indication, and the benefit rating only impacts pricing. [c]The Scottish Medicines Consortium accepted proposals for patient access schemes for crizotinib (Jun 2018) and entrectinib (Jan 2021). AEMPS, Spanish Agency of Medicines and Medical Devices; AIFA, Italian Medicines Agency; CDF, Cancer Drugs Fund (England); G-BA, Federal Joint Committee (Germany); HAS, Haute Autorité de Santé (France); HTA, health technology assessment; MEA, managed entry agreement; NICE, National Institute for Health and Care Excellence (England/Wales); SMR, structured medication review; TLV, Swedish Dental and Pharmaceutical Benefits Agency.

Methods

Projected rates of *ROS1* testing and positivity

- National rates of *ROS1* alteration testing and *ROS1* positivity were estimated between January 2018 and December 2022 in France, Germany, Italy, Spain, and the United Kingdom (UK; England, Wales, Scotland, and Northern Ireland) using the Oncology Dynamics database, a syndicated survey collecting comprehensive oncology patient data[12] (all patients with stage IIIB-IV NSCLC from a sample population in each country)
- Countries were selected based on available representative data and having a leading role in reimbursement decisions, adoption of innovative medicines, and implementation of novel biomarker testing
- Data from physician questionnaires collected in the Oncology Dynamics database were used to estimate *ROS1* testing and positivity rates on a quarterly cross-sectional basis; projection methodology was used to estimate patient numbers at a national level. Reported cases underwent quality checks and were used to estimate the prevalence of drug-treated patients based on physician workload data
- Projected *ROS1* testing and positivity data from the Oncology Dynamics database were analysed by year

Actual rates of *ROS1* testing and positivity

- Data derived from country-specific data sources were analysed for the entire study period from France (Épidémio-Stratégie Médico-Economique [ESME], 2015-2018), Italy (Istituto Romagnolo per lo Studio dei Tumori "Dino Amadori" [IRST] Meldola, 2018-2020), Spain (Grupo Espanol de Cancer de Pulmon [GECP], 2016-2020),[13] and England (Cancer Analysis System, 2016-2019)
- Data from Sweden (Swedish Lung Cancer Registry, January 2018-March 2022)[14] were analysed by year and for the overall study period
- Where available, testing and positivity rates were evaluated by histology (non-squamous vs squamous NSCLC)

Results

Patterns of testing for *ROS1* based on projected data

- Among all patients with advanced/metastatic NSCLC, the average (range) testing rate in 2022 was 69% (57%-75%)
- Testing rates increased between 2018 and 2022 in all countries, with the greatest increase observed in the UK (from 15% to 72%) (**Figure 1**)
- Testing rates were consistently higher over the entire period in Germany and France, compared with Italy, Spain, and the UK (**Figure 1**)
- The highest testing rates in 2022 were observed among patients with non-squamous histology (average [range], 78% [69%-86%]; **Figure 2**)
 - The largest increase in testing rates among patients with non-squamous histology was observed in the UK, where rates increased from 17% in 2018 to 82% in 2022 (**Figure 2**)
 - In 2022, the lowest testing rates among patients with non-squamous histology were seen in Italy (69%) and Spain (75%), in line with the lack of reimbursement of *ROS1* inhibitors (**Table 1** and **Figure 2**)
- Among patients with squamous histology, the average (range) testing rate in 2022 was 32% (16%-58%)
 - Testing rates among patients with squamous histology were higher across the study period in Germany (range, 39%-58%) vs all other countries (**Figure 3**)

Patterns of *ROS1* positivity based on projected data

- Based on projected data, the range of *ROS1* positivity rates over the study period and across countries was 1%-5% (**Table 2**)
- Increased *ROS1* testing resulted in more patients identified with *ROS1*+ NSCLC across the 5 countries, from 1096 in 2018 to 2090 in 2022 (**Table 2**)
- Across all countries, *ROS1* alterations were generally more frequent in patients with non-squamous vs squamous histology (**Table 2**)

Patterns of *ROS1* positivity based on country-specific data sources

- Among patients tested for *ROS1* alterations in populations included in the selected country-specific data sources, the proportions with *ROS1*+ NSCLC were 2% in France, 2% in Italy, 3% in Spain, 1% in England, and 1% in Sweden (**Figure 4a-e**)
- The actual number of patients testing positive for *ROS1* recorded in England increased substantially between 2016 and 2019 (< 5 patients in 2016 to 30 in 2019) (**Figure 4d**)
- In Sweden, 1% of patients had *ROS1*+ NSCLC in the overall and non-squamous populations, and 0% in the squamous population; 105 patients with *ROS1*+ NSCLC were diagnosed between 2018 and March 2022 (**Figure 4e**)

Figure 1. Temporal changes in the percentage of patients with advanced/metastatic NSCLC tested for *ROS1* alterations by country

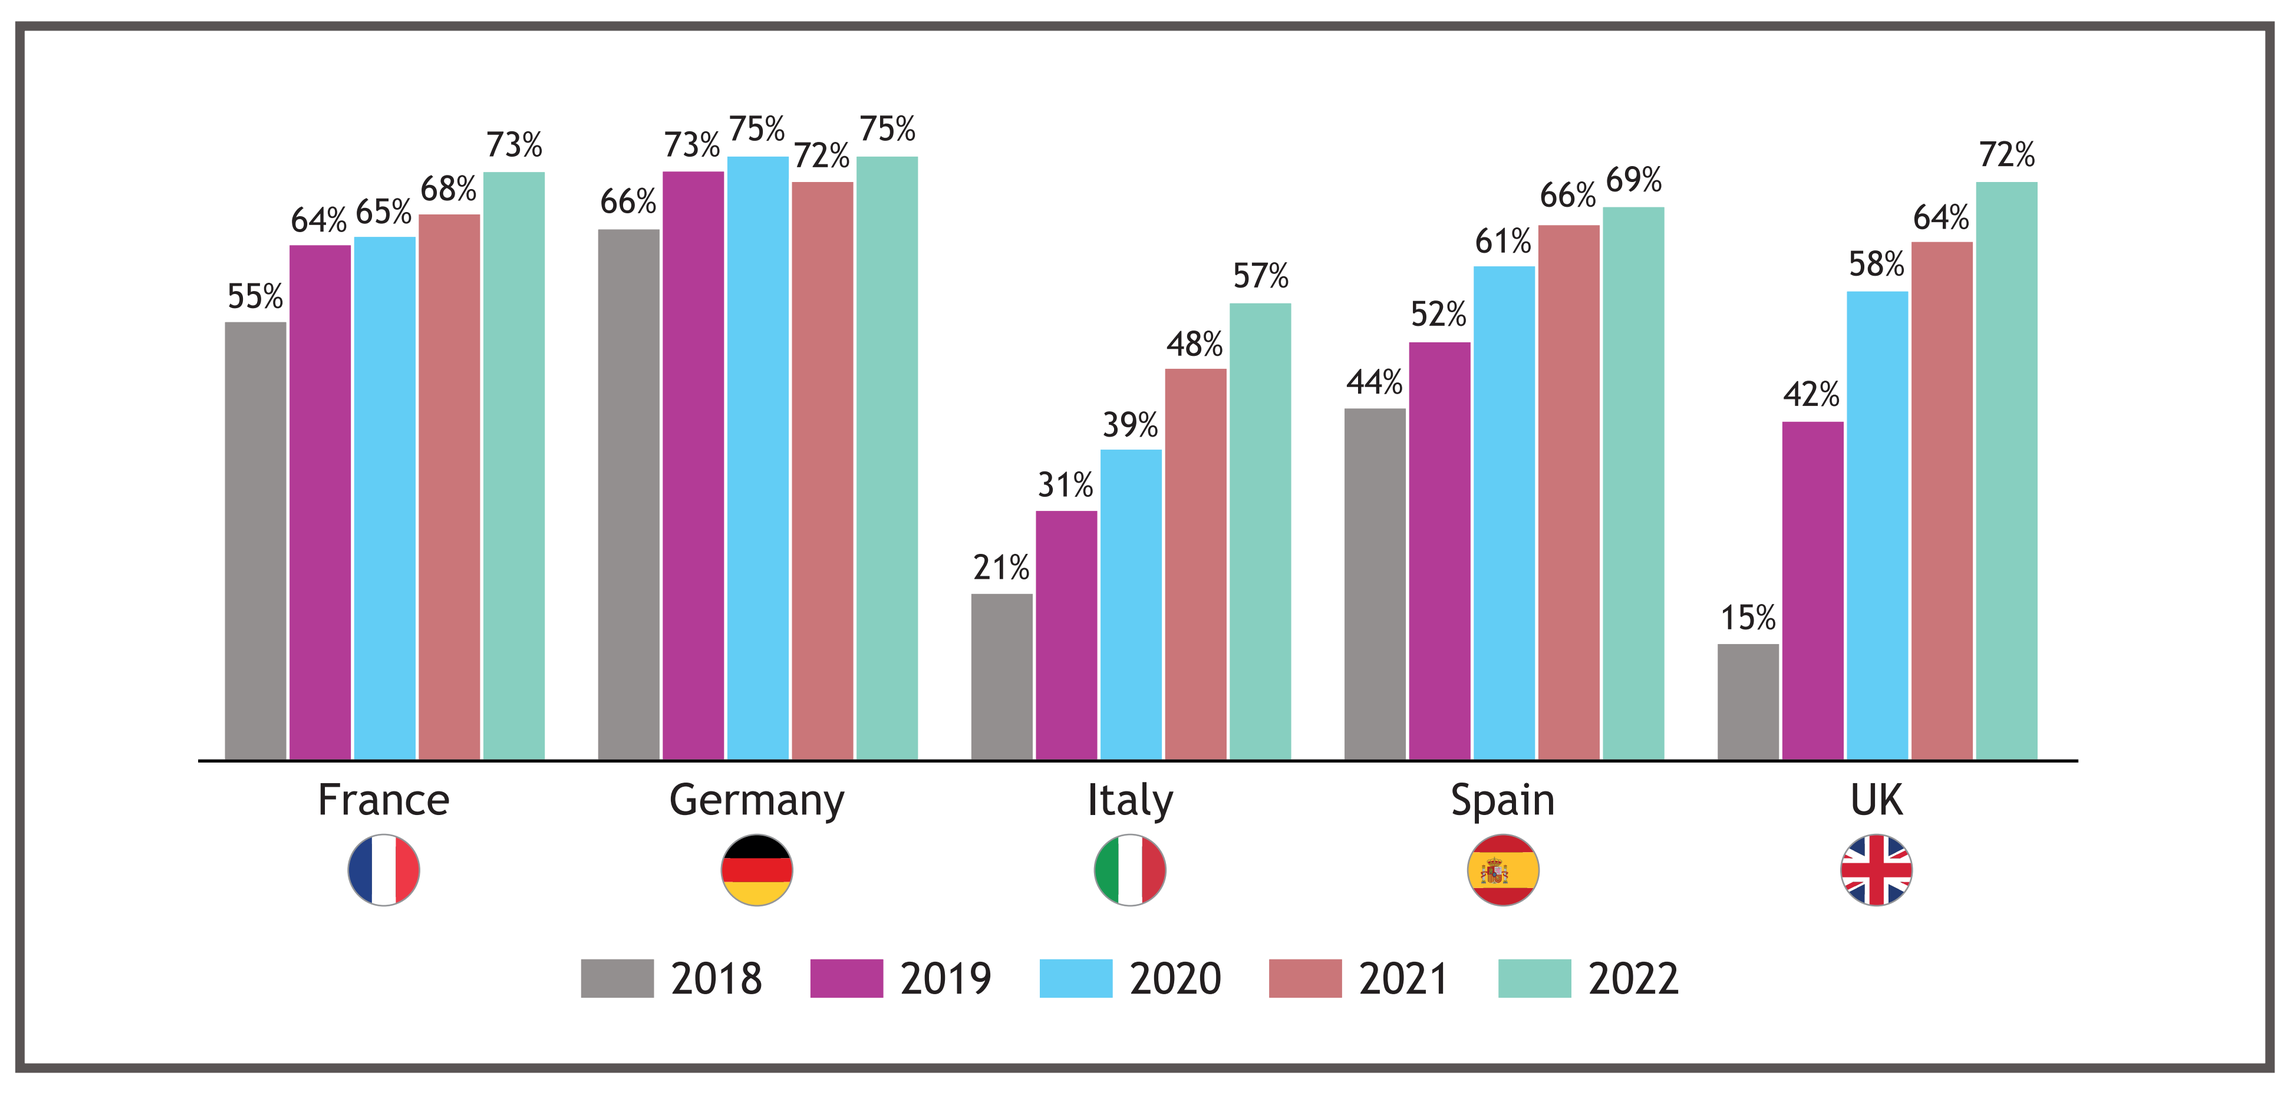

France: 55% 64% 65% 68% 73%
Germany: 66% 73% 75% 72% 75%
Italy: 21% 31% 39% 48% 57%
Spain: 44% 52% 61% 66% 69%
UK: 15% 42% 58% 64% 72%
2018 2019 2020 2021 2022

Figure 2. Temporal changes in the percentage of patients with advanced/metastatic non-squamous NSCLC tested for *ROS1* alterations by country

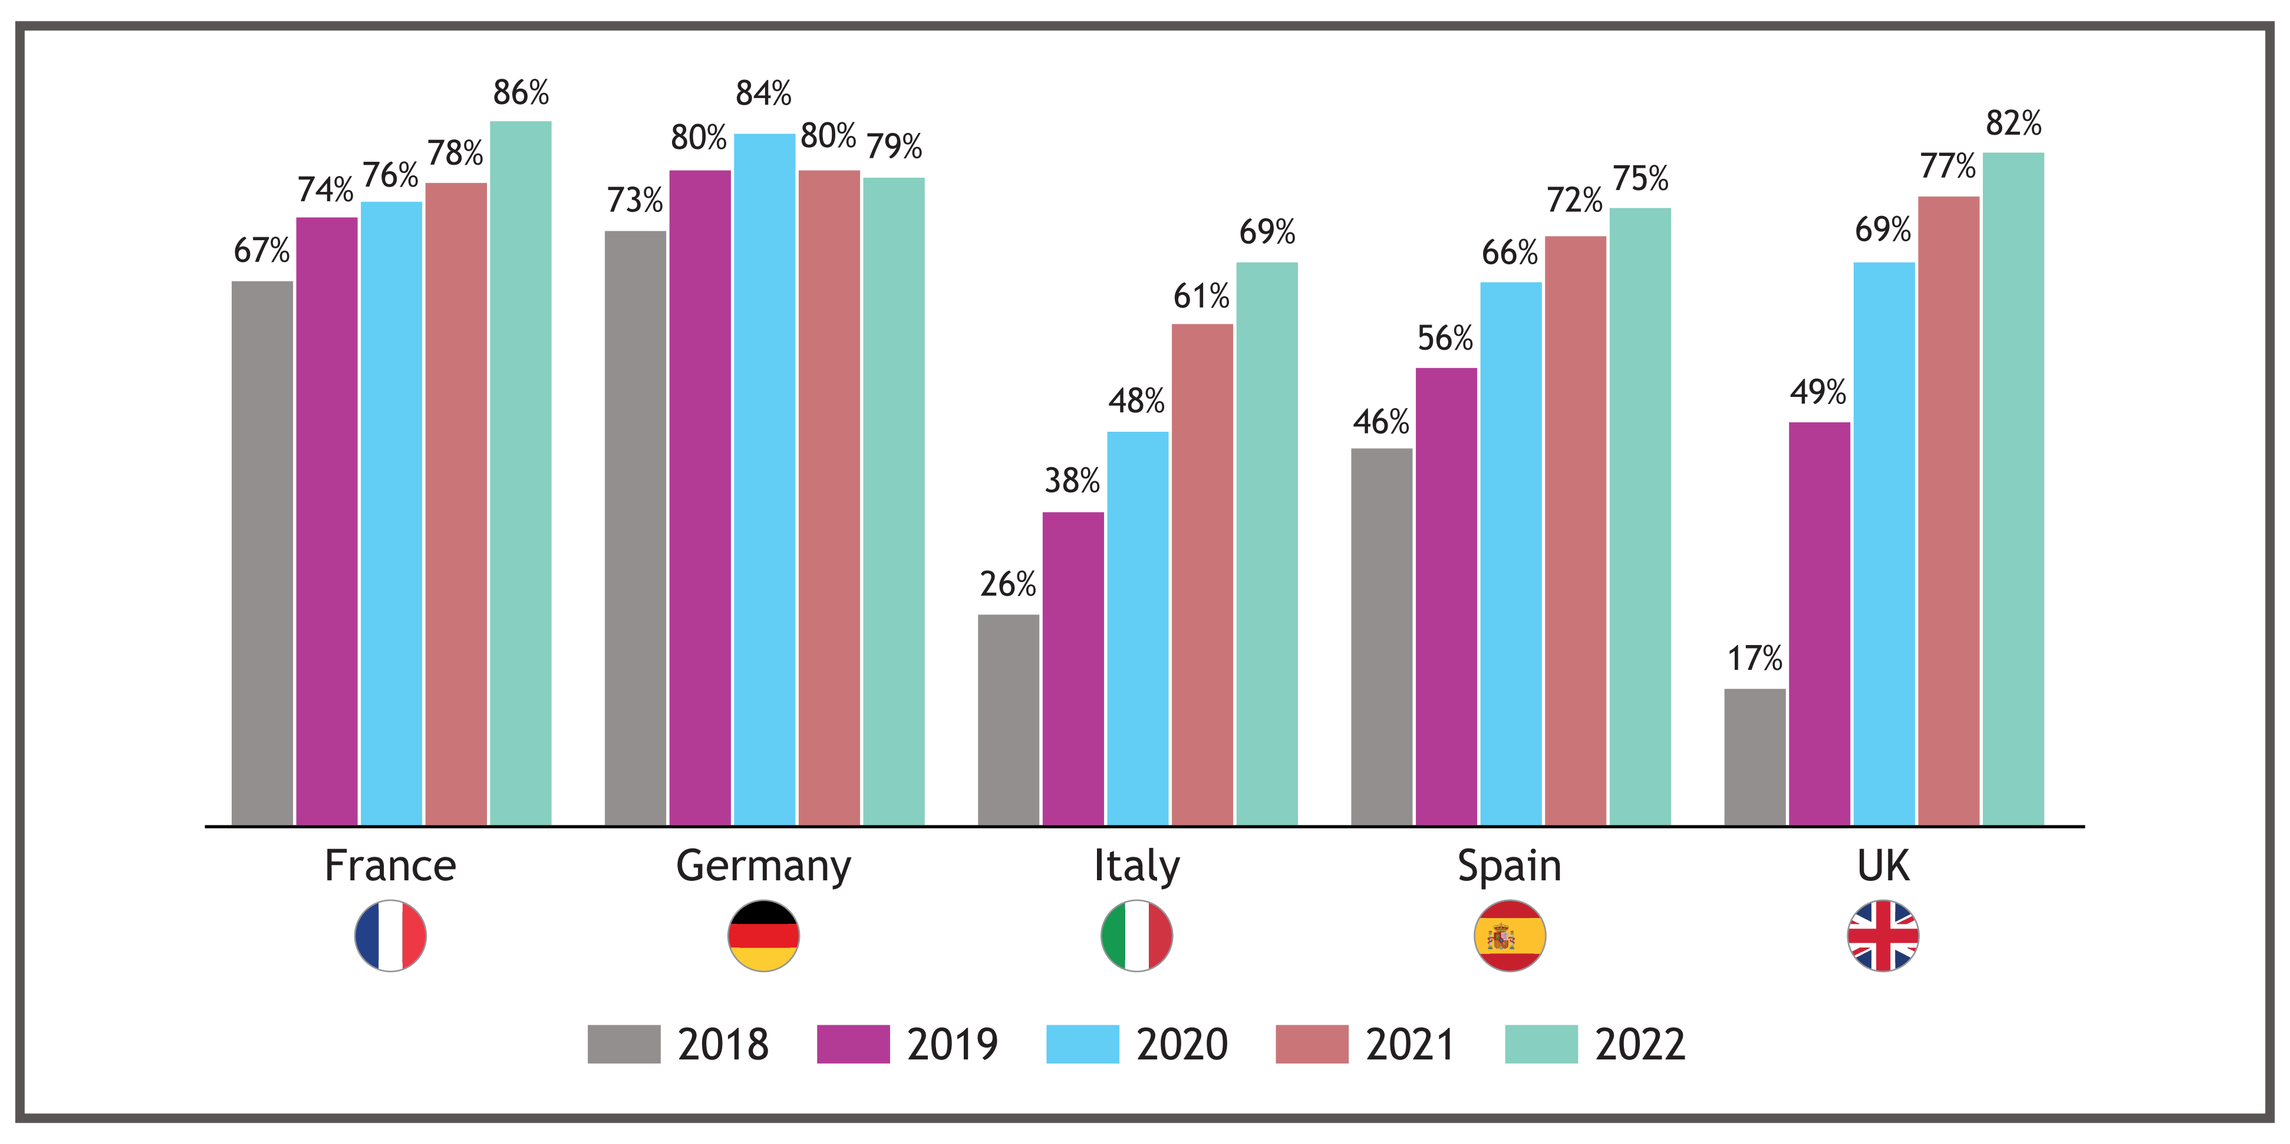

France: 67% 74% 76% 78% 86%
Germany: 73% 80% 84% 80% 79%
Italy: 26% 38% 48% 61% 69%
Spain: 46% 56% 66% 72% 75%
UK: 17% 49% 69% 77% 82%
2018 2019 2020 2021 2022

Figure 3. Temporal changes in the percentage of patients with advanced/metastatic squamous NSCLC tested for *ROS1* alterations by country

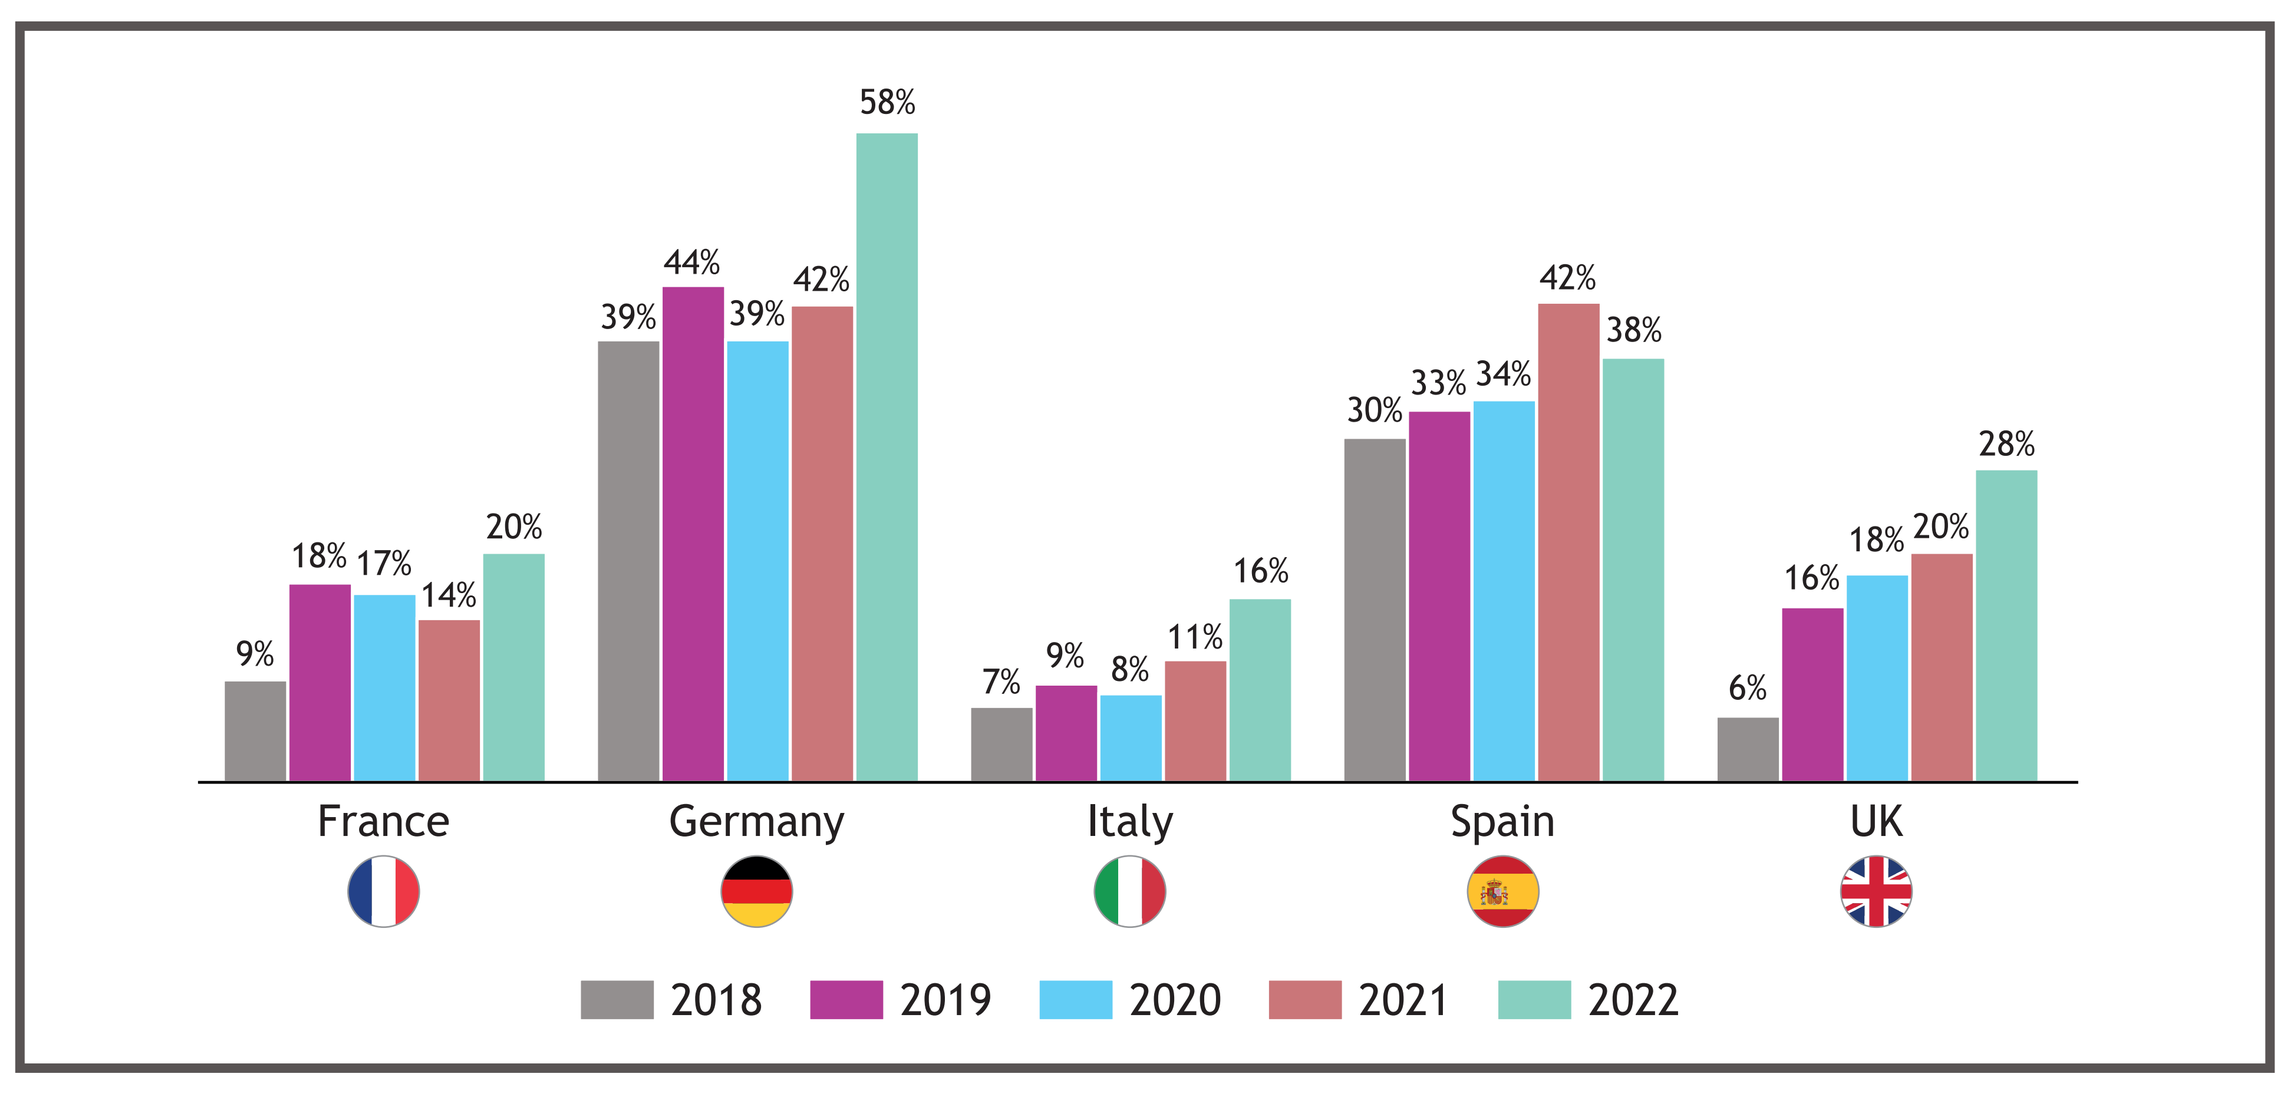

France: 9% 18% 17% 14% 20%
Germany: 39% 44% 39% 42% 58%
Italy: 7% 9% 8% 11% 16%
Spain: 30% 33% 34% 42% 38%
UK: 6% 16% 18% 20% 28%
2018 2019 2020 2021 2022

Table 2. Projected number and percentage of patients with advanced/metastatic NSCLC testing positive for *ROS1* alterations[a,b]

Patients, n (%)		2018	2019	2020	2021	2022
All	France	143 (1)	444 (3)	328 (2)	386 (2)	694 (4)
	Germany	578 (3)	701 (3)	620 (3)	421 (2)	481 (2)
	Italy	93 (2)	137 (2)	258 (3)	377 (4)	268 (3)
	Spain	219 (4)	256 (4)	280 (4)	262 (3)	249 (3)
	UK	63 (2)	398 (5)	398 (3)	596 (4)	398 (2)
Non-squamous histology	France	143 (1)	444 (3)	328 (2)	345 (2)	694 (4)
	Germany	534 (3)	646 (3)	583 (3)	421 (2)	481 (2)
	Italy	93 (3)	137 (3)	221 (3)	361 (4)	268 (3)
	Spain	219 (5)	248 (4)	280 (4)	253 (4)	249 (4)
	UK	63 (2)	398 (5)	398 (3)	596 (4)	398 (3)
Squamous histology[c]	France	0 (0)	0 (0)	0 (0)	41 (7)	0 (0)
	Germany	44 (2)	55 (2)	37 (1)	0 (0)	0 (0)
	Italy	0 (0)	0 (0)	36 (10)	15 (3)	0 (0)
	Spain	0 (0)	8 (1)	0 (0)	9 (1)	0 (0)
	UK	0 (0)	0 (0)	0 (0)	0 (0)	0 (0)

[a]Patient numbers are nationally projected. [b]Data presented as a percentage of all tested patients. [c]Due to low sample numbers in the squamous NSCLC patient population, the projected percentage of patients with *ROS1* positivity varied substantially across the study period; therefore, results should be interpreted with caution.

Strengths and limitations

Strengths

- Oncology Dynamics captures any type of *ROS1* testing technology used within biomarker panels facilitating a comprehensive assessment of testing rates
- Data were taken from high-quality real-world data sources and provide a holistic view of the testing landscape for patients with *ROS1*+ NSCLC in the respective countries
- The use of multiple data sources ensured that sufficient data were available for analysis of patients tested for *ROS1* alterations
- Projected data were supplemented with real-world data from country-specific sources

Limitations

- Absolute numbers of patients in the analysis were small due to the rarity of the *ROS1* alteration
- A high-level projection was carried out from the patient sample size to estimate patient numbers at a national level for each country; however, the projected calculations were based on low sample sizes and therefore should be interpreted with caution

References

1. Fois SS, et al. *Int J Mol Sci* 2021;22:612.
2. Novello S, et al. *Ann Oncol* 2016;27(suppl 5):v1-v27.
3. Planchard D, et al. *Ann Oncol* 2018;29(suppl 4):iv192-iv237.
4. Hendriks LE, et al. *Ann Oncol* 2023;34:339-357.
5. Griesinger F, et al. *Lung Cancer* 2021;152:174-184.
6. Provencio M, et al. *BMC Cancer* 2022;22:732.
7. Terrenato I, et al. *Cancers (Basel)* 2022;14:2971.
8. van de Ven M, et al. *J Mol Diagn* 2021;23:484-494.
9. de Jager VD, et al. *Lancet Reg Health Eur* 2024;38:100838.
10. NICE. Crizotinib for treating ROS1-positive advanced non-small-cell lung cancer. Accessed October 15, 2024. https://www.nice.org.uk/guidance/ta529
11. Ekman S, et al. *Future Oncol* 2019;15:1551-1563.
12. Alymova S, et al. *Int J Clin Pharmacol Ther* 2022;60:207-216.
13. Provencio M, et al. *BMC Cancer* 2022;22:732.
14. Swedish Lung Cancer Registry. Interactive report from the National Lung Cancer Registry. Accessed October 15, 2024. https://statistik.incanet.se/Lunga/

Acknowledgments

- This work was funded by Bristol Myers Squibb
- All authors contributed to and approved the presentation; professional writing and editorial assistance were provided by Becky O'Connor, PhD, of Parexel, funded by Bristol Myers Squibb

Figure 4. Patients with advanced/metastatic NSCLC testing positive for *ROS1* alterations by country

Figure 4a. France

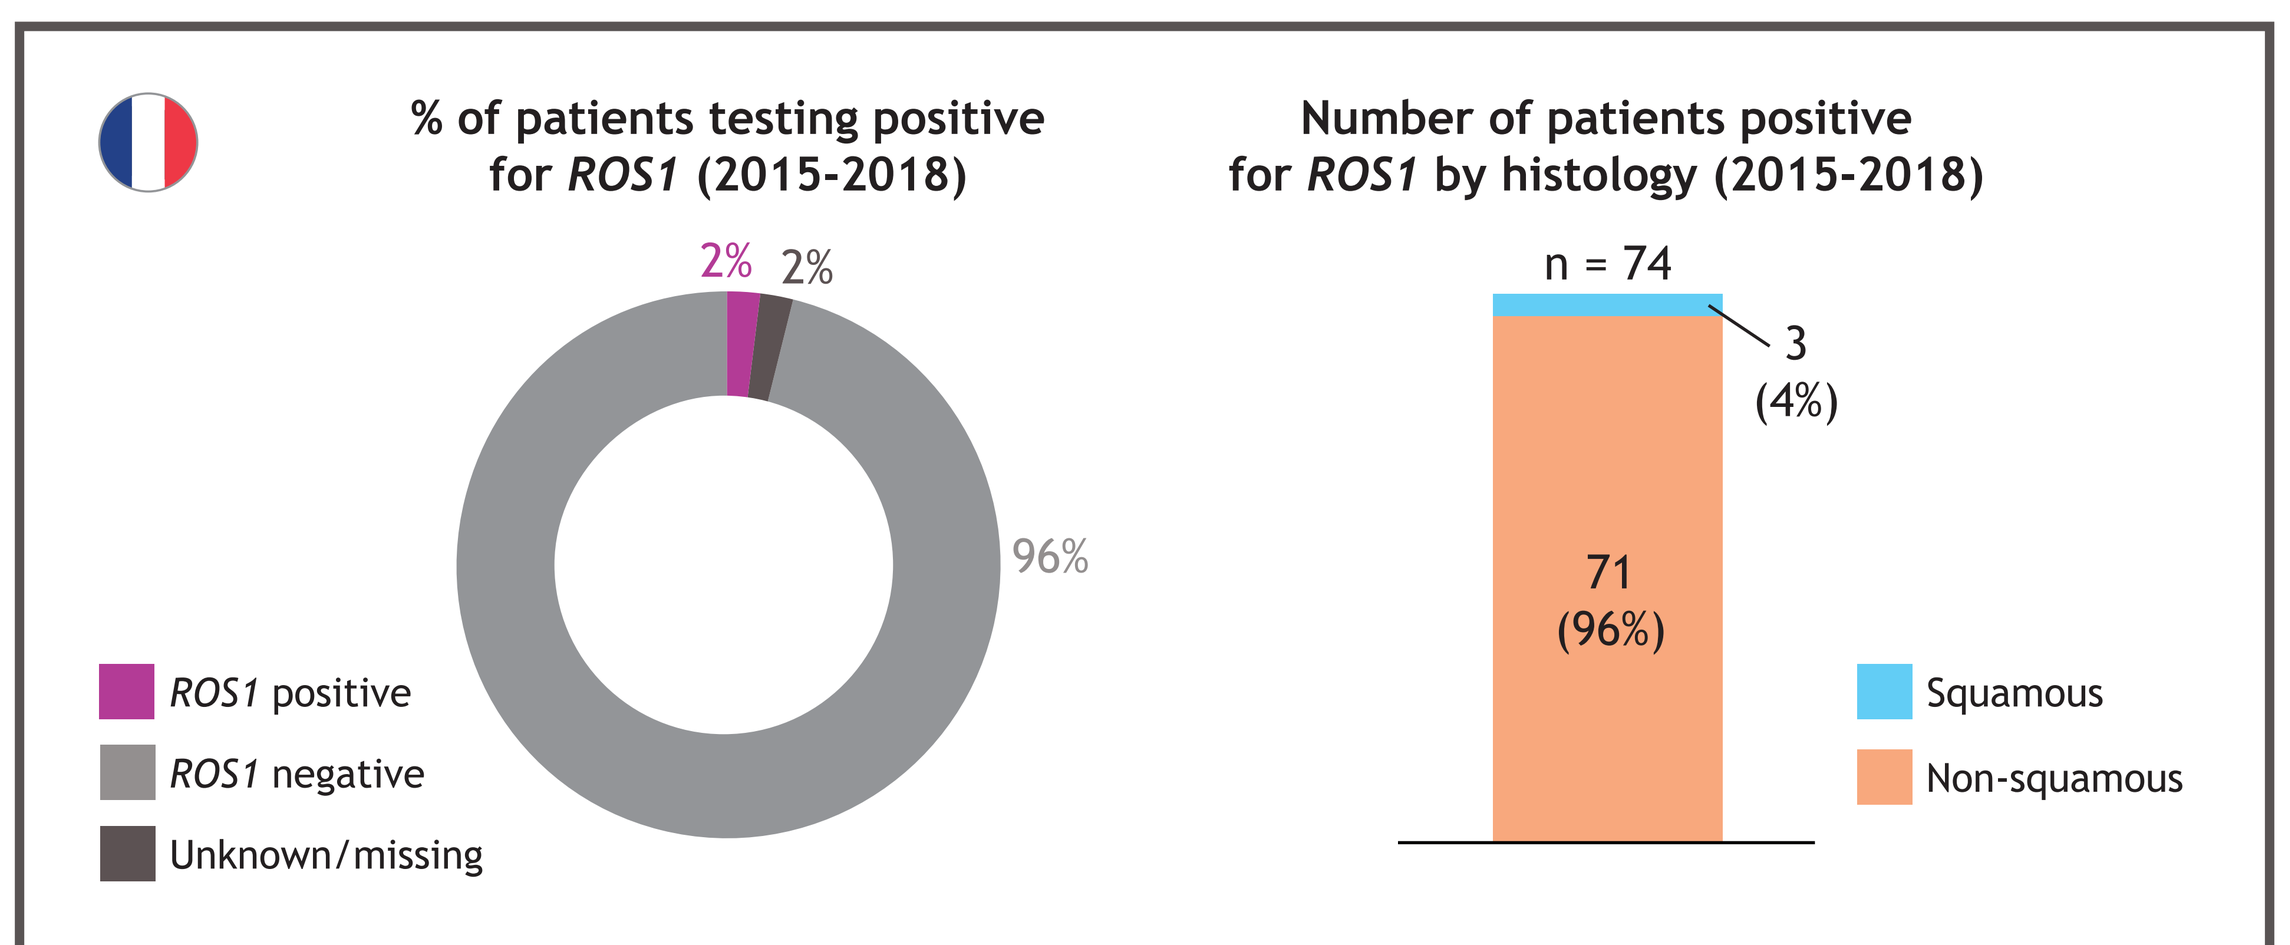

% of patients testing positive for ROS1 (2015-2018)
2% 2% 96%
ROS1 positive
ROS1 negative
Unknown/missing
Number of patients positive for ROS1 by histology (2015-2018)
n = 74
3 (4%)
71 (96%)
Squamous
Non-squamous

Tested population includes patients without a conclusive result. Data include patients with regional disease progression, metastatic disease progression, de novo locally advanced patients, and de novo stage IV NSCLC; data are based on the absence of a positive test result for an *ALK* or *EGFR* mutation which may impact *ROS1* positivity rates.

Figure 4b. Italy

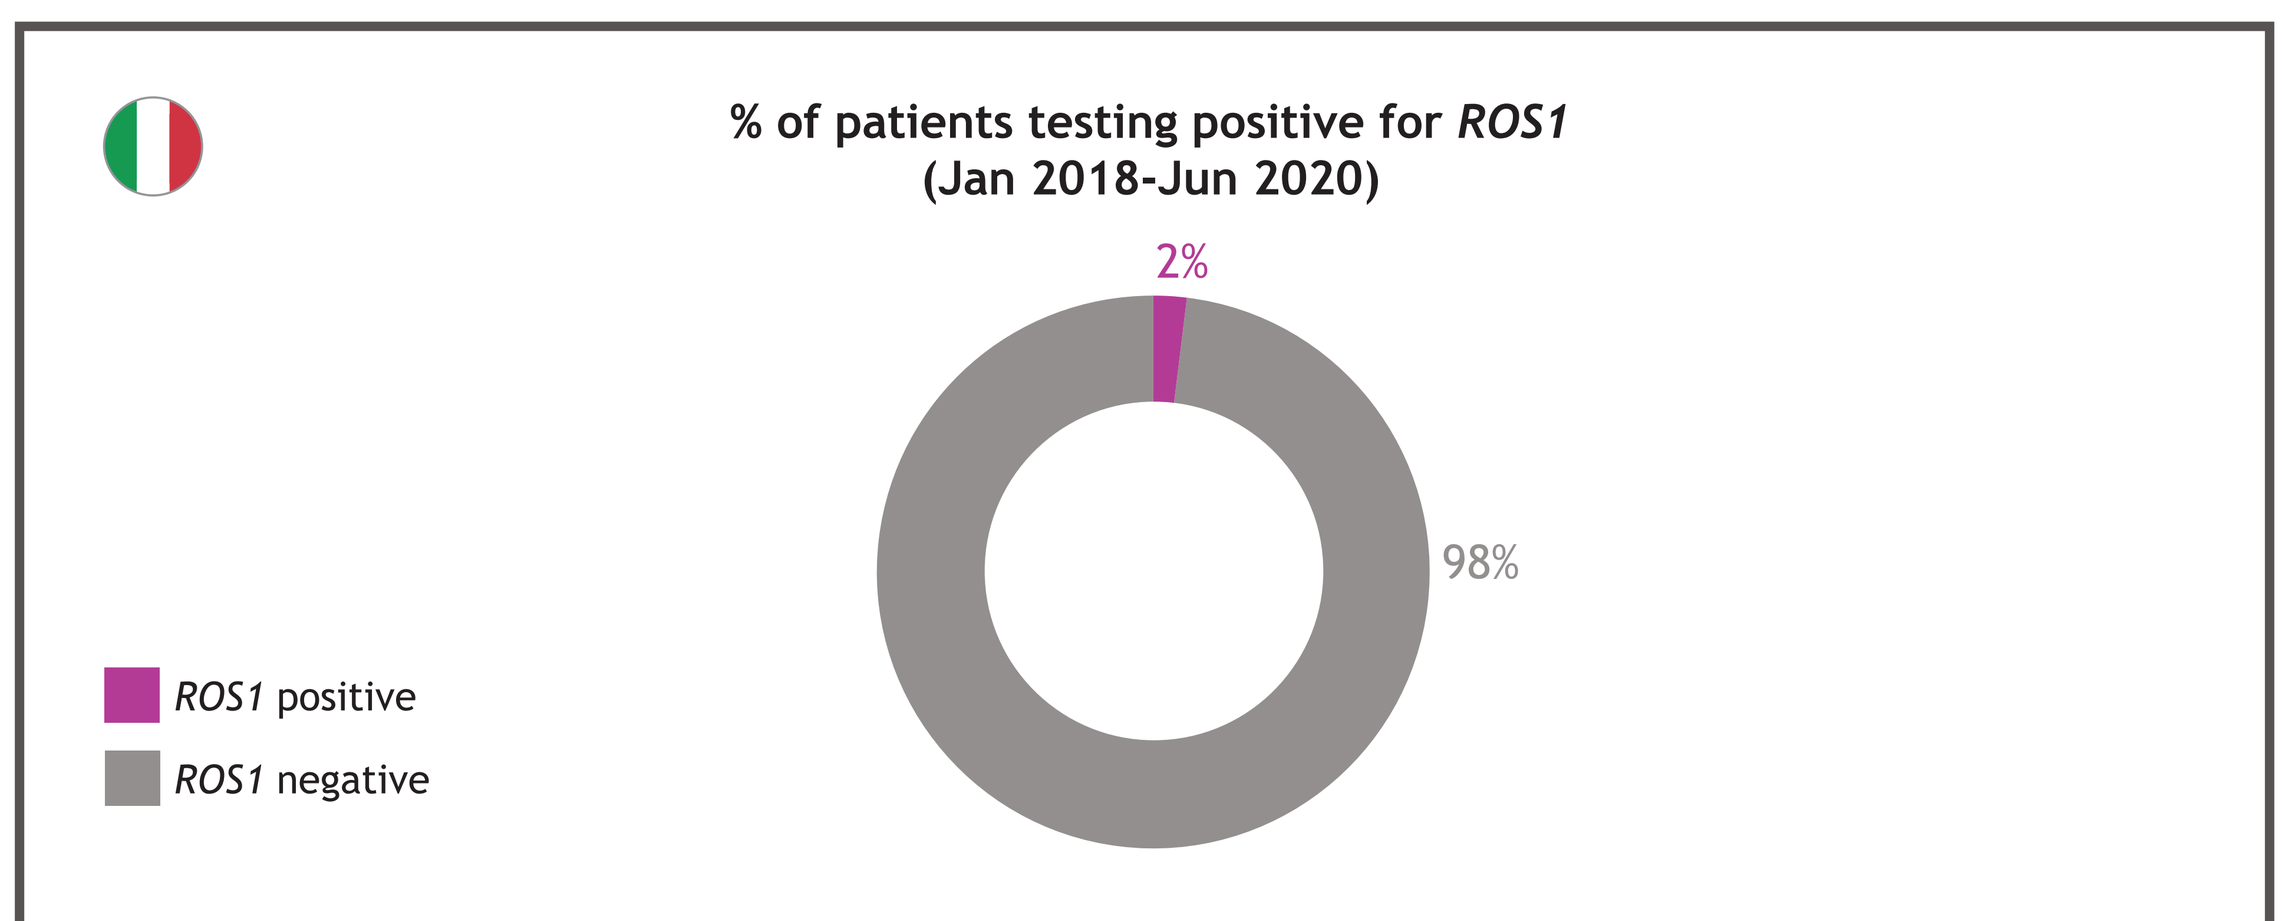

% of patients testing positive for ROS1 (Jan 2018-Jun 2020)
2% 98%
ROS1 positive
ROS1 negative

Tested population includes patients without a conclusive result. Data only include patients with NSCLC who were treated; therefore, patients who did not receive treatment or who chose to be treated at another center cannot be described.

Figure 4c. Spain

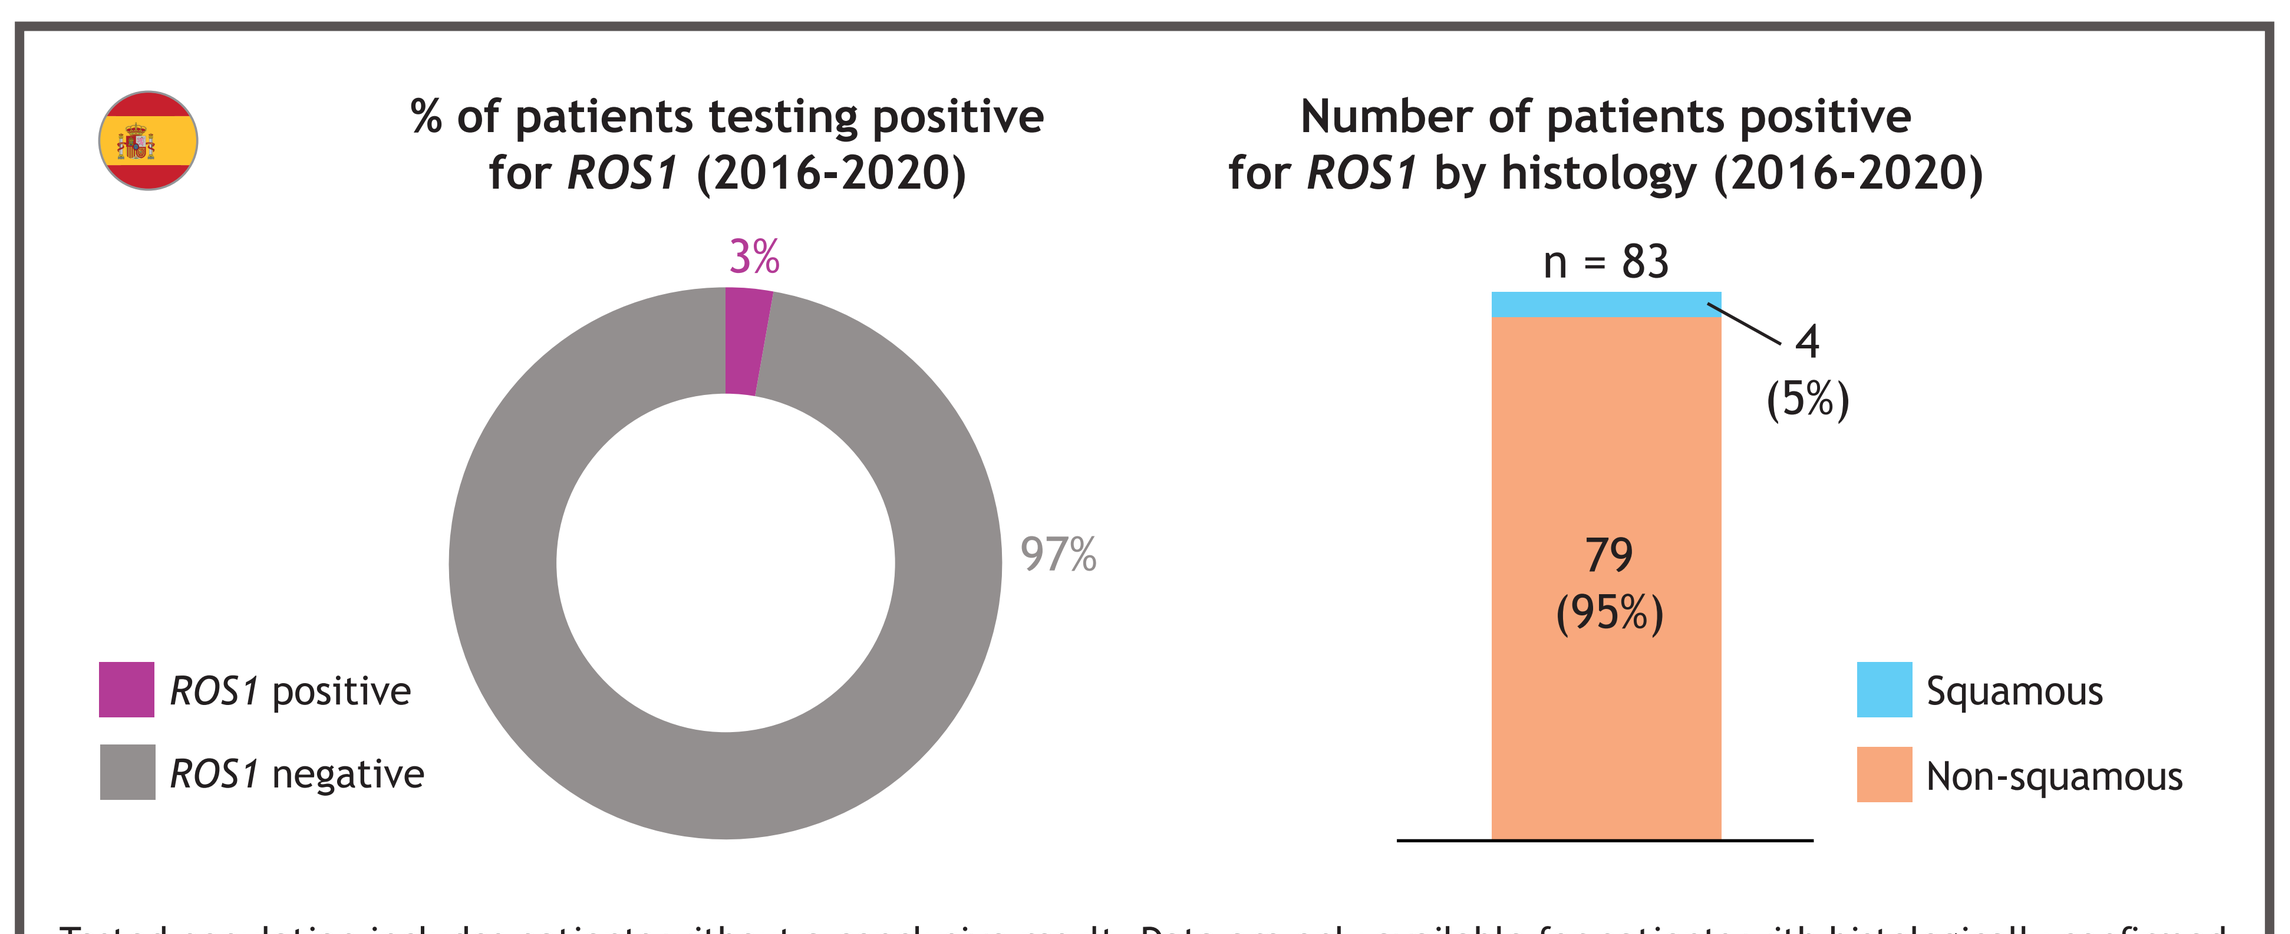

% of patients testing positive for ROS1 (2016-2020)
3% 97%
ROS1 positive
ROS1 negative
Number of patients positive for ROS1 by histology (2016-2020)
n = 83
4 (5%)
79 (95%)
Squamous
Non-squamous

Tested population includes patients without a conclusive result. Data are only available for patients with histologically confirmed stage IV NSCLC from 182 hospitals across the Spanish territory and represent a significant proportion of the territory but may not reflect countrywide practices.

Figure 4d. England

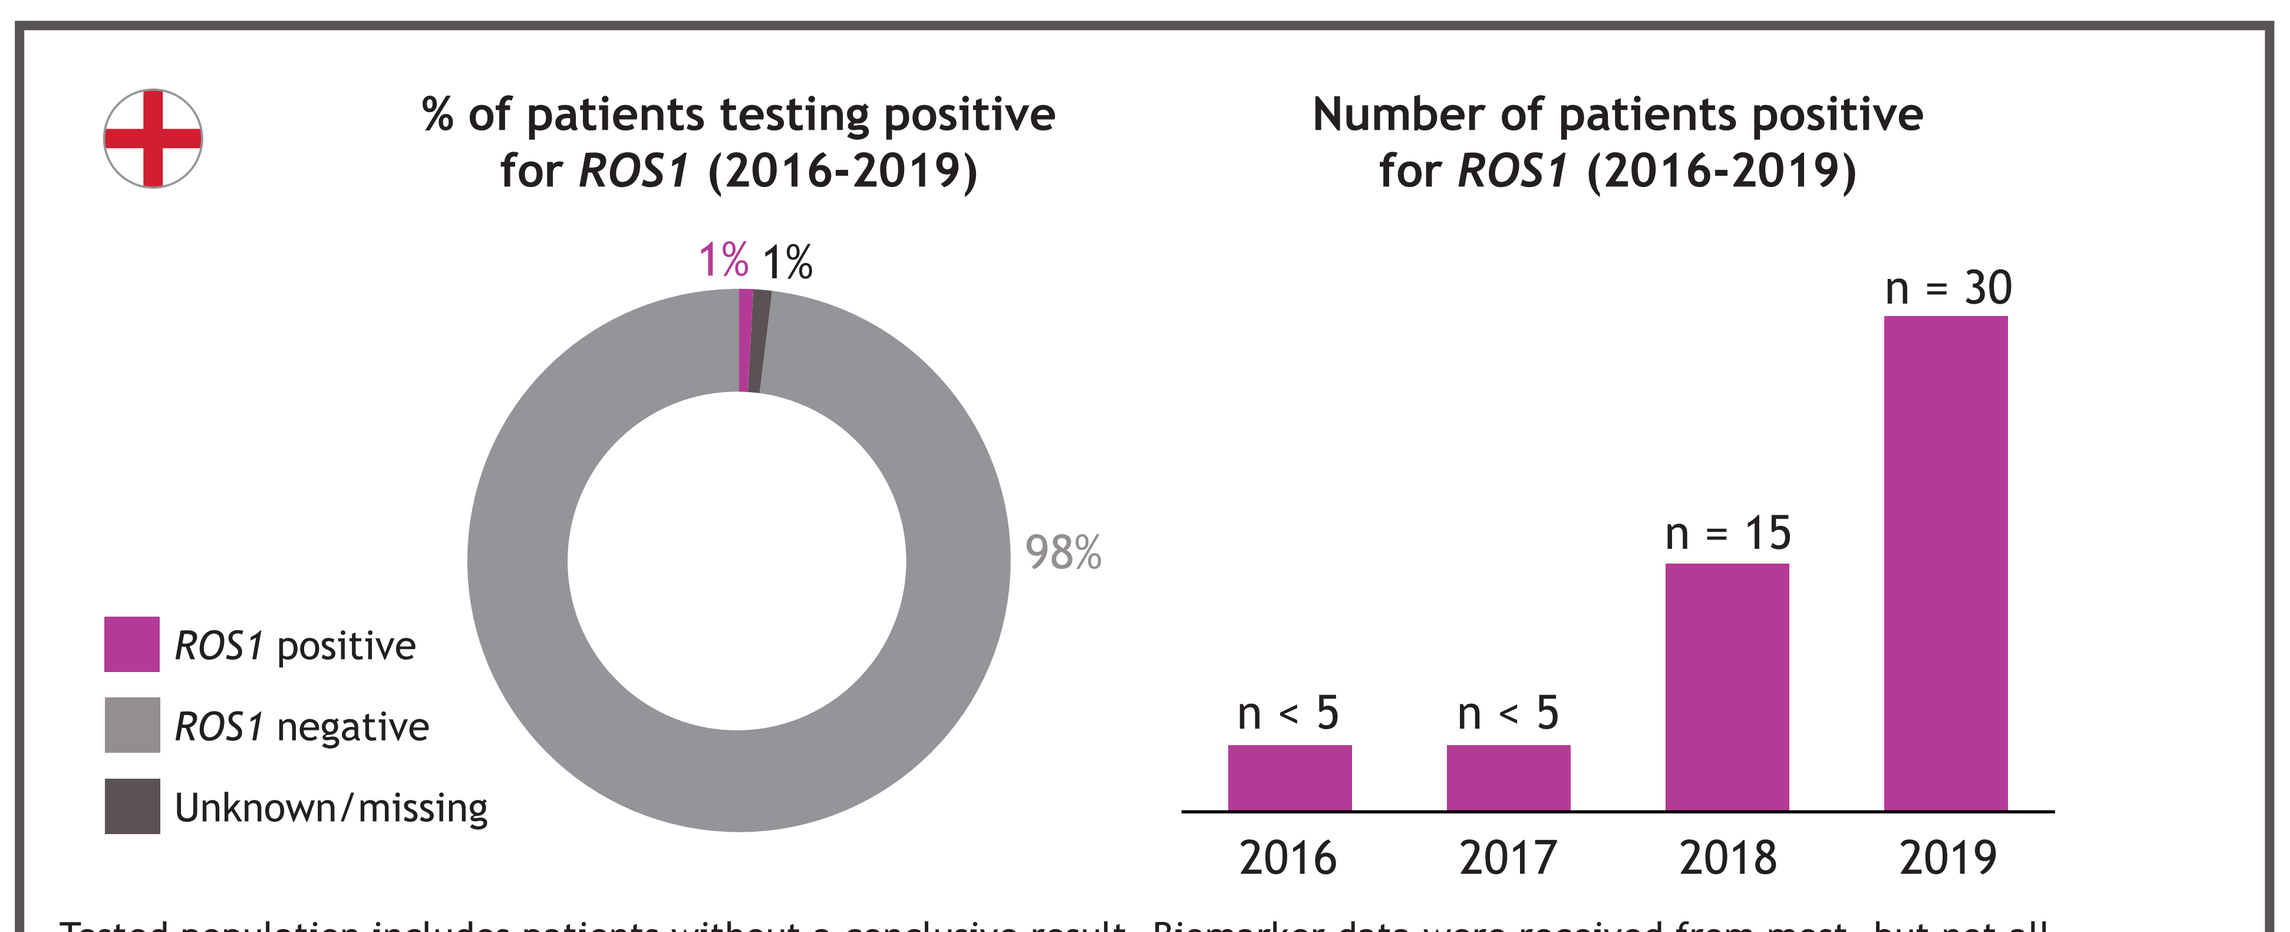

% of patients testing positive for ROS1 (2016-2019)
1% 1% 98%
ROS1 positive
ROS1 negative
Unknown/missing
Number of patients positive for ROS1 (2016-2019)
n < 5 n < 5 n = 15 n = 30
2016 2017 2018 2019

Tested population includes patients without a conclusive result. Biomarker data were received from most, but not all, molecular diagnostic laboratories in England, which may impact the total number of patients with *ROS1* positivity.

Figure 4e. Sweden

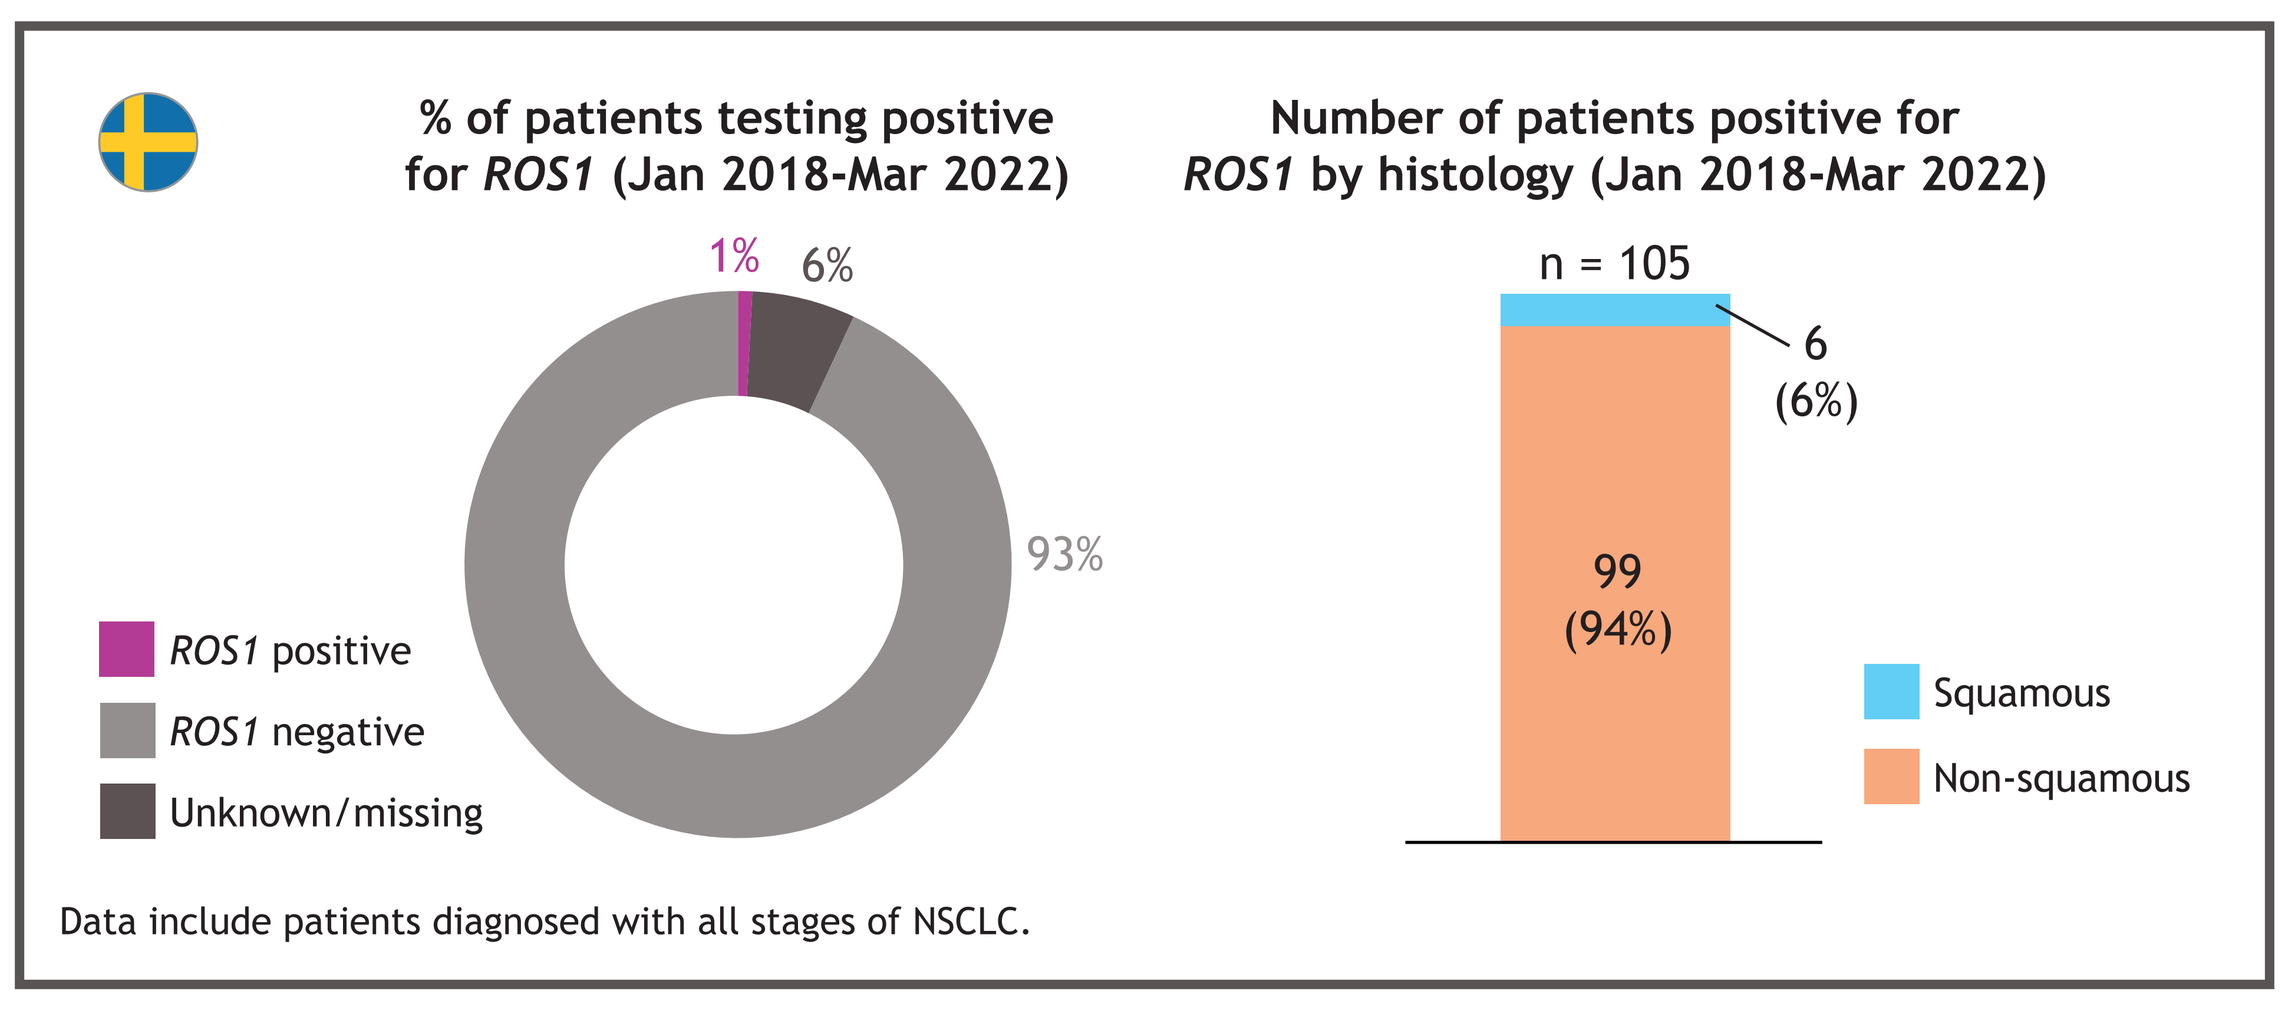

% of patients testing positive for ROS1 (Jan 2018-Mar 2022)
1% 6% 93%
ROS1 positive
ROS1 negative
Unknown/missing
Number of patients positive for ROS1 by histology (Jan 2018-Mar 2022)
n = 105
6 (6%)
99 (94%)
Squamous
Non-squamous

Data include patients diagnosed with all stages of NSCLC.

Conclusions

- These data provide evidence of increased *ROS1* testing across Europe and associated increased numbers of patients identified with *ROS1*+ NSCLC, highlighting a growing population of patients who may benefit from targeted therapy
- Countries with the highest rate of *ROS1* testing tended to be those that had the earliest introduction of national recommendations for biomarker testing (eg, France and Germany) and those with the lowest rate of testing corresponded with countries with a lack of reimbursement (eg, Spain and Italy)
 - The relatively high projected *ROS1* testing rates observed in countries without reimbursement in 2022 were likely a reflection of (i) testing for *ROS1* alterations as part of a standard genomic testing panel or (ii) specific *ROS1* testing for enrolment in clinical trials of *ROS1*-targeted therapy
- The reported actual data on *ROS1* positivity from country-specific data sources was consistent with the Oncology Dynamics projected data; the *ROS1* positivity rates projected (1%-5%) and observed (1%-3%) in this study were similar to the range of rates reported in other real-world studies (~0.5%-3%),[1,4-6] highlighting the validity of this approach to estimate patient numbers from multiple data sources
 - The actual numbers of patients recorded with *ROS1*+ NSCLC were lower than projected figures for the same period (where data were available)
- The differences in testing rates between countries likely reflect the variability in how and when international guidelines for treatment were adopted and when treatments are approved for reimbursement, and highlight that the implementation of guidelines and treatments are not always aligned
- Given the scarcity of European data on patients with *ROS1*+ NSCLC, this study provides useful insights into *ROS1* alteration testing and positivity among patients with NSCLC in Europe

Presented at International Society for Pharmacoeconomics and Outcomes Research (ISPOR) Europe 2024; November 17-20; Barcelona, Spain

Email: p.baas@nki.nl

Copies of this poster are for personal use only and may not be reproduced without written permission of the authors.