OP7

Challenges in Treating PNH Patients in Brazil's Public Healthcare System Due to the Frequency of Eculizumab Administration and Patients' Distance from Infusion Centers

W. Hirth[1]; J. Nacazume[1]; M. Mendoça[1]; L. Branco[2]; V. Mata[2]

[1] Valueconnected, São Paulo, São Paulo, Brazil; [2] F.Hoffmann-La Roche, São Paulo, Brazil

OBJECTIVES

The study aimed to describe the commute distances and times for patients to access infusion centers for eculizumab administration in the treatment of paroxysmal nocturnal hemoglobinuria (PNH) within the Brazilian public healthcare system (SUS).

METHODS

Public databases of healthcare facilities (CNES) and patient records (DataSUS) from January 2023 to February 2024 were analyzed to identify PNH patients treated with eculizumab and the centers administering the biweekly infusions. Since only four states specified the centers administering eculizumab, centers were identified based on capabilities, qualifications as blood or rare disease reference centers, and eculizumab dispensation procedure requests. Patients' geolocations were determined by postal codes or municipalities from DataSUS, and infusion centers' geolocations were obtained from CNES data. Distance and time between patients' homes and infusion centers were calculated using a distance matrix API.

RESULTS

Of the 376 PNH patients receiving eculizumab, 55% reside in a different municipality from their infusion center. Patients travel an average of 101 kilometers to reach the infusion center, with an average one-way commute time of 77 minutes (1-711 minutes), implying nearly 2.5 hours of commuting on infusion days. Patients in the Northeast, North, and South regions experience longer commutes averaging about 3 hours on infusion days.

Region	Number of patients	Average driving distance (Km)	Average driving time (min)
North	21	131	104
Northeast	114	118	96
South	75	114	83
Southeast	123	81	60
Midwest	43	71	55
Brazil	**376**	**101**	**77**

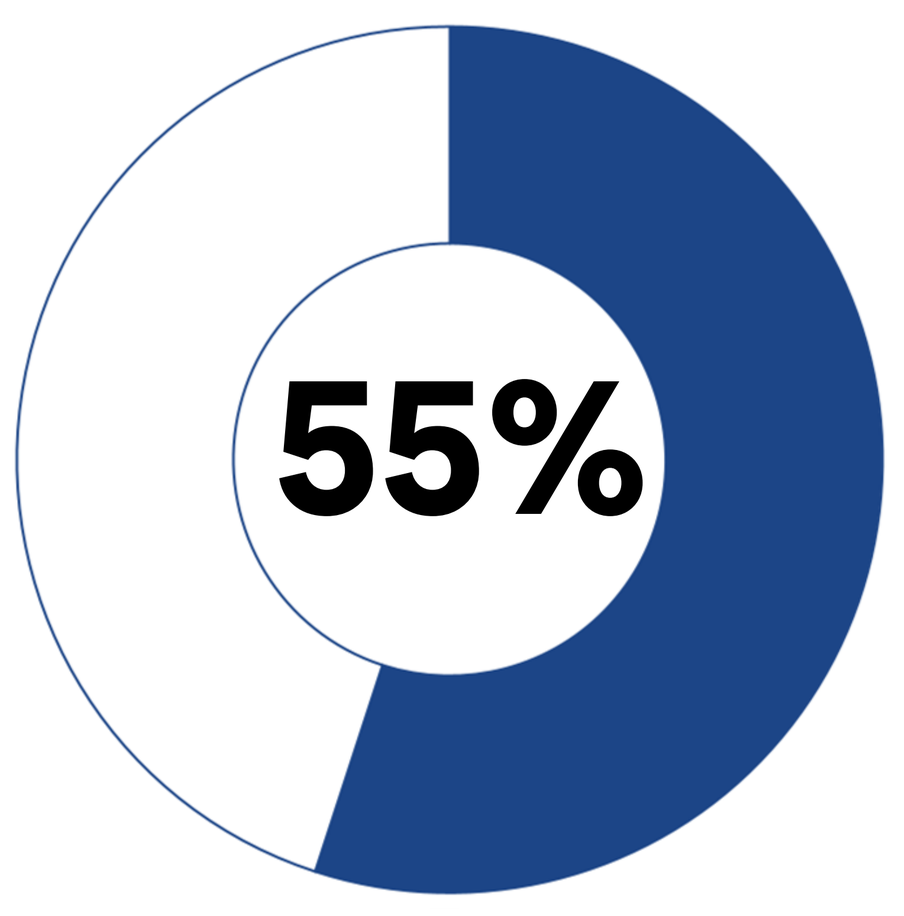
55%

Reside in **a different municipality** from their infusion center

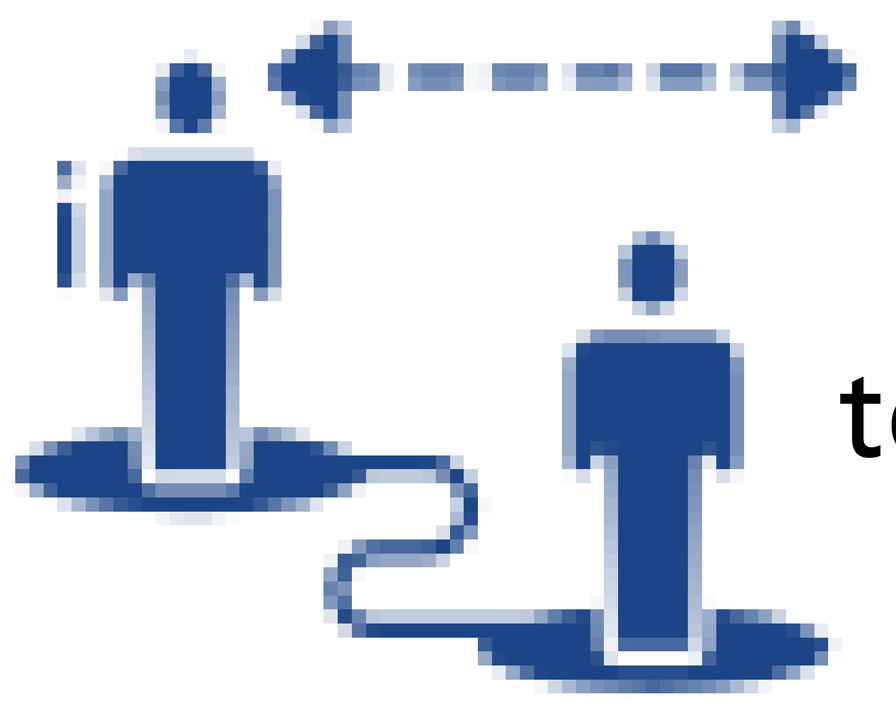

101 km
to reach the infusion center

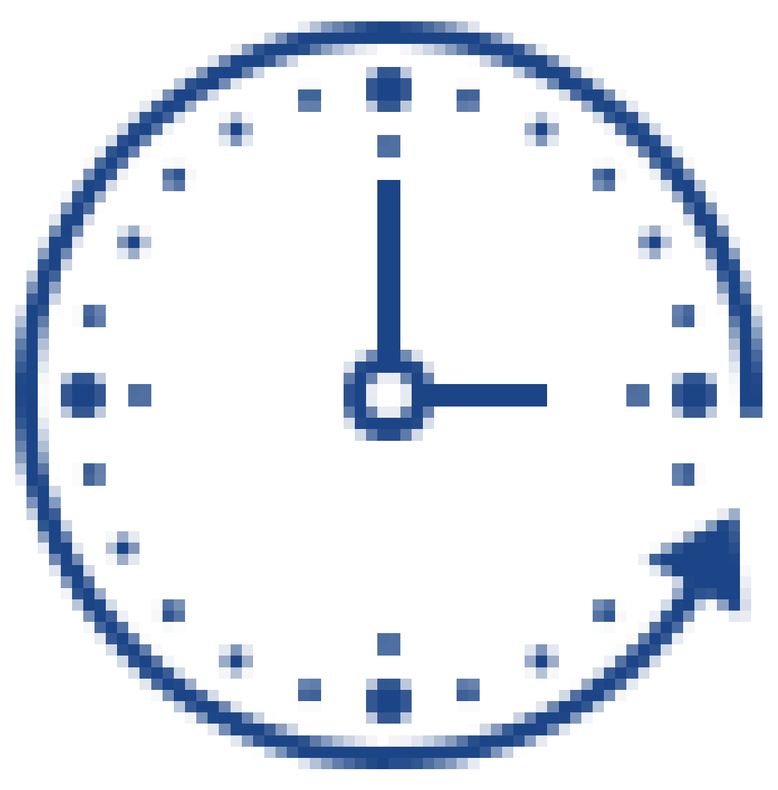

2.5 hours
of commuting on infusion days

CONCLUSIONS

The study shows, with real-world data, the logistical challenges that PNH patients face in accessing their current drug treatment, where one in two patients needs to travel biweekly to different municipalities to receive the treatment, with commutes lasting up to two hours on infusion days. However, the total time spent might be underestimated, as they do not consider variables like traffic conditions, transportation modes, or infusion duration, all of which can impact treatment time, patient quality of life and adherence. These findings underscore the need for more efficient treatment options that offer more convenient posology and sustainable healthcare logistics.

REFERENCE

Brasil. Protocolo clínico e diretrizes terapêuticas hemoglobinúria paroxística noturna. Brasília; 2019. Available from: http://portalms.saude.gov.br/protocolos-e-diretrizes. Accessed July 14, 2024.

Brasil. Ministério da Saúde. Portaria n.º 55, de 24 de fevereiro de 1999. Dispõe sobre a rotina do tratamento fora de domicílio no Sistema Único de Saúde - SUS, com inclusão dos procedimentos específicos na tabela de procedimentos do Sistema de Informações Ambulatoriais do SIA/SUS e dá outras providências. Brazil; 1999. Available from: https://bvsms.saude.gov.br/bvs/saudelegis/sas/1999/prt0055_24_02_1999.html. Accessed July 14, 2024.

Governo da Paraíba. Guia informativo - Hemoglobinúria Paroxística Noturna. Available from: https://paraiba.pb.gov.br/diretas/saude/cedmex-old/documentos-por-patologia-para-obter-medicamentos/hemoglobinuria-paroxistica-noturna/guia-informativo_hemoglubinuria-paroxistica-noturna/view. Accessed July 14, 2024.

Governo do Estado de São Paulo. Nota técnica CAF n.º 18/2023 - Distribuição, dispensação e administração do medicamento eculizumabe para tratamento de pacientes com hemoglobinúria paroxística noturna (HPN) no âmbito do Sistema Único de Saúde (SUS). Available from: https://saude.sp.gov.br/resources/ses/perfil/gestor/assistencia-farmaceutica/notas-tecnicas/sei_024.00160984_2023_02_-_nt_caf_18-2023.pdf. Accessed July 14, 2024.

Governo do Estado da Bahia. Fluxo de acesso para hemoglobinúria - Portaria Conjunta n.º 18. Available from: https://www.saude.ba.gov.br/wp-content/uploads/2024/01/FLUXO-DE-ACESSO-HEMOGLOBINIRIA.pdf. Accessed July 14, 2024.

Governo do Estado de Santa Catarina. Nota técnica conjunta n.º 01/2023 DIAF/SPS/SUR/SUH/SES/SC. Available from: https://www.saude.sc.gov-.br/index.php/componente-especializado-da-assistencia-farmaceutica-ceaf/informes-ceaf/informes-2023/20920-nota-tecnica-conjunta-01-2023-diaf-sps-sur-suh-ses-sc/file. Accessed July 14, 2024.

DATASUS (Departamento de Informática do SUS). c2008. Disponível em: http://www2.datasus.gov.br/DATASUS/index.php. Accessed April 30, 2024.

Feep!k images.