Economic Impact of Bimekizumab for Psoriatic Arthritis and Axial Spondyloarthritis in France

EE234

De Pouvourville Gérard[1], Fabre Esther[2], Schleret Thomas[2], Leproust Sandy[3], Chabani Bilel[3], Lyris Nikos[4], Willems Damon[5], McCarthy Peter[6], Goupille Philippe[7]

Objectives

This study aimed to assess the budget impact of the introduction of bimekizumab, a humanised monoclonal IgG1 antibody that selectively inhibits interleukin (IL)-17F in addition to IL-17A [1], as a treatment option for patients diagnosed with psoriatic arthritis (PsA) or axial spondyloarthritis (axSpA) in France.

Introduction

PsA is a long-term, systemic inflammatory condition where patients have musculoskeletal symptoms along with skin inflammation related to psoriasis [2].

AxSpA is an inflammatory rheumatic disease that impacts the axial skeleton, leading to severe pain, stiffness, and fatigue. AxSpA includes non-radiographic axial spondyloarthritis (nr-axSpA) and radiographic axial spondyloarthritis (r-axSpA) [3].

Methods

Model

A joint budget impact model with a 5-year time horizon was adapted to estimate the economic impact of introducing bimekizumab into the therapeutic strategy from the French payer perspective (Figure 1).

Target population & market share

Target populations assessed by the French Health Authority (HAS) were considered [4,5].

The main market share uptake assumption was a gain of bimekizumab over anti-IL-17A and anti-IL-23 treatments and was based on UCB market forecast estimates. Market shares were differentiated between patients who are biologic/synthetic (b/ts) DMARD-naïve and b/ts-DMARD in inadequate response (IR) (Table 1).

Comparators and costs

The budget impact analysis included all relevant comparators (anti-tumor necrosis factor (TNF)-alpha, anti-IL 17A, 12/23 and 23, and Janus kinase inhibitors (JAKi)) in both indications. Direct medical costs were considered, including treatment acquisition (list prices), administration, monitoring, and costs associated with adverse events (AE) (Table 2).

Clinical data

Clinical response rates are included in the model and obtained from published network meta-analyses (for the probability of response using the American College of Rheumatology score - ACR50 for PsA [6] and the Assessment of SpondyloArthritis International Society score - ASAS40 for axSpA [7]).

Patients who do not respond to treatment move to a new treatment as inadequate responders.

Based on experts' opinions, serious infections were included, and their probability of occurrence was based on the clinical trials of each treatment.

Sensitivity analysis

One-way sensitivity analyses were conducted by varying the model inputs (±10% of the original estimates).

Different scenarios were explored, with different market share uptakes for bimekizumab, and the inclusion of indirect costs to non-responders.

Results

Base case analysis

The number of patients treated with bimekizumab in the new market scenario was projected to rise from 845 (PsA: 630; axSpA: 215) patients in 2024 to 2581 (PsA: 1807; axSpA: 774) patients in 2028.

Through the years, the inclusion of bimekizumab into the market is projected to result in a total budget decrease of €7,670,990 for PsA (-2.24% compared to scenario without bimekizumab, amounting to €342,070,210) and increase of €1,572,426 for axSpA (+0.54% compared to scenario without bimekizumab, amounting to €289,652,288) over a 5-year time horizon. (Figure 2)

Introducing bimekizumab to the French rheumatology market is projected to lead to an average budget reduction of €1,219,713 per year, primarily driven by savings in drug acquisition costs (97.30%).

Sensitivity analysis

The model was most sensitive to changes in drug acquisition costs and the eligible population. Overall, sensitivity analyses revealed no significant deviations from the base case analysis. (Figure 3)

Conclusions

Based on this budget impact analysis, the introduction of bimekizumab to the PsA and axSpA markets in France is expected to have a neutral impact on the overall French health insurance budget over a 5-year time horizon.

Summary

The aim of this study was to assess the budgetary impact of introducing bimekizumab in France for treating axial spondyloarthritis and psoriatic arthritis, from the perspective of a public payer.

5-year budget impact		
PsA	axSpA	Pooled
-€7,670,990	€1,572,426	-€6,098,564

A minor budget increase was projected in the axSpA market (+0.54%); while the PsA market saw greater savings (-2.24%), resulting in a minor budget impact. This difference is mainly due to the variation in available treatments for the two diseases, affecting market adoption scenarios.

Scenario analysis including indirect costs showed increased cost savings (-40.58% budget in the PsA and axSpA market), highlighting the clinical benefits of bimekizumab in rheumatology, as its responder rate exceeded the average pre-market introduction response rate.

Overall, the introduction of bimekizumab to the PsA and axSpA markets in France is expected to have a neutral impact on the overall French health insurance budget (-0.97% of the total budget of €631,722,498).

Figure 1 Budget impact model structure

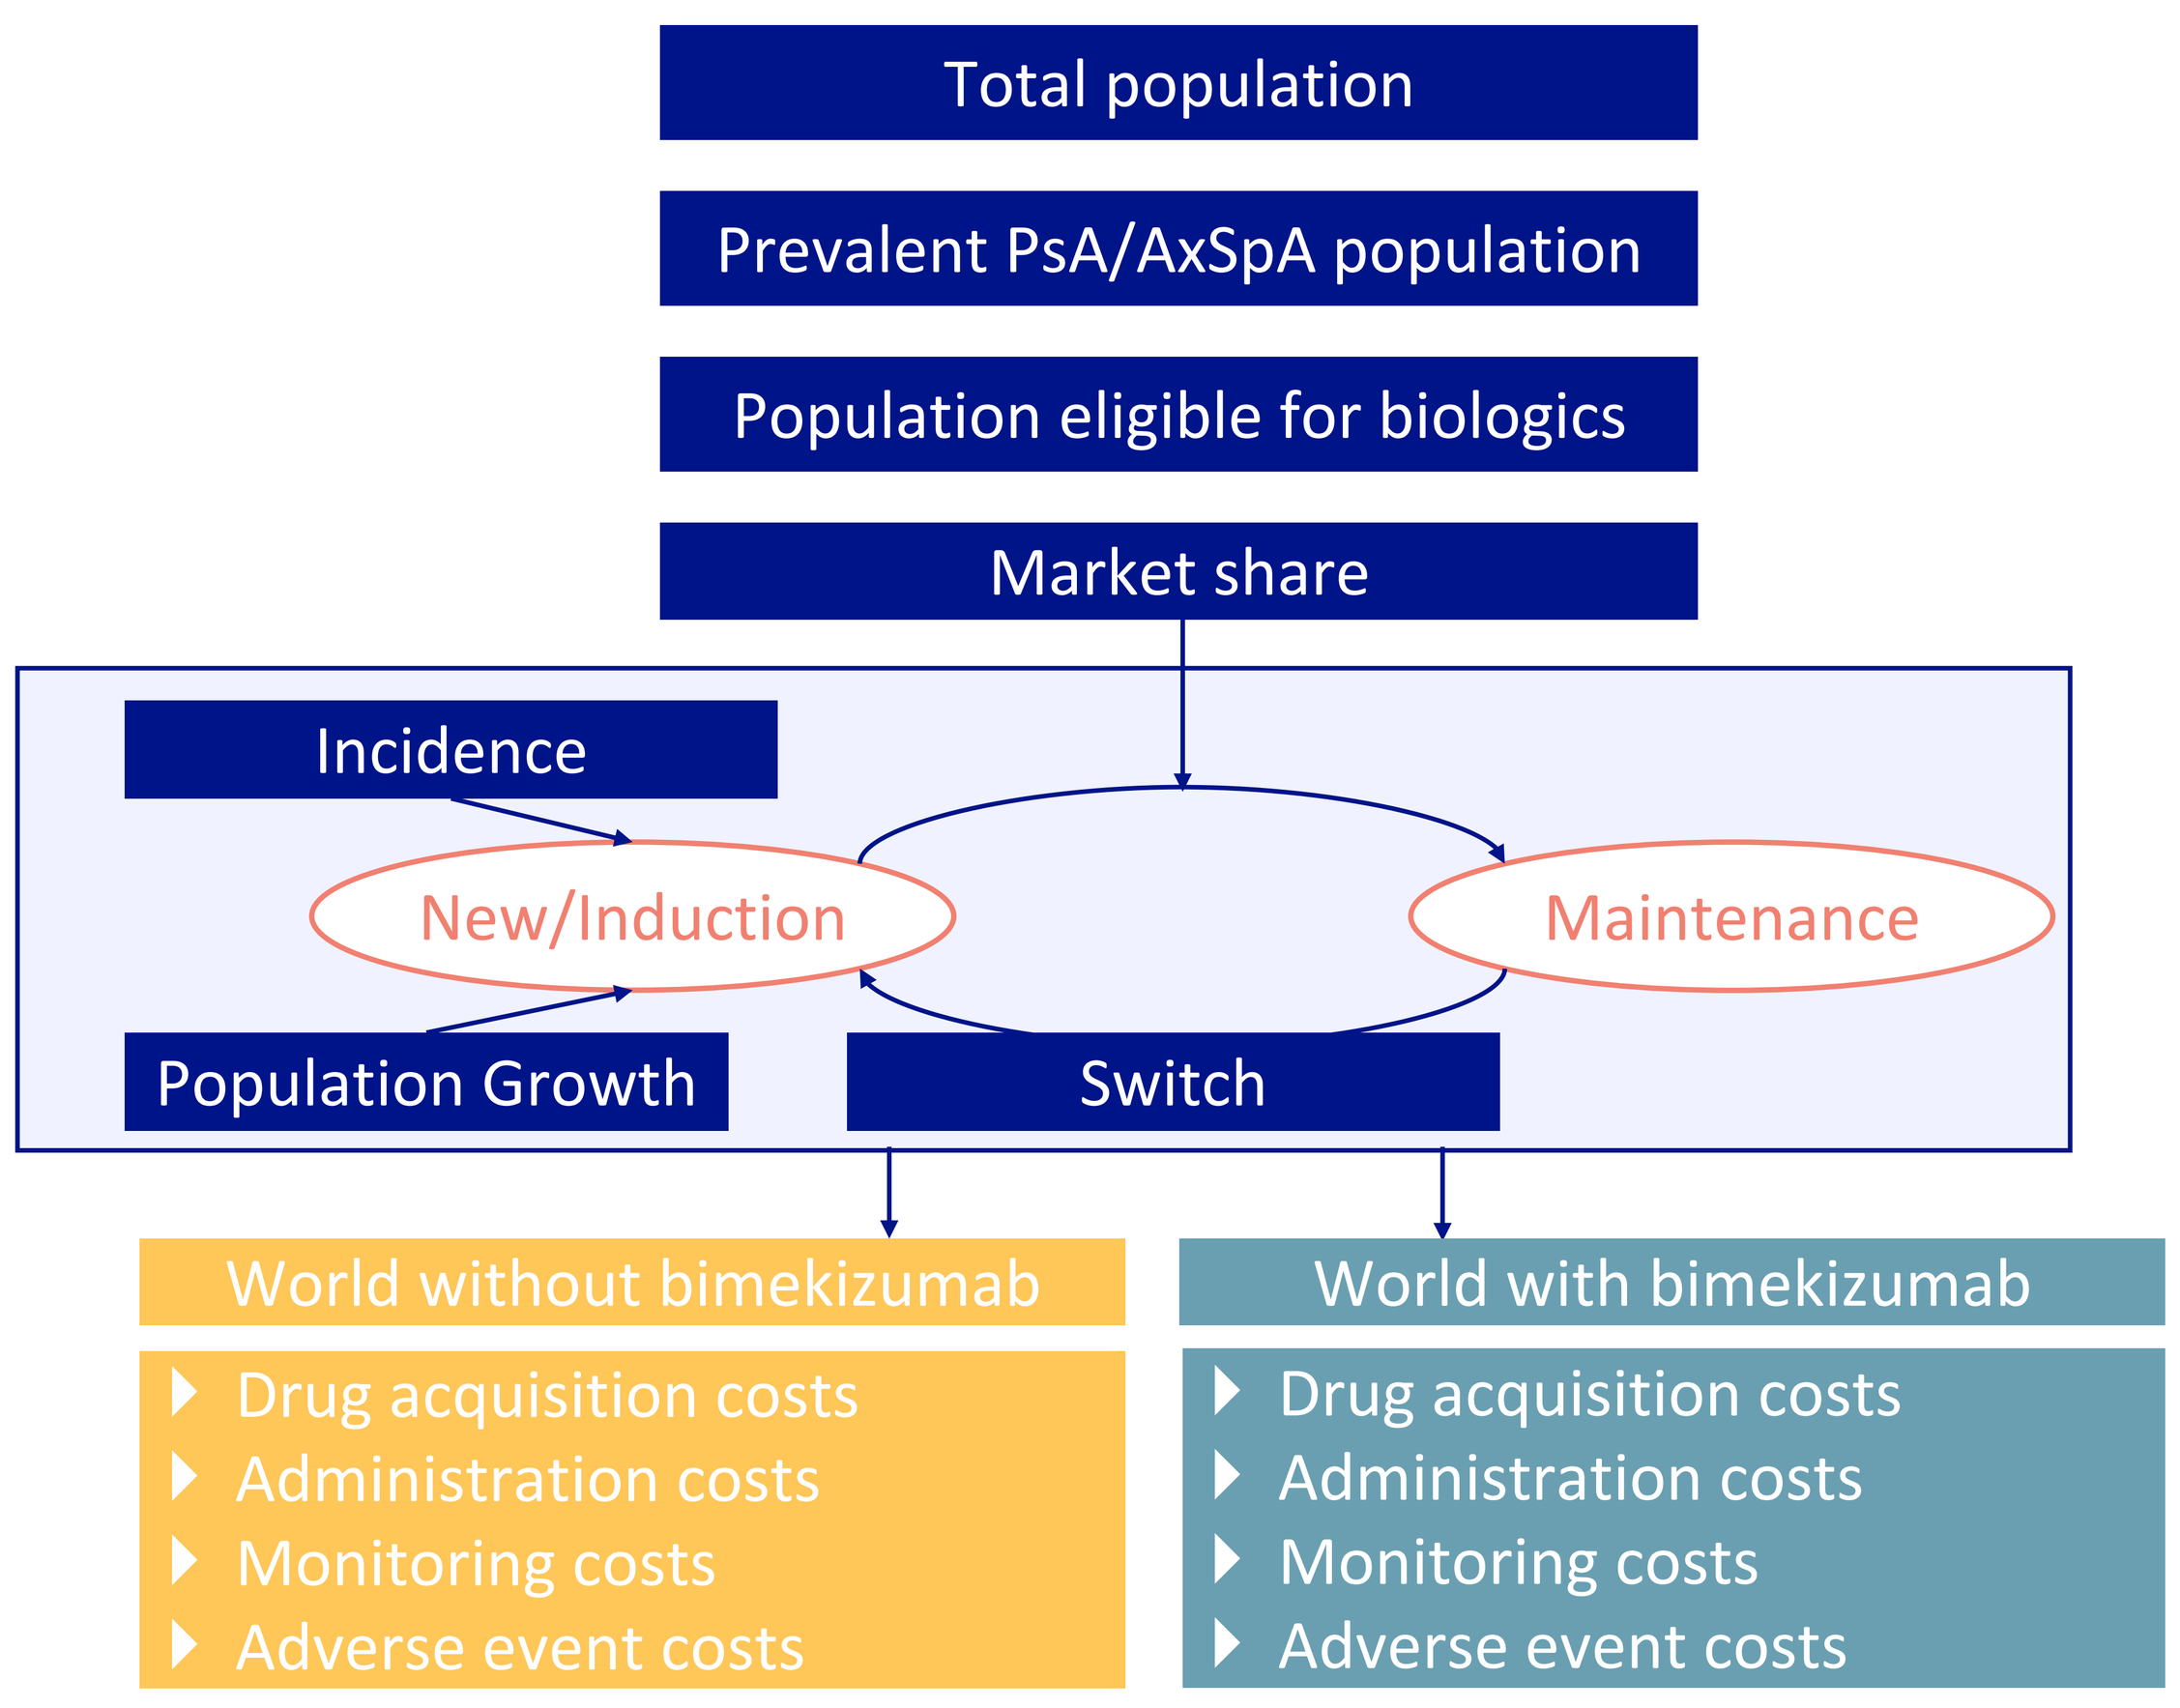

Total population
Prevalent PsA/AxSpA population
Population eligible for biologics
Market share
Incidence
New/Induction
Maintenance
Population Growth
Switch
World without bimekizumab
Drug acquisition costs
Administration costs
Monitoring costs
Adverse event costs
World with bimekizumab
Drug acquisition costs
Administration costs
Monitoring costs
Adverse event costs

Model assumption: The number of maintenance patients will be the prevalent population minus the new patients. The total population will be the same as the prevalent population.

Figure 2 Total annual incremental costs and cumulative costs

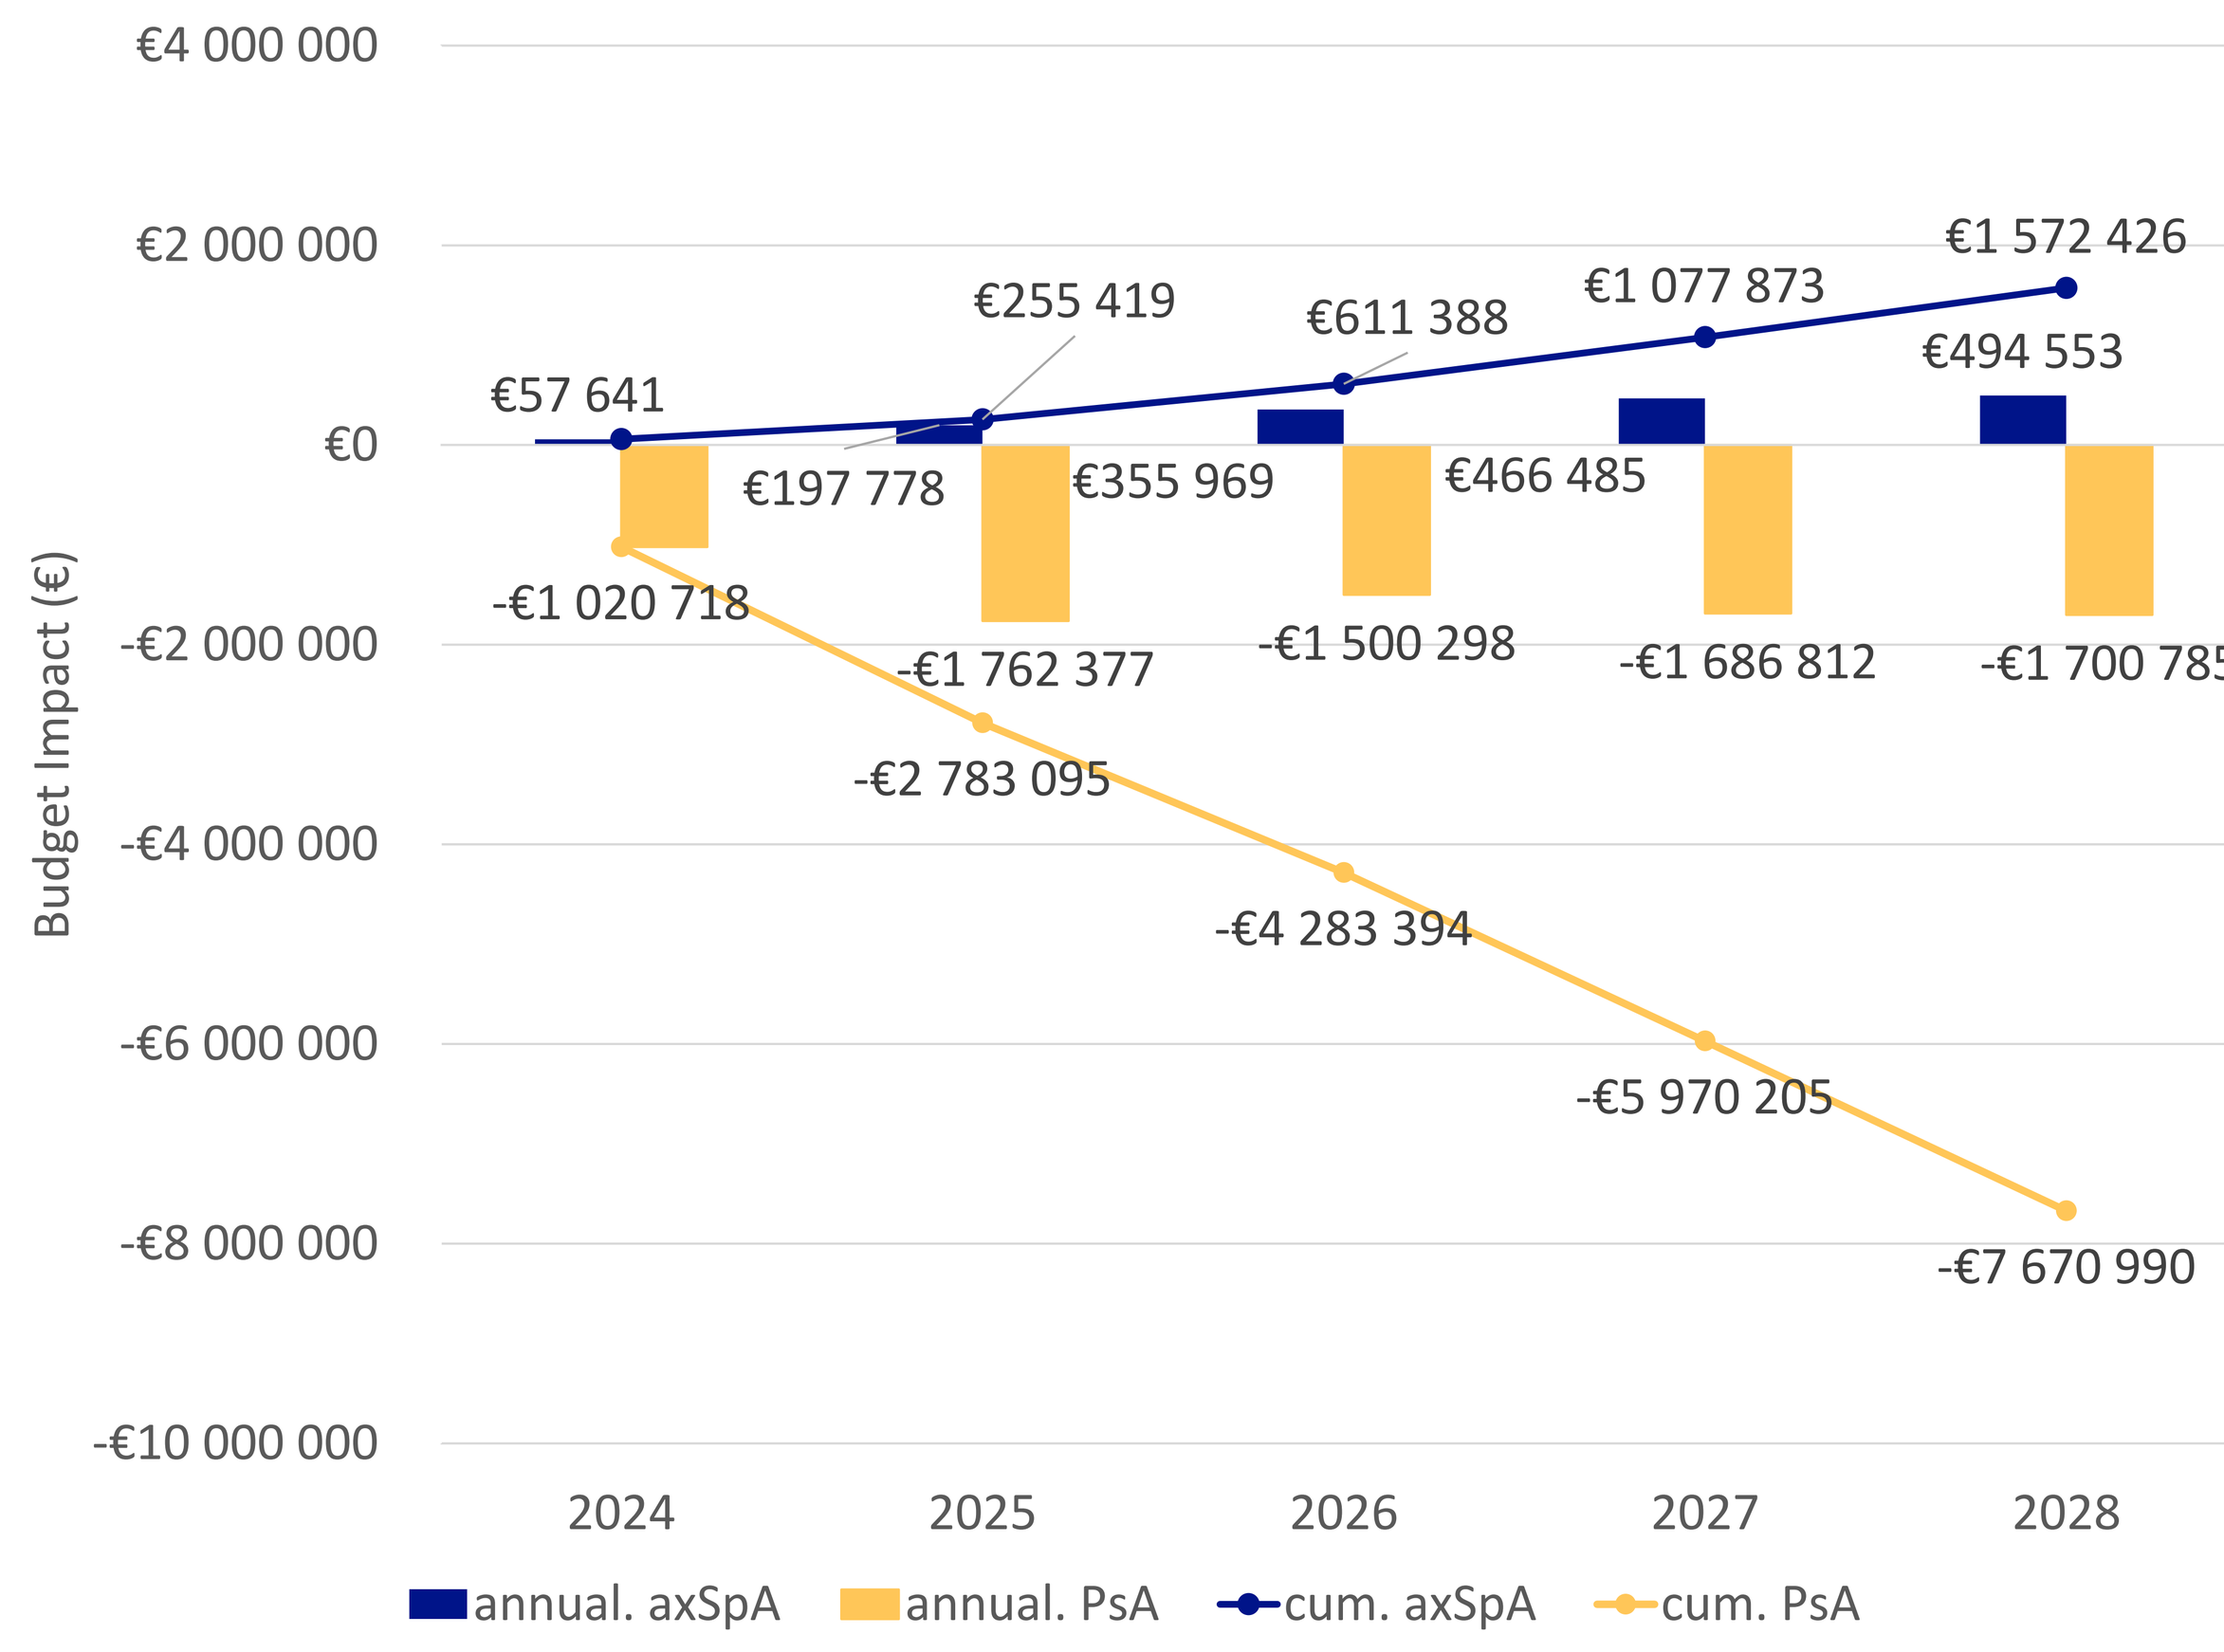

€4 000 000
€2 000 000
€0
-€2 000 000
-€4 000 000
-€6 000 000
-€8 000 000
-€10 000 000
Budget Impact (€)
€57 641
€255 419
€611 388
€1 077 873
€1 572 426
€197 778
€355 969
€466 485
€494 553
-€1 020 718
-€1 762 377
-€1 500 298
-€1 686 812
-€1 700 785
-€2 783 095
-€4 283 394
-€5 970 205
-€7 670 990
2024
2025
2026
2027
2028
annual. axSpA
annual. PsA
cum. axSpA
cum. PsA

Note: "annual." denotes the annual cost; "cum." denotes the cumulative cost

Figure 3 Univariate sensitivity analysis for the pooled axSpA and PsA 5-year budget impact

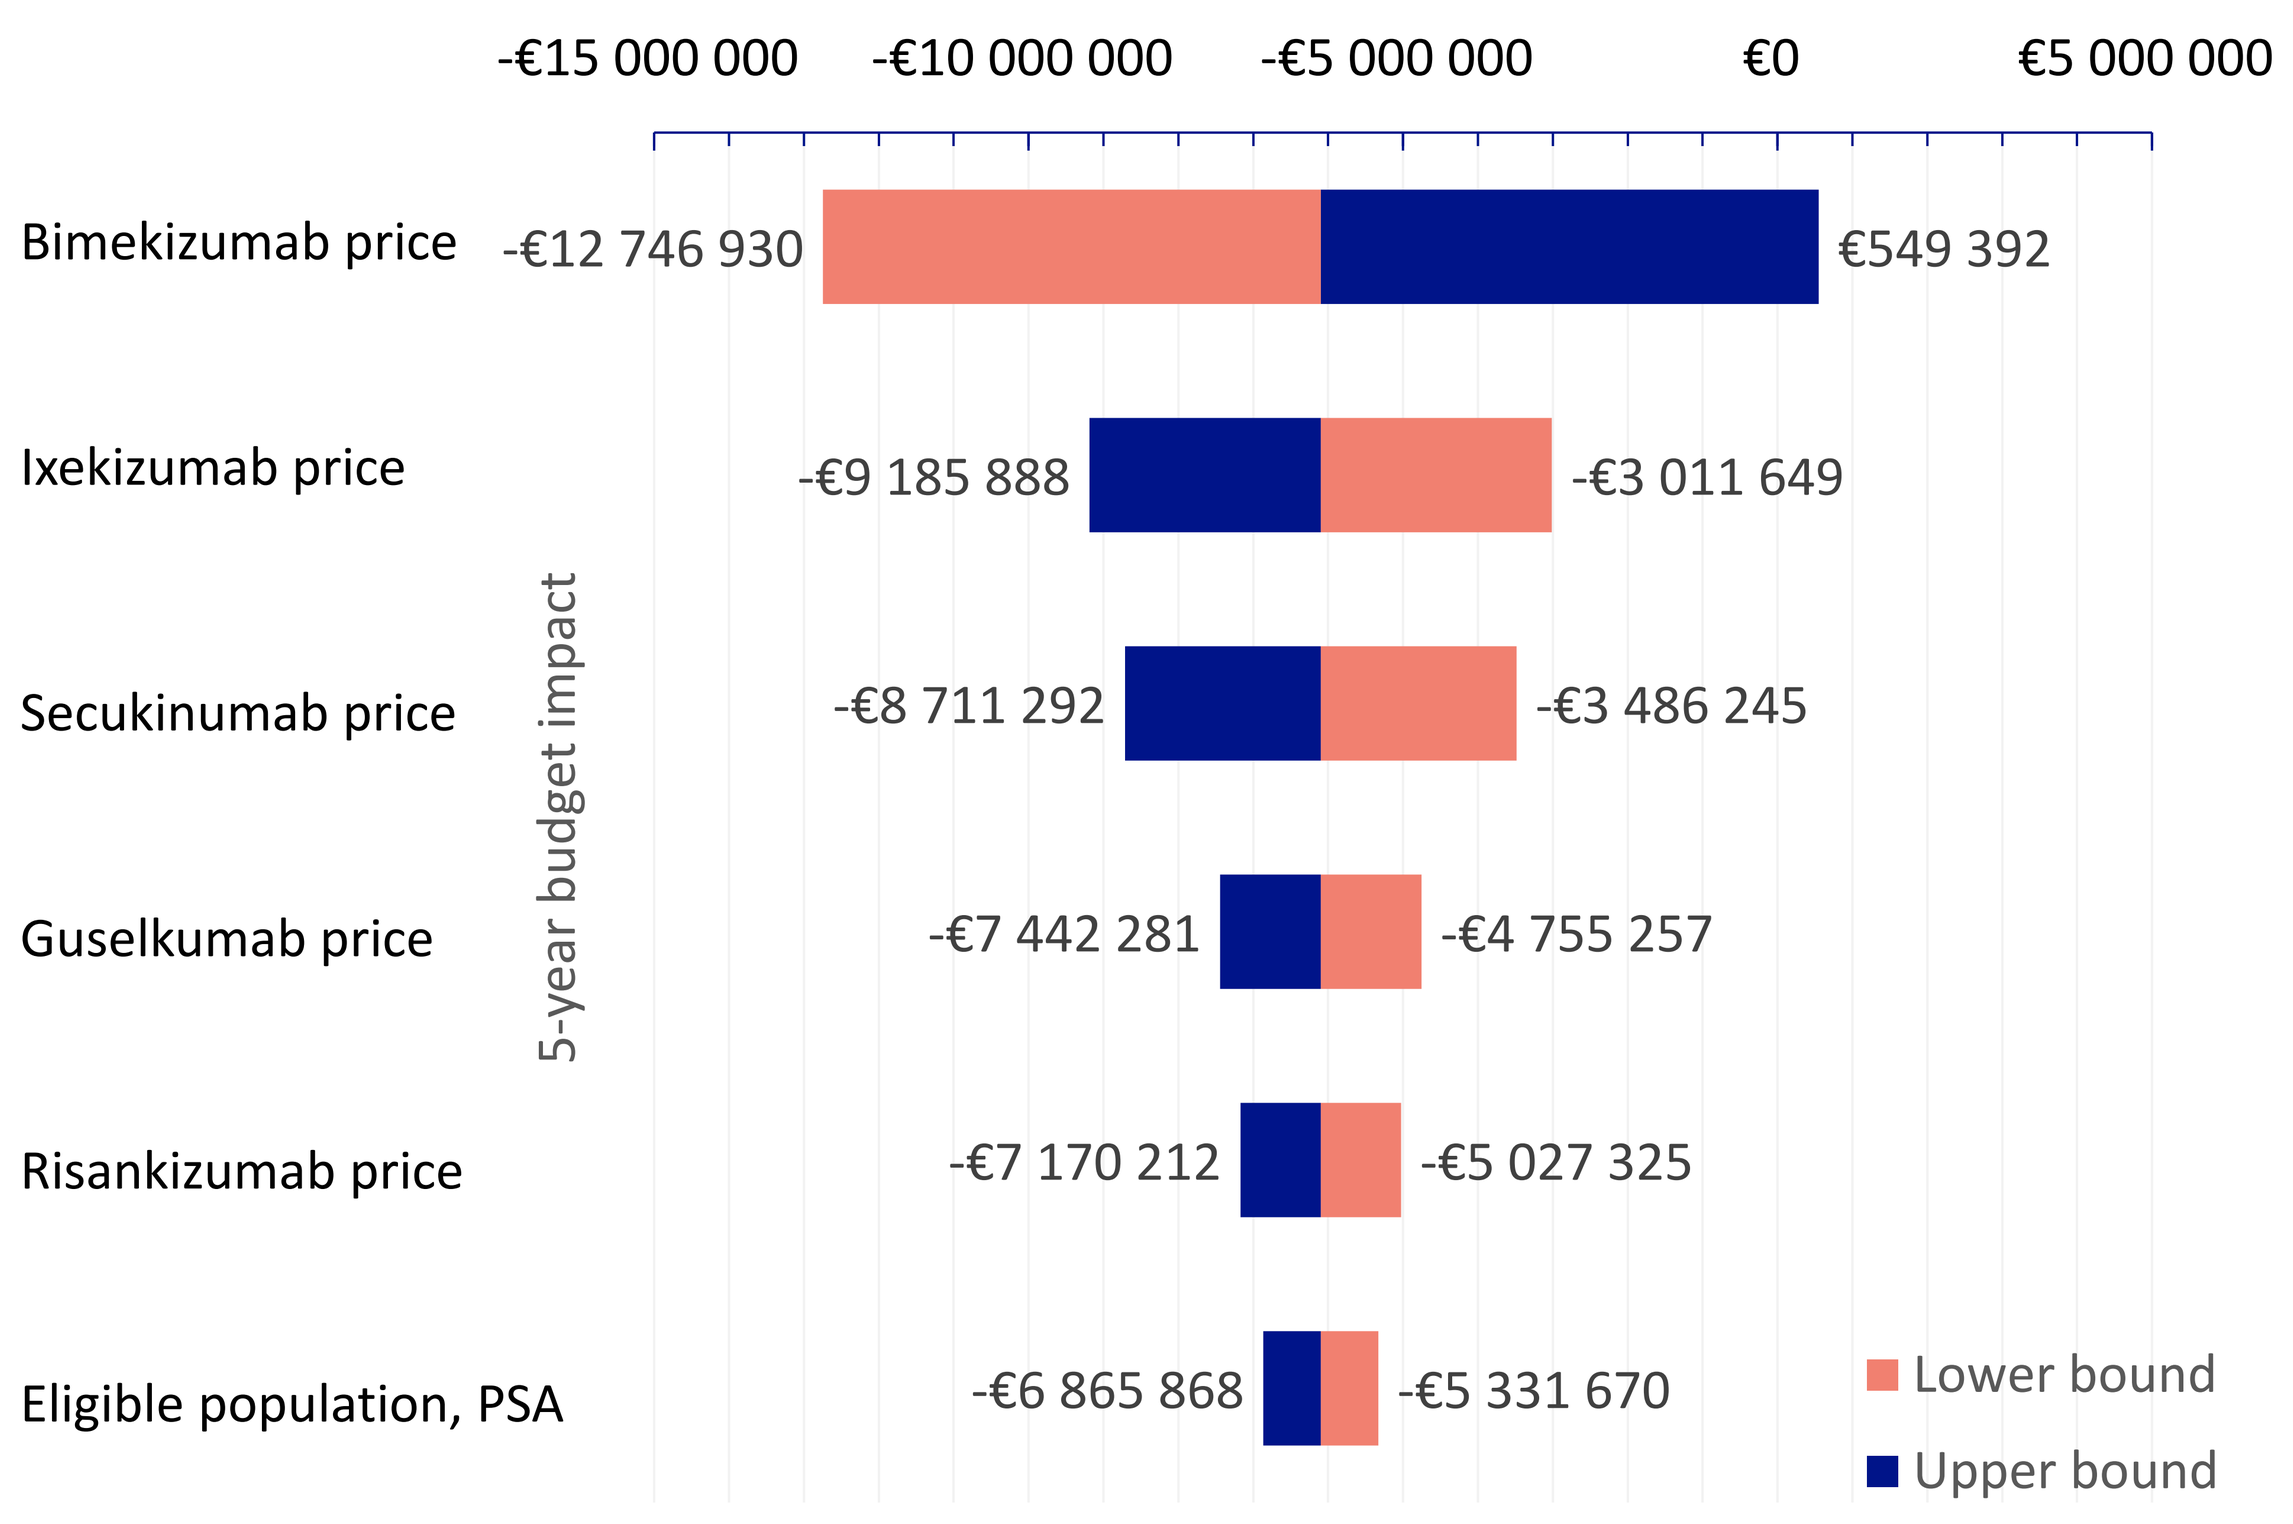

-€15 000 000
-€10 000 000
-€5 000 000
€0
€5 000 000
Bimekizumab price
-€12 746 930
€549 392
Ixekizumab price
-€9 185 888
-€3 011 649
Secukinumab price
-€8 711 292
-€3 486 245
Guselkumab price
-€7 442 281
-€4 755 257
Risankizumab price
-€7 170 212
-€5 027 325
Eligible population, PSA
-€6 865 868
-€5 331 670
5-year budget impact
Lower bound
Upper bound

Note: The vertical blue line represents the base case 5-year budget impact. "Lower bound" shows the budget impact for the lower bound of the parameter, "Upper bound" shows the budget impact for the upper bound of the parameter.

Table 1 Market share scenarios with and without bimekizumab impact

Bio-naïve patients

Treatment	Market share – World with bimekizumab 2023	2024	2025	2026	2027	2028	Market share – World without bimekizumab 2023	2024	2025	2026	2027	2028
Psoriatic arthritis												
Bimekizumab	0%	4%	10%	11%	13%	13%	-	-	-	-	-	-
Adalimumab	8%	8%	8%	7%	7%	7%	8%	8%	8%	7%	7%	7%
Adalimumab (biosimilar)	14%	14%	14%	14%	15%	15%	14%	14%	14%	14%	15%	15%
Certolizumab pegol	4%	3%	3%	3%	2%	2%	4%	3%	3%	3%	2%	2%
Etanercept	8%	7%	4%	4%	4%	4%	8%	7%	4%	4%	4%	4%
Etanercept (biosimilar)	9%	9%	9%	9%	9%	9%	9%	9%	9%	9%	9%	9%
Golimumab	3%	2%	1%	1%	1%	1%	3%	2%	1%	1%	1%	1%
Guselkumab	13%	13%	12%	12%	12%	12%	13%	13%	14%	15%	15%	15%
Infliximab	0%	0%	0%	0%	0%	0%	0%	0%	0%	0%	0%	0%
Infliximab (biosimilar)	3%	3%	3%	3%	3%	3%	3%	3%	3%	3%	3%	3%
Ixekizumab	9%	8%	7%	7%	7%	7%	9%	9%	9%	9%	9%	9%
Risankizumab	10%	9%	9%	9%	9%	9%	10%	11%	12%	12%	12%	12%
Secukinumab	9%	8%	8%	8%	8%	8%	9%	9%	11%	11%	12%	12%
Tofacitinib	0%	2%	2%	2%	2%	2%	0%	2%	2%	2%	2%	2%
Ustekinumab	9%	8%	7%	7%	5%	5%	9%	8%	7%	7%	6%	6%
Upadacitinib	1%	2%	3%	3%	3%	3%	1%	2%	3%	3%	3%	3%
Axial spondyloarthritis[a]												
Bimekizumab	0%	2%	4%	6%	8%	9%	-	-	-	-	-	-
Adalimumab	6%	6%	5%	5%	5%	5%	6%	6%	5%	5%	5%	5%
Adalimumab (biosimilar)	38%	38%	39%	39%	39%	39%	38%	38%	39%	39%	39%	39%
Certolizumab pegol	8%	7%	6%	6%	6%	6%	8%	7%	6%	6%	6%	6%
Etanercept	4%	4%	3%	3%	3%	3%	4%	4%	3%	3%	3%	3%
Etanercept (biosimilar)	10%	10%	9%	9%	9%	9%	10%	10%	9%	9%	9%	9%
Golimumab	8%	8%	7%	6%	6%	6%	8%	8%	7%	6%	6%	6%
Infliximab	0%	1%	1%	1%	1%	1%	0%	1%	1%	1%	1%	1%
Infliximab (biosimilar)	3%	4%	4%	5%	5%	5%	3%	4%	4%	5%	5%	5%
Ixekizumab	5%	5%	8%	6%	7%	6%	5%	6%	10%	9%	11%	11%
Secukinumab	12%	9%	8%	8%	5%	5%	12%	10%	10%	11%	9%	9%
Tofacitinib	3%	3%	3%	3%	3%	3%	3%	3%	3%	3%	3%	3%
Upadacitinib	3%	3%	3%	3%	3%	3%	3%	3%	3%	3%	3%	3%

Inadequate Response patients

Treatment	Market share – World with bimekizumab 2023	2024	2025	2026	2027	2028	Market share – World without bimekizumab 2023	2024	2025	2026	2027	2028
Psoriatic arthritis												
Bimekizumab	0%	7%	14%	15%	16%	17%	-	-	-	-	-	-
Adalimumab	6%	6%	6%	6%	5%	4%	6%	6%	6%	6%	5%	4%
Adalimumab (biosimilar)	11%	11%	11%	11%	12%	13%	11%	11%	11%	11%	12%	13%
Certolizumab pegol	7%	6%	6%	6%	6%	6%	7%	6%	6%	6%	6%	6%
Etanercept	6%	5%	4%	4%	4%	4%	6%	5%	4%	4%	4%	4%
Etanercept (biosimilar)	9%	9%	9%	9%	9%	9%	9%	9%	9%	9%	9%	9%
Golimumab	6%	5%	5%	5%	5%	5%	6%	5%	5%	5%	5%	5%
Guselkumab	11%	10%	8%	8%	8%	8%	11%	12%	13%	13%	13%	14%
Infliximab	0%	1%	1%	1%	1%	1%	0%	1%	1%	1%	1%	1%
Infliximab (biosimilar)	2%	2%	2%	2%	2%	2%	2%	2%	2%	2%	2%	2%
Ixekizumab	13%	12%	9%	8%	8%	7%	13%	14%	14%	15%	16%	14%
Risankizumab	5%	5%	5%	5%	5%	5%	5%	6%	7%	8%	8%	9%
Secukinumab	13%	11%	11%	10%	10%	10%	13%	12%	12%	10%	10%	10%
Tofacitinib	1%	2%	2%	2%	2%	2%	1%	2%	2%	2%	2%	2%
Ustekinumab	3%	2%	1%	2%	1%	1%	3%	3%	2%	2%	1%	1%
Upadacitinib	7%	6%	6%	6%	6%	6%	7%	6%	6%	6%	6%	6%
Axial spondyloarthritis[a]												
Bimekizumab	0%	4%	7%	9%	10%	11%	-	-	-	-	-	-
Adalimumab	6%	6%	5%	5%	4%	4%	6%	6%	5%	5%	4%	4%
Adalimumab (biosimilar)	18%	18%	18%	18%	19%	20%	18%	18%	18%	18%	19%	20%
Certolizumab pegol	8%	8%	7%	6%	5%	6%	8%	8%	7%	6%	5%	6%
Etanercept	6%	5%	4%	4%	4%	3%	6%	5%	4%	4%	4%	3%
Etanercept (biosimilar)	11%	11%	11%	11%	11%	11%	11%	11%	11%	11%	11%	11%
Golimumab	9%	9%	8%	7%	8%	7%	9%	9%	8%	7%	8%	7%
Infliximab	0%	1%	1%	1%	1%	1%	0%	1%	1%	1%	1%	1%
Infliximab (biosimilar)	2%	3%	4%	5%	5%	5%	2%	3%	4%	5%	5%	5%
Ixekizumab	16%	15%	18%	18%	18%	18%	16%	18%	22%	22%	22%	22%
Secukinumab	17%	13%	10%	9%	8%	7%	17%	14%	13%	14%	14%	14%
Tofacitinib	0%	2%	3%	3%	3%	3%	0%	2%	3%	3%	3%	3%
Upadacitinib	7%	5%	4%	4%	4%	4%	7%	5%	4%	4%	4%	4%

Note: The 2023 market share are based on observed sales in France (IQVIA Data); [a]the market shares for a nr-axSpA and r-axSpA are assumed to be identical.

Table 2 Cost and clinical inputs

	PsA	axSpA	
Target population [4,5], n	12,250	7,800	
Share of patients in Inadequate Response [8]	38%	38%	
PsA patients with PsO [9]	85%	-	
Annual discontinuation rate [10,7]	31.2%	r-axSpA	nr-axSpA
		5.0%	11.0%
	Cost		
Adverse event			
Serious infection[a]	€6,200.17		
Monitoring			
Rheumatologist consultation[b]	€29.50		
Biological check-up[c]	€21.38		
Chest X-ray[d]	€22.00		
Administration[e]			
Intravenous	€501.51		
Subcutaneous	€6.00		
Indirect costs[f] [11]	PsA	axSpA	
	€12,595.67	€15,367.20	

Technology	Cost per pack[g]	Pack size	Dose (mg) per unit	Response rate[l] [6,7] PsA ACR50	r-axSpA ASAS 40	nr-AxSpA ASAS 40
Bimekizumab	€1,678.83	2	160	41.6%	44.4%	44.2%
Adalimumab	€503.49	2	40	26.2%	42.3%	49.7%
Adalimumab (biosimilar)	€422.34	2	40	26.2%	42.3%	49.7%
Certolizumab pegol	€637.05	2	200	24.9%	44.0%	60.2%
Etanercept	€482.01	4	50	28.0%	50.4%	40.7%
Etanercept (biosimilar)	€472.65	4	50	28.0%	50.4%	40.7%
Golimumab	€633.22	1	50	21.7%	41.9%	50.8%
Guselkumab	€1,796.25	1	100	16.2%	-	-
Infliximab	€112.85	1	100	26.7%	49.9%	44.2%
Infliximab (biosimilar)	€190.40	1	120	26.7%	49.9%	44.2%
Ixekizumab[i]	€877.36	1	80	26.7%	49.9%	44.2%
Risankizumab	€2,481.85	1	150	26.5%	-	-
Secukinumab[j]	€948.25	2	150	27.1%	41.3%	28.6%
Tofacitinib	€672.57	56	5	24.2%	42.7%	44.2%
Ustekinumab[k]	€1,856.94	1	45	20.5%	-	-
Upadacitinib	€626.50	28	15	36.2%	49.4%	39.8%

[a]Serious infections were selected based on KOL opinion, the cost was based on the relevant DRG. [b]three visit during the first year, two in the subsequent years. [c]biological check up exams were validated by a clinical expert.[d]based on French tariffs. [e]Administration costs for IV injections are based on the relevant DRG alongside the cost of transport. Subcutaneous injection are based on the cost for an at-home nurse visit for a subcutaneous injection (1st injection only). [f]Indirect costs were estimated using the human capital approach and considered the cost sick leave and work disability of patients. They were only applied to non responders in a scenario analysis, additionally only patients of working age had indirect costs applied to them (based on the age distribution of the BE MOBILE 1 and 2 trials). [g]Value Added Tax (VAT) is included in the unit costs of the treatment, latest available price from the BdM_IT were used. [h]Based on the French average weight of 74 kg. [i]14% of patients are receiving an escalated annual maintenance dose. [j]74% of patients are receiving an escalated annual maintenance dose. [k]55% of patients are receiving an escalated annual maintenance dose. [l]if no data is available for a comparator, it was assumed to be as effective as bimekizumab.

AE: Adverse event; **ACR50**: American College of Rheumatology score**; ASAS40**: Assessment of SpondyloArthritis International Society score; **BDMIT**: Base des médicaments et informations tarifaires; **b/ts DMARD**: biologic or synthetic Disease-modifying anti-rheumatic drug; **Cum**: Cumulative; **DRG**: Diagnosis related groups**; IL**: Interleukin; **IR**: Inadequate response; **Ig**: Immunoglobulin; **JAKi**: Janus kinase inhibitors; **nr-axSpA**: Non-Radiographic Axial Spondyloarthritis; **PsA**: Psoriatic Arthritis; **PsO**: Psoriasis; **r-axSpA**: Radiographic axial Spondyloarthritis; **TNF**: Tumor necrosis factor; **VAT**: Value-added tax

Institutions: [1] ESSEC, Paris, France; [2] UCB, Colombes, France; [3] IQVIA, Courbevoie, France; [4] UCB, Slough, UK; [5] UCB, Brussels, Belgium; [6] REMAP, Zug, Switzerland; [7] CHU de Tours, Tours, France;

References: **[1]** European Medicines Agency (EMA). Bimzelx: EPAR-Product information Available at: https://www.ema.europa.eu/en/medicines/human/EPAR/bimzelx. **[2]** Van Den Bosch F, Coates L. Clinical management of psoriatic arthritis. The Lancet. 2018 Jun;391(10136):2285–94. **[3]** Sieper J, Poddubnyy D. Axial spondyloarthritis. The Lancet. 2017 Jul;390(10089):73–84. **[4]** Avis CT - Bimzelx dans la spondylarthrite axiale. Available from: https://www.has-sante.fr/upload/docs/evamed/CT-20360_BIMZELX_SpAax_PIC_EI_AvisDef_CT20360.pdf. **[5]** Avis CT - Bimzelx dans le rhumatisme psoriasique Available from: https://www.has-sante.fr/upload/docs/evamed/CT-20359_BIMZELX_RP_PIC_EI_AvisDef_CT20359.pdf. **[6]** Mease PJ, Gladman DD, Merola JF, Nash P, Grieve S, Laliman-Khara V, et al. Comparative efficacy and safety of bimekizumab in psoriatic arthritis: a systematic literature review and network meta-analysis. Rheumatology. 2024 Jul 1;63(7):1779–89. **[7]** Deodhar A, Machado PM, Mørup M, Taieb V, Willems D, Orme M, et al. Comparative efficacy and safety of bimekizumab in axial spondyloarthritis: a systematic literature review and network meta-analysis. Rheumatology. 2024 May 2;63(5):1195–205. **[8]** Haberhauer G, Strehblow C, Fasching P. Observational study of switching anti-TNF agents in ankylosing spondylitis and psoriatic arthritis versus rheumatoid arthritis. Wien Med Wochenschr. 2010;160(9-10):220-4. **[9]** S. Jara and S. Newmark "Psoriasis and Psoriatic Arthritis: What's the Connection?" [Internet]. Available from: https://creakyjoints.org/living-with-arthritis/symptoms/psoriasis-and-psoriatic-arthritis-connection/. **[10]** Pina Vegas L, Penso L, Sbidian E, Claudepierre P. Influence of sex on the persistence of different classes of targeted therapies for psoriatic arthritis: a cohort study of 14 778 patients from the French health insurance database (SNDS). RMD Open. 2023 Dec;9(4):e003570. **[11]** Huscher D, Merkesdal S, Thiele K, Zeidler H, Schneider M, Zink A, et al. Cost of illness in rheumatoid arthritis, ankylosing spondylitis, psoriatic arthritis and systemic lupus erythematosus in Germany. Annals of the Rheumatic Diseases. 2006 Sep 1;65(9):1175–83.
Acknowledgements: The authors acknowledge Rémi Monnier, IQVIA, Courbevoie, France for his assistance during the model adaptation. The authors acknowledge Susana Lobo Berastegui, UCB, Madrid, Spain for publication coordination and Charlotte Evans, Costello Medical, Bristol, UK for editorial assistance. Medical writing was provided by IQVIA and funded by UCB. Editorial services were provided by Costello Medical and funded by UCB. This study was sponsored by UCB. All costs associated with development of this poster were funded by UCB.

Presented at ISPOR EU 2024 | 17–20 November | Barcelona, Spain