#PCR79

Measuring the caregiver burden in children, adolescents and young adults with Achondroplasia: Results from a prospective, cross-sectional, multi-country study

Due. C[1] Quinn, J[2] and Butt, T[1].

[1]BioMarin, London, United Kingdom; [2]Patient Centered Access Consulting Ltd., London, United Kingdom

Background

Achondroplasia is a skeletal dysplasia and the most common cause of disproportionate short stature affecting more than 250,000 individuals globally[1]

- Achondroplasia is characterized by disproportionate short stature, with standing height often below the fifth percentile[1]
 - Throughout life, individuals with achondroplasia encounter a wide range of medical complications [2] a noticeable impacts on their health-related quality of life (HRQoL)3,4
- To date, studies that explore the full extent of caring for an individual with achondroplasia have been limited and focused on qualitative insights.
- Until recently, no disease modifying treatments have been available for achondroplasia. Vosoritide (Voxzogo™), a CNP analogue, targets overactive FGFR3 to stimulate endochondral bone growth. In infants and children with achondroplasia, vosoritide significantly improves growth from birth, resulting in sustained increases in height over time[5]
- Given the ever-shifting treatment landscape, it is important to better understand the impact of caring for a child with achondroplasia on the caregiver
- This study quantitatively assessed caregiver burden and explored whether it is impacted by the relative height vs. population mean (height z-score) of the person with achondroplasia.

Table 1. Participant Demographics

Characteristic/Concept	n (%) [a]
Age of the caregiver	
n (missing)	53 (0)
Mean (SD)	41.09 (8.34)
Median	40
Min - Max	29.00 - 62.00
Have you or the other primary/co-primary caregiver (i.e., child's other parent) also been diagnosed with achondroplasia?	
No, neither of the caregivers have achondroplasia	48 (90.6%)
Yes, I have achondroplasia	2 (3.5%)
Yes, the other caregiver has achondroplasia	3 (5.7%)
What is your current marital status?	
Single	2 (3.8%)
Divorced/separated	12 (22.6%)
Partnership/Co-habiting	36 (67.9%)
Married/De facto relationship	3 (5.7%)
Age of whom you are caring	
Missing	5 (9.4%)
< 3 years	12 (22.6%)
3-8 years	20 (37.7%)
> 8 years	16 (30.2%)

[a] Missing data included in calculation of percentages

Methods

An international, prospective, cross-sectional, non-interventional study.

- Participants were recruited from Australia, Austria and Sweden via patient advocacy groups (PAGs), recruitment agencies (Austria and Sweden) or clinical sites (Australia).
- An online survey captured socio-demographics, medical history, health state (EQ-5D-5L), well-being (SF-12), work productivity (WPAI:SPH), and impacts of caregiving on the family via the PedsQL Family Impact Module (PedsQL FM).
- Data was summarized using descriptive statistics. Continuous variables are presented as n, mean, standard deviation (SD), median, interquartile range (IQR), and minimum/maximum values. Categorical variables are presented as frequencies (n) and percentages (%).

Figure 1. EQ-5D 5L mean visual analogue scale (VAS) scores by Height Z-Score

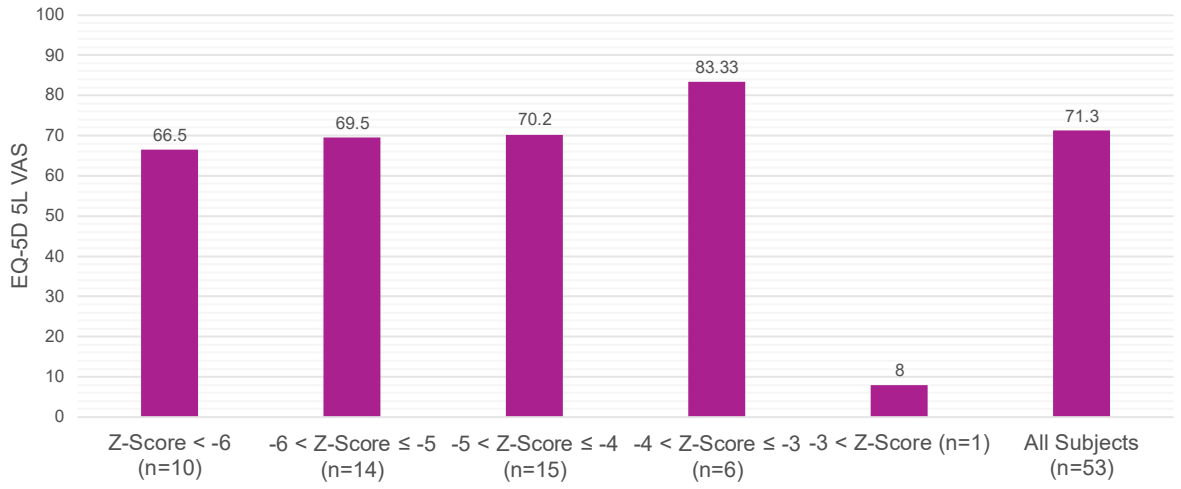
EQ-5D 5L VAS
100
90
80
70
60
50
40
30
20
10
0
66.5
69.5
70.2
83.33
8
71.3
Z-Score < -6 (n=10)
-6 < Z-Score ≤ -5 (n=14)
-5 < Z-Score ≤ -4 (n=15)
-4 < Z-Score ≤ -3 (n=6)
-3 < Z-Score (n=1)
All Subjects (n=53)

Results

- A total of 53 participants were enrolled from Austria (n=8), Australia (n=2) and Sweden (n=43) with a mean (SD) age of 41.09 (8.34).
- The majority of patients were married (36 (67.9%) with neither the participant or their spouse having a diagnosis of achondroplasia (90.6%, n=48).
- 67.9% (n=36) of participants were working full time. However, since the person they are caring for was diagnosed with achondroplasia:
 - 26.4 (n=14) have had to reduce their working hours; and
 - 35.8% stated their household income had decreased
- Work productivity was also impacted with mean (SD) WPAI:SHP overall productivity loss score of 33.91% (29.06).
 - Participants caring for a child or adolescent with a height z-score of Z-Score < -6 (n=10) reported mean (SD) absenteeism and presenteeism scores of 21.60 (22.18) and 32.86 (34.02) respectively, and overall work productivity loss of 47.36 (39.39).
- All Z-Score groups reported EQ-5D 5L UIS lower than the mean (SD) population norm of 0.91 (0.100)[6].
- Participants reported the lowest HRQoL in the SF-12 'Role, Emotional', Mental Health and General Health domains with mean (SD) of 42.89 (12.07), 43.49 (10.04) and 43.60 (10.36) respectively.
 - Mean Mental Component Score was 40.92 (11.79). When analyzed by height z-score, all groups reported MCS means below the population norm of 49.79 (SD=9.58)[7]
- The biggest PedsQL FM decrements in mean scores vs. population norms were see in the Communication and Worry domain where mean scores for all study subjects were respectively 24.7% and 29.5% lower than the mean for the general population.

Figure 2. SF 12 PCS and MCS by Height Z-Score vs. Population norms

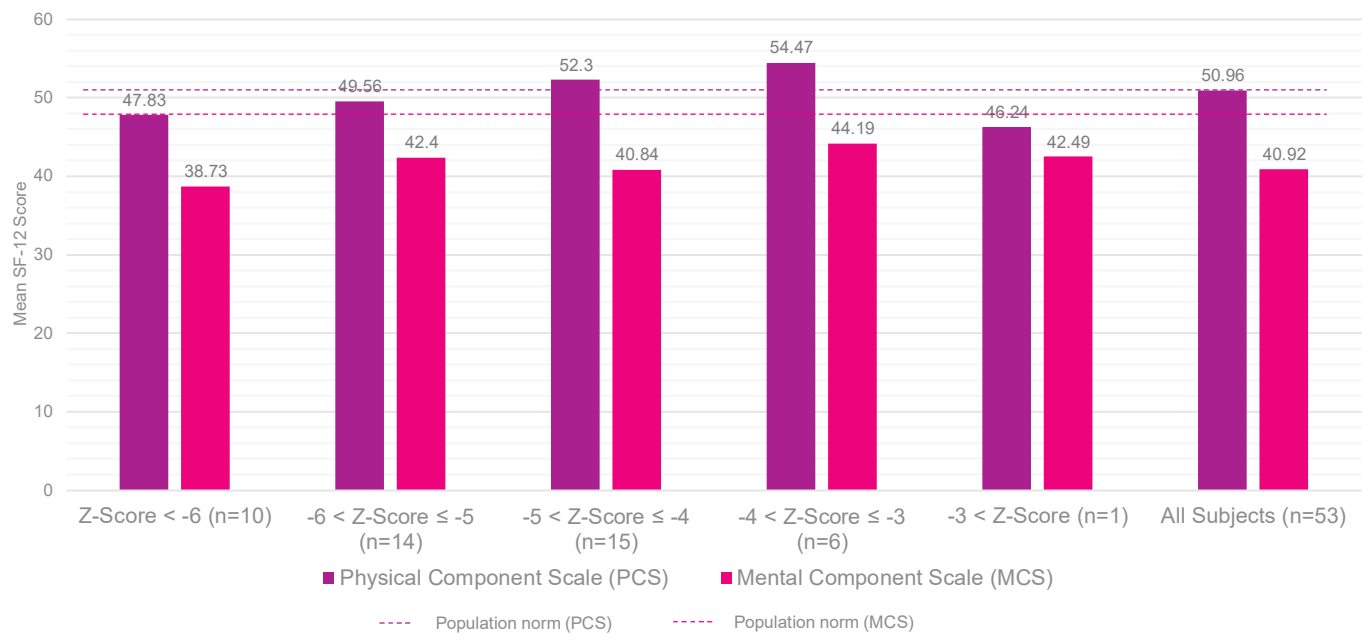
Mean SF-12 Score
60
50
40
30
20
10
0
47.83
38.73
49.56
42.4
52.3
40.84
54.47
44.19
46.24
42.49
50.96
40.92
Z-Score < -6 (n=10)
-6 < Z-Score ≤ -5 (n=14)
-5 < Z-Score ≤ -4 (n=15)
-4 < Z-Score ≤ -3 (n=6)
-3 < Z-Score (n=1)
All Subjects (n=53)
Physical Component Scale (PCS)
Mental Component Scale (MCS)
Population norm (PCS)
Population norm (MCS)

Figure 3. PedsQL FM mean score difference from population norm

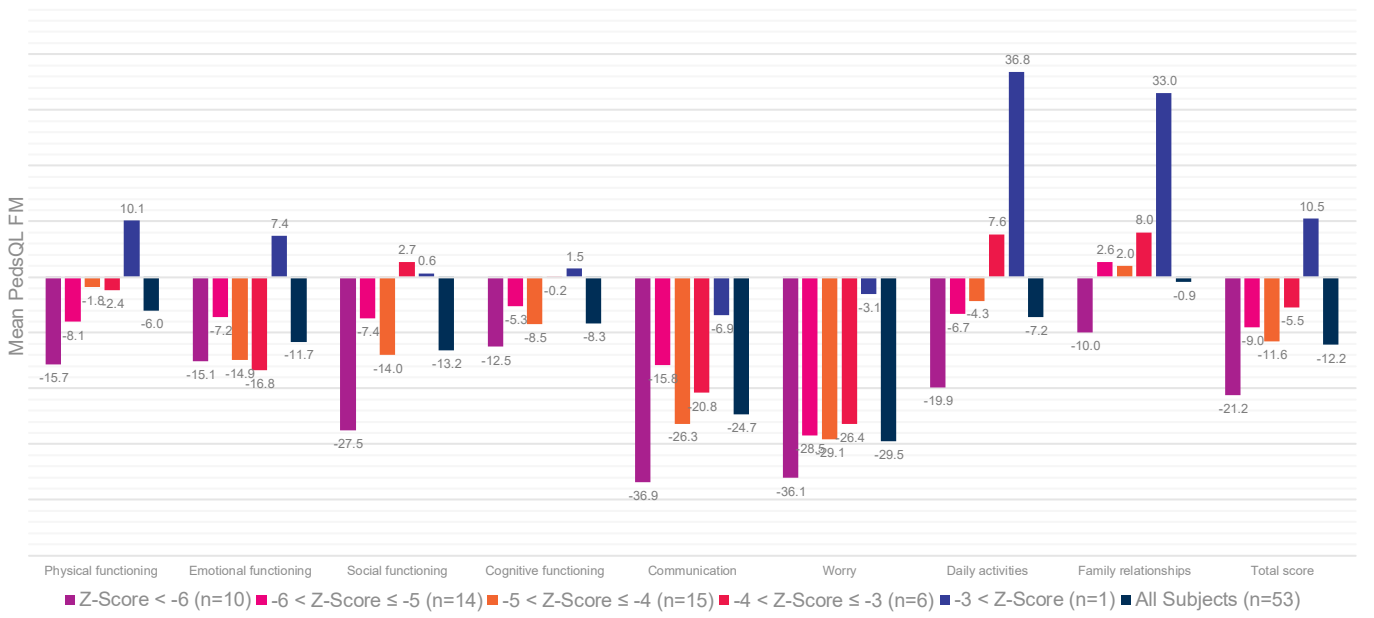
Mean PedsQL FM
-15.7
-8.1
-1.8
2.4
10.1
-6.0
-15.1
-7.2
-14.9
-16.8
7.4
-11.7
-27.5
-7.4
-14.0
2.7
0.6
-13.2
-12.5
-5.3
-8.5
-0.2
1.5
-8.3
-36.9
-20.8
-26.3
-6.9
-24.7
-36.1
-3.1
-26.4
-29.5
-19.9
-6.7
-4.3
7.6
36.8
-7.2
-10.0
2.6
2.0
8.0
33.0
-0.9
-21.2
-9.0
-11.6
-5.5
10.5
-12.2
Physical functioning
Emotional functioning
Social functioning
Cognitive functioning
Communication
Worry
Daily activities
Family relationships
Total score
Z-Score < -6 (n=10)
-6 < Z-Score ≤ -5 (n=14)
-5 < Z-Score ≤ -4 (n=15)
-4 < Z-Score ≤ -3 (n=6)
-3 < Z-Score (n=1)
All Subjects (n=53)

Conclusions

- Participants reported reduced working hours and a drop of income as a result of caregiver responsibilities.
- When grouped by height z-score, caregiver burden increased as height z-scores moved further away from population norms.
- None of the participants' children were being treated with Vosoritide (Voxzogo™) at the time of the study.
- These results describe the burden of caring for a person with achondroplasia on the caregiver's health, wellbeing, financial and employment status and demonstrate the relationship between height z-scores and caregiver outcomes.

References: 1. Pauli RM. Achondroplasia: a comprehensive clinical review. Orphanet J Rare Dis. 2019;14(1):1.. **2.** Horton WA, Hall JG, Hecht JT. Achondroplasia. Lancet. 2007;370(9582):162-72. **3.** Alade Y, Tunkel D, Schulze K, McGready J, Jallo G, Ain M, et al. Cross-sectional assessment of pain and physical function in skeletal dysplasia patients. Clin Genet. 2013;84(3):237-43. **4.** Maghnie M, Semler O, Guillen-Navarro E, Selicorni A, Heath KE, Haeusler G, et al. Lifetime impact of achondroplasia study in Europe (LIAISE): findings from a multinational observational study. Orphanet J Rare Dis. 2023;18(1):56. **5.** Savarirayan R, Tofts L, Irving M, Wilcox W, Bacino CA, Hoover-Fong J, et al. Once-daily, subcutaneous vosoritide therapy in children with achondroplasia: a randomised, double-blind, phase 3, placebo-controlled, multicentre trial. Lancet.**6.** Teni FS, Gerdtham UG, Leidl R, Henriksson M, Astrom M, Sun S, et al. Inequality and heterogeneity in health-related quality of life: findings based on a large sample of cross-sectional EQ-5D-5L data from the Swedish general population. Qual Life Res. 2022;31(3):697-712. 2020;396(10252):684-92. **7.** Mols F, Pelle AJ, Kupper N. Normative data of the SF-12 health survey with validation using postmyocardial infarction patients in the Dutch population. Qual Life Res. 2009;18(4):403-14.

Acknowledgments: We thank Louise Tofts for recruitment in Australia, Föreningen för Kortvuxna DHR – FKV for recruitment in Sweden and Bundesverband kleinwüchsiger Menschen und ihrer Familien – BKMF for recruitment in Austria

Disclosures: This research was funded by Biomarin Pharmaceutical Inc.

Presented at the ISPOR 2024 Annual Meeting: 18-20 November, Barcelona, Spain

©2024 BioMarin Pharmaceutical Inc. All rights reserved.