We introduce a unified and extensible framework for estimating **QALYs**, providing more **appropriate**, **robust**, and **accurate** estimates to be used in cost-effectiveness modelling.

MSR73

A Framework for Estimating Quality Adjusted Life Years Using Joint Models of Longitudinal and Survival Data

Michael J. Crowther[1,*], Alessandro Gasparini[1], Sara Ekberg[1], Federico Felizzi[2], Elaine Gallagher[3], Noman Paracha[3]

Objectives

Length and quality of life (utility score) are frequently combined in health technology assessments to derive a single, generic measure of health improvement resulting from a health care intervention. One such measure is given by quality-adjusted life years (QALY), typically calculated by assuming discrete health states and quality of life values.

Accurate estimation of QALYs is crucial for informed decision-making in healthcare policy and resource allocation: however, traditional methods often rely on assumptions that are sometimes, if not always, biologically and statistically inappropriate. For instance, quality of life data is often modelled as a time-varying covariate, changing value only when a new measurement is recorded: this is usually implausible. Moreover, this data suffers from measurement error, and it is necessary to account for the correlation between observations recorded on the same patient.

We aim to develop a framework for estimating QALYs that addresses these limitations.

Methods

We begin by fitting a multivariate joint model for survival, quality of life Q, and an ancillary longitudinal biomarker Y:

$$\begin{cases} h(t) = h_0(t)e^{X^h\gamma + \alpha^q g^q(Q) + \alpha^b g^b(Y)} \\ Q(t) = X^q\beta^q + Z^q b^q + \varepsilon^q \\ Y(t) = X^b\beta^b + Z^b b^b + \varepsilon^b \end{cases}$$

with outcome-specific covariates X^h, X^q, X^b, random effects Z^q, Z^b, and association structures g^q, g^b linking the submodels. Usual assumptions of joint longitudinal survival models apply for the remaining model components.

A lifetime QALY measure in continuous time is defined as:

$$QALY = \int_0^\infty Q(u)S(u)du$$

where Q and S denote quality of life and survival estimated from the joint model.

Similarly, a restricted QALY measure that does not rely on extrapolation (beyond a certain time point t) is defined as:

$$QALY(t) = \int_0^t Q(u)S(u)du$$

Both QALY and QALY(t) can be estimated from the joint model and rely on numerical integration techniques. Marginal and conditional versions of each measure can be defined, referring to, e.g., overall and treatment-specific estimates.

We illustrate the new methodology in practice using a synthetic prostate cancer trial dataset with overall survival as the time to event and quality of life and prostate-specific antigen measurements as the longitudinal outcomes.

Figure 1: Estimated, population-level trajectories of quality of life (utility score) from the joint model.

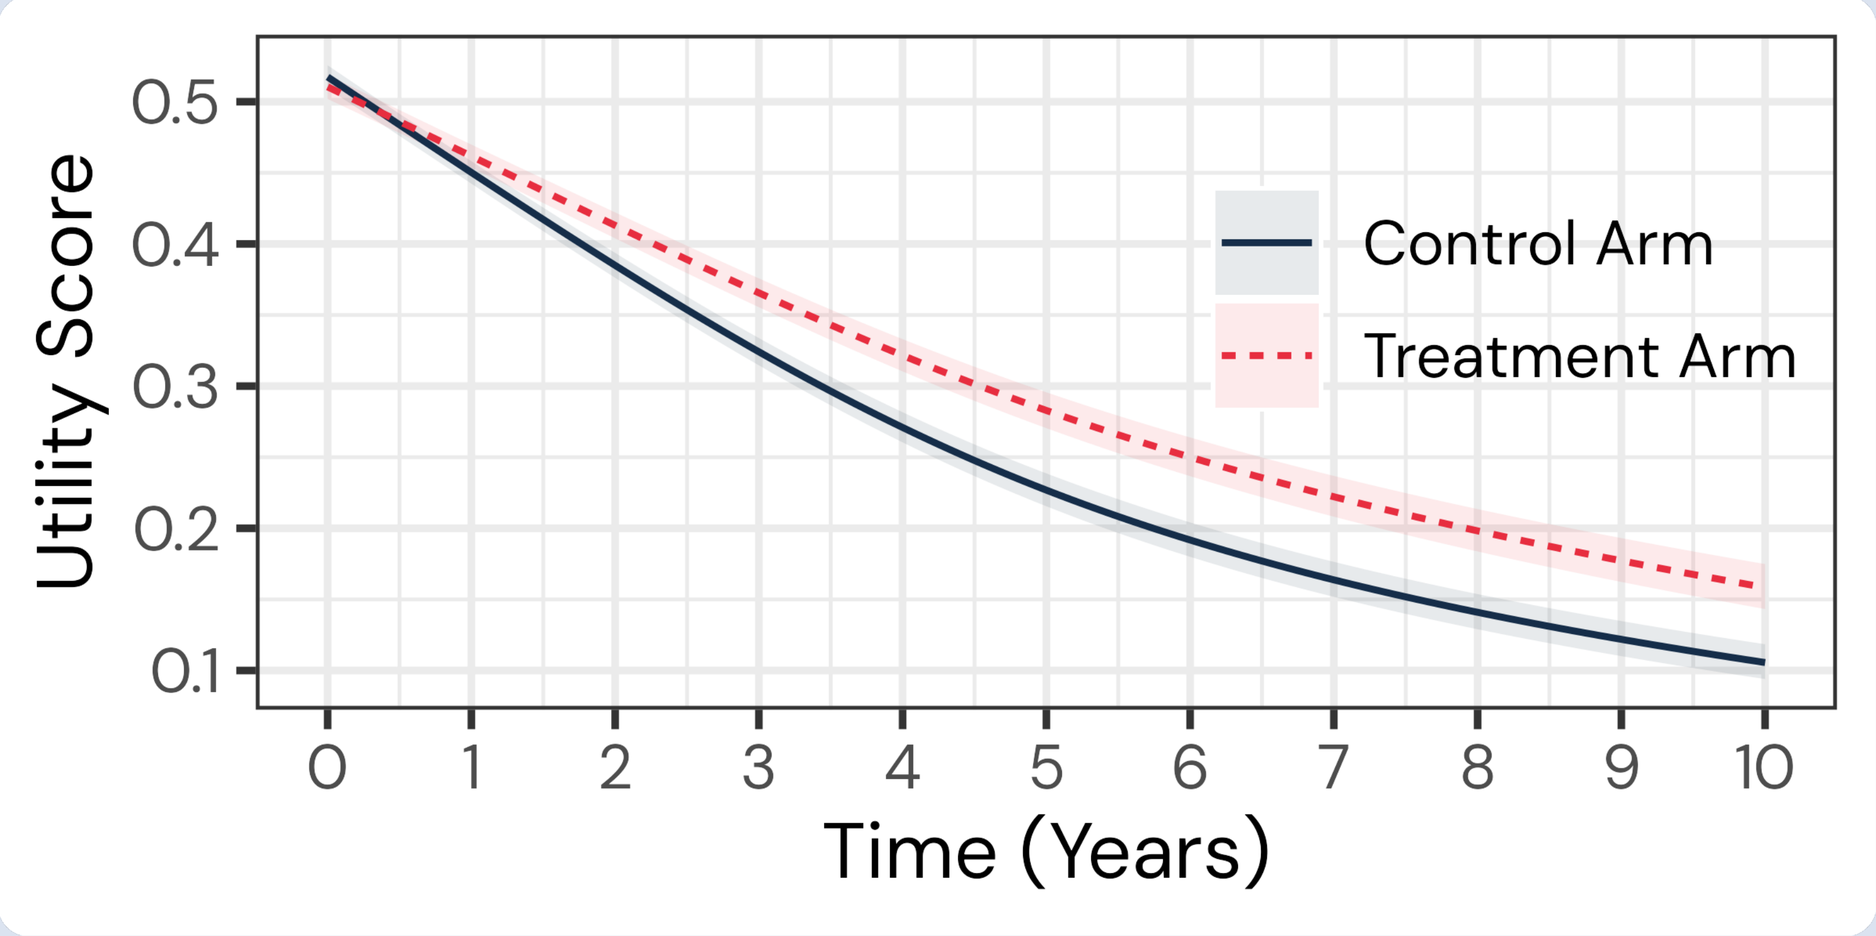
Utility Score
Time (Years)
Control Arm
Treatment Arm

Figure 2: Estimated standardised survival difference between the treatment and control arm.

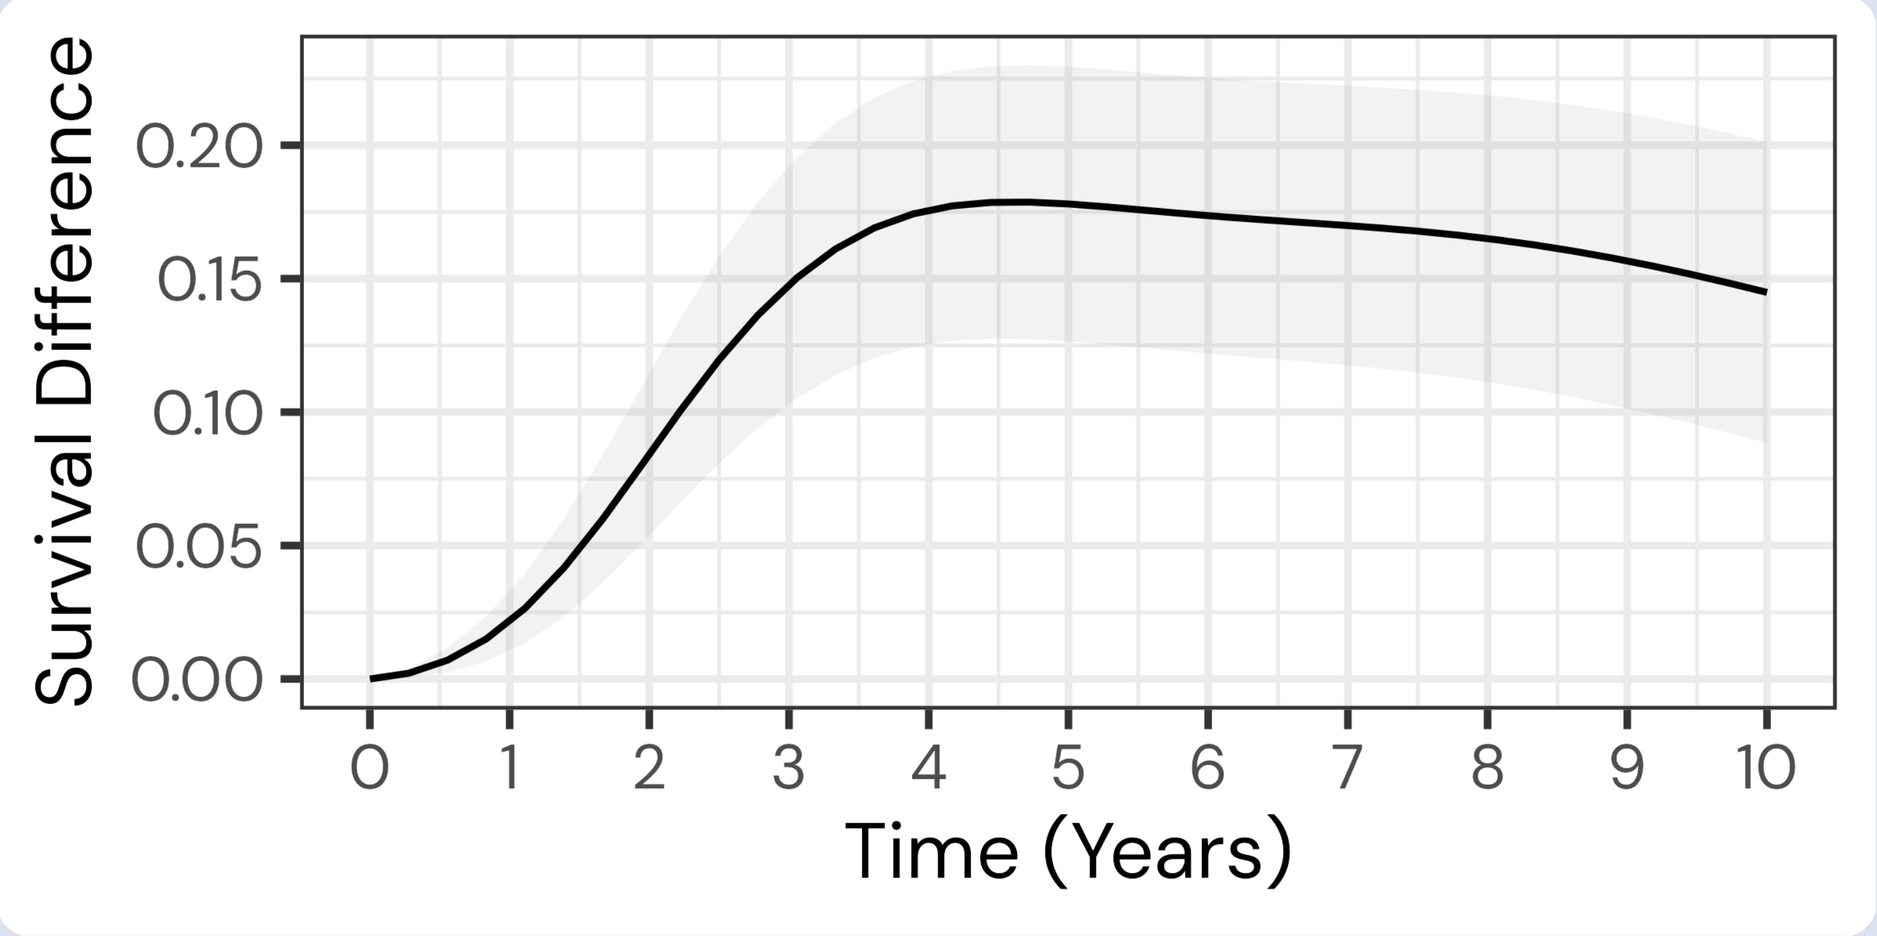
Survival Difference
Time (Years)

Figure 3: Estimated restricted QALYs by treatment arm and over time.

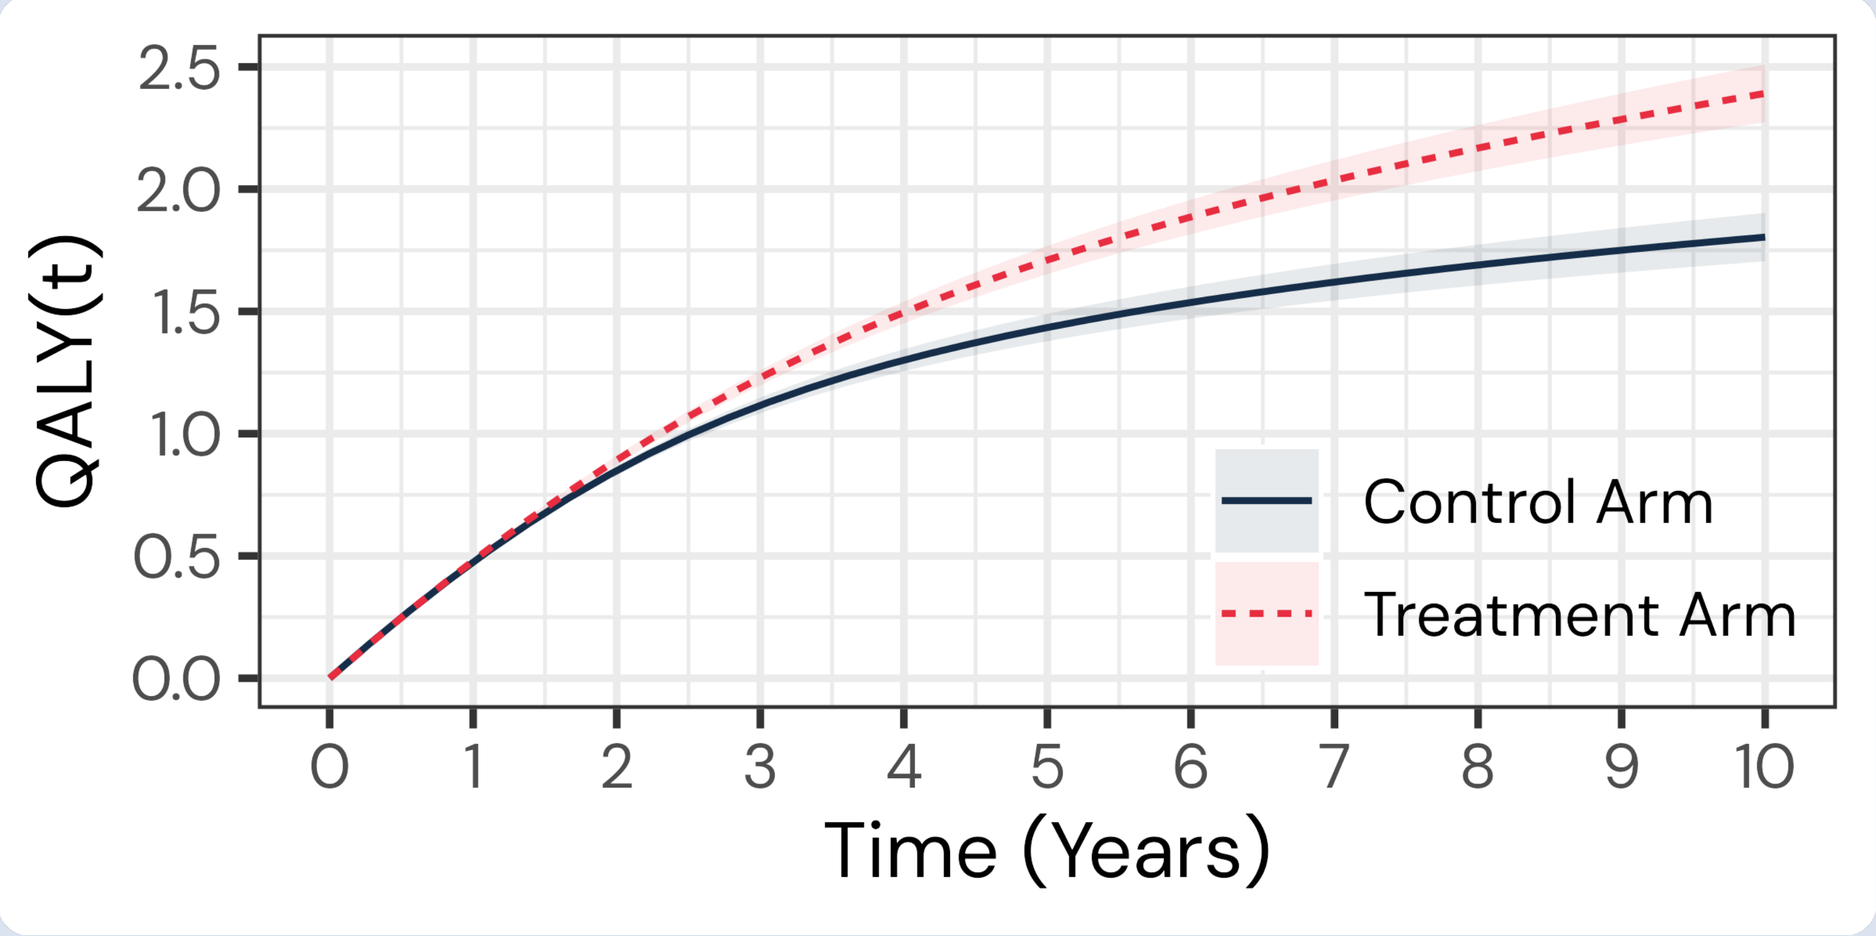
QALY(t)
Time (Years)
Control Arm
Treatment Arm

Figure 4: Difference in estimated restricted QALYs over time between the treatment and control arm.

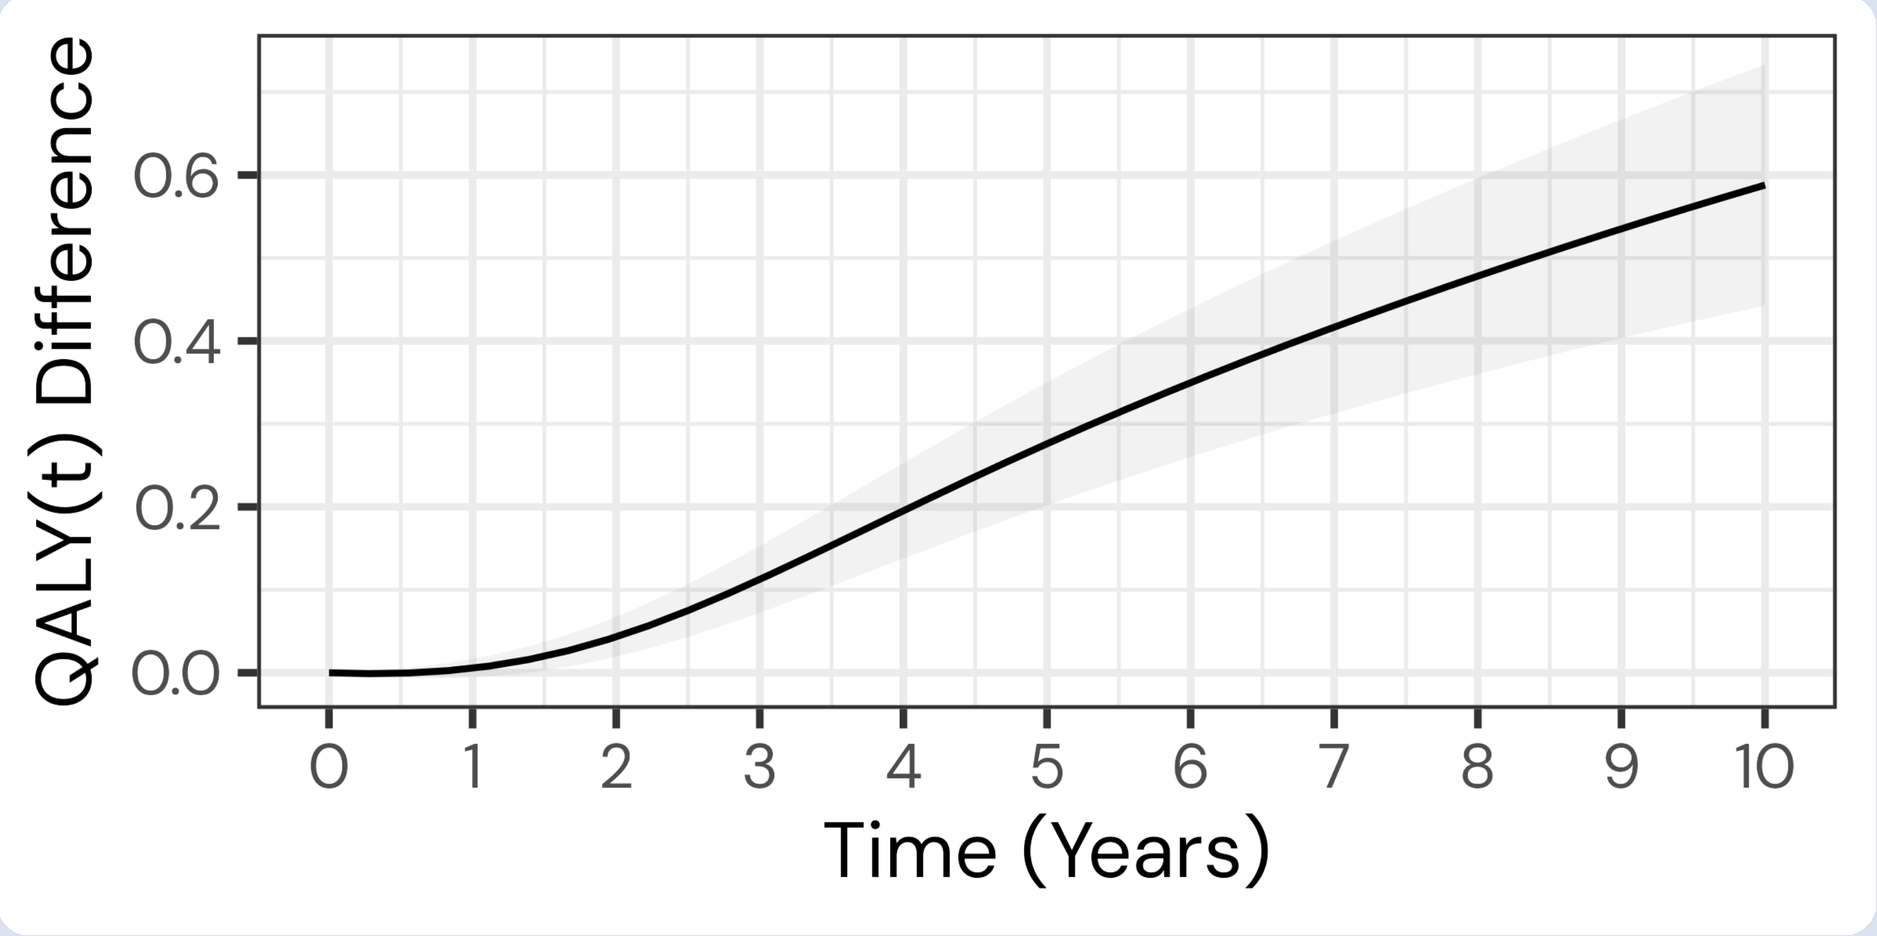
QALY(t) Difference
Time (Years)

Results

We could estimate from the joint model both population-level, marginal quality of life trajectories by treatment arm and over time (Figure 1) and differences between treatment arms in standardised survival probability (Figure 2). The former highlights that the experimental treatment is associated with improved quality of life over time (as compared with control), while the latter highlights a significant survival benefit of the treatment, peaking at about 18% after 4 years of follow-up.

We estimated 7.29 lifetime QALYs (95% C.I.: 4.02, 10.56) in the treatment arm and 4.23 (3.37, 5.10) in the control arm, for a difference of 3.06 (-0.20, 6.31) QALYs. Moreover, Figures 3 and 4 illustrate restricted QALYs by treatment arm, over time, and differences thereof. These predictions of restricted QALYs start to diverge after one year of follow-up, showing an increased benefit of the treatment (in terms of QALYs) increasing up to 0.6 QALYs after 10 years of follow-up. Overall, both lifetime and restricted QALYs highlight once again a significant benefit of the experimental treatment.

Conclusion

We have developed a new approach for the estimation of QALYs, based on multivariate joint models for longitudinal and survival data. The methodology provides a unified framework for modelling all components of QALYs jointly, incorporating all sources of heterogeneity and accounting for all the statistical and biological intricacies of the data. Disease burden can be accommodated as well, and further extension, e.g., to incorporate costs are also possible. Moreover, we focus on cancer, but the methodology is general in nature and can be applied in other disease areas as well.

In conclusion, the framework has the potential to enable more timely, accurate, and robust decision-making, significantly impacting clinical practice and health policy.

1. Red Door Analytics AB, Stockholm, Sweden
2. Department of Computer Science, ETH Zurich, Switzerland
3. Bayer Pharmaceuticals AG, Basel, Switzerland

* E-mail: <michael.crowther@reddooranalytics.se>

reddooranalytics.se
linkedin.com/company/red-door-analytics
youtube.com/@RedDoorAnalytics

Red Door Analytics