A Systematic Review of Pharmacological Interventions for Hidradenitis Suppurativa: A Focus on Clinical Response, Quality of Life, and Safety

srs HEOR evidence to access

Bisen R, Thakur L, Shivsingwale G, Gunde D, Mukta Y

SRS HEOR (A Division of SRS Pharmaceuticals Pvt. Ltd., Mumbai, India)

CO66

OBJECTIVES

- Hidradenitis Suppurativa (HS) also known as acne inversa, is a condition that causes small painful nodules, lumps, abscesses, and tunnelling in areas where skin rubs together, such as the armpits, groin, buttocks, and breasts.
- This systematic review aimed to evaluate the efficacy and safety of pharmaceutical therapies for mild, moderate, and severe HS patients.

METHODS

- A comprehensive searches of MEDLINE®, Embase®, Evidence-based Medicine Reviews, and grey literature were conducted.
- All records were screened against predefined inclusion criteria (**Table 1**).
- Bibliographic lists of relevant SLRs were also conducted.
- All included studies were extracted and evaluated using NICE's critical appraisal checklist.

Table 1: Eligibility criteria

Population	**Patients diagnosed with mild/ moderate/ severe HS**
Intervention	All approved or investigational pharmacological interventions used for the treatment of HS
Comparator	▪ Placebo ▪ BSC (author defined) ▪ Any other pharmacological/non-pharmacological intervention
Outcome	▪ Clinical response (HiSCR, HiSCR50, HiSCR75, HiSCR90, HiSCR100) ▪ DLQI ▪ Pain score ▪ Severity outcomes ▪ Incidence of adverse events ▪ Study/treatment discontinuation
Study design	▪ RCT ▪ SLR*
Language	English language

Key: BSC, best supportive care; DLQI, dermatological quality of life index; HiSCR, hidradenitis suppurativa clinical response; HS, hidradenitis suppurativa; MA, meta-analysis; RCT, randomized controlled trial; SLR, systematic literature review
* Bibliographies of existing systematic reviews were reviewed to ensure that all relevant studies were identified for inclusion in the SLR

RESULTS

Figure 1: PRISMA diagram of included and excluded publications

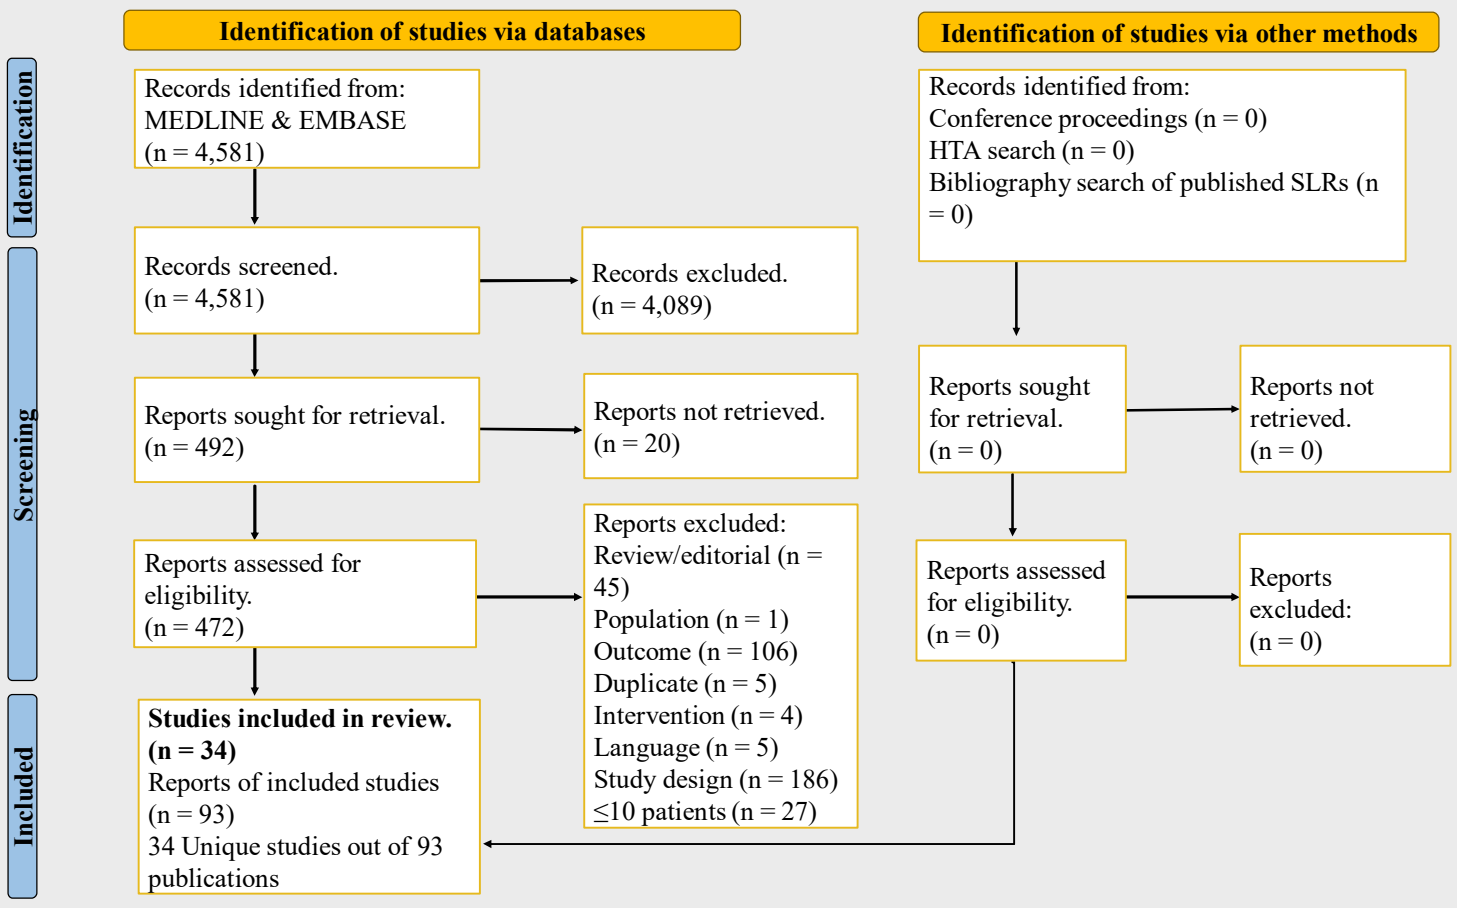
Identification of studies via databases
Identification of studies via other methods
Identification
Screening
Included
Records identified from: MEDLINE & EMBASE (n = 4,581)
Records screened. (n = 4,581)
Records excluded. (n = 4,089)
Reports sought for retrieval. (n = 492)
Reports not retrieved. (n = 20)
Reports assessed for eligibility. (n = 472)
Reports excluded: Review/editorial (n = 45) Population (n = 1) Outcome (n = 106) Duplicate (n = 5) Intervention (n = 4) Language (n = 5) Study design (n = 186) ≤10 patients (n = 27)
Studies included in review. (n = 34) Reports of included studies (n = 93) 34 Unique studies out of 93 publications
Records identified from: Conference proceedings (n = 0) HTA search (n = 0) Bibliography search of published SLRs (n = 0)
Reports sought for retrieval. (n = 0)
Reports not retrieved. (n = 0)
Reports assessed for eligibility. (n = 0)
Reports excluded: (n = 0)

- Of 4,581 publications screened, 34 studies were RCT which met inclusion criteria.
- Out of the 34 RCTs included in this SLR, 28 were double-blinded, 3 were triple-blinded, 2 were open-label, and 1 was assessor-blinded.
- PIONEER I and PIONEER II were pivotal trials for HS, as discussed in **(Table 2)**.

Table 2: Results of PIONEER I and PIONEER II

	PIONEER I [33]		PIONEER II [33]	
Study summery	• Phase 3, double-blind, multicentre • Length of follow up: 36 weeks		• Phase 3, double-blind, multicentre • Length of follow up: 36 weeks	
Population	**Adalimumab EW (N=153):** • Mean age: 37.8 years • Male: 40.5% • Female: 59.5% • BMI: 33 Kg/m2 • Race, white: 75.8% • Race, black: 21.6%	**Placebo (N=154):** • Mean age: 36.2 years • Male: 31.8% • Female: 68.2% • BMI: 34.5 Kg/m2 • Race, white: 76.6% • Race, black: 18.8%	**Adalimumab EW (N=163):** • Mean age: 34.9 years • Male: 33.7% • Female: 66.3% • BMI: 31.3 Kg/m2 • Race, white: 87.7% • Race, black: 5.5%	**Placebo (N=163):** • Mean age: 36.1 years • Male: 50% • Female: 69.3% • BMI: 32.9 Kg/m2 • Race, white: 79.8% • Race, black: 12.3%
Treatment	Adalimumab EW vs placebo (1:1)		Adalimumab EW vs placebo (1:1)	
HiSCR (%)	41.8%	26%	58.9%	27.6%
Change in mSS from baseline at week 12: Mean (95%CI)	-24.4 (-19.7 to 2.4)	-15.7	-28.9 (-28.6 to -10.1)	-9.5
Change in DLQI from baseline at week 12: Mean (95%CI)	-5.4 (-3.8 to -1.1)	-2.9	-5.1 (-4.1 to -1.5)	-2.3
Change in total AN count from baseline at week 12: Mean	-5.47	-2.81	-5.64	-2.24
Any AEs (week 12)	50.3%	58.6%	57.1%	63.2%
SAE (week 12)	1.3%	1.3%	1.8%	3.7%

Key: AE, adverse events; AN, abscesses and inflammatory nodules; BMI, body mass index; DLQI, dermatological quality of life index; EW, every weekly; HiSCR, hidradenitis suppurativa clinical response; mSS, modified sartorius score; SAE, serious adverse events

Hidradenitis Suppurativa Clinical Response (HiSCR):

- Adalimumab, anakinra, apremilast, avacopan, bimekizumab, doxycycline, guselkumab, IFX-1, MABp1, PF-06650833, PF-06700841, PF-06826647, povorcitinib, risankizumab, RIST4721, secukinumab, upadacitinib all reported notable clinical benefits, with HiSCR ranged from 10% (anakinra at week 24) to 88% (povorcitinib-90 mg at week 8).[7-8,10,13,17-18,21,24,26-29,31]
- At week 12, a significantly greater proportion of patients receiving Anakinra (78%; 7 of 9 patients) achieved a positive HiSCR outcome compared to the placebo group (30%; 3 of 10 patients) (p=0.04). This trend reversed by week 24, with similar proportions of patients achieving positive HiSCR in both the anakinra (10%; 1 of 10 patients) and placebo (33%; 3 of 9 patients) groups (p=0.28).[29]
- At week 8, HiSCR rates were observed as 56%, 56%, 88%, and 57% among patients treated with povorcitinib at doses of 30 mg, 60 mg, 90 mg, and placebo, respectively.[26]

Modified sartorius score (mSS):

- The mean change from baseline in mSS was -32 (SD, 9.5), -40.2 (SD, 9.8; p value [weekly vs placebo=0.097]), and -17.2 (SD, 9.8) for patients receiving adalimumab EOW, adalimumab EW, and placebo respectively. Difference between adalimumab EOW vs placebo was -14.8 (95% CI, -41.0 to 12.1) and between adalimumab weekly vs placebo was -22.0 (95% CI, -50.1 to 4.2).[12]
- In AURORA trial, at week 12, the mean change from baseline in mSS was higher in avacopan 30 mg BID than in avacopan 10 mg BID, and placebo {-14.4 (SD, 29.96) vs -12.6 (SD, 25.98) vs and -11.2 (SD, 35.69)} respectively.[27]

Total abscesses and inflammatory nodules (AN) count:

- Significant improvements were observed in mean change from baseline in AN count (range: -52.58 for PF-06700841 at week 16 to 60.3 for adalimumab at week 12).[10,12]

Draining fistula count:

- Similarly, significant improvements were observed in mean change from baseline in draining fistula count (range: -40.1 for spesolimab at week 12 to 31.1 for adalimumab at week 16).[1,12]

Skin pain:

- The mean change from baseline in skin pain at week 16 was (range: -43.88 for PF-06700841 to -0.1 for IFX-1 400 mg).[10,20]

Dermatological quality of life index (DLQI):

- The mean change from baseline in DLQI score was (range: -7.5 for PF-06700841 at week 16 to 5.6 for adalimumab weekly-EOW at week 52).[10,12]

Safety:

- A low rate of serious adverse events (SAEs) was observed in most of the studies.
- Only upadacitinib (12.2%) and secukinumab (10.5%) had more SAEs.[7,31]
- In contrast, no SAEs were detected in spesolimab or anakinra, while bimekizumab had similar safety profiles to adalimumab.[1,17,29]

Discontinuation:

- In the DETERMINE trial, at 68 weeks, 78.8% of patients in the risankizumab 180 mg→360 mg group discontinued treatment. Similarly, 85.2% of patients in the risankizumab 360 mg→360 mg group and 94.6% of patients in the placebo→risankizumab 360 mg group discontinued treatment. Discontinuations were due to various reasons, including adverse events (AEs), patient withdrawal, loss to follow-up, lack of efficacy, and other unspecified reasons.[18]
- None of the patients receiving spesolimab discontinued treatment, and anakinra demonstrated a very low rate of discontinuation.[1,29]

Figure 2: Results of study quality assessment using NICE critical appraisal checklist[34]

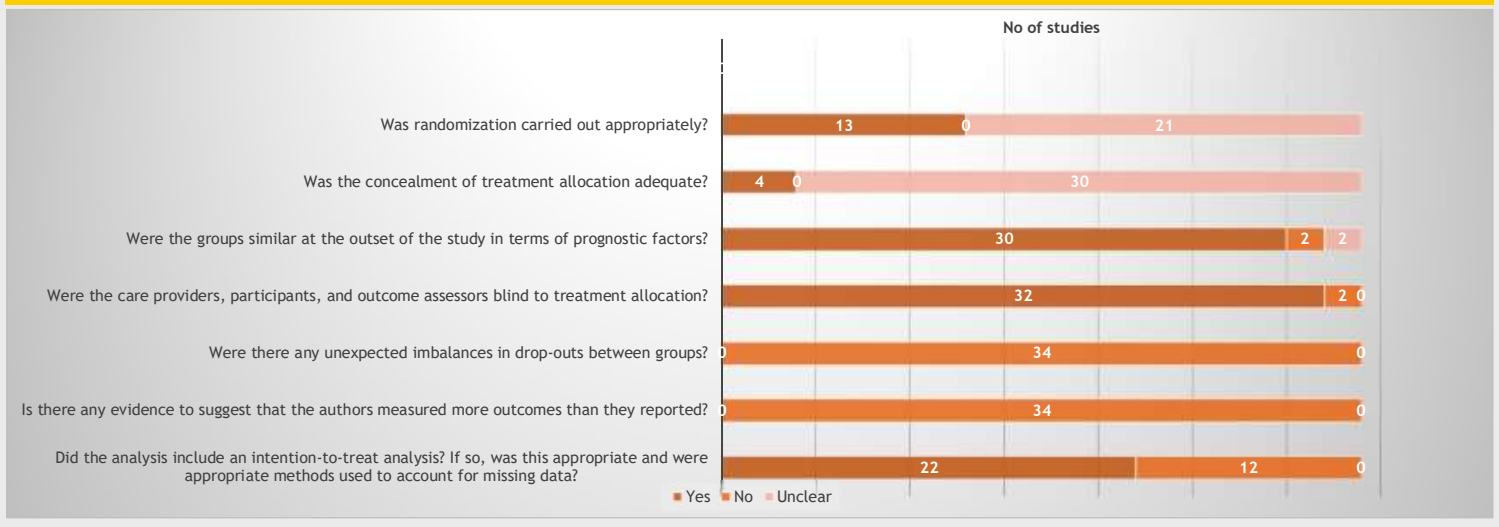
No of studies
Was randomization carried out appropriately? 13 0 21
Was the concealment of treatment allocation adequate? 4 0 30
Were the groups similar at the outset of the study in terms of prognostic factors? 30 2 2
Were the care providers, participants, and outcome assessors blind to treatment allocation? 32 2 0
Were there any unexpected imbalances in drop-outs between groups? 0 34 0
Is there any evidence to suggest that the authors measured more outcomes than they reported? 0 34 0
Did the analysis include an intention-to-treat analysis? If so, was this appropriate and were appropriate methods used to account for missing data? 22 12 0
Yes
No
Unclear

CONCLUSIONS

This systematic review highlights the significant advances in pharmacological therapies for the management of HS, particularly with treatments like adalimumab and povorcitinib showing promising clinical responses. While these therapies offer improved outcomes for patients with mild to severe HS, the review emphasizes the need for further research into long-term efficacy and safety. The findings support the development of personalized treatment strategies, aiming to enhance clinical response, quality of life, and management of adverse events, but there remains a critical need for continued exploration into novel therapeutic options for sustained HS care.

REFERENCES

1. Alavi A. et al. JAAD 2023; 89(3):AB89; 2. ClinicalTrials.gov identifier: NCT04988308, Accessed April 28, 2024; 3. Mortimer PS. Et al. BJD 1986; 115(3):263-268; 4. Miller I. et al. BJD 2011; 165(2):391-398; 5. Sayed CJ. ClinicalTrials.gov identifier: NCT02781818, Accessed April 28, 2024; 6. ClinicalTrials.gov identifier: NCT04493502, Accessed April 28, 2024; 7. ClinicalTrials.gov identifier: NCT04430855, Accessed April 28, 2024; 8. ClinicalTrials.gov identifier: NCT05348681, Accessed April 28, 2024; 9. ClinicalTrials.gov identifier: NCT04019041, Accessed April 28, 2024; 10. ClinicalTrials.gov identifier: NCT04092452, Accessed April 28, 2024; 11. Kimball AB. ClinicalTrials.gov identifier: NCT00722800, Accessed April 28, 2024; 12. Kimball AB. Et al. Annals of internal medicine 2012; 157(12):846-855; 13. Vossen ARJV. et al. JAAD 2019; 80(1):80-88; 14. Lee RA. ClinicalTrials.gov identifier: NCT01838499, Accessed April 28, 2024; 15. Grimstad O. et al. American journal of clinical dermatology 2020; 21(5):741-748; 16. Aarts P. et al. Dermatology 2023; 239(4):670-674; 17. Glatt S. et al. JAMA dermatology 2021; 157(11):1279-1288; 18. Kimball AB. Et al. Dermatology and therapy 2023; 13(5):1099-1111; 19. Kirby J.S. et al. JAAD 2023; 90(3) :521-529; 20. Zenker O. ClinicalTrials.gov identifier: NCT03487276, Accessed April 28, 2024; 21. Kontochristopoulos G. et al. Skin appendage disorders 2022; 8(6):476-481; 22. Hunger R. ClinicalTrials.gov identifier: NCT02421172, Accessed April 28, 2024; 23. Efthymiou O. Et al. Experimental dermatology 2022; 31, pp.71; 24. Kimball AB. et al. JEADV 2023; 37(10):2098-2108; 25. Grant A. et al. JAAD 2010; 62(2):205-217; 26. Alavi A. et al. BJD 2022; 186(5):803-813; 27. Kirby JS. Et al. JID 2021; 141(9):B19; 28. Kanni T. et al. JID 2018; 138(4):795-801; 29. Tzanetakou V. et al. JAMA Dermatology 2016; 152(1) 52-59; 30. ClinicalTrials.gov identifier: NCT03713632, Accessed April 28, 2024; 31. ClinicalTrials.gov identifier: NCT03713619, Accessed April 28, 2024; 32. Clemmensen OJ. Et al. International journal of dermatology 1983; 22(5):325-328; 33. Kimball AB. Et al. NEJM 2016; 375(5):422-434; 34. NICE, https://www.nice.org.uk/article/pmg24/chapter/4-Clinical-effectiveness#quality-assessment-of-the-relevant-randomised-controlled-trials, Accessed April 28, 2024.

Financial Support: This study was funded by SRS Pharmaceuticals Pvt Ltd, Mumbai, India

Poster presented at the ISPOR EU 2024; 17-20 November 2024; Barcelona, Spain