Hospital Healthcare Resource Utilization in People with CF Treated with Elexacaftor/Tezacaftor/Ivacaftor in Portugal: a Non-interventional, Retrospective Study

Carl Baxter[1], Gabriela Vega-Hernandez[1], Ana Ordaz[1], Heike Wöhling[1], Margarida Bártolo[2], Margarida Ribeiro[2], João Raposo[2], Humberto Melo[2], Pilar Azevedo[3]

[1] Vertex Pharmaceuticals Incorporated, Boston, MA, USA; [2] HMR – Health Market Research, Sintra, Portugal;
[3] Hospital de Santa Maria, Centro Hospitalar Lisboa Norte EPE, and Universidade de Lisboa, Lisboa, Portugal

Poster EE543

BACKGROUND

- The safety and efficacy of elexacaftor/tezacaftor/ivacaftor (ELX/TEZ/IVA) have been demonstrated in clinical trials in people with cystic fibrosis (CF)[1-6]
- Safety and effectiveness have also been demonstrated in real-world studies in people with CF aged ≥12 years in several countries, including Germany,[7] France,[8] and the US[9-11]
- ELX/TEZ/IVA was granted marketing authorization by the European Commission in August 2020, and became available in Portugal first through Exceptional Use Authorization and then reimbursement, which now covers people with CF aged ≥6 years who have at least one *F508del* variant (*F/any* genotypes)
- We conducted a real-world evidence study in people with CF receiving ELX/TEZ/IVA in Portugal that evaluated healthcare resource utilization (HCRU) and prescription medications using the Health Market Research (HMR) consumption database

METHODS

Study Design

Objective: To describe the impact of ELX/TEZ/IVA on HCRU before and after treatment initiation among people with CF in Portugal

Design: Observational, retrospective study using data from the HMR hospital medication consumption database

Inclusion Criteria (at Index[a])

- Initiation with ELX/TEZ/IVA between August 21, 2020, and June 30, 2022
- At least 12 months of data availability prior to ELX/TEZ/IVA initiation
- At least 12 months of data availability post ELX/TEZ/IVA initiation and on ELX/TEZ/IVA treatment

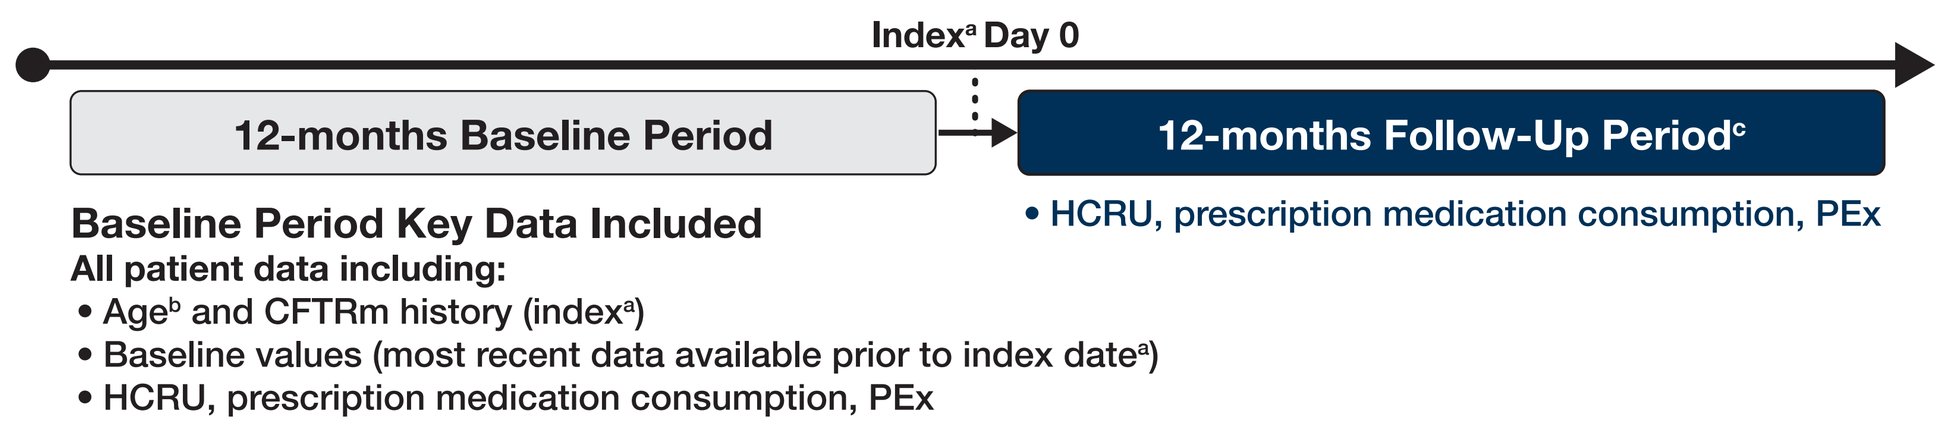
Index[a] Day 0
12-months Baseline Period
12-months Follow-Up Period[c]
Baseline Period Key Data Included
All patient data including:
• Age[b] and CFTRm history (index[a])
• Baseline values (most recent data available prior to index date[a])
• HCRU, prescription medication consumption, PEx
• HCRU, prescription medication consumption, PEx

CFTRm, cystic fibrosis transmembrane conductance regulator modulator; **ELX/TEZ/IVA,** elexacaftor/tezacaftor/ivacaftor; **HCRU,** healthcare resource utilization; **PEx,** pulmonary exacerbation (proxied by IV antibiotic treatment episodes)

[a] Defined as the ELX/TEZ/IVA initiation date;
[b] The specific age of each patient is not provided. Contained within the database are age intervals (0-14y, 15-39y, 40-59y, 60-79y, ≥80y) and for this study all the patients from these age intervals were considered for the analysis;
[c] Follow-up outcomes were collected from hospital consumption data

Data analysis and definitions used in this study

- Age interval* (0-14y, 15-39y, 40-59y, 60-79y, ≥80y)
- CFTRm history (yes/no)
- Hospital HCRU – Episode type and specialty
 - Outpatient Visit
 - Hospitalization/Inpatient
 - Day Hospital Visit
 - Emergency Room
- PEx proxied by any hospital IV antibiotic treatment episodes
- Prescription medication consumption
 - Antacid/Antiulcer
 - Antibiotics
 - Bronchodilators
 - Antifungals
 - Anti-inflammatory agents
 - Digestive or pancreatic enzymes
 - Mucus thinners/mucolytics
 - Osmotic Agents
 - Ursodeoxycholic acid
 - Vitamins

*The specific age of each patient is not provided, contained within the database are age intervals and for this study all the patients from these age intervals were considered for the analysis
CFTRm, cystic fibrosis transconductance regulator modulator; **HCRU,** healthcare resource utilization; **IV,** intravenous; **PEx,** pulmonary exacerbation

Definitions for hospital HCRU:

- **Episode:** an episode relates to a medication consumption record from the hospital pharmacy
- **Outpatient:** patient visits the Hospital Pharmacy to have the pharmacist dispense free medication that is reimbursed for the given pathology. This consumption does not have to be related to a department visit; it could be collection of a prescription at the hospital pharmacy only
- **Inpatient:** when the patient stays under observation and/or needs care that can only be provided in a hospital environment with monitoring by healthcare professionals. The patient may be admitted to various departments in the hospital
 - Admission to the intensive care unit (ICU)
 - Surgery may or may not be ambulatory, and may or may not require subsequent hospitalization
 - Inpatient-Other: any other hospitalization/inpatient besides ICU or Surgery

RESULTS

Figure 1. Demographic characteristics of the study cohort[a]

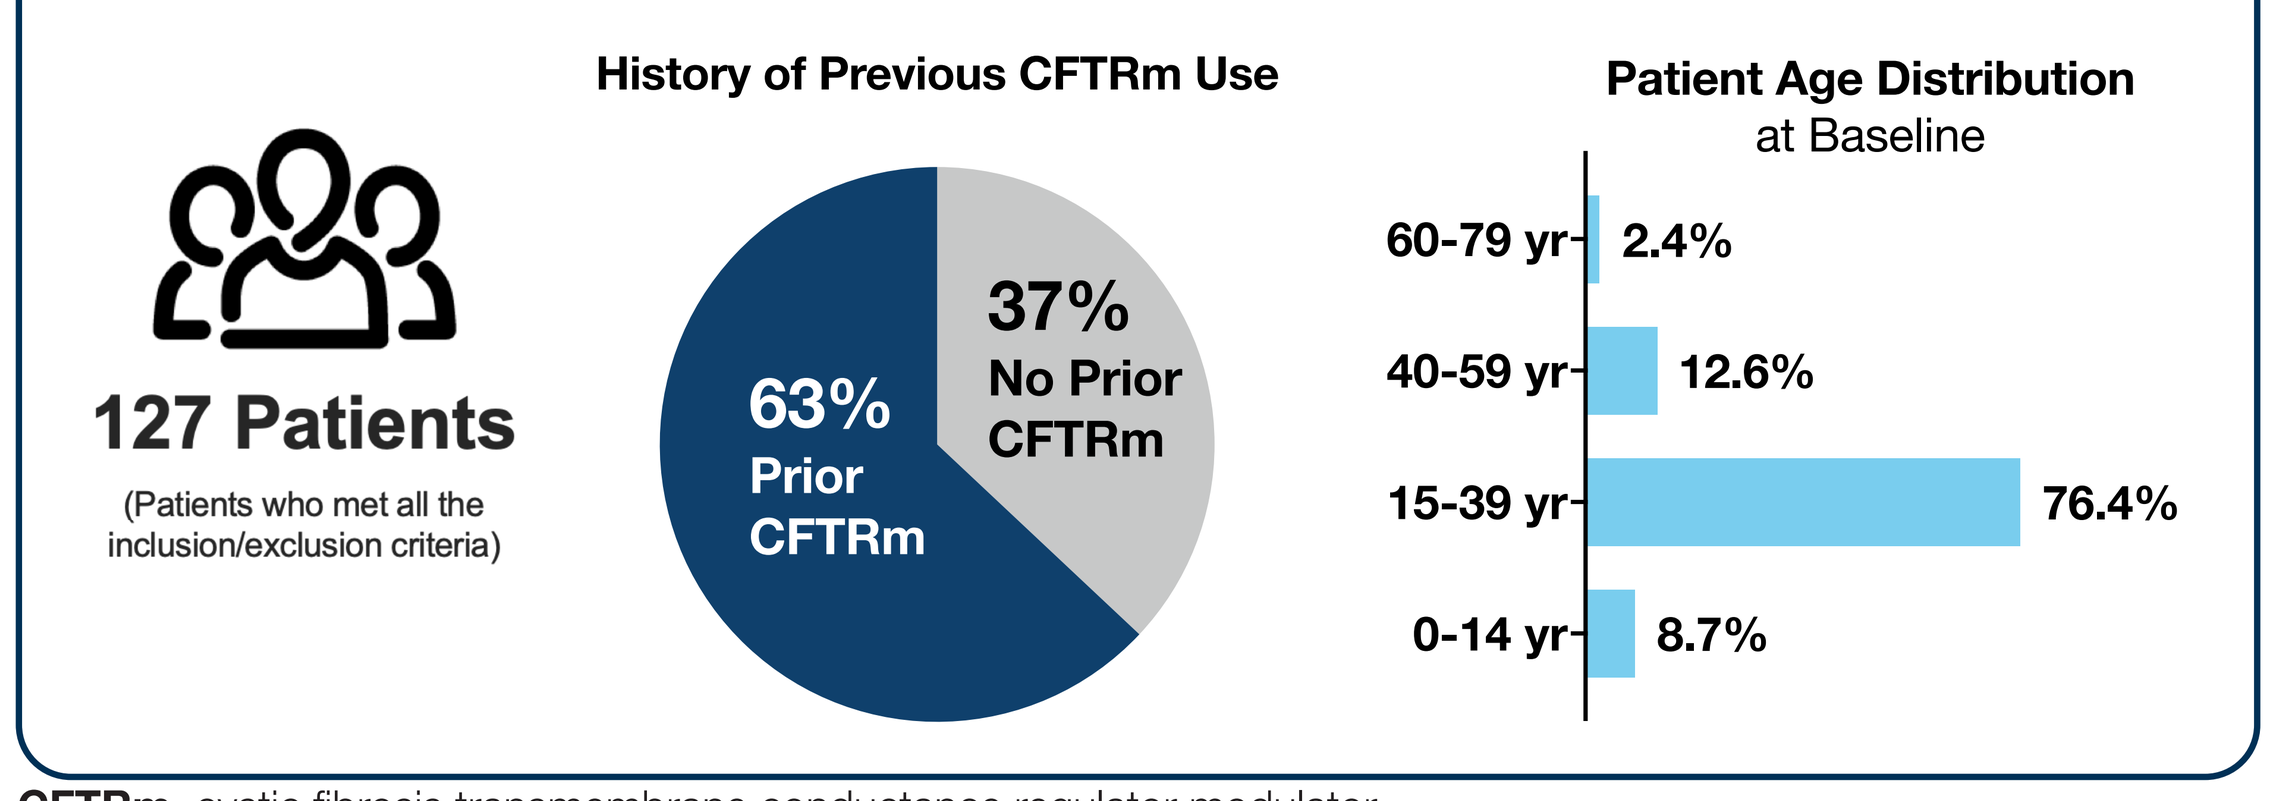
127 Patients
(Patients who met all the inclusion/exclusion criteria)
History of Previous CFTRm Use
63% Prior CFTRm
37% No Prior CFTRm
Patient Age Distribution at Baseline
60-79 yr 2.4%
40-59 yr 12.6%
15-39 yr 76.4%
0-14 yr 8.7%

CFTRm, cystic fibrosis transmembrane conductance regulator modulator
[a] Study period: August 21, 2019, and June 30, 2023. Baseline characteristics were obtained from the most recent record prior to ELX/TEZ/IVA initiation (index)

- For outpatient visit: Outpatient visit episodes increased post-ELX/TEZ/IVA initiation (people with CF need to visit hospital pharmacy every month to collect prescription medication, with consumption increasing)
- For inpatient-other: the number of participants with at least one episode decreased from 26 (20.5%) during baseline to 12 (9.4%) during follow-up, with a corresponding decrease in the total number of episodes from 44 during baseline period to 21 during follow-up period (risk ratio [RR] 0.48 [95% CI: 0.28, 0.80])
- Other hospital types (day hospital visit, emergency room, hospitalization/inpatient [surgery and ICU] didn't meet the sample size to proceed with analysis of the data [n≤5])
- Most patients (>85%) with at least one episode were seen within the pneumology and pediatrics departments

Figure 2. Hospital inpatient HCRU episodes decreased following ELX/TEZ/IVA initiation, whilst outpatient visit episodes increased

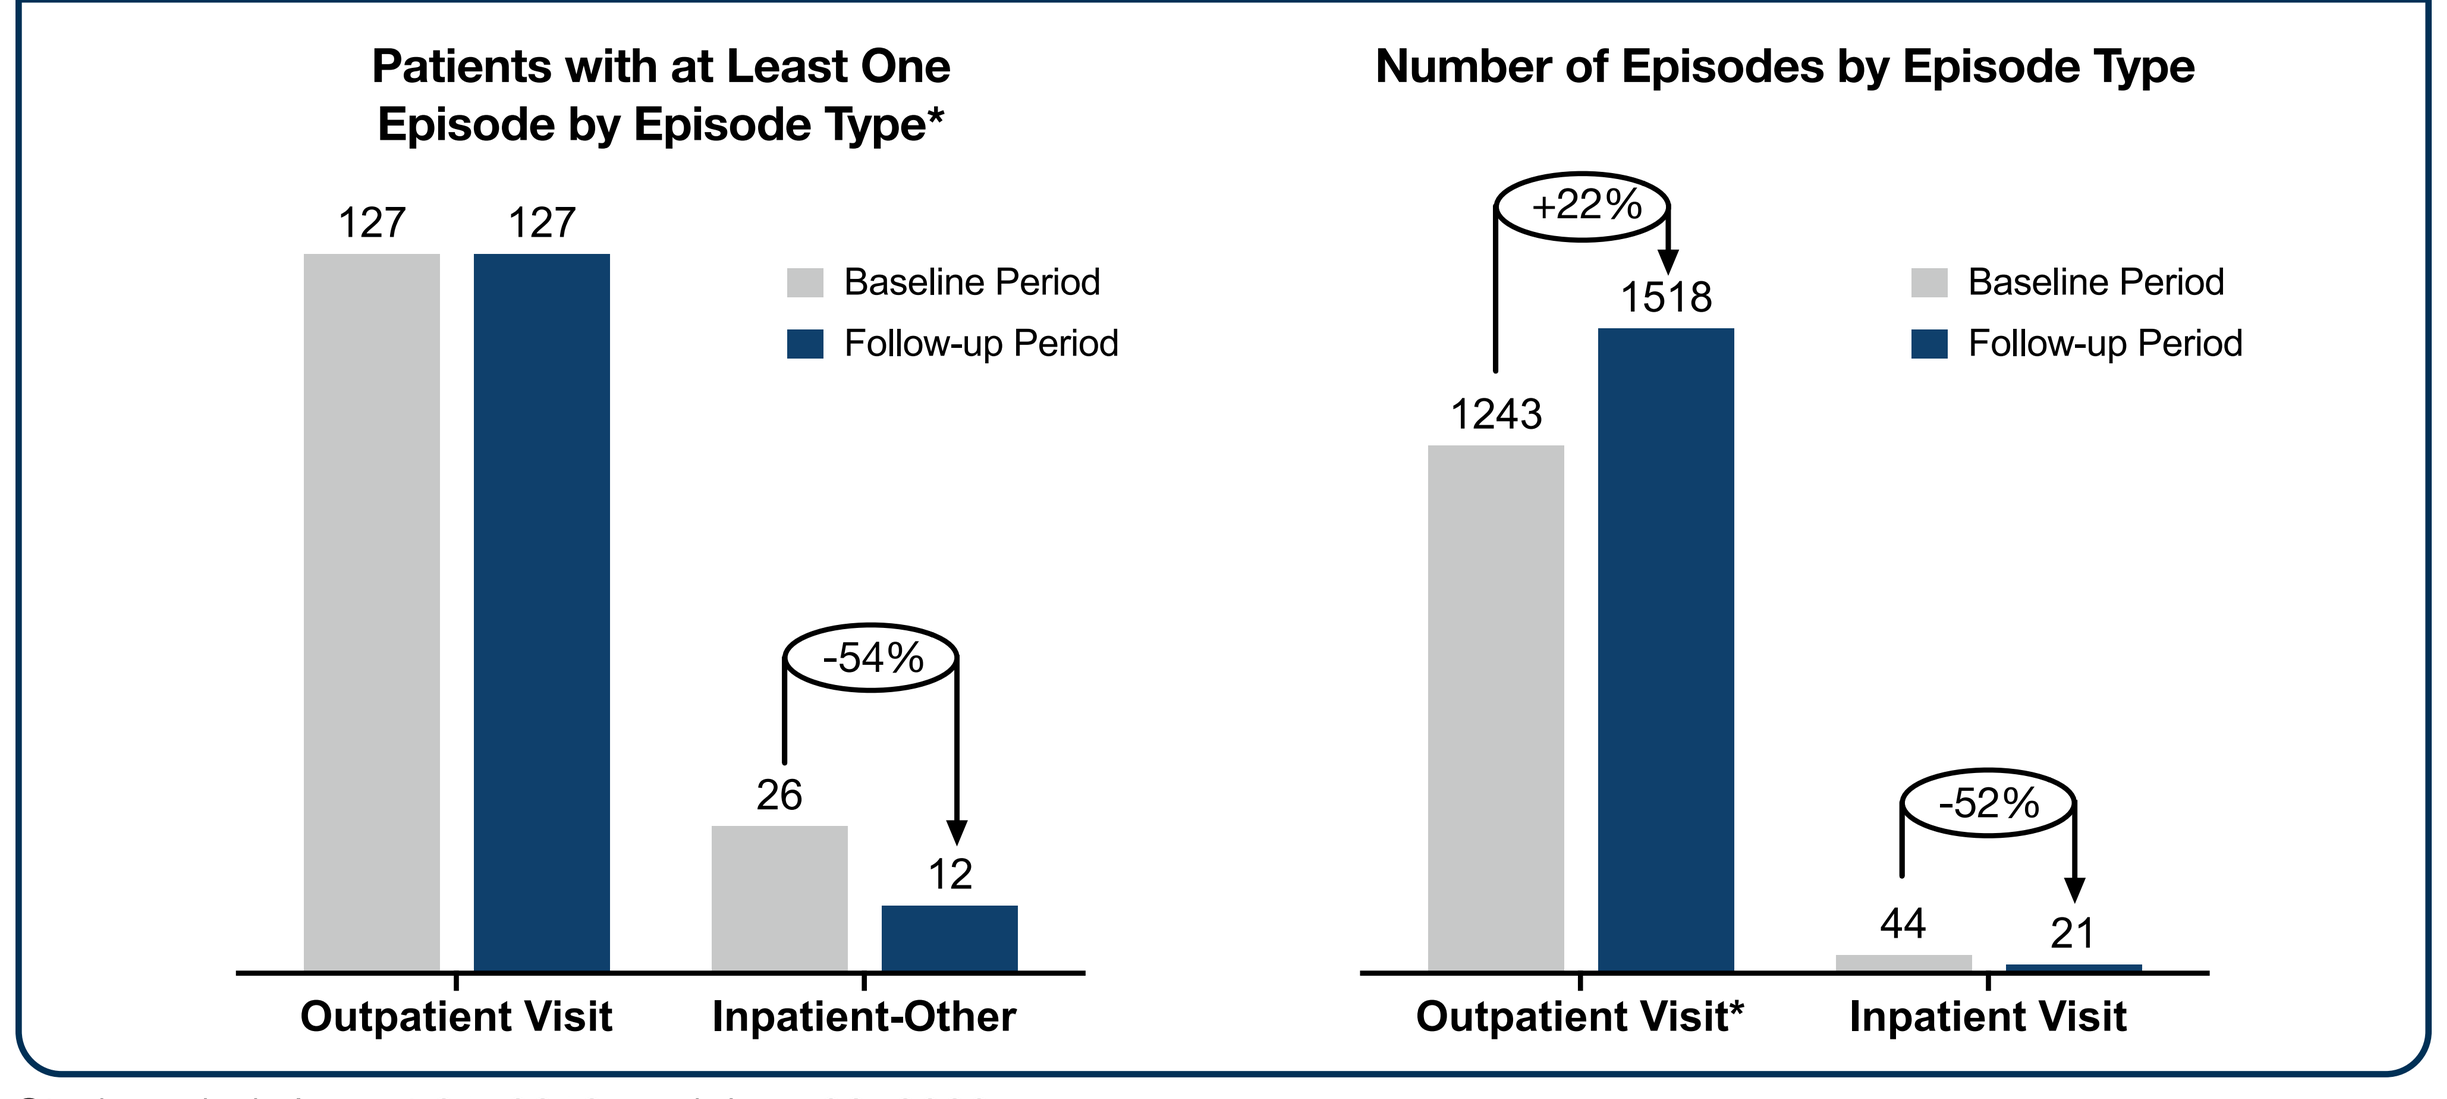
Patients with at Least One Episode by Episode Type*
127
127
Baseline Period
Follow-up Period
-54%
26
12
Outpatient Visit
Inpatient-Other
Number of Episodes by Episode Type
+22%
1243
1518
Baseline Period
Follow-up Period
-52%
44
21
Outpatient Visit*
Inpatient Visit

Study period: August 21, 2019, and June 30, 2023
ELX/TEZ/IVA, elexacaftor/tezacaftor/ivacaftor; **HCRU,** healthcare resource utilization
*The patients in this analysis are unique patients and are not counted more than once

Figure 3. Prescription medication consumption

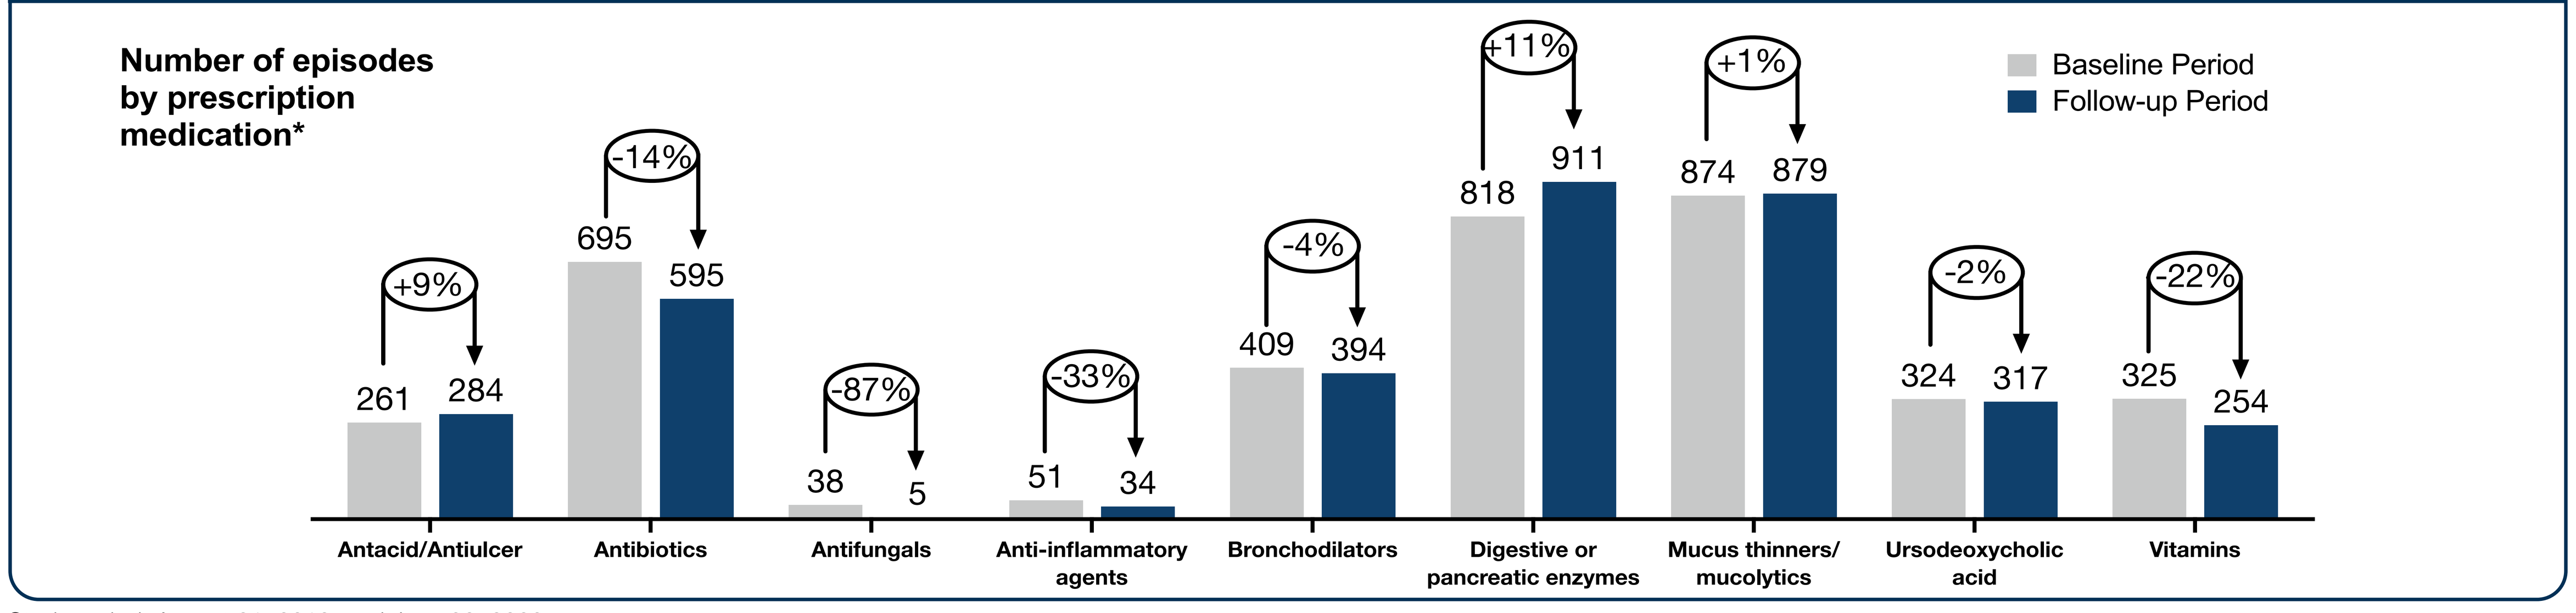
Number of episodes by prescription medication*
Baseline Period
Follow-up Period
+9%
261
284
Antacid/Antiulcer
-14%
695
595
Antibiotics
-87%
38
5
Antifungals
-33%
51
34
Anti-inflammatory agents
-4%
409
394
Bronchodilators
+11%
818
911
Digestive or pancreatic enzymes
+1%
874
879
Mucus thinners/ mucolytics
-2%
324
317
Ursodeoxycholic acid
-22%
325
254
Vitamins

Study period: August 21, 2019, and June 30, 2023
* Analgesics and antivirals reported ≤3 episodes

Table 1. Number and rate of PEx (identified by all IV antibiotic treatment episodes) declined after ELX/TEZ/IVA initiation

	ELX/TEZ/IVA N = 127[a]	
	Baseline (pre-initiation) Period	Follow-up (post-initiation) Period
Number of patients with at least one episode, n (%)	20 (15.8)	7 (5.5)
Number of PEx episodes, n	37	10
Annualized rate PEx, Mean (SE) [95% CI]	0.29 (0.05) [0.20 - 0.39]	0.08 (0.02) [0.03 - 0.13]
Rate ratio [95% CI]	0.27 [0.13, 0.54]	

Annualized rate of PEx declined by 73% in people treated with ELX/TEZ/IVA

Study period: August 21, 2019, and June 30, 2023. 95% confidence interval represents an exact 95% Poisson confidence interval
CI, confidence interval; **ELX/TEZ/IVA,** elexacaftor/tezacaftor/ivacaftor; **IV,** intravenous; **PEx,** pulmonary exacerbation; **SE,** standard error
[a] People with available data in the 12 months before and after index

Study Limitations

- The observational, real-world evidence study design did not control for potential confounding variables or other changes over time
- The main outcomes of ELX/TEZ/IVA were limited to 1-year of follow-up period after initiation
- Due to the nature of HMR database being a hospital consumption database:
 - Consumption data can be subject to overestimation due to prescriptions made in order to assure the patient has some medication in stock at home to avoid discontinuation
 - HMR database does not contain any specific clinical information, including diagnosis, *CFTR* genotype, lung function, etc.
 - HMR database does not capture people with CF that are not receiving a CFTRm and are naïve to treatment
- To ensure patient confidentiality, a non-reversible anonymization algorithm is employed before data is made available to HMR. This process may lead to an underestimation of the consumptions of some drugs
- Given that the database comprises data sourced from hospital consumption, discrepancies may stem from hospital recording practices
- Any consumption made by the patient outside the hospital context will not be included in this database, which might lead to an underestimation of the consumptions of some drugs
- Due to the short period of follow-up in this study, it is expected that observing changes in prescription medication use will be challenging

CONCLUSIONS

- **In this study, people with CF in Portugal who initiated ELX/TEZ/IVA had decreases in hospital pharmacy consumption with reductions in inpatient hospital admissions, prescription medication usage, and PEx**
- **These data are consistent with the results of previous clinical trials and other real-world evidence and further support the positive impact of ELX/TEZ/IVA on the lives of people with CF**
- **Longer follow-up is needed to confirm these early findings**

References

1. Middleton PG, et al. *N Engl J Med*. 2019;381(19):1809-1819.
2. Heijerman HGM, et al. *Lancet*. 2019;394(10212):1940-1948.
3. Barry PJ, et al. *N Engl J Med*. 2021;385(9):815-825.
4. Zemanick ET, et al. *Am J Respir Crit Care Med*. 2021;203(12):1522-1532.
5. Mall MA, et al. *Am J Respir Crit Care Med*. 2022;206(11): 1361-1369.
6. Sutharsan S, et al. *Lancet Respir Med*. 2022;10(3):267-277.
7. Mainz JG, et al. *Front Pharmacol*. 2022;13:877118.
8. Martin C, et al. *Respir Med Res*. 2021;80:100829.
9. DiMango E, et al. *J Cyst Fibros*. 2021;20:460-463.
10. Fajac I, et al. *J Cyst Fibros*. 2023;22(1):119-123.
11. Thompson A, et al. *Pediatr Pulmonol*. 2020;294-295. Abstract 655.

Author Disclosures

MB, MR, JR, and **HM** are currently, or were previously employed at HMR when the work was carried out; HMR provided statistical analysis, and reporting preparation in accordance with the agreed protocol and analysis plan developed in collaboration between Vertex and HMR

CB, GV-H, AO, and **HW** are employees of Vertex Pharmaceuticals Incorporated and may own stock or stock options in the company

PA received speaker honoraria and other fees from Vertex, Viatris and Zambon

Acknowledgments

This work was performed as part of a collaboration between HMR and Vertex Pharmaceuticals Incorporated. The authors thank all the people with cystic fibrosis who participated in this study

Editorial support was provided by Shivali Arora, PhD (SA) under the guidance of the authors and graphic design support was provided by Jonathan Kirk, MS (JK). **SA** and **JK** are employees of Vertex Pharmaceuticals and own stock/stock options in the company

The authors thank Luciana Saturnino (Vertex HEOR) for helpful discussions during design and interpretation, and Vasilis Nikolaou (Vertex Biostatistics) for helpful review and discussion of data

The authors thank Miguel Lopes (HMR Project Manager) for project management support

Presented at ISPOR EUROPE, Barcelona, Spain, 17-20 November 2024

Sponsored by Vertex Pharmaceuticals Incorporated