Qualitative literature review to explore the patient experience of rheumatoid arthritis and unmet treatment needs to inform clinical trial patient-reported and digital endpoints

Samantha Wratten[1], Dane McCarrick[2], Elizabeth Collins[1], Aikaterini Vardavoulia[1], Helen Kitchen[1], Anna Fishbein[3], Frédérique Chézalviel-Guilbert[4], Veleka Allen[5], Adeebah Adams[6], Ernest H Choy[7], Jeffrey R Curtis[8], Benoit Arnould[9], Jane R Wells[2]

[1]Clarivate, London, UK; [2]Sanofi, Reading, Berkshire, UK; [3]Sanofi, Cambridge, MA, USA ; [4]Sanofi, Paris, France ; [5]Sanofi, Bridgewater, NJ, USA; [6]Sanofi, Johannesburg, South Africa; [7]Cardiff University, Cardiff, UK; [8]University of Alabama at Birmingham, Birmingham, AL, USA; [9]Sanofi, Lyon, France

Copies of this poster obtained through Quick Response (QR) Code are for personal use only

Background

- Clinical trials are underway to develop SAR441566, an oral tumor necrosis factor receptor 1 signal inhibitor, as a treatment for rheumatoid arthritis (RA).[1]
- The use of patient-reported outcomes (PRO) assessments and digital sensors/wearables provide an opportunity to measure the patient experience within the context of a clinical trial.[2,3]
- It is essential that the outcomes measured using PRO and digital tools reflect the health aspects that are meaningful to patients.[4]
- Therefore, research to understand and define what constitutes meaningful treatment benefit to patients, and how to optimize the measurement of these outcomes, is both timely and warranted throughout the measurement development process.[2]

Objective

- To characterize the patient experience of RA via a targeted literature review (TLR) to develop disease conceptual models, including models of meaningful aspects of health (MAH) that patients want to prevent, improve, or avoid worsening.

Methods

- A TLR was conducted in November 2023 to identify peer-reviewed qualitative studies published in the past five years describing the experience of RA from the patient perspective (**Figure 1**).

Figure 1: Overview of study methods

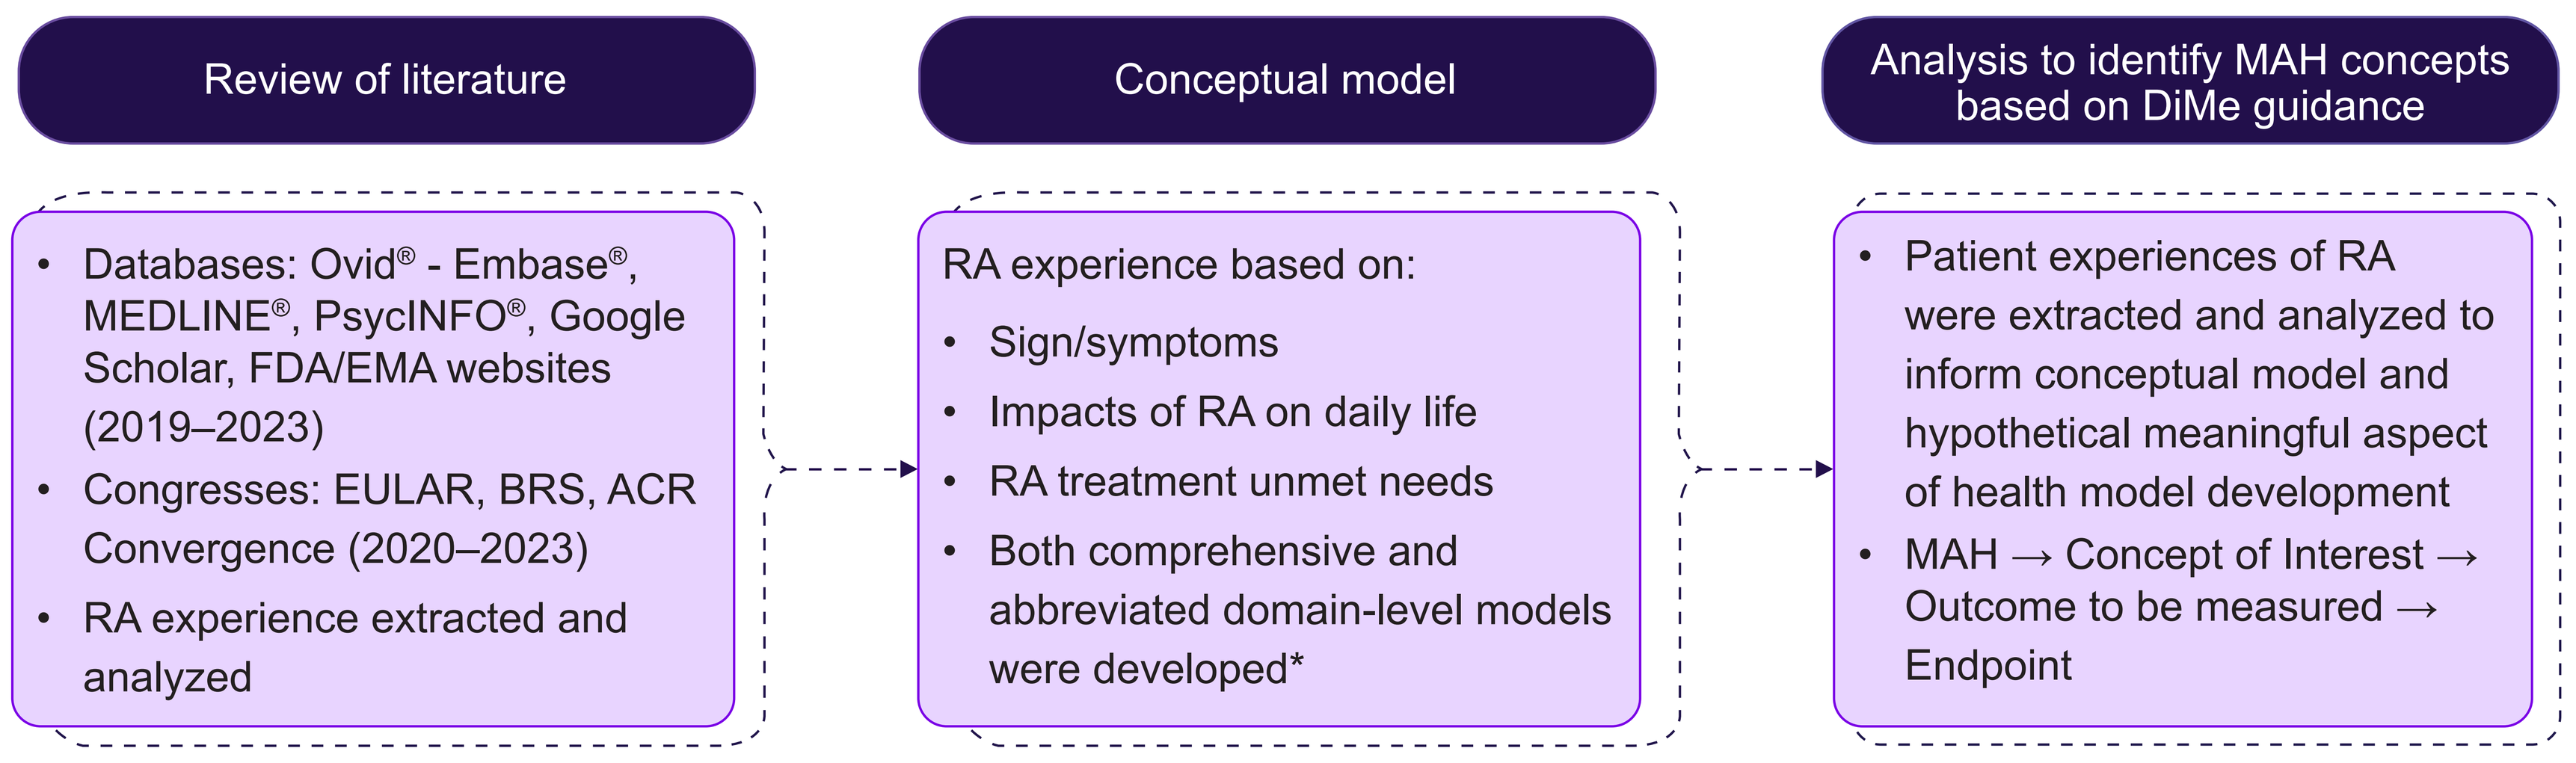
Review of literature
Conceptual model
Analysis to identify MAH concepts based on DiMe guidance
• Databases: Ovid® - Embase®, MEDLINE®, PsycINFO®, Google Scholar, FDA/EMA websites (2019–2023)
• Congresses: EULAR, BRS, ACR Convergence (2020–2023)
• RA experience extracted and analyzed
RA experience based on:
• Sign/symptoms
• Impacts of RA on daily life
• RA treatment unmet needs
• Both comprehensive and abbreviated domain-level models were developed*
• Patient experiences of RA were extracted and analyzed to inform conceptual model and hypothetical meaningful aspect of health model development
• MAH → Concept of Interest → Outcome to be measured → Endpoint

ACR: American College of Rheumatology; BRS: British Society for Rheumatology; DiMe: Digital Medicine Society; EMA: European Medicines Agency; EULAR: The European Alliance of Associations for Rheumatology; FDA: Food and Drug Administration; MAH: Meaningful Aspects of Health; RA: Rheumatoid arthritis.
* Abbreviated version is presented in the poster.

- The included studies were critically appraised utilizing Critical Appraisal Skills Programme (CASP).[5]
- Data extraction focused on patient descriptions of symptoms and impacts of RA, perceived associations between concepts, and unmet treatment needs.
- Draft conceptual models (conceptual model and MAH) were constructed in line with the Digital Medicine Society (DiMe) framework to guide the development of digital health endpoints that assess MAH.[2] The models were revised following expert rheumatologists' inputs.

Results

Literature review

- Our keyword-based search yielded a total of 2,971 citations from EMBASE (*n* = 2,238), Medline (*n* = 653), and PsycINFO (*n* = 80). After reviewing the titles and abstracts and removing duplicates, 74 studies remained for further evaluation. Of these, 42 articles were excluded due to discordance with the inclusion criteria, resulting in 26 studies included for the full review.
- The studies mainly reported individual interviews involving patients with RA (*n* = 22/26) from several regions (**Figure 2**). The average sample size of patients among the included studies was 28 (range: 7 – 90).

Figure 2: Geographic distribution of included studies

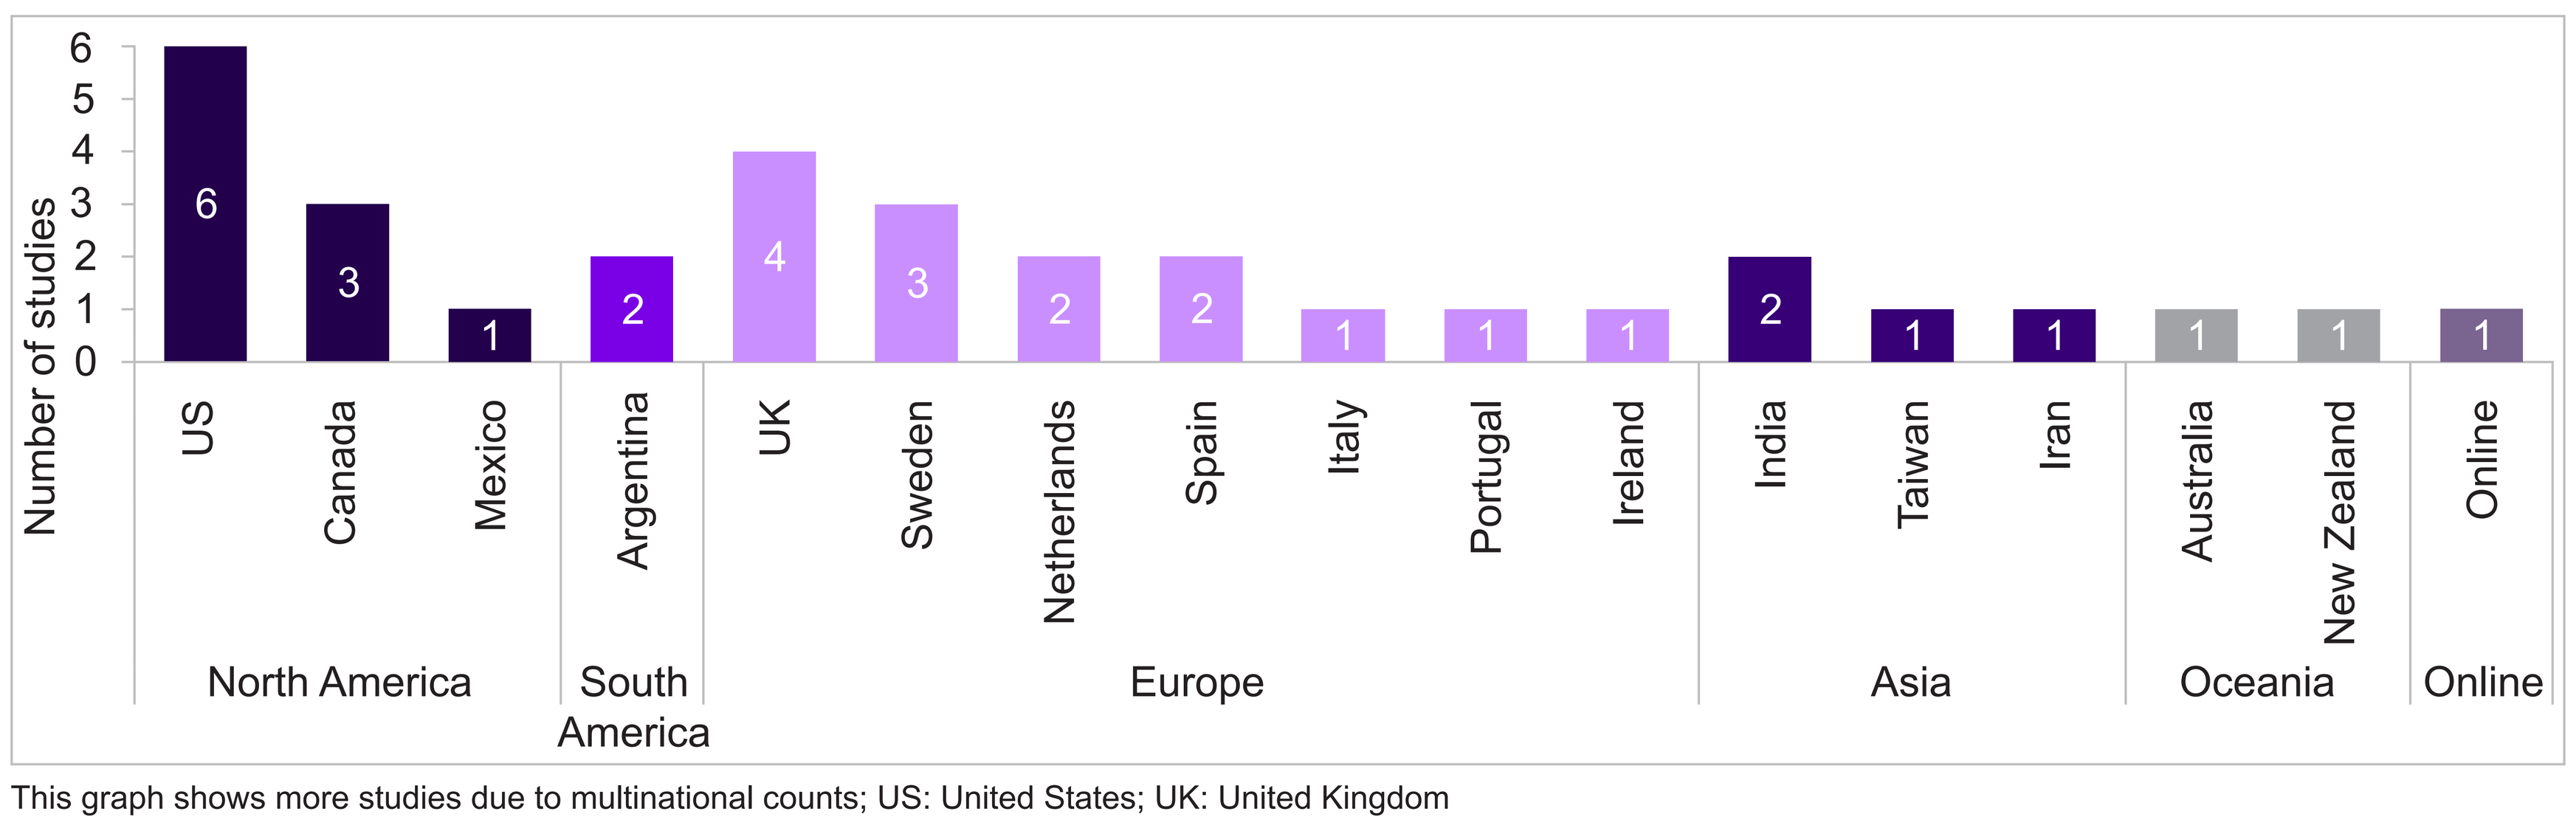
Number of studies
6
5
4
3
2
1
0
6
3
1
2
4
3
2
2
1
1
1
2
1
1
1
1
1
US
Canada
Mexico
Argentina
UK
Sweden
Netherlands
Spain
Italy
Portugal
Ireland
India
Taiwan
Iran
Australia
New Zealand
Online
North America
South America
Europe
Asia
Oceania
Online

This graph shows more studies due to multinational counts; US: United States; UK: United Kingdom

Development of conceptual model

- The review identified concepts, including signs/symptoms of RA, impacts of RA on health-related quality of life domains, and unmet treatment needs (**Figure 3**).

Figure 3: Domain-level conceptual model of the patient disease experience of rheumatoid arthritis

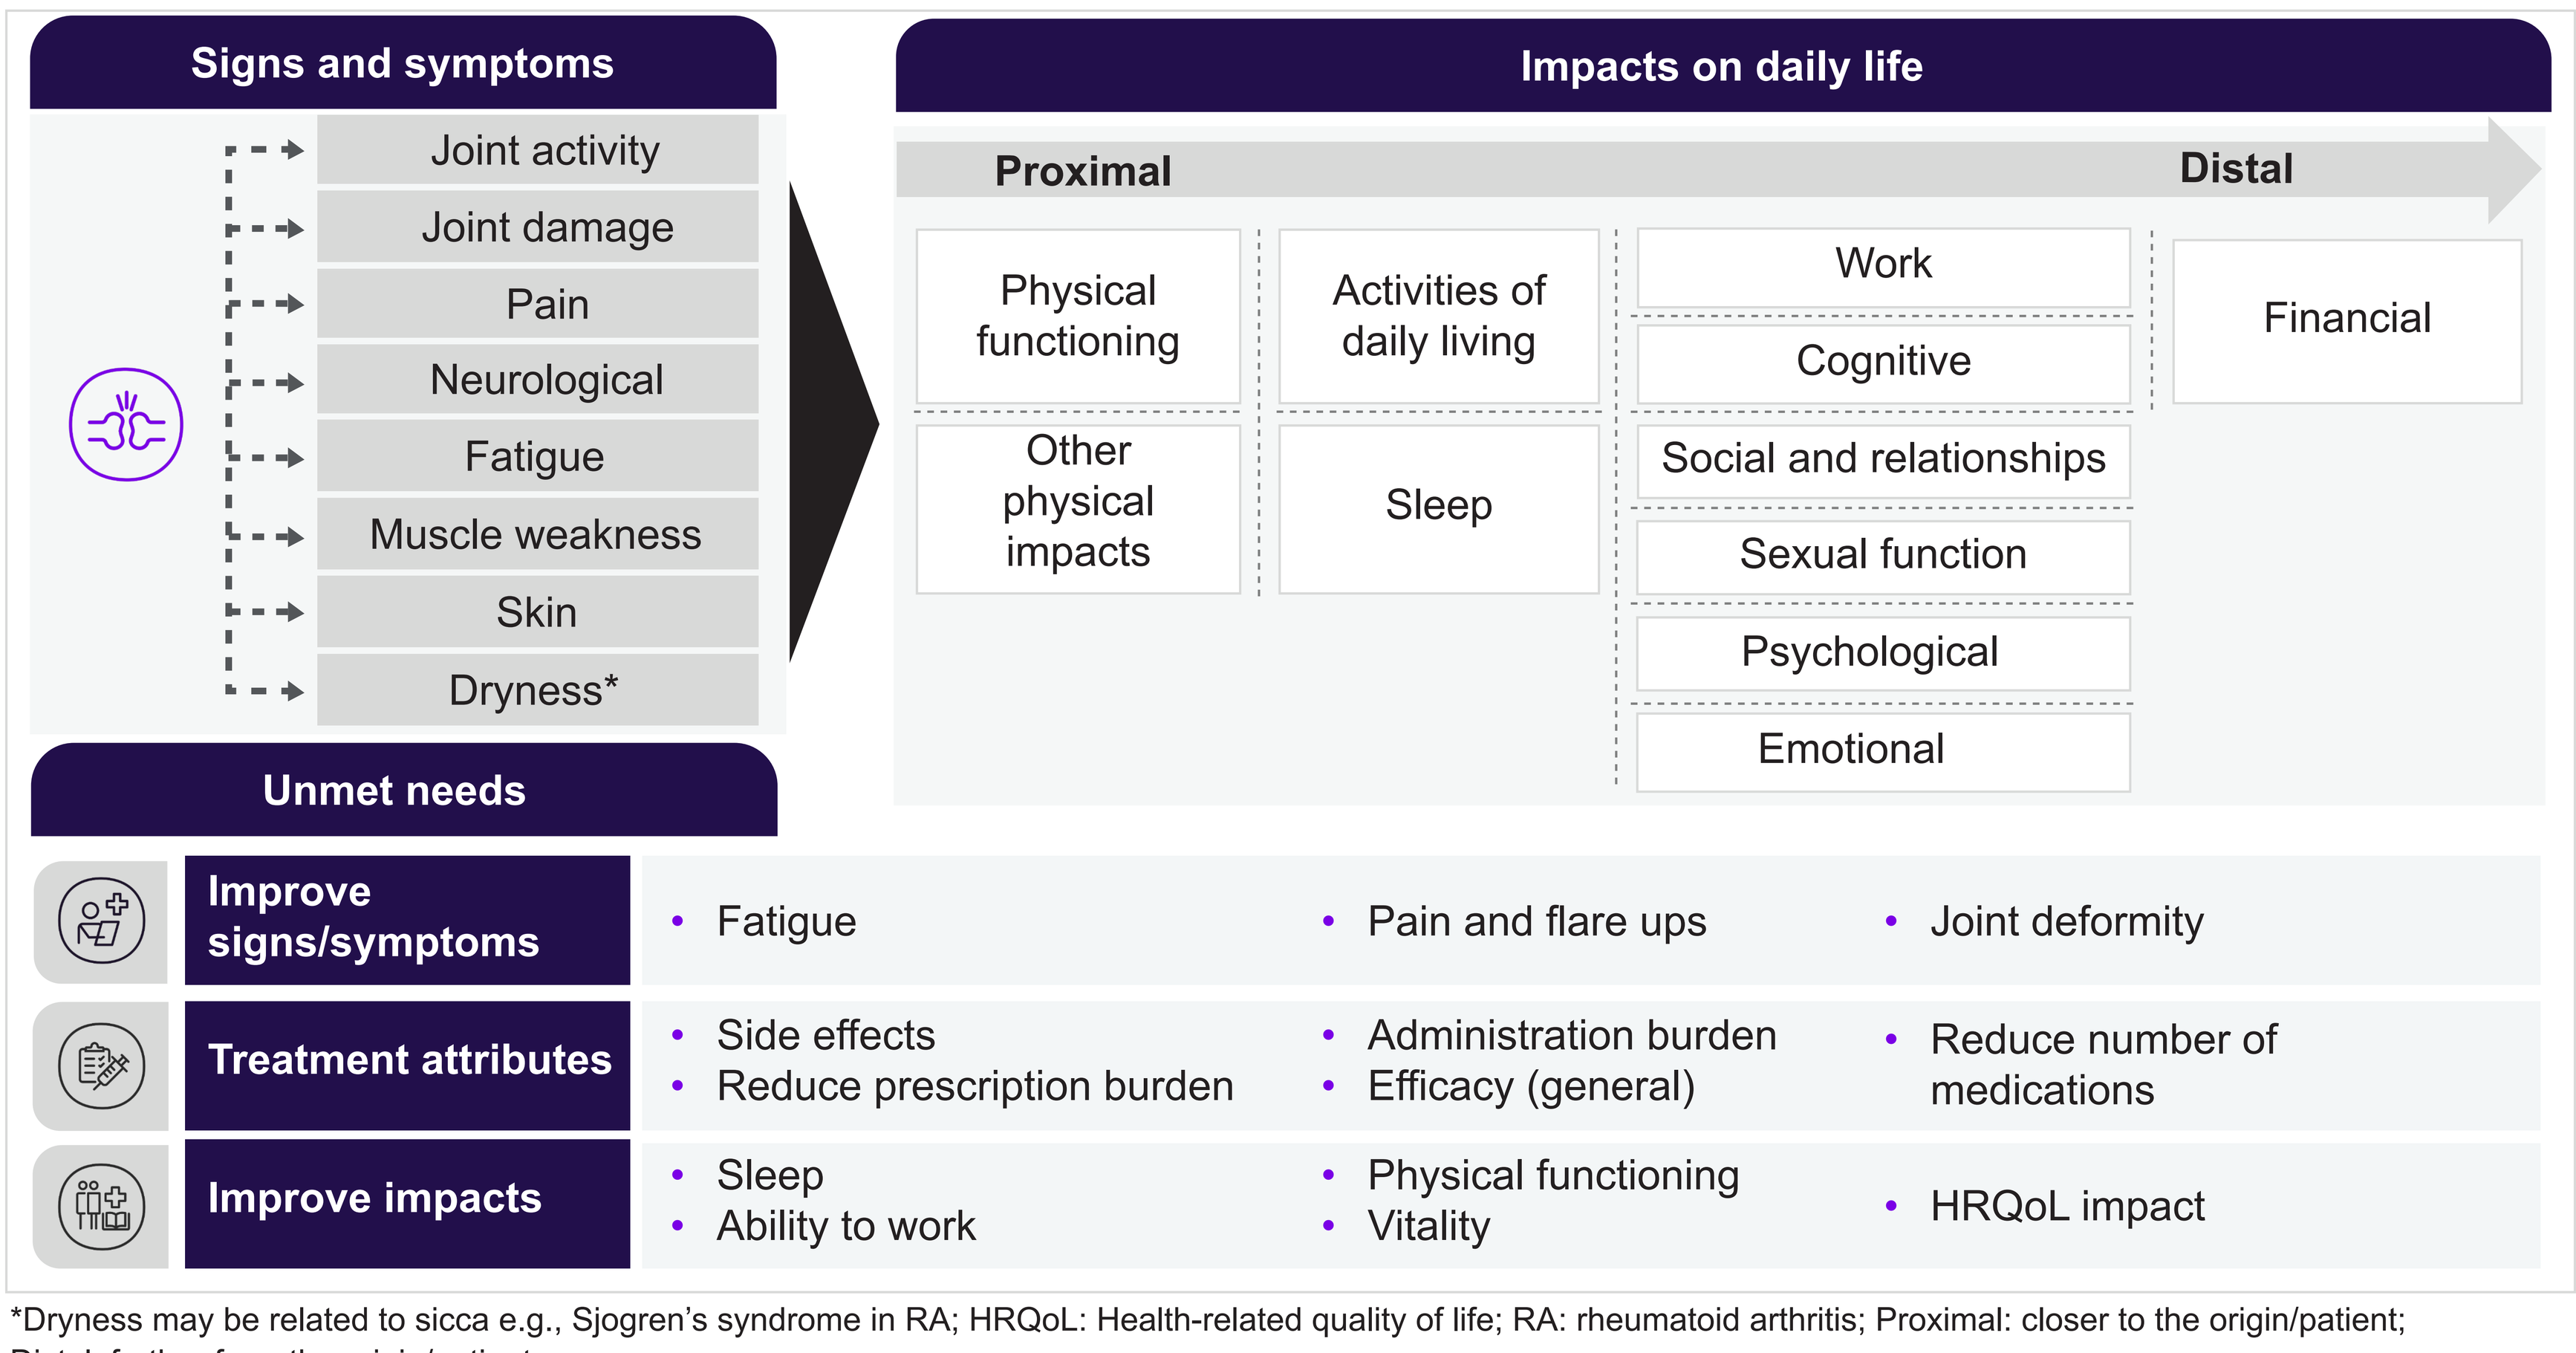
Signs and symptoms
Joint activity
Joint damage
Pain
Neurological
Fatigue
Muscle weakness
Skin
Dryness*
Impacts on daily life
Proximal
Distal
Physical functioning
Activities of daily living
Work
Cognitive
Financial
Other physical impacts
Sleep
Social and relationships
Sexual function
Psychological
Emotional
Unmet needs
Improve signs/symptoms
• Fatigue
• Pain and flare ups
• Joint deformity
Treatment attributes
• Side effects
• Reduce prescription burden
• Administration burden
• Efficacy (general)
• Reduce number of medications
Improve impacts
• Sleep
• Ability to work
• Physical functioning
• Vitality
• HRQoL impact

*Dryness may be related to sicca e.g., Sjogren's syndrome in RA; HRQoL: Health-related quality of life; RA: rheumatoid arthritis; Proximal: closer to the origin/patient; Distal: further from the origin/patient.

- Most bothersome and/or important to improve concepts were extracted when explicitly reported in the literature (**Figure 4**).

Figure 4: Conceptual model supporting quotes: most bothersome and/or important to improve symptoms and impacts

SYMPTOMS

Pain and pain flare ups (reduction in pain and flare ups) – "*I can run my life if I am not in so much pain…*" "*That would be my outcome, not having a flare, when my joints flare up it is excruciating.*"[6]

Joint deformity (slow joint deformity) – "*I have seen the long-term progression in others like deformity, and some are not able to walk.*"[6]

Treatment side effects ("organ function", "medication side effects [weight gain, dry nails, hair loss, changes in mood, stomach cramps, cancer, cataracts, diabetes]).[7]

Fatigue "*I wish I had more energy to do things that I enjoy doing. Like I said I used to do a lot of yard work with as my therapy, working in the yard and I used to walk a lot for exercise. I don't do that anymore. I'm just kind of isolated because on the weekends I'm so tired.*"[7]

IMPACTS

Physical functioning* (better function / movement) – "*I want to move my hands and be able to lift my grandkids.*" "*I would prefer both my hands to work and my feet and my knees.*"[6]

Sleep & vitality "*…their preferred treatment outcome was expressed as an increased desire to experience vitality through enthusiasm, energy, good sleep, and good spirits.*"[8]

Daily activities & ability to work "*The most important thing for me is to function, yeah to be able to work, and do the things I am used to and such.*"[9]

Financial impact "*The patients' preferences at both time points were to be able to work and to maintain their economic standard*"[8]

*Including mobility, lifting objects, hand movements.
KEY: Concept [Author reported "treatment priority name"] or [Author reported "domain important to patient"] – "*patient quote*"

Development of meaningful aspects of health models

- Although some bothersome and important to improve concepts were identified, the reviewed data sources did not consistently report an association between MAH and symptoms or functional impacts.
- Expert rheumatologist review indicated that relevant MAHs may not have been identified in the literature, so hypothetical models have been developed combining the data and expert insights (**Figure 5 and 6**).

Figure 5: Hypothesized MAH models – fine motor skills

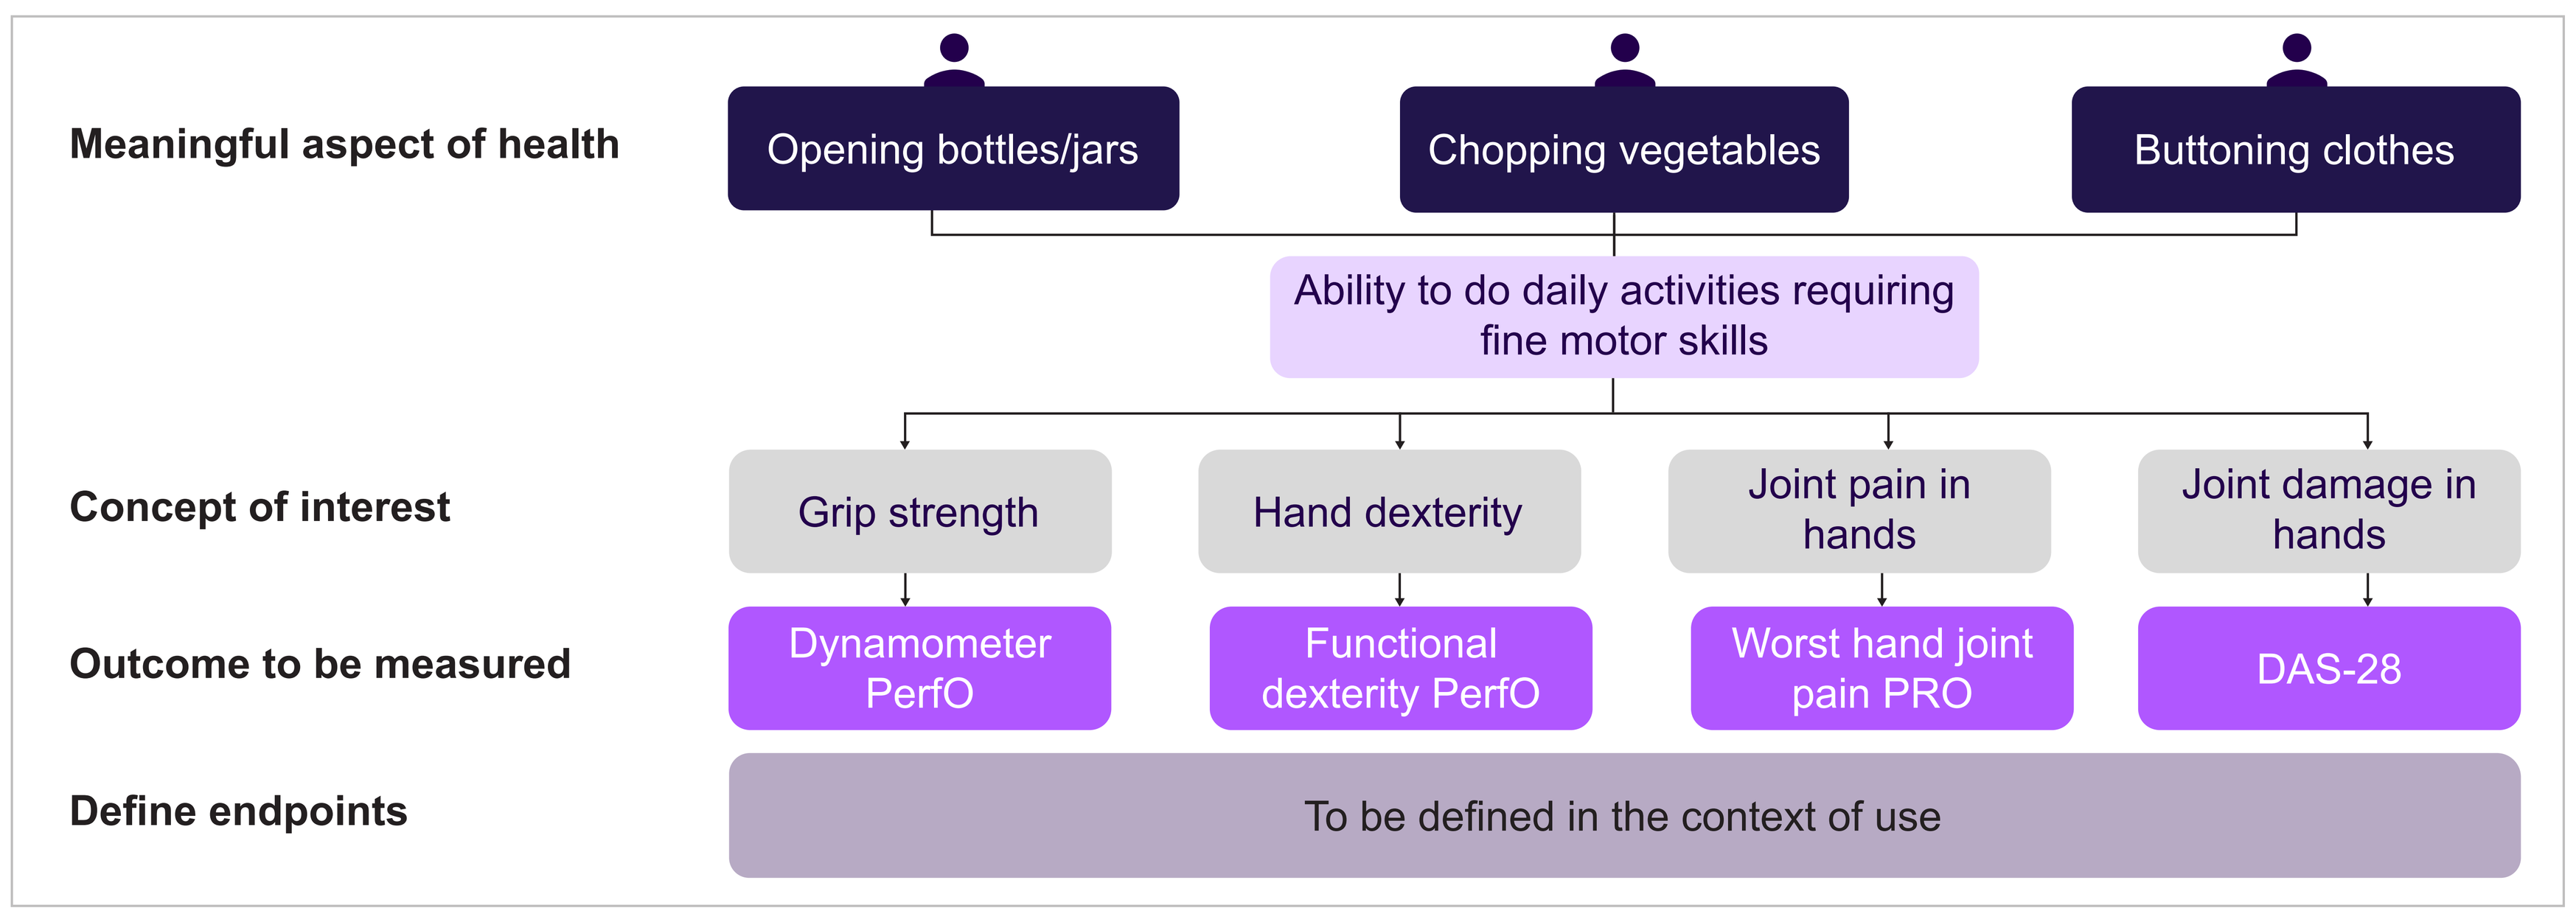
Meaningful aspect of health
Opening bottles/jars
Chopping vegetables
Buttoning clothes
Ability to do daily activities requiring fine motor skills
Concept of interest
Grip strength
Hand dexterity
Joint pain in hands
Joint damage in hands
Outcome to be measured
Dynamometer PerfO
Functional dexterity PerfO
Worst hand joint pain PRO
DAS-28
Define endpoints
To be defined in the context of use

DAS: Disease Activity Score; MAH: meaningful aspects of health; PerfO: performance outcome; PRO: patient-reported outcomes.

Figure 6: Hypothesized MAH models – lower limb exercises

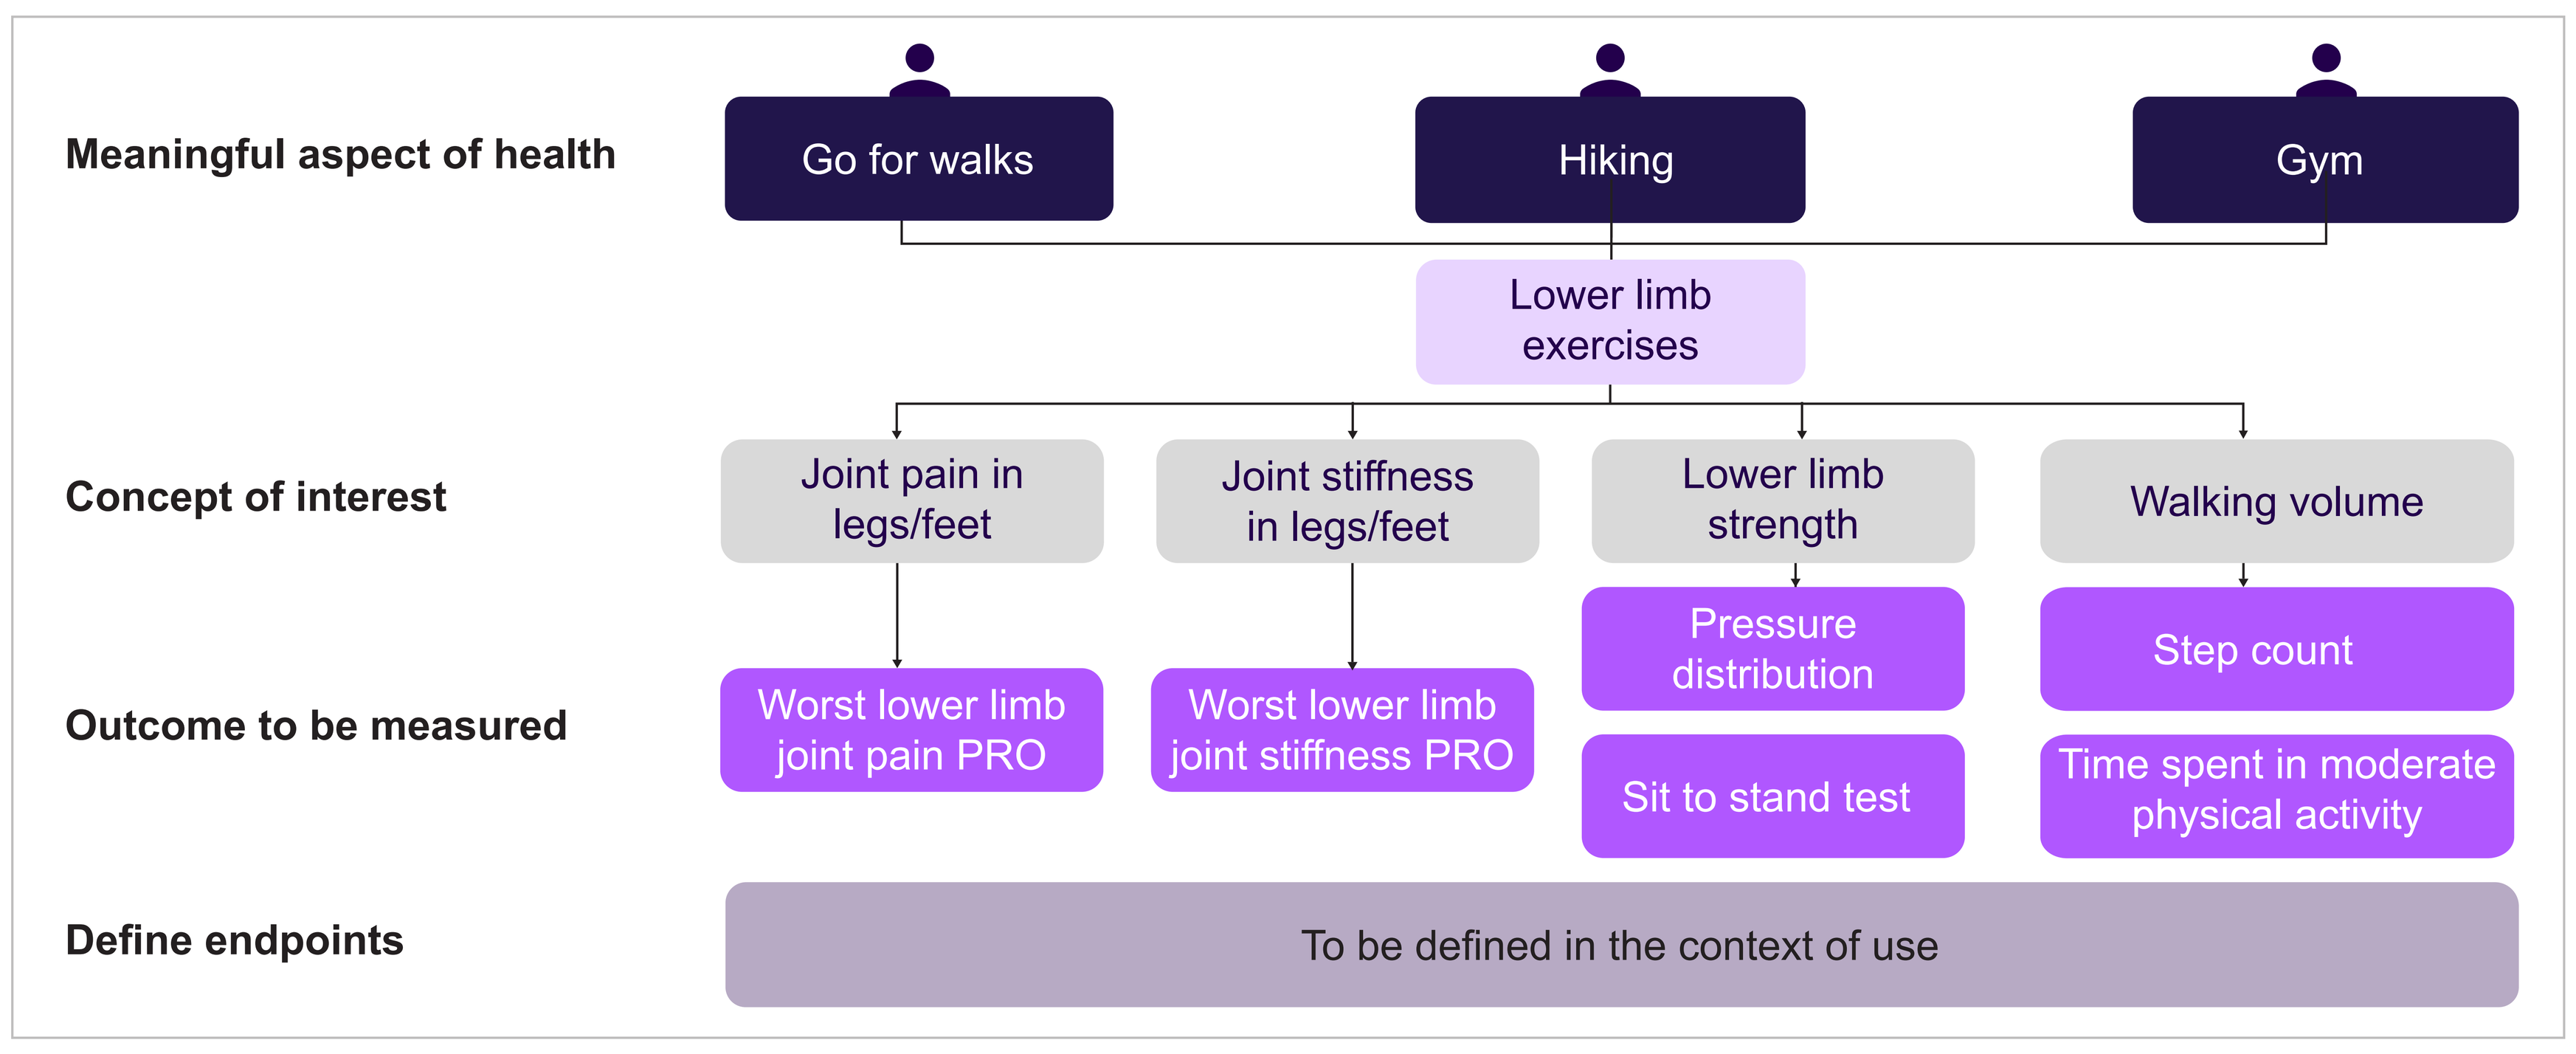
Meaningful aspect of health
Go for walks
Hiking
Gym
Lower limb exercises
Concept of interest
Joint pain in legs/feet
Joint stiffness in legs/feet
Lower limb strength
Walking volume
Outcome to be measured
Worst lower limb joint pain PRO
Worst lower limb joint stiffness PRO
Pressure distribution
Sit to stand test
Step count
Time spent in moderate physical activity
Define endpoints
To be defined in the context of use

MAH: meaningful aspect of health; PerfO: performance outcome; PRO: patient-reported outcomes.

Limitations

- One limitation we encountered was insufficient data on MAH, the models we developed for MAH are currently hypothetical. This lack of empirical data limits the robustness and accuracy of these models, highlighting the need for further research and data collection in this area.
- Another limitation was the necessity of a five-year publication cut-off. This criterion was essential to ensure our model remained contemporary and relevant. However, it also means that some potentially valuable data might have been excluded.

Conclusions

- These models hypothesize how MAH may be linked to measurable concepts of interest, and possible outcome measures. These in turn would be linked to context-specific endpoints.
- These models provide a proposed structure for mapping aspects of health meaningful to RA patients to appropriate digital and clinical outcome endpoints in RA clinical trials.
- Multiple clinical outcome assessments (including PROs and clinician reported outcomes) and digital health technologies may be needed to capture the MAH and outcomes that were identified in this literature review. The best way to capture these concepts will be examined in the next stage of the research and analysis.
- In the subsequent research phase, interviews with RA patients will be conducted to gain further insights into their experiences and to guide the development of models and the selection of clinically relevant PROs and digital endpoints.

REFERENCES
1. ClinicalTrials.gov.NCT06073093. Accessed 2024-10-08; 2. Manta et al. *Digital Biomarkers*. 2020;4(3):69–77; 3. Kruzinga et al. *Pharmacological Reviews*. 2020;72(4):899–909; 4. Orbai et al. *Current rheumatology reports*. 2015;17(1)–10; 5. Long et al. *Research Methods in Medicine & Health Sciences*. 2020;1(1):31-42; 6. Cozad et al. *Clinical nursing research*. 2023;32(1):40–48; 7. Becker et al. *Journal of Patient-Reported Outcomes*. 2021;5(1)–14; 8. Landgren et al. *Patient Preference and Adherence*. 2020;1421–1433; 9. Bywall et al. *Clinical Rheumatology*. 2022; 41(3):695-704.

FUNDING: This study was sponsored by Sanofi.

ACKNOWLEDGEMENT: Medical writing support was provided by Deepshikha Pawar, Sunita Rana, and Rahul Nikam from Sanofi.

CONFLICTS OF INTEREST: DM, AF, FCG, VA, AA, JRW and AB are employees of Sanofi and may hold shares and/or stock options in the company. SW, EC, AV, HK are employees of Clarivate, the consultancy who received funding from Sanofi to conduct the study described in this submission. HK owns stocks in Clarivate. JC and EHC were paid by Sanofi to input into the study described in this submission.

The poster was presented at ISPOR EU 2024, Nov 17–20, 2024, Barcelona, Spain

Poster Number: PCR250