EE286

Cost-effectiveness of nirmatrelvir/ritonavir in Swedish adults, stratified by age, comorbidity and vaccination status, compared to no anti-viral treatment

Fredrik Nilsson[1], Martina Aldvén[1], Tendai Mugwagwa[2], Christian Rappe Gerdesköld[1]

Affiliations: **1** Pfizer AB, Stockholm, Sweden, **2** Pfizer Inc., Tadworth, Surrey, United Kingdom

OBJECTIVE

Nirmatrelvir/ritonavir (NMV/r) is an antiviral agent indicated for adults at increased risk for progression to severe COVID-19, regardless of vaccination status, and is reimbursed in Sweden according to label. Both the pivotal clinical trial EPIC-HR [1] and real-world evidence (RWE) [*i.a.* 2,3,4] have shown that the NMV/r is effective in preventing hospitalization and death regardless of vaccination status and COVID-19 variant. This study assessed the cost-effectiveness of NMV/r for various risk groups, stratified by age, comorbidity, and vaccination status, compared to standard-of-care (SoC) in Sweden.

METHODS

A previously published and validated cost-effectiveness model was utilized and adapted to the Swedish setting [5]. The model used a short-term decision-tree (1 year) followed by a lifetime 2-state Markov model. The model accounted for symptom days, hospitalizations, intensive care unit admissions, quality of life, mortality, and treatment costs. It had a conservative approach by excluding the considerable, but still uncertain, costs related to post-acute covid syndrome. Overview of the model is shown in **Figure 1** and model inputs used in **Table 1**.

Figure 1. Model overview.

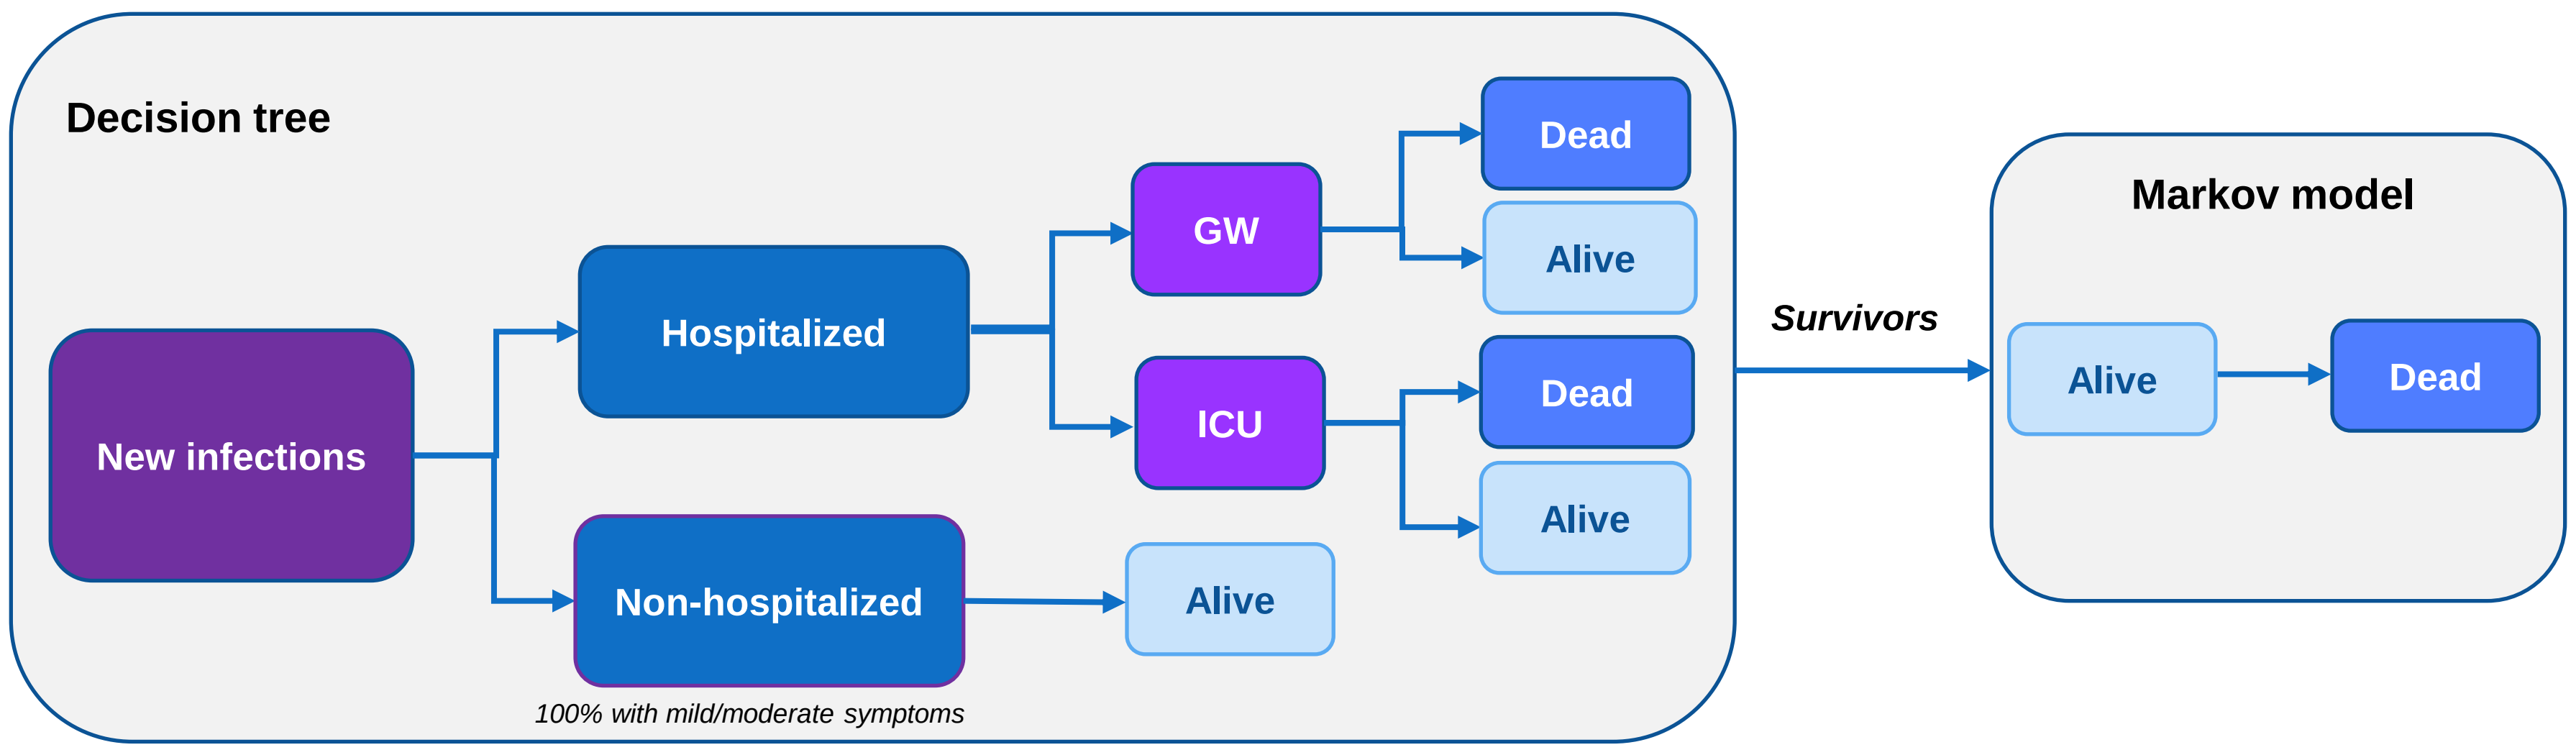
Decision tree
New infections
Hospitalized
Non-hospitalized
100% with mild/moderate symptoms
GW
ICU
Dead
Alive
Dead
Alive
Alive
Survivors
Markov model
Alive
Dead

Table 1. Model inputs.

BASE CASE INPUTS	
Annual discount rate (costs & health benefits) [9]	3%
Medication cost per case – Paxlovid [10]	SEK 9,965
Cost per day at GW [11]	SEK 11,824
Cost per day at ICU [11]	SEK 71,473
Disutility, per day – Non-Hospitalized [12]	-0.290
Disutility, per day – Hospitalized [12]	-0.640
Disutility, total QALY loss – PACS [12]	-0.340
Proportion ICU [13]	2.40%
Proportion PACS in non-hospitalized [14]	5.7%
Proportion PACS in hospitalized [14]	17.5%
Symptom days in non-hospitalized [15]	6.87
Length of stay at GW (days) [16]	6
Length of stay at ICU (days) [16]	7
TREATMENT EFFECTIVENESS	
Reduction in number of hospitalizations and deaths, treatment within 5 days since the onset of symptoms [2]	79.6% (33.9-93.8)
Reduction in symptom days non-hospitalized [17]	20%
Reduction of length of stay, hospitalization [3]	30%
Reduction in proportion requiring ICU [3]	65%

The baseline absolute risks of hospitalization and death were collected from published data for 54 risk groups, unique in its granularity, detailing the risk for COVID-19 hospitalization and deaths in Sweden by age, vaccination status and comorbidity burden, based on nationwide data from the first quarter of 2022 [6]. The relative risk-reduction was set to 79.6%, based on RWE to reflect omicron vaccinated era [2]. Baseline utility for different age groups was sourced from Ara & Brazier [7].

Cost-effectiveness was assessed for a range of patient profiles (n=54) with different combinations of vaccination status, comorbidity profile and age. Vaccination status was categorized as ‘vaccinated within past 180 days’, ‘vaccinated before past 180 days’ and ‘unvaccinated’, comorbidity profiles assessed were ‘no comorbidities’, low- and high comorbidity status and age was varied in 10-year intervals from below 40 to above 80 years.

Quality-adjusted life years and costs were accumulated over the patients’ life expectancy and the incremental cost-effectiveness ratio (ICER) was calculated. A willingness-to-pay threshold of SEK 500,000 per QALY was used to determine cost-effectiveness of NMV/r treatment. This is a commonly used threshold in Sweden for moderate conditions [8].

RESULTS

The ICERs for different patient profiles show significant variation, from almost nine million SEK to being dominant (i.e. cost saving with higher QALY gains vs SoC), see **Table 2**. NMV/r was a dominant treatment option in 12 risk groups. Utilizing a willingness to pay threshold of SEK 500.000 resulted in NMV/R being cost-effective in 30 risk groups. NMV/r was cost-effective in all patient groups older than 50 years with high comorbidity status. Only among patients younger than 40 years there were no risk groups in which NMV/r was cost-effective. NMV/r was cost-effective for all patients older than 70 years, except for those with no comorbidities that had been vaccinated in the prior 180 days.

Table 2. ICER for different patient profiles (in SEK): ICER below WTP level marked in bold

Vaccination status	Comorbidity burden	Age[a]					
		<40 years	40-49 years	50–59 years	60–69 years	70–79 years	80+ years
Vaccinated within past 180 days	none	7,659,802	6,028,560	9,003,026	4,432,556	580,257	**53,463**
	low	8,868,918	803,207	831,289	668,331	**103,836**	**29,808**
	high	8,146,790	**156,407**	**140,263**	**44,585**	**16,588**	**3,062**
Vaccinated before past 180 days	none	8,995,119	3,372,696	4,644,583	8,688,525	**95,190**	**Dominant**
	low	8,766,810	**484,904**	730,606	**303,373**	**Dominant**	**Dominant**
	high	8,300,464	8,223,517	**104,539**	3,397,720	**Dominant**	**Dominant**
Unvaccinated	none	5,720,717	2,505,151	1,675,717	**303,373**	**Dominant**	**Dominant**
	low	1,905,049	**152,522**	**172,498**	**14,809**	**Dominant**	**Dominant**
	high	6,083,939	**16,006**	**20,142**	**Dominant**	**Dominant**	**Dominant**

Notes:
ICER: Incremental cost-effectiveness ratio, SEK: Swedish krona, WTP: willingness to pay (500,000 SEK)
a. One specific age (not age range) had to be used to run the analysis, for <40 age 40 was used, for the intervals the midpoint, i.e. 45, 55, etc. was used, for 80+ age 85 was used to run the analysis.

CONCLUSION

This study suggests that utilization of nirmatrelvir/ritonavir is cost-effective compared to SoC for most patient groups, and especially for patients older than 70 years for whom it often is a cost-saving treatment. Even for several patient groups belonging to age cohorts as young as 40-49 years may NMV/r be considered a cost-effective treatment option.

REFERENCES

1. Hammond et al. Oral Nirmatrelvir for High-Risk, Nonhospitalized Adults with Covid-19. N Engl J Med. 2022; 386:397-1408 **2.** Lewnard et al. Effectiveness of nirmatrelvir-ritonavir against hospital admission or death: a cohort study in a large US healthcare system. Lancet Infect Dis. 2023;23:806-15 **3.** Aggerwal et al. Real-world use of nirmatrelvir-ritonavir in outpatients with COVID-19 during the era of omicron variants including BA.4 and BA.5 in Colorado, USA, Lancet Infect Dis. 2023;6:696-705 **4.** Dryden-Peterson et al. Nirmatrelvir Plus Ritonavir for Early COVID-19 in a Large US Health System. Ann Intern Med 2023;176:77-84 **5.** Carlson, J., et al., Cost-Effectiveness of Oral Nirmatrelvir/Ritonavir in Patients at High Risk for Progression to Severe COVID-19 in the United States. Value Health, 2024. 27(2): p. 164-172. **6.** Wahlström, E., et al. Who was at increased risk of severe covid-19 during the first omicron wave? 2023 [cited 2024 06 June]; Available from: https://lakartidningen.se/opinion/debatt/2023/05/vilka-hade-forhojd-risk-for-svar-covid-19-under-forsta-omikronvagen/. **7**. Ara & Brazier. Using health state utility values from the general population. Value in Health. 2011;14(4):539-45 **8.** Viollet, J., et al., HTA228 Willingness to Pay for Different Severity Levels in Sweden: An Analysis of TLV Decisions (2014-2022). Value in Health, 2022. 25(12): p. S341. **9.** The Dental and Pharmaceutical Benefits Agency. Tandvårds- och läkemedelsförmånsverkets allmänna råd. 2017. **10.** The Dental and Pharmaceutical Benefits Agency - Price & decision database – Paxlovid. 2024. **11.** Södra sjukvårdsregionen, Regionala priser och ersättningar för Södra sjukvårdsregionen 2023/2024 **12.** Goswami et al. Cost-Effectiveness Analysis of Molnupiravir Versus Best Supportive Care for the Treatment of Outpatient COVID-19 in Adults in the US. Pharmacoeconomics. 2022;40(7):699-714 **13.** Socialstyrelsen. Övergripande statistik över slutenvårdade med covid-19. 2024 02 September 2024]; Available from: https://www.socialstyrelsen.se/statistik-och-data/statistik/statistik-om-covid-19/. **14.** Xie Y, Choi T, Al-Aly Z. Association of Treatment With Nirmatrelvir and the Risk of Post-COVID-19 Condition. JAMA Intern Med. 2023 Jun 1;183(6):554-564. **15.** Menni et al. Symptom prevalence, duration, and risk of hospital admission in individuals infected with SARS-CoV-2 during periods of omicron and delta variant dominance: a prospective observational study from the ZOE COVID Study. Lancet. 2022;399:1618-24 **16.** Socialstyrelsen. Övergripande statistik covid-19. 2024 02 September 2024]; Available from: https://www.socialstyrelsen.se/globalassets/sharepoint-dokument/dokument-webb/statistik/statistik-covid19-overgripande.xlsx. **17.** Pfizer - data on file, EPIC HR clinical study report, release 08 March 2023 (Section 6). 2023.

ACKNOWLEDGEMENTS

We acknowledge HEOR UK for their contributions to the model conceptualisation and development.

DISCLOSURES

This study was funded by Pfizer AB. C Gerdesköld, M Aldvén and F Nilsson are employed by Pfizer AB, Sweden. T Mugwagwa is employed by Pfizer Inc., UK.