CO69

Indirect Treatment Comparison of Iptacopan versus Eculizumab and Ravulizumab for Patients with Paroxysmal Nocturnal Hemoglobinuria Naive to C5 Inhibitors

Régis Peffault de Latour,[1] Phillip Scheinberg,[2] Paul Spin,[3] Maria-Magdalena Balp,[4] Katharina Pannagl,[5] Anggie Wiyani,[5] Olivier Somenzi,[4] Josefin Snellman,[4] Pearl Wang,[3] Jason Steenkamp,[3] Austin Kulasekararaj[6]

[1]French Référence Center for Aplastic Anemia and Paroxysmal Nocturnal Hemoglobinuria, Paris, France; [2]Hospital A Beneficência Portuguesa, São Paulo, Brazil; [3]EVERSANA, Burlington, Ontario, Canada; [4]Novartis Pharma AG, Basel, Switzerland; [5]Novartis Pharmaceuticals UK Ltd, London, UK; [6]King's College Hospital NHS, London, UK

KEY FINDINGS & CONCLUSIONS

- The results of this ITC suggest that iptacopan is more effective than eculizumab and ravulizumab, respectively, in reducing LDH levels and transfusion rate in C5i-naive PNH patients.
- Conclusions from this study should be interpreted in the context of this being an ITC.

This research was funded by Novartis Pharma AG.
Poster presented at the International Society for Pharmacoeconomics and Outcomes Research (ISPOR), Barcelona, Spain, 17-20 November 2024.

INTRODUCTION

- Paroxysmal nocturnal hemoglobinuria (PNH) is a rare blood condition characterized by hemolysis, anemia and thrombosis.[1]
- The first approved drugs for PNH were complement 5 inhibitor (C5i) infusions; eculizumab,[2] and later ravulizumab.[3]
- Iptacopan is the first-in-class, oral monotherapy, factor B inhibitor recently approved for adults with PNH who have hemolytic anemia[4,5] which was not compared in a head-to-head (H2H) trial vs the above drugs in patients who were not previously treated with C5i (named hereafter C5i-naive).
- In the absence of H2H trials, indirect treatment comparisons (ITC) are commonly performed to compare efficacy of two treatments in the context of Health Technology Assessment.[6-8]

AIM

- To conduct an ITC of iptacopan vs eculizumab and ravulizumab for the treatment of patients with PNH who were C5i-naive.

METHODS

- A systematic literature review identified Study 301 (NCT02946463; ravulizumab vs eculizumab),[9] a phase 3 trial, as the most relevant comparator trial for the single-arm phase 3 trial of iptacopan APPOINT-PNH (NCT04820530),[10] in the target population.
- ITC feasibility assessment (trial design, inclusion/exclusion criteria, and endpoints) concluded that unanchored matching-adjusted indirect comparison (MAIC) was most suitable, using individual patient data (IPD) from APPOINT-PNH[10] for iptacopan (N=40) and published aggregated data from Study 301[9] for eculizumab (N=121) and ravulizumab (N=125).
- Outcomes assessed were lactate dehydrogenase (LDH; mean % change from baseline [CFB]) and transfusion rate per patient-month and reported as difference between iptacopan vs eculizumab and iptacopan vs ravulizumab.

Statistical Analysis MAIC

- Due to high overlap in the eligibility criteria of the trials, no patient in APPOINT-PNH was excluded for this analysis.
- Patients from APPOINT-PNH were re-weighted via entropy balancing[7] to match patient characteristics in Study 301; adjusting for age, gender, % transfusion-free 12 months prior, LDH at baseline and history of major adverse vascular events (MAVE) after consulting with clinicians and health economics experts.
- The percent change from baseline LDH was derived by fitting a mixed model for repeated measures to the reweighted IPD from APPOINT-PNH. The treatment effect between iptacopan vs eculizumab and iptacopan vs ravulizumab was then derived as the difference between the adjusted mean CFB for iptacopan and the published mean CFB for eculizumab and ravulizumab.
- Transfusion rate per patient-month was reported by adjusting for differences in assessment windows between trials and were modelled using a log-Poisson distribution. This approach makes the assumption of a constant rate of transfusion events throughout the follow-up period.

RESULTS

- After matching and adjusting, baseline characteristics were similar across trials, with an effective sample size of 31 for APPOINT-PNH (**Table 1**).

Table 1. Baseline characteristics in APPOINT-PNH and Study 301

Baseline characteristics	Study 301, N=246	Before matching and adjusting		After matching and adjusting	
	Eculizumab, N=121 Ravulizumab, N=125	APPOINT-PNH Iptacopan, N=40	SMDs*	APPOINT-PNH Iptacopan, ESS=31	SMDs*
Age, mean (SD)	45.5 (15.7)	42.1 (15.9)	0.216	45.5 (15.7)	0.000
Gender, male, n (%)	134 (54.5)	23 (57.5)	0.061	54.5	0.001
Transfusion free 12 months prior, n (%)	44 (17.9)	12 (30.0)	0.342	17.8	0.000
LDH, U/L, mean (SD)	1606.4 (752.7)	1698.8 (683.3)	0.129	1606.4 (684.7)	0.000
History of MAVE, n (%)	42 (17.1)	5 (12.5)	0.129	17.1	0.001

*SMD ≤ 0.1 indicates small difference. ESS: effective sample size; LDH: lactate dehydrogenase; MAVE: major adverse vascular events; SD: standard deviation; SMD: standardized mean difference

Change from baseline in LDH

- The mean % CFB in LDH (reduction in LDH) was higher for iptacopan (-85.08%) compared to eculizumab (-76.02%) and ravulizumab (-76.84%) (**Figure 1**).
- The mean difference in % CFB in LDH was significantly in favor of iptacopan compared to eculizumab and ravulizumab.
 - Iptacopan vs. eculizumab: -9.061%, p=0.0005
 - Iptacopan vs. ravulizumab: -8.241%, p=0.0013

Figure 1. Percent change from baseline in LDH, and mean difference between treatments

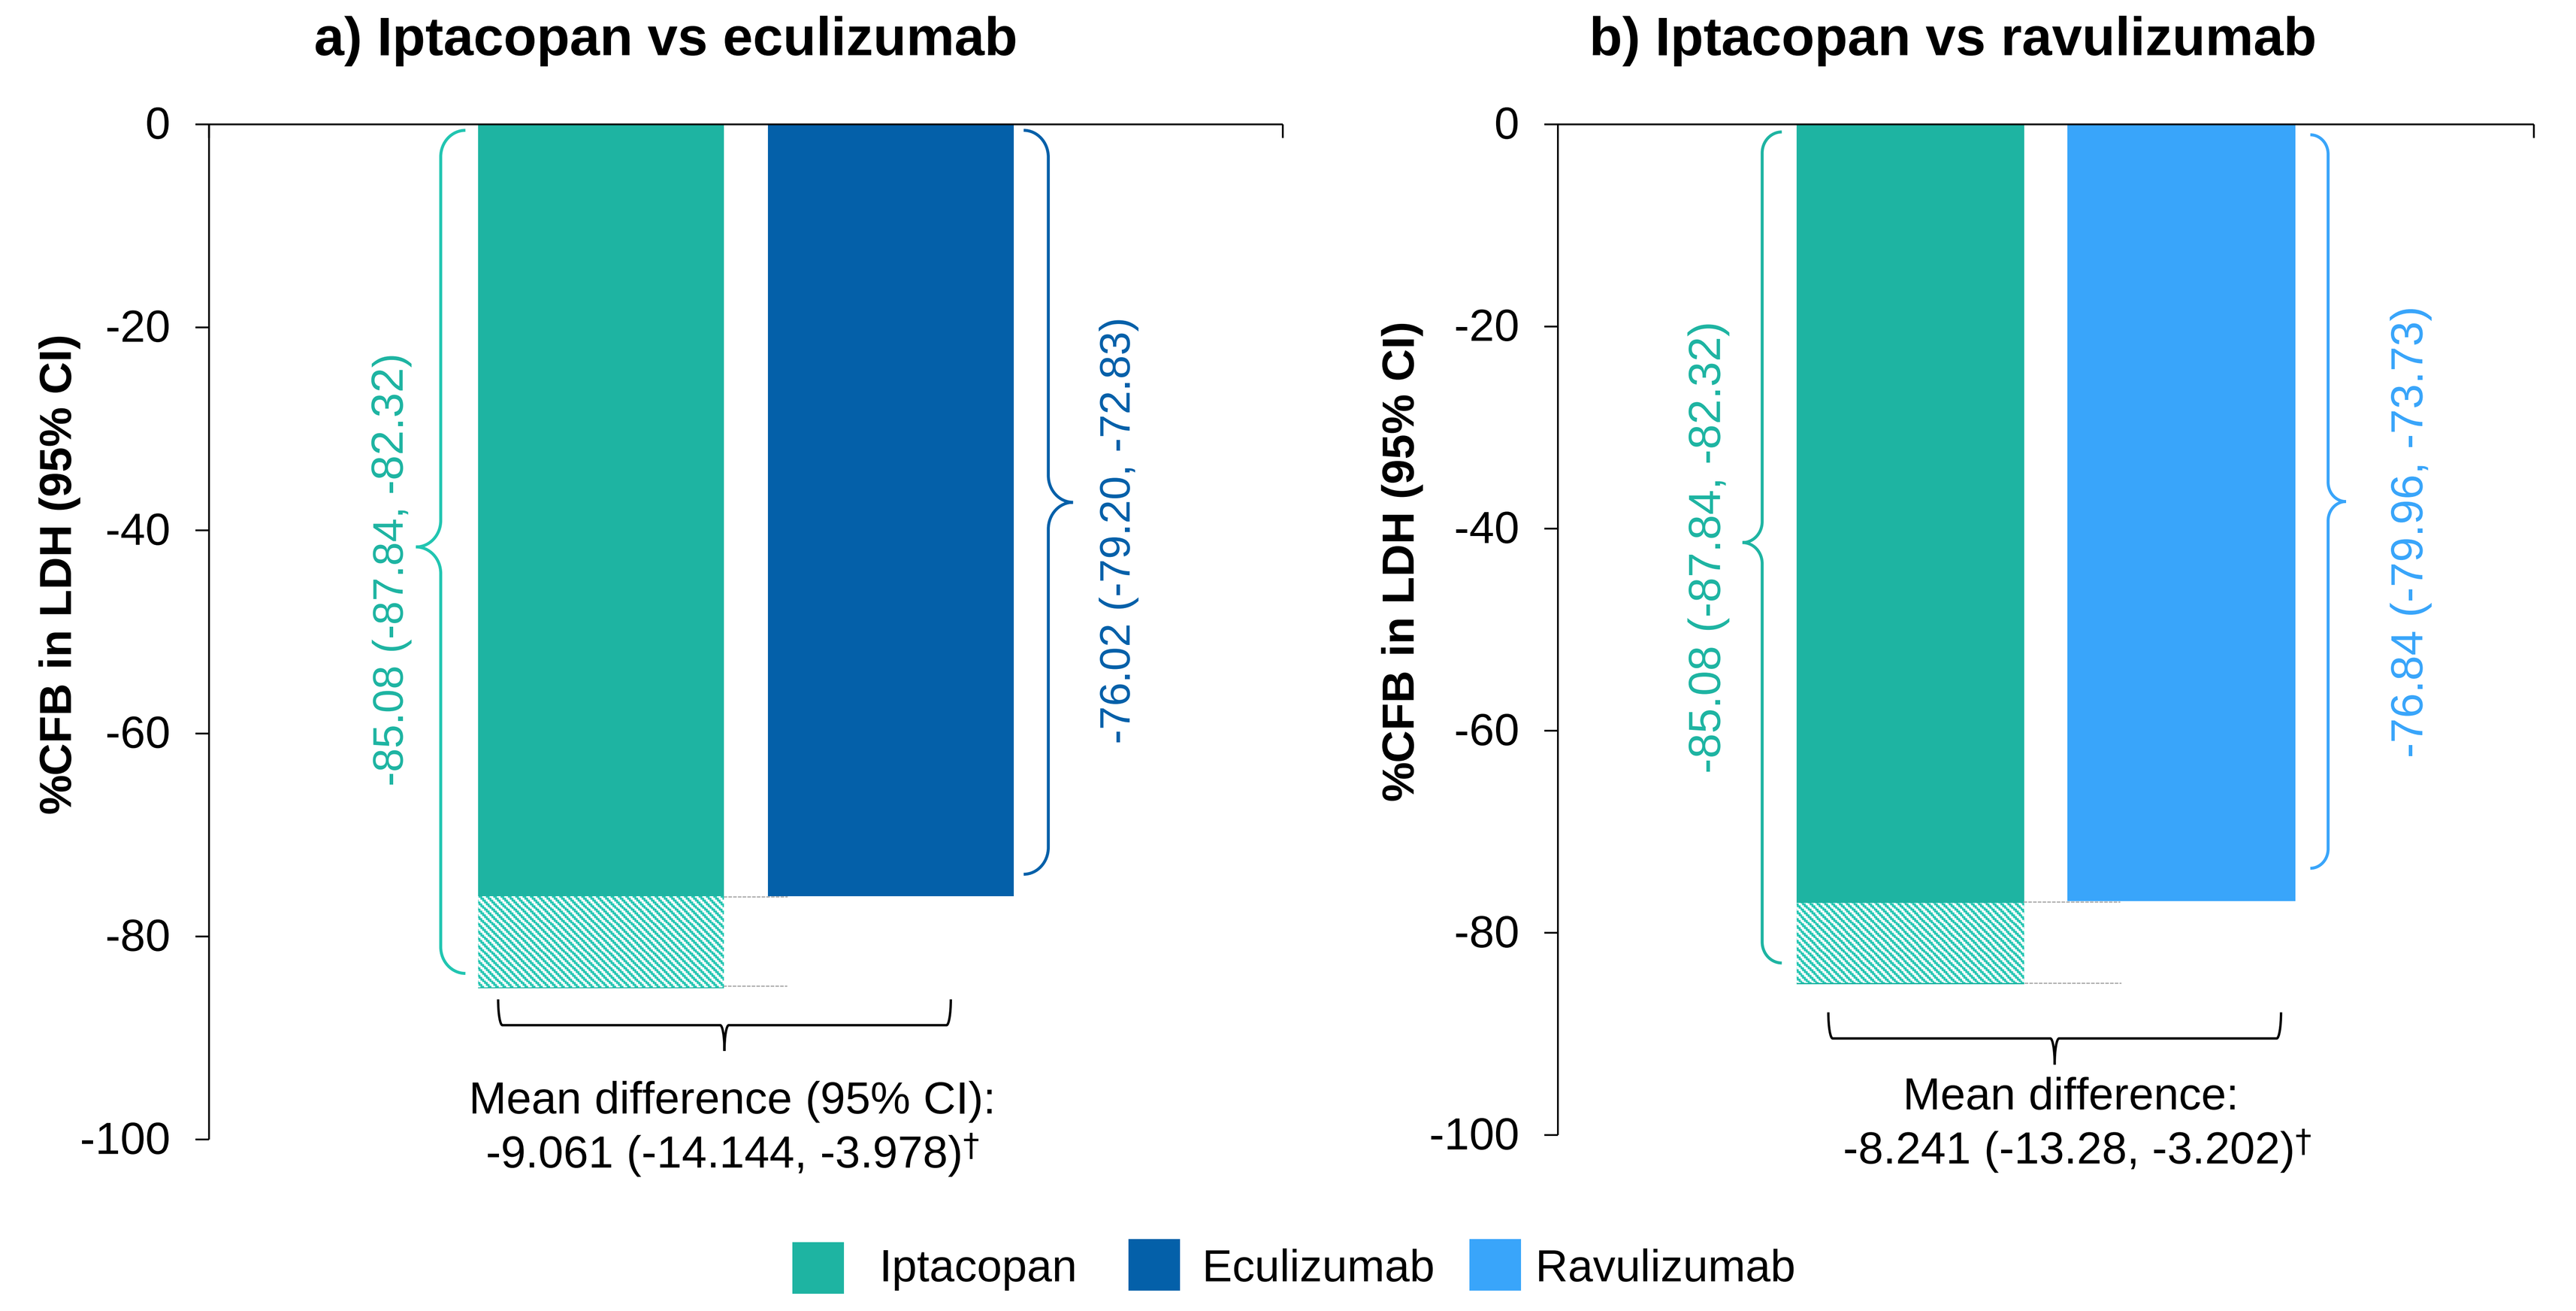
a) Iptacopan vs eculizumab
b) Iptacopan vs ravulizumab
%CFB in LDH (95% CI)
0
-20
-40
-60
-80
-100
-85.08 (-87.84, -82.32)
-76.02 (-79.20, -72.83)
-76.84 (-79.96, -73.73)
Mean difference (95% CI):
-9.061 (-14.144, -3.978)†
Mean difference:
-8.241 (-13.28, -3.202)†
Iptacopan
Eculizumab
Ravulizumab

CFB: change from baseline; CI: confidence interval; LDH: lactate dehydrogenase. †P < 0.05

Transfusion rate

- The transfusion rate per patient-month was lower for iptacopan (0.00037) compared to eculizumab (0.050) and ravulizumab (0.045) (**Table 2**).
- The rate ratio of iptacopan vs. eculizumab, and vs. ravulizumab, respectively, <1 suggests a significantly lower need for transfusions for patients treated with iptacopan (**Table 2**).

Table 2. Transfusion rate per patient-month and rate ratio

Transfusion rate	Eculizumab	Ravulizumab	Iptacopan
Transfusion rate per patient-month (95% CI)	0.050 (0.041, 0.060)	0.045 (0.033, 0.060)	0.00037 (0.00025, 0.00053)
Rate ratio (95% CI)†; p-value			
Iptacopan vs eculizumab	-	-	0.0065 (0.0042, 0.0101) p=0.0004
Iptacopan vs ravulizumab	-	-	0.0082 (0.0052, 0.0131) p=0.0008

†Rate ratio <1 implies a lower rate of transfusion for iptacopan. Results are statistically significant when the CI does not contain the null value (1.00). CI: confidence interval.

References

1. Brodsky RA et al. *Blood*. 2014;124:2804–11.
2. Food and Drug Administration (FDA). Soliris (eculizumab) package insert. Accessed on: September 23, 2024. Available at: https://www.accessdata.fda.gov/drugsatfda_docs/label/2020/125166s434lbl.pdf.
3. Food and Drug Administration (FDA). Ultomiris (ravulizumab) package insert. Accessed on: September 23, 2024. Available at: https://www.accessdata.fda.gov/drugsatfda_docs/label/2022/761108s021lbl.pdf.
4. European Medicines Agency (EMA). First oral monotherapy for patients with paroxysmal nocturnal haemoglobinuria. Accessed on: September 23, 2024. Available at: https://www.ema.europa.eu/en/news/first-oral-monotherapy-patients-paroxysmal-nocturnal-haemoglobinuria#:~:text=First%20oral%20monotherapy%20for%20patients%20with%20paroxysmal%20nocturnal%20haemoglobinuria,-Share&text=EMA%20has%20recommended%20granting%20.
5. Food and Drug Administration (FDA). Fabhalta (iptacopan) package insert. Accessed on: September 23, 2024. Available at: https://www.accessdata.fda.gov/drugsatfda_docs/label/2024/218276s001lbl.pdf#page=19.
6. NICE health technology evaluations: the manual. Available from: https://www.nice.org.uk/process/pmg36/resources/nice-health-technology-evaluations-the-manual-pdf-72286779244741.
7. Phillippo DM et al. NICE DSU Technical Support Document 18: Methods for population-adjusted indirect comparisons in submission to NICE. 2016. Available from www.nicedsu.org.uk.
8. Phillippo DM et al. *Int J Technol Assess Health Care*. 2019;35(3): 221-28.
9. Lee JW et al. *Blood*. 2019 Feb 7;133(6):530-9.
10. Peffault de Latour R et al. *N Engl J Med*. 2024 Mar 14;390(11):994-1008.

Acknowledgements

The authors acknowledge **Kolli Narotham Reddy** (Novartis, Hyderabad) for preparing poster content and **Srinivas Kanchipati** for designing the poster layout. The final responsibility for the content lies with the authors.

Scan to obtain:
- Poster

https://www.medicalcongressposters.com//Default.aspx?doc=a5d91
Copies of this poster obtained through Quick Response (QR) code are for personal use only and may not be reproduced without permission of the authors.