MSR148

Partial Adjustment for Treatment Switching to Represent Expected Switching in Clinical Practice

Bell Gorrod, H[1], Mt-Isa, S[2], White, IR[3], Xuan, J[3], Malbecq, W[4], Vandormael, K[5], Yorke-Edwards, V[6], Latimer, NR[1,7]

1 Sheffield Centre for Health and Related Research, University of Sheffield 2 Biostatistics and Research Decision Sciences (BARDS) Health Technology Assessment (HTA) Statistics, MSD, Zurich 3 MRC Clinical Trials Unit at UCL, University College London 4 Department of Mathematics, University of Brussels 5 Biostatistics and Research Decision Sciences (BARDS) Health Technology Assessment (HTA) Statistics, MSD, Brussels 6 Centre for Advanced Research Computing, University College London 7 Delta Hat Limited, UK

Background:

Treatment switching is when patients in a randomised control trial (RCT) switch from the treatment they were randomised to receive, onto another treatment or treatments.[1-4]

In Health Technology Assessment:

- If the treatment switched onto is unavailable in clinical practice (CP), then ***full adjustment*** should be made to estimate outcomes in the absence of treatment switching
- If patients make a switch to a treatment available in CP, but the proportion of switchers in the randomised controlled trial (RCT) differs from the proportion of switchers expected in CP, a ***partial adjustment*** for treatment switching may provide suitable estimates.

Study Objectives:

- We introduce a **new estimand**.
 If there is x% of switchers in an RCT, but we expect y% of switchers in CP, methods to partially adjust for treatment switching can estimate what would have happened if y% had switched in the RCT.
- We **describe methods** to partially adjust for treatment switching, which involve adaptations to existing inverse probability of censoring weights (IPCW) and two-stage estimation (TSE) methods.
- We **assess the performance** of adapted methods for the new estimand in a simulation study.

Methods:

Partial adjustment for treatment switching

Step 1: Assign non-switching switchers (NSS)

- To partially adjust for treatment switching, a proportion of switchers in the RCT, (x-y)%, must be assigned as **non-switching switchers (NSS)**, where x is the proportion of switchers in the RCT and y is the proportion of switchers expected in CP.
- By assigning switchers as NSS, we are selecting these switchers to be adjusted to represent non-switchers, to allow estimation of the target estimand.
- We **describe 2 approaches for selecting NSS,** see Figure 1 and Table 1 of online supplementary material for details.
 1. **Random allocation** with bootstrapping
 2. Allocation based on **modelled probability** of switch

Step 2: Apply adjustment method

- Adapted versions of inverse probability of censoring weights (IPCW) and the Two-stage estimation (TSE) method, using each approach to assign NSS, are described in the online supplementary materials.

Figure 1: Visualisation of non-switching switcher (NSS) assignment

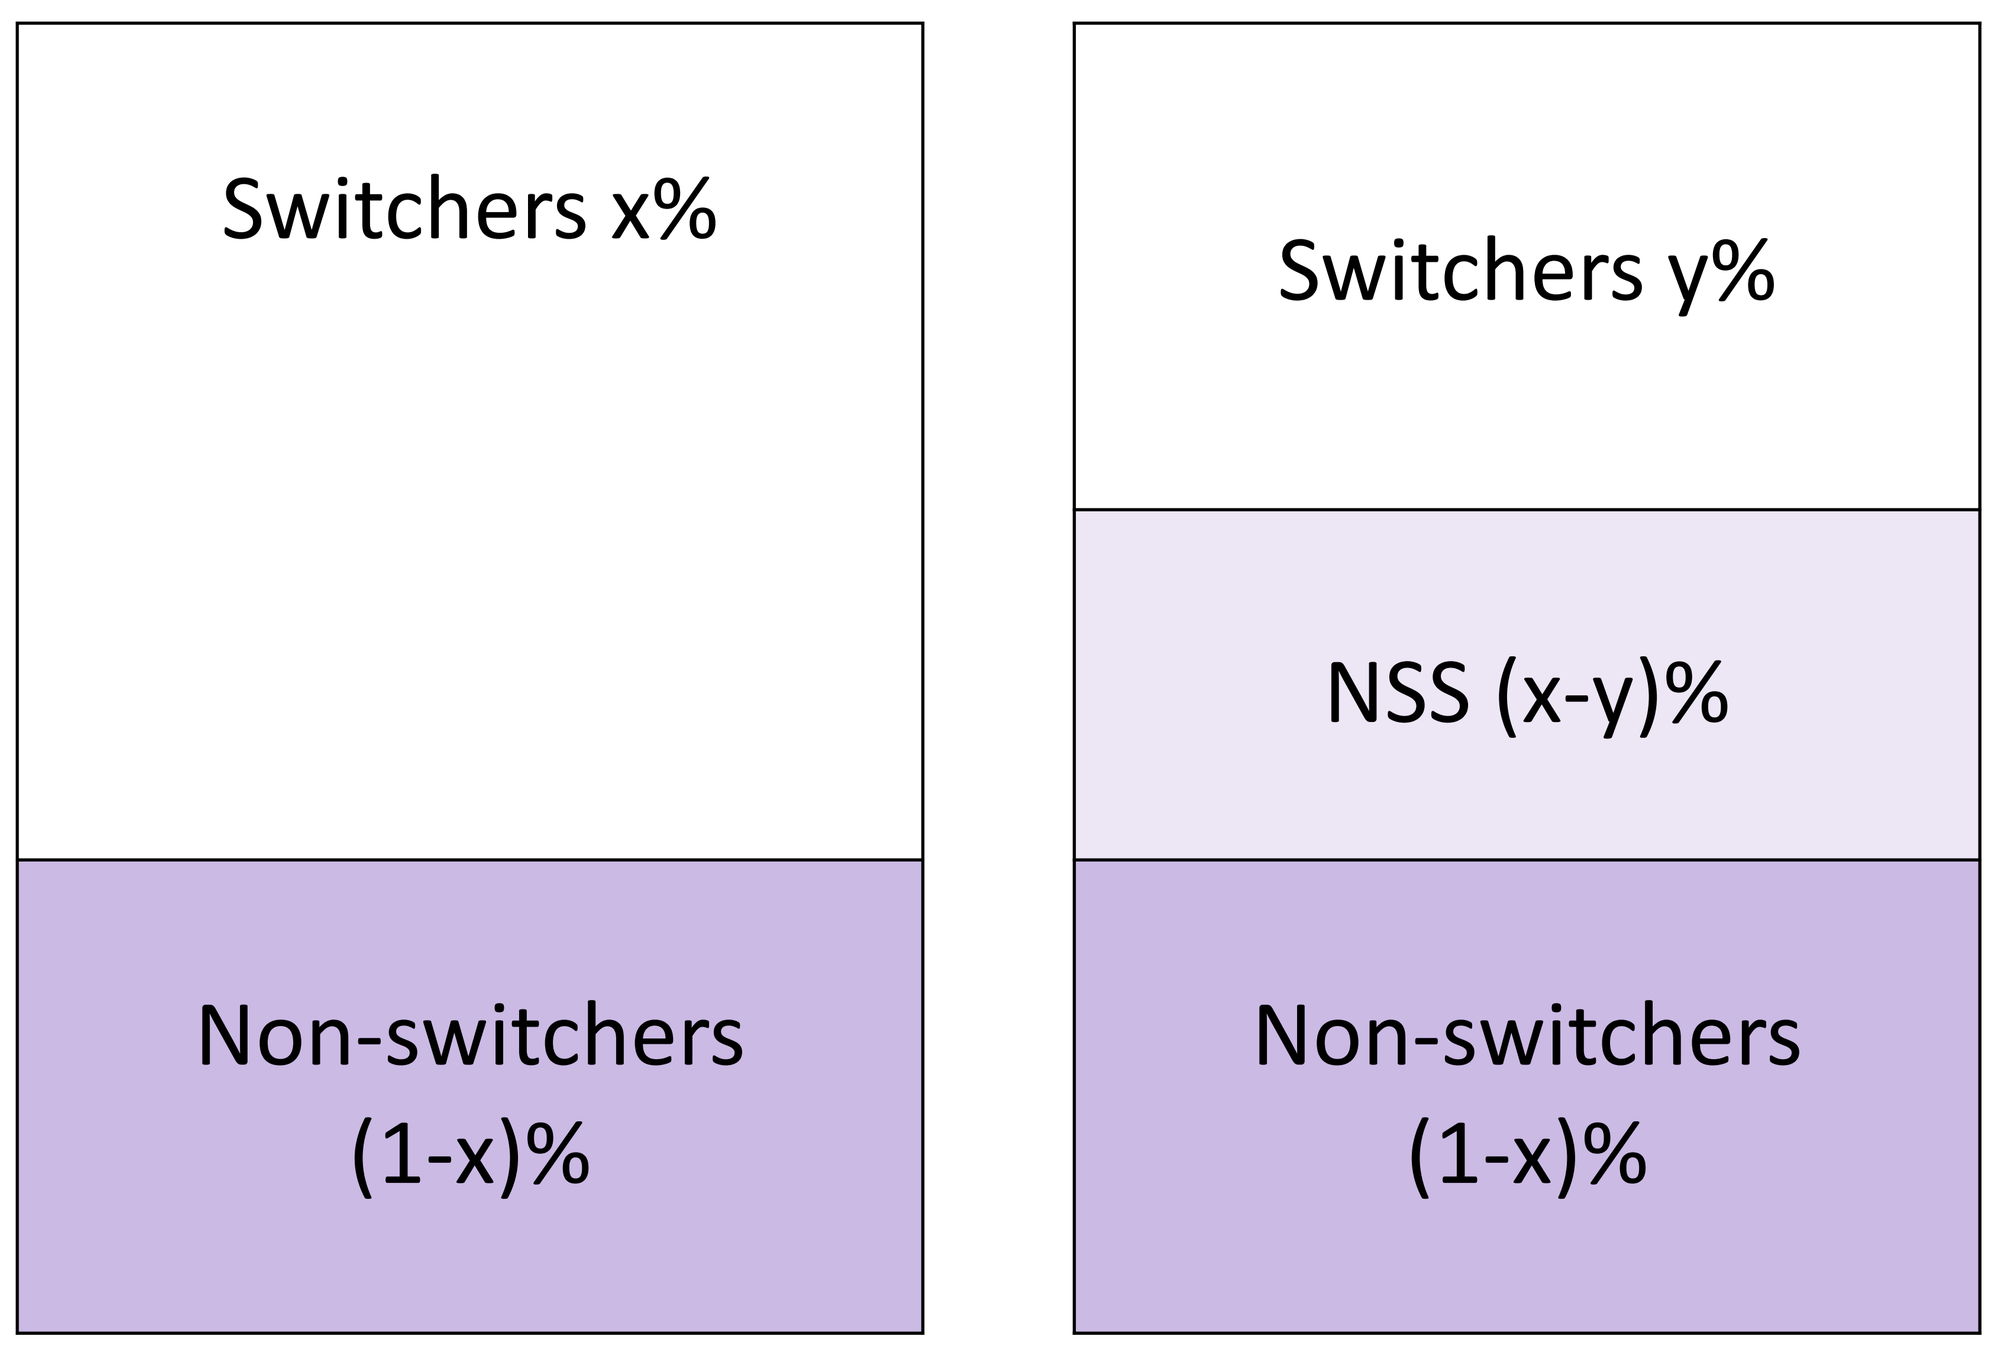
Switchers x%
Non-switchers (1-x)%
Switchers y%
NSS (x-y)%
Non-switchers (1-x)%

Figure 1(a) Figure 1(b)

x% represents the proportion of switchers observed in one arm of an RCT, y% represents the proportion of switchers expected in clinical practice. i.e. the proportion of switchers in our target estimand. Figure 1(a) represents the observed proportions of switchers and non-switchers in one arm of an RCT. Figure 1(b) shows (x-y)% of switchers assigned as NSS and y% of switchers now represents what is expected in CP.

Glossary

RCT –randomised controlled trial
CP – Clinical Practise
NSS – Non-switching Switcher
ADEMP - aims, data-generating mechanisms, estimands, methods, and performance measures
DGM – Data generating mechanism
RMST – Restricted mean survival time

Treatment switching adjustment methods

IPCW – Inverse probability of censoring weights
- Involves applying weights to non-censored patients so they represent themselves and censored patients

TSE – Two-stage estimation
- Involves estimating a post-progression treatment effect, then using that to adjust survival times.

For more information on the methods in the context of partial adjustment, see Table 1 in the online supplementary material.

Online Supplementary material available at https://sites.google.com/sheffield.ac.uk/supplementarymaterial-msr148/home or using QR code

Simulation Study Design

- The study was designed according to the ADEMP structure recommended by Morris et al (2019).[5]
- **Aims** To test the performance of two different approaches to partial treatment switching adjustment, applied to IPCW and TSE methods.
- **Performance** is tested by applying adjustment methods to simulated RCT data to assess how close the results are to the simulated truth.
- **Data Generating Mechanisms (DGM)**
 1) generate data with an assigned switching proportion reflecting that expected in CP. Calculate the restricted mean survival time (RMST) in this data. This forms our "truth"
 2) generate simulated RCT data with a switching proportion that is different from that expected in CP.
- **Estimand** RMST in the control group that would have been observed with the proportion of treatment switching expected in CP.
- **Methods** Adapted versions of IPCW and TSE using two different approaches for allocating NSS, as described in Table 1 of the online supplementary materials.

Parameters We varied the proportion of switchers in the RCT, the proportion of switchers in the CP, the underlying treatment effects and the treatment effect of the switched onto treatment. See Table 2 of online supplementary material for more details.

Results:

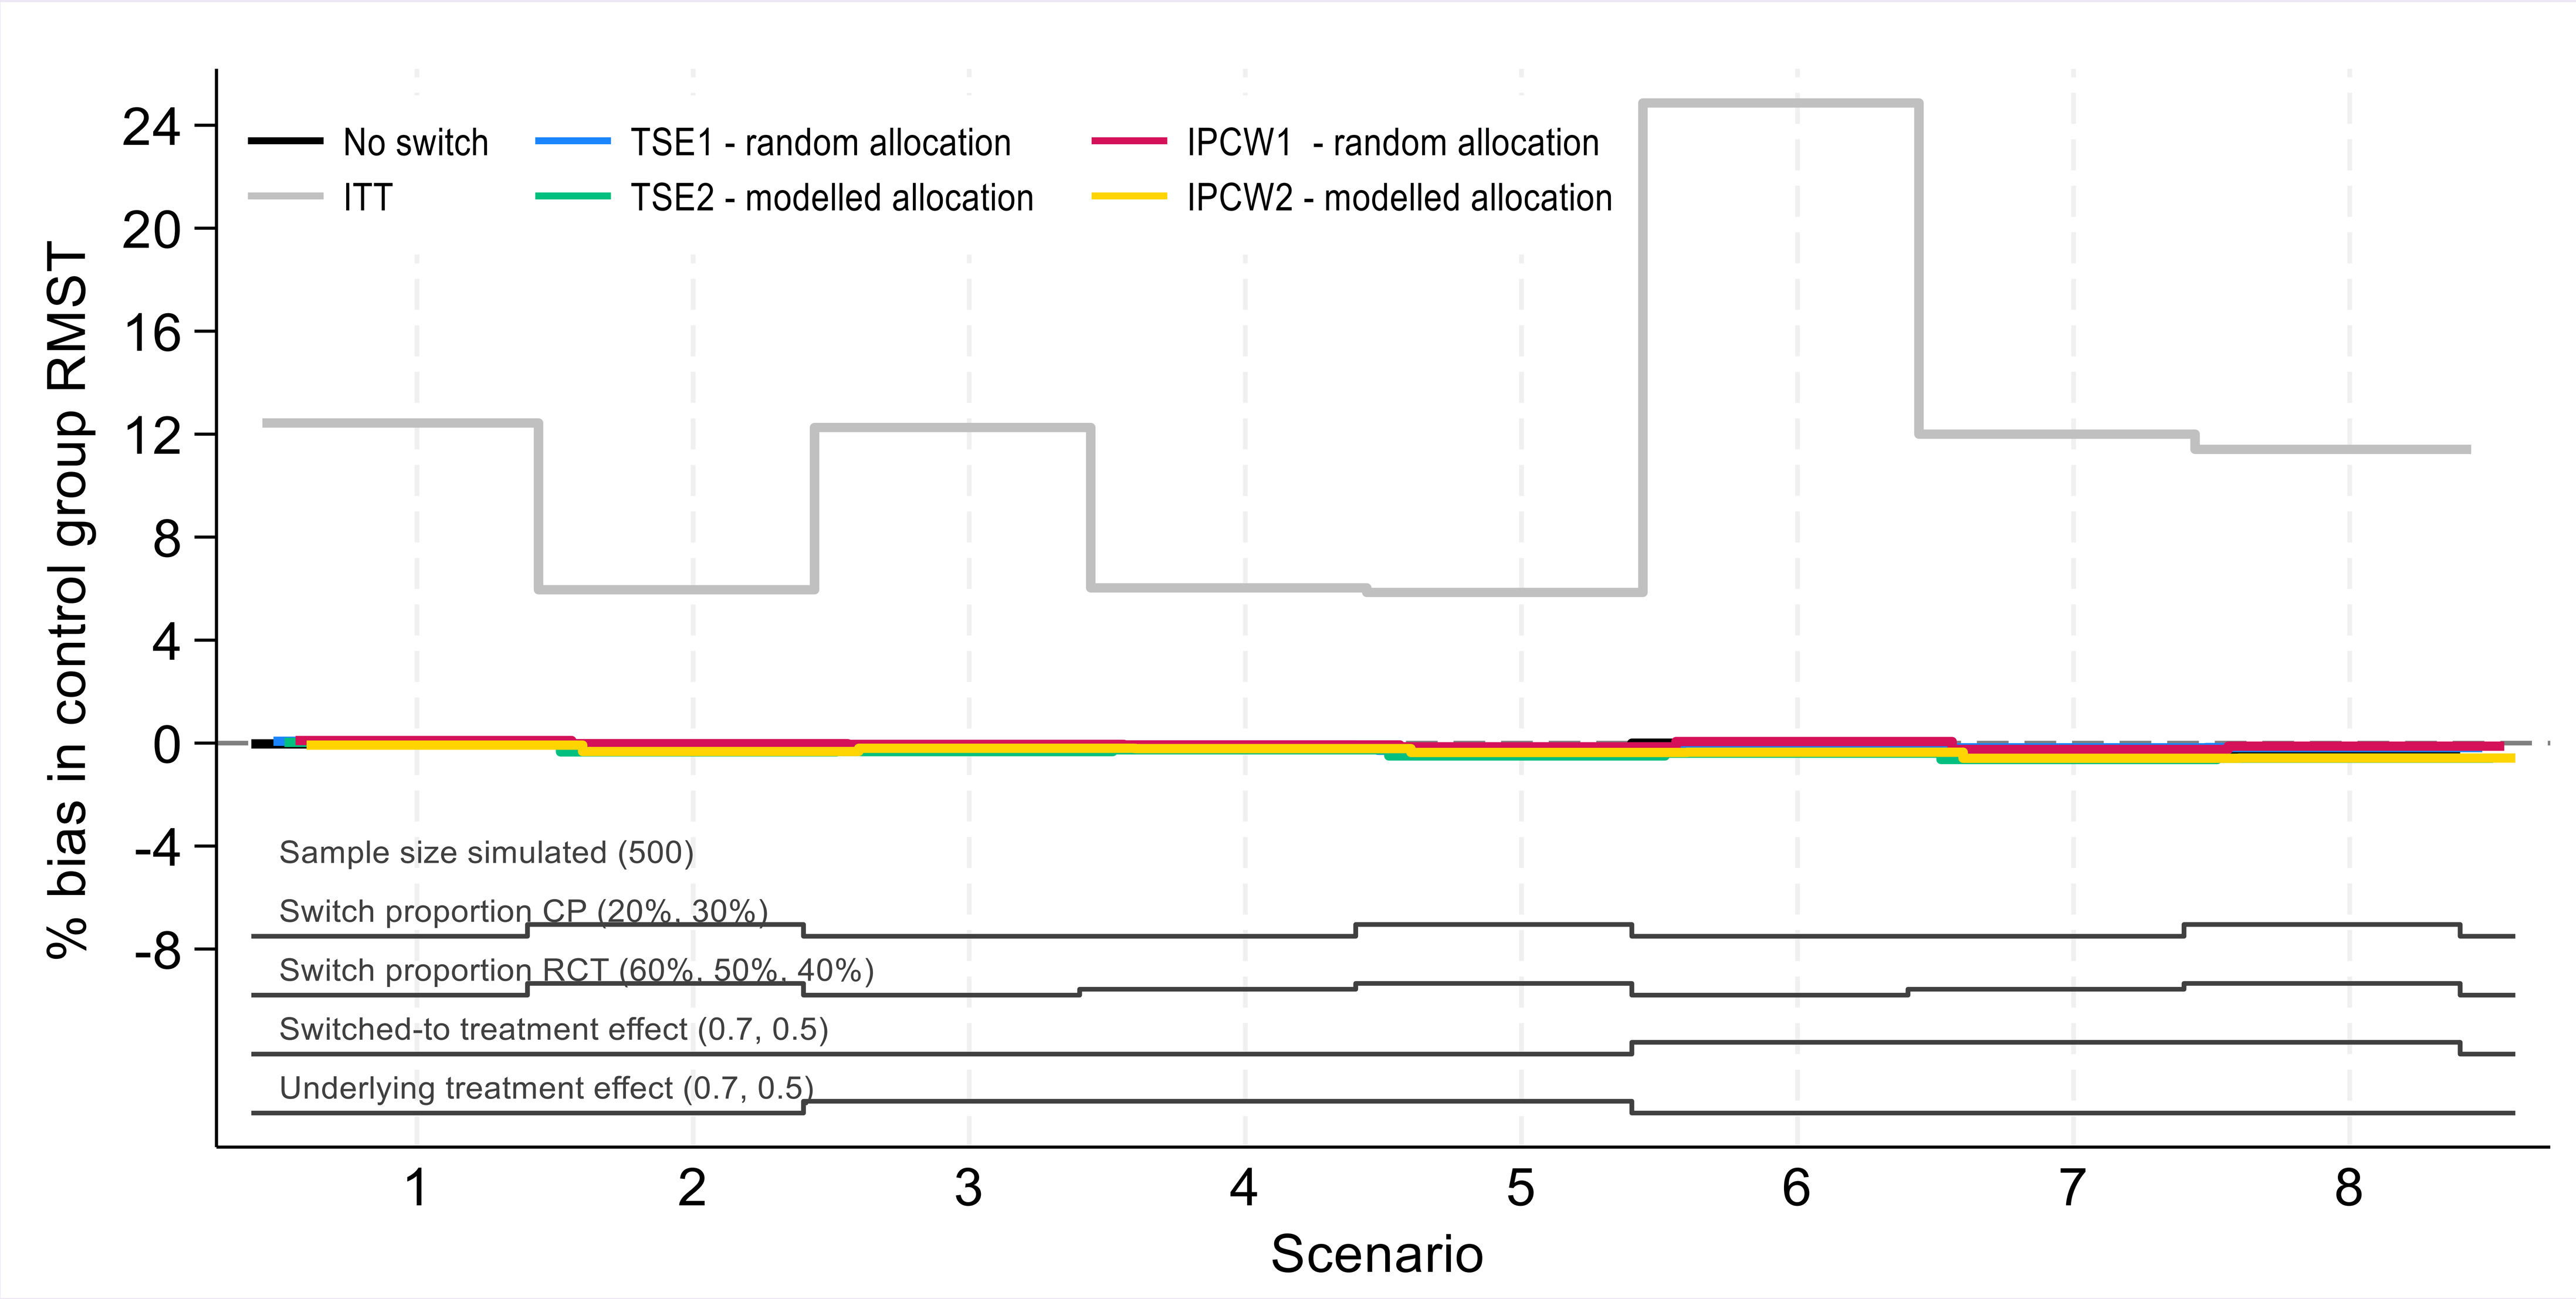
No switch
ITT
TSE1 - random allocation
TSE2 - modelled allocation
IPCW1 - random allocation
IPCW2 - modelled allocation
% bias in control group RMST
24
20
16
12
8
4
0
-4
-8
Sample size simulated (500)
Switch proportion CP (20%, 30%)
Switch proportion RCT (60%, 50%, 40%)
Switched-to treatment effect (0.7, 0.5)
Underlying treatment effect (0.7, 0.5)
1
2
3
4
5
6
7
8
Scenario

- TSE1, TSE2, IPCW1 and IPCW2 produced similar results across scenarios.
- All methods produced less bias that the RCT intention-to-treat (ITT) analysis.

Conclusions:

- We introduced a new estimand for situations when the proportion of switchers in an RCT does not reflect the proportion of switchers expected in CP.
- We introduced adapted IPCW and TSE methods to adjust for partial treatment switching.
- Method performance was compared in a simulation study.
- All methods tested produced less biased estimates of the simulated CP truth than the RCT ITT analysis.
- The random and modelled approaches performed similarly well.

Directions for further research:

- Scenarios 1-8 assume that switching can only take place at the time of disease progression. Additional scenarios could allow switching to also take place after disease progression.
- We assumed for simplicity that all patients in the RCT experience disease progression. The methods could be extended for the situation where only some patients experience disease progression.
- In scenarios 1-8, the proportion of switchers in the RCT is greater than the proportion of switchers expected in CP. The methods can be adapted for the situation where the proportion of switchers in CP is greater than the proportion of switchers in the RCT.

References: 1. Latimer, N.R., et al., Adjusting survival time estimates to account for treatment switching in randomized controlled trials—an economic evaluation context: methods, limitations, and recommendations. Medical Decision Making, 2014. 34(3): p. 387-402. 2. Latimer, N.R. and K.R. Abrams, NICE DSU technical support document 16: adjusting survival time estimates in the presence of treatment switching. Report by the Decision Support Unit, 2014. 3. Bell Gorrod, H., N.R. Latimer, and K.R. Abrams, NICE DSU Technical Support Document 24 Adjusting survival time estimates in the presence of treatment switching: An update. Report by the Decision Support Unit, forthcoming. 4. Latimer, N.R., et al., Adjusting for treatment switching in randomised controlled trials–a simulation study and a simplified two-stage method. Statistical methods in medical research, 2017. 26(2): p. 724-751. 5. Morris TP, White IR, Crowther MJ. Using simulation studies to evaluate statistical methods. Statistics in medicine. 2019 May 20;38(11):2074-102.

MSD INVENTING FOR LIFE
University of Sheffield
UKRI MRC Clinical Trials Unit
UCL