Comparative effectiveness of echinocandins in the treatment of invasive candidiasis: A systematic review and network meta-analysis

CO145

L. Linsell[1], JC Thompson[1], EJ Manalastas[1], S. Dickerson[2], I. Bielicka[2]
[1]Visible Analytics, Oxford, Oxfordshire, UK; [2]Mundipharma Research Ltd, Cambridge, UK.

INTRODUCTION AND OBJECTIVES

- Invasive candidiasis is a severe invasive fungal infection associated with high mortality, morbidity and prolonged hospital stay.
- Echinocandins are recommended for first-line treatment due to their efficacy and low rates of toxicity.
- The aim of the study was to determine the relative efficacy of a new, weekly echinocandin (rezafungin) compared to daily echinocandins, on mortality and response.

METHODS

- A systematic literature review (SLR) was conducted in MEDLINE, EMBASE and CENTRAL to identify randomised controlled trials (RCTs) of licenced antifungal treatments reporting clinical outcomes in patients with invasive candidiasis.
- Studies evaluating azoles and polyenes were retained if they provided network connectivity for echinocandin treatments.
- Separate network meta-analyses (NMAs) were conducted for 4 clinical endpoints using Bayesian Markov chain Monte Carlo (MCMC) methods.
- Random-effects models using informative priors[1] for between-study heterogeneity were used to compare treatment effects where possible, otherwise fixed-effects models were fitted.

SYSTEMATIC REVIEW RESULTS

- The SLR was conducted on 18 October 2023 and identified 16 RCTs published between 1993 and 2021 (Figure 1).
- Of the 16 unique RCTs included in qualitative synthesis, six studies allowed for within-class echinocandins connectivity.
- A summary of the studies and the treatment arms included in the NMA are shown in Table 1.

Figure 1. PRISMA flow diagram

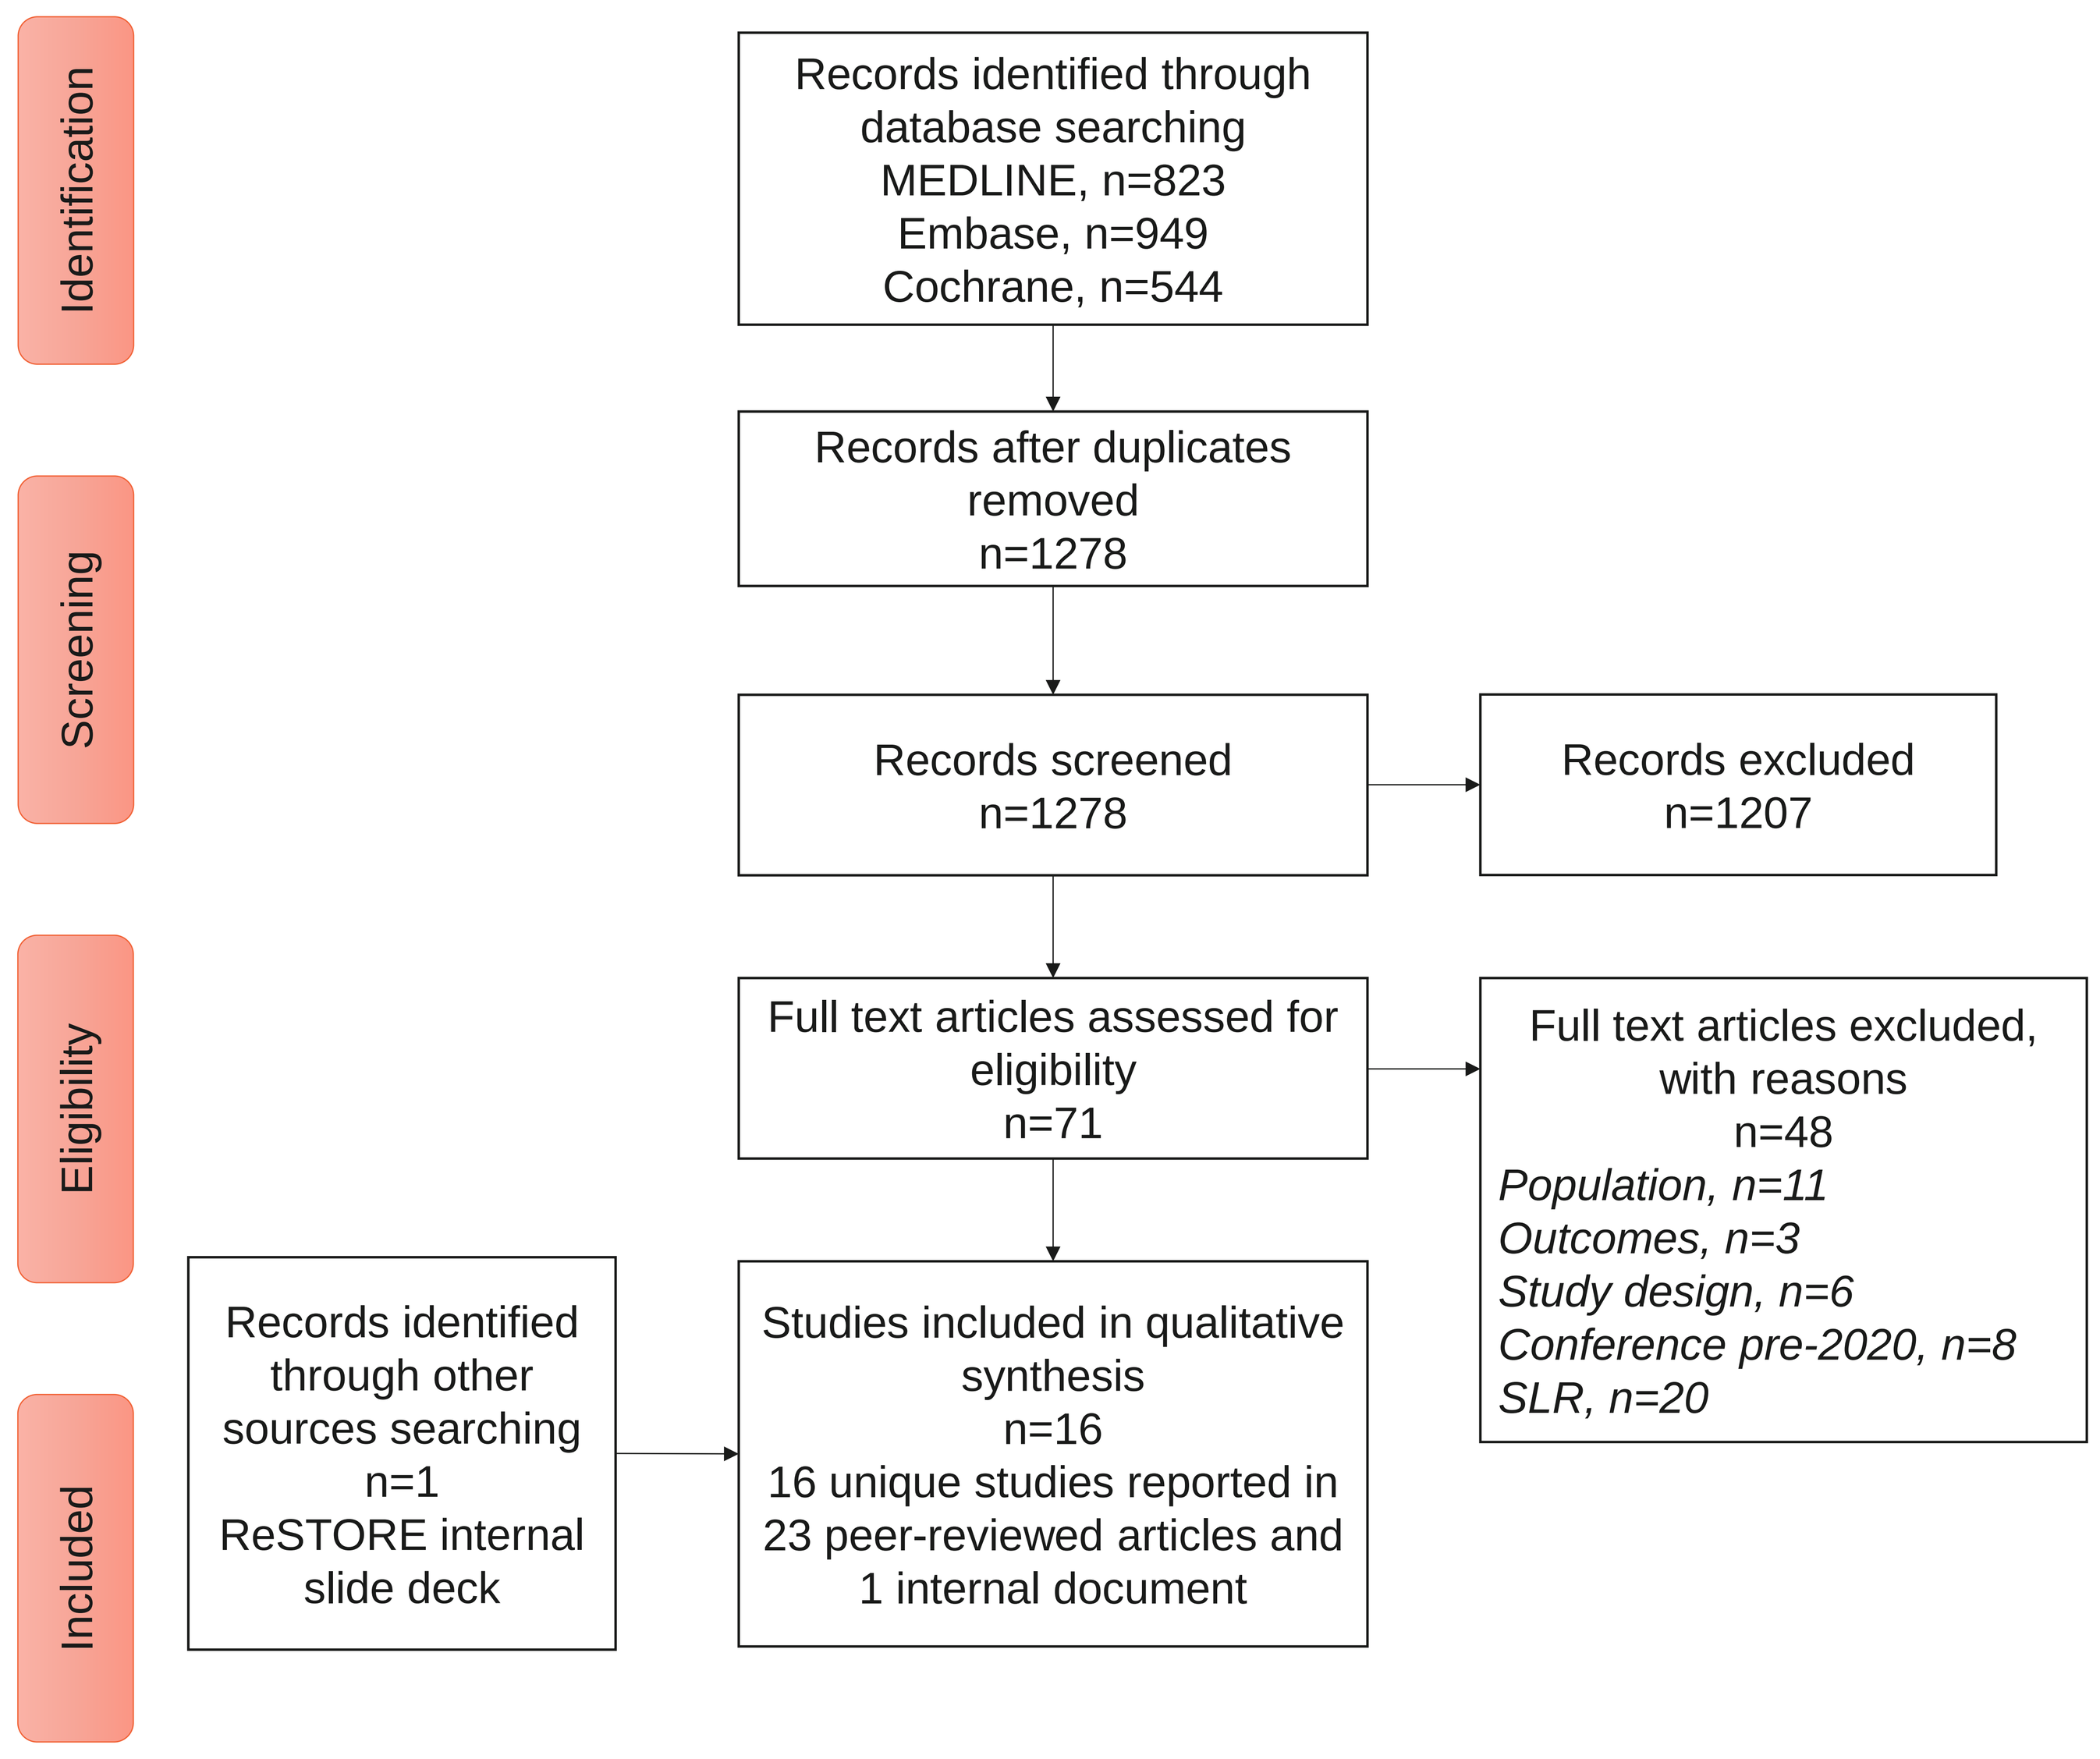
Identification
Screening
Eligibility
Included
Records identified through database searching
MEDLINE, n=823
Embase, n=949
Cochrane, n=544
Records after duplicates removed
n=1278
Records screened
n=1278
Records excluded
n=1207
Full text articles assessed for eligibility
n=71
Full text articles excluded, with reasons
n=48
Population, n=11
Outcomes, n=3
Study design, n=6
Conference pre-2020, n=8
SLR, n=20
Records identified through other sources searching
n=1
ReSTORE internal slide deck
Studies included in qualitative synthesis
n=16
16 unique studies reported in 23 peer-reviewed articles and 1 internal document

Table 1. Summary of the 6 studies and trial arms included in the NMA

StudyID	Study design	Country	Period of conduct	Treatment	Treatment class	Number of patients
ReSTORE[2] (Thompson 2023)	Phase 3 double-blind RCT	18 countries	Oct-18 to Oct-21	Rezafungin 400/200 Caspofungin 70/50	Echinocandin Echinocandin	100 99
STRIVE[3] (Thompson 2021)	Phase 2 double-blind RCT	10 countries (USA/ Europe)	Jul-16 to Apr-19	Rezafungin 400/200 Caspofungin 70/50	Echinocandin Echinocandin	57 69
Pappas 2007[4]	Phase 3 double-blind RCT	15 countries	Aug-04 to Apr-06	Micafungin 100 Caspofungin 70/50	Echinocandin Echinocandin	191 188
Mora-Duarte 2002[5]	Double-blind RCT	20 countries	Nov-97 to Jun-01	Caspofungin 70/50 Amphotericin B	Echinocandin Polyene	109 115
Reboli 2007[6]	Phase 3 double-blind RCT	USA	Mar-03 to Oct-04	Anidulafungin 100 Fluconazole 400	Echinocandin Azole	127 118
Phillips 1997[7]	RCT	Canada	Apr-91 to Jul-94	Fluconazole 400 Amphotericin B	Azole Polyene	50 53

NETWORK META-ANALYSIS RESULTS

- Six studies[2-7] were included in the NMA for mortality and global response (n=1276 randomised patients, Figure 2a), and three studies[2-4] were included for mycological and clinical response (n=704 randomised patients, Figure 2b).
- Comparators included once-daily: caspofungin (70/50mg) (reference treatment), micafungin (100mg) and anidulafungin (100mg) and once-weekly rezafungin (400/200mg).
- There were no significant differences between echinocandins for any of the endpoints, however the rezafungin trials were based on early eradication response (day 14) compared to end of treatment response in the other trials.

Figure 2. Network diagrams

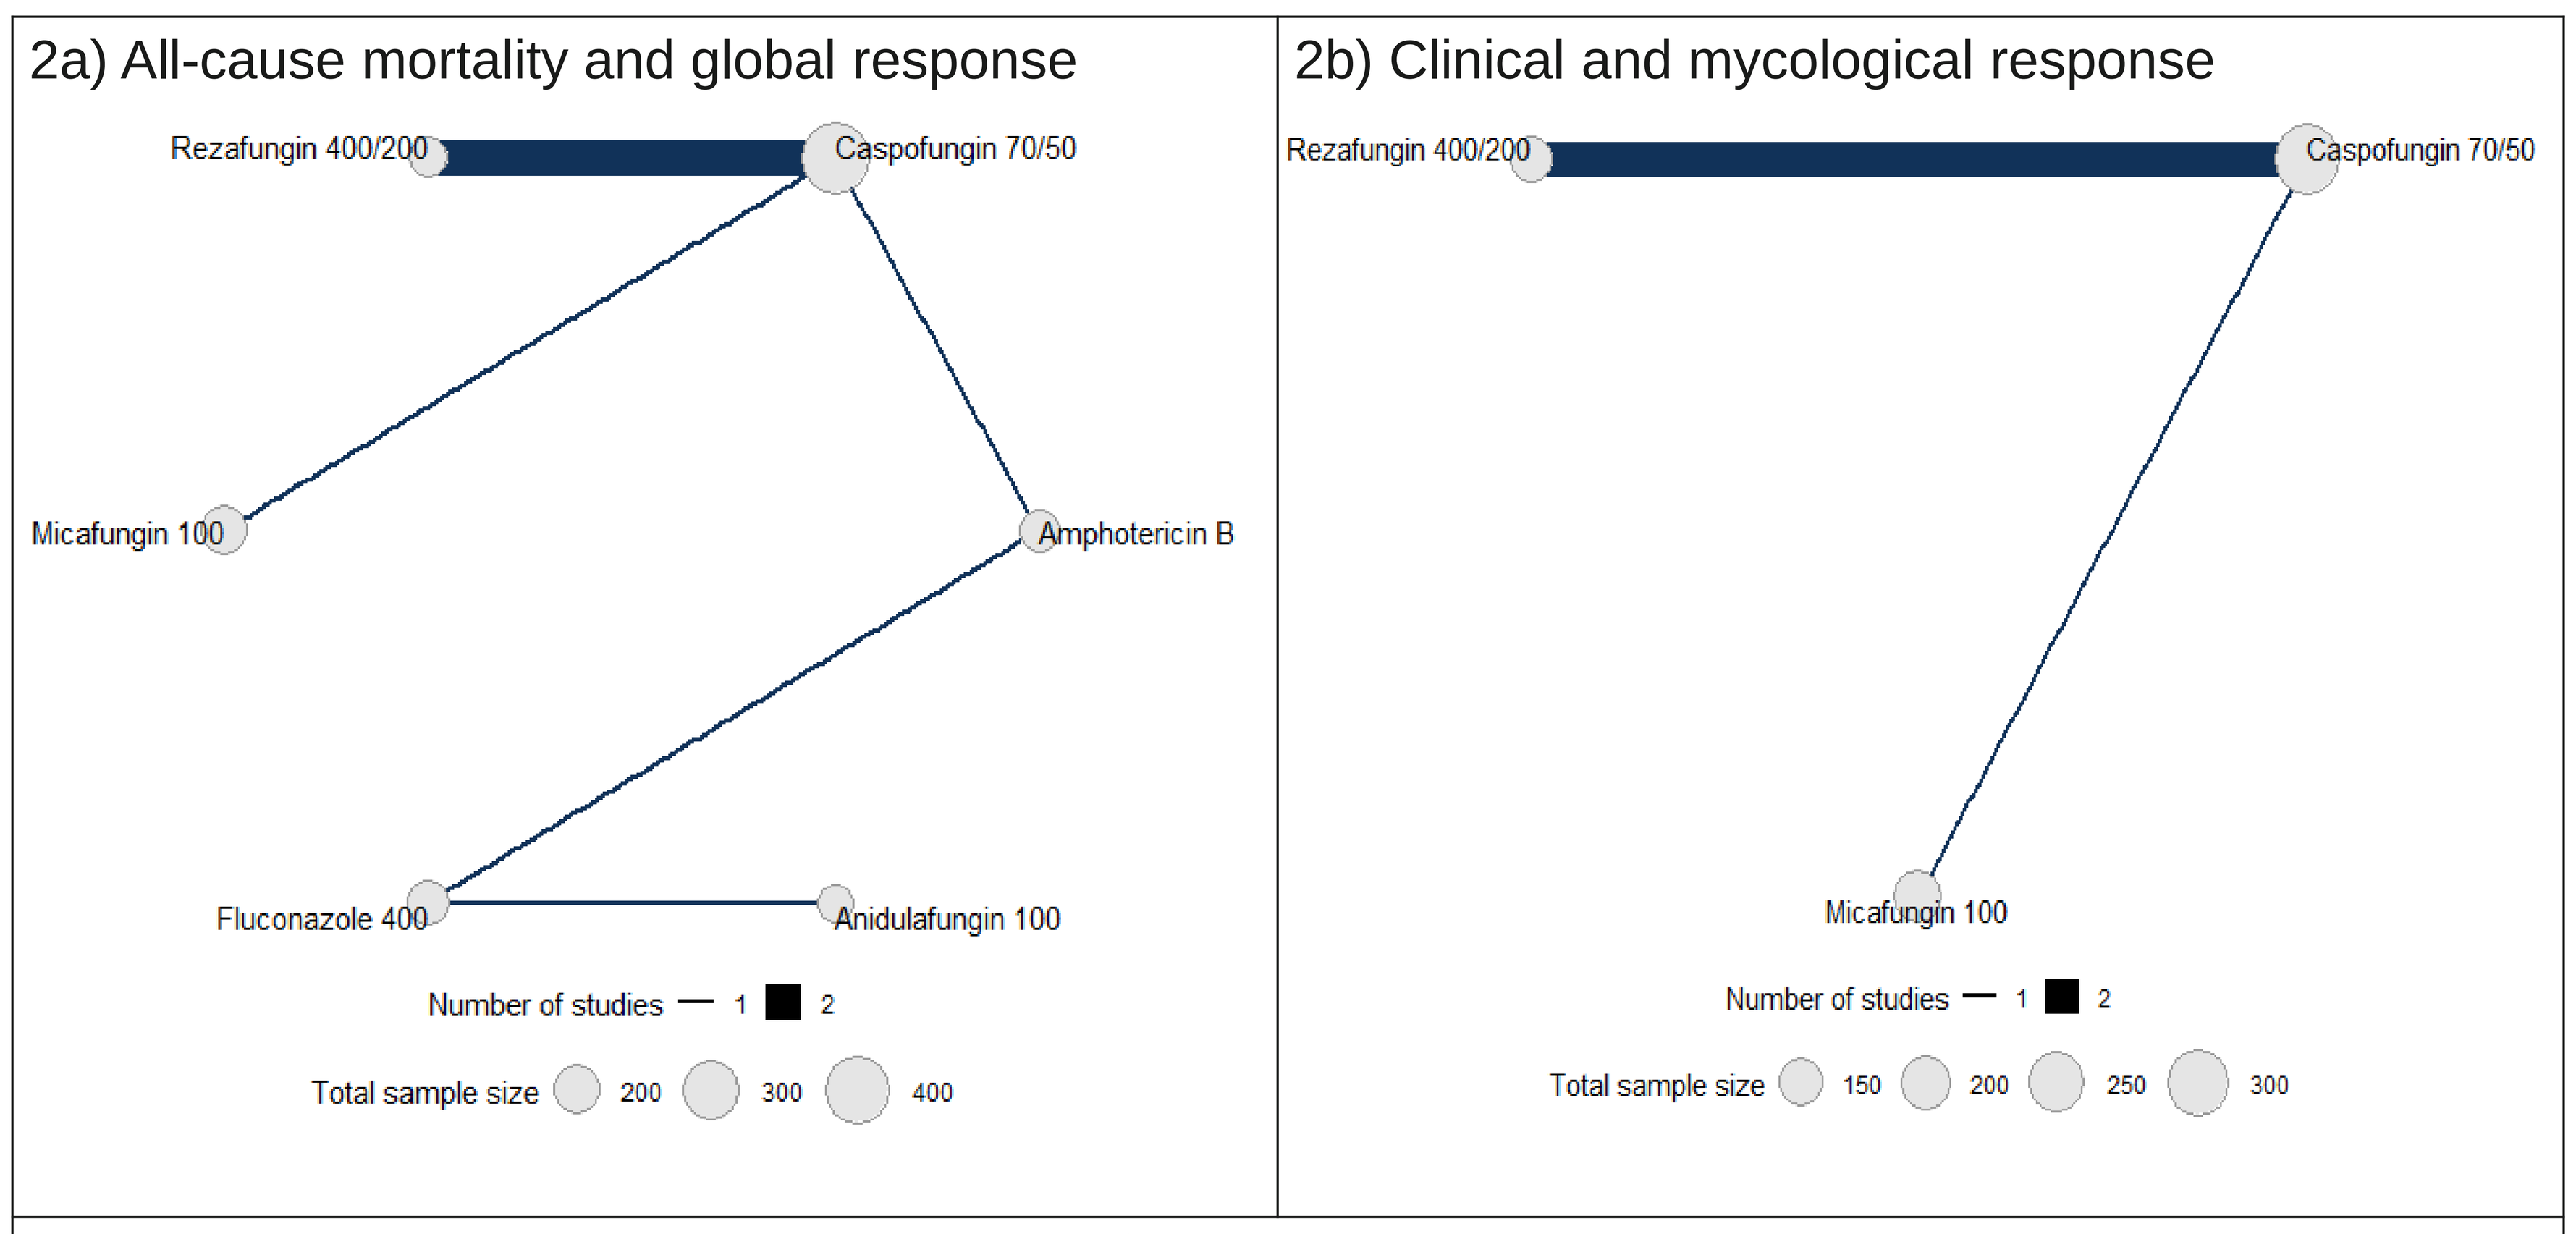
2a) All-cause mortality and global response
Rezafungin 400/200
Caspofungin 70/50
Micafungin 100
Amphotericin B
Fluconazole 400
Anidulafungin 100
Number of studies 1 2
Total sample size 200 300 400
2b) Clinical and mycological response
Rezafungin 400/200
Caspofungin 70/50
Micafungin 100
Number of studies 1 2
Total sample size 150 200 250 300

Each circle represents a treatment; connecting lines indicate pairs of treatments that have been directly compared in studies. The line thickness is proportional to the number of studies making that comparison. Circle diameters are proportional to the number of patients randomized to that treatment.

Figure 3. NMA forest plots of treatment effects

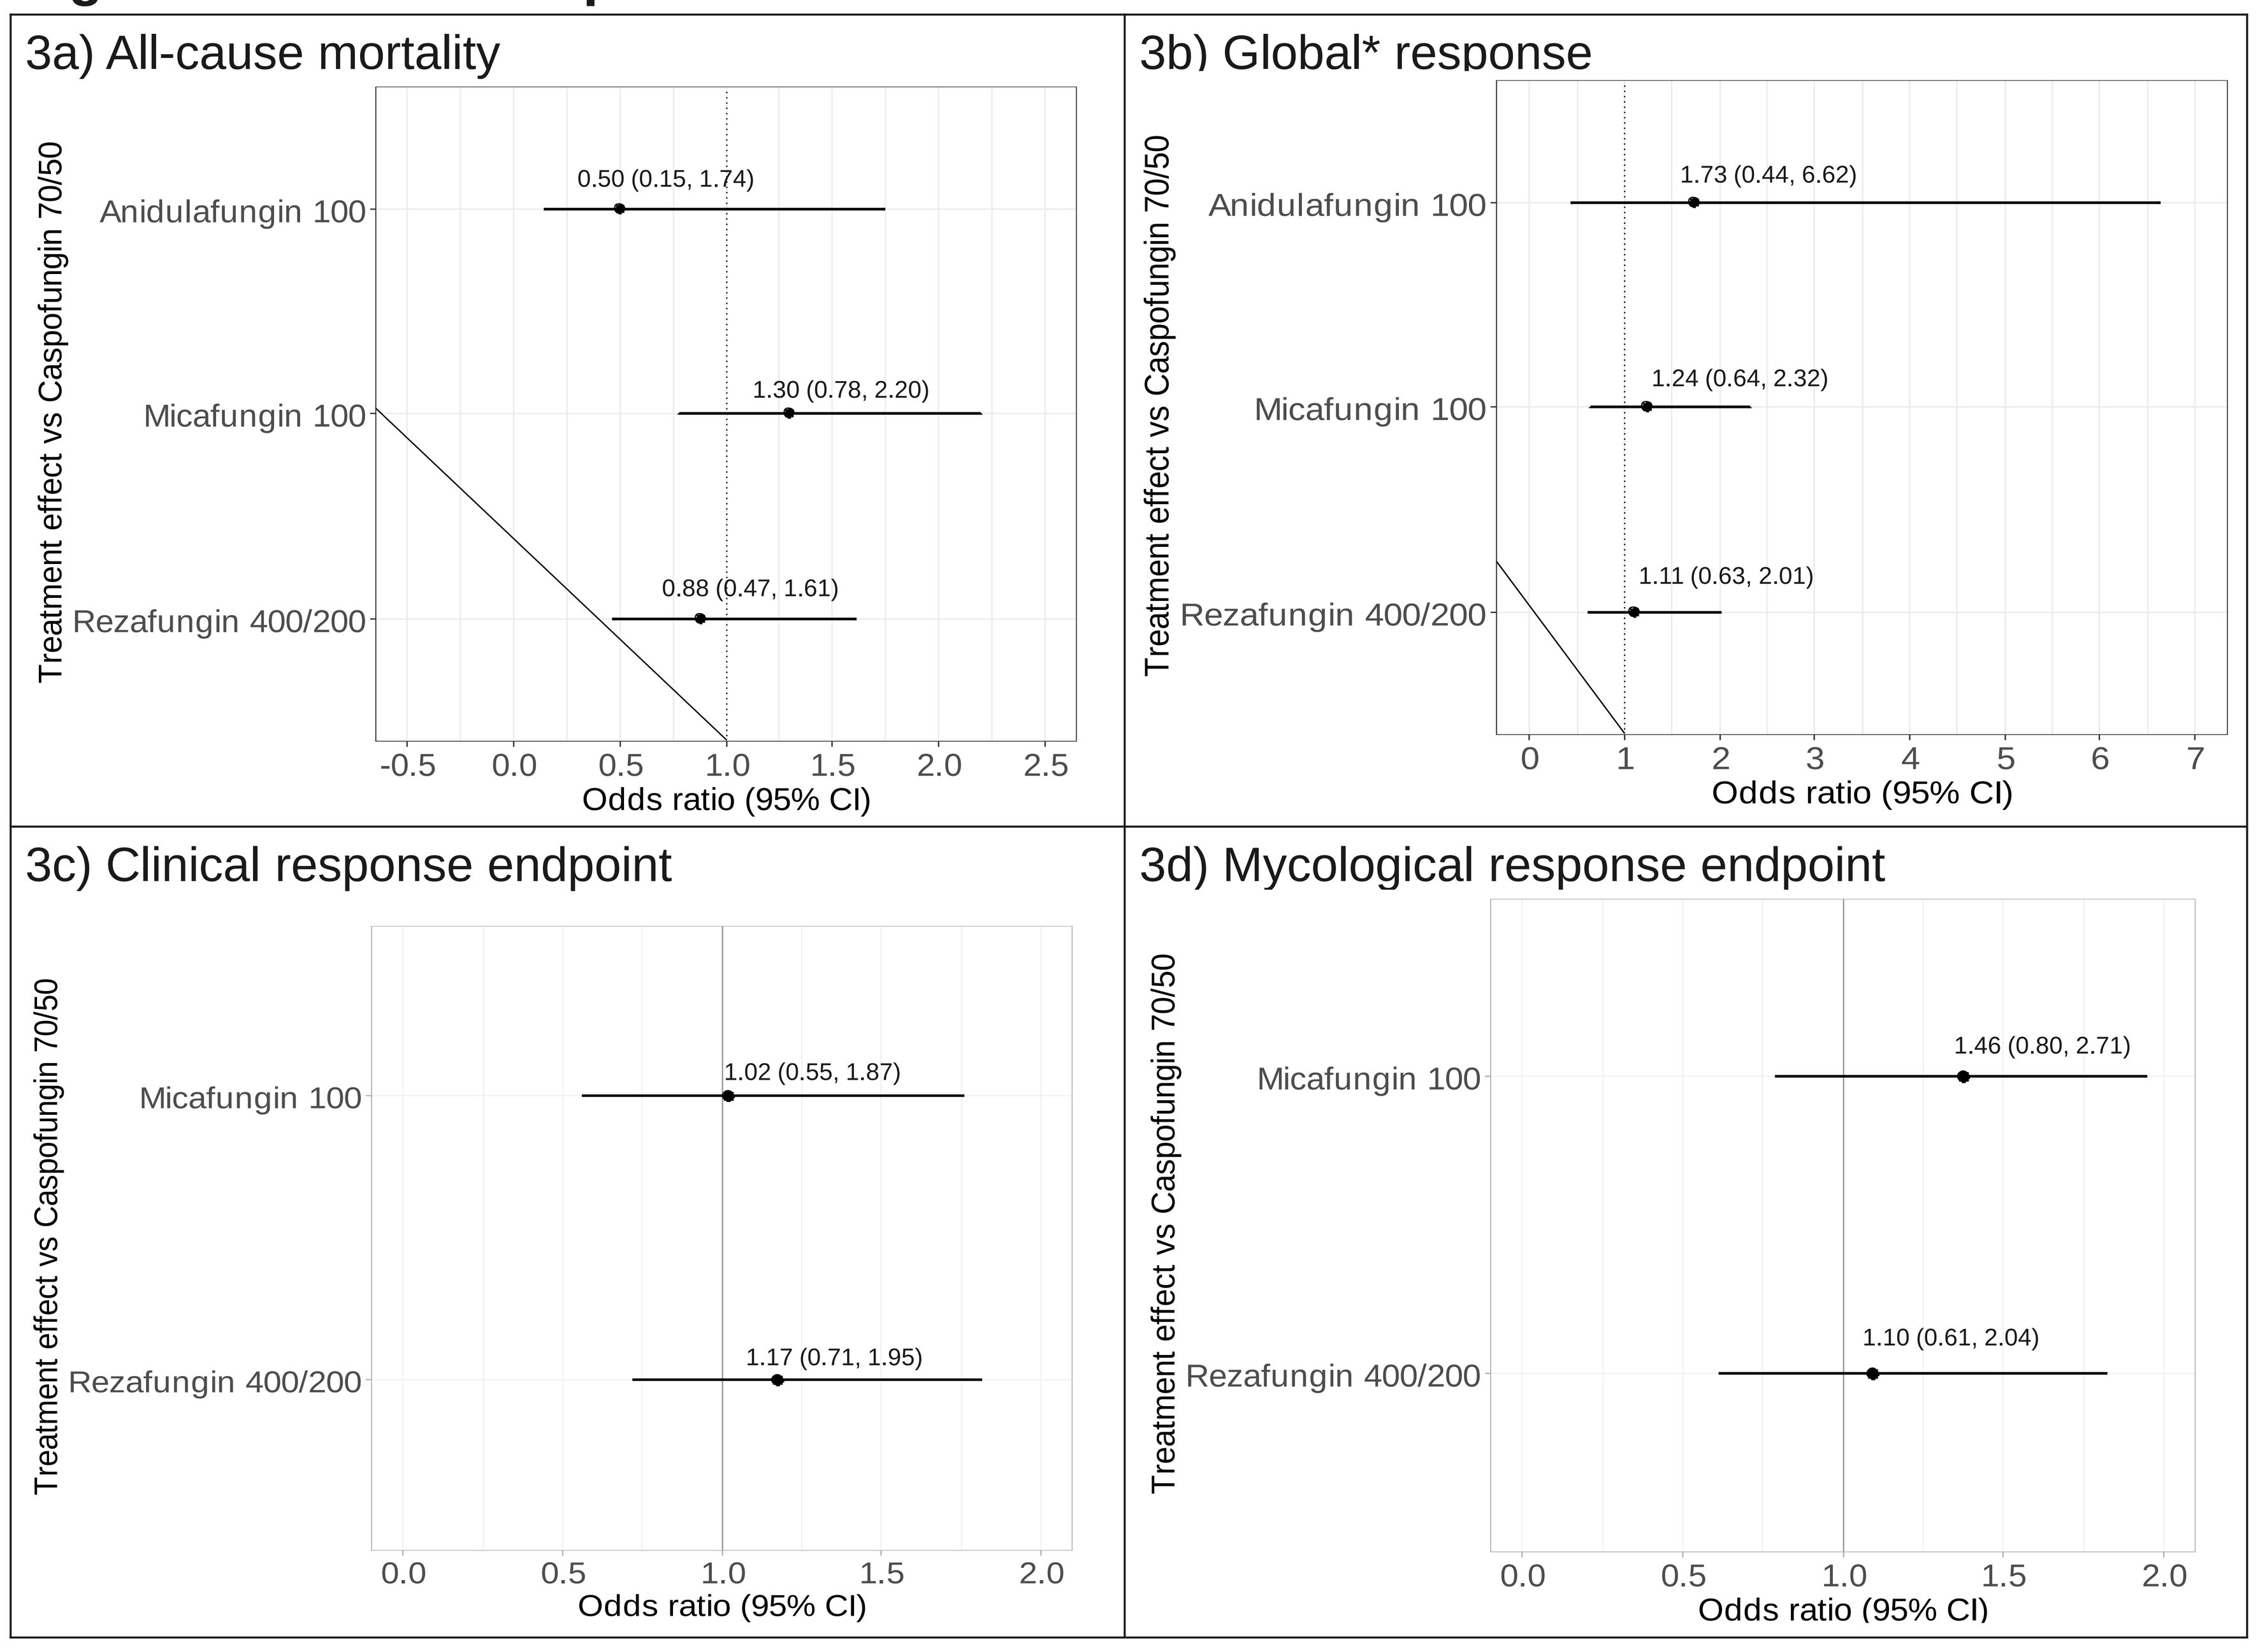
3a) All-cause mortality
Treatment effect vs Caspofungin 70/50
Anidulafungin 100
0.50 (0.15, 1.74)
Micafungin 100
1.30 (0.78, 2.20)
Rezafungin 400/200
0.88 (0.47, 1.61)
-0.5 0.0 0.5 1.0 1.5 2.0 2.5
Odds ratio (95% CI)
3b) Global* response
Treatment effect vs Caspofungin 70/50
Anidulafungin 100
1.73 (0.44, 6.62)
Micafungin 100
1.24 (0.64, 2.32)
Rezafungin 400/200
1.11 (0.63, 2.01)
0 1 2 3 4 5 6 7
Odds ratio (95% CI)
3c) Clinical response endpoint
Treatment effect vs Caspofungin 70/50
Micafungin 100
1.02 (0.55, 1.87)
Rezafungin 400/200
1.17 (0.71, 1.95)
0.0 0.5 1.0 1.5 2.0
Odds ratio (95% CI)
3d) Mycological response endpoint
Treatment effect vs Caspofungin 70/50
Micafungin 100
1.46 (0.80, 2.71)
Rezafungin 400/200
1.10 (0.61, 2.04)
0.0 0.5 1.0 1.5 2.0
Odds ratio (95% CI)

*Most studies defined global response based on mycological and clinical response criteria, however, ReSTORE had a slightly more stringent definition incorporating radiological cure.

LIMITATIONS

- There was heterogeneity among studies with respect to the definitions used for the response endpoints, the timing of assessments and patient populations.
- Between-study heterogeneity was incorporated in the random-effects models for the two larger networks (mortality and global response), but credible intervals were very wide and highly sensitive to prior information.

CONCLUSIONS

- There was no evidence that any treatment included in the NMAs was more effective than the reference treatment caspofungin 70mg once/50mg QD at reducing all-cause mortality or improving global, mycological or clinical response.
- However, rezafungin is a differentiated echinocandin with a low clearance and long half-life, enabling a once weekly front-loaded dosing regimen which may confer additional benefits not recognized herein.

References

1. Turner RM et al. Predictive distributions for between-study heterogeneity and simple methods for their application in Bayesian meta-analysis. *Statistics in Medicine*. 34, 984–998 (2015).
2. Thompson GR et al. Rezafungin versus caspofungin for treatment of candidaemia and invasive candidiasis (ReSTORE): a multicentre, double-blind, double-dummy, randomised phase 3 trial.*The Lancet.* 401(10370), 49-59 (2023).
3. Thompson GR et al. Rezafungin versus caspofungin in a phase 2, randomized, double-blind study for the treatment of candidemia and invasive candidiasis: The STRIVE Trial. *Clin. Infect. Dis. Off. Publ. Infect. Dis. Soc. Am.* 73, e3647–e3655 (2021).
4. Pappas PG et al. Micafungin versus caspofungin for treatment of candidemia and other forms of invasive candidiasis. *Clin. Infect. Dis.* 45, 883–893 (2007).
5. Mora-Duarte J.et al. Comparison of caspofungin and amphotericin B for invasive candidiasis. *N. Engl. J. Med.* 347, 2020–2029 (2002).
6. Reboli AC et al. Anidulafungin versus fluconazole for invasive candidiasis. *N. Engl. J. Med.* 356, 2472–2482 (2007).
7. Phillips P.et al. Multicenter randomized trial of fluconazole versus amphotericin B for treatment of candidemia in non-neutropenic patients. *Eur. J. Clin. Microbiol. Infect. Dis.* 16, 337–345 (1997).

Visible Analytics from data to decisions™

mundipharma | market access

ISPOR Europe 2024 | Barcelona, Spain | 17 – 20 November