Comparative effectiveness of mRNA-1273 vs BNT162b2 in Canadians: a systematic literature review and GRADE meta-analysis

Xuan Wang, MD, MSc[1]; Ankit Pahwa, MS[2]; Pawana Sharma, MSc[3]; Anushri Chitkara, MPH[2]; Nitendra Kumar Mishra, MS[2]; Mia Malmenäs[1]; Sonam Vats, MPharma[2]; Paridhi Jain, MSc[2]; Ritu Gupta, MPH, BDS[2]; Ekkehard Beck, PhD[4]; Kavisha Jayasundara PhD[5]; Michelle Blake PhD[5]

[1]ICON plc, Stockholm, Sweden; [2]ICON plc, Bengaluru, India; [3]ICON plc, London, UK; [4]Moderna, Inc., Cambridge, MA, USA; [5]Moderna Biopharma Canada, Toronto, Canada

BACKGROUND

- As Canada's public healthcare payers transition to planning COVID-19 vaccine programs in an endemic context, the question of the preferred **mRNA vaccine** between the **Moderna** and **BioNTech/Pfizer** products persists.[1]
- To address this gap, this **systematic literature review (SLR) and GRADE meta-analysis** (INPLASY registration: INPLASY202460071) aims to summarize published studies on the **comparative effectiveness** of mRNA COVID-19 vaccines among **Canadians**.

METHODS

- The main search for studies was conducted in Embase, Medline and MEDLINE In-Process, e-pubs ahead of print, the Cochrane Database of Systematic Reviews (CDSR) and Cochrane Central Register of Controlled Trial (CCRT) on April 4, 2024. The search considered English studies only based on criteria in Table 1.
- Outcomes of interest were SARS-CoV-2 infection (symptomatic or asymptomatic with positive test of COVID-19 diagnosis), laboratory-confirmed symptomatic SARS-CoV-2 infection, severe SARS-CoV-2 infection (as defined by the study), COVID-19–related hospitalization, and COVID-19–related death.

Table 1. Study Eligibility Criteria

Inclusion Criteria	
Population	Canadians aged ≥ 16 years
Intervention	mRNA-1273 (Moderna mRNA COVID-19 vaccine)
Comparator	BNT162b2 (Pfizer/BioNTech mRNA COVID-19 vaccine)
Outcomes	SARS-CoV-2 infection, symptomatic SARS-CoV-2 infection, severe SARS-CoV-2 infection (only included studies with defined severe infection), hospitalization, and death associated with SARS-CoV-2 infection
Study type	Clinical trials and real-world studies
Geography	Canada only (studies conducted in Canada + studies with Canadian-specific data)

- A pairwise meta-analysis was conducted using risk ratio (RR) as the data input, derived either from number of events and sample size per arm or, for cases in which these were not reported, from vaccine effectiveness data.
- Random-effects meta-analysis models were used to pool RRs across studies. Heterogeneity was evaluated using the I^2 measure with corresponding chi-square statistics. Certainty of evidence and importance of outcomes was assessed per the GRADE framework.

RESULTS

Figure 1. PRISMA flow diagram

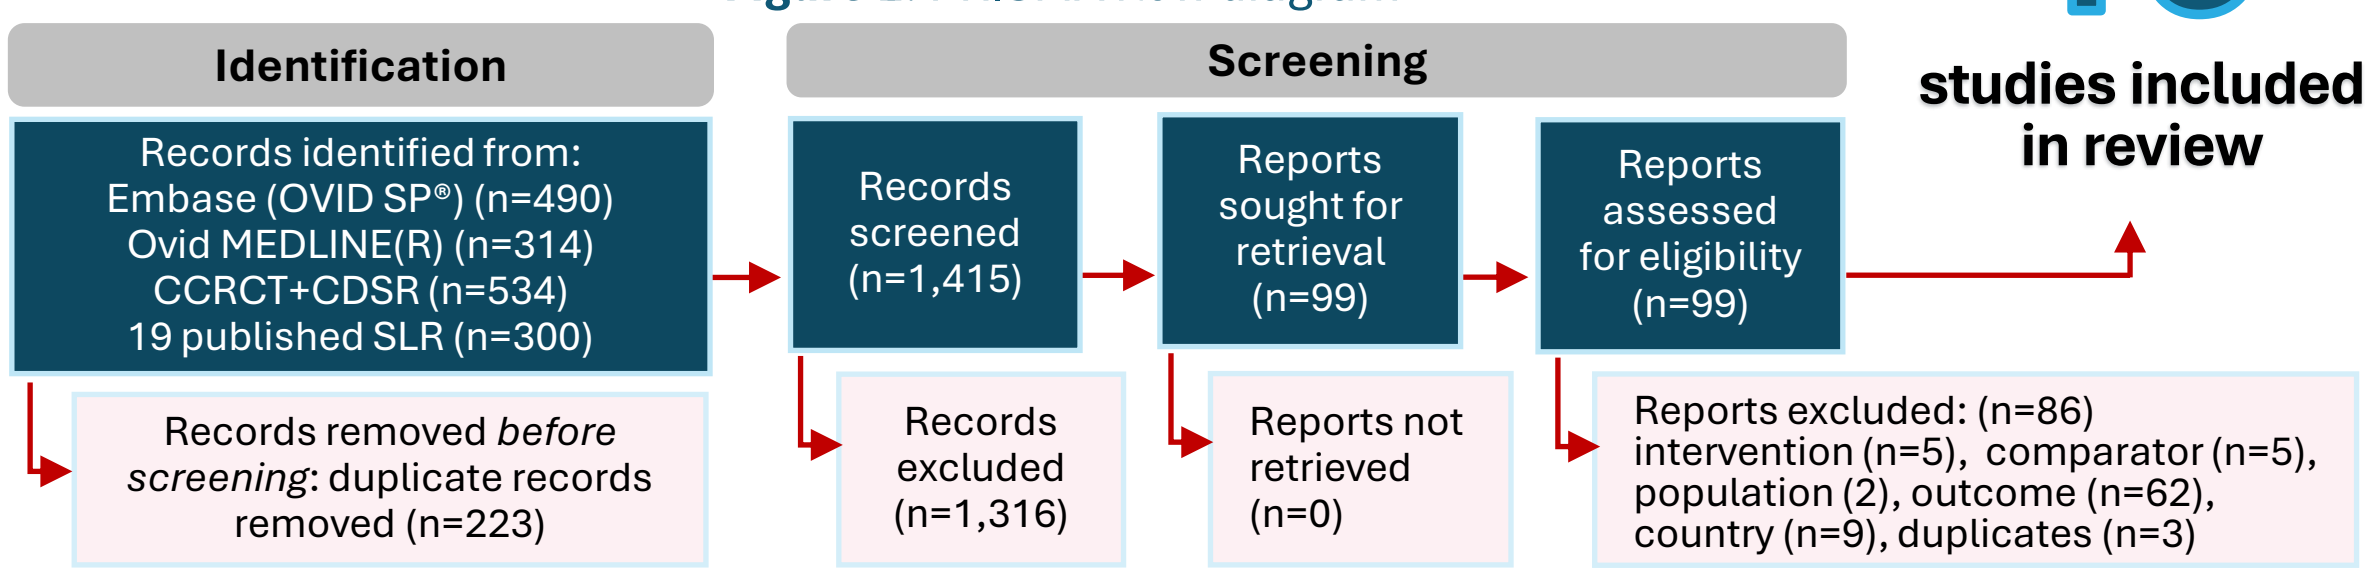
Identification
Screening
Records identified from:
Embase (OVID SP®) (n=490)
Ovid MEDLINE(R) (n=314)
CCRCT+CDSR (n=534)
19 published SLR (n=300)
Records removed *before screening*: duplicate records removed (n=223)
Records screened (n=1,415)
Records excluded (n=1,316)
Reports sought for retrieval (n=99)
Reports not retrieved (n=0)
Reports assessed for eligibility (n=99)
Reports excluded: (n=86)
intervention (n=5), comparator (n=5), population (2), outcome (n=62), country (n=9), duplicates (n=3)

13
studies included in review

Figure 2. Populations of included studies
(CKD, chronic kidney disease; LTC, long-term care)

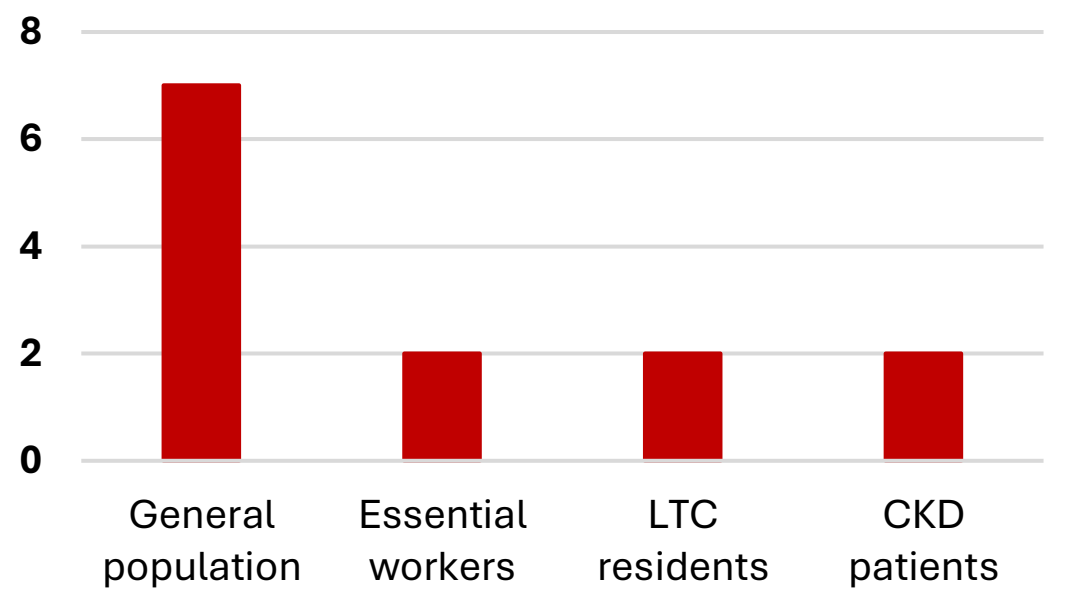
8
6
4
2
0
General population
Essential workers
LTC residents
CKD patients

Overall population

- Overall, the studies included 552,619 Canadians (aged ≥16 years) vaccinated with mRNA-1273 and 1,778,942 vaccinated with BNT162b2.
- In the overall population, mRNA-1273 was associated with significantly lower risk of SARS-CoV-2 infection (any infection, laboratory-confirmed symptomatic infection) and a lower risk of severe infection (Figure 3).
- There was limited data for COVID-19-related hospitalization and no data for related death to draw any conclusion on these outcomes for the general population in Canada.

Figure 3. Summary of meta-analysis results on clinical effectiveness outcomes of mRNA-1273 vs. BNT162b2 in the overall population of Canadian adults

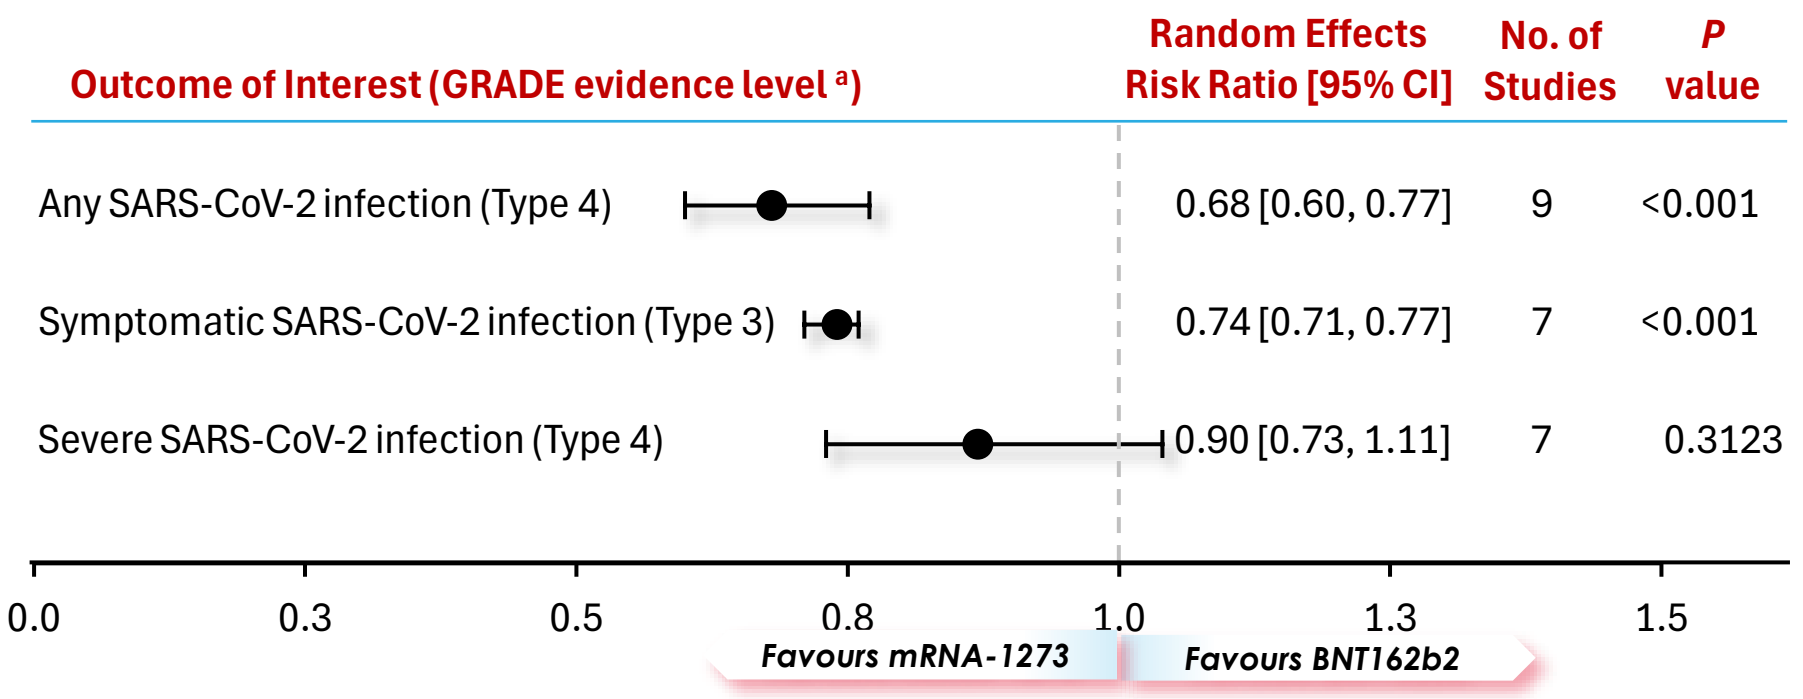
Outcome of Interest (GRADE evidence level a)
Random Effects Risk Ratio [95% CI]
No. of Studies
P value
Any SARS-CoV-2 infection (Type 4)
0.68 [0.60, 0.77]
9
<0.001
Symptomatic SARS-CoV-2 infection (Type 3)
0.74 [0.71, 0.77]
7
<0.001
Severe SARS-CoV-2 infection (Type 4)
0.90 [0.73, 1.11]
7
0.3123
0.0
0.3
0.5
0.8
1.0
1.3
1.5
Favours mRNA-1273
Favours BNT162b2

[a] Certainty type 3 = Type 3= Observational studies, or RCTs with notable limitations; type 4= Clinical experience and observations, observational studies with important limitations, or RCTs with several major limitations

Subgroup analysis

Figure 4. High-risk groups of interest for whom data from the included studies were and were not available to conduct a meta-analysis (risk ratio [95% confidence interval]). Where available, the RRs of any SARS-CoV-2 infection associated with mRNA-1273 vs. COMIRNATY are reported.

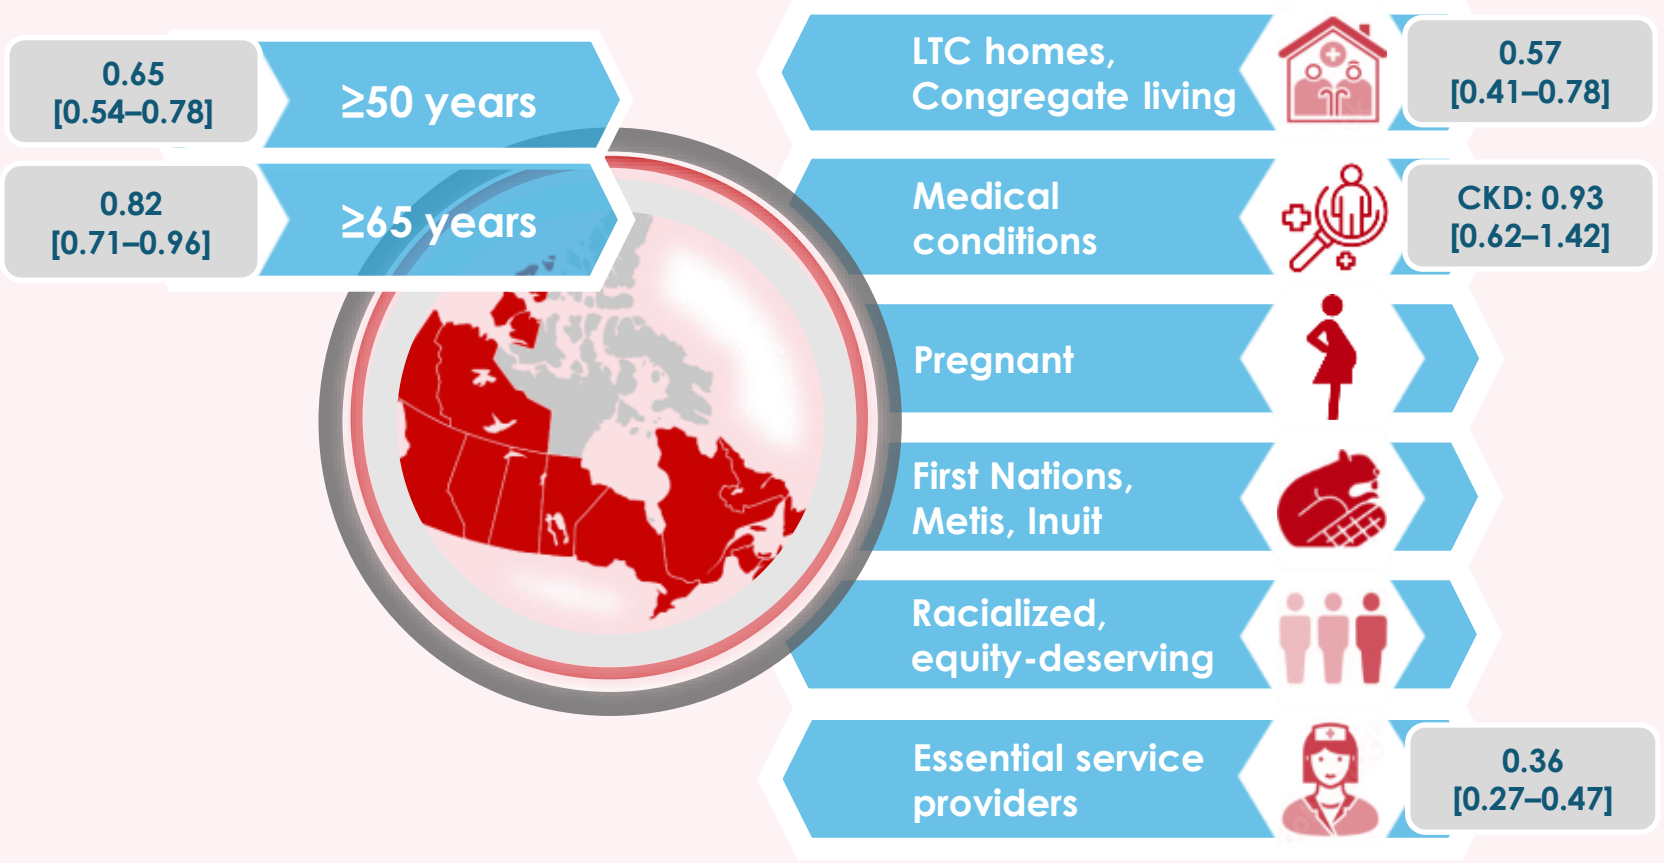
0.65 [0.54–0.78]
≥50 years
0.82 [0.71–0.96]
≥65 years
LTC homes, Congregate living
0.57 [0.41–0.78]
Medical conditions
CKD: 0.93 [0.62–1.42]
Pregnant
First Nations, Metis, Inuit
Racialized, equity-deserving
Essential service providers
0.36 [0.27–0.47]

- Compared with BNT162b2, mRNA-1273 was associated with a statistically significantly reduced risk for any COVID-19 infection in groups at high risk for COVID-19 severe disease based on age and living in LTC/congregate settings (Figure 4).

LIMITATIONS & CONCLUSION

- mRNA-1273 was associated with a significantly lower risk of SARS-CoV-2 infection and symptomatic SARS-CoV-2 infection, as well as a lower risk of severe SARS-CoV-2 infection, compared with BNT162b2. The results were robust across subgroups of individuals aged ≥50 years, ≥65 years, essential community services, and LTC residents.
- The lower risk of severe SARS-CoV-2 infection associated with mRNA-1273 was not statistically significant, and likely related to limitation of small number of included studies and high variability in the outcome, given severe infection is a rare event. The association, however, is directionally consistent with larger SLRs & GRADE meta-analyses of real-world studies that have investigated the comparative effectiveness of mRNA-1273's vs. BNT162b2 against COVID-19 outcomes amongst populations across a range of geographies.[1-3]
- Key limitations included low certainty of evidence for the outcomes, which were based on a mixed dosing (primary vs. booster) and strain formulations that may not capture recent updates of COVID-19 vaccines; inadequate data for hospitalizations or death due to COVID-19 to draw any conclusion on these outcomes for general population

References

1. Kavikondala et al. Infect Dis Ther. 2024 Apr;13(4):779-811. doi: 10.1007/s40121-024-00936-z. Epub 2024 Mar 18.
2. Wang et al. Front Immunol. 2023 Sep 12;14:1204831. doi: 10.3389/fimmu.2023.1204831
3. Wang et al. https://doi.org/10.1101/2024.09.13.24313632

Full list of articles included in analysis available upon request

Additional information

Please scan the QR code for a PDF copy of the poster. Copies of the poster obtained through this QR code are for personal use only and may not be reproduced without written permission of the authors. For additional information, please contact Michelle Blake (michelle.blake@modernatx.com)

Disclosures

X Wang, A Pahwa, P Sharma, A Chitkara, N Mishra, M Malmenas, S Vats, P Jain, and R Gupta are employees of ICON plc, a clinical research organization paid by Moderna, Inc., to conduct the study.

E Beck, K Jayasundra, M Blake are employed by Moderna, Inc., and hold stock/stock options in the company.

Presented at the International Professional Society for Health Economics and Outcomes Research (ISPOR) Annual Congress
November 18-20, 2024; Barcelona, Spain