Survival Benefit of Oral Systemic Monotherapy Treatment in Heavily Pre-treated Metastatic Colorectal Cancer: A Meta-analysis

CO203

Victoria Paly, MHS,[1] Chiara Cremolini, MD, PhD,[2] Elena Elez, MD, PhD,[3] Sarah Ronnebaum, PhD,[4] Morodoluwa Akin-Fajiye, PhD,[5] Paola Stefan, PharmD, MPH,[1] Jennifer Elliott, PhD,[1] Luis Hernandez, PhD, MPH, MSc[1]

[1]Takeda Pharmaceuticals America, Inc., Lexington, MA, USA; [2]University of Pisa, Pisa, Italy; [3]Vall d'Hebron Institute of Oncology, Barcelona, Spain; [4]Evidera Inc., Bethesda, MD, USA; [5]Evidera Inc., Montreal, Canada

Question

Do oral systemic therapies provide a survival benefit, relative to placebo and/or BSC, for patients with heavily pre-treated mCRC?

Methods

Systematic literature review

Databases (searched Oct 4, 2023):

- **MEDLINE**
- **MEDLINE In-Process**
- **Embase**
- **Cochrane Database of Systematic Reviews**
- **Cochrane Central Register of Controlled Trials**

PICOS criteria:

- **Patients:** Heavily pre-treated mCRC
- **Interventions:** Fruquintinib, regorafenib, TAS-102 ± bevacizumab, and chemotherapy/targeted therapy re-challenge
- **Comparators:** Placebo and/or BSC
- **Outcomes:** Median OS and median PFS
- **Study design:** Phase III RCTs

Meta-analyses

- **Median OS** and **median PFS** were meta-analyzed across studies comparing **oral systemic therapies with placebo and/or BSC**
- **Meta-analyses** were conducted using **random effects** and **fixed effects** models

Key take aways

Oral systemic therapies provide significant survival benefits for patients with heavily pre-treated mCRC

Figure 1: Meta-analysis of difference in median OS

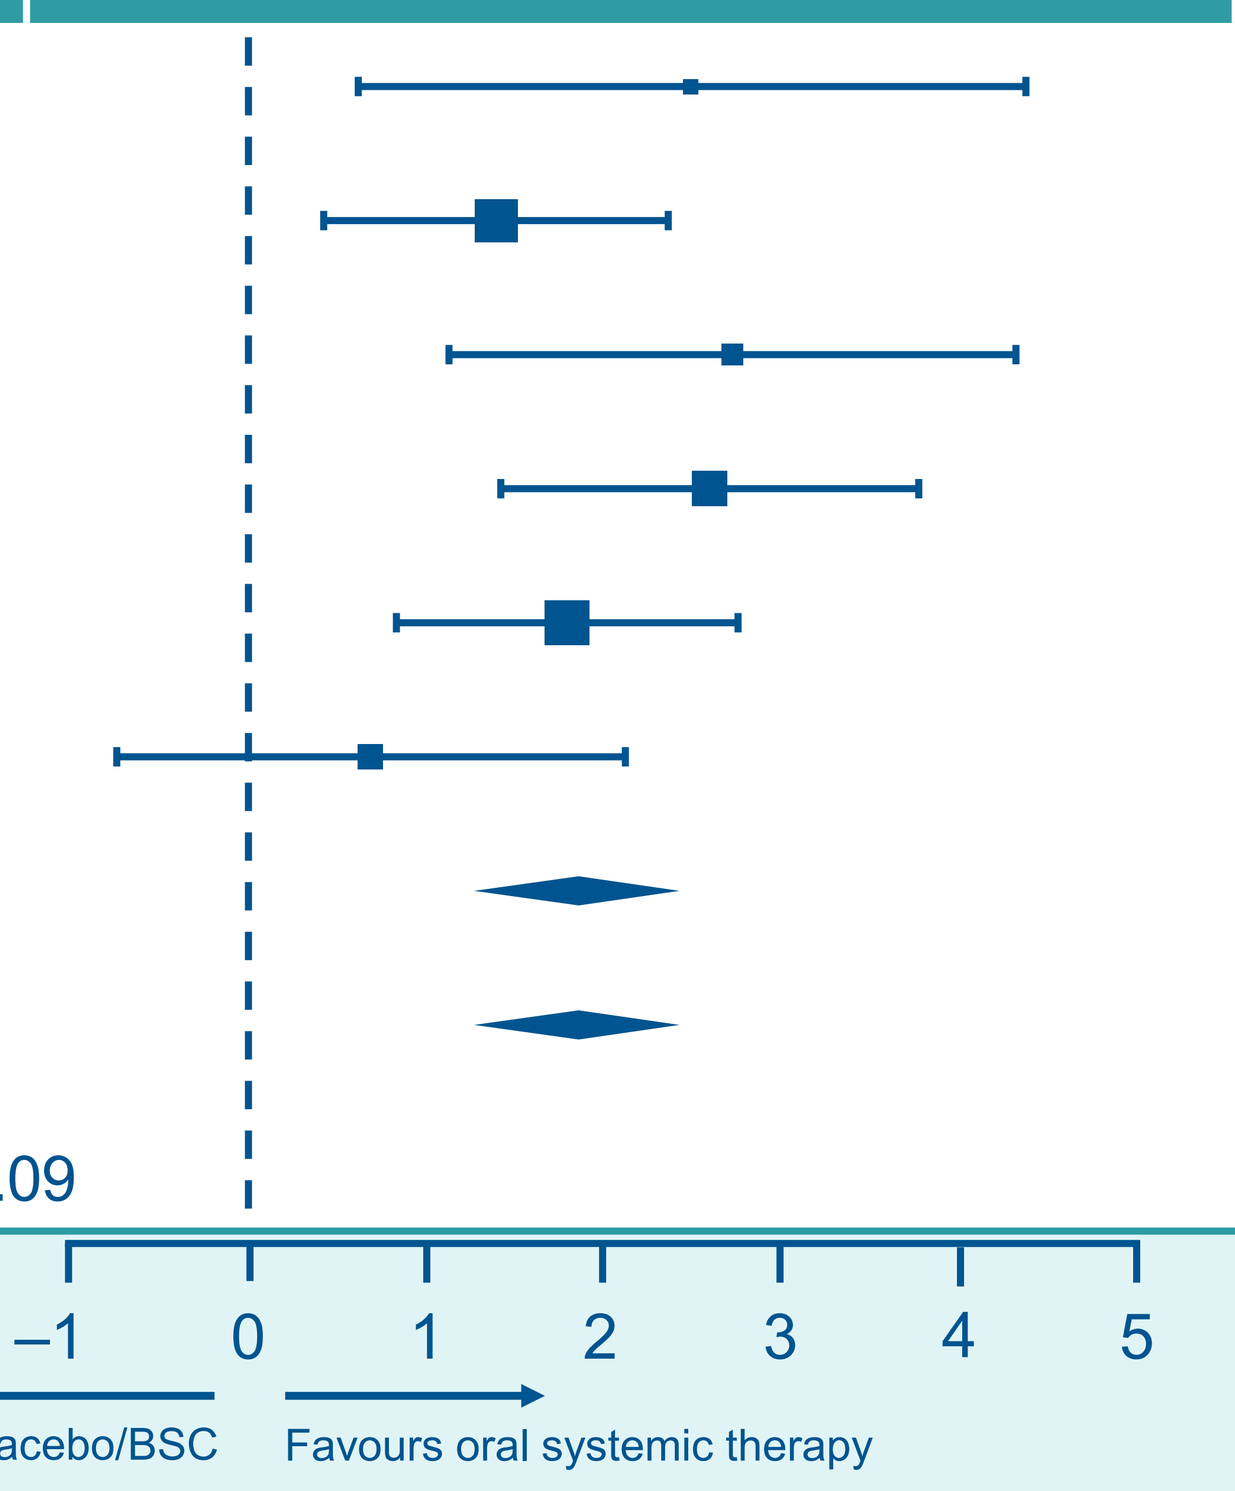
Mean difference in median OS, months
–1
0
1
2
3
4
5
Favours placebo/BSC
Favours oral systemic therapy

Study	MD (95% CI)
CONCUR*	2.50 (0.62, 4.38)
CORRECT	1.40 (0.43, 2.37)
FRESCO*	2.73 (1.14, 4.32)
FRESCO-2	2.60 (1.43, 3.77)
RECOURSE	1.80 (0.84, 2.76)
TERRA*	0.70 (–0.73, 2.13)
RE Model	1.86 (1.30, 2.42)
FE Model	1.84 (1.35, 2.34)

Heterogeneity: I^2=18.63%, τ^2=0.09

*Trial was conducted only in Asia.

Background & Objective

- Colorectal cancer (CRC) often presents at an advanced stage. Approximately 23% of patients having developed metastatic disease by the time of diagnosis,[1] while up to 50% of patients with localized CRC at diagnosis eventually develop metastases[2]
- The prognosis of patients with metastatic colorectal cancer (mCRC) is poor and worsens as patients receive multiple lines of therapy
 - Based on randomized controlled trials (RCTs), the expected median overall survival (OS) of patients treated with ≥2 prior lines of therapy receiving best supportive care (BSC) is 4.8−7.1 months[3–8]
- Based on the European Society for Medical Oncology (ESMO) Metastatic Colorectal Cancer Living Guideline for third- and further-line treatment, fruquintinib, regorafenib, or trifluridine/tipiracil (T/T) ± bevacizumab are recommended for patients who had previously received chemotherapy, anti-vascular endothelial growth factor (VEGF) therapy, and/or anti-epidermal growth factor receptor (EGFR) therapy if *RAS* wild type[9]
- The objective of this study was to characterize the survival benefit associated with oral systemic therapy relative to no active therapy (placebo and/or BSC) for patients with heavily pre-treated mCRC

Methods

- A systematic literature review (SLR) was conducted to summarize the available evidence concerning treatment efficacy in adult patients with mCRC who have been previously treated with fluoropyrimidine-, oxaliplatin-, and irinotecan-based chemotherapy, anti-VEGF therapy, and, if *RAS* wild type, an anti-EGFR therapy
- The SLR was conducted using OvidSP to identify relevant peer-reviewed studies in the following electronic databases: MEDLINE, MEDLINE In-Process, Embase, Cochrane Database of Systematic Reviews, and Cochrane Central Register of Controlled Trials (searches conducted October 4, 2023). The literature search was limited to citations published in the English language, and conference abstracts were limited to those published in or after 2020
- The inclusion criteria used to identify relevant studies are shown in **Table 1**
- Median OS and median progression free survival (PFS) were meta-analyzed across the included studies that compared oral systemic therapy to placebo and/or BSC
- Fixed effects (FE) and random effects (RE) frequentist meta-analyses were conducted for both outcomes
 - Each analysis estimates the mean effect and its standard error and 95% confidence interval (CI), and each RE analysis also estimates the value of τ, the proportion of observed variation caused by RE variance (the I^2 measure), and tests the hypothesis τ=0 using a χ^2 test of Q
 - Statistical heterogeneity was investigated by assessing the test of homogeneity and consideration of the size of I^2
- All meta-analyses were conducted using a restricted maximum-likelihood approach, using the *metafor* package (version 4.4)[10] for the R software environment (version 4.4.1)

Table 1: PICOS criteria for the SLR

Criteria	Inclusion criteria
Population	Patients with mCRC (received ≥2 prior lines of systemic chemotherapies) who have been previously treated with or are not considered candidates for available therapies, including fluoropyrimidine-, oxaliplatin-, and irinotecan-based chemotherapy, an anti-VEGF therapy, and, if *RAS* wild type, an anti-EGFR therapy
Interventions	• Fruquintinib • Regorafenib • T/T ± bevacizumab* • Rechallenge using treatments including but not limited to panitumumab, cetuximab, FOLFOX, FOLFIRI, CAPOX, bevacizumab, or other chemotherapy/targeted therapy treatments
Comparators	• Placebo • BSC
Outcomes	• Median OS • Median PFS
Study Design	• Phase III RCTs

PICOS, population, intervention, comparator, outcome, and study design.
*Trials investigating T/T + bevacizumab were not included in the final analysis.

Results

Systematic Literature Review

- The electronic database search identified 4,165 publications. After deduplication (1,143 records removed), the titles and abstracts of the remaining 3,022 records were screened. Of these, the full texts of 516 publications were reviewed and six phase 3 RCTs met all inclusion criteria (**Table 2**)
 - CONCUR[5] and CORRECT:[6] regorafenib + BSC versus placebo + BSC
 - FRESCO[3] and FRESCO-2:[4] fruquintinib + BSC versus placebo + BSC
 - RECOURSE[7] and TERRA:[8] T/T + BSC versus placebo + BSC
- All included studies required patients to have failed on ≥2 lines of standard chemotherapies (fluoropyrimidine-, oxaliplatin-, and irinotecan-based chemotherapy) for advanced disease, or to have received all current locally approved standard therapies. Patients may have also received anti-VEGF therapy, and, if *RAS* wild type, an anti-EGFR therapy
 - The FRESCO-2 trial was the only study to also require that patients had previously received regorafenib and/or T/T[4]
- The identified RCTs included populations with 39%−61% of patients who had progressed after ≥4 lines of chemotherapy (**Table 2**)
- Of note, the T/T + bevacizumab combination was only assessed in the SUNLIGHT trial with T/T monotherapy as the comparator, and thus could not be included in the meta-analysis of systemic treatment versus placebo and/or BSC.[11] Additionally, the SUNLIGHT trial had a less heavily pre-treated patient population than the other included studies (97% of participants had received ≤2 lines of prior treatment)[11]

Table 2: Summary of included study characteristics

Trial	Region	Intervention and comparator*	n	Median age, years (IQR)	Male, %	ECOG PS 0 / 1, %	Number of metastatic sites, %	Prior lines of treatment, %
CONCUR[5]	Asia	Regorafenib	136	58 (50–66)	63	26 / 74	1: 21 ≥2: 79	1–2: 35‡ 3: 24‡ ≥4: 38‡
		Placebo	68	56 (49–62)	49	22 / 78	1: 22 ≥2: 78	1–2: 35‡ 3: 25‡ ≥4: 40‡
CORRECT[6]	Global	Regorafenib	505	61 (54–67)	62	52 / 48	NR	1–2: 27‡ 3: 25‡ ≥4: 49‡
		Placebo	255	61 (54–68)	60	57 / 43	NR	1–2: 25‡ 3: 28‡ ≥4: 47‡
FRESCO[3]	Asia	Fruquintinib	278	55 (23–75)†	57	28 / 72	1: 5 ≥2: 95	≤3: 79‡ >3: 21‡
		Placebo	138	57 (24–74)†	70	27 / 73	1: 3 ≥2: 97	≤3: 78‡ >3: 22‡
FRESCO-2[4,12]	Global	Fruquintinib	461	64 (56–70)	53	43 / 57	1: 13 ≥2: 87	≤3: 27‡ >3: 73‡
		Placebo	230	64 (56–69)	61	44 / 56	1: 18 ≥2: 82	≤3: 28‡ >3: 72‡
RECOURSE[7,13]	Global	T/T	534	63 (27–82)†	61	56 / 44	1–2: 61 ≥3: 39	2: 18 3: 22 ≥4: 60
		Placebo	266	63 (27–82)†	62	55 / 45	1–2: 58 ≥3: 42	2: 17 3: 20 ≥4: 63
TERRA[8]	Asia	T/T	271	58 (26–81)†	63	24 / 76	1–2: 61 ≥3: 39	2: 23 3: 27 ≥4: 50
		Placebo	135	56 (24–80)†	62	22 / 78	1–2: 61 ≥3: 39	2: 19 3: 27 ≥4: 55

*All treatment arms were given in combination with BSC. †IQR was not reported for median age; range is reported instead. ‡Prior lines of treatment in metastatic disease.
ECOG; Eastern Cooperative Oncology Group; IQR, interquartile range; NR, not reported.

Meta-Analyses

- At the trial level, mean differences in median OS for oral systemic therapy versus placebo + BSC ranged from 0.70 months (95% CI: −0.73, 2.13) for T/T in TERRA[8] to 2.73 months (95% CI: 1.14, 4.32) for fruquintinib in FRESCO.[3] Median OS benefit for regorafenib and T/T varied across their respective trials,[5–8] whereas the benefit seen for fruquintinib was consistent across trials (**Summary Panel, Figure 1**)[3,4]
- In the meta-analysis, the mean difference in median OS associated with oral systemic therapy versus placebo + BSC was 1.86 months (95% CI: 1.30, 2.42) using the RE model and 1.84 months (95% CI: 1.35, 2.34) using the FE model (**Summary Panel, Figure 1**)
 - A low degree of statistical heterogeneity (I^2=18.63%) was observed for the OS analysis
 - As a higher proportion of patients had received >3 lines of prior therapy in FRESCO-2 compared with the other trials included,[4] this was excluded in sensitivity analyses. When FRESCO-2 was excluded, the improvement in median OS across all the remaining studies was 1.68 months (95% CI: 1.13, 2.23) using the RE model and 1.68 months (95% CI: 1.13, 2.23) using the FE model
- At the trial level, mean differences in median PFS ranged from 0.20 months (95% CI: −0.25, 0.65) for T/T in TERRA[8] to 1.90 months (95% CI: 1.74, 2.06) for fruquintinib in FRESCO-2.[4] Median PFS benefit was consistent across trials for fruquintinib and T/T, whereas the benefit varied in trials investigating regorafenib[5–8] (**Figure 2**)
- In the meta-analysis, the mean difference in median PFS observed was 0.97 months (95% CI: 0.28, 1.66) using the RE model and 0.63 months (95% CI: 0.56, 0.70) using the FE model (**Figure 2**)
 - A high degree of statistical heterogeneity (I^2=98.63%) was observed for the PFS analysis
 - When FRESCO-2 was excluded, the mean difference in median PFS was 0.77 months (95% CI: 0.07, 1.46) using the RE model and 0.31 (95% CI: 0.23, 0.38) using the FE model

Figure 2: Meta-analysis of difference in median PFS

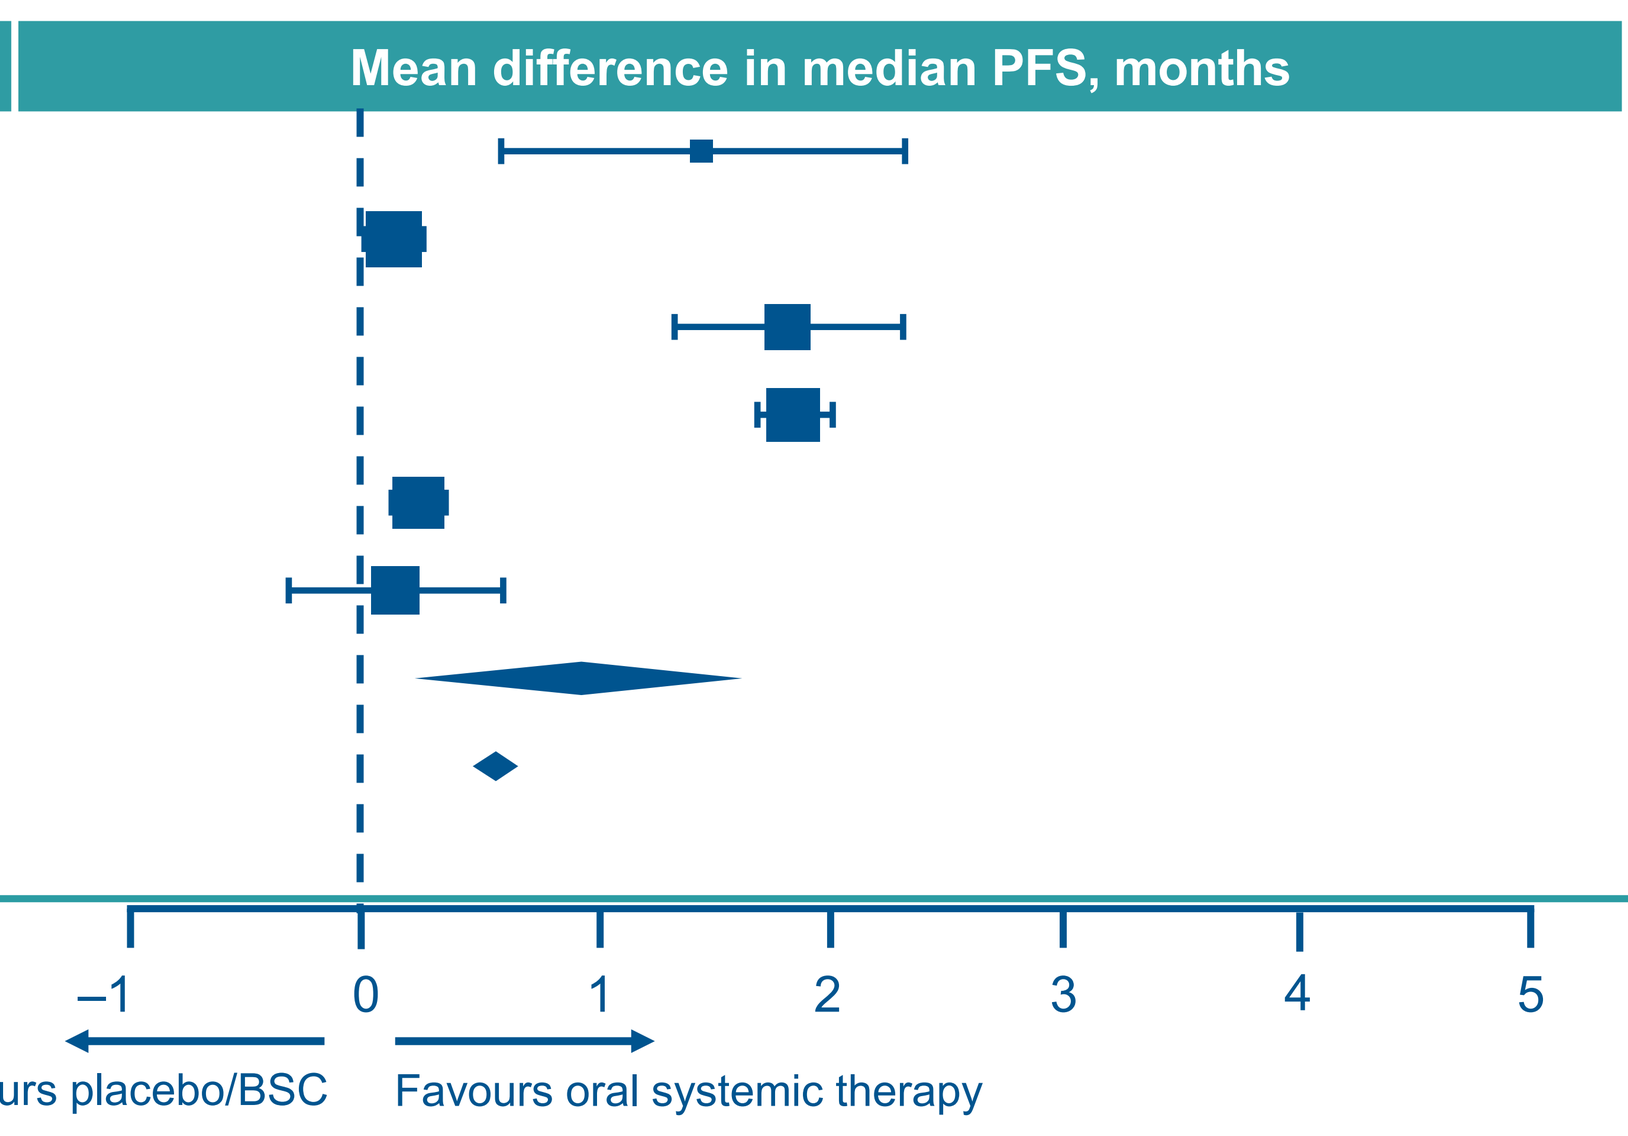
Mean difference in median PFS, months
–1
0
1
2
3
4
5
Favours placebo/BSC
Favours oral systemic therapy

Study	MD (95% CI)
CONCUR*	1.50 (0.64, 2.36)
CORRECT	0.20 (0.08, 0.32)
FRESCO*	1.87 (1.38, 2.36)
FRESCO-2	1.90 (1.74, 2.06)
RECOURSE	0.30 (0.19, 0.41)
TERRA*	0.20 (–0.25, 0.65)
RE Model	0.97 (0.28, 1.66)
FE Model	0.63 (0.56, 0.70)

Heterogeneity: I^2=98.63%, τ^2=0.7

*Trial was conducted only in Asia.
MD, mean difference.

Limitations

- The aim of this study was to characterize the survival benefit of patients receiving oral systemic therapy versus no active therapy; the study was not designed to indirectly compare the efficacy of the oral systemic therapies
- These analyses do not capture the impact of adverse events associated with systemic therapy or the impact on quality of life of choosing an oral systemic therapy over BSC. These are important considerations from the patient perspective when assessing the meaningfulness of survival gains
- Aside from differences in the number and types of prior therapies, as noted above in relation to FRESCO-2, there may be other sources of cross-trial heterogeneity that have not been explored here (e.g., trials conducted in Asia only vs global trials). Additionally, high statistical heterogeneity was present in the analyses of PFS specifically

Conclusions

- Oral systemic therapies may provide significant survival benefits for heavily pre-treated mCRC. In this meta-analysis, the improvement in median OS associated with oral systemic therapies versus placebo + BSC was <2 months, indicating that the increases in median OS with oral systemic monotherapies versus placebo + BSC observed in RCTs, in heavily pre-treated mCRC, are meaningful

References

1. National Cancer Institute. Cancer Stat Facts: Colorectal Cancer. 2022; https://seer.cancer.gov/statfacts/html/colorect.html. (Accessed October 08, 2024)
2. Ciardiello F, et al. *CA Cancer J Clin*. 2022;72:372–401
3. Li J, et al. *JAMA*. 2018;319:2486−96
4. Dasari A, et al. *Lancet*. 2023;402:41−53
5. Li J, et al. *Lancet Oncol*. 2015;16:619−29
6. Grothey A, et al. *Lancet*. 2013;381:303−12
7. Mayer RJ, et al. *N Engl J Med*. 2015;372:1909−19
8. Xu J, et al. *J Clin Oncol*. 2018;36:350−8
9. A. Cervantes, et al, on behalf of the ESMO Guidelines Committee. *Ann Oncol*. 2023;34:10−32 (v1.2, updated September 2024)
10. Viechtbauer W. *J Stat Softw*. 2010;36:1−48
11. Prager GW, et al. *N Engl J Med*. 2023;388:1657−67
12. Tabernero J, et al. *Ann Oncol*. 2024;35:S15−6
13. Tabernero J, et al. *ESMO Open*. 2020;5:e000752

Acknowledgments

Editorial support for the development of this poster, under the direction of the authors, was provided by Christian Jones, PhD, of Ashfield MedComms, an Inizio Company, funded by Takeda Pharmaceuticals U.S.A., Inc., Lexington, MA, and complied with the Good Publication Practice (GPP) guidelines (DeTora LM, et al. *Ann Intern Med*. 2022;175:1298–304).

Disclosures

VP: employment and stocks/shares with Takeda.

All remaining author disclosures can be accessed by scanning the QR code.

To view a copy of this poster on your own device, scan this quick response (QR) code or visit: https://tiny.one/ivX1754JrZ. Copies of this poster obtained through QR Code are for personal use only and may not be reproduced without permission from the author of this presentation.

Presented at the International Society for Pharmacoeconomics & Outcomes Research Annual European Congress (ISPOR EU), 17–20 November 2024, Barcelona, Spain. For questions or comments, please contact Dr Victoria Paly: victoria.paly@takeda.com.