EE481

Cost-Effectiveness Analysis of Upadacitinib in Patients with Moderately to Severely Active Ulcerative Colitis in Greece

Charalampos Tzanetakos[1], Marina Psarra[1], Ilias Kotsis[2], George Gourzoulidis[1]

[1]Health Through Evidence G.P., Athens, Greece; [2]AbbVie Pharmaceuticals S.A., Athens, Greece

OBJECTIVE

To evaluate the cost-effectiveness of upadacitinib in patients with moderately to severely active ulcerative colitis (UC), who have had an inadequate response, lost response or were intolerant to either conventional therapy (bio-naive) or a biologic agent (bio-exposed), in Greece

CONCLUSIONS

Upadacitinib was estimated to be more effective and cost-effective therapy versus all biologics and small molecule agents in the treatment of moderately to severely active UC in Greece

The present cost-effectiveness findings underpin the potential of upadacitinib as a promising treatment option in UC patients, where despite the currently available treatments, a large unmet need still exists

This study was funded by AbbVie Pharmaceuticals S.A.. AbbVie participated in the conceptualization, review, and approval of the publication. AbbVie had no interference in the study design, data collection, data analysis and interpretation, the writing of this study or the decision to submit it for publication. All authors critically reviewed this publication for important intellectual content and gave their approval for this version to be published. Authors would like to thank AbbVie Pharmaceuticals S.A. for funding this study.

Ilias Kotsis is an employee of AbbVie Pharmaceuticals S.A.. All authors declare no other competing interests.

References: 1. Levine, J.S. and R. Burakoff, Gastroenterol Hepatol (N Y), 2011. 7(4): p. 235-41. 2. Gajendran, M., et al. Dis Mon, 2019. 65(12): p. 100851. 3. Burisch, J., et al. Lancet Gastroenterol Hepatol, 2020. 5(5): p. 454-464. 4. Peyrin-Biroulet, L., et al. Aliment Pharmacol Ther, 2016. 44(8): p. 807-16. 5. Kennedy, N.A., et al. Aliment Pharmacol Ther, 2016. 43(8): p. 910-923. 6. Long, M.D., et al. Inflamm Bowel Dis, 2020. 26(6): p. 941-948. 7. D'Amico, F., et al. Therapeutic Advances in Gastroenterology, 2019. 12: p. 1-10. 7. European Medicines Agency (EMA). EPAR-Product information 2023.; Available from: https://www.ema.europa.eu/en/medicines/human/EPAR/Rinvoq. 8. Danese, S., et al. Lancet, 2022. 399(10341): p. 2113-2128. 9. NICE. Technology appraisal guidance [TA856]. 2023. 10. Panaccione, R., et al. Crohns Colitis 360, 2023. 5(2): p. otad009. 11. NICE. Technology appraisal guidance [TA633]. 2020. 12. Misra, R., et al. Can J Gastroenterol Hepatol, 2016. 2016: p. 8723949. 13. Woehl A, et al. Gut. 2008:A1-A172. 14. Arseneau, K.O., et al. Clin Gastroenterol Hepatol, 2006. 4(9): p. 1135-42. 15. Stevenson, M., et al. Health Technol Assess, 2016. 20(35): p. 1-610. 16. Greek Ministry of Health. Drug Price Bulletin. Available from: http://www.moh.gov.gr. Accessed June 2023. 17. Vellopoulou, K., et al. Eur J Gastroenterol Hepatol, 2021. 33(3): p. 325-333. 18. International Monetary Fund (IMF). World Economic Outlook Database. April 2023.

INTRODUCTION

- UC is a chronic inflammatory bowel disease (IBD) that affects the mucosa, the innermost lining of the intestinal wall in the large bowel (i.e., the colon and rectum)[1]
- Significant clinical, humanistic and economic burden is associated with UC[1-4]
- Despite the availability of multiple treatment options, many patients with UC do not achieve disease remission at 52 weeks with current advanced therapies (biologics and small molecule agents) and unmet therapeutic need remains in patients with moderately to severely active disease[5-7]
- Based on its clinical evidence, upadacitinib represents a promising oral advanced therapy (JAK inhibitor) in the therapeutic arsenal against this inflammatory disease[7,8]

METHODS

- A cost-effectiveness model was locally adapted from a public payer perspective[9]
- Response to treatment in the induction and maintenance phase was obtained from a network meta-analysis[10]
- Probabilities for surgical events and utility values were retrieved from published literature[11-15]
- Drug acquisition, administration, disease management, and adverse events costs were considered[16,17]

Induction phase decision tree

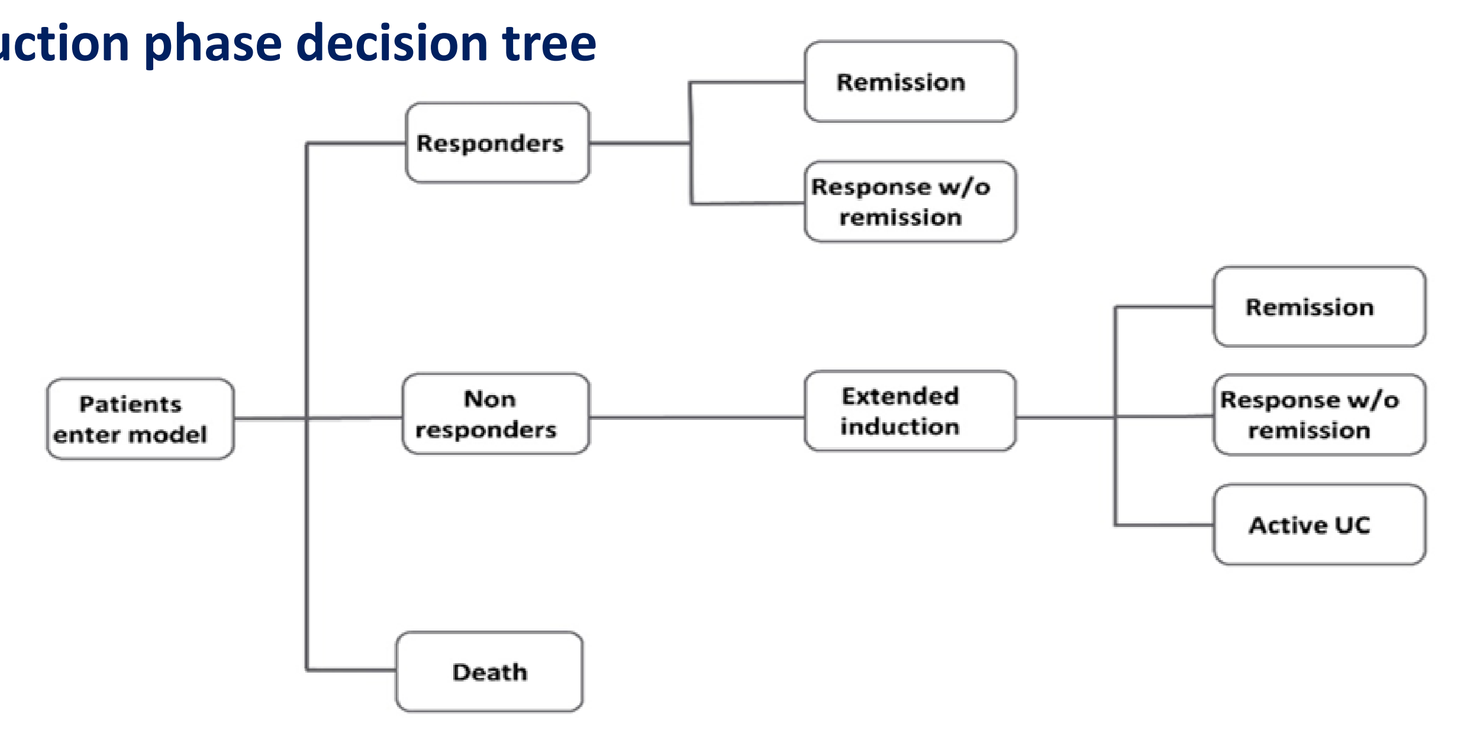
Patients enter model
Responders
Remission
Response w/o remission
Non responders
Extended induction
Remission
Response w/o remission
Active UC
Death

Long-term maintenance phase Markov model

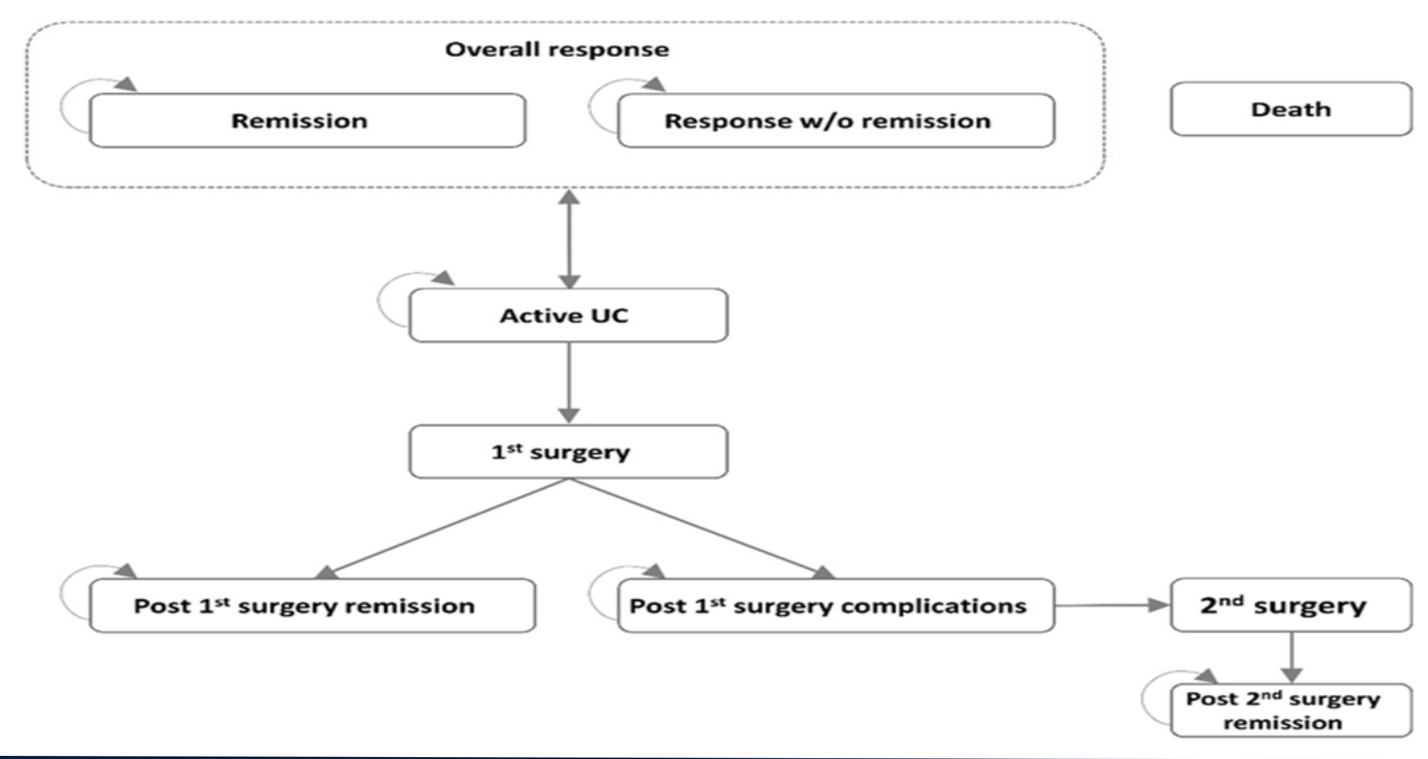
Overall response
Remission
Response w/o remission
Death
Active UC
1st surgery
Post 1st surgery remission
Post 1st surgery complications
2nd surgery
Post 2nd surgery remission

RESULTS

Base case pairwise results for bio-naïve and bio-exposed population

Therapy	Lifetime Total costs	Lifetime Total QALYs	Upadacitinib versus comparator (bio-naïve population)		
			Incremental costs	Incremental QALYs	Cost per QALY gained[a]
Upadacitinib	€ 112,780	9.030	-	-	-
Adalimumab	€ 97,279	8.198	€ 15,501	0.833	€ 18,618
Infliximab	€ 100,789	8.359	€ 11,991	0.671	€ 17,864
Golimumab	€ 98,262	8.361	€ 14,518	0.670	€ 21,682
Vedolizumab	€ 104,298	8.509	€ 8,481	0.522	€ 16,263
Tofacitinib	€ 103,339	8.716	€ 9,441	0.314	€ 30,061
Ustekinumab	€ 105,402	8.453	€ 7,378	0.577	€ 12,776
Ozanimod	€ 100,529	8.247	€ 12,251	0.783	€ 15,637

Therapy	Lifetime Total costs	Lifetime Total QALYs	Upadacitinib versus comparator (bio-exposed population)		
			Incremental costs	Incremental QALYs	Cost per QALY gained[a]
Upadacitinib	€ 109,730	8.928	-	-	-
Adalimumab	€ 96,882	8.144	€ 12,848	0.784	€ 16,396
Vedolizumab	€ 99,749	8.209	€ 9,981	0.719	€ 13,881
Tofacitinib	€ 100,955	8.414	€ 8,776	0.514	€ 17,074
Ustekinumab	€ 101,791	8.204	€ 7,940	0.723	€ 10,975
Ozanimod	€ 100,210	8.231	€ 9,520	0.697	€ 13,661

Notes: [a] All ICERs fall well within the cost-effectiveness threshold of €42,000 per QALY gained and most of them even lying below €21,000 per QALY gained [1 x GDP/capita] [18]
Abbreviations: ICER, Incremental cost- effectiveness ratio; QALY, Quality Adjusted Life Year.

Cost-effectiveness acceptability curves (multiple comparisons)

Bio-naïve population

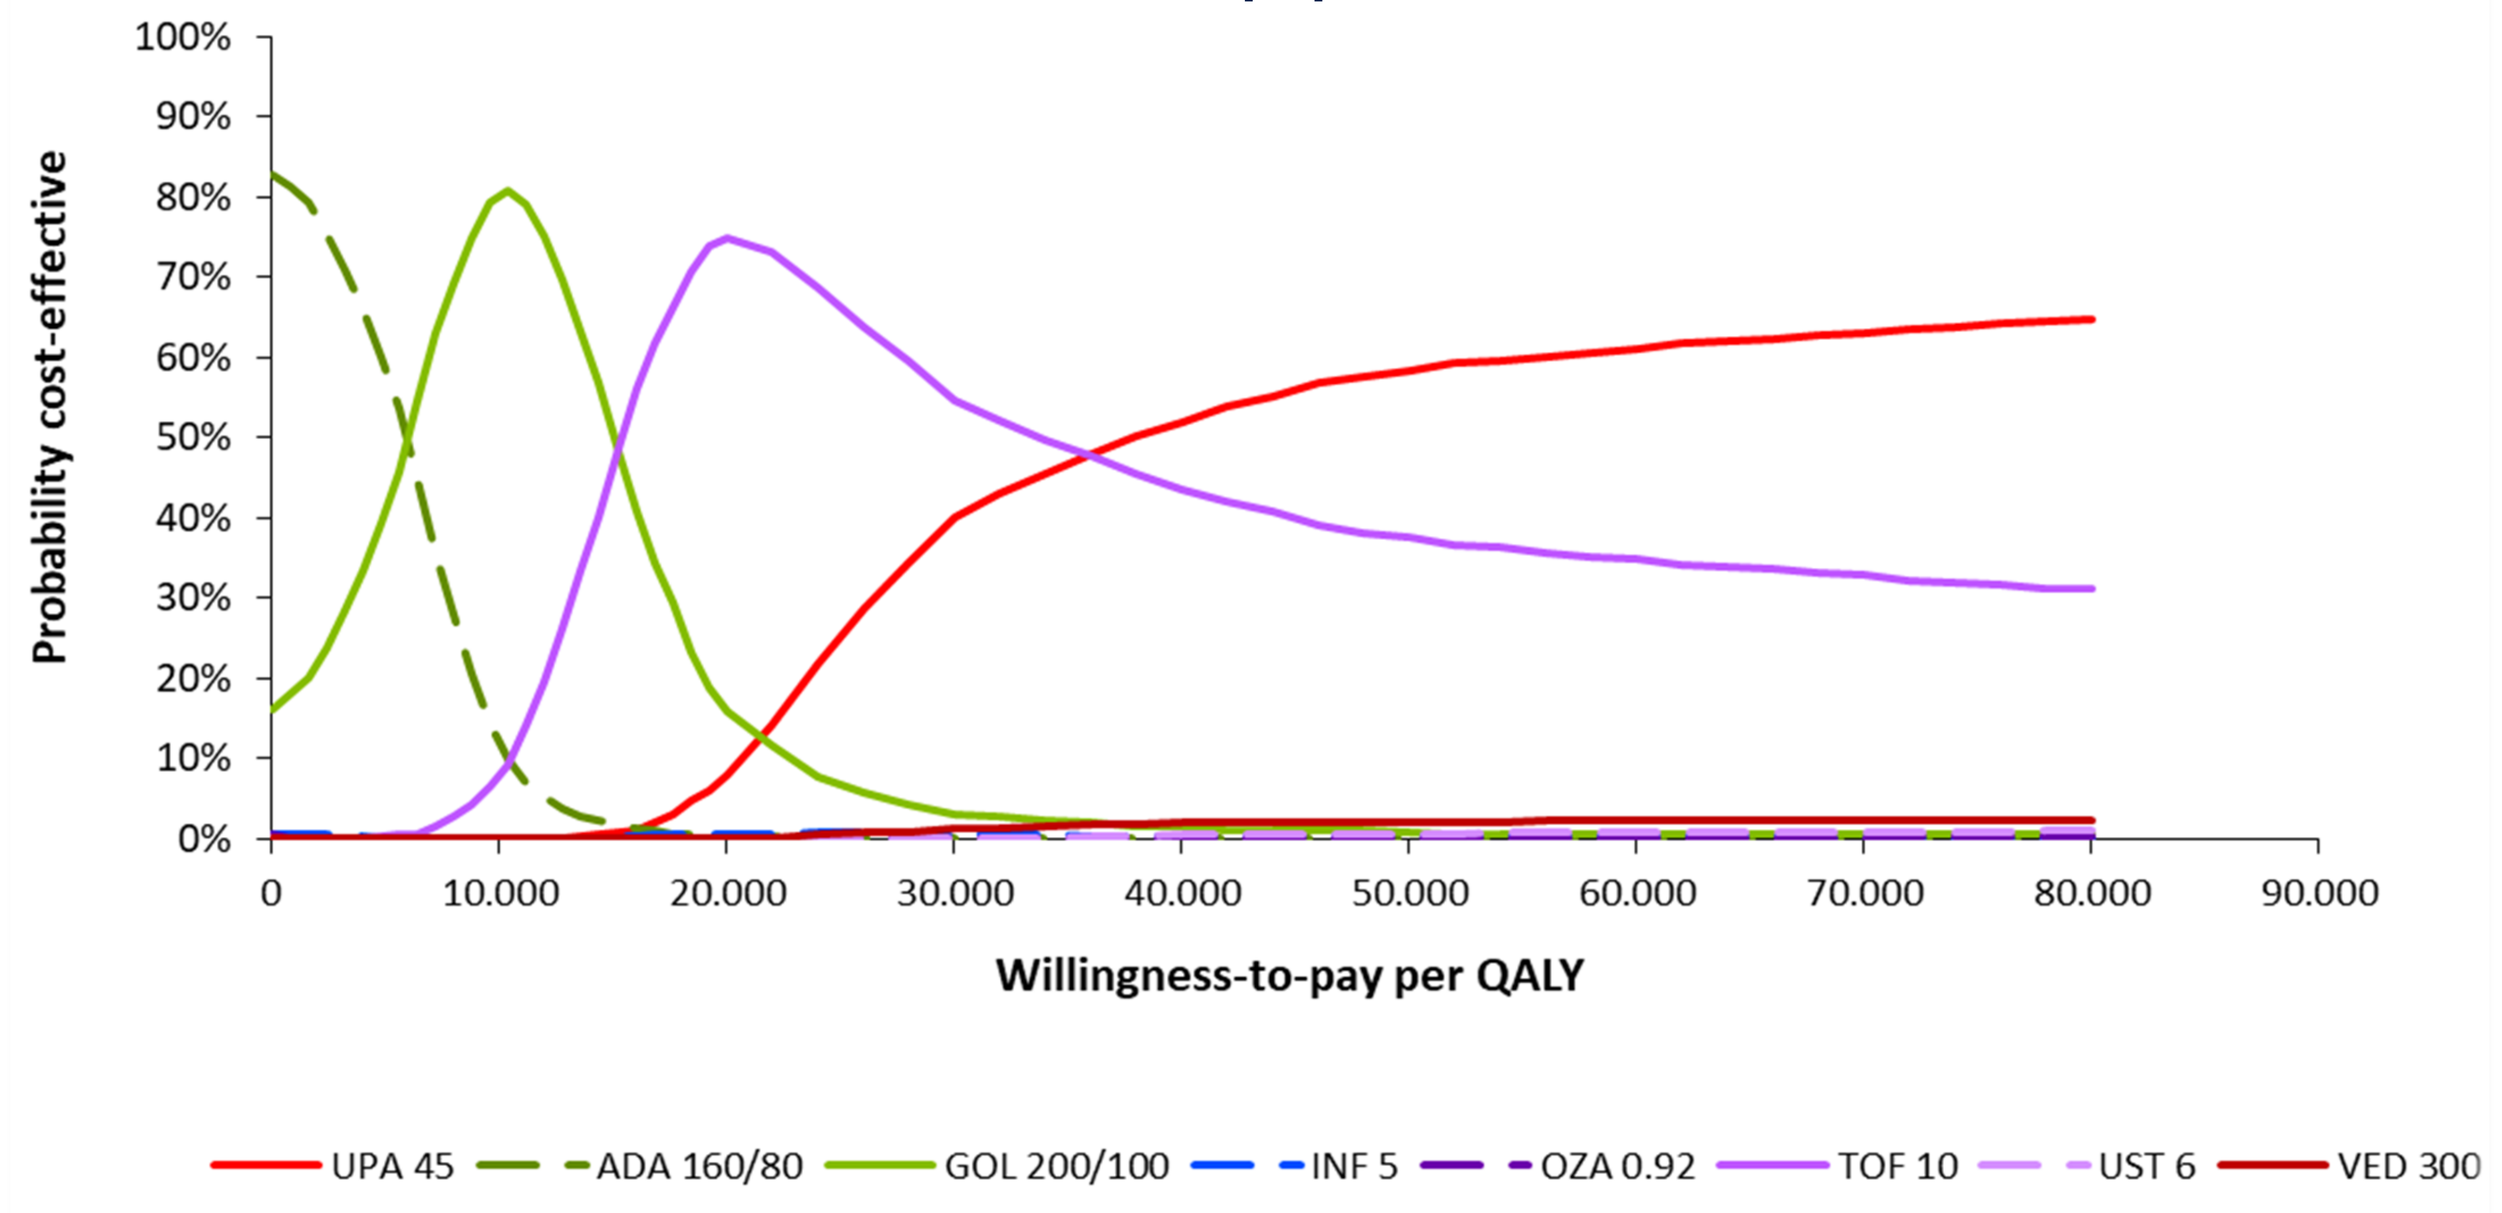
Probability cost-effective
100%
90%
80%
70%
60%
50%
40%
30%
20%
10%
0%
0
10.000
20.000
30.000
40.000
50.000
60.000
70.000
80.000
90.000
Willingness-to-pay per QALY
UPA 45
ADA 160/80
GOL 200/100
INF 5
OZA 0.92
TOF 10
UST 6
VED 300

Bio-exposed population

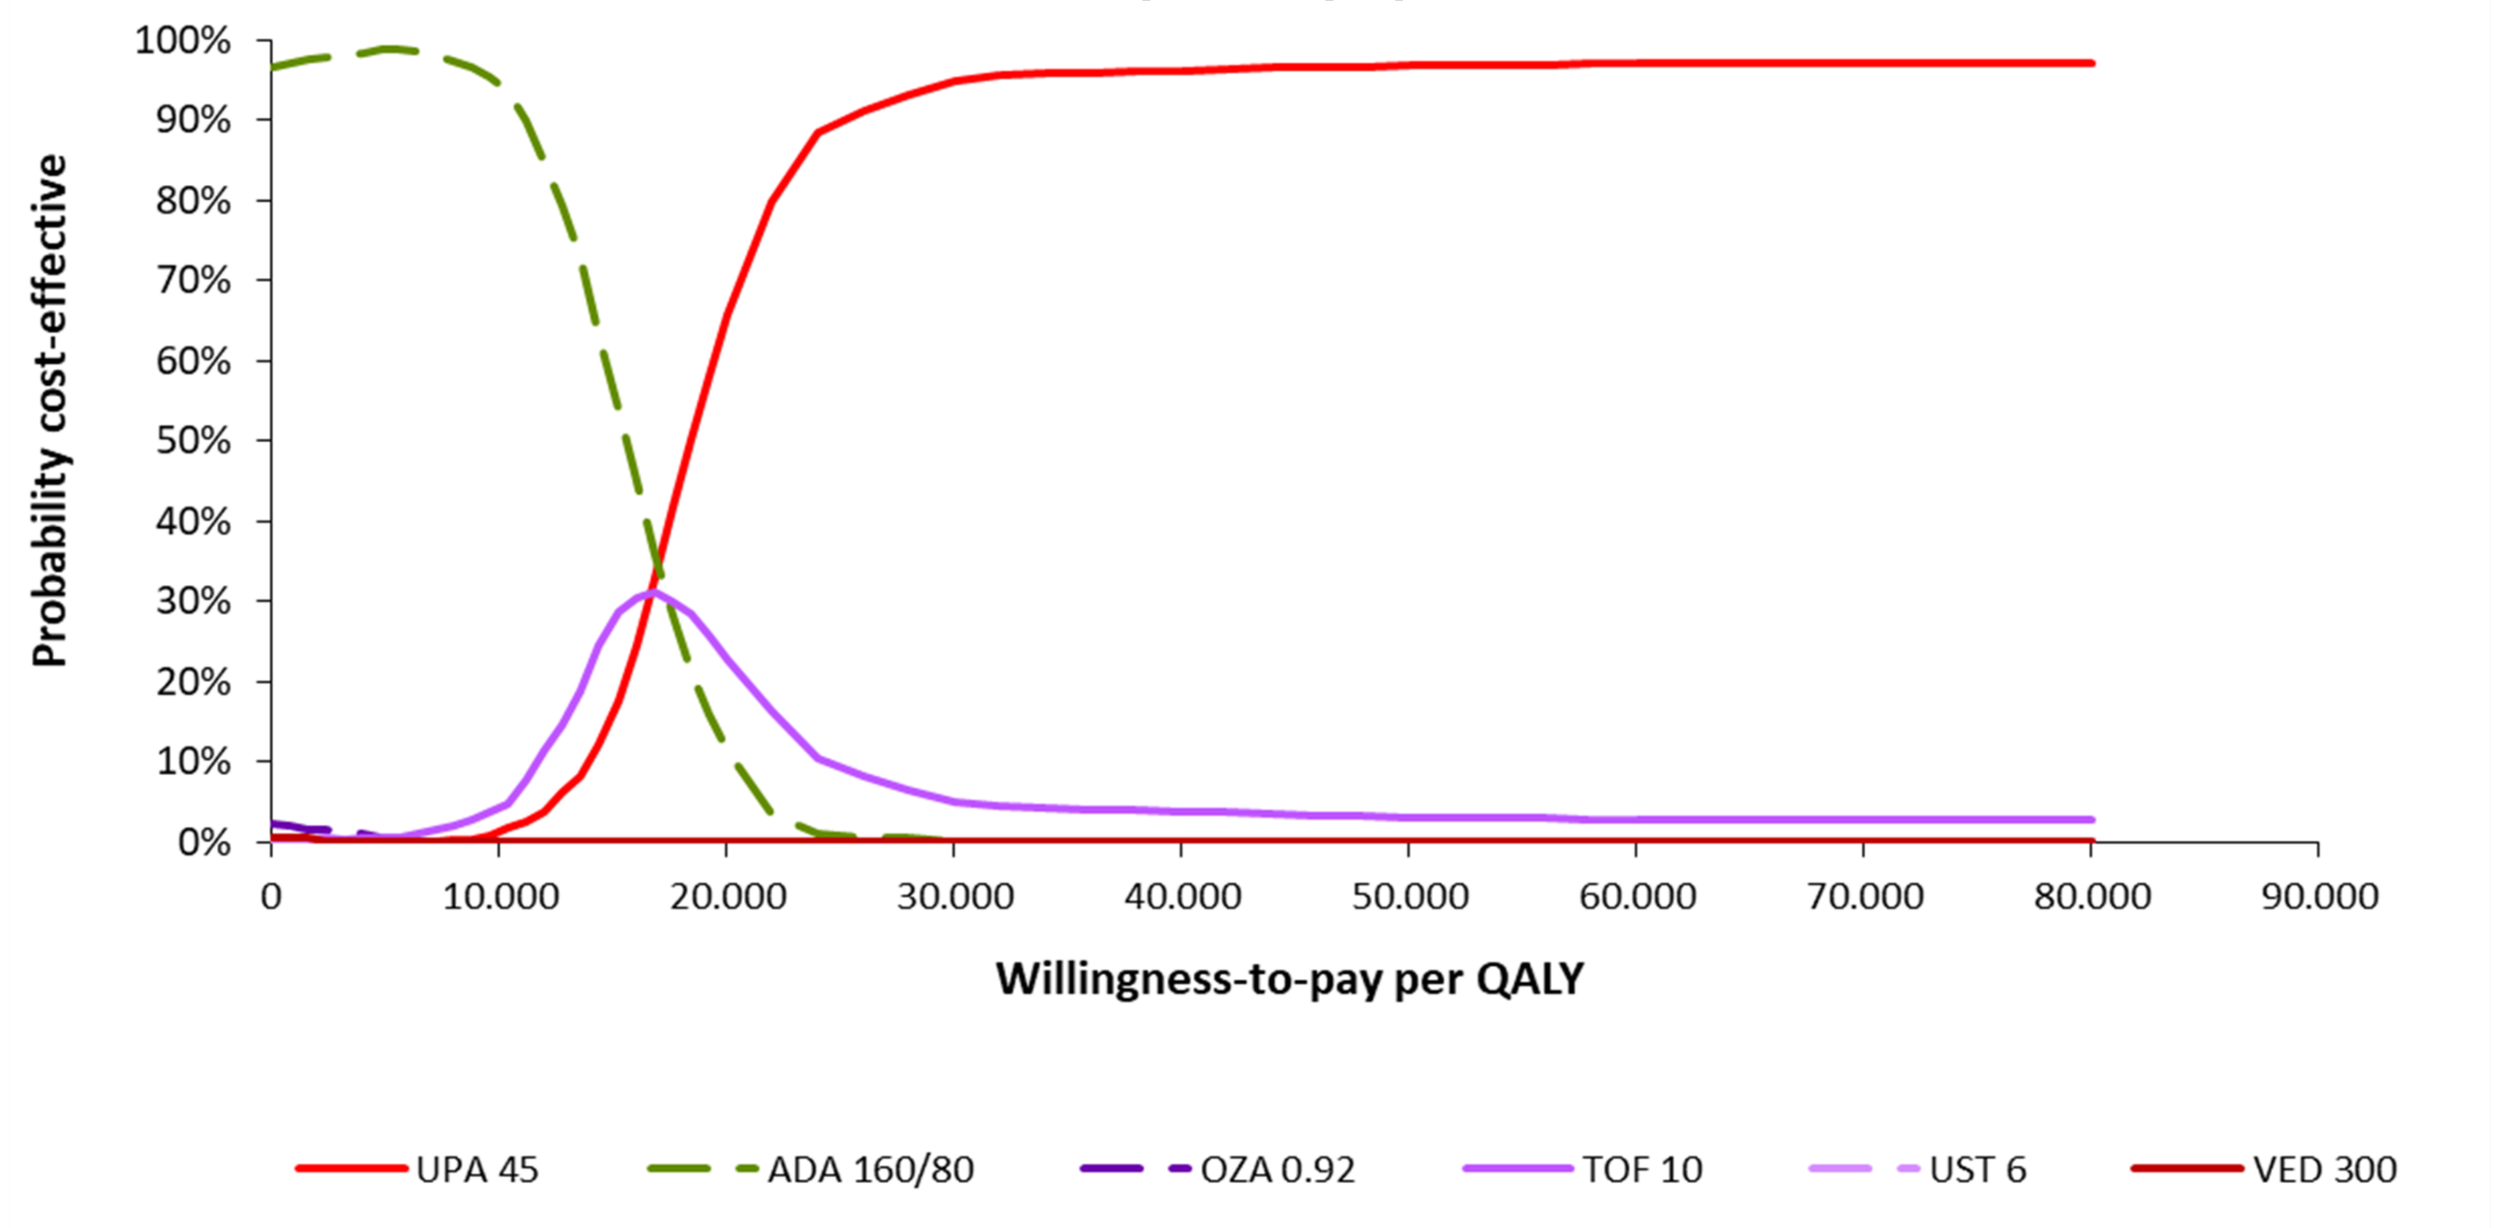
Probability cost-effective
100%
90%
80%
70%
60%
50%
40%
30%
20%
10%
0%
0
10.000
20.000
30.000
40.000
50.000
60.000
70.000
80.000
90.000
Willingness-to-pay per QALY
UPA 45
ADA 160/80
OZA 0.92
TOF 10
UST 6
VED 300

Deterministic and probabilistic sensitivity analysis (Upadacitinib versus most utilized biological [Infliximab] in Greece)

Tornado diagram - 20 Most Influential Parameters on net monetary benefit (NMB)

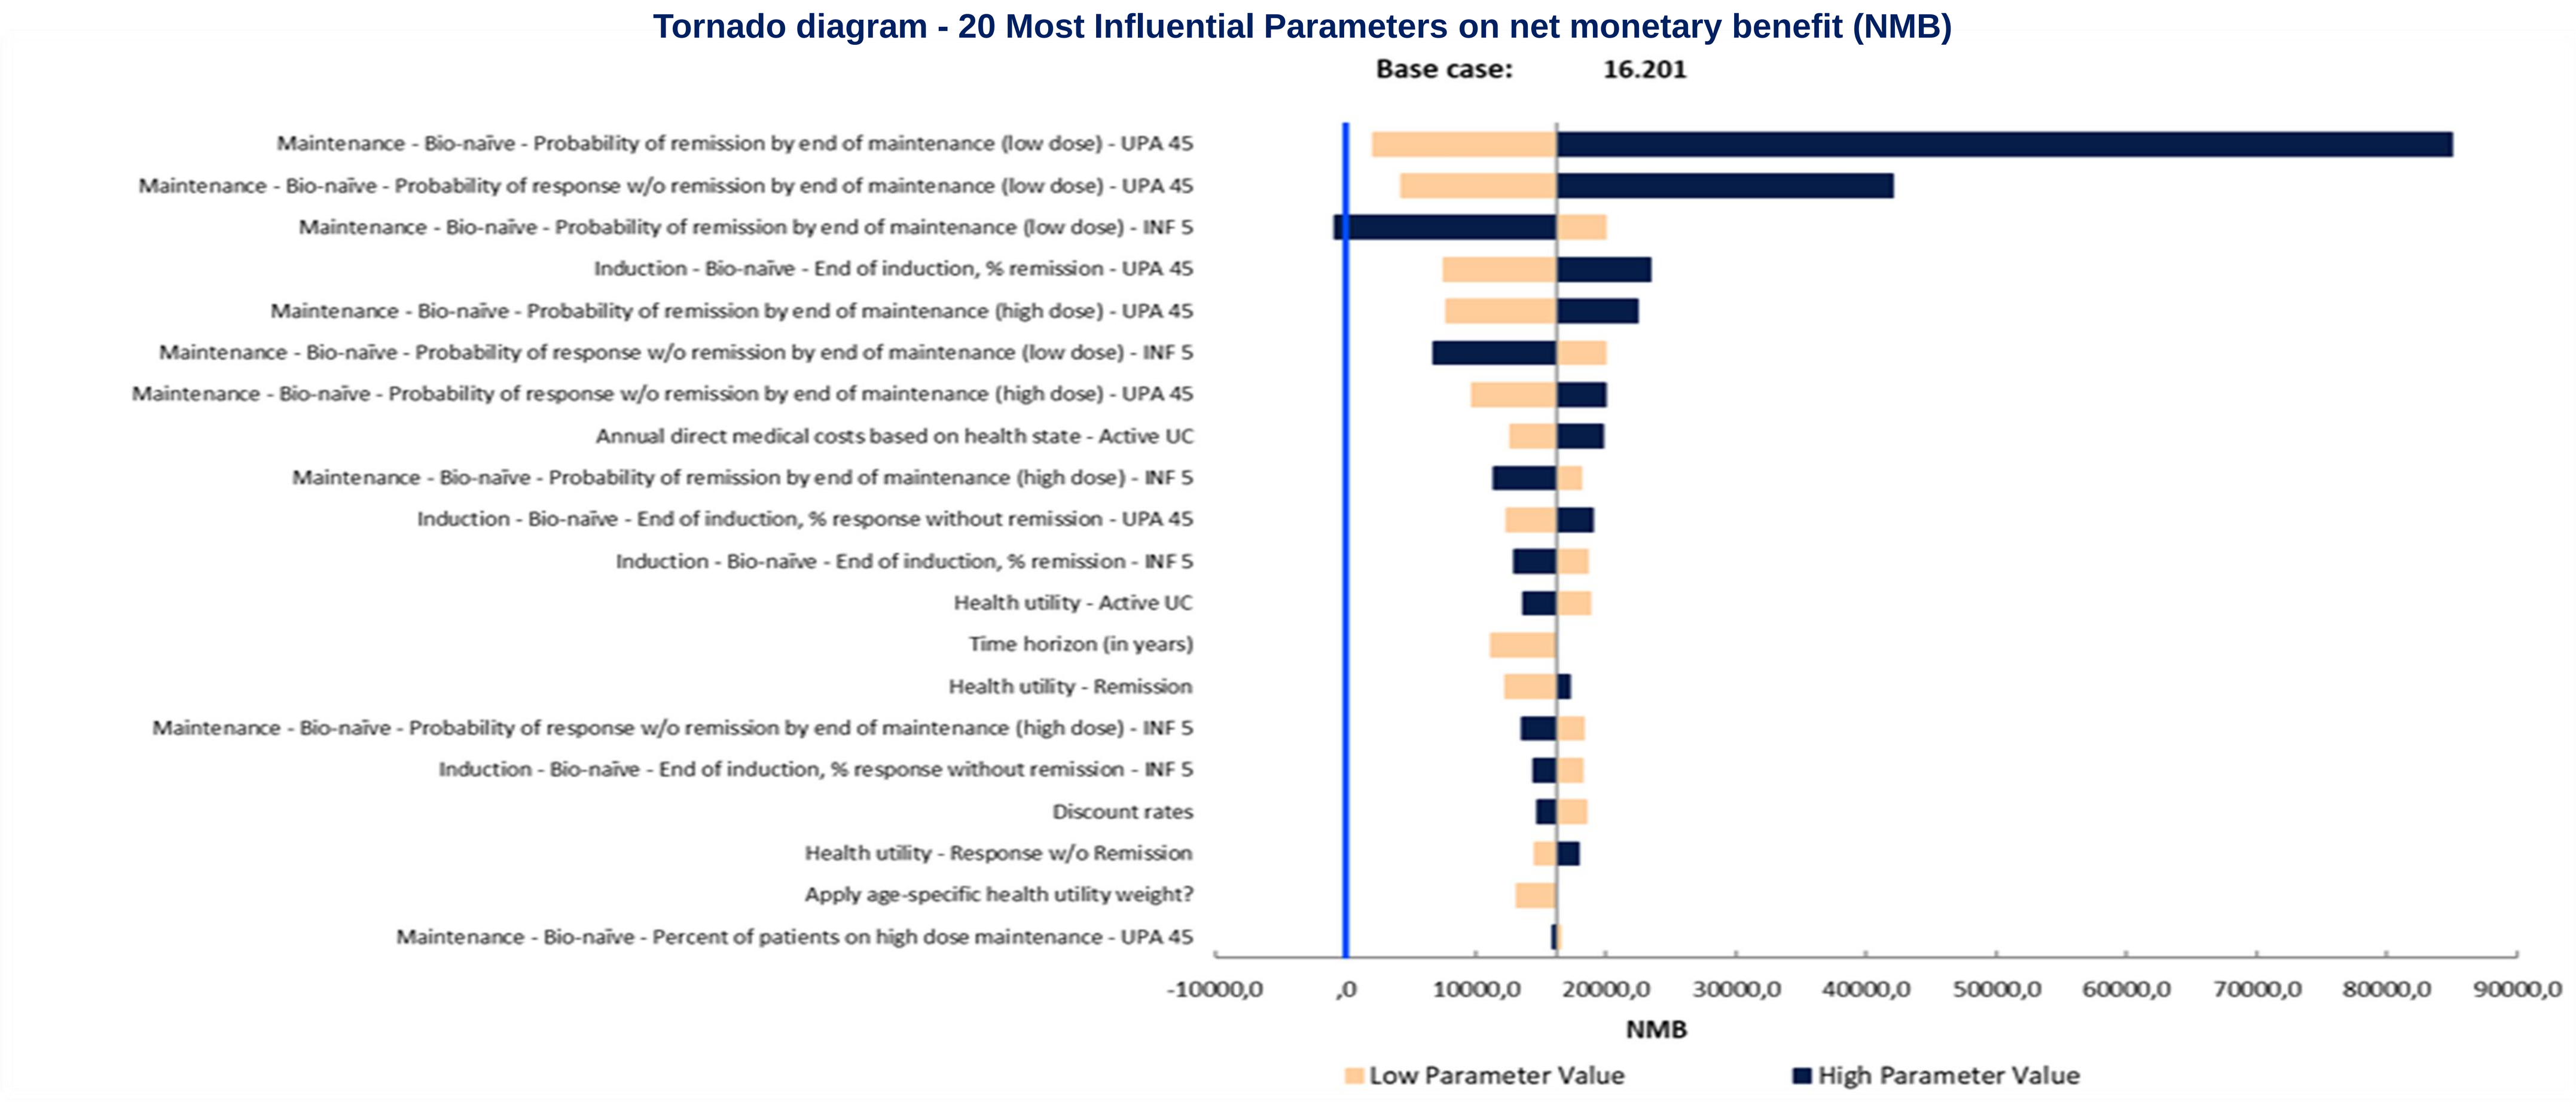
Base case: 16.201
Maintenance - Bio-naïve - Probability of remission by end of maintenance (low dose) - UPA 45
Maintenance - Bio-naïve - Probability of response w/o remission by end of maintenance (low dose) - UPA 45
Maintenance - Bio-naïve - Probability of remission by end of maintenance (low dose) - INF 5
Induction - Bio-naïve - End of induction, % remission - UPA 45
Maintenance - Bio-naïve - Probability of remission by end of maintenance (high dose) - UPA 45
Maintenance - Bio-naïve - Probability of response w/o remission by end of maintenance (low dose) - INF 5
Maintenance - Bio-naïve - Probability of response w/o remission by end of maintenance (high dose) - UPA 45
Annual direct medical costs based on health state - Active UC
Maintenance - Bio-naïve - Probability of remission by end of maintenance (high dose) - INF 5
Induction - Bio-naïve - End of induction, % response without remission - UPA 45
Induction - Bio-naïve - End of induction, % remission - INF 5
Health utility - Active UC
Time horizon (in years)
Health utility - Remission
Maintenance - Bio-naïve - Probability of response w/o remission by end of maintenance (high dose) - INF 5
Induction - Bio-naïve - End of induction, % response without remission - INF 5
Discount rates
Health utility - Response w/o Remission
Apply age-specific health utility weight?
Maintenance - Bio-naïve - Percent of patients on high dose maintenance - UPA 45
-10000,0
,0
10000,0
20000,0
30000,0
40000,0
50000,0
60000,0
70000,0
80000,0
90000,0
NMB
Low Parameter Value
High Parameter Value

Cost-effectiveness acceptability curve

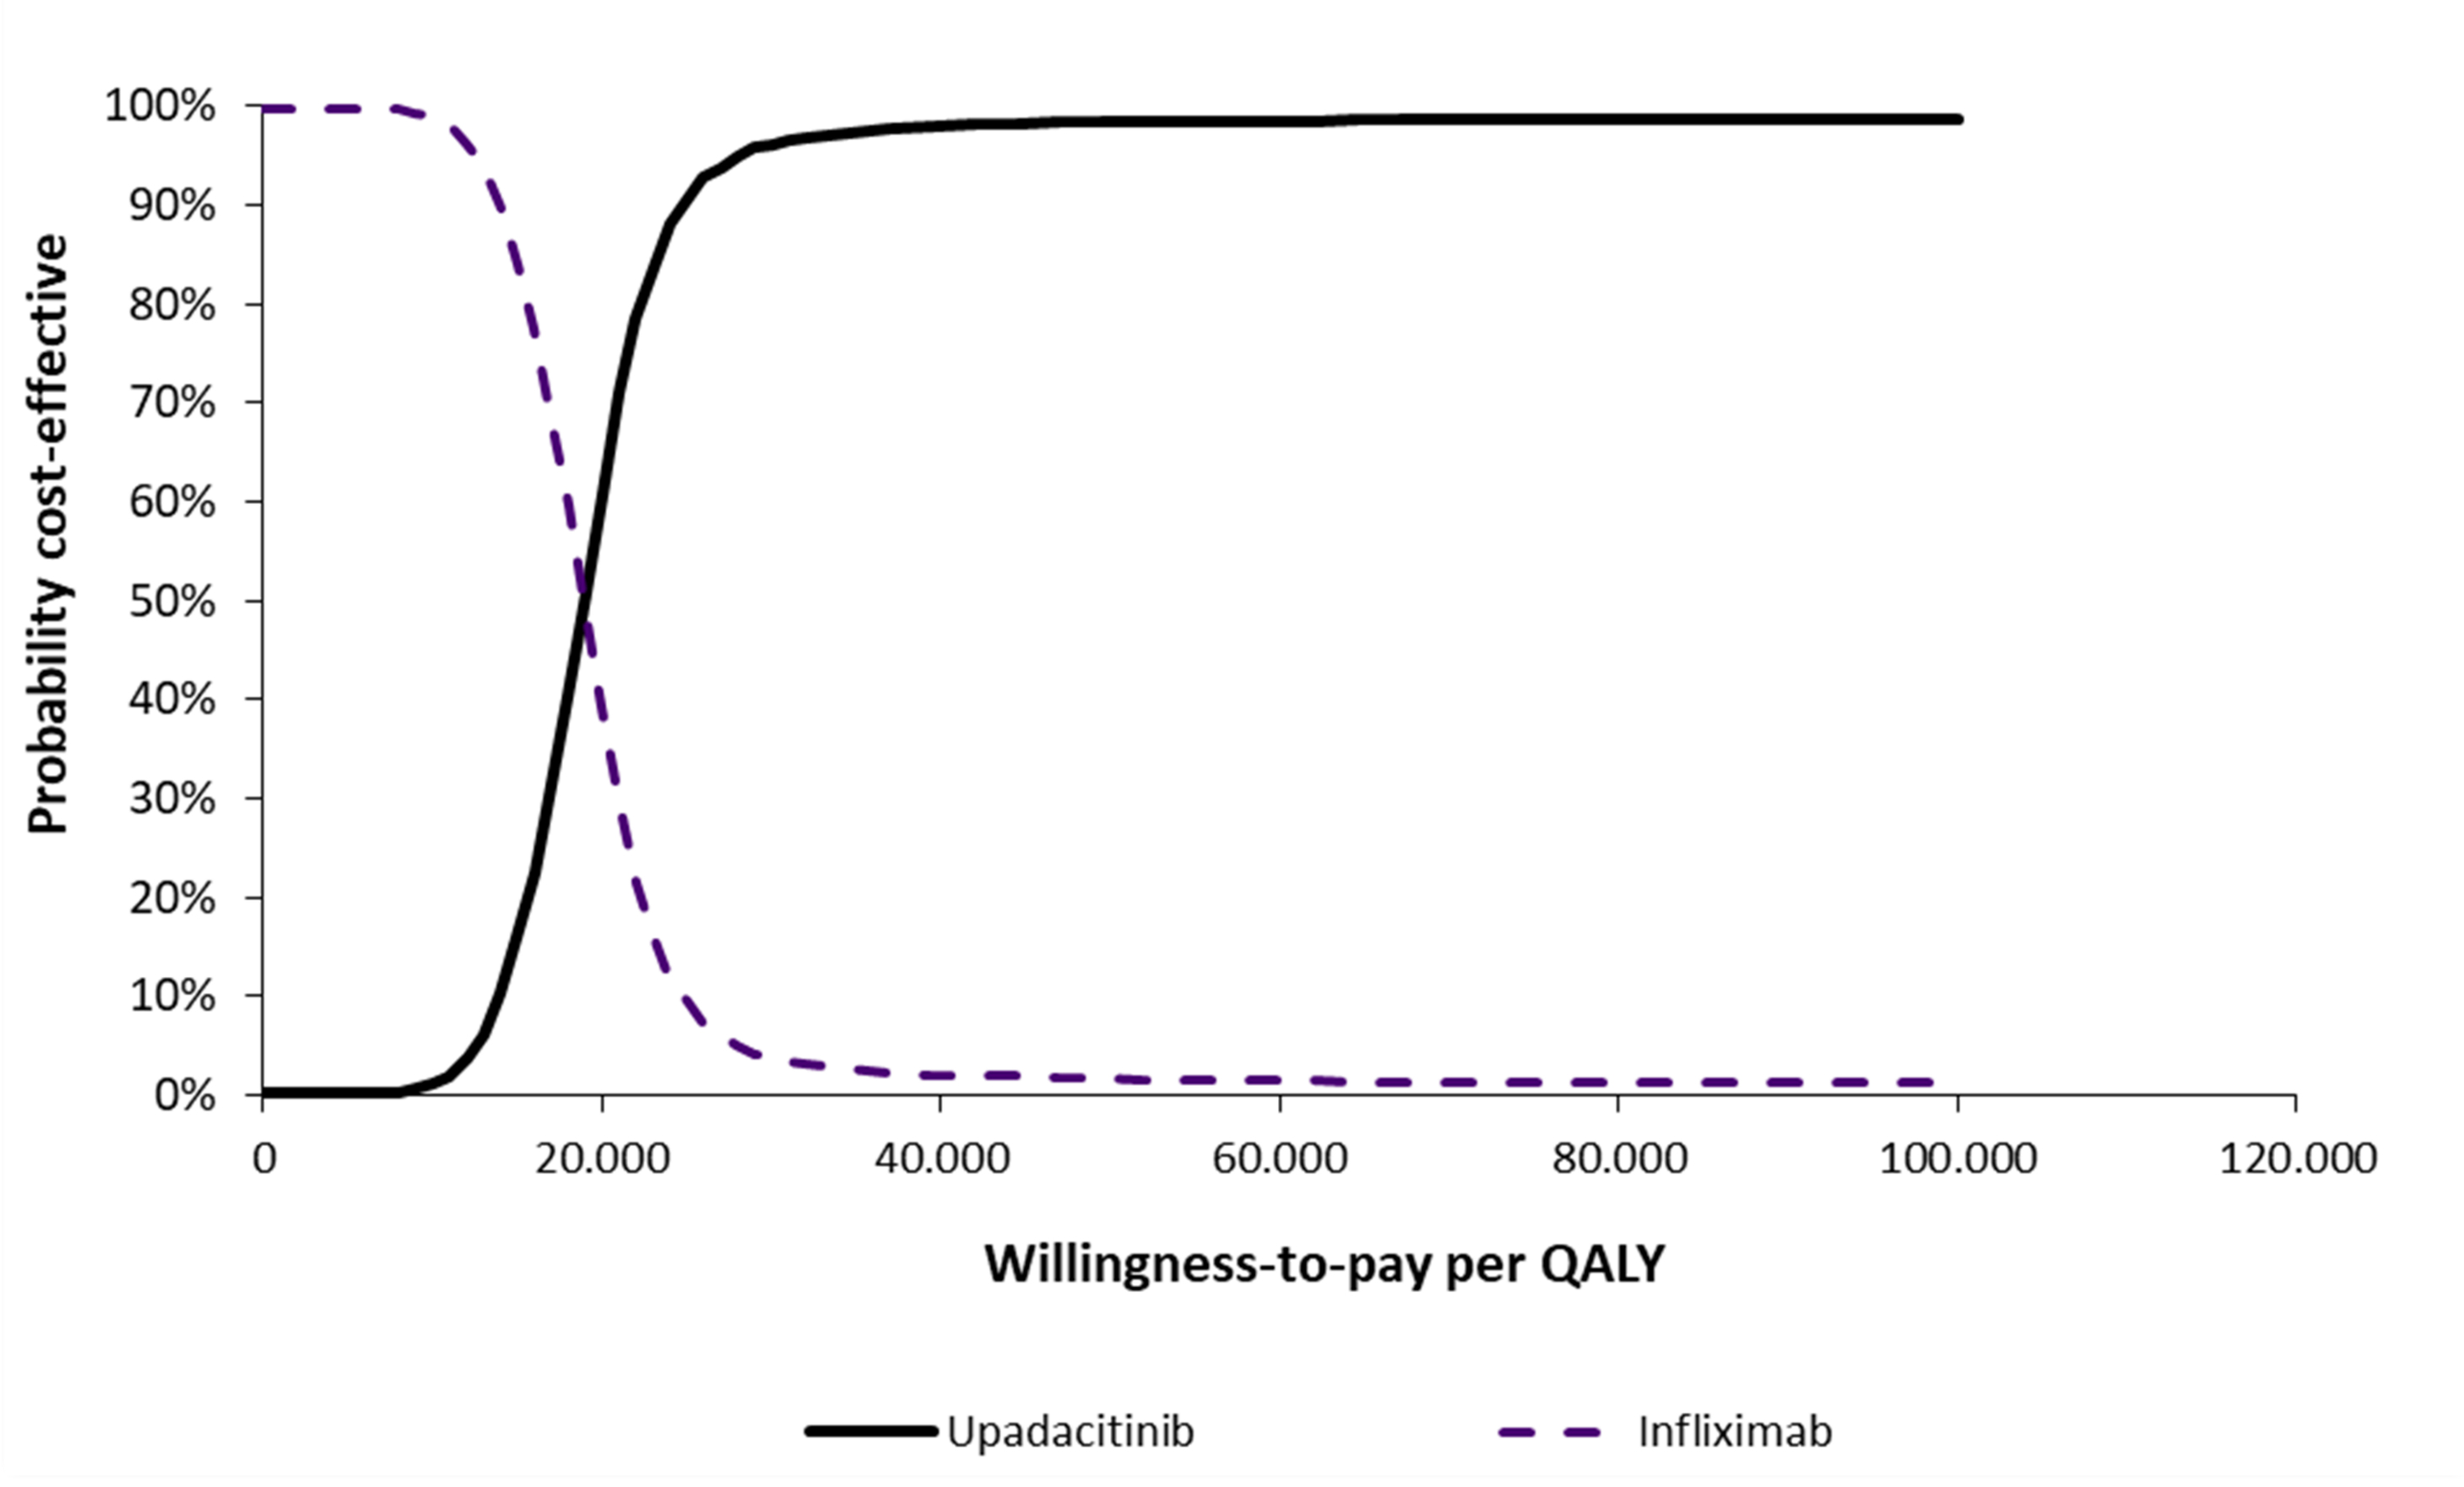
Probability cost-effective
100%
90%
80%
70%
60%
50%
40%
30%
20%
10%
0%
0
20.000
40.000
60.000
80.000
100.000
120.000
Willingness-to-pay per QALY
Upadacitinib
Infliximab

ISPOR ANNUAL EUROPEAN CONGRESS, 12 – 15 NOVEMBER 2023, COPENHAGEN, DENMARK
Contact details: c.tzanetakos@hte.gr