Cost-effectiveness of pembrolizumab plus chemotherapy, with or without bevacizumab for the first-line treatment of PD-L1-positive patients with persistent, recurrent, or metastatic cervical cancer in France

Isabelle Ray-Coquard[1], Patricia Pautier[2], Dominic Muston[3], Matthew Montberg[3], Shilpi Swami[4], Olivia Chaker[5], Lionel Bensimon[5], Lucile Marié[6], Suzy Lange[6], Margarita Kulakova[7]

1. Centre Léon Bérard
2. Gustave Roussy
3. Merck & Co., Inc., Rahway, NJ, USA
4. MSD (UK) Limited, London, UK
5. MSD France, Puteaux, France
6. Steve consultants, Lyon, France
7. Lumanity, Sheffield, UK

Background

- Keytruda® (pembrolizumab) is a humanized monoclonal antibody designed to block the Programmed Death-1 (PD-1) receptor, a negative regulator of T-cell anti-tumor defense.
- Pembrolizumab was recently approved by the EMA in association with chemotherapy with or without bevacizumab for the treatment of adults with persistent, recurrent, or metastatic cervical cancer PD-L1-positive.
- Approval was based on the results of the KEYNOTE-826 trial, a phase III trial that included a total of 617 patients with persistent, recurrent or metastatic cervical cancer. Patients were randomized to receive either pembrolizumab or placebo, with chemotherapy ± bevacizumab intravenously in a 1:1 ratio. The median follow-up was 18.2 months (range 0.5 to 29.4) in the pembrolizumab plus chemotherapy ± bevacizumab arm.
- There was a statistically significant improvement in overall survival in favor of pembrolizumab plus chemotherapy ± bevacizumab with a 36.0% reduction in the risk of recurrence or death (HR=0.64, IC95%: [0.50;0.81]).
- The French Health Technology Assessment (HTA) agency requires to assess the cost-effectiveness for innovative therapies, in order to help decision making regarding the drug price.

Objective

- The aim of the present analysis was to assess the cost-effectiveness of pembrolizumab with chemotherapy with or without bevacizumab (PEM + SoC) versus chemotherapy with or without bevacizumab (SoC) in PD-L1-positive (CPS≥1) patients with persistent, recurrent, or metastatic cervical cancer, from the French healthcare perspective.

Method

Economic model

- A 3-state semi-Markov model was developed to estimate, costs, effectiveness, and incremental cost-effectiveness ratio (ICER) of pembrolizumab with chemotherapy (with or without bevacizumab) versus chemotherapy (with or without bevacizumab) following French HTA (Haute Autorité de Santé (HAS)) guidelines[1].

Figure 1. Model structure

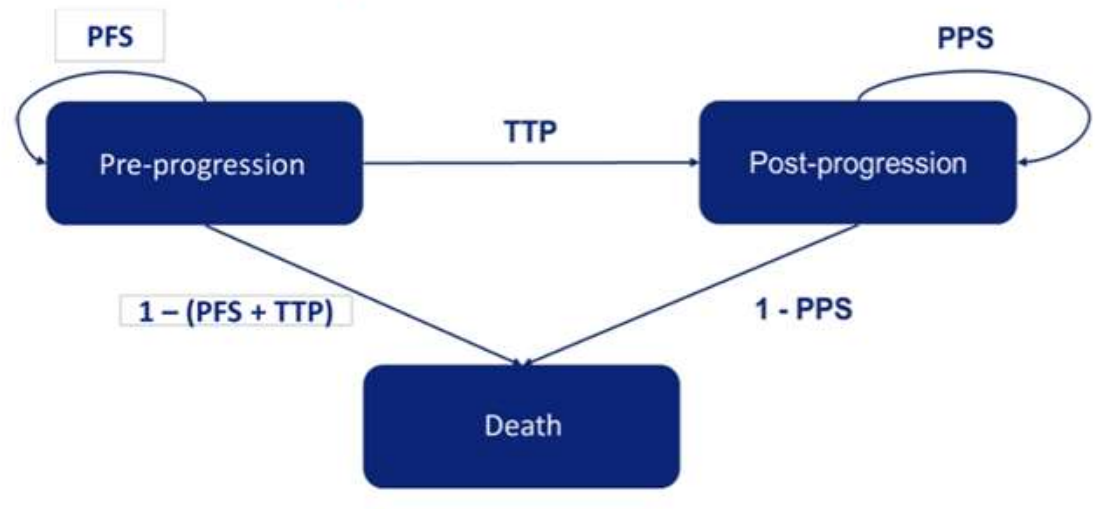
PFS
PPS
Pre-progression
TTP
Post-progression
1 – (PFS + TTP)
1 - PPS
Death

PFS : Progression-free survival ; PPS : Post-progression survival ; TTP : Time-to-progression

- Costs and health outcomes were projected over a 7-year time horizon (based on a trade-off between expected proportion of surviving patients in each treatment arm and uncertainty generated by extrapolations following HAS guideline) and were discounted at 2.5% per year. The duration of each cycle was 1 week.

Clinical parameters – efficacy and safety

- Transition between health states were extrapolated from progression-free survival (PFS), time to progression (TTP), and post-progression survival (PPS) from the KEYNOTE-826 study (data cutoff May 03, 2021). Piecewise parametric extrapolations were used.
- Grade 3+ all cause adverse events (AE) with an occurrence ≥2% in any of the treatment arms during the trial were modelled.

Clinical parameters – utility scores

- Utility data were collected from the KEYNOTE-826 study using EQ-5D-5L questionnaires and were converted to French population-based utilities using the French value set[2]. Mixed models for repeated measures (MMRM) were used.
- QALY loss related to the tolerance of pembrolizumab was considered.

Cost parameters

- Medical costs (in €2022) were assessed, from a health system perspective, taking into account all French heath system stakeholders.
- Costs included drug costs of first and later-line treatments (including acquisition and administration), follow-up, AE management, surgery, transportation and end of life.

Results

Base case analysis

- Over a 7-year time horizon, PEM + SoC was projected to increase (discounted) average life expectancy of 0.82 years (9.94 months) with an absolute gain of 0.66 QALYs (7.94 months spent in perfect health) versus the current standard of care.
- The average total cost of care over a 7-year time horizon for PEM + SoC was €161,014 (discounted) vs. €49,673 for chemotherapy ± bevacizumab (incremental cost of €111,341). These costs are mainly attributable to the drug cost (acquisition and administration), partially offset by savings in later-line treatments and end-of life expenses.
- ICER of PEM + SoC vs SoC was €168,076/QALY.

Table 1. Results of the base case analysis

Therapeutic strategy	Costs	LYs	QALYs	ICER (€/LY)	ICER (€/QALY)
SoC	€49,673	2.09	1.68		
PEM + SoC	€161,015	2.92	2.35	€134,390/LY	€168,076/QALY

Sensitivity analyses

- The univariate deterministic sensitivity analysis showed that the different numerical parameters of the model had limited impact on ICER (<5%).
- Probabilistic sensitivity analysis estimated a mean ICER of pembrolizumab plus chemotherapy ± bevacizumab vs. chemotherapy ± bevacizumab at €167,600/QALY (-0.3%).
- The acceptability curve shows that pembrolizumab plus chemotherapy ± bevacizumab has more than 80% probability of being cost-effective beyond the willingness-to-pay (WTP) threshold of €225,000/QALY compared to the current standard of care (Figure 2).

Figure 2. Cost-effectiveness acceptability curves

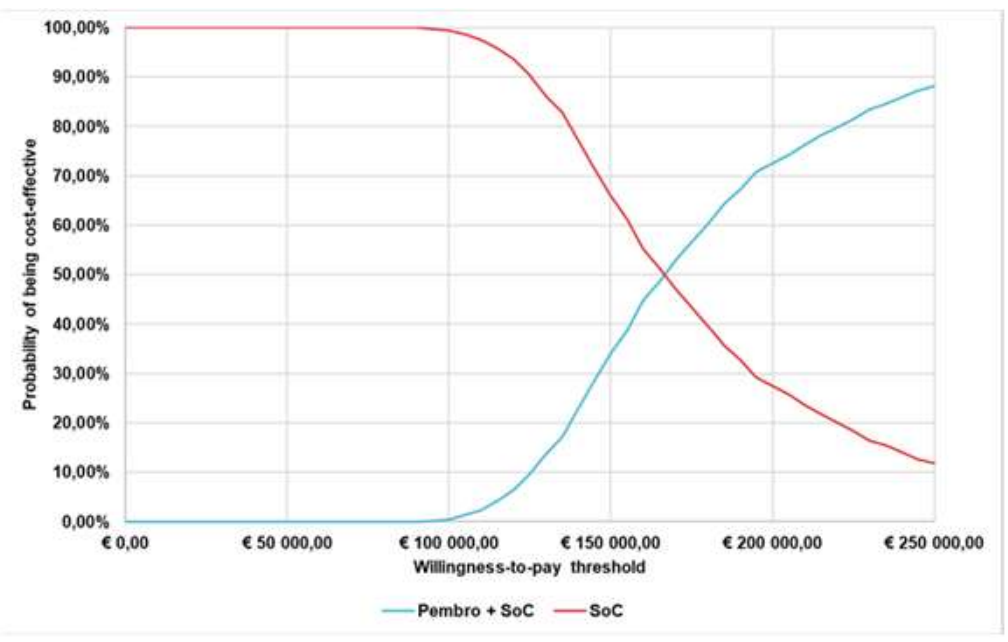
100,00%
90,00%
80,00%
70,00%
60,00%
50,00%
40,00%
30,00%
20,00%
10,00%
0,00%
Probability of being cost-effective
€ 0,00
€ 50 000,00
€ 100 000,00
€ 150 000,00
€ 200 000,00
€ 250 000,00
Willingness-to-pay threshold
Pembro + SoC
SoC

Scenario analyses

- The scenario with the greatest impact on the ICER was obtained when applying a treatment waning after 24 months of treatment (+34.66%).
- Exploratory subgroups analyses relative to the use of bevacizumab are associated with ICERs from €158,738/QALY in patients receiving bevacizumab to €214,450/QALY in patients not receiving bevacizumab.

Conclusion

Model-based analysis suggests that combining pembrolizumab with chemotherapy, regardless of the use of bevacizumab improves life expectancy and has more than 80% probability of being cost-effective versus the current standard of care assuming a WTP under €225,000/QALY. Results were robust to scenario analyses testing structural and methodological assumptions and were accepted by the French HTA agency.

1. HAS. Methodological recommendation – Methodological choices for cost-effectiveness assessment2020.
2. Andrade, L.F., et al., A French Value Set for the EQ-5D-5L. Pharmacoeconomics, 2020. 38(4): p. 413-425

ISPOR Europe 2023, 12-15 Nov, Copenhagen, Denmark

Copyright © 2023 Merck & Co., Inc., Rahway, NJ, USA and its affiliates. All rights reserved.