Estimating Health-Related Quality of Life in Fabry Disease for Patients Treated with Enzyme Replacement Therapy in the BALANCE Randomized Controlled Trial

PCR86

Author Names: Lee B[a], Hemstock M[a], Stevenson A[b], Stork R[b], Azimpour K[c]
Author affiliations: [a] Lumanity, Sheffield, UK, [b] Chiesi Limited, Manchester, UK, [c] Chiesi, Boston, MA, USA

Study Objectives

This utility analysis aimed to estimate the health-related quality of life (HRQoL) of patients with Fabry disease (FD) in the BALANCE randomized controlled trial. Furthermore, our analysis aimed to understand the impact that adverse events have on a patient's HRQoL.

Background

Fabry disease

FD is a rare, progressive X-linked lysosomal storage disorder involving renal, cardiovascular, respiratory, skin and gastrointestinal systems.[1] Real-world data from the UK Clinical Practice Research Datalink (2000-2019) reported an FD point prevalence of 3.7 per 100,000 persons in England.[1]

FD is caused by mutations in the galactosidase alpha gene, resulting in deficiency of α-galactosidase A (α-Gal A) enzyme.[2] Significant reduction/absence of α-Gal A results in progressive accumulation of glycolipids in cells throughout the body. As FD is X-linked, men have only one copy of the defective gene, so they are more likely to develop symptomatic disease. In general, people with FD have either:

- **Markedly deficient (< 5%) α-Gal A activity**: Generally considered to be classic FD[3]. Symptoms usually develop during childhood and can be severe
- **Some α-Gal A activity**: Generally considered to be non-classic FD, characterized by a more variable disease course, in which patients are generally less severely affected and disease manifestations may be limited to a single organ.[4] Patients can remain asymptomatic for many years before being diagnosed with FD[5]

Of note, some women with FD can have normal α-Gal A activity. Women generally have later symptom onset than men, but they can develop the classic phenotype due to a skewed X-chromosome inactivation pattern.[2]

Treatment options

Given the heterogeneous nature of FD, patients are treated on an individual basis.[5, 6] UK guidelines recommend adults diagnosed with FD are treated with enzyme-replacement therapies (ERTs), agalsidase beta or agalsidase alfa, and patients with an amenable mutation can also be treated with miglastat.[7] There are several limitations with current ERTs, such as a short circulatory half-life and ability to induce neutralizing anti-drug antibodies. Therefore, there is a clear need for additional ERT options for patients with FD.

Pegunigalsidase alfa

Pegunigalsidase alfa (PA) (Elfabrio®) is a newly approved pegylated ERT for the treatment of adults with FD, designed to offer a prolonged half-life.[8,9] PA has been studied previously in a comprehensive trial programme in 142 patients with FD, including those previously treated with ERTs and treatment-naïve patients.

The BALANCE trial

BALANCE[10,11] is the largest Phase III randomized active control trial conducted in FD. BALANCE compared PA with agalsidase beta (both dosed every 2 weeks) in patients with renal impairment, previously treated with agalsidase beta. 77 patients received either PA (n=52) or agalsidase beta (n = 25).

To estimate HRQoL and generate utility values, patient characteristics need to be considered and potentially adjusted for. Key baseline characteristics are presented in Table 1 for patients in the BALANCE trial.

Table 1. Patient baseline characteristics in BALANCE*

Characteristic	Type	PA (n=50)	Agalsidase beta (n=25)	Total (n=75)
Age	Mean (SD)	44.1 (11)	45.1 (10)	44.5 (10)
Sex	Female, n (%)	22 (44)	7 (28)	29 (39)
	Male, n (%)	28 (56)	18 (72)	46 (61)
Country	USA, n (%)	32 (64)	18 (72)	50 (67)
	Ex-USA, n (%)	18 (36)	7 (28)	25 (33)
Weight	Mean (SD)	77.7 (17)	81.2 (19)	78.9 (18)
Disease type**	Classic, n (%)	26 (52)	14 (56)	40 (53)
	Non-classic, n (%)	24 (48)	11 (44)	35 (47)
eGFR	Mean (SD)	73.7 (20)	74.2 (21)	73.9 (20)
	eGFR ≤ 60, n (%)	12 (24)	8 (32)	20 (27)
	60 < eGFR ≤ 90, n (%)	27 (54)	11 (44)	38 (51)
	eGFR ≥ 90, n (%)	11 (22)	6 (24)	17 (23)
UPCR	UPCR ≤ 0.5 gr/gr, n (%)	35 (70)	20 (80)	55 (73)
	0.5 < UPCR < 1 gr/gr, n (%)	8 (16)	2 (8)	10 (13)
	UPCR ≥ 1 gr/gr, n (%)	7 (14)	3 (12)	10 (13)

*The presented baseline characteristics are for patients that completed one or more follow-up EQ-5D questionnaire(s).
**In order to be classified as FD classic, a patient had to have ≤ 5% mean of lab normal ranges residual enzymatic activity in plasma or leukocytes at the baseline visit, and at least one Fabry-specific symptom: cornea verticillata, acroparesthesias, or angiokeratomas.

Adverse events in BALANCE

Adverse events are assumed to impact utility and therefore were explored in our analysis. For this utility analysis, four types of adverse event were considered; pain-related, neuropathic pain-related, severe adverse events, and Fabry clinical events (FCEs). The incidence of FCEs was defined based on adverse events, regardless of their seriousness/severity, and events were counted irrespective of their relatedness to the drug. The FCEs can also be split into the subcategories of cardiac, cerebrovascular and renal events. However, for this utility analysis, the FCE subcategories were grouped together as it was considered that there were not sufficient data to investigate FCEs by subcategory.

Key: α-Gal A, α-galactosidase A; AE, adverse events; Agal, Agalsidase; eGFR, estimated glomerular filtration rate; ERT, enzyme- replacement therapy; EQ-5D-5L, EuroQol 5 Dimension 5 Level; FCE; Fabry clinical event; FD, Fabry disease; HRQoL, health-related quality of life; ITT, intention to treat; n, number; NICE, National Institute for Health and Care Excellence; SD, standard deviation; PA, pegunigalsidase alfa; UPCR, urine protein creatinine ratio.

References: **1.** K Malottki, et al. Value in Health. 2022; 25 (12). **2.** Germain DP, et al. Genet Med. 2019; 21(9): 1987-97. **3.** M Arends, et al. Am J Nephrol. 28(5): 1631-1641. **4.** NICE. Highly Specialised Technology Evaluation Migalastat for treating Fabry disease [ID868]. 2016. **5.** Wanner C, et al. Mol Genet Metab. 2018; 124(3): 189-203. **6.** Wanner C, et al. Mol Genet Metab. 2019; 126(3): 210-1. **7.** British Inherited Medical Disease Guidelines. Guidelines for the treatment of Fabry Disease. 2020. **8.** Kizhner T, et al. Mol Genet Metab. 2015; 114(2): 259-267. **9.** Ruderfer I, et al. Bioconjug Chem. 2018; 29(5): 1630-9. **10.** Bernat J. Genet Med Open. 2023; 1 (1) supplement. **11.** Wallace E, et al. Safety and Efficacy BALANCE poster, protalix biotherapeutics website, 2022 **12.** NICE health technology evaluations: the manual. 2022. **13.** Alava MH, et al. Estimating the relationship between EQ-5D-5L and EQ-5D-3L: Results from an English Population Study. University of Sheffield & University of York. 2020.

Methods

In BALANCE, patients were randomized to either PA or agalsidase beta; however, in this utility analysis, data were pooled from both arms to increase the sample size. This approach was taken as the analysis' objective was to assess the overall HRQoL of patients with FD, rather than assess treatment impact on HRQoL.

Patients in BALANCE completed EQ-5D-5L questionnaires to measure their HRQoL at baseline and every 6 months up to Month 24. These data were then mapped to EQ-5D-3L using the EEPRU algorithm as per NICE guidance.[12, 13] Linear mixed-effects models were then fitted to this data. To account for multiple quality of life (QoL) observations per patient, patient IDs were modelled using a random intercept. Covariates that could be considered clinically relevant were used within a forwards selection algorithm. The 11 covariates considered for the regression analysis are: baseline utility score, age (continuous), sex (male/female), FD class (classic/non-classic), baseline eGFR (eGFR ≤ 60 / 60 < eGFR ≤ 90 / eGFR > 90), UPCR (UPCR ≤ 0.5 / 0.5 < UPCR < 1 / UPCR ≥ 1), treatment (PA/agalsidase beta), severe adverse events (pre/post event), pain-related adverse events (pre/post event), neuropathic pain adverse events (pre/post event), and Fabry clinical events (pre/post event).

Results

Correlation matrices

Before fitting a regression model, the correlation between the 11 covariates was assessed using matrices of correlation coefficients, such as Figure 1. This showed that 'FD class' and 'sex' were highly correlated. In particular, the correlation coefficient was calculated at 0.85, and equivalently -0.85, indicating that females in the trial are more likely to have non-classic FD. This level of correlation could lead to confounding in the final model. As such, sex was determined to be a more important covariate, and the 'FD class' covariate was excluded.

Figure 1. Matrix of correlation coefficients for baseline assessments of utility

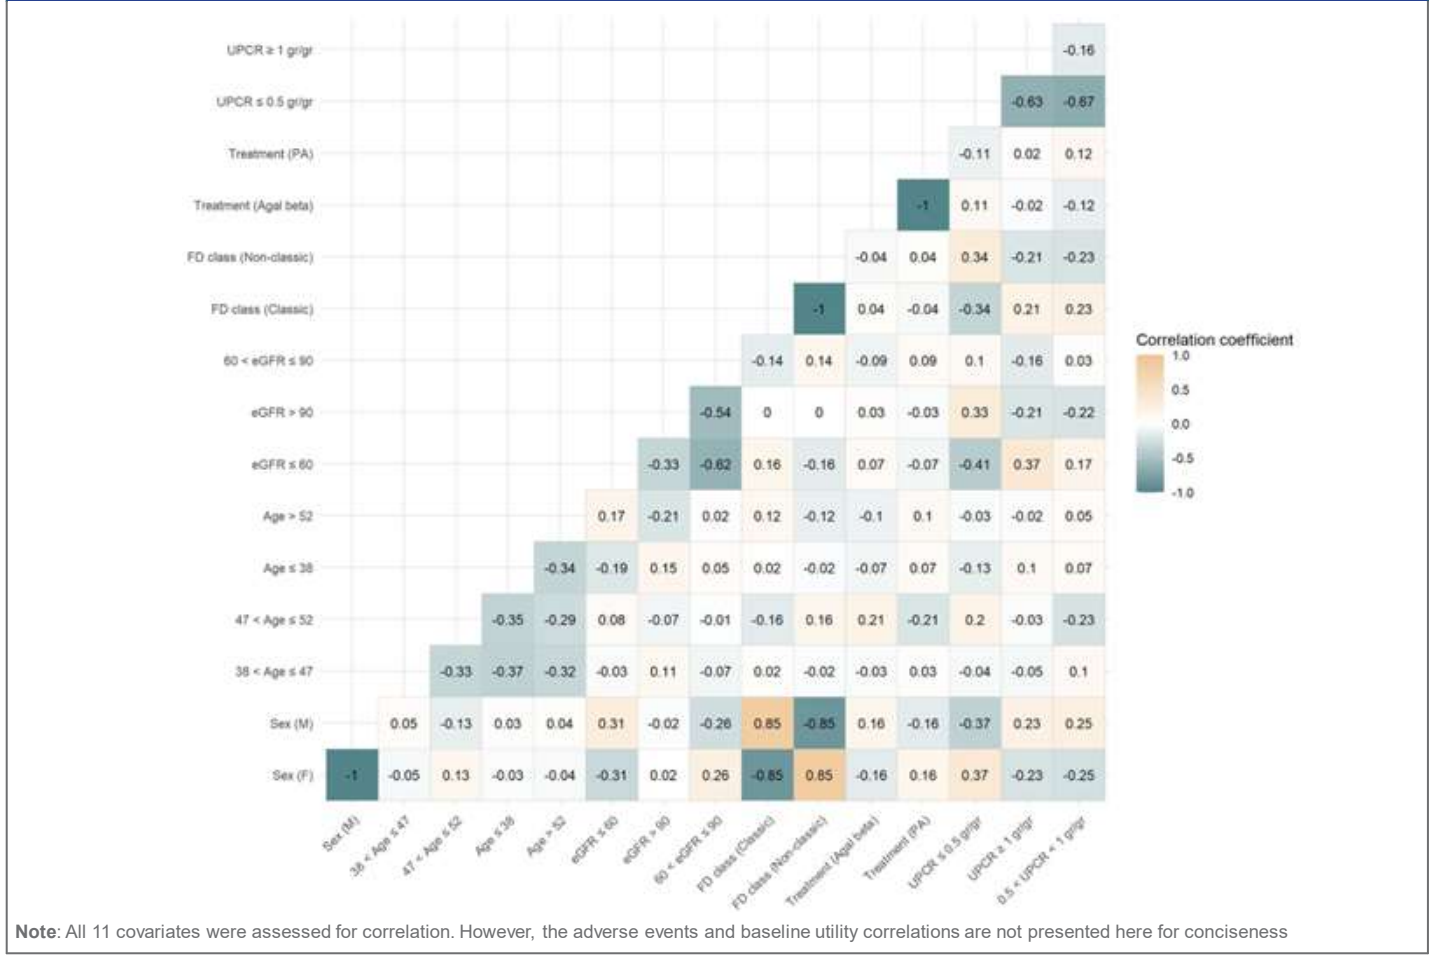
UPCR ≥ 1 gr/gr
UPCR ≤ 0.5 gr/gr
Treatment (PA)
Treatment (Agal beta)
FD class (Non-classic)
FD class (Classic)
60 < eGFR ≤ 90
eGFR > 90
eGFR ≤ 60
Age > 52
Age ≤ 38
47 < Age ≤ 52
38 < Age ≤ 47
Sex (M)
Sex (F)
Correlation coefficient
1.0
0.5
0.0
-0.5
-1.0

Note: All 11 covariates were assessed for correlation. However, the adverse events and baseline utility correlations are not presented here for conciseness

Model selection and results

The stepwise forward selection algorithm determined that the covariates baseline utility and FCEs should be included in the regression model. Both covariates were statistically significant (p-values were below 0.05 in Table 2.) Including these covariates also improved model fit (based on Akaike and Bayesian information criteria) versus models with additional covariates.

Table 2. Model results

Coefficient	Coefficient value	Standard error	p-value
Intercept	0.214	0.039	< 0.001
Baseline utility	0.740	0.049	< 0.001
FCE: yes (reference: no)	-0.093	0.033	0.005

In BALANCE, the mean baseline utility was 0.764. By utilizing this value in the regression model, a utility value for patients prior to experiencing an FCE was estimated at 0.779. The model predicted that a patient's utility would be lower after experiencing an FCE at 0.686. These results are summarized in Table 3.

Table 3. Utility estimates by health state

Health state	Utility value
FCE = No	0.779
FCE = Yes	0.686

Discussion

From the stepwise regression approach, a simple mixed-effects model with adjustment for baseline utility and FCE was selected. However, it should be noted that the definition of FCE used in the model is quite general; subcategories of FCEs (cardiac, cerebrovascular and renal adverse events) were grouped, and the severity of the events were not accounted for due to a low number of observations. It is possible that different subcategories/severity of FCEs along with other factors may influence a patient's utility value. However, FD is rare, and the low number of patients results in the analysis being underpowered to detect additional meaningful differences.

One point to note is that patients included in the BALANCE trial had advanced disease, including worsening kidney function. Therefore, it is possible that this utility model may underestimate utility for the wider FD population. However, this may be mitigated due to the baseline utility covariate, which allows the regression model to be applied to populations with a higher baseline utility estimate. The utility values presented here can be of use in cost-effectiveness analyses for FD and other lysosomal storage disorders.

Conclusions

The results demonstrate that renally impaired patients with FD who experience an FCE adverse event are expected to report a reduced HRQoL. Given the renally impaired population of BALANCE, estimates of HRQoL may be lower than for the general Fabry population.