CO123

An Indirect Comparison of Efgartigimod Versus Rituximab for Generalized Myasthenia Gravis

Lorenzo Celico[1], Erik Spaepen[1], Maria De Francesco[1], Sergio Iannazzo[2]

[1] HEOR Value Hub, Brussels, Belgium; [2] argenx, Geneva, Switzerland

Background

- gMG is a rare, chronic, neuromuscular autoimmune disease, mediated by pathogenic IgG autoantibodies[1].
- Two treatments for AChR-Ab+ gMG patients, efgartigimod and rituximab, have been separately compared with placebo in independent RCTs, not restricting time from generalized symptoms onset (Table 1).

Table 1 – Description of RCTs included in the analysis

	Efgartigimod	Rituximab
Drug type	Humanized IgG1 antibody Fc fragment vs IgG autoantibody	Anti-CD20 biologic drug
RCT	ADAPT[1]	BeatMG[2]
AChR-Ab+ gMG patients, n	129	52

- In BeatMG, all patients were treated with prednisone ≥15 mg/d at baseline.
- An understanding of the comparative efficacy of these two therapies would support decision-making in gMG treatment, yet no direct comparative evidence exists.
- We estimated the relative effect of efgartigimod vs rituximab through two indirect treatment comparisons anchored to the placebo arm and including only AChR-Ab+ patients.

Methods

Bucher's adjusted comparison

- The relative effect of efgartigimod vs placebo and the relative effect of rituximab vs placebo were compared.
- No further adjustment was made to correct for differences in treatment effect modifiers between the compared cohorts.
- This approach preserves the within-trial randomization effect, but assumes that treatment effect modifiers are balanced between the compared cohorts. This is unlikely to be true, so the Bucher's adjusted comparison results should be interpreted with caution.

Matching Adjusted Indirect Comparison (MAIC)

- Published aggregate data from BeatMG and individual patient data from ADAPT were used.
- The ADAPT population was restricted to align with the eligibility criteria for BeatMG, therefore 54 ADAPT participants were included in the analysis.
- ADAPT data were then weighted to match the baseline characteristics (only treatment effect modifiers) of BeatMG population (Table 2).
- This allows estimation of the relative effect of efgartigimod vs placebo as if efgartigimod was administered to the population used in the BeatMG study.

Table 2 – Variables used for weighting

Study	ADAPT		BeatMG	
Treatment	Efgartigimod	Placebo	Rituximab	Placebo
Number of patients at baseline	65	64	25	27
Years since diagnosis, mean (SE)	9.7 (8.3)	8.9 (8.2)	6.7 (6.5)	4.4 (5.3)
Use of prednisone as monotherapy at baseline, n (%)	7 (11)	10 (16)	17 (68)	17 (63)
Use of prednisone in combination with other NSIDs at baseline, n (%)	22 (34)	20 (31)	8 (32)	10 (37)
MG-ADL score at baseline, mean (SE)	9.0 (2.5)	8.6 (2.1)	5.8 (3.6)	4.0 (3.4)

Endpoint in the analysis

- MG-ADL change from baseline to the time of best response (week 4 for efgartigimod and week week 52 for rituximab) compared with placebo.

Statistical model

- In ADAPT, the difference in MG-ADL change from baseline between efgartigimod and placebo was calculated using a multivariate linear regression, with MG-ADL change from baseline as the dependent variable, and treatment arm, baseline MG-ADL and baseline use of NSID as covariates.
- For the MAIC, the model included the estimated weights as frequency weights.
- For the Bucher's comparison, the model was unweighted.

Results

MAIC reweighting

- The effective sample size was 1.3 (2.44% of the included sample size) so the results were based on a very small sample.
- The relative weights were heavily positive skewed. A few extreme outliers on the right side were driving the results of the analysis (Figure 1).

Figure 1 – Distribution of the relative weights estimated by means of the MAIC analysis

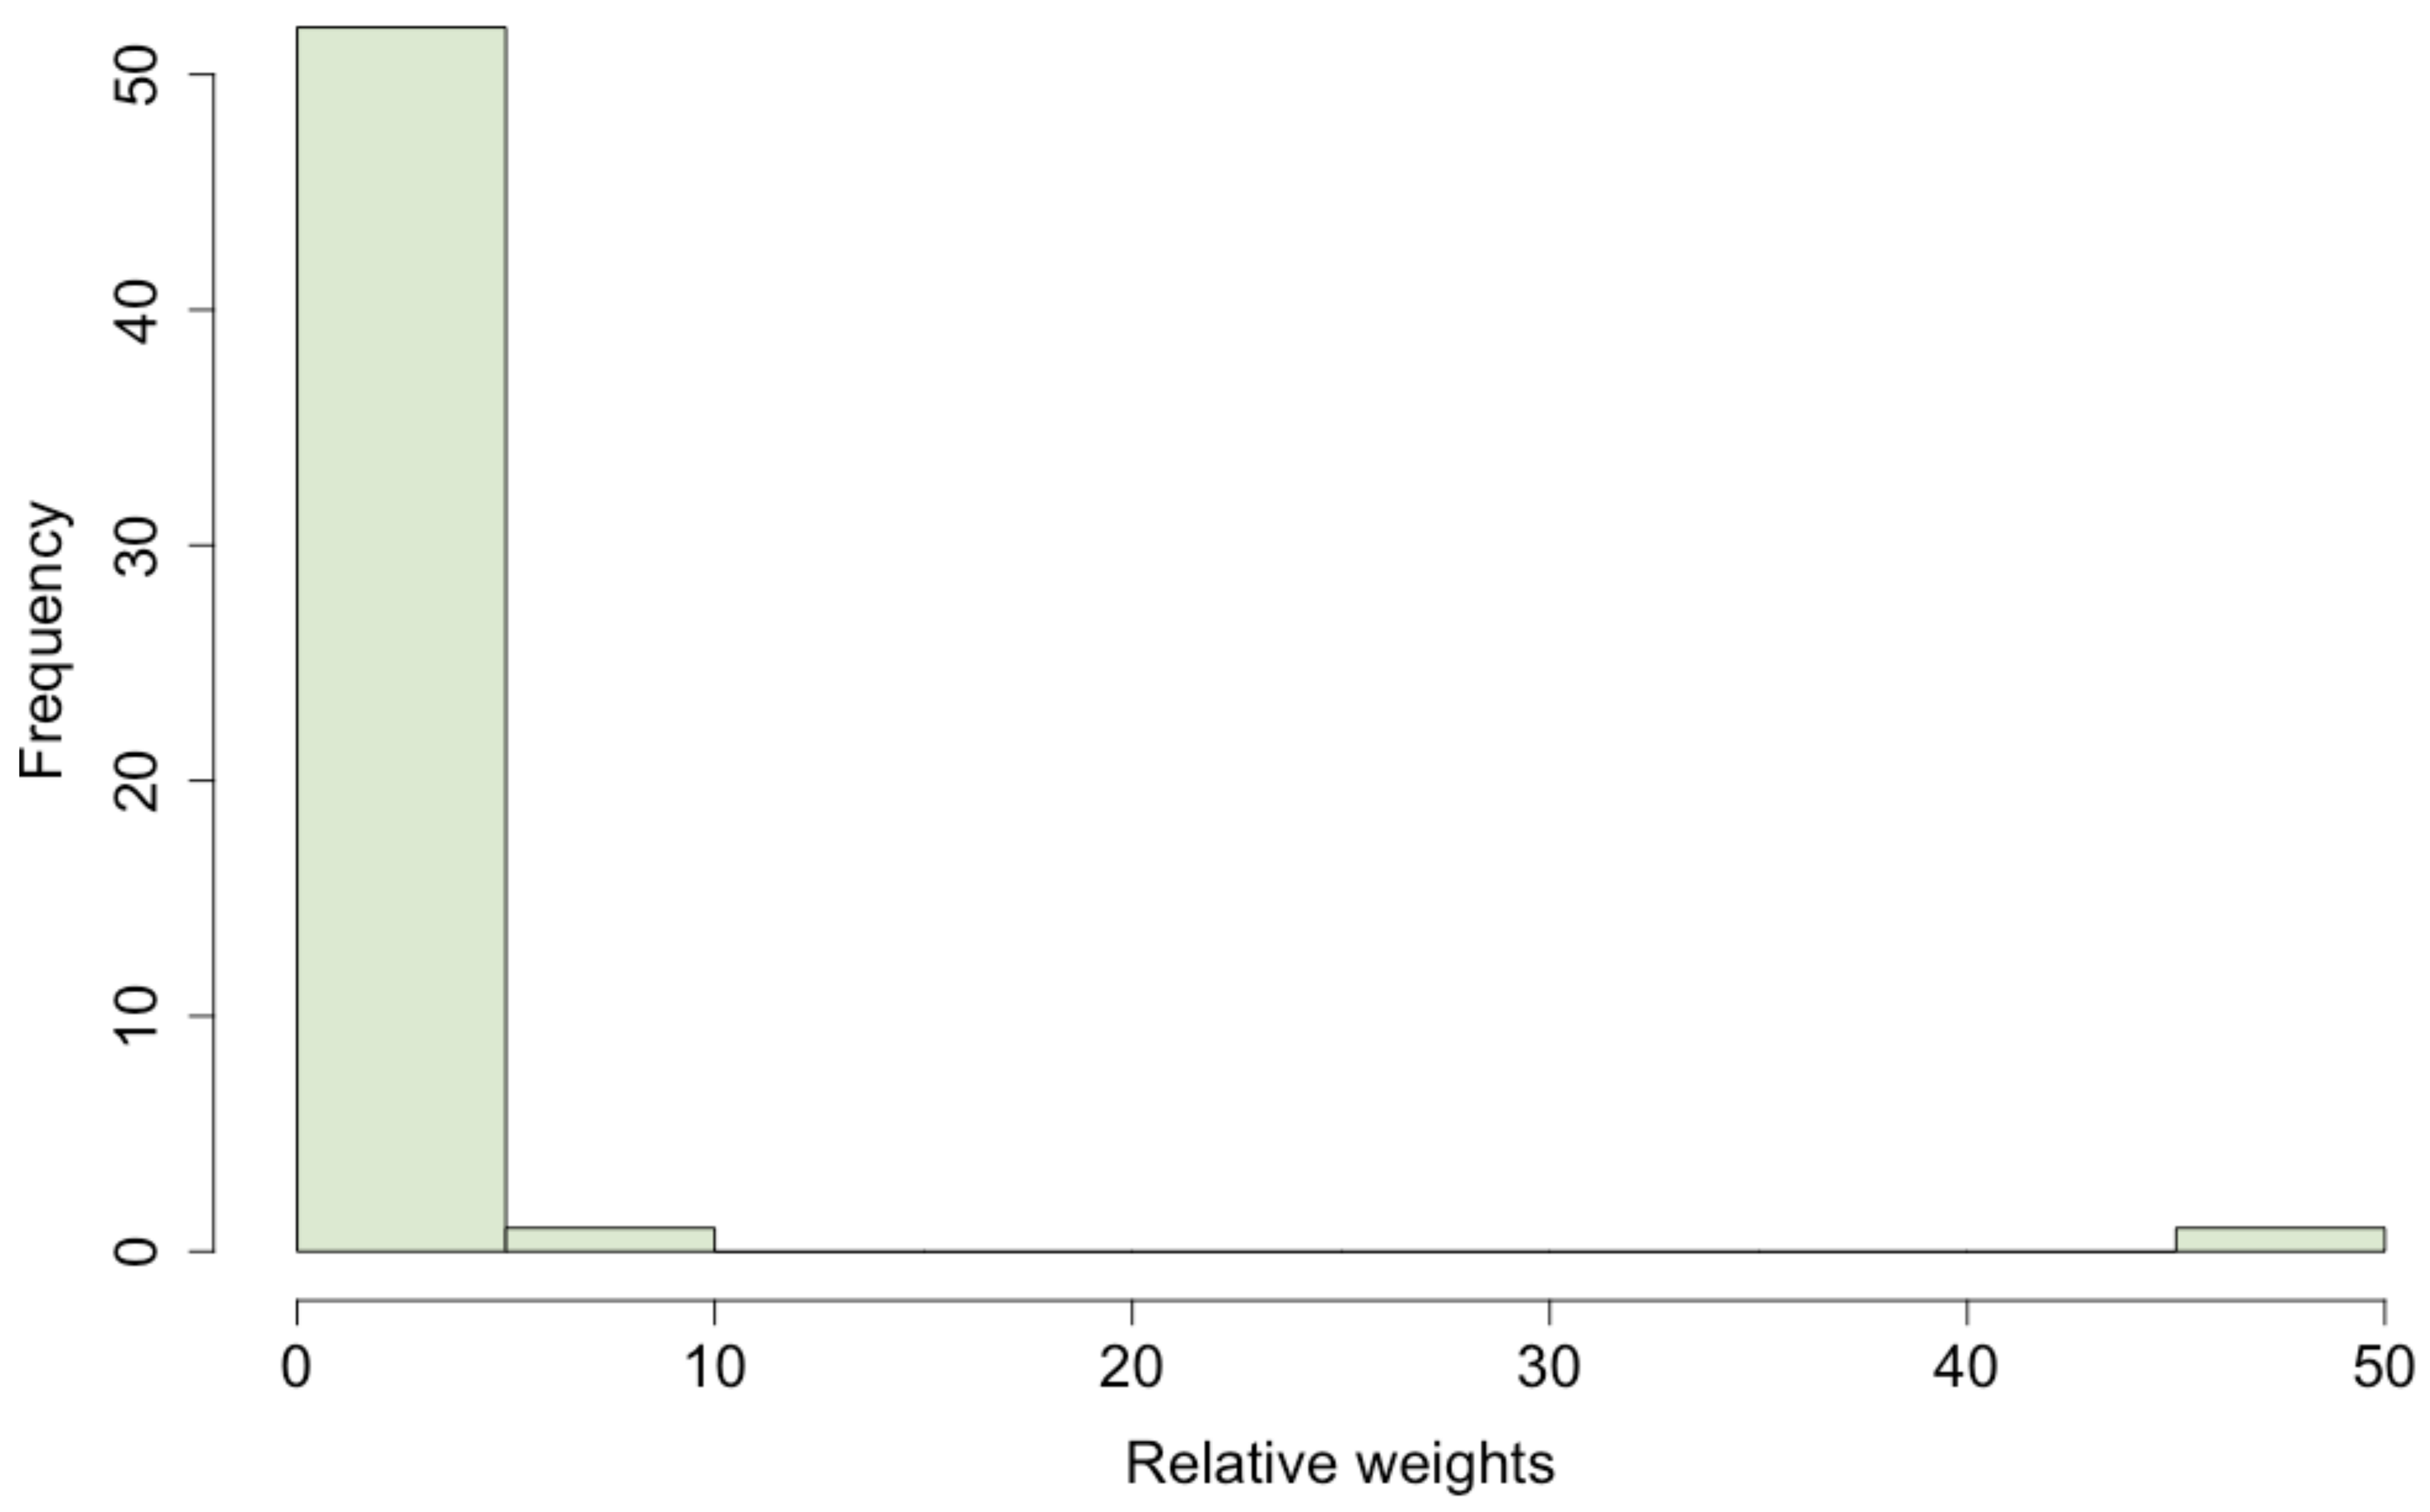
Frequency
0
10
20
30
40
50
Relative weights

Results of the comparison using Bucher approach

- At the time of best response, the reduction in MG-ADL from baseline vs placebo was, on average, 2.9 points greater (SE=0.8, 95% CI=[1.2, 4.6], p<0.001) for efgartigimod than rituximab.

Results of the comparison using the MAIC approach

- At the time of best response, the reduction in MG-ADL from baseline vs placebo was, on average, 3.2 points greater (SE=0.7, 95% CI=[1.8, 4.6], p<0.001) for efgartigimod than rituximab.

Figure 2 –MG-ADL change from baseline for efgartigimod vs rituximab (negative difference indicates greater MG-ADL reduction for efgartigimod than rituximab)

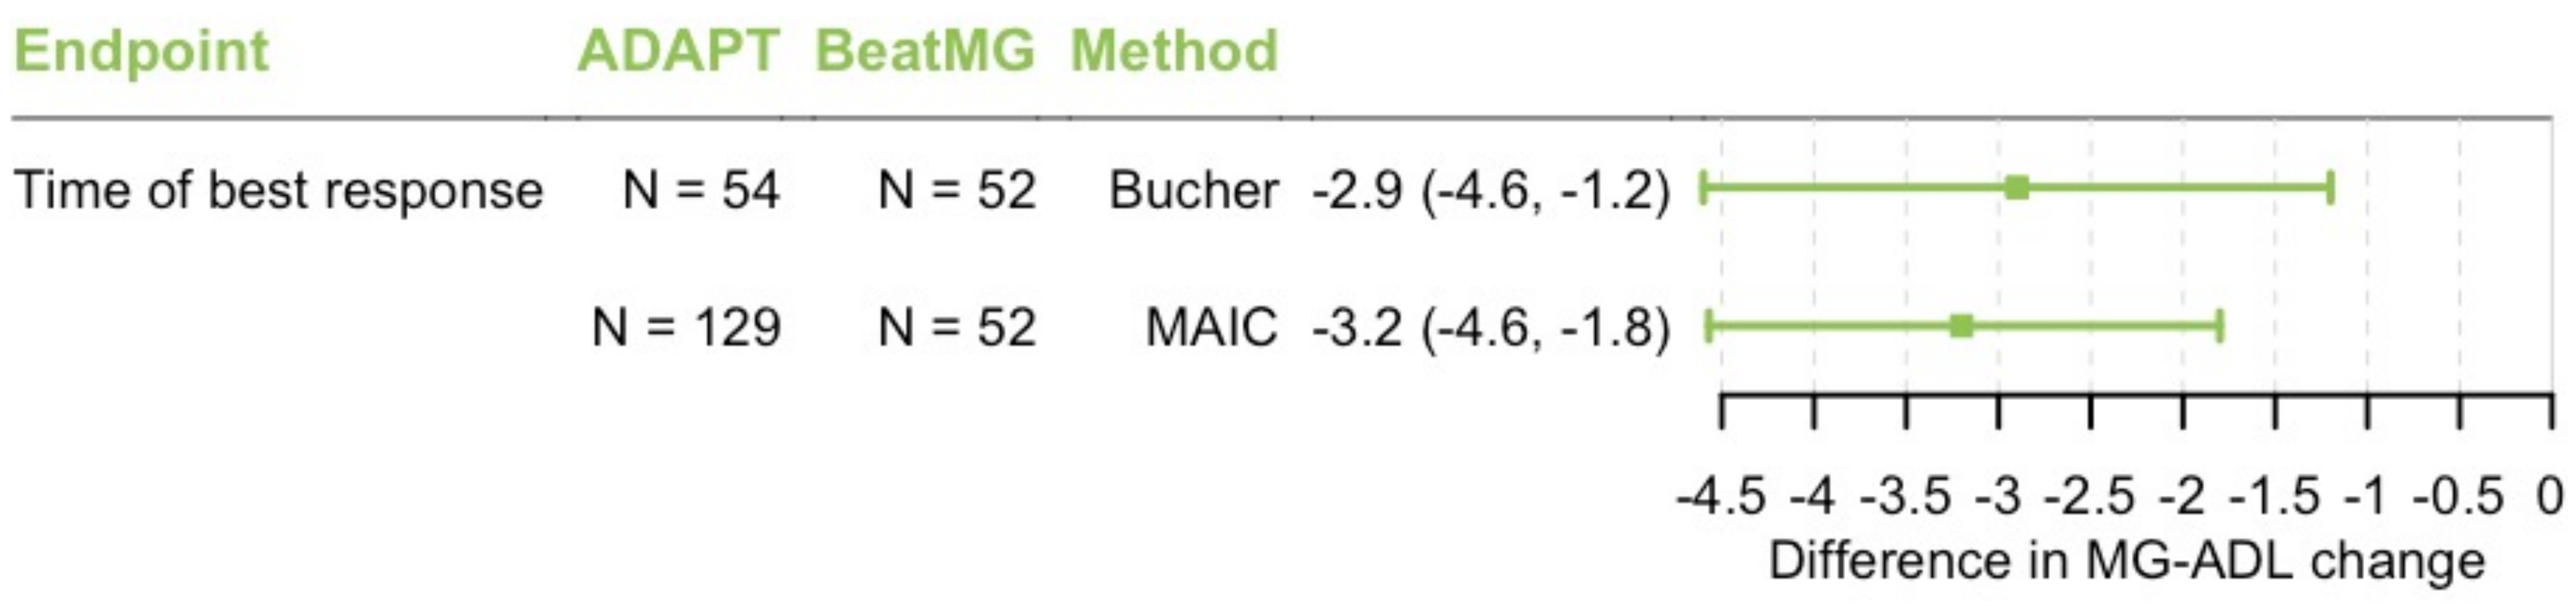
Endpoint
ADAPT
BeatMG
Method
Time of best response
N = 54
N = 52
Bucher
-2.9 (-4.6, -1.2)
N = 129
N = 52
MAIC
-3.2 (-4.6, -1.8)
-4.5 -4 -3.5 -3 -2.5 -2 -1.5 -1 -0.5 0
Difference in MG-ADL change

Conclusions

- We provide the first study comparing efgartigimod and rituximab in treating gMG.
- The included sample size for the MAIC consisted of 54 AChR-Ab+ patients, but the MAIC was based on an effective sample size of only 1.3 patients (2.44% of the original sample).
- Both MAIC and Bucher comparisons suggest greater efficacy of efgartigimod vs rituximab.
- Due to the limited effective sample size in the MAIC and the strong assumptions underlying the Bucher's adjusted comparison, these results should be interpreted with caution.
- Since this study was conducted, a new study (RINOMAX) was published. A comparison of RINOMAX and ADAPT cannot be performed because the population and comparator are different. RINOMAX showed that rituximab did not statistically significantly reduce MG-ADL reduction vs placebo, therefore confirming the evidence from the BeatMG used in the current indirect treatment comparison.

ABBREVIATIONS:

AChR-Ab+ = Acetylcholine Receptor Autoantibodies Positive
gMG = Generalized myasthenia gravis (gMG)
MAIC = Matching-Adjusted Indirect Comparison
MGFA = Myasthenia Gravis Foundation of America
MG-ADL = Myasthenia Gravis Activities of Daily Living
RCT = Randomized controlled trial

REFERENCES: 1. Howard J. F., et al. (2021); *The Lancet Neurology*, *20*(7), 526–536; **2.** Vu T., et al. (2022); NEJM Evidence, 1(5)

ACKNOWLEDGEMENTS: Formatting and editing assistance were provided by HEOR Value Hub funded by argenx. The authors would like to thank the patients enrolled in the study, as well as all patients with gMG and the investigators integral to treating these patients.

Presented at ISPOR Europe 2022; 6-9 November 2022; Vienna, Austria.