POSC24

Surrogate endpoints for overall survival (OS) in advanced melanoma: A targeted literature review (TLR) of correlation meta-analyses

Pourrahmat M, BSc[1]; Leung L, MSc[1]; Kurt M, PhD[2]; Chou E, MSc[2]; Kanters S, PhD[1]; Moshyk A, MSc[2]; Hamilton M, MPH[2]

[1]Evidinno Outcomes Research Inc., Vancouver, BC, Canada; [2]Bristol Myers Squibb, Princeton, NJ, USA

Introduction

- Melanoma is the fifth most common cancer in the United States (US). In 2021, an estimated 106,110 new cases of melanoma will have been diagnosed in the US, and 7,180 American patients with melanoma are estimated to die.[1]
- There has been a growing interest in recent years to understand the landscape of evidence surrounding the relationship between OS, the gold standard endpoint for oncology trials, and other endpoints.
- If surrogate endpoints (SEs) are consistently reported to be valid and useful, then they can be used to estimate the OS benefit of new interventions in present and future trials, which can potentially reduce the duration of patient follow-up times.
- Prior meta-analyses have assessed the surrogacy between OS and alternative time-to-event outcomes in advanced or metastatic melanoma.
- To understand and synthesize different methodologies used by these studies, we aimed to conduct a targeted literature review (TLR) of published analyses that investigated the validity of SEs for OS in the context of advanced melanoma within any treatment setting.

Objective

- To review existing correlation analyses evaluating candidate SEs for OS in advanced melanoma within any treatment setting.

Methods

- A TLR was conducted using methodology adapted from the Cochrane Handbook for Systematic Reviews of Interventions.[2]
- Embase and MEDLINE® were searched for relevant entries up to October 19, 2020 (with no restriction on the start date), using a mix of Medical Subject Headings (MeSH) and free-text key words.
- A grey literature search using Embase was also conducted for abstracts between 2018-2020 from the conferences below:
 - International Society for Pharmacoeconomics and Outcomes Research (ISPOR)
 - ISPOR EU
 - American Society of Clinical Oncology (ASCO)
 - European Society for Medical Oncology (ESMO)
- Included studies had to be systematic literature reviews and meta-analyses evaluating SEs of phase II/phase III melanoma randomized control trials.
- The outcomes of interest were correlation measures between OS and potential SEs, including but not limited to progression-free survival (PFS), disease-free survival (DFS), and objective response rate (ORR).

Figure 1. PRISMA diagram

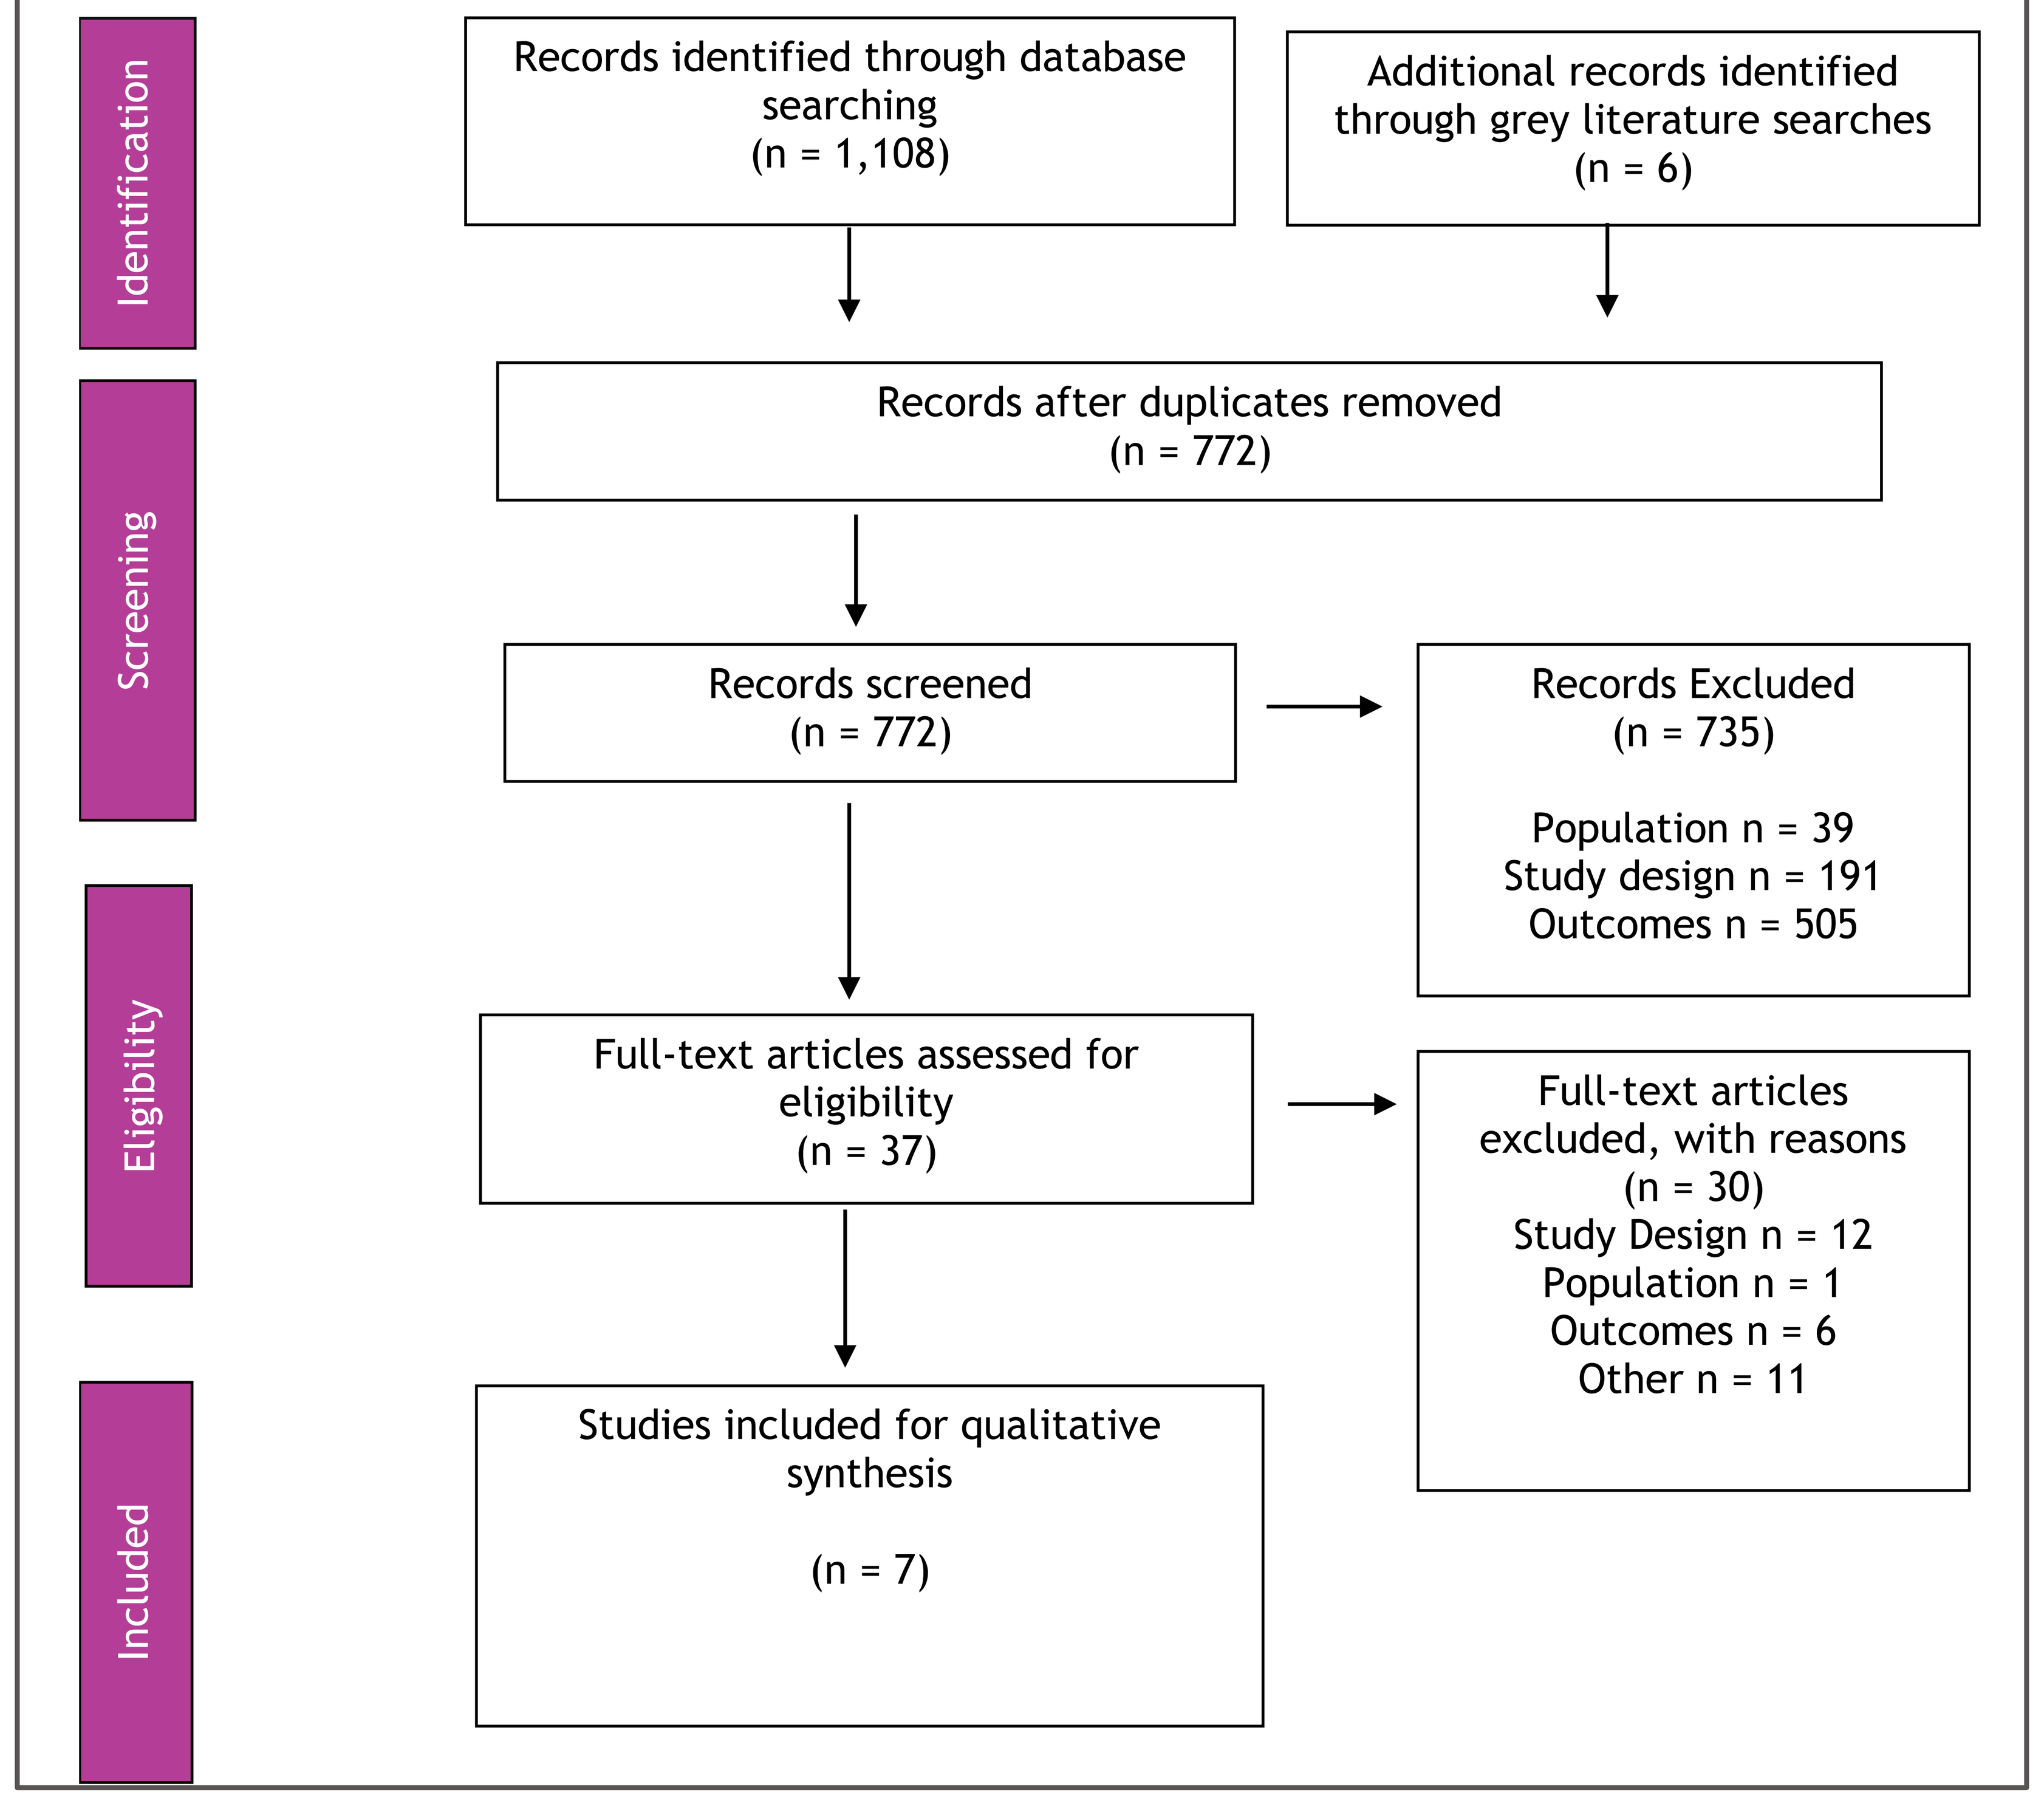
Identification
Screening
Eligibility
Included
Records identified through database searching
(n = 1,108)
Additional records identified through grey literature searches
(n = 6)
Records after duplicates removed
(n = 772)
Records screened
(n = 772)
Records Excluded
(n = 735)
Population n = 39
Study design n = 191
Outcomes n = 505
Full-text articles assessed for eligibility
(n = 37)
Full-text articles excluded, with reasons
(n = 30)
Study Design n = 12
Population n = 1
Outcomes n = 6
Other n = 11
Studies included for qualitative synthesis
(n = 7)

Results

- Out of 1,114 records that were identified by the main database search and grey literature searches that underwent title abstract screening, 37 citations were included for full-text screenings, of which 7 publications were eligible to be included in qualitative evidence synthesis (**Figure 1**).
- Summary characteristics of the included studies are presented in **Table 1**.
- All publications studied PFS as an SE. Five studies conducted trial-level surrogacy analysis only, while two also conducted arm-level (i.e., summarized endpoint estimates by treatment arms) surrogacy analysis. Only two studies reported validation analysis (see **Supplementary Table** attached in the Handout).
- Brief summaries of each included study are provided below, in order of publication date.

Flaherty et al 2014[3]

- Flaherty et al conducted a meta-analysis of 12 dacarbazine-controlled randomized trials (RCT) with metastatic melanoma patients to assess hazard ratio (HR) of PFS (HR_{PFS}) as a potential surrogate for HR of OS (HR_{OS}). From each trial, the authors extracted sample size and HRs along with their 95% confidence intervals (CIs) for PFS and OS. Statistical analysis was based on trial-level data.
- The authors suggest that HR_{PFS} can be used as a robust SE for HR_{OS} in dacarbazine-controlled RCTs of metastatic melanoma.

Kaufman et al 2017 (conference abstract)[4]

- The authors evaluated odds ratio (OR) of objective responsive rate (OR_{ORR}), OR of disease control rate (OR_{DCR}), and HR_{PFS} as surrogates for HR_{OS} in patients receiving immunotherapies from 18 RCTs. Statistical analysis was based on trial-level data.
- Eligible studies were RCTs that studied immune checkpoint blockers targeting programmed death proteins (PD-1/PD-L1) and cytotoxic T-lymphocyte-associated protein 4 (CTLA-4). The included studies reported their relative effect on OS and on at least one of the clinical endpoints mentioned in the first bullet.
- The authors did not observe a clear correlation between the relative treatment effects of conventional clinical endpoints and OS for checkpoint blockers.

Mushti et al 2018[5]

- The authors evaluated RECIST criteria-based OR of ORR (OR_{ORR}), and HR_{PFS} as SEs for HR_{OS}.
- Data from 13 multicenter, active-controlled immunotherapy trials of anti-PD-1/PD-L1 agents were used. A correlation meta-analysis on PFS for OS in melanoma patients was conducted at the trial level. Authors did not report any surrogacy analyses for ORR to OS specifically for the melanoma cohort, hence this information is not available.
- The authors concluded that PFS is not a valid surrogate for OS; however, they noted that the number of studies in the evidence base and the respective sample sizes of the studies were too small to draw definitive conclusions.

Nie et al 2019[6]

- The authors assessed RECIST criteria-based ORR, DCR, and median PFS as SEs for median OS. They also assessed OR_{ORR}, OR_{DCR}, and HR_{PFS} as SEs for HR_{OS}. The authors included 43 phase II and phase III trials of anti-PD-1/PD-L1 drugs for advanced or recurrent solid tumors including but not limited to melanoma, small cell lung cancer, and renal cell carcinoma. While the authors used individual level patient data from the clinical trials, only a trial-level correlation meta-analysis on PFS and OS was reported for melanoma patients.
- Surrogacy relationship between ORR and OS, and DCR and OS, as well as a series of sensitivity analyses were performed. However, none of them were reported separately for the melanoma cohort.
- The authors concluded that none of the candidate endpoints or durations could be a valid surrogate for OS due to the lack of strong correlation between the HR estimates on trial-level.
- The positive association observed in the melanoma subgroup may have led the authors to conduct further analyses focused on this population – described below in Nie et al 2020.[7]

Nie et al 2020[7]

- The authors assessed surrogacy of RECIST criteria-based OR_{ORR}, OR_{DCR}, and HR_{PFS} for HR_{OS} in trials of melanoma. They included 8 phase II/III trials of unresectable, advanced, or recurrent melanoma that investigated PD-1/PD-L1 inhibitors in the experimental arm and any therapy in the control arm. Statistical analysis was done at the trial level.
- The authors defined that the correlation between OS and SEs would be considered strong if R^2 exceeded 0.75 without providing a reference or guidance for this classification.
- The authors proposed using HR_{PFS} as an SE for HR_{OS} in anti-PD-1/PD-L1 trials for advanced melanoma after detecting a strong correlation between the treatment effects on PFS and OS. Unlike HR_{PFS}, the authors did not observe strong correlations between OR_{DCR}/OR_{ORR} and HR_{OS}.

Petrelli et al 2016[8]

- The authors aimed to evaluate median PFS, and 1- and 2-year OS rates as SEs for median OS via a meta-analysis of 13 trials evaluating immune checkpoint inhibitors (ICIs) in any line of therapy for metastatic melanoma. Statistical analysis was based on arm-level data and trial-level data.
- However, trial-level PFS surrogacy was not performed due to only four eligible RCTs reporting HR_{PFS} and HR_{OS}.
- The authors concluded that 1-year OS rate could be regarded as a potential surrogate for median OS in novel immunotherapy trials of metastatic melanoma.

Hopkinson et al 2018 (conference abstract)[9]

- The authors evaluated alternative endpoints to median OS in advanced melanoma using data from 25 trials evaluating immunotherapies or targeted therapies. Statistical analysis was done at the arm-level and the trial-level.
- The authors mentioned they relied on guidelines provided from HTA agencies, including IQWiG, and concluded that median PFS, 1-year OS rate, and 2-year OS rate are statistically viable alternative endpoints.

Conclusions

- Multiple studies have investigated the validity of PFS and other outcome measures as surrogates for OS in melanoma studies.
- While there is general agreement on methods, it is worth pointing out that there are some differences between the analyses — for example, Flaherty et al 2014 included only one immunotherapy trial while all analyses in all other studies were based almost exclusively on immunotherapy trials.
- Our study highlights the variability among the results of the existing analyses and limited consensus for surrogacy between PFS and OS.
- Improved correlation estimates between PFS and OS were seen with increased study sample size, restricting the evidence base to only phase III studies, or to studies with minimal or no crossover.
- PFS may be a promising SE for OS in advanced melanoma, but further research is needed to confirm this.
- This research area continues to grow with new publications (Sheth et al 2021[10] and Branchoux et al 2020[11]) which were published after the completion date of our search.
 - Sheth et al 2021 concluded that ORR may not be an appropriate surrogate for OS or PFS. The unclear relationship between the endpoints could be due to restricting the trials to only those submitted to the FDA.
 - Branchoux et al 2020 used data from the CheckMate-067 trial and concluded that time to next treatment (TNT) may be a valuable SE in previously untreated advanced melanoma patients treated with ICIs. As the research area continues to develop, new promising SEs such as TNT can be expected in the future.

Table 1. Characteristics of the included studies

Study	Surrogate Endpoints*	True Endpoint	Endpoint Definition	Association Measures	# of Included RCTs	# of Included Patients	Time of Search†	Patient Population
Studies with only Trial-level Associations								
Flaherty et al 2014	HR_{PFS}	HR_{OS}	As defined and reported by each trial.	R	12	4,416	September 2013	Trials investigating dacarbazine as the control arm in advanced melanoma patients
Kaufman et al 2017 (conference abstract)	HR_{PFS}, OR_{DCR}, OR_{ORR}	HR_{OS}	NR	Adjusted R^2	18	7,140	January 2005-November 2016 (PubMed, Embase); 2014-2016 (conference proceedings)	Trials of immune checkpoint blockers targeting programmed death proteins (PD-1/PD-L1) and cytotoxic T-lymphocyte-associated protein 4 (CTLA-4)
Mushti et al 2018	HR_{PFS}, OR_{ORR}	HR_{OS}	PFS: the time since randomization to progression or death, whichever occurred first. ORR: the proportion of confirmed complete response (CR) or partial response (PR) at the point of best overall response.	R^2	13	6,722	2014-2016	Trials of anti-PD-1/PD-L1 agents in melanoma, NSCLC, renal cell carcinoma and head and neck cancer
Nie et al 2019	HR_{PFS}	HR_{OS}	PFS: the time from randomization to the first event (progressive disease or death from any cause). OS: the time from randomization to death from any cause.	R^2	43	15,088	June 2018	Phase II/III trials of anti-programmed death-1 (PD-1)/programmed death ligand 1 (PD-L1) in advanced or recurrent solid tumors
Nie et al 2020	HR_{PFS}, OR_{DCR}, OR_{ORR}	HR_{OS}	PFS: the time from randomization to progressive disease or death from any cause. DCR: the percentage of confirmed CR, PR, or stable disease at the point of best overall response. ORR: the proportion of confirmed CR or PR at the point of best overall response. OS: the time from randomization to death from any cause.	R^2	8	4,110	June 2019	Phase II/III trials of unresectable, advanced, or recurrent melanoma
Studies with both Arm-level Associations/Outcome Surrogacy and Trial-level Associations								
Petrelli et al 2016	Median PFS, 1-year OS rate, 2-year OS rate	Median OS	NR	R; R^2	13	3,373	July 3, 2015	Phase II/III trials of immunotherapy as the treatment for advanced melanoma
	Delta in 1-year OS rate, delta in 2-year OS rate	HR_{OS}						
Hopkinson et al 2018 (conference abstract)	Median PFS, 1-year OS rate, 2- year OS rate	Median OS	NR	R	25	NR	NR	Trials of immunotherapies or targeted therapies in advanced melanoma
	HR_{PFS}, Delta in 1-year OS rate, Delta in 2-year OS rate	HR_{OS}						

DCR – Disease control rate, delta – difference in 1 and 2-year OS rate between the experimental and the control arms, HR – Hazard ratio, NR – Not reported, NSCLC – non-small cell lung cancer, OR – Odds ratio, ORR – Objective response rate, OS – Overall survival, PFS – Progression-free survival, R – Pearson's correlation coefficient, R^2 – Coefficient of determination

References

1. Siegel RL, Miller KD, Fuchs HE, Jemal A. Cancer Statistics, 2021. CA: A Cancer Journal for Clinicians. 2021;71(1):7-33.
2. Higgins JP, Green S. *Cochrane handbook for systematic reviews of interventions*. Vol 4: John Wiley & Sons; 2011.
3. Flaherty KT, Hennig M, Lee SJ, et al. Surrogate endpoints for overall survival in metastatic melanoma: a meta-analysis of randomised controlled trials. *The Lancet Oncology*. 2014;15(3):297-304.
4. Kaufman H, Schwartz LH, William WN, et al. Evaluation of clinical endpoints as surrogates for overall survival in patients treated with immunotherapies. *Journal of Clinical Oncology Conference*. 2017;35(15 Supplement 1).
5. Mushti SL, Mulkey F, Sridhara R. Evaluation of overall response rate and progression-free survival as potential surrogate endpoints for overall survival in immunotherapy trials. *Clinical Cancer Research*. 2018;24(10):2268-2275.
6. Nie RC, Chen FP, Yuan SQ, et al. Evaluation of objective response, disease control and progression-free survival as surrogate end-points for overall survival in anti-programmed death-1 and anti-programmed death ligand 1 trials. *European Journal of Cancer*. 2019;106:1-11.
7. Nie RC, Yuan SQ, Wang Y, et al. Surrogate endpoints for overall survival in anti-programmed death-1 and anti-programmed death ligand 1 trials of advanced melanoma. *Therapeutic Advances in Medical Oncology*. 2020;12:1758835920929583.
8. Petrelli F, Coinu A, Cabiddu M, et al. Early analysis of surrogate endpoints for metastatic melanoma in immune checkpoint inhibitor trials. *Medicine (United States)*. 2016;95 (26) (no pagination)(e3997).
9. Hopkinson D, Jones C, Chadwick C. Validation of Alternative "Surrogate" Clinical Endpoints in Advanced Melanoma. *Value in Health*. 2018;21 (Supplement 3):S356.
10. Sheth M, Ko J. Exploring the relationship between Overall Survival (OS), Progression Free Survival (PFS) and Objective Response Rate (ORR) in patients with advanced melanoma. *Cancer Treatment and Research Communications*. 2021;26:100272.
11. Branchoux S, Sofeu CL, Gaudin AF, et al. PCN29 Time to Next Treatment As a Candidate Surrogate Endpoint for Overall Survival in Previously Untreated Advanced Melanoma Patients Treated with Immune-Checkpoint Inhibitors. *Value in Health*. 2020;23:S426.

Acknowledgments

- This study was funded by Bristol Myers Squibb (Princeton, NJ, USA) and conducted by Evidinno Outcomes Research Inc. (Vancouver, BC, Canada).
- Mir-Masoud Pourrahmat and Lisa Leung report employment, and Steve Kanters reports contract, with Evidinno Outcomes Research Inc. Murat Kurt, Engels Chou, Andriy Moshyk, and Melissa Hamilton report employment with Bristol Myers Squibb. Authors report no other conflicts of interest.
- All authors contributed to and approved the presentation; poster development was supported by Andi Qian (Evidinno Outcomes Research Inc.)

Presented at Virtual ISPOR Europe 2021, November 30–December 3

Email: mpourrahmat@evidinno.com

Copies of this poster are for personal use only and may not be reproduced without written permission from the author of this poster.