Cost-effectiveness analysis of avelumab + best supportive care (BSC) vs BSC alone as a first-line (1L) maintenance treatment for patients with locally advanced or metastatic urothelial carcinoma in Taiwan

W.-C. Chang,[1] Y. Xiao,[2] A. Lin,[3]* P.-J. Su,[1] C. Goh,[3] E. Wu,[4] K. Liu,[4] P. Chou,[5] K. Kuo,[5] R. Palencia,[6] J. Chang,[7] M. Kearney,[6] V. Kapetanakis,[2] A. Benedict[8]

[1]Chang Gung Memorial Hospital, Linkou, Taiwan; [2]Evidera, London, UK; [3]Merck Ltd., Taipei, Taiwan, an affiliate of Merck KGaA, Darmstadt, Germany; [4]Pfizer, Taipei, Taiwan; [5]IQVIA Solutions Taiwan Ltd., Taipei, Taiwan; [6]the healthcare business of Merck KGaA, Darmstadt, Germany; [7]Pfizer, New York, NY, USA; [8]Evidera, Budapest, Hungary

*Affiliation at the time the analysis was conducted.

SCOPE

- **This analysis aimed to adapt a global cost-effectiveness model in Taiwan and present the results of the adaptation in terms of the incremental cost-effectiveness ratio (ICER), life-years (LYs), and quality-adjusted life-years (QALYs) of a partitioned survival economic model for avelumab + BSC vs BSC alone over a 20-year time horizon**

CONCLUSIONS

- **JAVELIN Bladder 100 demonstrated avelumab + BSC as an effective 1L maintenance treatment after completion of 1L platinum-containing chemotherapy**
- **The ICER fell below the threshold of 2 times the gross domestic product (GDP) per capita and demonstrated the cost-effectiveness of avelumab + BSC vs BSC alone in Taiwan**
 - **This was supported by deterministic and probabilistic sensitivity analyses.**
- **This analysis may be used by national payers when considering reimbursement for avelumab for the treatment of patients with locally advanced/metastatic urothelial carcinoma (LA/mUC) who did not show disease progression with 1L chemotherapy in Taiwan**

GET POSTER PDF

Copies of this poster obtained through this **hyperlink** or quick response (QR) code are for personal use only and may not be reproduced without permission from ISPOR Europe and the author of this poster.

Correspondence: Patrick Chou, **patrick.chou@iqvia.com**

INTRODUCTION

- UC accounts for >90% of bladder cancers in Taiwan[1]
- Current standard of care (SOC) in 1L, platinum-containing chemotherapy, is associated with limited duration and impacts quality of life[2]
- JAVELIN Bladder 100 is the only international, multicenter, phase 3 trial in over 20 years to demonstrate a significant increase in overall survival (OS) in patients with LA/mUC with 1L treatment[3]
- In patients with LA/mUC without disease progression after 4-6 cycles of 1L platinum-containing chemotherapy, avelumab + BSC increased the 1-year survival rate by 12.9% (avelumab + BSC, 71.3% [95% CI, 66.0%-76.0%] vs BSC, 58.4% [95% CI, 52.7%-63.7%])[3]
 - Median OS was significantly increased by 7.1 months (avelumab + BSC, 21.4 months [95% CI, 18.9-26.1 months] vs BSC, 14.3 months [95% CI, 12.9-17.9 months]; hazard ratio, 0.69 [95% CI, 0.56-0.86]; p=0.001)[3]
 - Median progression-free survival (PFS) was significantly increased by 1.7 months (avelumab + BSC, 3.7 months [95% CI, 3.5-5.5 months] vs BSC, 2.0 months [95% CI, 1.9-2.7 months]; hazard ratio, 0.62 [95% CI, 0.52-0.75])[3]

METHODS

- A global partition survival cost-effectiveness model after 1L platinum-containing chemotherapy was adapted to Taiwan from the National Health Insurance Administration (NHIA) perspective. The 3 primary health states are PFS, post-progression survival, and death (**Figure 1**). A patient in the model was considered to be in 1 of these 3 health states at any time
- Patient-level data on efficacy, safety, utility values, and treatment exposure, including subsequent therapies, were obtained from JAVELIN Bladder 100 to provide parameters for the model[3]
- Results are presented as total costs, LYs, and QALYs gained. For utilities, a mixed-effects model was generated, and the base-case utility model with 3 covariates (baseline utility, progression status, and proximity to death status) was fitted to the data
- For OS, PFS, and time to treatment discontinuation, log-normal, Weibull, and exponential distributions were used, respectively
- The costs of drug acquisition and adverse events were identified from the NHIA medication online website[4] and the NHIA annual medication report.[5] The costs of monitoring and healthcare resource use (HCRU) were identified from Taiwan-specific sources such as the NHIA Medical Service Online[6]
- Estimates of HCRU frequency were initially obtained via literature review and validated by Taiwan oncology experts (**Table 1**)
- A 20-year time horizon and 1-week cycle length were used, and a 3% discount rate was applied to both costs and outcomes
- A willingness-to-pay threshold of 3 times the per capita GDP was adopted according to the World Health Organization definition
- Sensitivity analyses were performed to characterize uncertainties in the expected outcomes

RESULTS

- In the modeled base case, avelumab + BSC increased survival vs BSC alone by 0.79 LYs (2.93 vs 2.14) and 0.61 QALYs (2.15 vs 1.54) (**Table 2**)
- The ICER for avelumab + BSC vs BSC alone fell below the threshold of 2 times the gross domestic product
- Results of the scenario analysis indicated that LY and QALY gains were most sensitive to alternative survival extrapolations for both avelumab + BSC and BSC alone
- The exponential distribution was chosen as the best-fitting distribution for time-to-treatment discontinuation (TTD) for avelumab + BSC based on median treatment duration observed in JAVELIN Bladder 100 (**Figure 2**)
- One-way sensitivity analyses showed that the ICER decreased when the following parameters increased:
 - Time on subsequent immune checkpoint inhibitors (ICIs) following progression with BSC and percentage of patients receiving subsequent BSC
- However, the ICER increased when the following parameter increased:
 - Median duration of treatment with avelumab (**Figure 3**)
- The scatterplot with 1,000 repetitions showed the uncertainty surrounding the estimates of expected incremental cost and expected incremental effect (QALYs gained) when comparing avelumab + BSC vs BSC alone (**Figure 4A**)
- 72% and 92% of the probabilistic sensitivity analyses fell within 2 times and 3 times the acceptable cost-effectiveness threshold, respectively[7] (**Figure 4B**)

Figure 1. Partitioned survival model structure

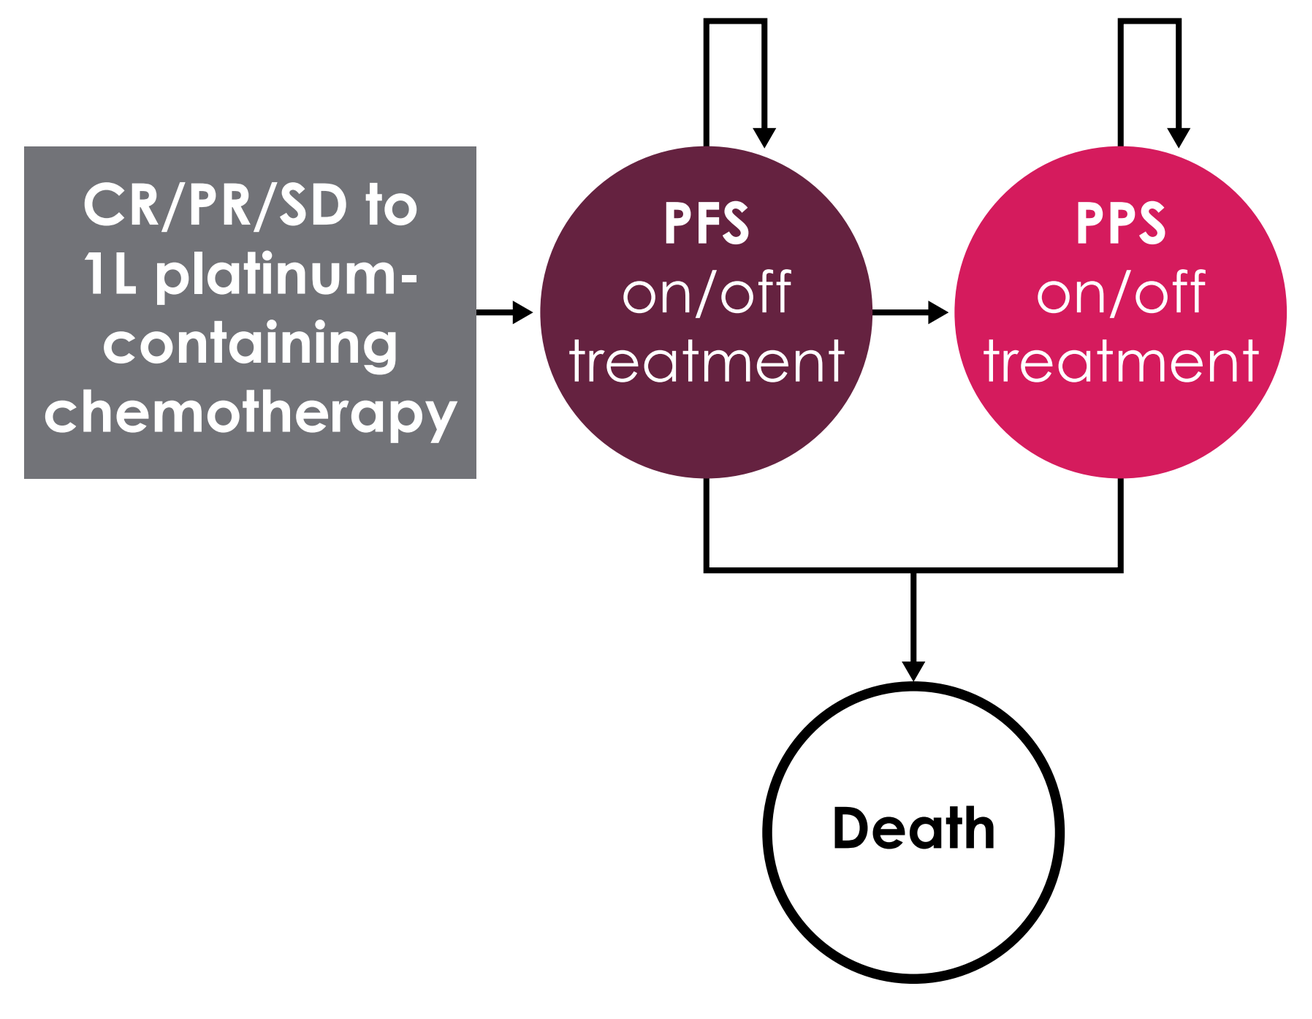
CR/PR/SD to 1L platinum-containing chemotherapy
PFS on/off treatment
PPS on/off treatment
Death

Notes: Arrows indicate transitions. Arrows that curve back to the same state indicates remaining in the same state.

CR, complete response; **PFS**, progression-free survival; **PPS**, post-progression survival; **PR**, partial response; **SD**, stable disease.

Figure 2. TTD projection for avelumab + BSC vs BSC alone

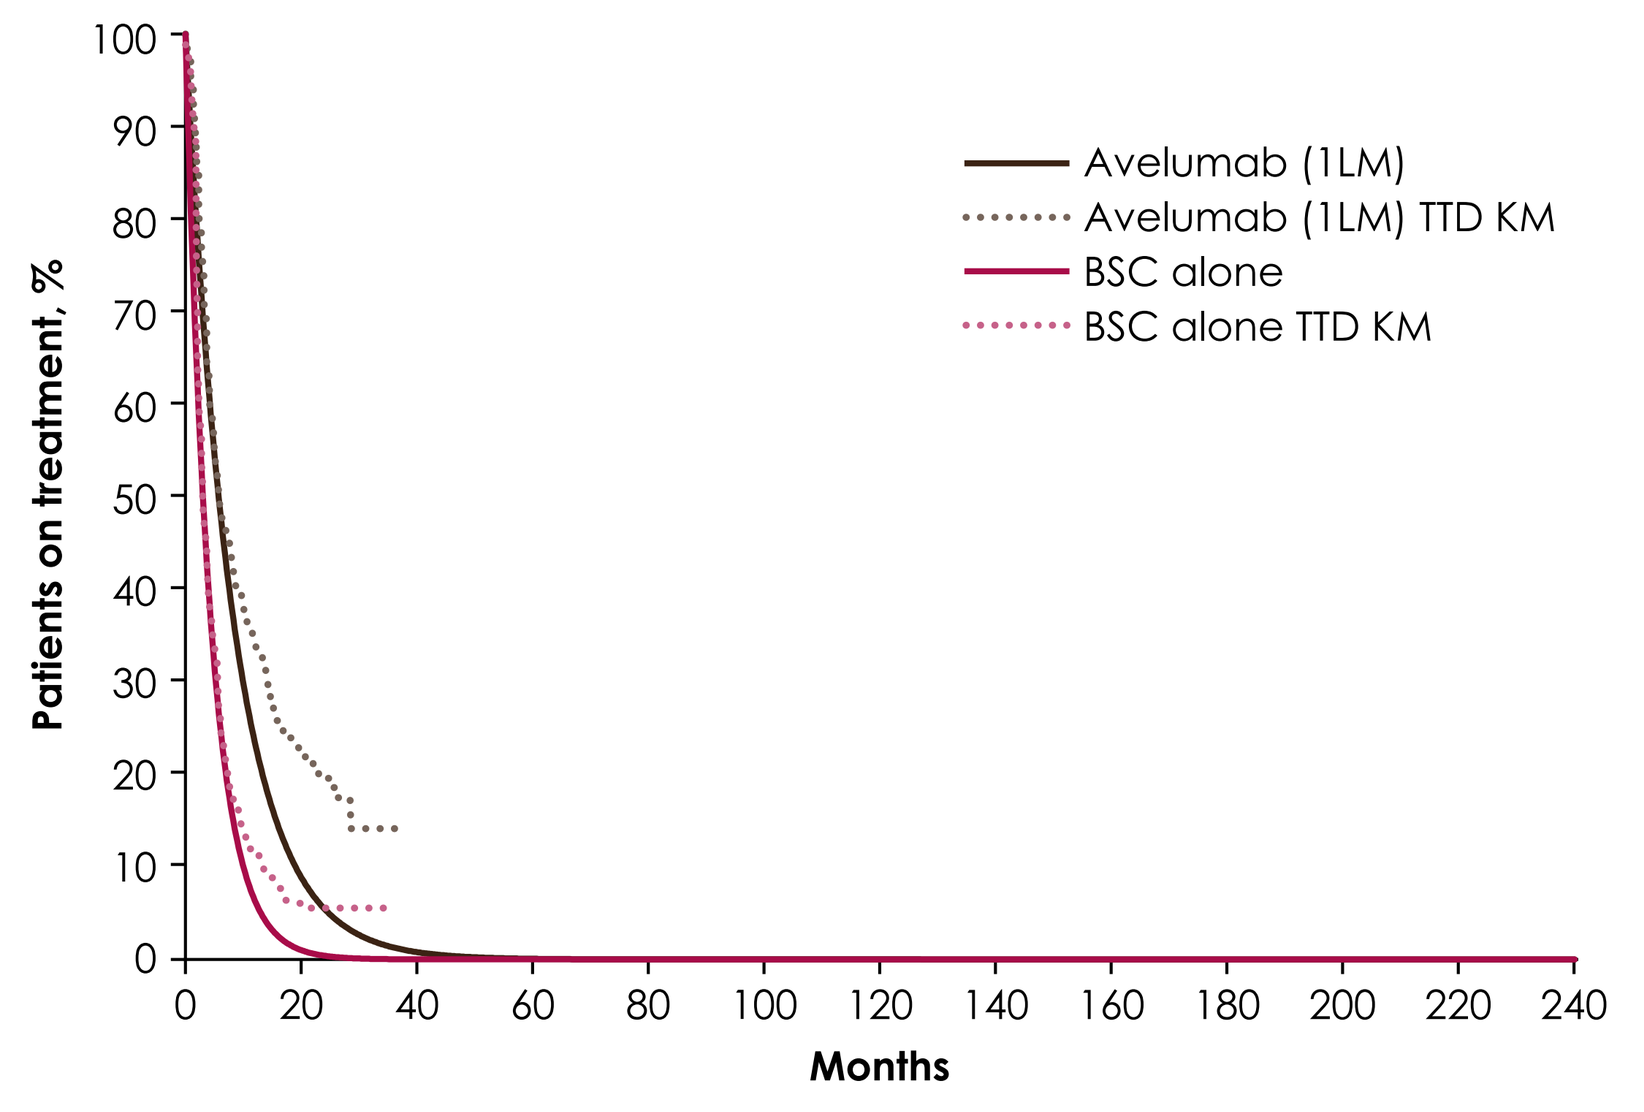
Patients on treatment, %
Months
Avelumab (1LM)
Avelumab (1LM) TTD KM
BSC alone
BSC alone TTD KM

1LM, first-line maintenance; **BSC**, best supportive care; **KM**, Kaplan-Meier; **TTD**, time to treatment discontinuation.

Figure 3. One-way sensitivity analyses

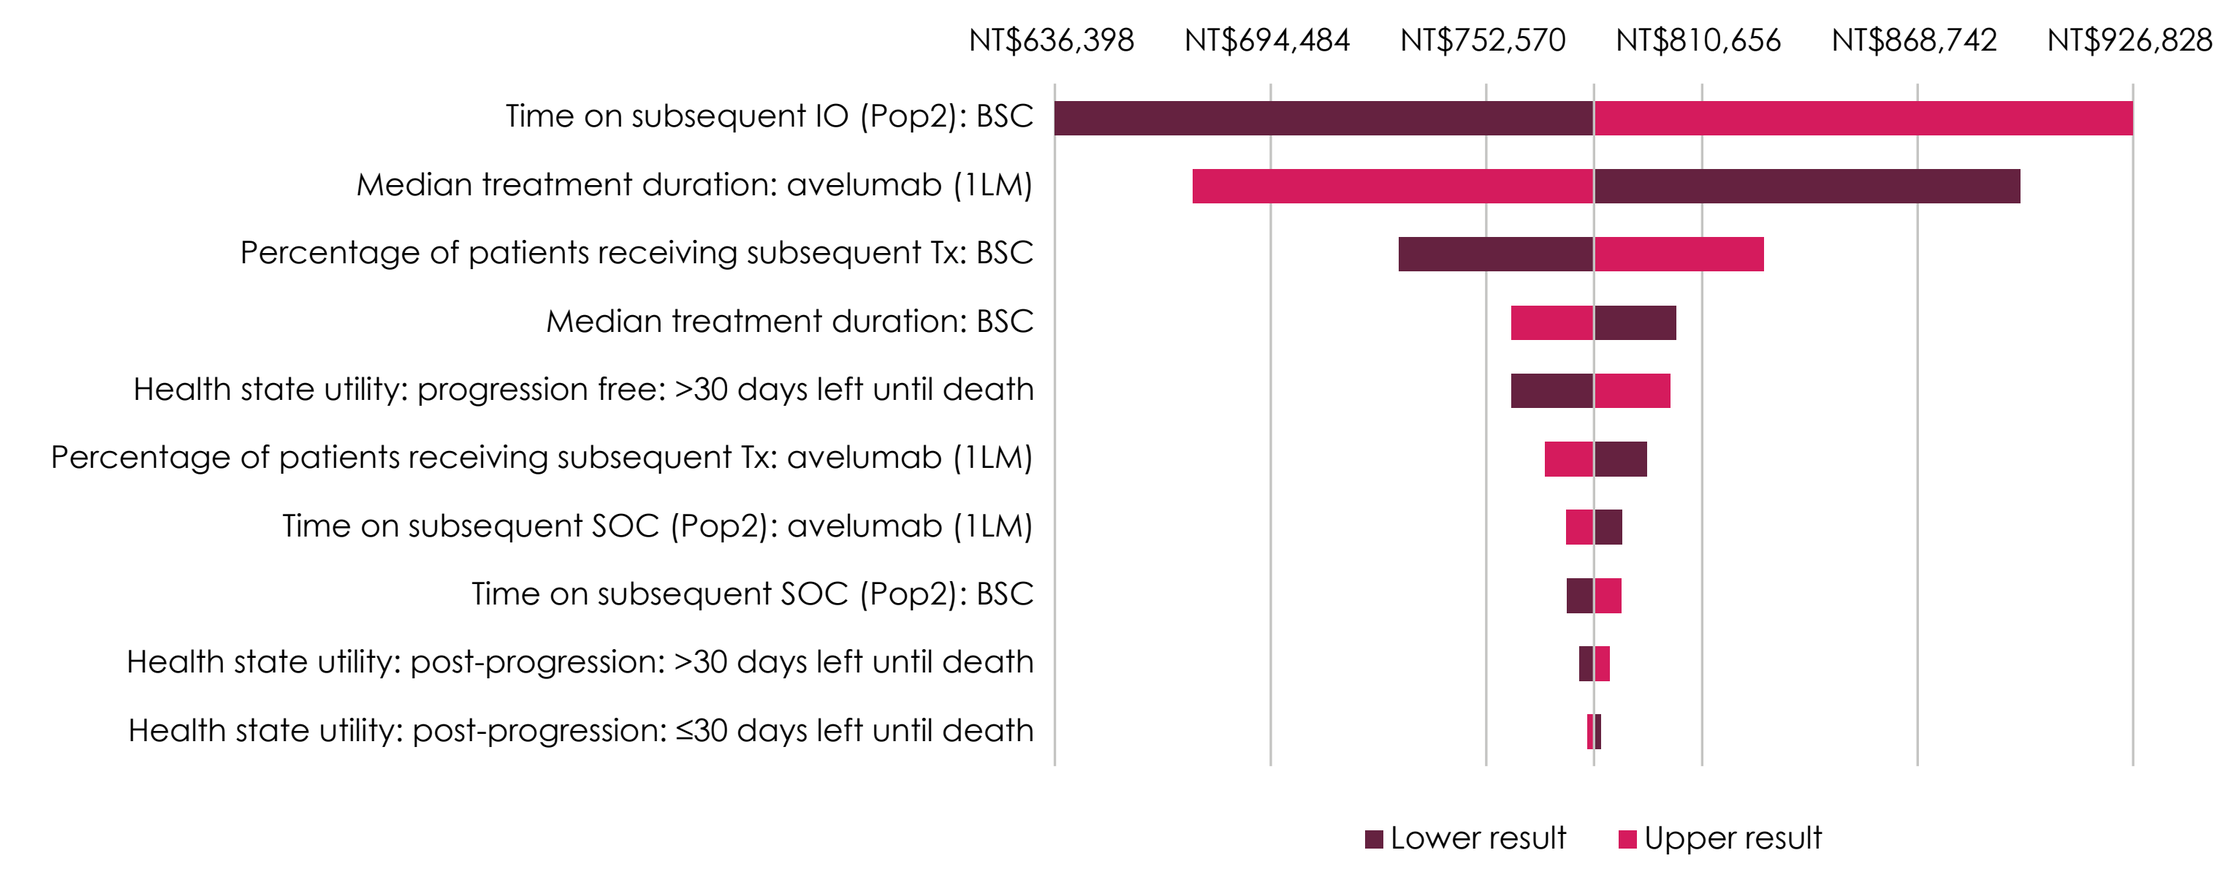
INMB = (incremental benefit × ICER threshold) - incremental cost. (i.e., 0.6 × 2,533,961-incremental cost).
NT$636,398
NT$694,484
NT$752,570
NT$810,656
NT$868,742
NT$926,828
Time on subsequent IO (Pop2): BSC
Median treatment duration: avelumab (1LM)
Percentage of patients receiving subsequent Tx: BSC
Median treatment duration: BSC
Health state utility: progression free: >30 days left until death
Percentage of patients receiving subsequent Tx: avelumab (1LM)
Time on subsequent SOC (Pop2): avelumab (1LM)
Time on subsequent SOC (Pop2): BSC
Health state utility: post-progression: >30 days left until death
Health state utility: post-progression: ≤30 days left until death
Lower result
Upper result

1LM, first-line maintenance; **BSC**, best supportive care; **ICER**, incremental cost-effectiveness ratio; **INMB**, incremental net monetary benefit; **IO**, immuno-oncology; **NT$**, new Taiwan dollar; **Pop2**, first-line maintenance population; **SOC**, standard of care; **Tx**, treatment.

Figure 4. (A) Incremental cost-effectiveness plane and (B) cost-effectiveness acceptability curve

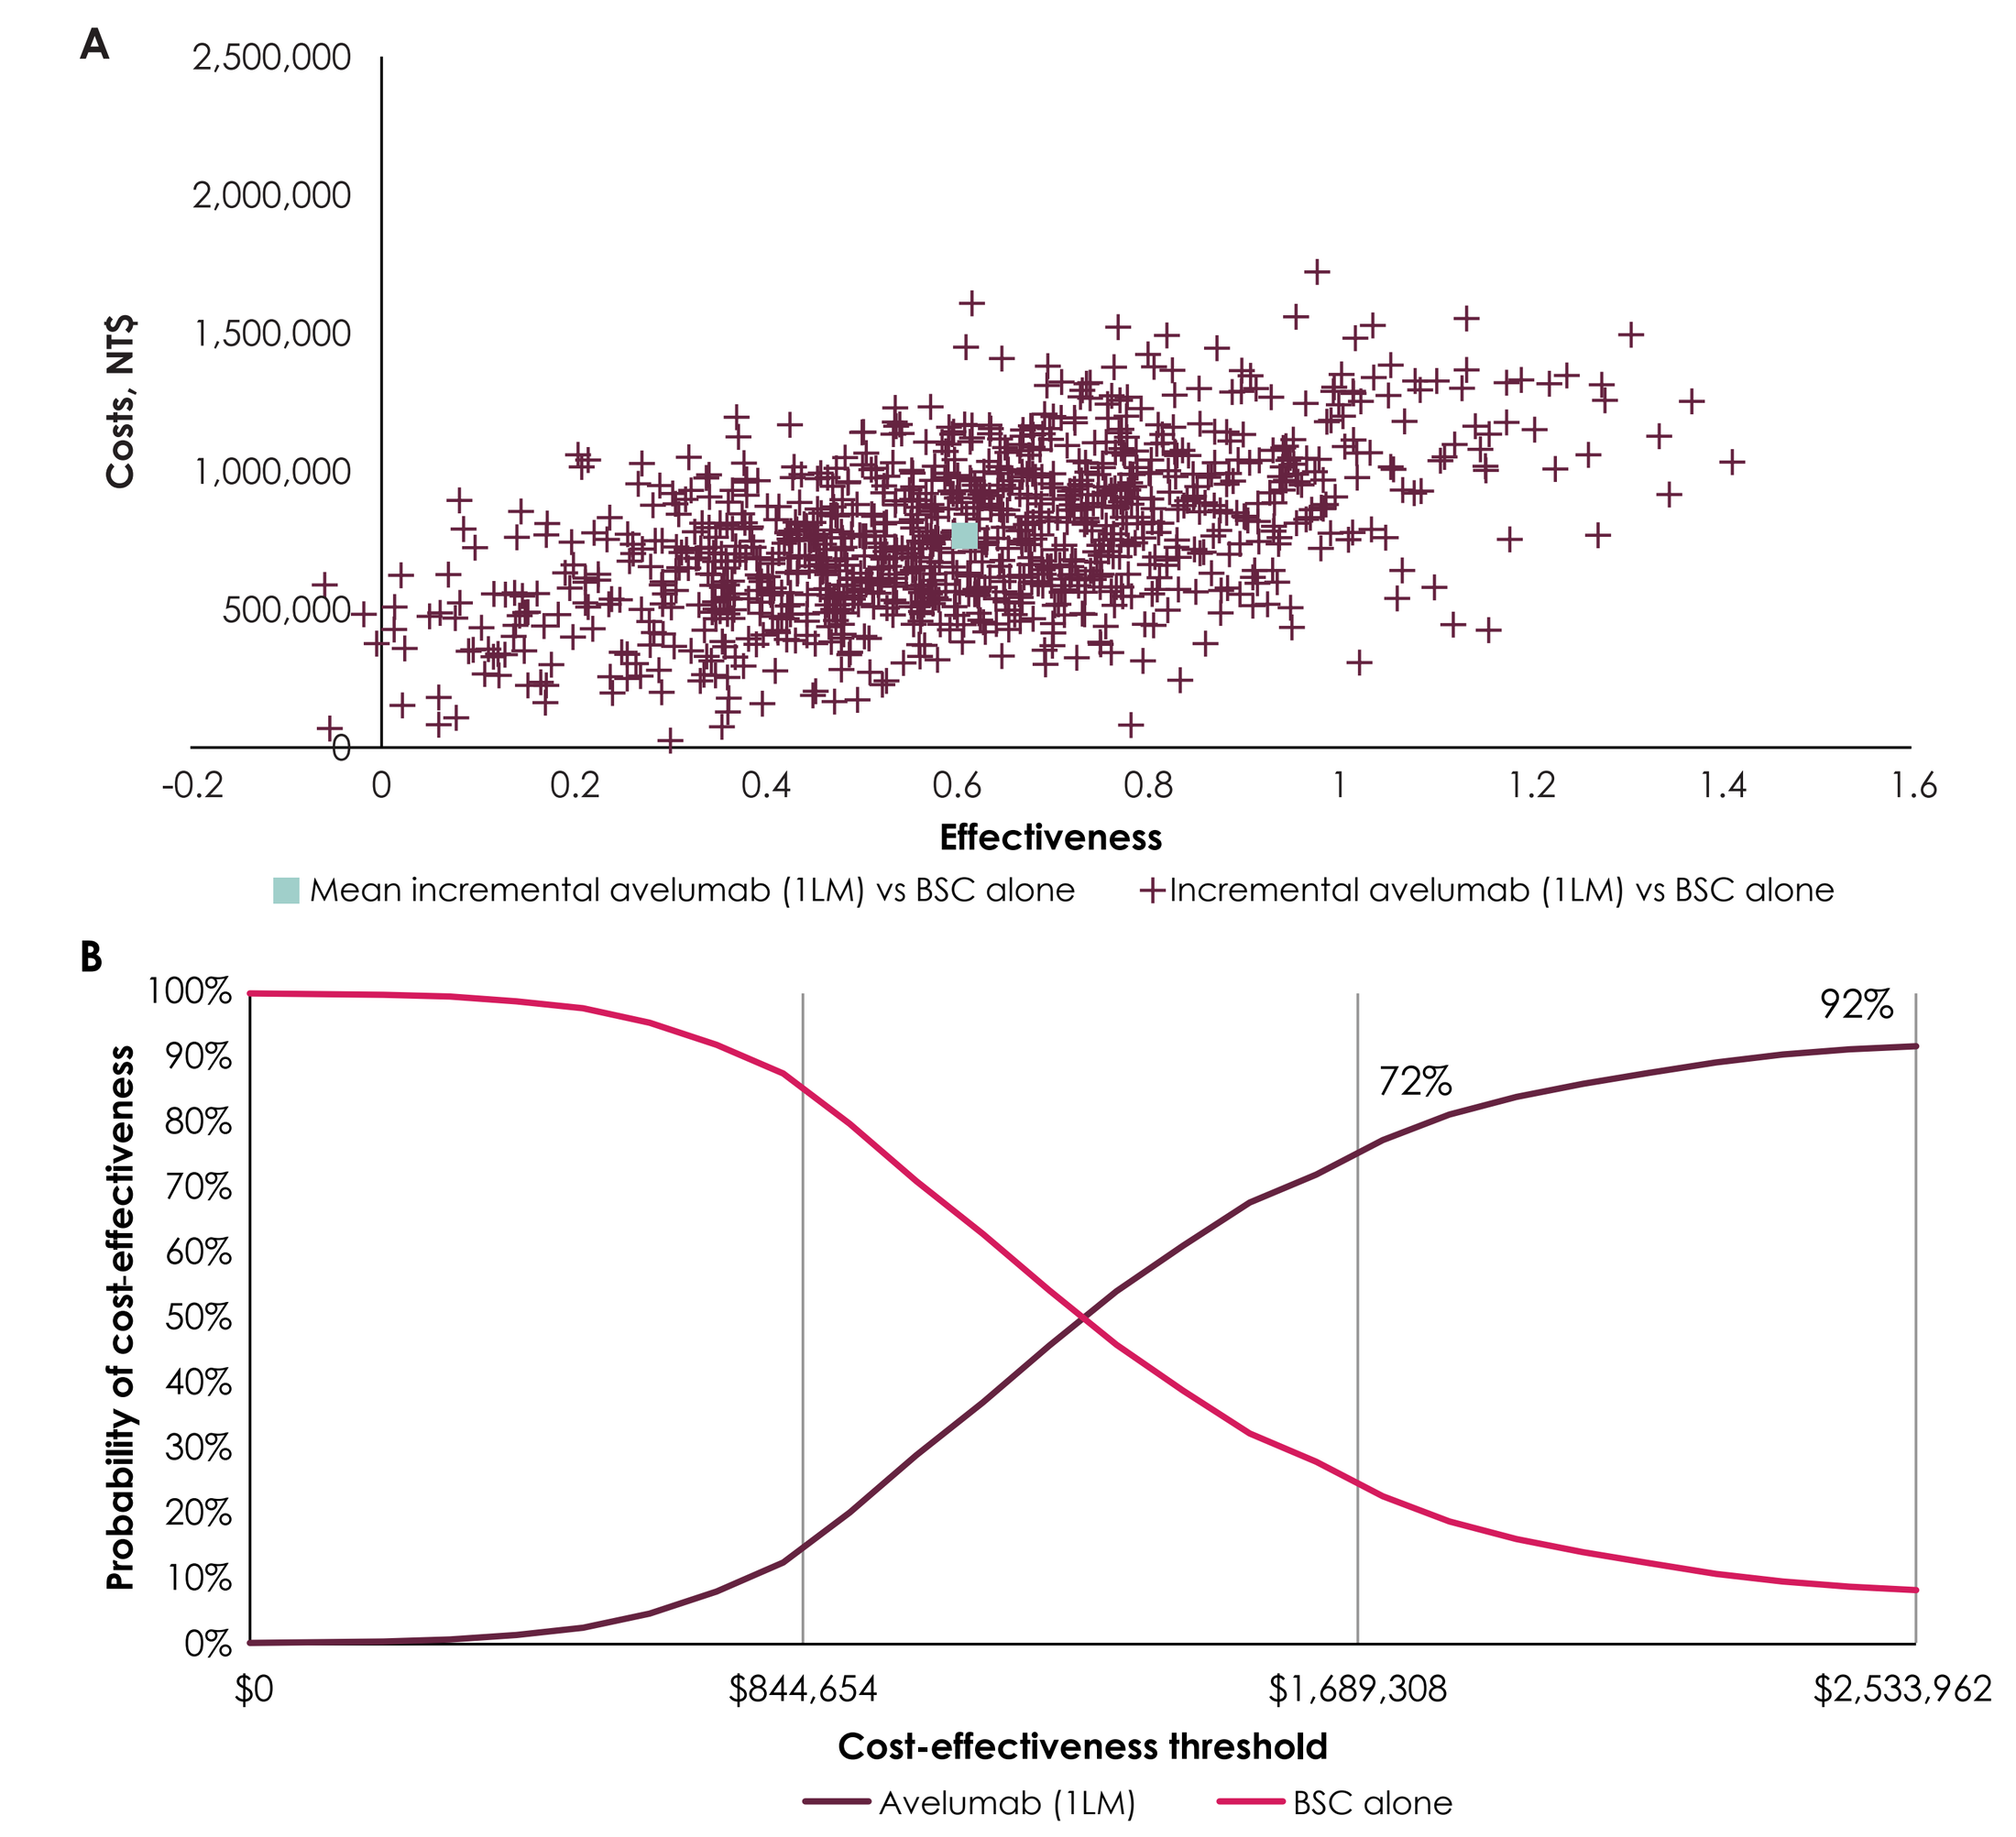
A
Costs, NT$
Effectiveness
Mean incremental avelumab (1LM) vs BSC alone
Incremental avelumab (1LM) vs BSC alone
B
Probability of cost-effectiveness
72%
92%
$0
$844,654
$1,689,308
$2,533,962
Cost-effectiveness threshold
Avelumab (1LM)
BSC alone

Current Taiwan GDP = NT$844,653; 2GDP = NT$1,689,308; 3GDP = NT$2,533,961.

1LM, first-line maintenance; **2GDP**, twice the GDP; **3GDP**, 3 times the GDP; **BSC**, best supportive care; **GDP**, Gross Domestic Product; **NT$**, new Taiwan dollar.

Table 1. First-line maintenance setting: HCRU costs per cycle in 2020

		Cost input, NT$	
Health states	**HCRU Categories**	**Avelumab + BSC**	**BSC alone**
PFS on/off treatment	1L maintenance treatment	1,059	220
	Off 1L maintenance treatment	1,192	1,192
	On subsequent treatment	708	708
PPS on/off treatment	Disease progression (on/off)	1,286	1,286
	Progressive disease (in addition to on/off treatment)	9,259	9,259

Notes: Estimates of HCRU were initially obtained via literature review and validated by Taiwan oncology experts.

1L, first line; **BSC**, best supportive care; **NT$**, new Taiwan dollar; **PFS**, progression-free survival; **PPS**, post-progression survival.

Table 2. Effectiveness and cost results

	Avelumab + BSC (1LM)	**BSC alone**
Total LYs	2.93	2.14
Progression free	1.38	0.69
Post-progression	1.55	1.45
Time on treatment	0.68	0.36
Total QALYs	2.15	1.54
Progression free	1.06	0.53
Post-progression	1.09	1.01
Costs, NT$		
Drug acquisition cost 1LM	1,400K-1,500K	Not applicable
Drug administration cost 1LM	17K-18K	Not applicable
AE management cost 1LM	3K-4K	0-1K
Disease progression cost (one-off)	1K-2K	1K-2K
Disease monitoring cost	887K-888K	784K-785K
Subsequent treatment cost	69K-70K	862K-863K
Terminal care costs	63K-64K	65K-66K

1LM, first-line maintenance; **BSC**, best supportive care; **K**, NT$1,000; **LYs**, life-years; **NT$**, new Taiwan dollar; **QALYs**, quality-adjusted life-years.

REFERENCES 1. Hung CF, et al. Jpn J Clin Oncol. 2016;46(7):605-9. **2.** Choueiri TK, et al. J Clin Oncol. 2012;30(5):507-12. **3.** Powles T, et al. N Engl J Med. 2020;383(13):1218-30. **4.** National Health Insurance Administration. Accessed September 2021. https://www.nhi.gov.tw/QueryN/Query1.aspx. **5.** Ministry of Health and Welfare. Accessed September 2021. https://dep.mohw.gov.tw/dos/cp-5034-57477-113.html. **6.** National Health Insurance Administration. Accessed September 2021. https://www.nhi.gov.tw/query/query2.aspx?n=6A7346FAE90CAF3F&sms=36A0BB334ECB4011&topn=D39E2B72B0BDFA15. **7.** Directorate-General of Budget, Accounting and Statistics, Executive Yuan. Accessed January 2021. https://www.dgbas.gov.tw/point.asp?index=1. **DISCLOSURES A. Lin** was an employee of Merck Ltd., Taipei, Taiwan, an affiliate of Merck KGaA, Darmstadt, Germany at the time the analysis was conducted. **C. Goh** is an employee of Merck Ltd., Taipei, Taiwan, an affiliate of Merck KGaA, Darmstadt, Germany. **E. Wu**, **K. Liu**, and **J. Chang** are employees of Pfizer. **R. Palencia** and **M. Kearney** are employees of the healthcare business of Merck KGaA, Darmstadt, Germany. **ACKNOWLEDGMENTS** This analysis was funded by the healthcare business of Merck KGaA, Darmstadt, Germany (CrossRef Funder ID: 10.13039/100009945), as part of an alliance between the healthcare business of Merck KGaA, Darmstadt, Germany and Pfizer. The authors would like to thank Dr Wen-Cheng Chang, assistant professor at Chang Gung University, Taiwan, for his participation and expertise.

Poster No. POSA128. Presented at Virtual ISPOR Europe 2021, 30 November - 3 December 2021.