Cost-effectiveness of nivolumab in patients with pre-treated advanced renal cell carcinoma (aRCC) in the United States: impact of >5 years of follow-up data from CheckMate 025

PDG20

May JR[1], Malcolm B[1], Ejzykowicz F[2], Kurt M[2], Borrill J [1], Theriou C[3], Teitsson S[3]

[1]Bristol Myers Squibb, London, United Kingdom; [2]Bristol Myers Squibb, New Jersey, United States;
[3]Parexel International, London, United Kingdom

Introduction

Renal cell carcinoma (RCC)

- Across Europe, kidney cancer was the 8th most common cancer in 2018. There were an estimated 136,515 newly diagnosed cases and 54,709 deaths [1]
- In the US, SEER program estimates that for the year 2020, the number of new cases of kidney cancer is 73,750 and deaths is 14,830, accounting for 2.4% of all cancer-related deaths [2]
- The median age at diagnosis is 64 years, with about 50% of all tumors found incidentally.[3] RCC represents ~80% of all kidney cancers [4]
- Despite introduction of immuno-oncologic (IO)-containing combination therapies in 1L, many patients still receive TKI monotherapy, highlighting the continuing need for improved efficacy and toxicity of 2L+ therapies

Clinical background of nivolumab

- Nivolumab (NIVO), a programmed cell death-1 (PD-1) inhibitor, is the only IO therapy approved for the treatment of pre-treated patients with aRCC [5,6]
- CheckMate 025 (ClinicalTrials.gov Identifier: NCT01668784) is a randomized phase III trial investigating NIVO versus everolimus (EVE) in pre-treated aRCC with the longest follow-up of any phase III study of immunotherapy in RCC (≥64 months)
- The clinical data for NIVO show superiority compared with EVE in the primary endpoint of overall survival (OS) and also in objective response rate (ORR); the responses were found to be durable [7]
 - Data from the initial database lock (DBL) (minimum follow-up of 14 months; June 2015) confirmed the trial had met its primary endpoint of OS and was considered final. The trial demonstrated OS benefit with NIVO over EVE (hazard ratio [HR], 0.73; [98.5% confidence interval [CI], 0.57–0.93; *P* = 0.002]) [8]
 - With additional follow-up data from the latest DBL (July 2019), superior OS was maintained with NIVO versus EVE over a minimum of 64 months of follow-up (HR, 0.73; 95% CI, 0.62–0.85; *P* < 0.0001) [7]
- Premised on both efficacy and safety data from Checkmate 025, NIVO provided a paradigm shift for the treatment landscape of pre-treated aRCC, where few treatment options were previously available
 - Fewer NIVO-treated patients experienced treatment-related adverse events (AEs) compared with patients treated with EVE, and no new safety signals were observed with longer follow-up [7]
- Following the approval of nivolumab, based on the CheckMate 025 results, NIVO has been widely established as the standard of care in pre-treated patients who have not received IO therapy in 1L [9,10]

Figure 1. OS Kaplan-Meier curves for NIVO and EVE from the 64-month minimum follow-up data-cut in CheckMate 025 [7]

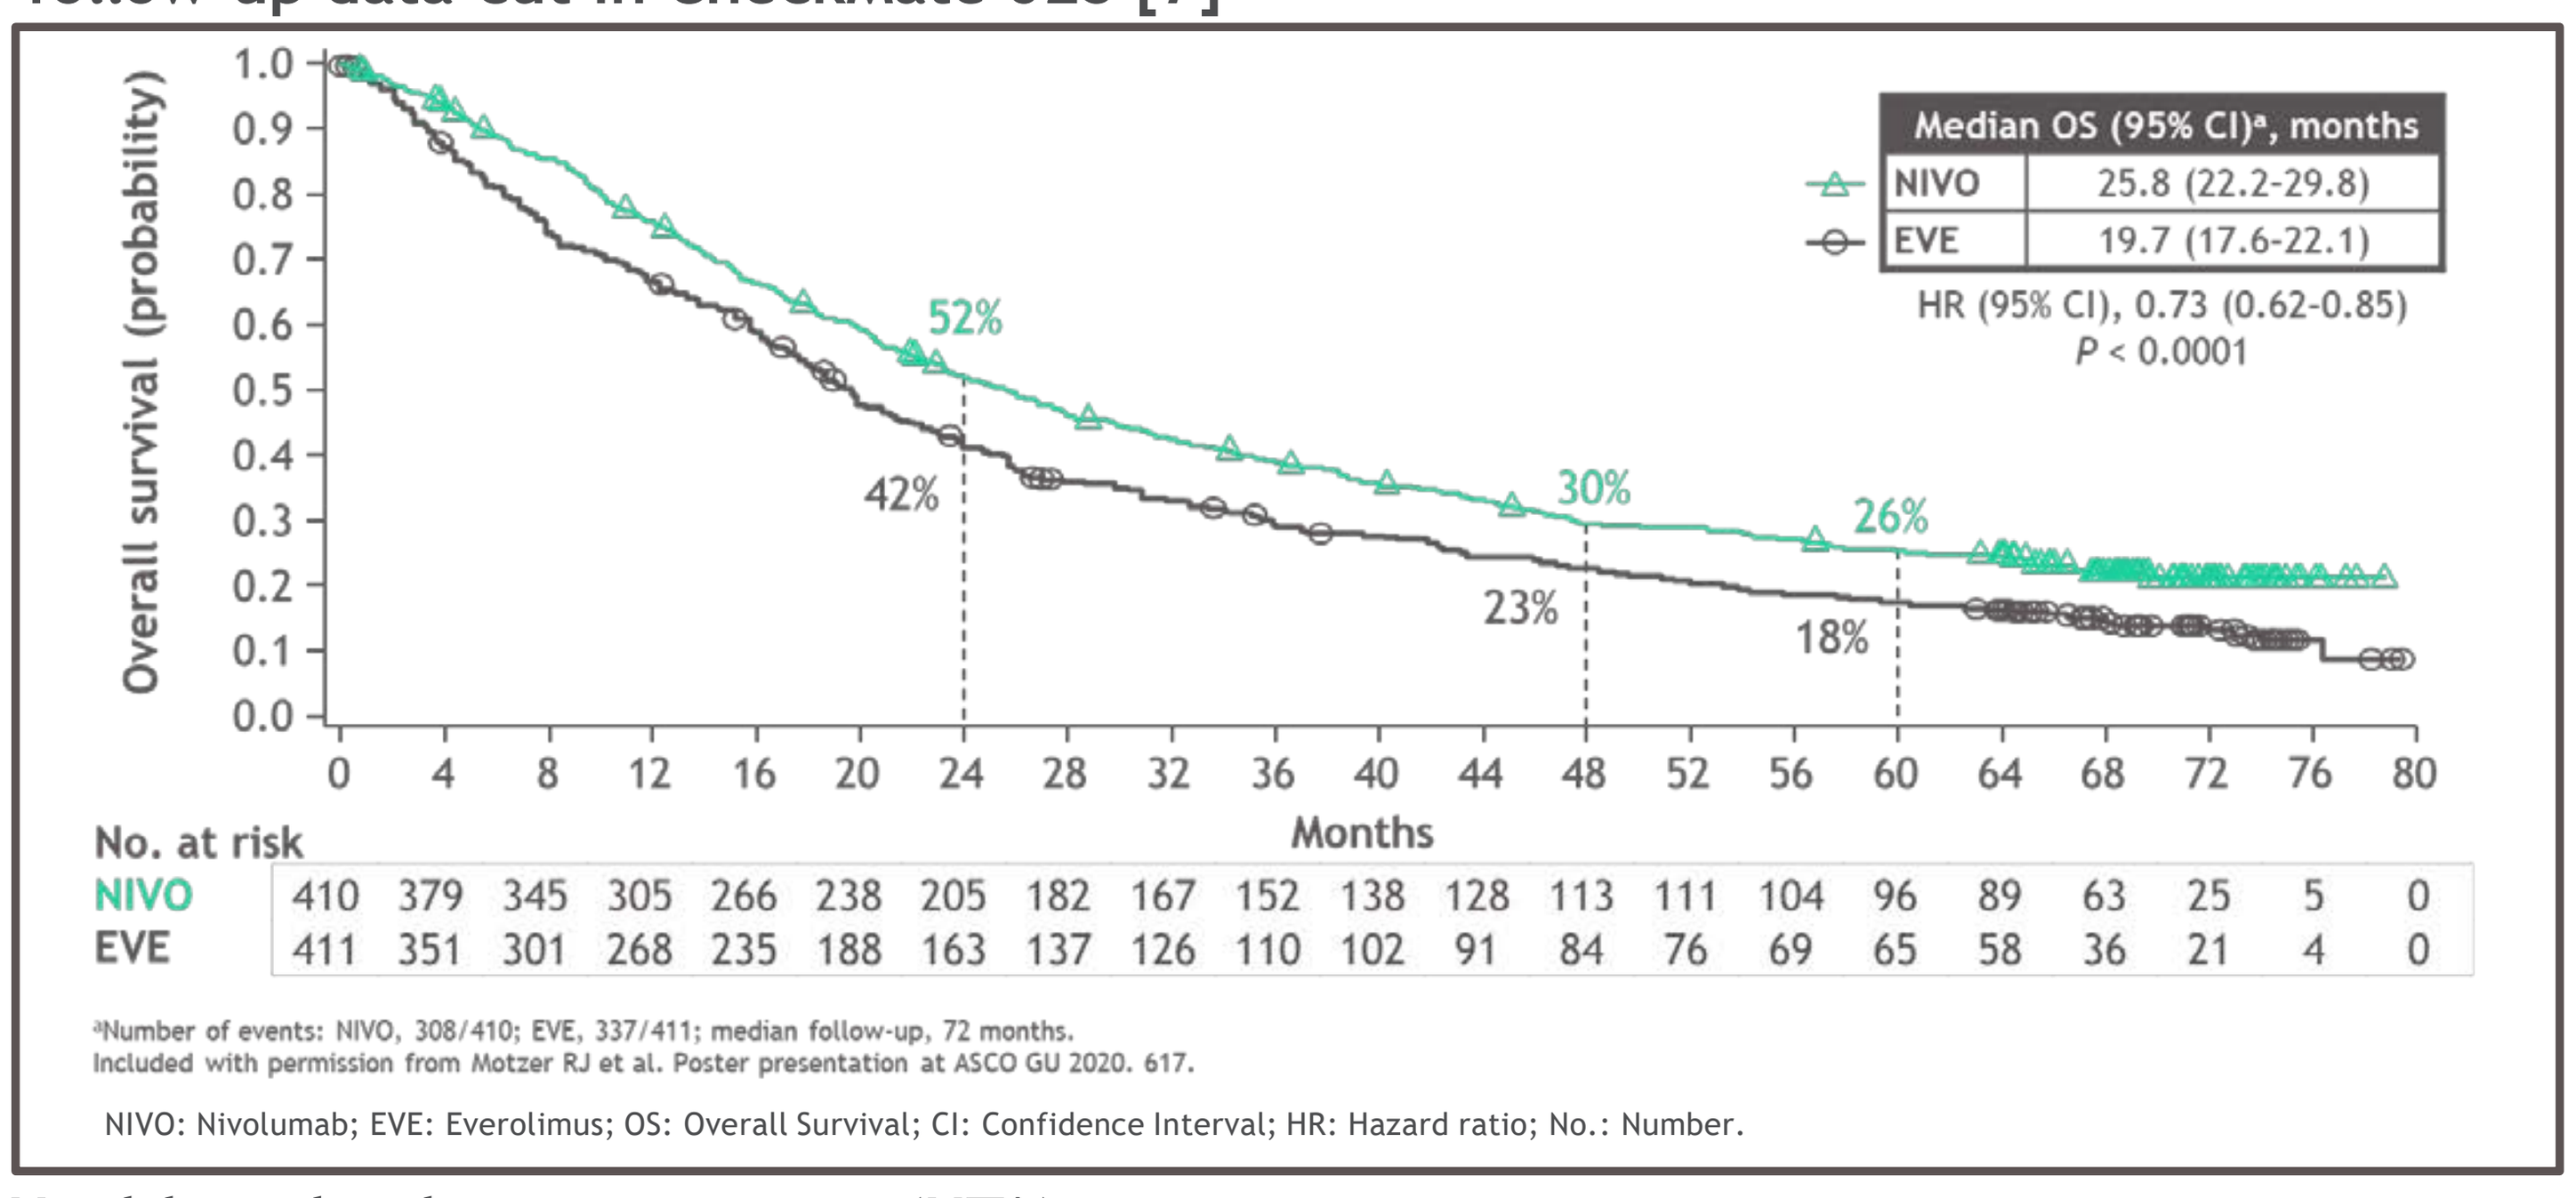
Overall survival (probability)
Months
Median OS (95% CI)a, months
NIVO 25.8 (22.2-29.8)
EVE 19.7 (17.6-22.1)
HR (95% CI), 0.73 (0.62-0.85)
P < 0.0001
52%
42%
30%
23%
26%
18%
No. at risk
NIVO 410 379 345 305 266 238 205 182 167 152 138 128 113 111 104 96 89 63 25 5 0
EVE 411 351 301 268 235 188 163 137 126 110 102 91 84 76 69 65 58 36 21 4 0

aNumber of events: NIVO, 308/410; EVE, 337/411; median follow-up, 72 months.
Included with permission from Motzer RJ et al. Poster presentation at ASCO GU 2020. 617.

NIVO: Nivolumab; EVE: Everolimus; OS: Overall Survival; CI: Confidence Interval; HR: Hazard ratio; No.: Number.

Health technology assessment (HTA)

- Manufacturers often seek reimbursement of new oncology treatments based on early clinical trial results
- Oncology trials often have limited follow-up at the time of HTA submission and therefore extrapolating the available trial survival data using parametric survival models is necessary to estimate lifetime survival outcomes for cost-effectiveness analysis
- In addition, data for other cost-effectiveness model inputs, such as patient utility (particularly after progression of the disease) and subsequent treatment, can often be limited from early trial results
- Maturity of clinical trial data can have implications on the estimated input values used in a cost-effectiveness analysis, and ultimately impacts cost-effectiveness results and reimbursement decisions
- As the primary endpoint of CheckMate 025 trial was met with a minimum of 14 months of follow-up, Health Technology Assessment (HTA) submissions were mostly based on an evidence package from the initial DBL

Objective

- This study compares predicted survival outcomes and cost-effectiveness results when using data from the initial and latest DBLs with a minimum follow-up of 14 and 64 months, respectively, from CheckMate 025

Methods

Cost-effectiveness model

- A partitioned survival cost-effectiveness model (CEM) consisting of three health states (progression-free [PF], progressive disease [PD], and death) was developed to evaluate the incremental cost-utility ratio (ICUR) and incremental cost-effectiveness ratio (ICER) of NIVO versus EVE [11]
- The model took a United States payer perspective, with a 25-year time horizon, 4-week cycle length, and 3% annual discounting for costs (2020 US$) and outcomes
- Standard parametric models and spline-based models were fitted to observed progression-free survival (PFS), OS and time-to-treatment discontinuation (TTD) data from CheckMate 025
- Survival analysis was conducted following guidance from Technical Support Document #14 published by the National Institute for Health and Care Excellence (NICE) Decision Support Unit [12]
- EuroQol-5 Dimensions (EQ-5D-3L) data from CheckMate 025 were analyzed to inform health state utility values (HSUVs) in the CEM
 - Patients receiving NIVO in the CheckMate 025 trial showed statistically significant improvement in EQ-5D scores compared with EVE patients for both PF and PD health states (Table 1); therefore, treatment-specific HSUVs were applied in the CEM
 - Utility values for the PF health state were weighted according to the ORR observed in CheckMate 025
- Time on treatment for both NIVO and EVE was estimated by extrapolating observed TTD data from the CheckMate 025 trial and capped at 7 years for both NIVO and EVE

Analysis

- Two different DBLs from CheckMate 025 are used in this analysis, with a minimum follow up of 14 months (initial DBL) and 64 months (latest DBL)
- The CEM based on the latest DBL was updated with trial-specific data from the initial DBL, one after the other in a stepwise fashion, to explore the impact of differences in maturity in the CheckMate 025 trial data on cost-effectiveness results and predicted survival
- Compared CEM outcomes included life-years (LYs), quality-adjusted life-years (QALYs) and total costs
- Due to the high impact of modelled OS on the CEM results, and to fully explore the estimated survival benefits of NIVO versus EVE, we further assessed differences in the restricted mean survival time (RMST) from the parametric survival models fitted to data from both DBLs (Table 3) over a 25-year time horizon

Trial-specific inputs used in the CEM

- The following trial-specific data inputs used in the CEM were considered for this analysis: PFS, OS and TTD curves, HSUVs, ORR, and subsequent treatment patterns (Table 1)
- Dependent log-logistic curves (proportional hazards assumption was upheld) were used to model NIVO and EVE OS in the base case analysis for both DBLs as they provided the best statistical goodness of fit, a good visual fit of curves, and represented one of the most conservative incremental long-term differences between the NIVO and EVE arms
- A dependent 2-knot spline hazard model was applied for NIVO and EVE PFS when using data from the initial DBL but an independent 2-knot spline hazard model was used when using the latest DBL as the PH assumption was violated
- Independent 2-knot spline normal model was used to model NIVO TTD across both DBLs, and an independent log-normal model was used for EVE TTD for both DBLs
- Input values for HSUVs and ORR used in the cost-effectiveness model, sourced from the initial and the latest DBL of the CheckMate 025 trial, are shown in Table 1

Table 1. Differences in HSUVs and ORR between the initial and latest DBL

Data input	Initial DBL		Latest DBL	
	NIVO	EVE	NIVO	EVE
PF utility value: Responders	0.898 (N=1,322)	0.879 (N=240)	0.901 (N=2,332)	0.881 (N=406)
PF utility value: Non-responders	0.848 (N=1,531)	0.844 (N=1,762)	0.860 (N=1,884)	0.844 (N=2,297)
PD utility value	0.830 (N=1,473)	0.804 (N=1,303)	0.819 (N=1,588)	0.797 (N=1,123)
ORR %a	25.1a, [8]	5.4a, [8]	22.9b, [7]	4.1b, [7]
Odds ratio (95% CI) for ORR: NIVO vs. EVE	5.98 (3.68-9.72)		6.86 (4.01-11.74)	

aInvestigator assessed response; bConfirmed response; DBL: Database lock; NIVO: Nivolumab; EVE: Everolimus; PF: Progression-free; ORR: Objective response rate; PD: Progressed disease; CI: Confidence interval

- Fewer NIVO patients received subsequent treatment than EVE patients in both DBLs. Subsequent therapy use in the initial DBL was 55% and 63% for NIVO and EVE6, respectively, compared with 67% and 72% in the latest DBL for NIVO and EVE, respectively [7]
- The mean duration of subsequent treatment in the CEM (assumed same for all treatments) was 3.65 months and was sourced from GOLD trial [13]
- Distribution of subsequent treatments was similar across both study arms. Use of subsequent nivolumab and cabozantinib treatments increased between the initial and latest DBL for patients in both trial arms, likely due to the availability of these therapies

Results

Survival outcomes

- With OS trial data extrapolated over a lifetime horizon (25 years), total LYs for NIVO and EVE based on the initial DBL were marginally underestimated by 3.12% (1.63 months) and 0.57% (0.23 months), respectively, when compared with estimations based on the latest DBL (Table 2, Figure 2)
- Mean PFS estimated from the initial DBL was 25.40% and 5.86% lower for NIVO and EVE, respectively, when compared with mean PFS based on extrapolations from the latest DBL (Table 2, Figure 2)
- This confirms that the base case parametric models from the initial DBL conservatively estimated the benefit of NIVO versus EVE when compared with extrapolations based on longer term data

Table 2. Comparison of RMST in months from parametric models fitted to data from both DBLs

DBL	PFS			OS		
	NIVO	EVE	Incremental	NIVO	EVE	Incremental
Initial DBL (RMST in months)	11.96	7.57	4.09	50.75	39.79	10.96
Latest DBL (RMST in months)	15.63	8.04	7.59	52.38	40.02	12.36
Initial DBL – Latest DBL (months)	-3.97	-0.47	-3.5	-1.63	-0.23	-1.40
% difference	-25.40%	-5.86%	-19.54%	-3.12%	-0.57%	-2.55%

RMST: Restricted mean survival time; DBL: Database lock; PFS: Progression-free survival; OS: Overall survival; NIVO: Nivolumab; EVE: Everolimus;

Figure 2. Parametric PFS and OS curves for NIVO and EVE from initial and latest DBL of CheckMate 025 trial

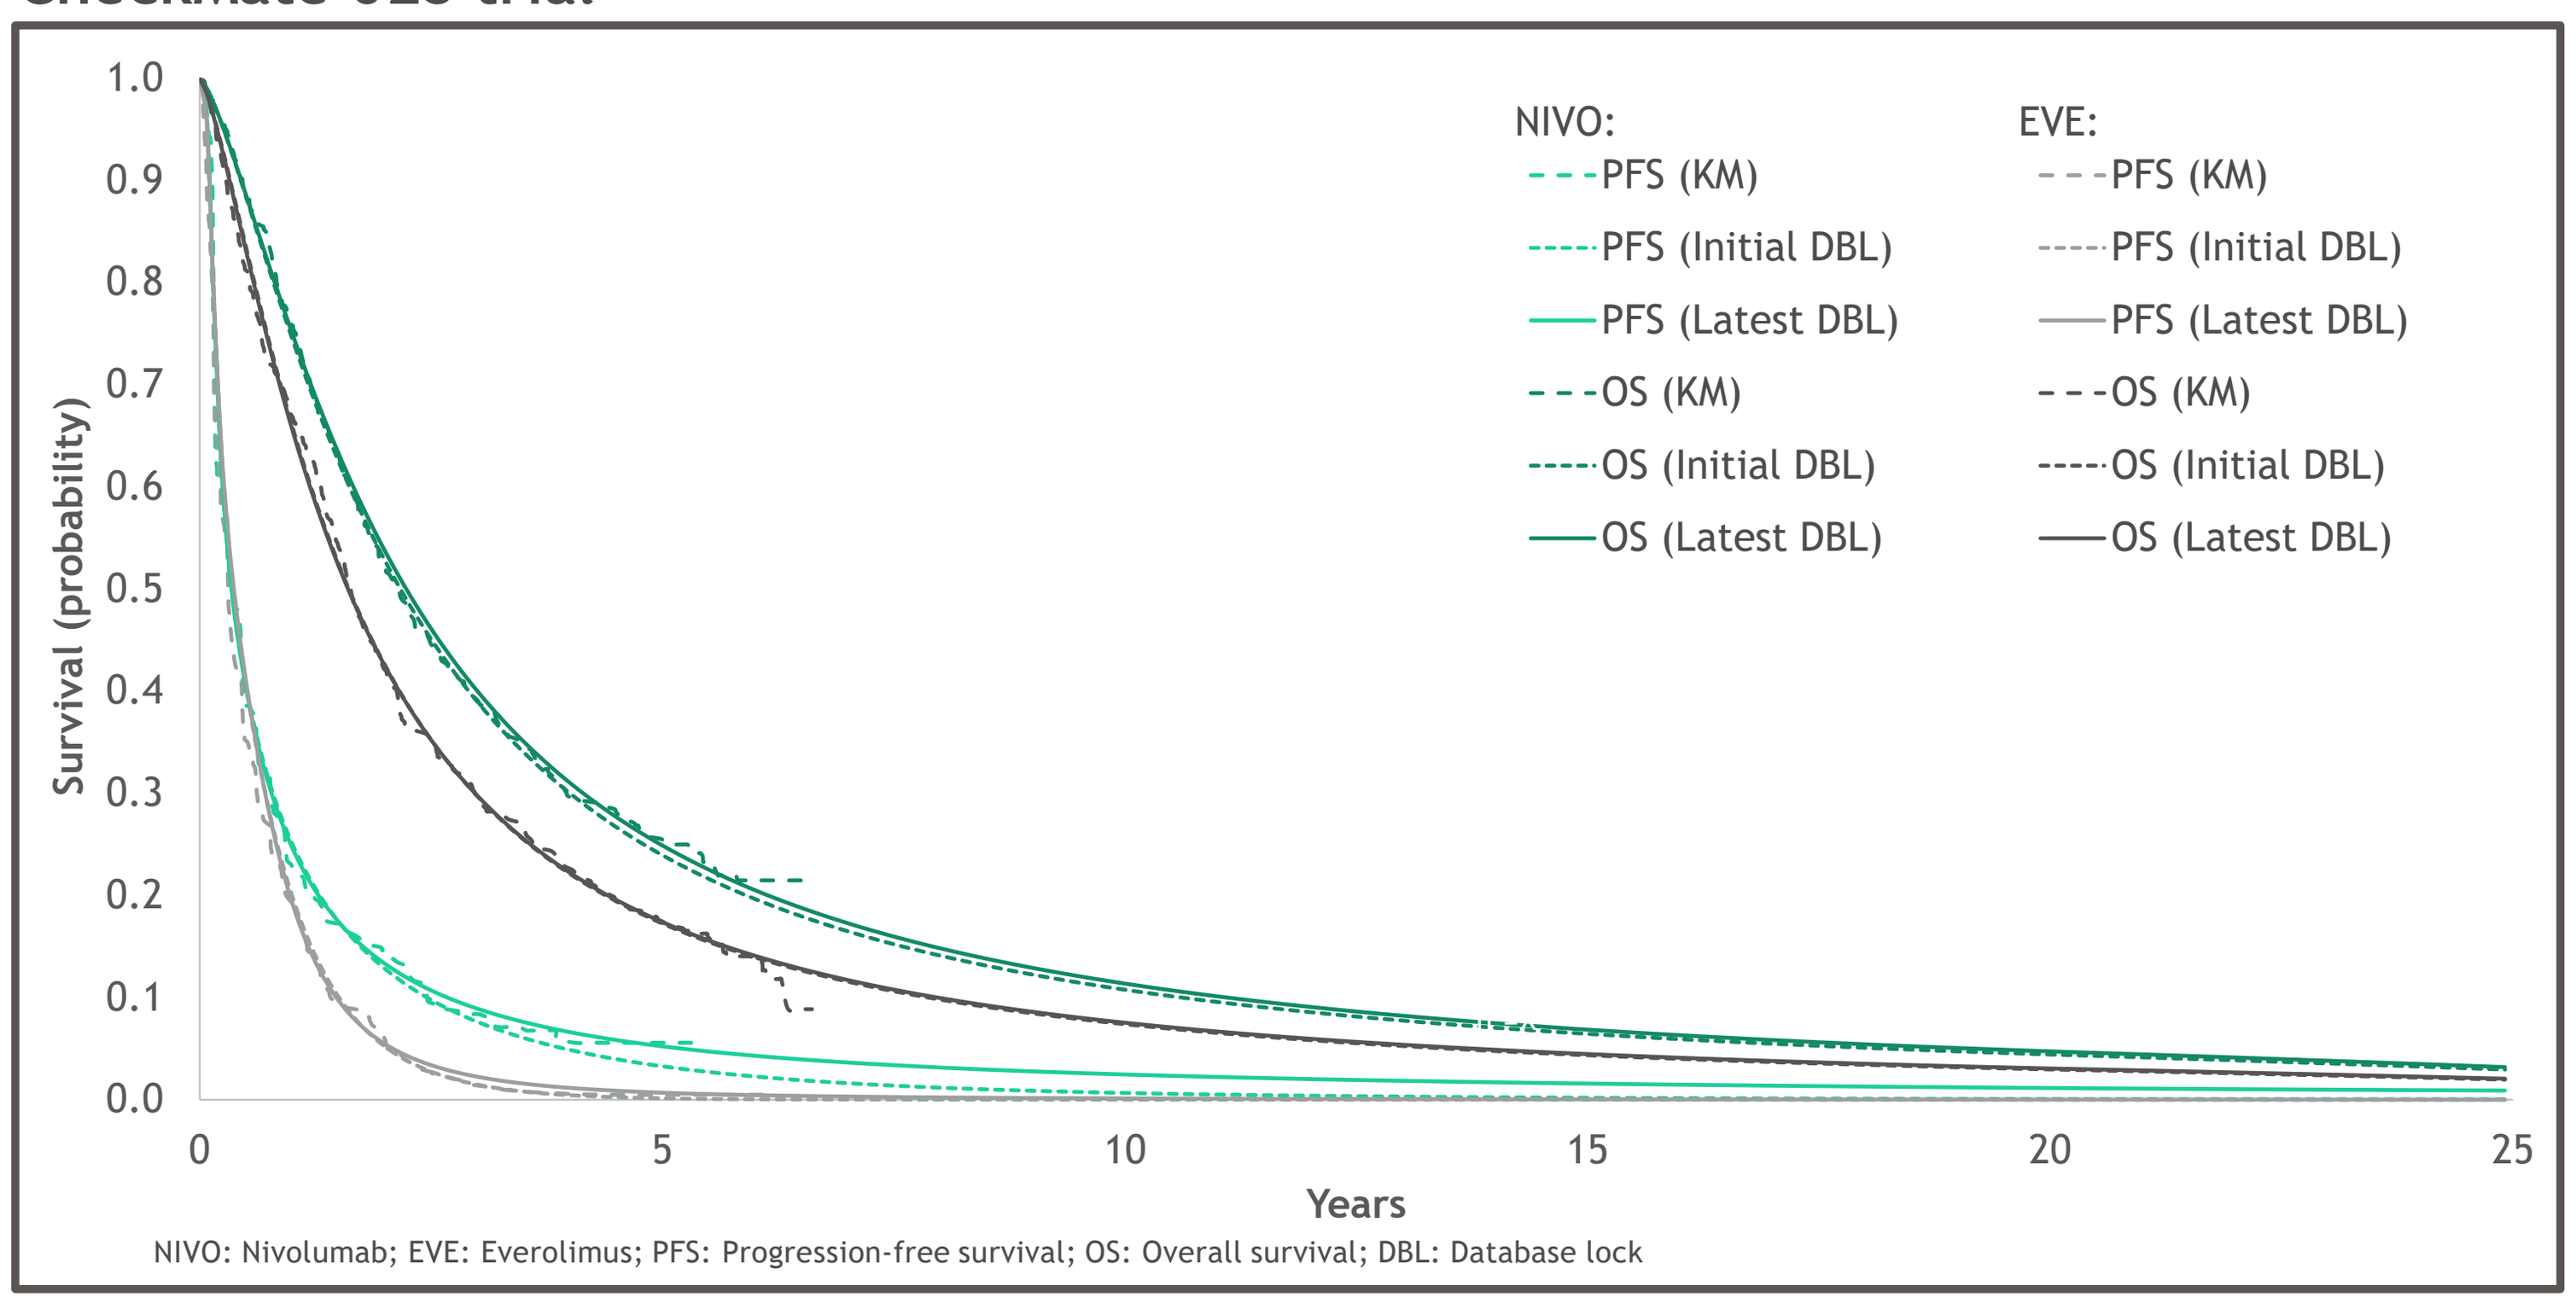
Survival (probability)
Years
NIVO:
PFS (KM)
PFS (Initial DBL)
PFS (Latest DBL)
OS (KM)
OS (Initial DBL)
OS (Latest DBL)
EVE:
PFS (KM)
PFS (Initial DBL)
PFS (Latest DBL)
OS (KM)
OS (Initial DBL)
OS (Latest DBL)

NIVO: Nivolumab; EVE: Everolimus; PFS: Progression-free survival; OS: Overall survival; DBL: Database lock

- The underestimation in the initial PFS and OS extrapolations compared with those based on data from the latest DBL, is further highlighted by 5-year survival rate comparisons (Table 3)

Table 3. Comparison of 5-year OS and PFS rates for nivolumab between observed trial data and extrapolations from the initial and latest DBLs

Outcome	Observed data	Original DBL (extrapolated)	Latest DBL (extrapolated)
OS	26%	24%	25%
PFS	5%	3%	5%

OS: Overall survival; PFS: Progression-free survival; DBL: Database lock.

Base case CEM results

- Using longer term trial data to inform the CEM inputs resulted in a modest reduction in the ICUR and ICER for NIVO versus EVE (Table 4)
- This was mainly driven by the initial DBL estimating lower incremental LY and QALY gains for NIVO versus EVE (by 11.4% and 11.0%, respectively) compared with the latest DBL, mainly explained by the conservative PFS and OS extrapolations from the initial DBL

Table 4. Incremental CEM results when using trial-specific data from the initial and latest DBL to inform CEM inputs

DBL	Treatment	Total costs	Total LYs	Total QALYs	Inc. costs	Inc. LYs	Inc. QALYs	ICER	ICUR
Initial DBL	NIVO	$250,449	4.229	3.089	-	-	-	-	-
	EVE	$199,353	3.316	2.383	$51,096	0.913	0.706	$55,946	$72,389
Latest DBL	NIVO	$287,938	4.365	3.172	-	-	-	-	-
	EVE	$232,053	3.335	2.379	$55,885	1.030	0.793	$54,240	$70,490

DBL: Database lock; LY: Life-years; QALYs: Quality-adjusted life-years; ICER: Incremental cost-effective ratio; ICUR: Incremental cost-utility ratio; NIVO: Nivolumab; EVE: Everolimus

- Although extrapolations from the initial DBL underestimated PFS benefit for NIVO versus EVE more so than OS, the marginal differences in OS predictions between DBLs had greater impact on the CEM results
 - Changes in PFS mainly impacted QALYs and disease management costs in the PF health state
- Using data from the latest DBL resulted in higher incremental costs for NIVO versus EVE but this was offset by the increase in LY and QALY gains for NIVO with longer follow-up data
 - The higher incremental costs were mainly driven by differences in the proportion of patients receiving subsequent treatment in the NIVO and EVE arms of the CheckMate 025 trial, as well as the distribution of subsequent treatment options in each arm between the two DBLs
 - TTD extrapolations were similar between the DBLs and therefore did not drive the higher incremental costs
- Changes in HSUVs and ORR between DBLs had limited impact on the CEM results

Limitation

- In the EVE arm, 65 patients (16.4%) crossed over to the NIVO extension phase according to a protocol amendment after the primary analysis. Adjustments for crossover were not undertaken in this analysis.

Conclusions

- NIVO has demonstrated superior and sustained OS compared with EVE for pre-treated RCC patients consistent between the initial DBL (0.913 LYs gained) and latest DBL (1.030 LYs gained)
- Parametric survival models fitted to OS data from the initial DBL were reasonably accurate but conservatively estimated the OS benefit of NIVO relative to EVE when compared with observed trial data and extrapolations from the latest DBL; the same functional form was best fitting for both DBLs
- Extrapolations from the initial DBL also underestimated the PFS benefit of NIVO versus EVE (-19.54%) when compared with predictions using the latest DBL, but this had less impact on the ICUR and ICER
- Higher incremental costs were observed with longer follow-up, mainly driven by data for subsequent treatment. However, this was offset by increased LY and QALY gains between the DBLs
- This study suggests that the initial DBL used in most HTA submissions for NIVO in aRCC conservatively estimated both survival benefit and ICUR for NIVO versus EVE

References

[1] World Health Organization. Cancer fact sheets: Europe, 2018.
[2] Howlander N, et al. SEER cancer statistics review 1975-2016. Natl. Cancer Institute. 2019.
[3] Ferlay J, et al. *Int J Cancer*. 2010;127(12):2893-2917.
[4] Escudier B, et al. *Ann. Oncol*. 2019;30(1):706–720.
[5] Food and Drug Administration, Prescribing information for Opdivo (nivolumab). U.S. Food and Drug Administration (FDA), 2014. Reference ID: 4421379
[6] European Medicines Agency. Opdivo (nivolumab): Summary of product characteristics https://www.ema.europa.eu/en/documents/product-information/opdivo-epar-product-information_en.pdf (accessed 01/07/2020)
[7] Motzer RJ, et al. Poster presentation at the Genitourinary Cancers Symposium; February 13–15, 2020; San Francisco, CA, USA. Abstract #617.
[8] Motzer RJ, et al. *N Engl J Med*. 2015;373:1803–1813.
[9] NCCN. 2019. Guidelines for patients: Kidney Cancer, https://www.nccn.org/patients/guidelines/kidney/files/assets/common/downloads/files/kidney.pdf
[10] EAU. 2018. EAU Guidelines on Renal Cell Carcinoma. https://uroweb.org/wp-content/uploads/EAU-RCC-Guidelines-2018-large-text.pdf
[11] McCrea C, et al. *Exp. Hematol. Oncol*. 2018;7(1).
[12] Latimer N. Sheffield: Report by the Decision Support Unit. 2013 (2011)
[13] Motzer, RJ, et al. *J Clin.Onco*. 2014; 33.13:1430

Acknowledgments

- This study was sponsored by Bristol Myers Squibb

Presented at Virtual ISPOR Europe 2020, November 16-19, 2020
Email: Jessica.May@bms.com
This poster may not be reproduced without permission from the author.