In the Current COVID-19 Reality, Can Independent Injection of Somatostatin Analogues Lead to Greater Health Savings? A Budget Impact Analysis in the UK

Harrow B.[1], Cristeau O.[2], Clay E.[2], Francois C.[3], Frankcom I.[4], Marteau F.[5]

[1]Ipsen, Cambridge, MA USA; [2]Creativ-Ceutical, Paris, France; [3]Aix-Marseille University, Marseille, France; [4]Ipsen, Slough, UK; [5]Ipsen, Boulogne-Billancourt, France

Background

Lanreotide autogel (LAN) and octreotide long-acting release (OCT) are:

- Long-acting somatostatin analogues (LA-SSAs).
- Used for the treatment of acromegaly and gastroenteropancreatic neuroendocrine tumours (GEP-NETs).
- Approved for the treatment of GEP-NETs for LAN or midgut NETs for OCT.[1,2]

Their budget impact in the treatment of these diseases was previously studied considering their differences regarding the following delivery attributes:

- Injection route (deep subcutaneous injection for LAN vs intramuscular injection for OCT);
- Pharmaceutical forms (respectively, pre-filled ready-to-use syringe vs powder + solvent for suspension for injection);
- Mode of administration (respectively, possibility of self-/partner- injection [independent injection] vs injection by a healthcare professional only);
- Real-life dosing intervals.[3]

Previous findings demonstrate potential savings in terms of visits avoided which could be of interest in the Coronavirus Disease 2019 (COVID-19) situation.[4,5]

Following the recommendations of the 'COVID-19 Medicines Guidance for Endocrine System Disorders' published by the UK National Health Service (NHS), delivery modes were reorganised with an increased use of independent injection.[6,7]

Objectives and Methods

Objectives

To assess the specific budget impact of increasing the uptake of independent administration for the treatment of acromegaly and NETs in the UK.

Methods

Model structure

A decision tree was developed considering the following delivery attributes of LAN and OCT:

- Administration time for nurses;
- Risk of needle clogging;
- Proportion of patients by administration type: independent administration; nurse at hospital; or nurse at home.
- Administration schedule.

Model population

- We considered all patients with GEP-NETs treated with SSA in the UK NHS, a population estimated at 3,921 from a NET prevalence rate of 35 per 100,000 individuals, 25% with metastatic non-resectable GEP-NET, of whom approximately 67% are treated with a SSA (calibrated to match current sales).[8-11]
- All patients with acromegaly receiving SSA treatment in the UK NHS were also included. This represents 2,073 patients, estimated from a prevalence of 8.6 per 100,000 individuals, considering 51.2% being treated pharmacologically of whom 70.4% are treated with SSA (calibrated to match current sales).[11-13]

Model inputs

- Model inputs were based on publicly available sources including drug acquisition and administration costs (**Tables 1 and 2**).

Model inputs

- Model inputs were based on publicly available sources including drug acquisition and administration costs (**Tables 1 and 2**).

Comparisons

The model provided overall budget impact and compared three situations (**Table 1**):

- The pre-COVID-19 era (65% market share for LAN and 12.6% of patients independently injecting, 20.0% at hospital);
- The COVID-19 era (65% market share for LAN, 24.5% of patients independently injecting, 8.1% at hospital), assuming that patients newly independently injecting would be previously treated at hospital;
- A hypothetical future COVID-19 scenario (75% market share for LAN), considering that increasing LAN usage is the only way to reduce patients' exposure, and assuming that the proportion of patients independently injecting remains at the same rate for LAN.

Analyses were conducted from the NHS England perspective, considering drug acquisition and administration costs for the whole population. Additional outcomes including the number of nurse visits potentially avoided and nurse time saved, in the interests of clinical commissioning groups (CCGs) and local institutions (hospitals), were also estimated.

Scenario analyses

- A dedicated scenario was designed to understand the potential savings generated when a patient treated with OCT switches to LAN administered via independent injection.

Table 1: Model inputs

	LAN			OCT LAR
Attributes	Pre-COVID-19	COVID-19	Future	
Market shares[14]	65%	65%	75%	
Administration time for nurses[15]	2.5 min			6.2 min
Risk of needle clogging[16]	0.0%			2.6%
Proportion of patients by administration type[17]				
Independent administration	12.6%	24.5%	24.5%	0.0%
Nurse at hospital	20.0%	8.1%	8.1%	30.0%
Nurse at home	67.4%	67.4%	67.4%	70.0%
Costs				
Drug acquisition cost per dose[18,19]	120 mg: £937.00			30 mg: £998.40
Administration cost				
Nurse visit at home	£0.00			£0.00
Nurse visit at hospital[20]	£124.00			£124.00

COVID-19: coronavirus disease 2019; LAN: lanreotide autogel; OCT LAR: octreotide long-acting release.

Table 2: Treatment schedules

Schedule	GEP-NET[21]	Acromegaly[3]
LAN		
120 mg / 4 weeks*	91.0%	88.0%
120 mg / 3 weeks	9.0%	0.0%
120 mg / 5 weeks	0.0%	2.5%
120 mg / 6 weeks	0.0%	4.7%
120 mg / 7 weeks	0.0%	0.2%
120 mg / 8 weeks	0.0%	4.6%
OCT LAR		
30 mg / 4 weeks*	74.0%	100.0%**
30 mg / 3 weeks	26.0%	0.0%

*Normal dosing and schedule for the indication; **Assumption. GEP-NET: gastroenteropancreatic neuroendocrine tumour; LAN: lanreotide autogel; OCT LAR: octreotide long-acting release.

In the Current COVID-19 Reality, Can Independent Injection of Somatostatin Analogues Lead to Greater Health Savings? A Budget Impact Analysis in the UK

Harrow B.[1], Cristeau O.[2], Clay E.[2], Francois C.[3], Frankcom I.[4], Marteau F.[5]

[1]Ipsen, Cambridge, MA USA; [2]Creativ-Ceutical, Paris, France; [3]Aix-Marseille University, Marseille, France; [4]Ipsen, Slough, UK; [5]Ipsen, Boulogne-Billancourt, France

Results

- In the COVID-19 pandemic scenario, the higher proportion of patients independently injecting treatment may have generated savings of £759,760 in administration costs over one year, reducing the annual overall healthcare expenditure from £79.5M to £78.8M (**Figure 1A**). Further, 6,030 hospital visits could have been avoided over a year, as well as 251 hours of hospital nurse time (representing 100 LAN injections) per year (4.2 hours for 100 patients; **Figure 1B**).
- With overall healthcare expenditures of £77.5M, the hypothetical future scenario, with the proportion of patients treated with LAN increased to 75% (increasing the overall proportion of patients independently injecting accordingly), would generate £1.3M of additional overall savings compared to the current COVID-19 situation. The future scenario would result in a budget reduction of £2.1M compared to the pre-COVID-19 situation, representing a 2.6% reduction of the overall budget (**Figure 1A**).
- Compared to the pre-COVID-19 situation, 8,569 nurse contacts could be avoided per year, including 7,863 hospital visits, in the future scenario (**Figure 1C**). Additionally, a total of 487 hours of hospital nurse time could be saved per year (8.1 hours for 100 patients; **Figure 1B**). In this future scenario, 52.7% of the savings resulted from reduction in treatment acquisition costs and 47.3% resulted from reduction in administration costs (**Figure 1D**).
- If a patient usually treated with OCT switches to LAN administered via independent injection, the overall expenses could be cut by 16.4% per patient with GEP-NETs (£2,458 saved per year) and 9.1% for a patient with acromegaly (£1,262 saved per year). In terms of patient exposure in one year, 14.5 nurse contacts, including four hospital visits, could be avoided for each patient with GEP-NETs, while 13.3 nurse contacts could be avoided per patient with acromegaly.

Conclusion

- We demonstrate that the reorganisation of health care for patients with GEP-NET or acromegaly during the first wave of the COVID-19 pandemic may have generated substantial savings in the UK. A shift to home-based, independently administered SSA treatment may also have helped to reduce exposure and to increase autonomy and independence for patients.
- These benefits could be substantially extended with increased uptake of LAN and independent administration. The estimated 8,000 hospital visits avoided in the future scenario could help to ease pressure on the NHS and allow for better allocation of hospital resources.
- Any pandemic-enforced increase may, if adopted long-term, translate into substantial ongoing savings.

Figure 1: Budget impact results

A) Overall healthcare expenditures over a year

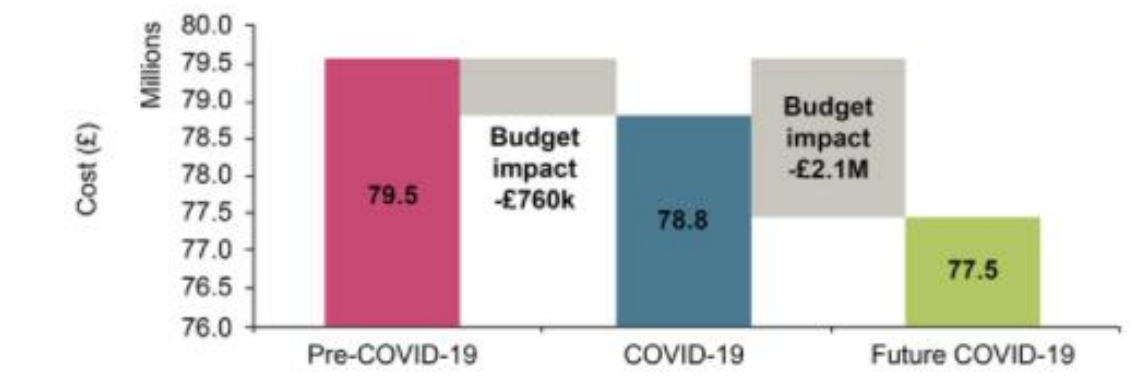
Cost (£)
Millions
80.0
79.5
79.0
78.5
78.0
77.5
77.0
76.5
76.0
79.5
Budget impact -£760k
78.8
Budget impact -£2.1M
77.5
Pre-COVID-19
COVID-19
Future COVID-19

C) Number of nurse contacts (hospital + home)

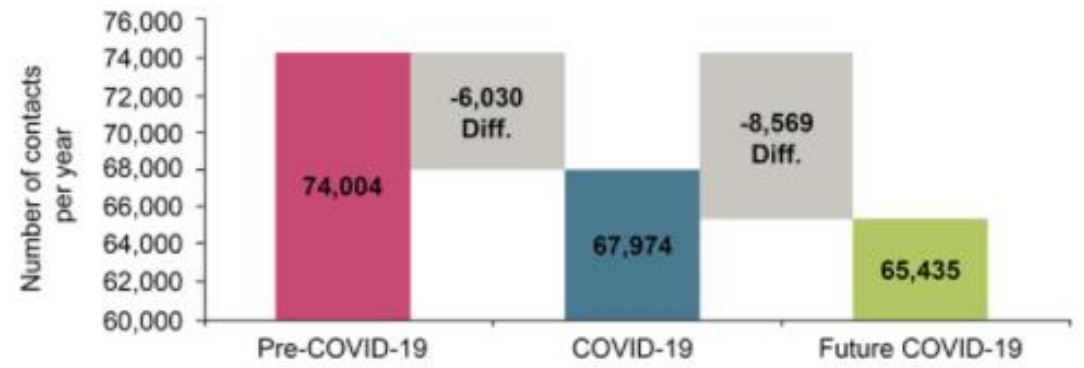
Number of contacts per year
76,000
74,000
72,000
70,000
68,000
66,000
64,000
62,000
60,000
74,004
-6,030 Diff.
67,974
-8,569 Diff.
65,435
Pre-COVID-19
COVID-19
Future COVID-19

COVID-19: coronavirus disease 2019; Diff: difference; LA-SSA: long-acting somatostatin analogue.

B) Nurse time

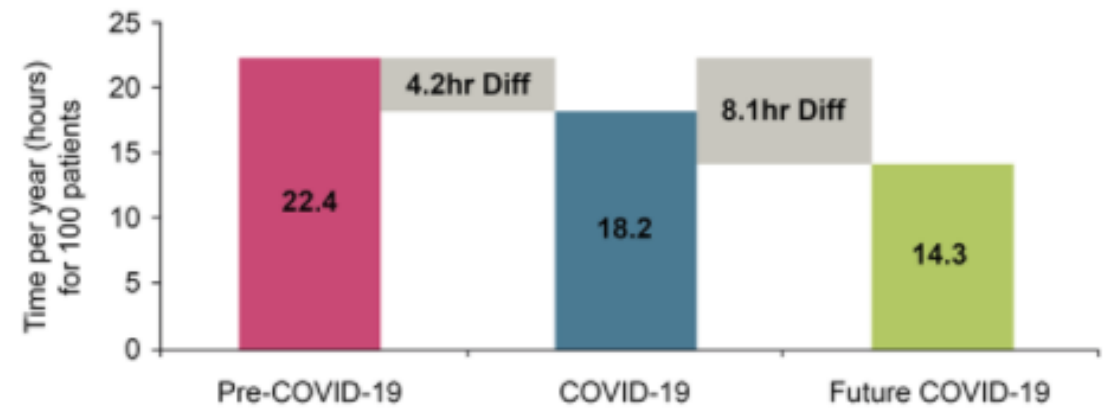
Time per year (hours) for 100 patients
25
20
15
10
5
0
22.4
4.2hr Diff
18.2
8.1hr Diff
14.3
Pre-COVID-19
COVID-19
Future COVID-19

D) Saving components: future versus pre-COVID-19

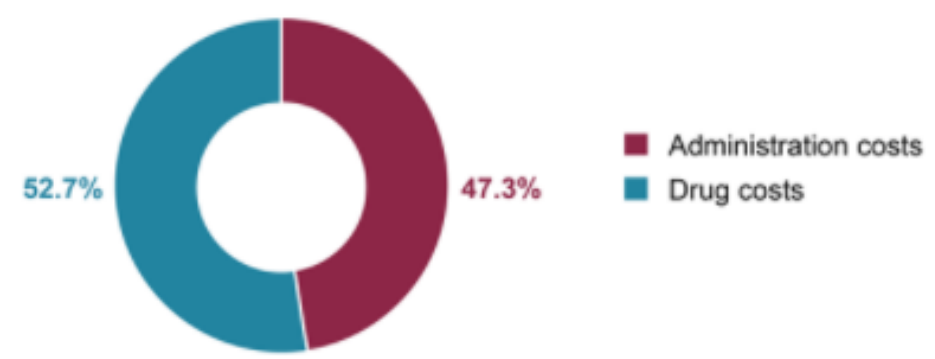
52.7%
47.3%
Administration costs
Drug costs